Establishing the Internal hERG Safety Margin Threshold Using a Series of Reference Drugs to Support the Integrated QTc Risk Assessment

Yuewen Cao, Rongfei Wu, Liwen Gao, Millie Chen
WuXi AppTec, 1318 Wuzhong Avenue, Suzhou, Jiangsu, China

Background

As ICH E14/S7B Q&As was adopted in February 2022[1], the pharmaceutical industry paid more attention to best practice considerations and hERG safety margin threshold. ICH E14/S7B Q&As require to use a hERG safety margin threshold to define negative hERG assays. Given that inter-laboratory variability in hERG IC50 persists even after implementing standardized protocols and adhering to ICH S7B Q&A best practices, it is wiser for each laboratory to establish its own hERG safety margin threshold rather than relying on a published one.

Method

Cell line: hERG-transfected CHO cells

Reference drugs: Cisapride, Dofetilide, Moxifloxacin, and Ondansetron

Electrophysiology:

- Voltage protocol: FDA recommended voltage protocol command (stimulation frequency: 0.2 Hz)

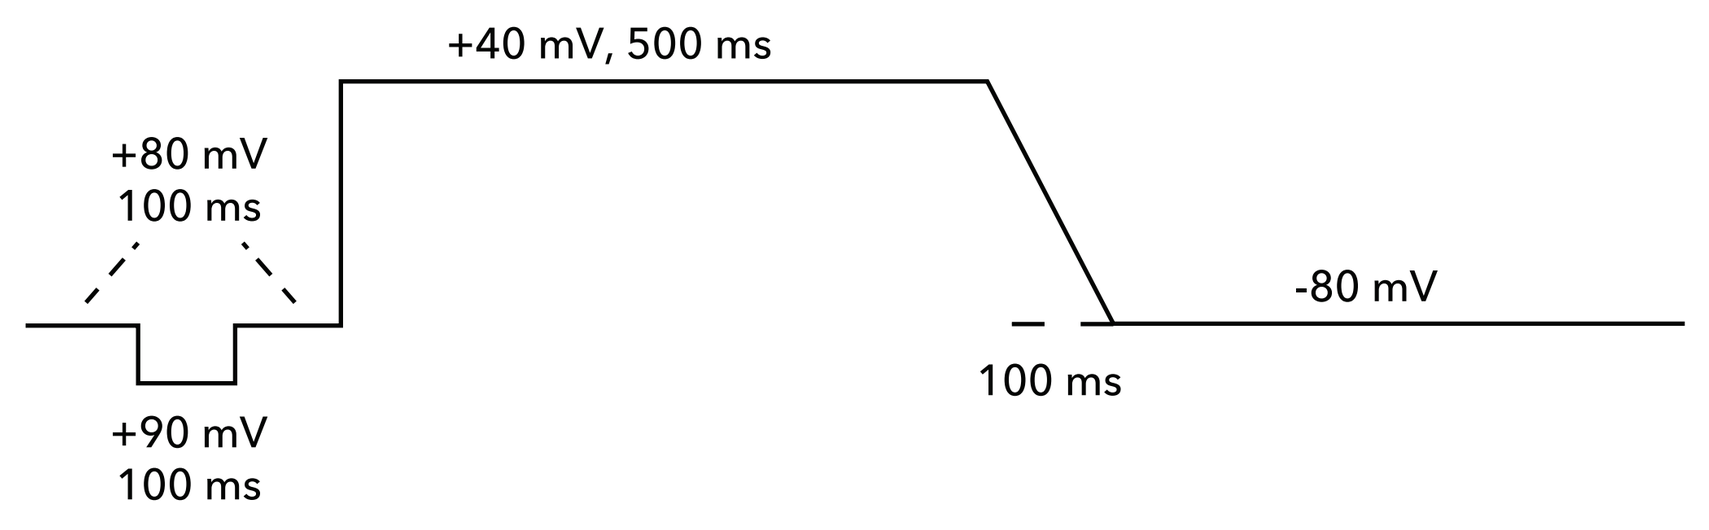
+40 mV, 500 ms
+80 mV
100 ms
-80 mV
100 ms
+90 mV
100 ms

Voltage protocol for hERG testing

- Recording temperature: near physiological temperature (35–37°C)
- Platforms: two laboratories both using manual patch-clamp system

Full blocker: 1 μM E-4031

Statistical analysis methodology[2,5]

- The analyses utilized 13 hERG IC50 values for the four selected reference drugs. Data from across drugs, laboratories, and repeated experiments were pooled using a random-effect meta analysis using the metafor package in R software.
- Log-normal distributions were assumed for both plasma concentration and hERG IC50.
- Using the mean and standard deviation to create large random data distributions.
- The margin is calculated by

$$\text{Margin} = \frac{\text{hERG } IC_{50}}{\text{Concentration} \times \text{Unbound Fraction}}$$

where Unbound Fraction=1-Plasma Protein Binding Fraction.

