Establishing Reference Ranges for Lymphocyte Subsets in Healthy Bama Minipigs for Immunotoxicity Assessment

Qinlei Gu, Mingdong Fu, Xiaochen Guo, Jinpeng Li, Xiaoying Jin
WuXi AppTec, 1318 WuZhong Avenue, Suzhou, Jiangsu, China

WuXi AppTec

Background and Purpose

During drug development and safety evaluation, Immunophenotyping serves as a critical assay for identifying immune cell populations affected by test articles. Characterization of peripheral whole blood matrix in nonclinical studies is especially informative, offering potentially translatable biomarkers for clinical studies.

Minipigs are emerging as one alternative non-rodent animal model in regulated toxicology studies due to their anatomical and physiological similarities to human features, relevant in metabolism, dermatology, cardiovascular research, and pediatric medicine. Among popular minipig breeds in laboratory, despite their long history of domestication and utilization, Bama minipig remains poorly characterized in term of the baseline immune cell populations.

In this study, we surveyed Bama minipig breed for immune cell populations at baseline, in order to establish a reliable reference range for lymphocyte population. Comparison between Bama minipig breed and published data of popular Gottingen minipig breed and common livestock, the Large White swine breed also reveals possible immune differences to other porcine species.

Materials and Methods

Animals

A total of 91 healthy adult Bama Minipig, 46 males and 45 females, originated from China were used in this study. All animals were naïve and maintained in compliance with the Guide for the Care and Use of Laboratory Animals. The mean age is 4.65±0.4 years, and the mean body weight ranged 12.3±1.49 kg. All animal research was approved by the Institutional Animal Care and Use Committee in WuXi AppTec.

Immunophenotyping

Blood samples were collected with anti-coagulant potassium EDTA and washed with 2 mL of DPBS by centrifugation for 5 min at 400 g. Red blood cells were then lysed with BD Pharm Lyse lysing solution. Live/dead was stained with Fixable Viability Stain 510 for 10 -15 minutes. Antibody cocktail was then added, which includes: FITC anti-pig CD45, PE-Cy™7 anti-pig CD3ε, Alexa Fluor® 647 anti-pig CD16, and PE anti-pig CD79a. Cells were incubated with antibody cocktail at room temperature for 30 minutes protected from white light. Cells were then washed and re-suspended with DPBS and analyzed using BD LSRFortessa™ flow cytometer with FACS Diva software.

Statistics

Two-sample t test was to calculated p-value for normally distributed data, and Mann-Whitney test was used for datasets that are not normal.

References

1. Tina Rubic-Schneider, Brigitte Christen, Dominique Brees, Michael Kammüller. Minipigs in Translational Immunosafety Sciences: A Perspective. 8-color whole blood immunophenotyping panel for the character. 2016. https://journals.sagepub.com/doi/full/10.1177/0192623315621628
2. Lunney JK, Van Goor A, Walker KE, Hailstock T, Franklin J, Dai C. Importance of the pig as a human biomedical model. Science Translational Medicine. 2021 Nov 24;13(621):eabd5758.
3. OMIP-107: characterization and quantification of lymphocyte subsets and monocytes in swine - Arrigucci - 2024 - Cytometry Part A. https://onlinelibrary.wiley.com/doi/full/10.1002/cyto.a.
4. Pernold CPS, Lagumdzic E, Stadler M, Mair KH, Jäckel S, Schmitt MW, et al. Characterization of the immune system of Ellegaard Göttingen Minipigs - An important large animal model in experimental medicine. Front Immunol. 2022 Sep 20;13:1003986.
5. Sipos W, Duvigneau CJ, Hartl RT, Schwendenwein I. Exploratory reference intervals on hematology and cellular immune system of multiparous Large White sows. Vet Immunol Immunopathol. 2011 Jun 15;141(3–4):307–11.

