Investigation of Embryo-Fetal Development Toxicity of Cyclophosphamide in Mice

Shuijin Yang, Jing Yan, Lei Guo, Lei Pan, Sue McPherson, Xiaoying Jin
WuXi AppTec, Suzhou 215104, PR China

WuXi AppTec

Poster #: P203

Introduction

Cyclophosphamide, one of the anti-cancer drugs, that is widely used as the positive control for development toxicity studies. The study was performed to investigate the embryo-fetal developmental toxicity of cyclophosphamide following ICH S5 (R3).

Experimental Procedures

One hundred mated female mice were randomly assigned to 4 groups of 25/group, and were administered cyclophosphamide at dosages of 0, 5, 10, or 20 mg/kg/day by intra-peritoneal injection from GD 11 to 13. The mated females were c-sectioned on GD 18.

Evaluation

Parameters	Feed Study
Mortality	✓
Clinical Observation	✓
Body Weight	✓
Food Consumption	✓
Macroscopic Observation	✓
Litter Data	✓
Fetal External, Visceral, and Skeletal Examinations	✓

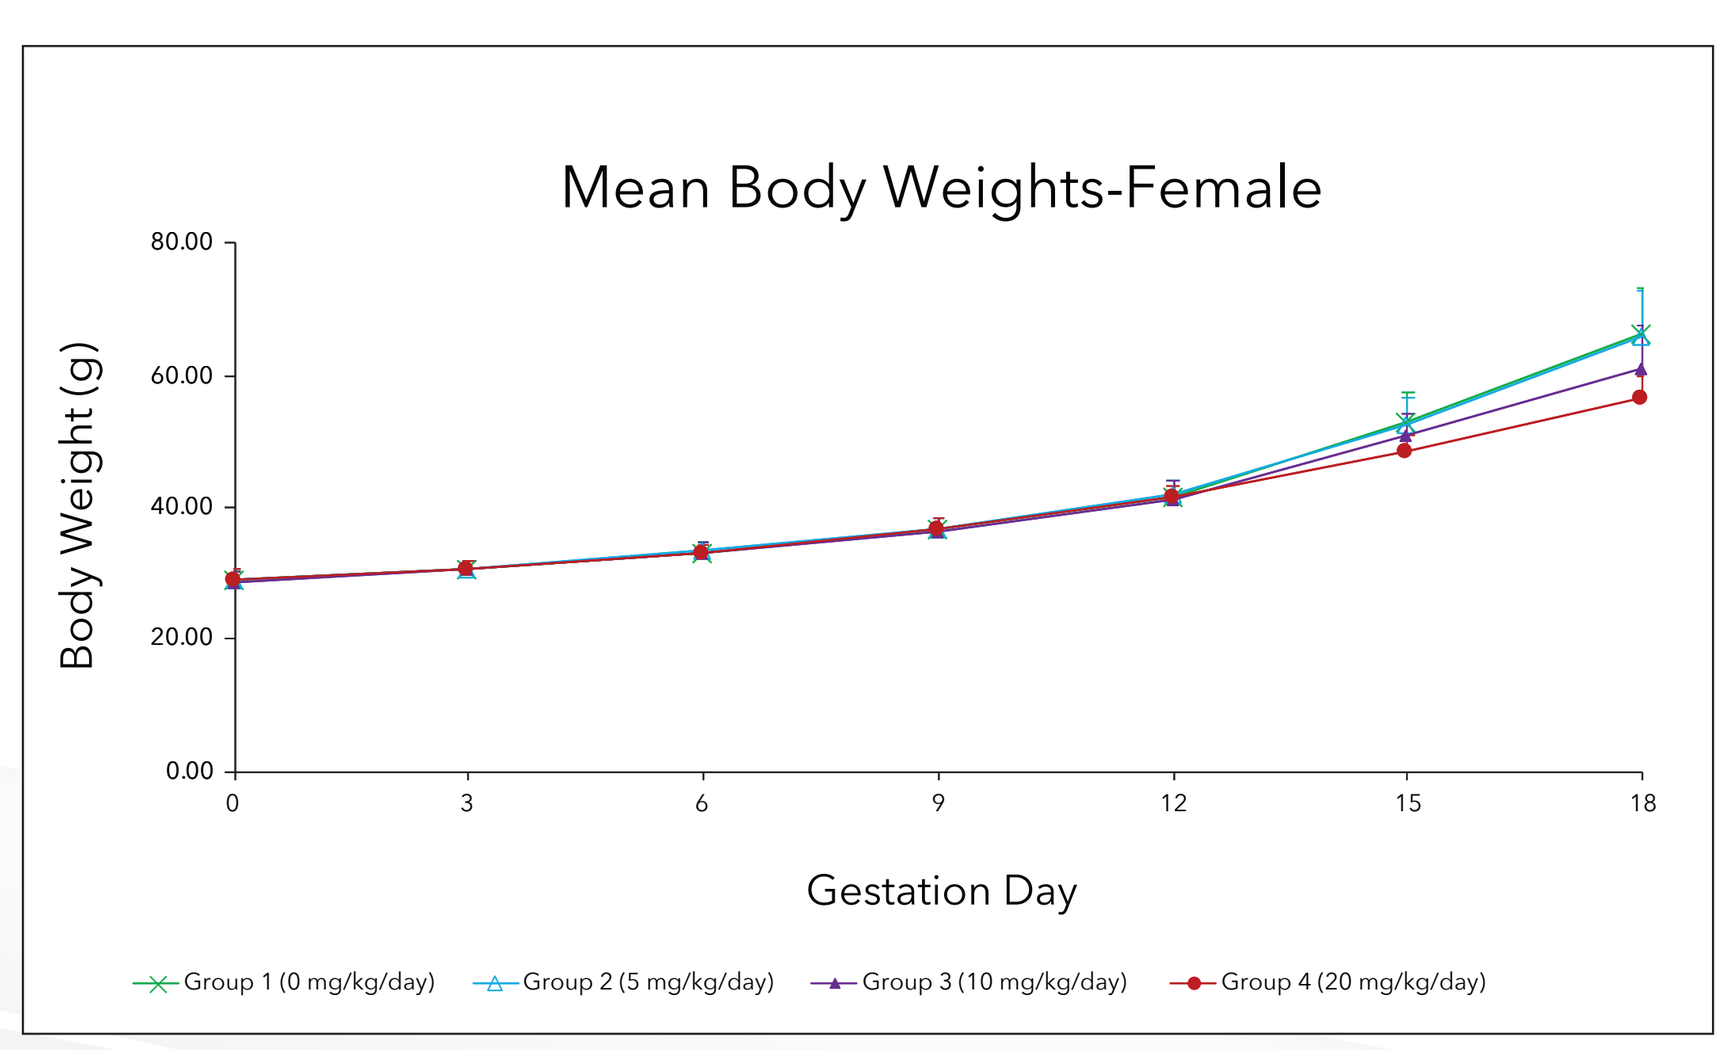
Mean Body Weights-Female
Body Weight (g)
Gestation Day
Group 1 (0 mg/kg/day)
Group 2 (5 mg/kg/day)
Group 3 (10 mg/kg/day)
Group 4 (20 mg/kg/day)