References

1. ICH E14/S7B Questions and Answers: Clinical and Nonclinical Evaluation of QT/QTc Interval Prolongation and Proarrhythmic Potential, 21 Feb 2022.
2. Leishman, Derek J et al. "Supporting an integrated QTc risk assessment using the hERG margin distributions for three positive control agents derived from multiple laboratories and on multiple occasions."
3. Pfeiffer, Emily R et al. "Specific prediction of clinical QT prolongation by kinetic image cytometry in human stem cell derived cardiomyocytes."
4. Meuldermans, W et al. "Excretion and biotransformation of cisapride in dogs and humans after oral administration."
5. ICH E14/S7B Q&As Training Material Examples Supplemental File.

Results

The four reference drugs were conducted for 2 to 5 times (Cisapride: 2; Dofetilide: 3; Moxifloxacin: 5; and Ondansetron: 3) in accordance with best practices in two laboratories. The typical current trace for the four reference drugs were presented in Figure 1, reflected the significant dose-dependent response. The inhibition of selective blocker (E-4031)was sufficiently high, that can indirectly reflect cell quality. The concentration-response curves for the four reference drugs were presented in Figure 2. The IC50 value and hill coefficient was reported as mean ±95% confidence interval to evaluate the uncertainty estimation. hERG safety margin threshold defined by reference drugs were presented in Table 1, including the critical concentration, Protein Binding, molecular weight, IC_{50} Distribution; shown as shown as 50th (2.5th, 97.5th) percentile.

Figure 1 Typical hERG current trace for the four reference drugs. A to D represents the following drugs: A for Cisapride, B for Dofetilide, C for Moxifloxacin, and D for Ondansetron