Results

Lymphocyte immunophenotyping was conducted in peripheral blood collected from 46 untreated male and 45 untreated female Bama minipigs. The baseline percentage of lymphocyte subpopulations was reported as total T cell (CD45+CD3ε+), B cell (CD45+CD3- CD16-CD79a+), and natural killer (NK) cell (CD45+CD3-CD16+CD79a-). To assess the distribution of the data, a variety of parameters were calculated and presented in **Table 1**, including the mean, standard deviation, minimum, 5th percentile, 25th percentile, median, 75th percentile, 95th percentile, and maximum. In summary, the 90% reference range (from 5th to 95th percentile) for lymphocyte subset proportion was as follows: total T cells range from 79.4% to 94.1% in male and from 79.2% to 97.2% in female; B cells range from 2.4% to 11.5% in male and from 1.6% to 12.8% in female; NK cells range from 2.7% to 12.4% in male and from 1.0% to 9.4% in female. In addition, a difference between male and female minipig was observed in NK cell baseline percentage (p-value=0.0047), with a higher NK proportion in male minipigs.

Table 1 Reference range for the percentage of lymphocyte subpopulations in Bama minipigs

Percentage of Lymphocyte Subpopulation						
	% T cells		% B cells		% NK cells	
Parameter	Female (n = 45)	Male (n = 46)	Female (n = 45)	Male (n = 46)	Female (n = 45)	Male (n = 46)
Mean	87.8	87.7	5.9	4.8	5.1	6.6
Standard Deviation	6.9	4.1	4.2	2.6	2.2	2.6
Minimum	50.9	74.5	1.3	1.1	0.8	1.9
5th Percentile	79.2	79.4	1.6	2.4	1.0	2.7
25th Percentile	85.3	85.2	3.2	3.2	3.9	4.5
50th Percentile	89.2	88.1	4.9	4.2	4.9	6.3
75th Percentile	91.3	90.7	7.7	5.2	6.2	8.0
95th Percentile	97.2	94.1	12.8	11.5	9.4	12.5
Maximum	97.6	95.0	25.5	15.8	10.9	13.9
Reference Range (90% Confidence Interval)	79.2 -97.2	79.4 -94.1	1.6 -12.8	2.4 -11.5	1.0 -9.4	2.7 -12.5

Figure 1 Percentage of lymphocyte subpopulations in Bama minipigs versus Ellegaard Göttingen Minipigs and Large White breed. Bar graph of A CD3 T cell, B B cell and C NK cell percentages for male and female animals.| Data of EGM breed and Large White breed were cited from published studies[4-5].