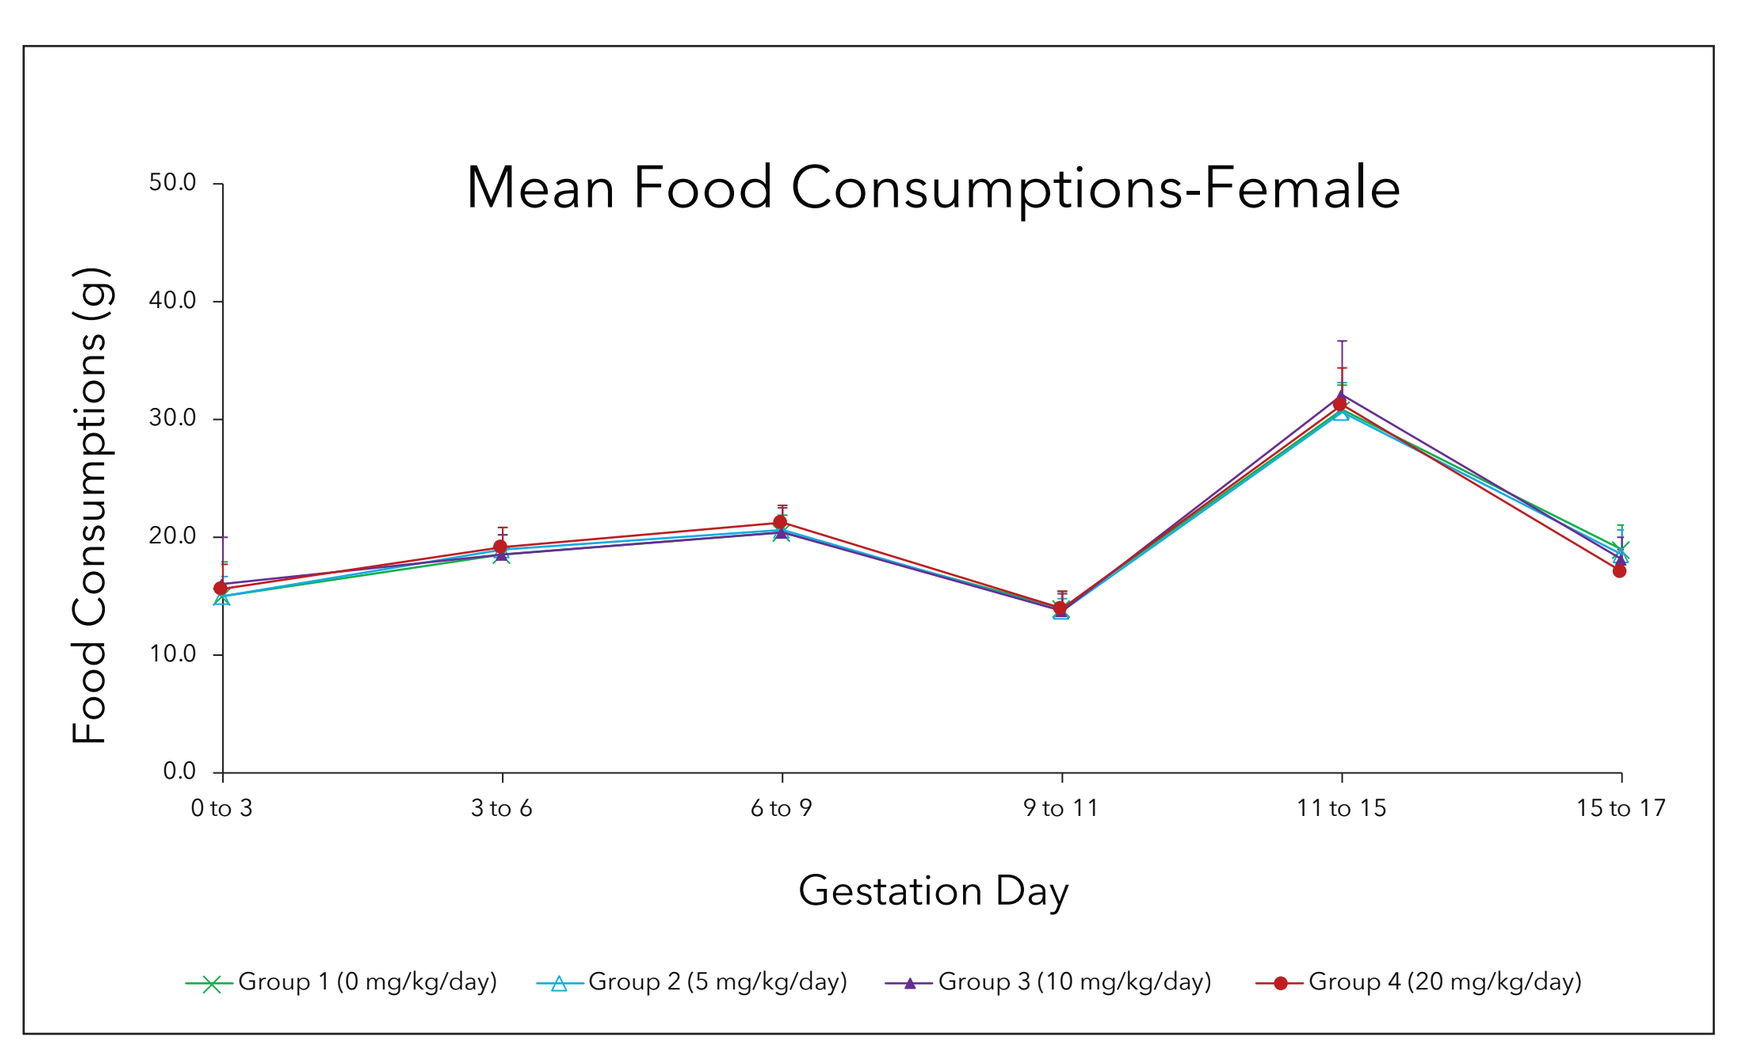
Mean Food Consumptions-Female
Food Consumptions (g)
Gestation Day
Group 1 (0 mg/kg/day)
Group 2 (5 mg/kg/day)
Group 3 (10 mg/kg/day)
Group 4 (20 mg/kg/day)