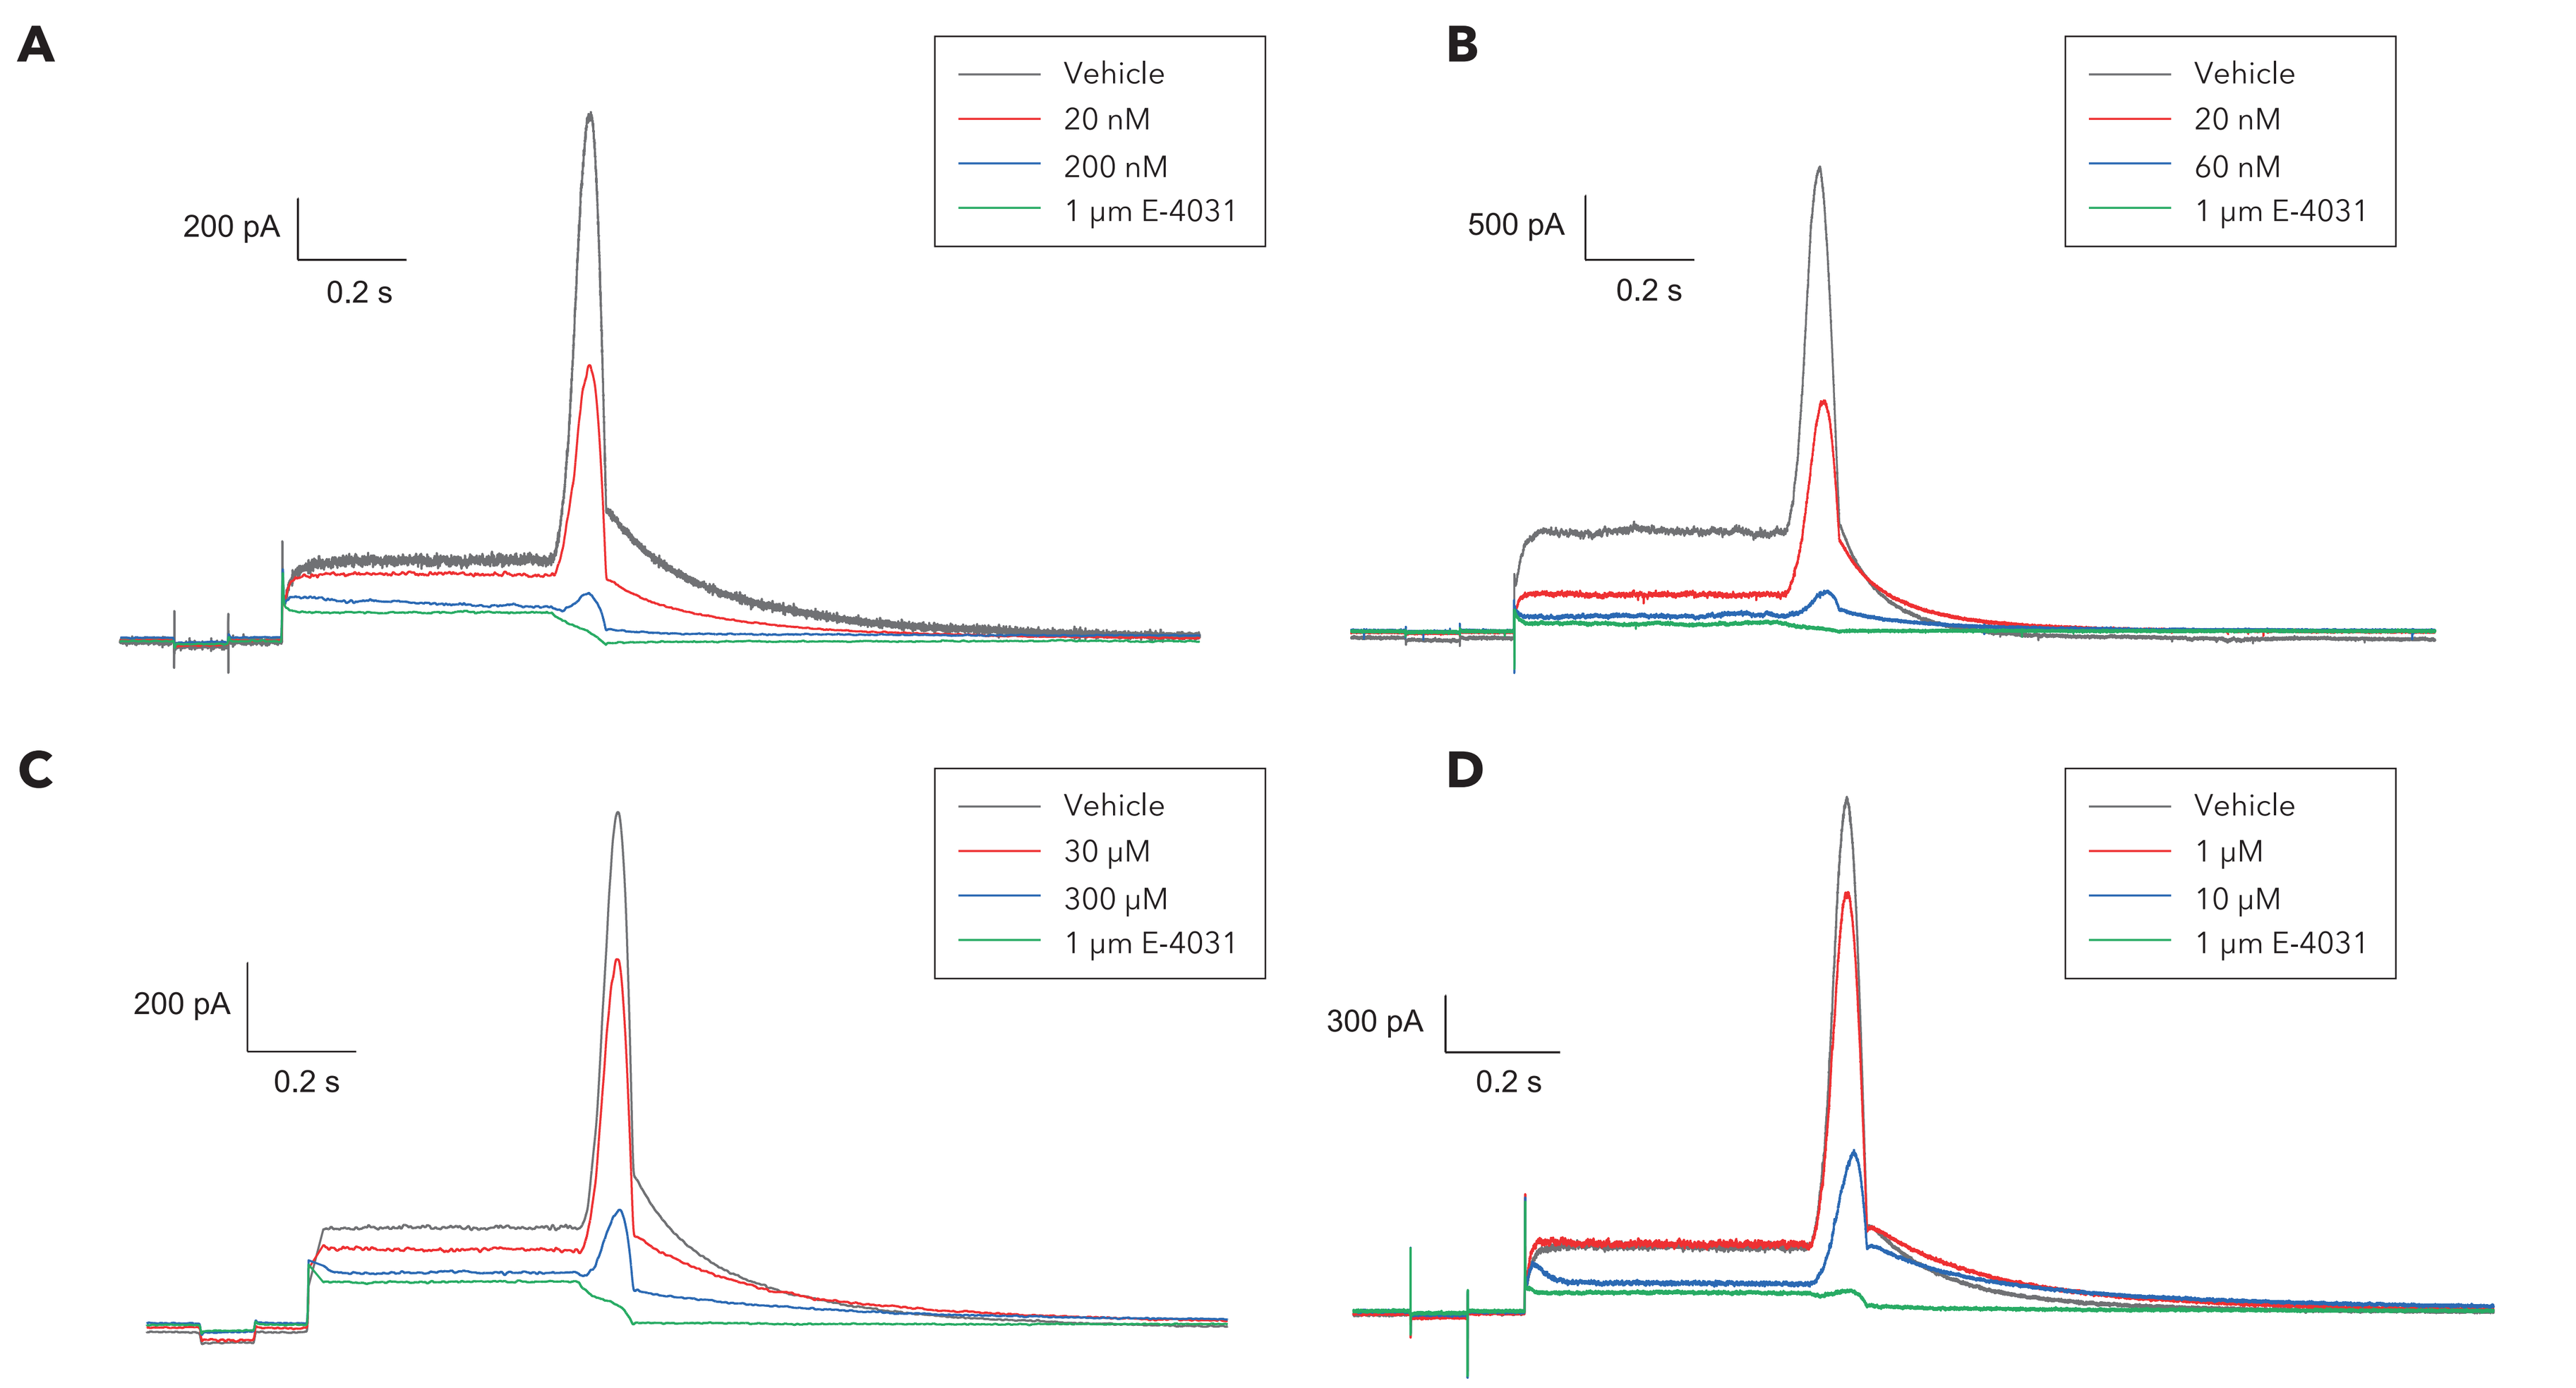
A
Vehicle
20 nM
200 nM
1 μm E-4031
200 pA
0.2 s
B
Vehicle
20 nM
60 nM
1 μm E-4031
500 pA
0.2 s
C
Vehicle
30 μM
300 μM
1 μm E-4031
200 pA
0.2 s
D
Vehicle
1 μM
10 μM
1 μm E-4031
300 pA
0.2 s