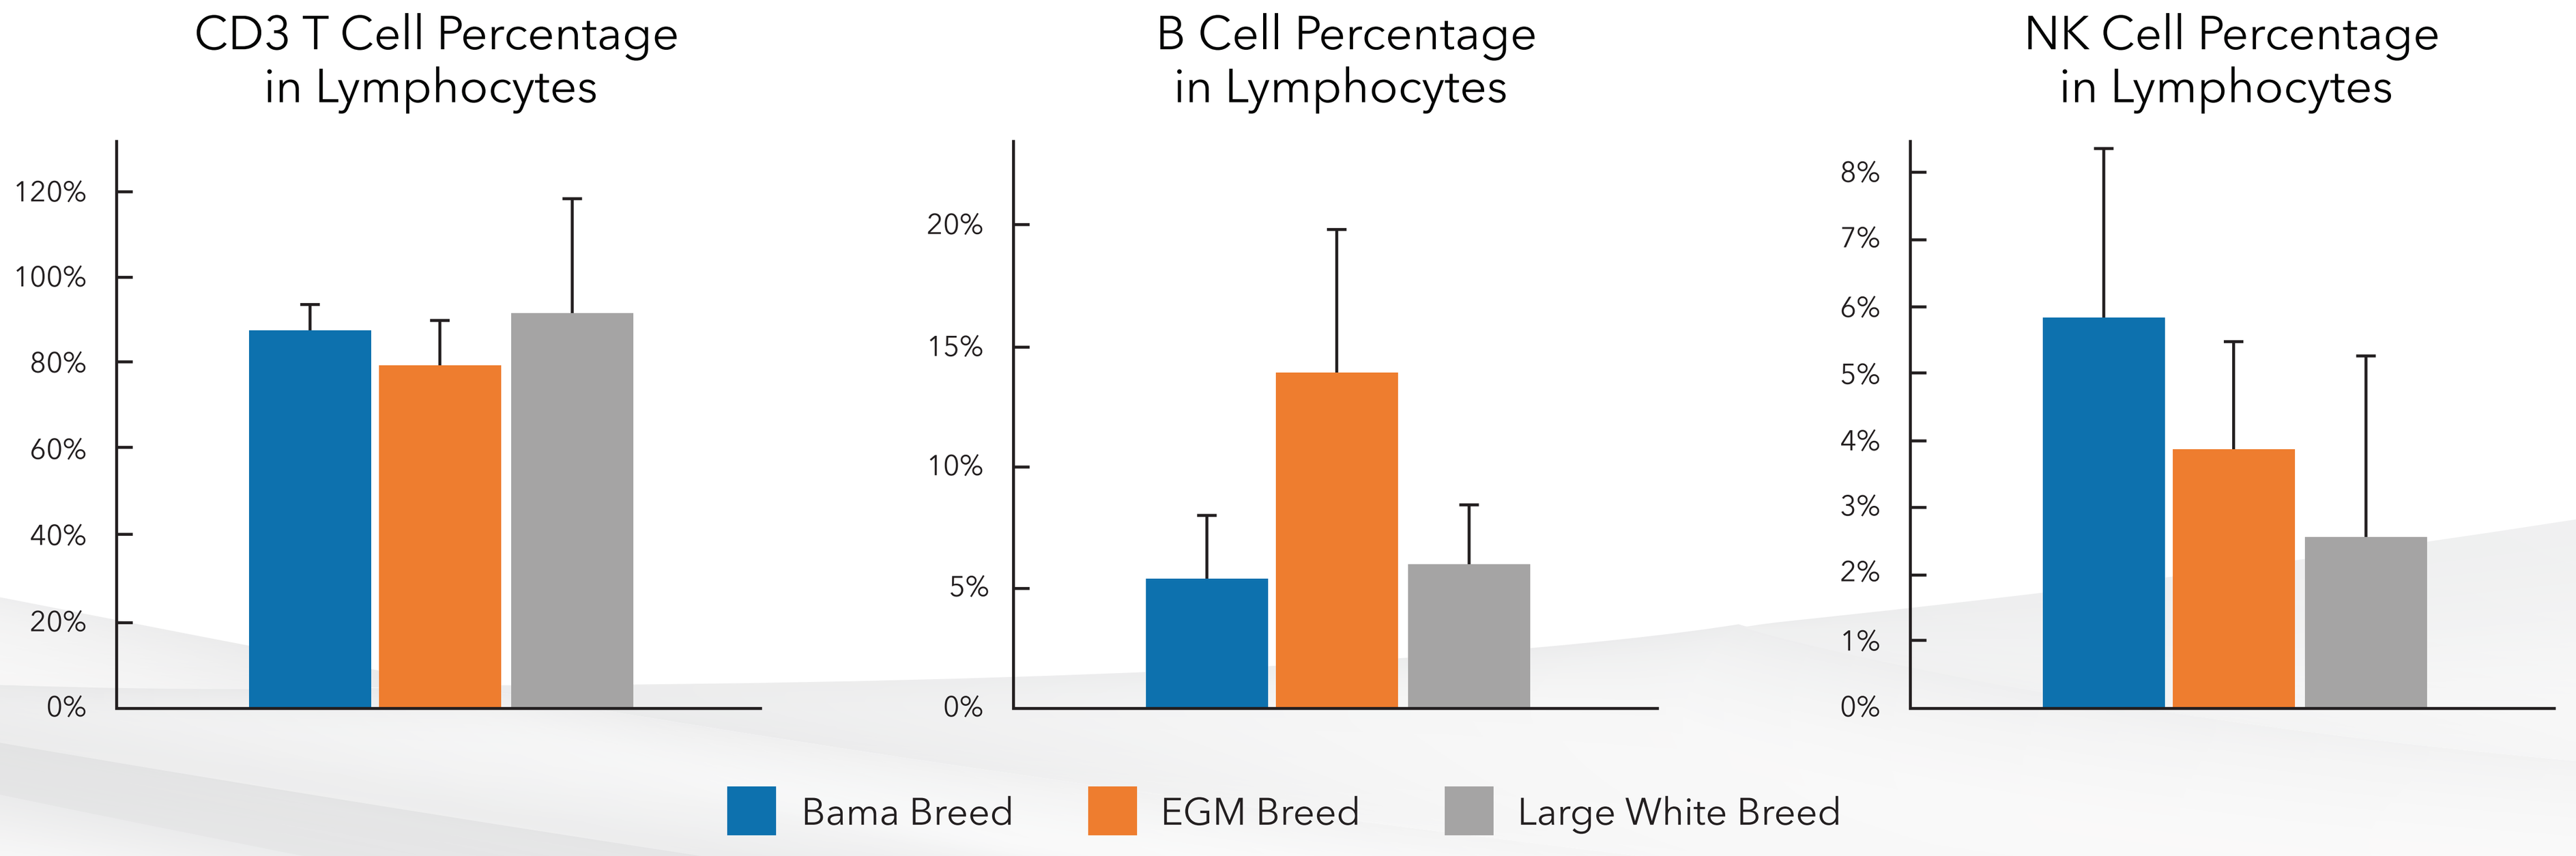
CD3 T Cell Percentage in Lymphocytes
B Cell Percentage in Lymphocytes
NK Cell Percentage in Lymphocytes
120%
100%
80%
60%
40%
20%
0%
20%
15%
10%
5%
0%
8%
7%
6%
5%
4%
3%
2%
1%
0%
Bama Breed
EGM Breed
Large White Breed