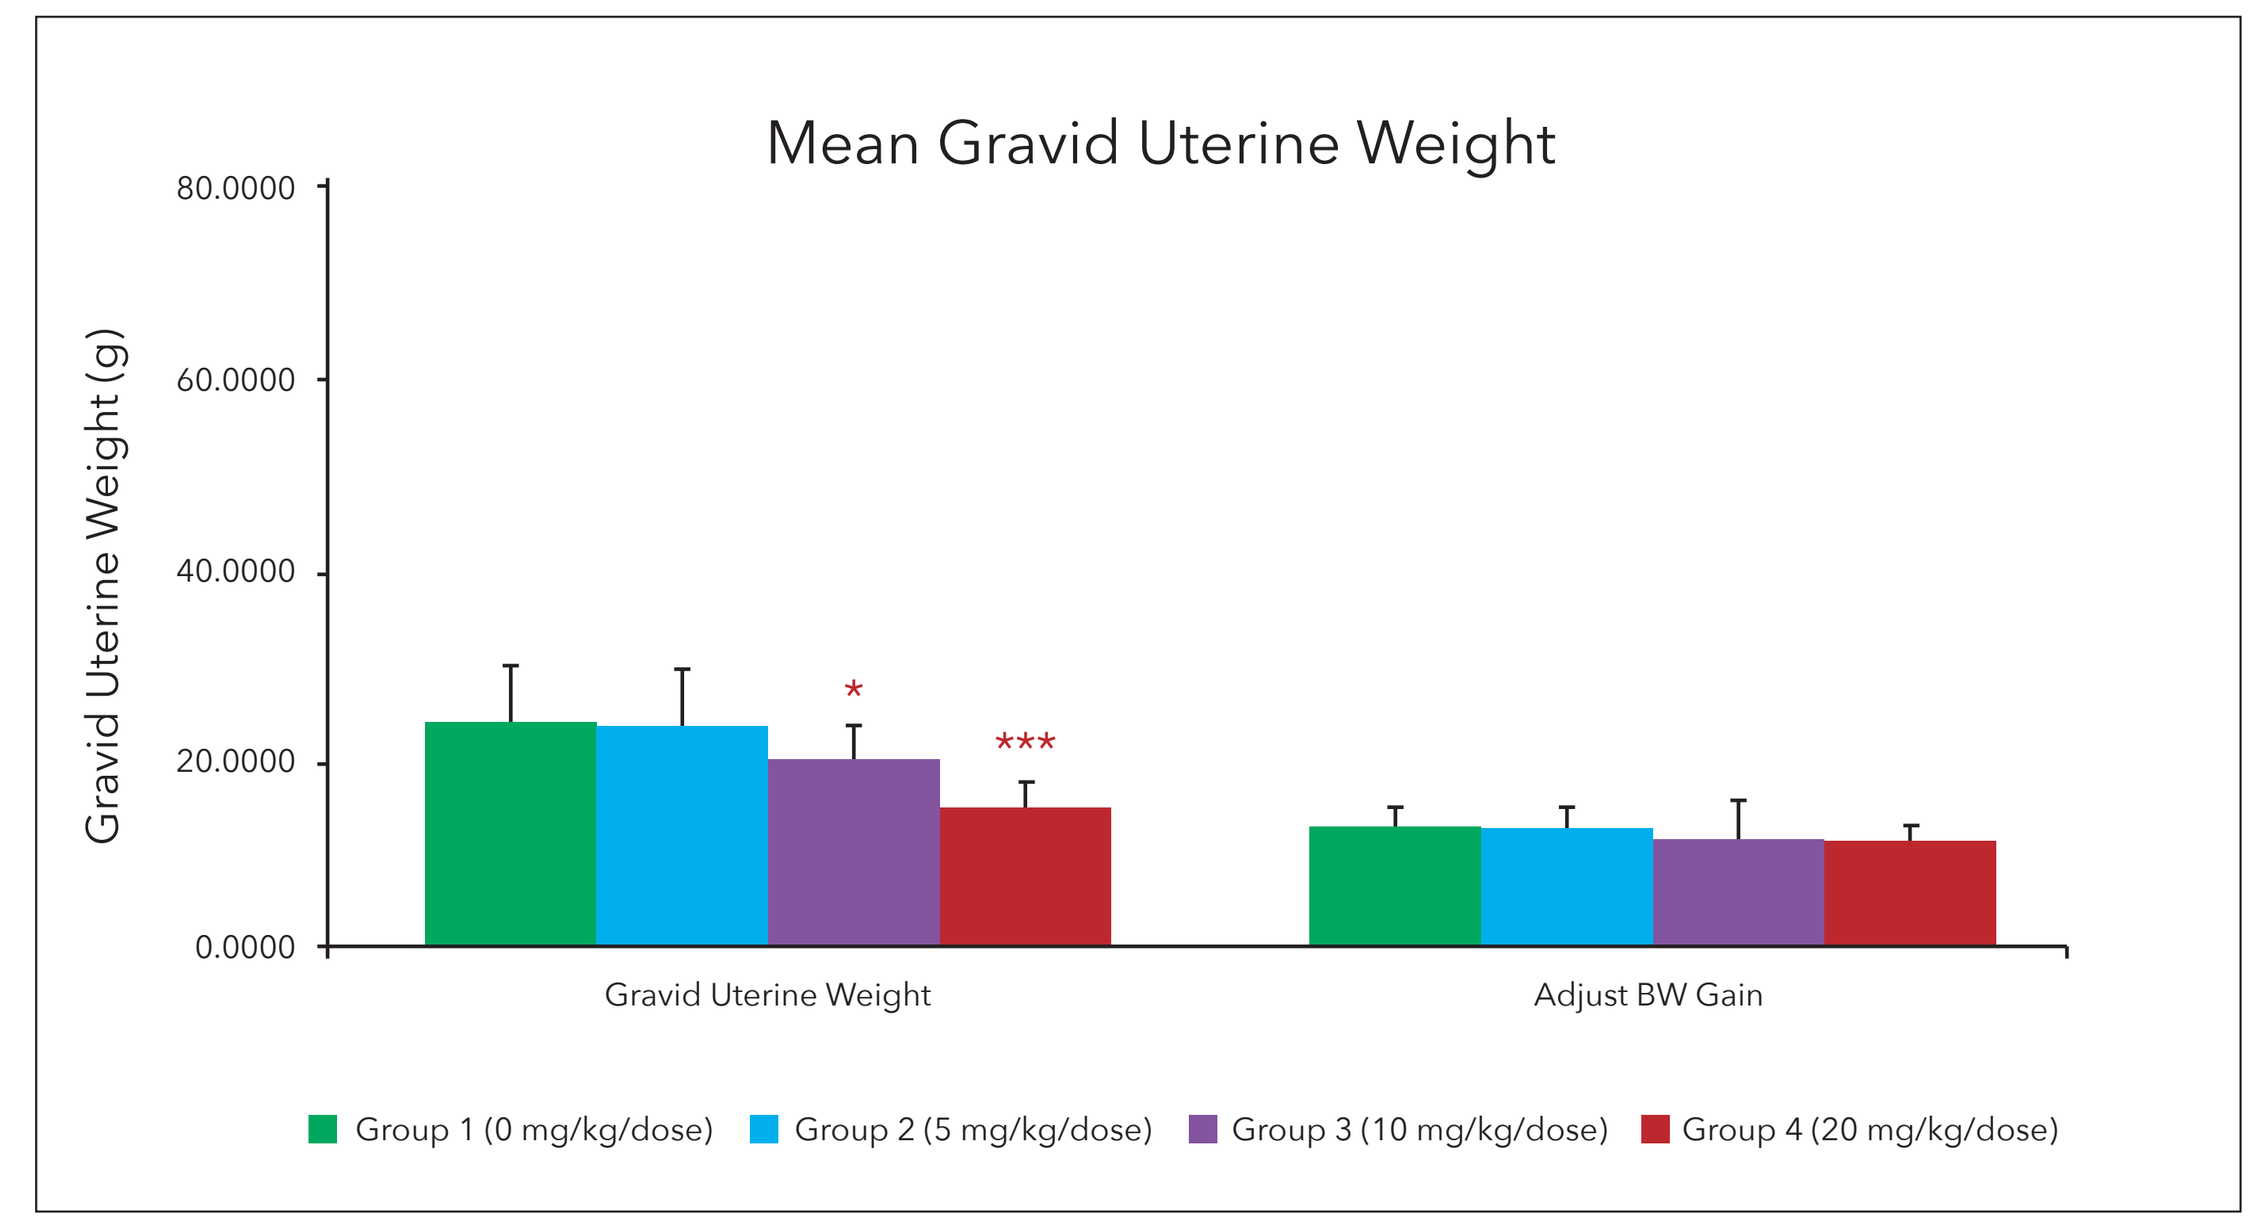
Mean Gravid Uterine Weight
Gravid Uterine Weight (g)
Gravid Uterine Weight
Adjust BW Gain
Group 1 (0 mg/kg/dose)
Group 2 (5 mg/kg/dose)
Group 3 (10 mg/kg/dose)
Group 4 (20 mg/kg/dose)