Figure 2 Concentration-response relationships for the four reference drugs. A to D represents the following drugs: A for Cisapride, B for Dofetilide, C for Moxifloxacin, and D for Ondansetron.

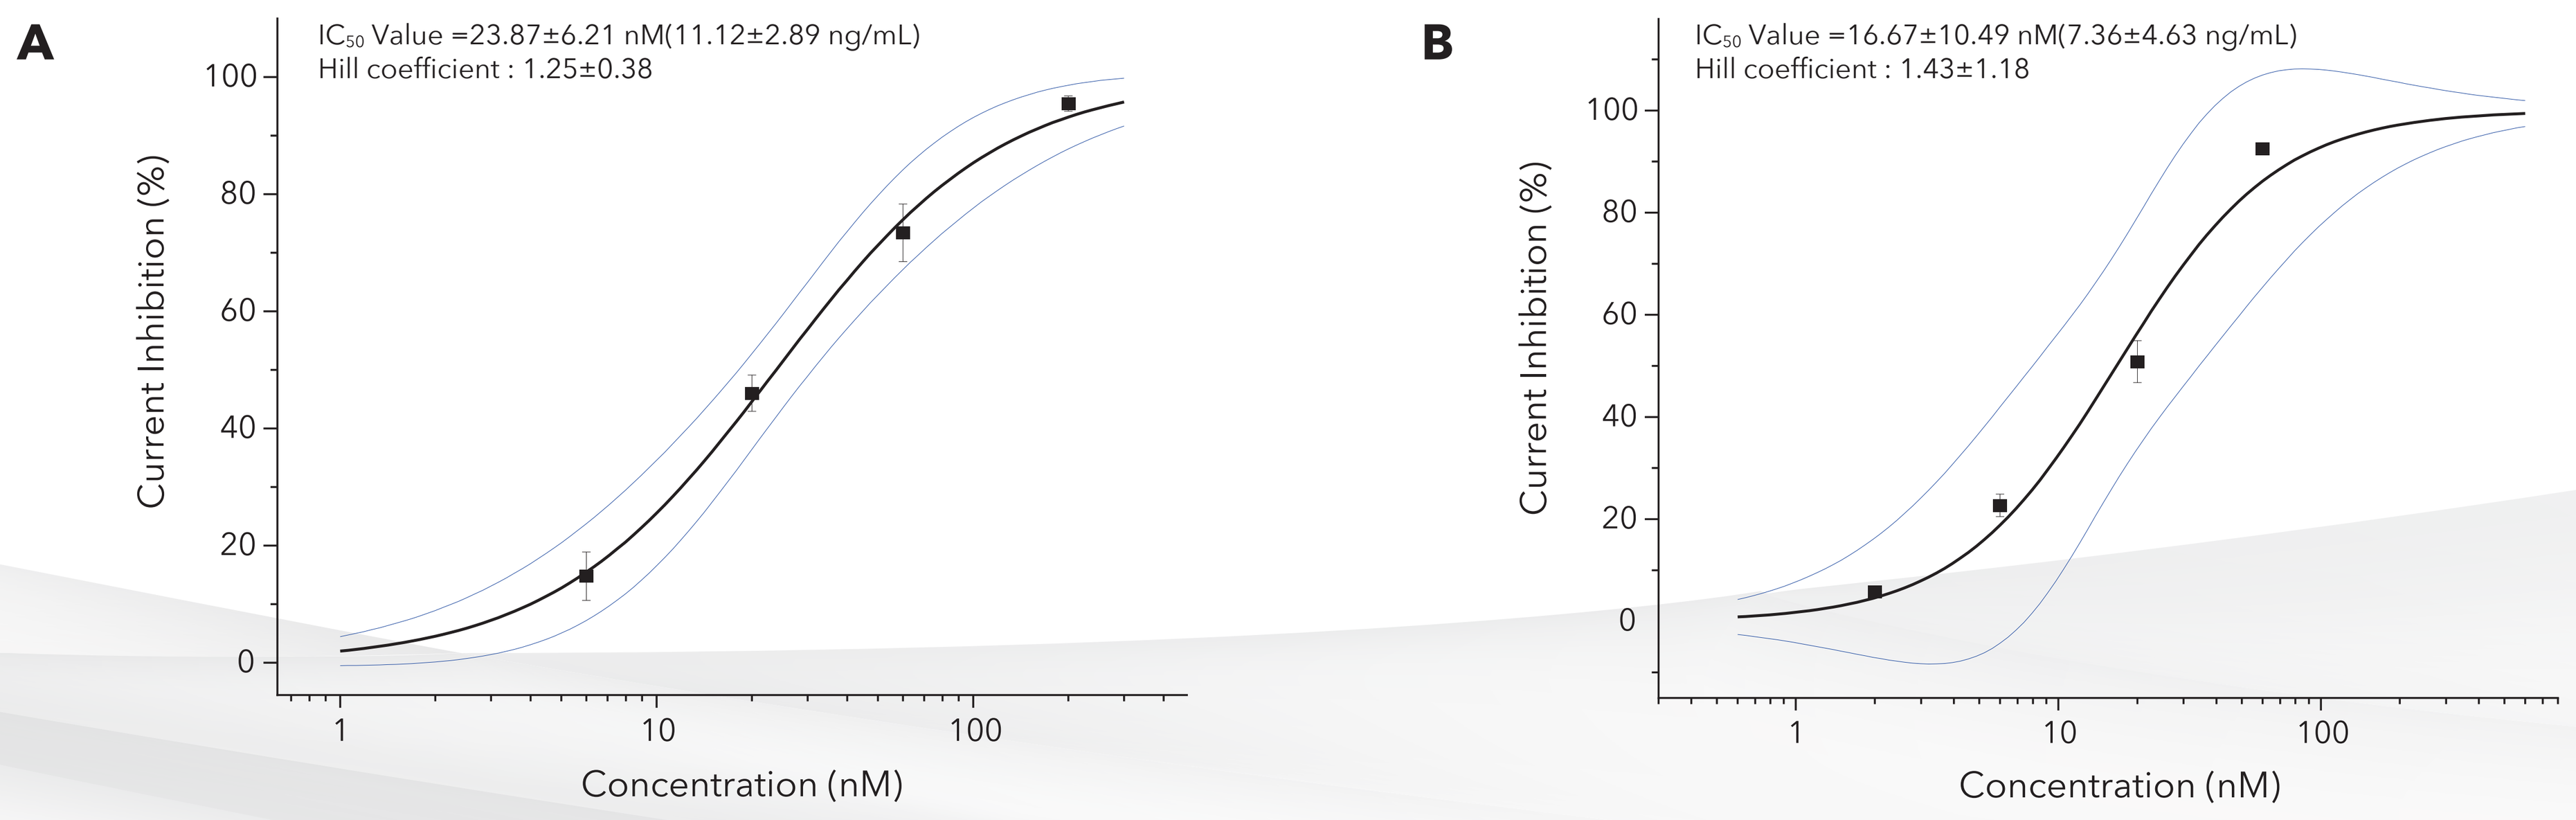
A
IC50 Value =23.87±6.21 nM(11.12±2.89 ng/mL)
Hill coefficient : 1.25±0.38
Current Inhibition (%)
Concentration (nM)
B
IC50 Value =16.67±10.49 nM(7.36±4.63 ng/mL)
Hill coefficient : 1.43±1.18
Current Inhibition (%)
Concentration (nM)