Figure 2 Percentage of lymphocyte subpopulations in male and female Bama mini pigs. Histogram and density plot of **A** T cell, **B** B cell and **C** NK cell percentages and violin plots for **D** females and **E** males are presented. Dotted lines represent 25th, 50th, and 75th percentile.

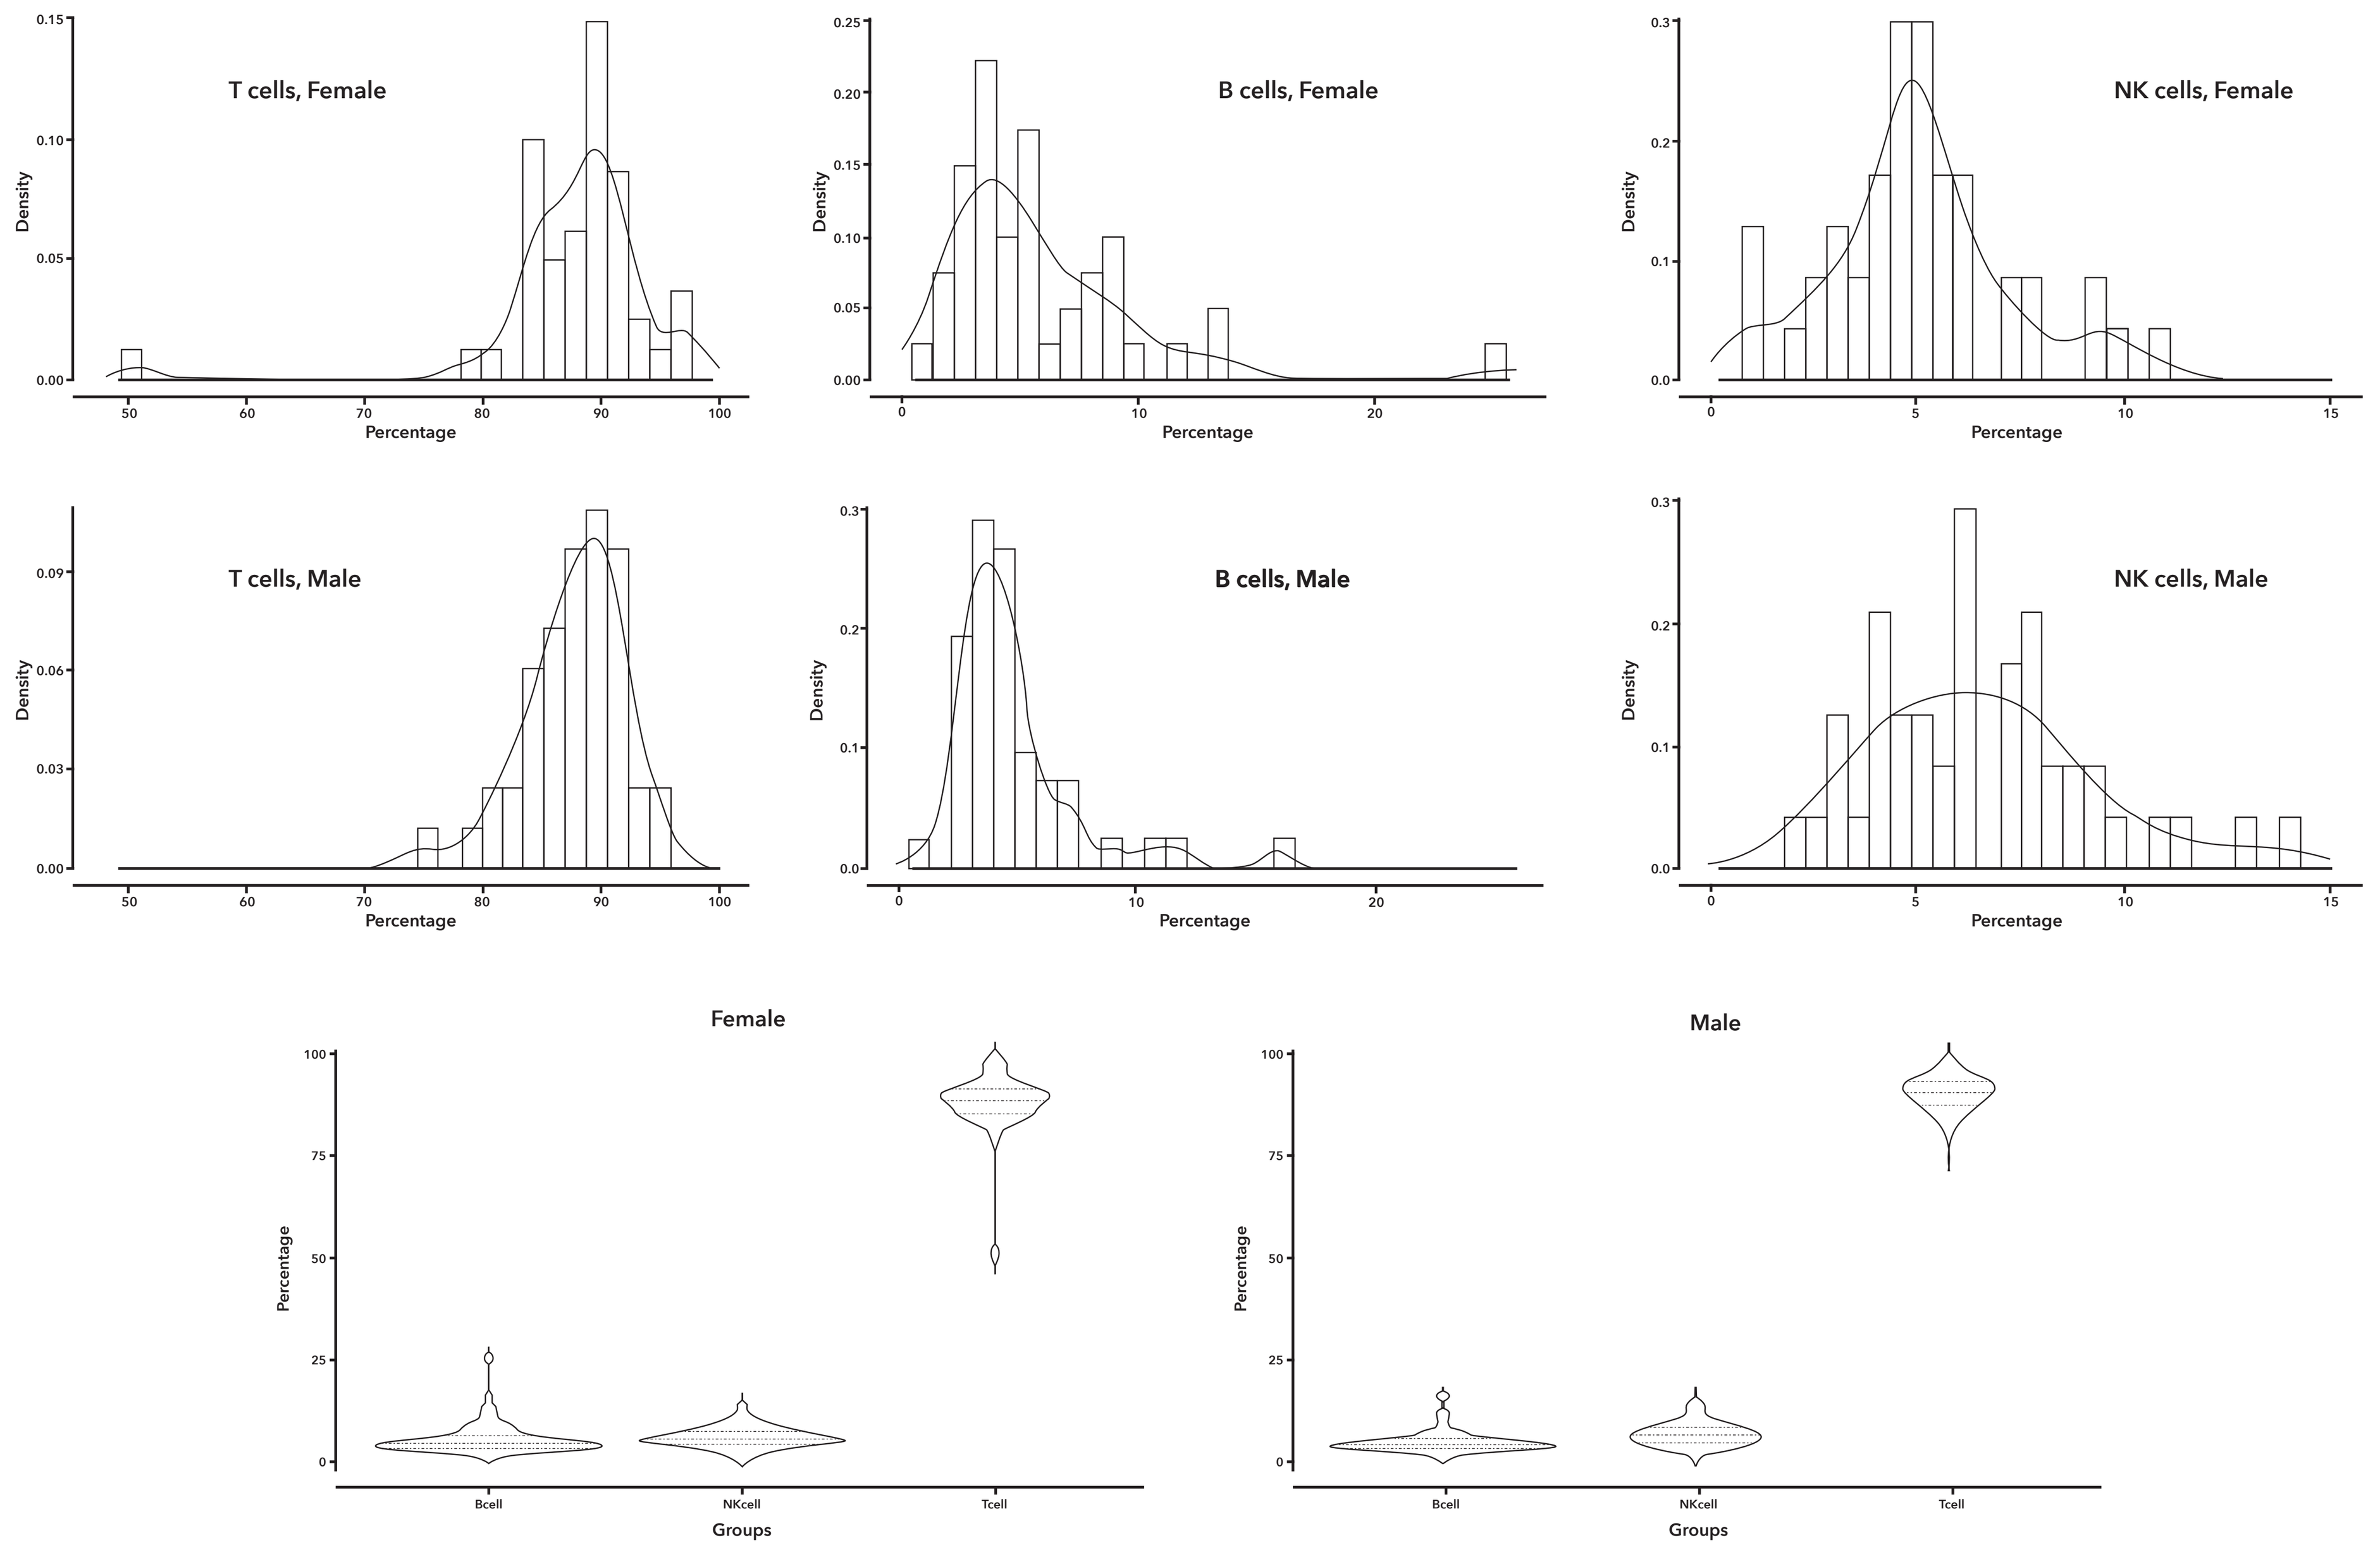
T cells, Female
B cells, Female
NK cells, Female
T cells, Male
B cells, Male
NK cells, Male
Density
Percentage
Female
Male
Bcell
NKcell
Tcell
Groups

When compared with published data from the widely used Göttingen minipig breed (Pernold et al., 2022) and the common livestock breed, the Large White (Sipos et al., 2011) in **Figure 1**, Bama minipigs exhibited a TBNK profile more closely aligned with the Large White than the Göttingen minipig breed for T and B cells, with slightly higher mean NK cell percentage than the other breeds.

Histograms and density lines in **Figure 2** showed that T cell percentage from Bama mini pigs demonstrateda right-heavy tail distribution, and the B cell percentage showed left-heavy tail distribution. Both T cell and B cell parameters appears not following normal (Gaussian) distribution, whereases NK cell percentage distributed normally. Histograms for both male and female Bama mini pigs are included, and their distributions for each parameter were similar.

Conclusion

Based on lymphocyte immunophenotyping data from 91 individual animals, a reference range for total T cells, B cells and NK cells in minipigs at homeostatic immune state has been established. This database could serve as a reliable reference, aiding the interpretation of preclinical immunotoxicity findings.

Scan for a copy