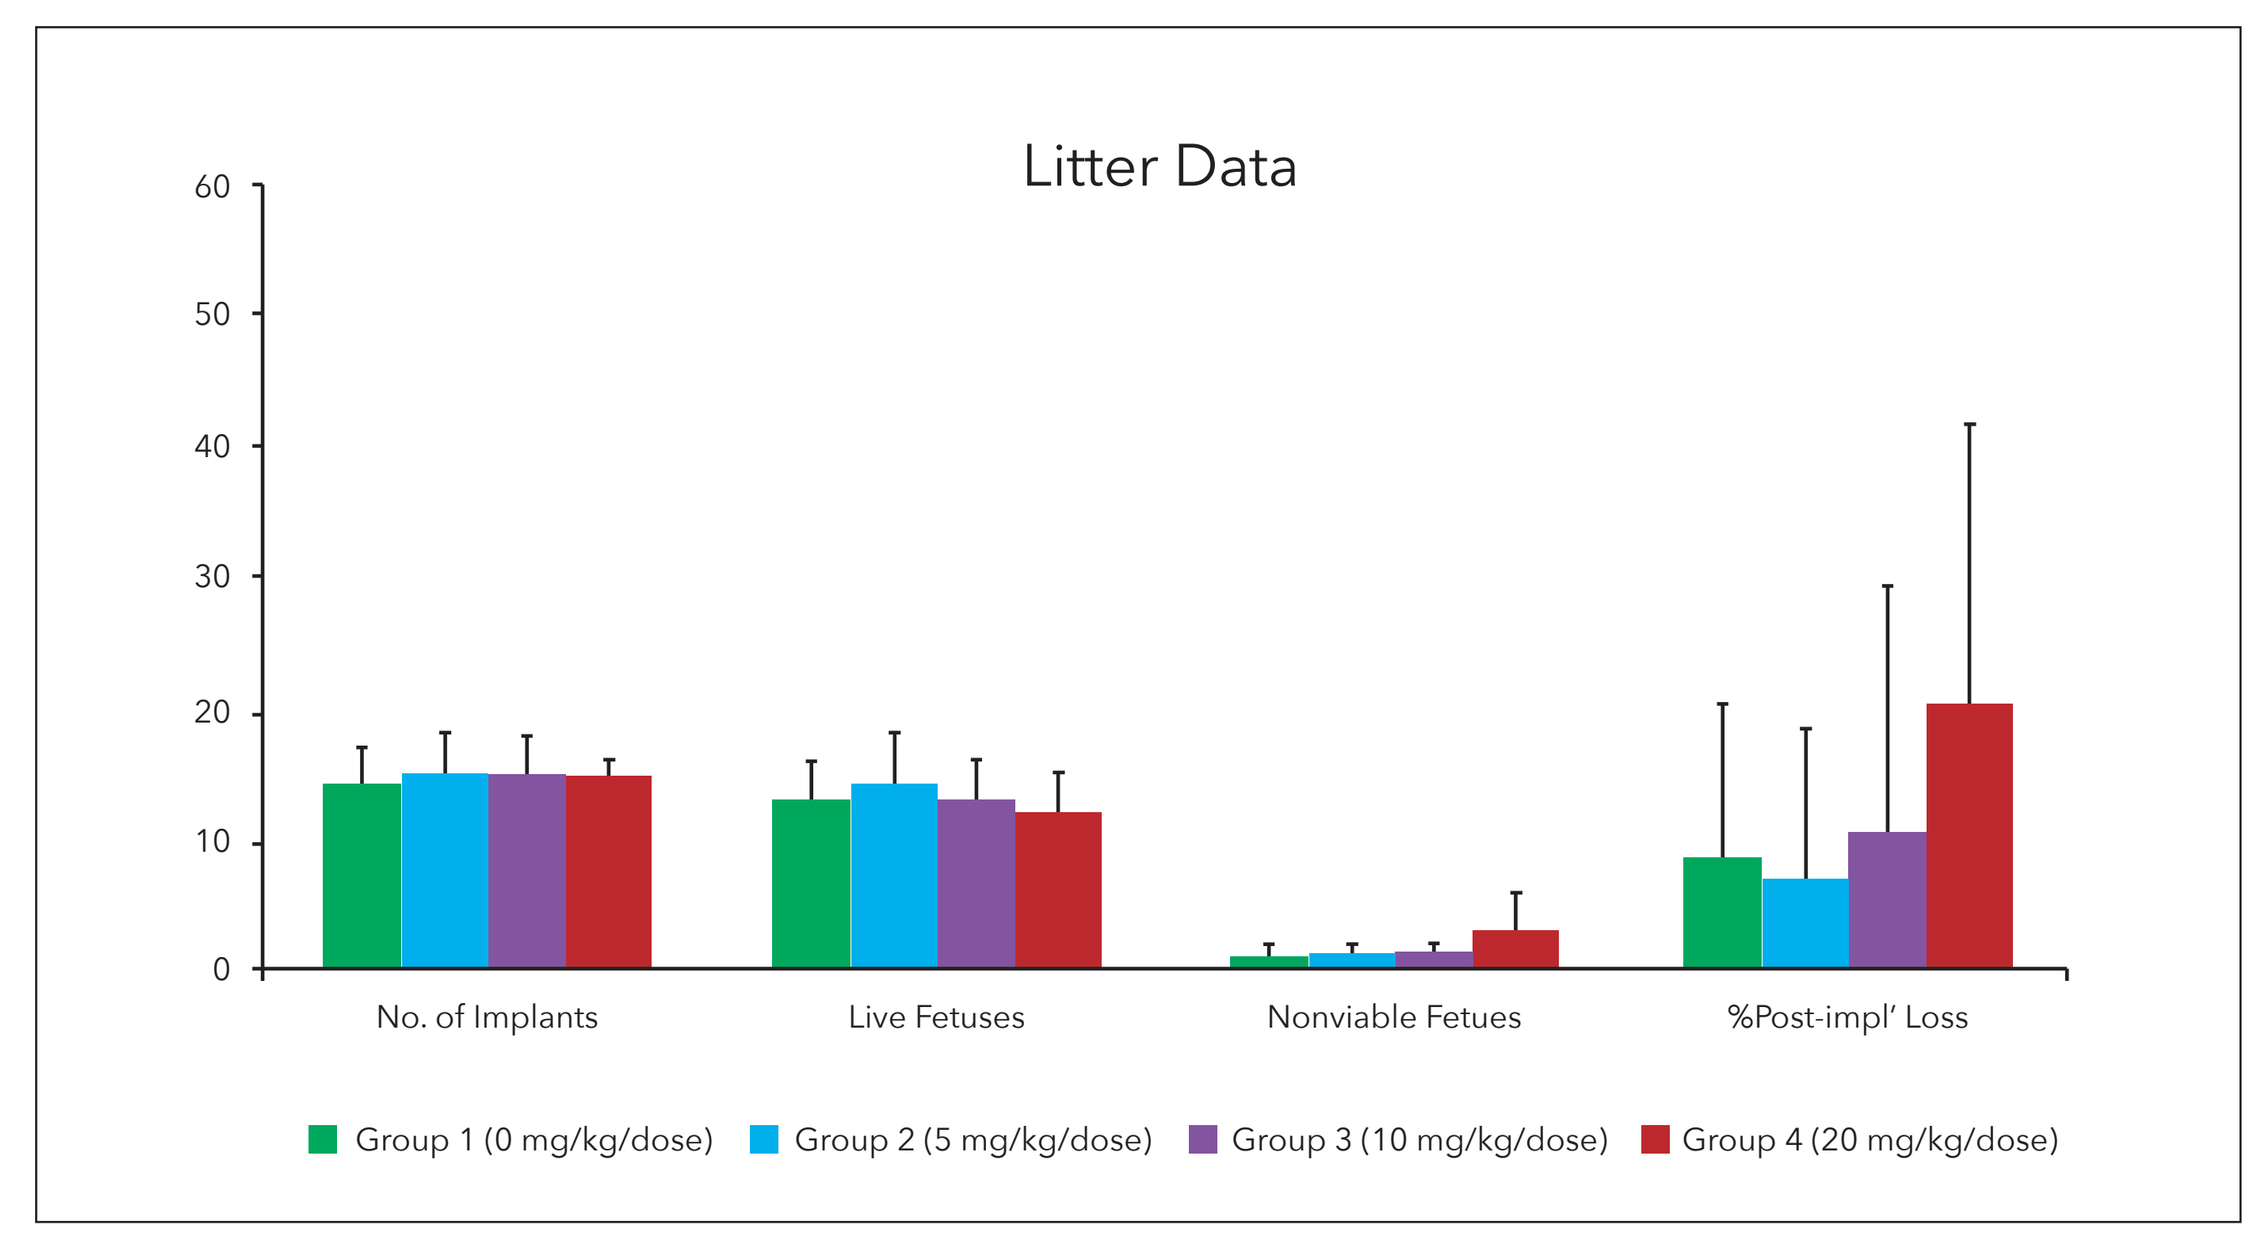
Litter Data
No. of Implants
Live Fetuses
Nonviable Fetues
%Post-impl' Loss
Group 1 (0 mg/kg/dose)
Group 2 (5 mg/kg/dose)
Group 3 (10 mg/kg/dose)
Group 4 (20 mg/kg/dose)