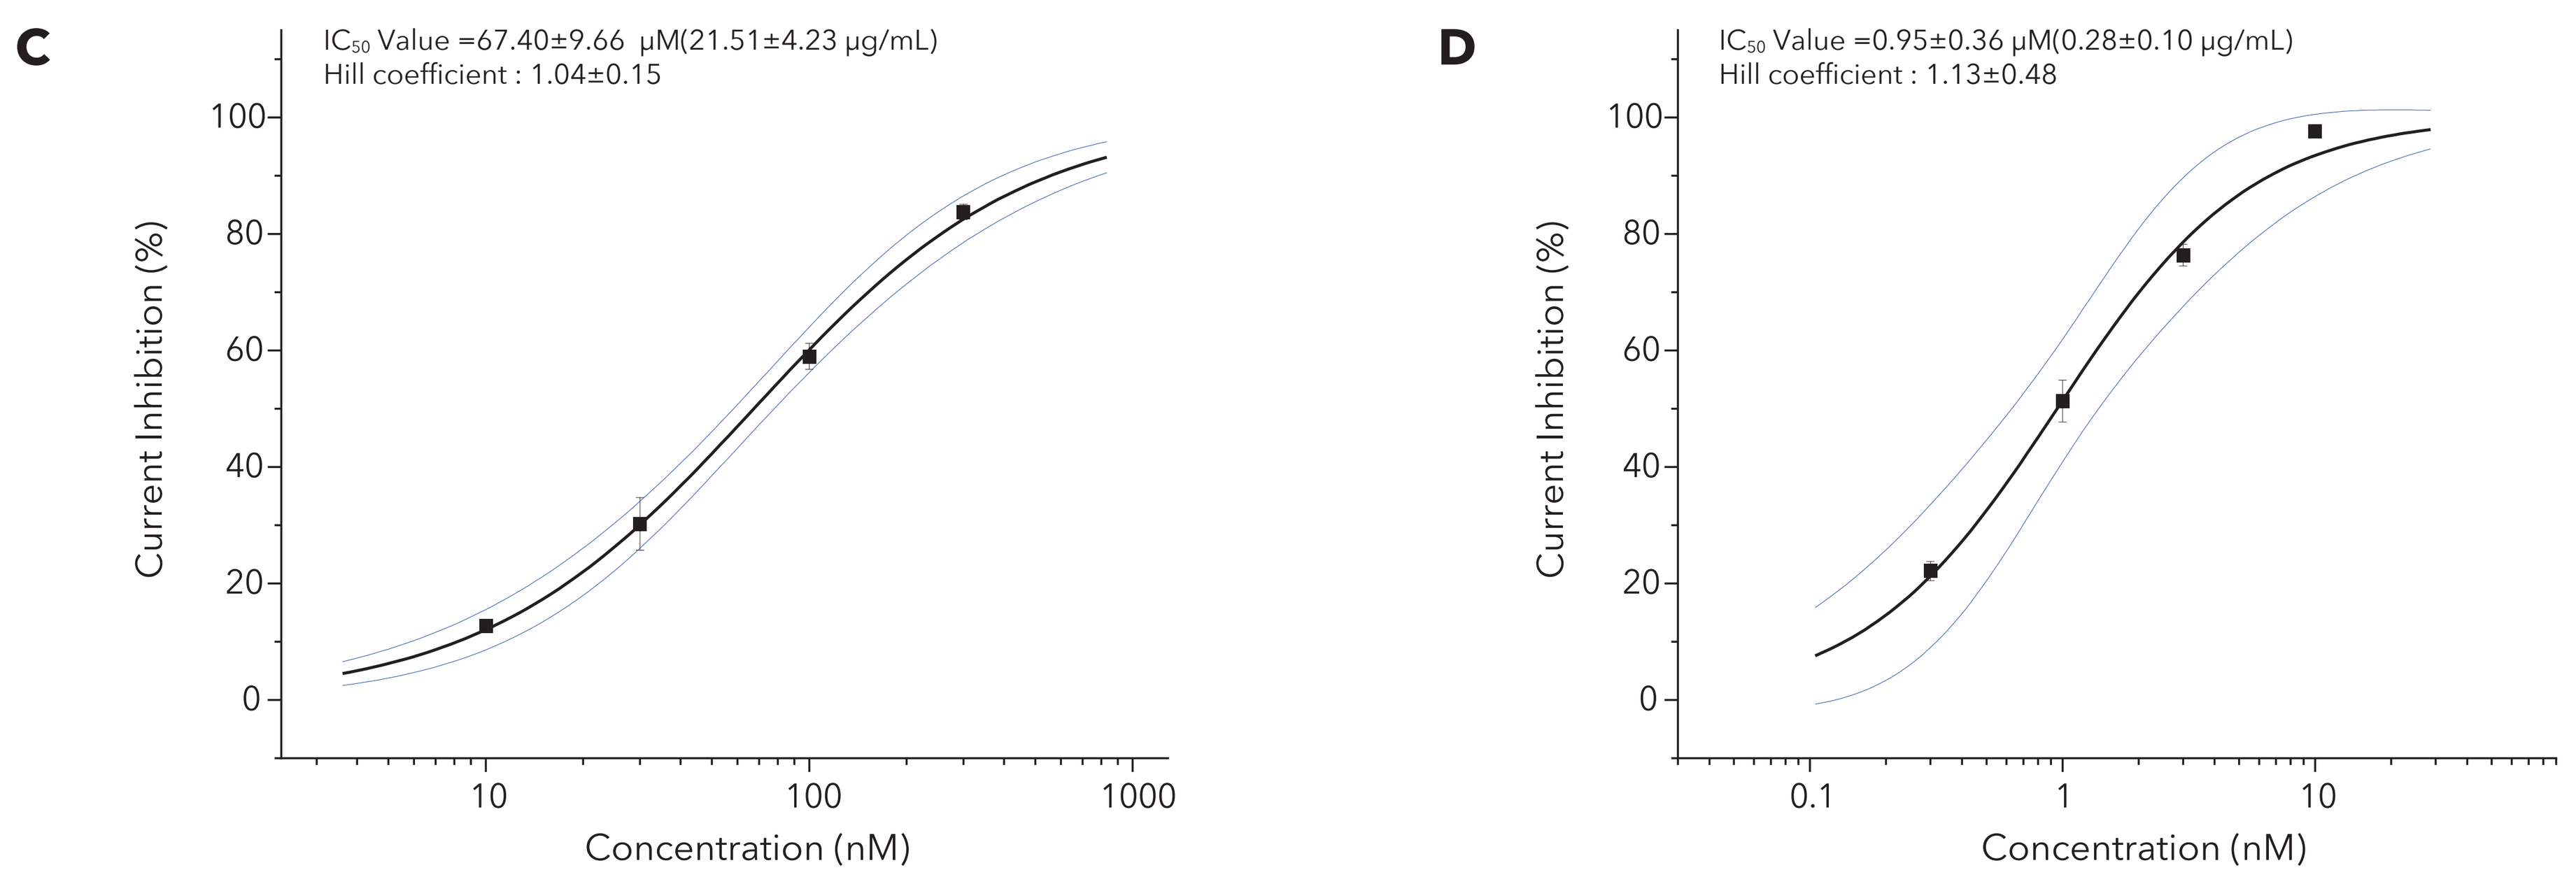
C
IC50 Value =67.40±9.66 μM(21.51±4.23 μg/mL)
Hill coefficient : 1.04±0.15
Current Inhibition (%)
Concentration (nM)
D
IC50 Value =0.95±0.36 μM(0.28±0.10 μg/mL)
Hill coefficient : 1.13±0.48
Current Inhibition (%)
Concentration (nM)