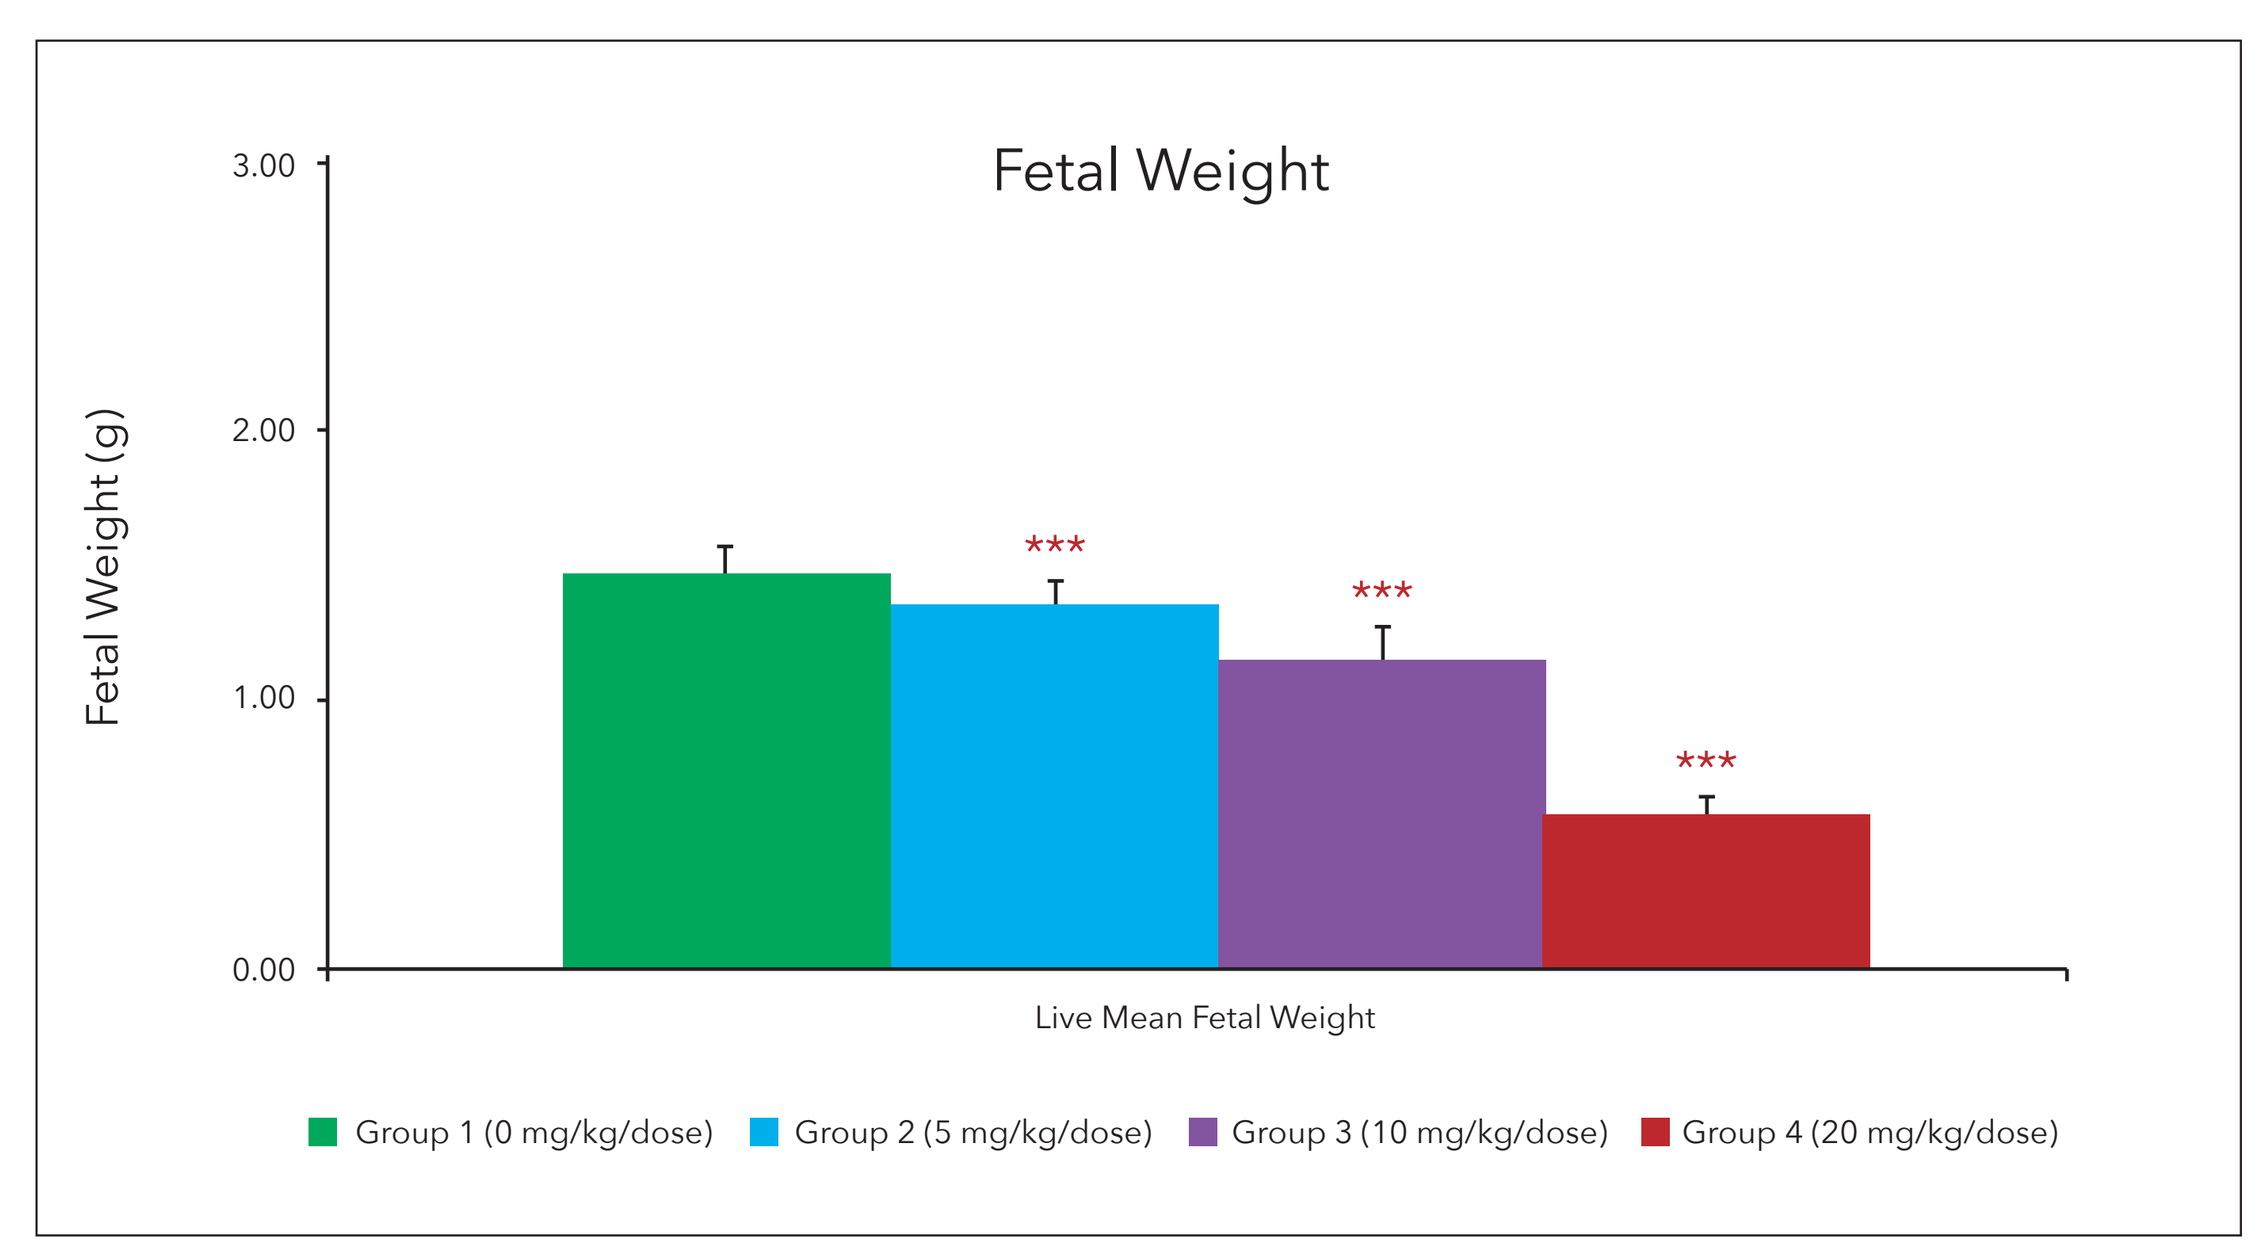
Fetal Weight
Fetal Weight (g)
Live Mean Fetal Weight
Group 1 (0 mg/kg/dose)
Group 2 (5 mg/kg/dose)
Group 3 (10 mg/kg/dose)
Group 4 (20 mg/kg/dose)

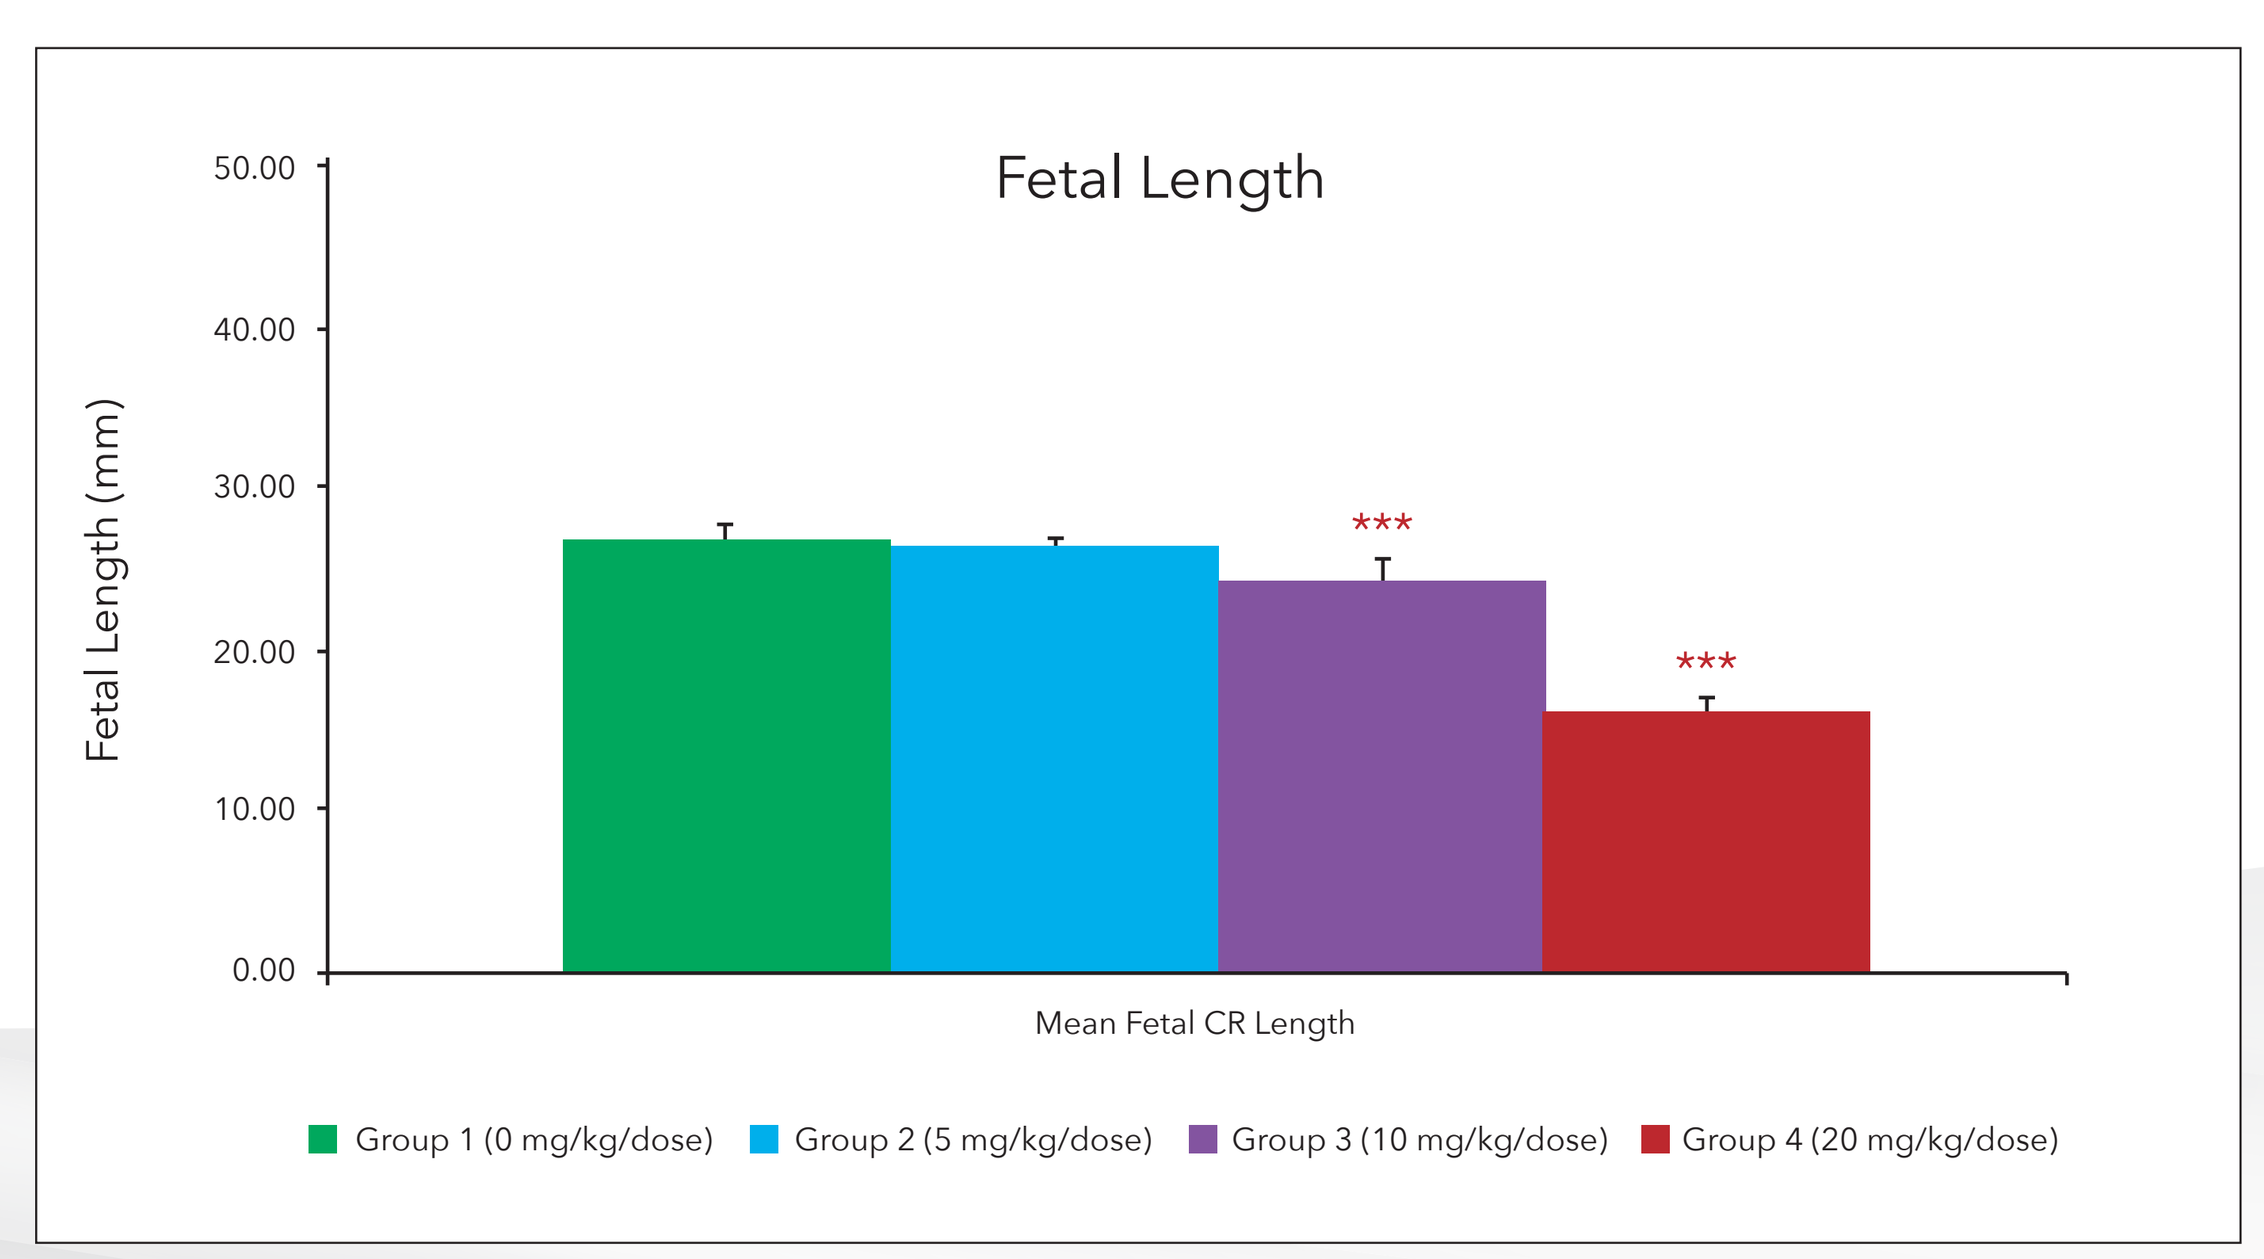
Fetal Length
Fetal Length (mm)
Mean Fetal CR Length
Group 1 (0 mg/kg/dose)
Group 2 (5 mg/kg/dose)
Group 3 (10 mg/kg/dose)
Group 4 (20 mg/kg/dose)