Table 1 hERG safety margin threshold defined by reference drugs

Reference Drugs	Critical Concentration (ng/mL)	Protein Binding, %	Mol Wt (g/mol)	IC_{50} Distribution (μM)	Safety Margin
Cisapride	135 (117, 155)[3]	97.5 (97.1, 97.9)[4]	466	0.02659 (0.02395, 0.02951) N=2	3.681 (2.910, 4.656)
Dofetilide	0.37 (0.24, 0.55)	64 (62, 66)	442	0.02562 (0.02383, 0.02756) N=3	84.79 (55.43, 129.7)
Moxifloxacin	1866 (1591, 2188)	40 (37, 43)	401	62.40 (55.56, 70.0958) N=5	22.31 (18.23, 27.31)
Ondansetron	249 (152, 412)	73 (71,76)	293	1.312 (0.9743, 1.767) N=3	5.696 (3.171, 10.23)
Pooled Safety Margin for Reference Drugs					14.11 (3.507, 56.78)
Threshold					>56.78

The validated outcomes, including simulated denominator exposure distributions and estimated margin distributions, were consistent with the ICH E14/S7B example, demonstrating the reliability of the methodology.

The mean and 95% confidence interval of the pooled hERG safety margin, as defined by reference drugs in our laboratories, are 14.11 (3.507, 56.78).

Conclusion

The IC_{50} values and variability were estimated using the best practice hERG protocol adhering ICH E14/S7B Q&As.

The threshold of approximately 57 (the upper bond of 95% confidence interval) in our laboratories is approaching the threshold of 51 proposed by the ICH S7B Q&A example, suggesting that a ~50-fold margin could offer sufficient discriminatory performance in an integrated risk assessment to identify drugs with low or no QTc liability.

Scan for a copy