Results

Test article-related slight decreased food consumption were observed at ≥10 mg/kg/day (4.7%, 9.8%) during GD 15-17, which correlated with lower mean body weight (up to 8.0%, 15.1%) during GD 15-18. The decreased maternal body weights were secondary to decreased gravid uterine weights (16.0%, 36.4%) because the adjusted body weights on GD 18 (excluded gravid uterine weight) were comparable to control and were not considered adverse.

Test article-related increased post-implantation loss (induced by decreased live fetuses and increased nonviable fetuses) was noted at 20 mg/kg/day (20.1% vs 8.4% of control). Additionally, test article-related decreased fetal weight (7.95%, 20.60%, 61.79% at ≥5 mg/kg/day) and crown-rump length (9.13%, 39.57% at ≥10 mg/kg/day) were observed.

Test article-related external/skeletal malformations were observed at ≥5 mg/kg/day. All live fetuses at 20 mg/kg/day were noted with external malformations including pinna misshapen/malpositioned, open eye, small lower jaw, mouth misshapen/small, cleft palate, misshapen tongue, generalized/localized subcutaneous edema, red discolored fetus, skin lesion, exencephaly, meningo-encephalocele, hemimelia, limbs absent/hyperextension/hyperflexion/malrotated, digit/paw/claw/anus absent, digit few, paw malrotated, bent/ blunt-tipped/hooked/-malpositioned/misshapen tail, distended abdomen, omphalocele, genital tubercle misshapen and umbilicus hernia. Open eye, meningo-encephalocele/exencephaly, malrotated hindlimb, digit few, bent/hooked tail, anus absent, omphalocele or supernumerary digit were observed at 5 and 10 mg/kg/day. Test article-related skeletal malformations included fused sternebra at ≥5 mg/kg/day and humerus/radius/ulna/femur bent, small thoracic vertebra at 10 mg/kg/day.

Table 1 Summary of Affected Body Area with External Malformations

Dose Level (mg/kg/day)	5	10	20
Number of Body Area with malformations	3 (Eye, head, paw/digit)	6 (Eye, head, limb, paw/digit, tail, trunk)	9 (Ear, eye, face, general, head, limb, paw/digit, tail, trunk)

There were no test article-related changes on clinical signs, macroscopic observation, placental examination and visceral examination.

Conclusion

In conclusion, the No-Observed-Adverse-Effect-Level (NOAEL) of cyclophosphamide for maternal toxicity was 20 mg/kg/day and the NOAEL for embryo-fetal developmental toxicity was less than 5 mg/kg/day.

References

1. Torchinsky A, Savion S, Gorivodsky M, Shepshelovich J, Zaslavsky Z, Fein A, Toder V. Cyclophosphamide-induced teratogenesis in ICR mice: the role of apoptosis. Teratog Carcinog Mutagen. 1995; 15 (4):179-90.

Acknowledgements

The authors would like to acknowledge the contributions made by the team at WuXi AppTec.