Retrospective Study of Survivability and Histopathology Findings in Cannulated Rodents

Zhimei Wang, Lei Guo, Sue McPherson
WuXi AppTec

Poster #: P119

Purpose

A retrospective study was performed to determine the survivability and operation related pathology findings in control intravenous (IV) (femoral vein) cannulated rodents. IV cannulation is a technique in which a cannula is placed inside a vein to provide venous access, which is typically used for long duration administration and/or alleviating the irritation problem of dose formulations. Data were collected from 281 females and 306 males, aged 6 to 10 weeks, from 32 studies (mostly 5 to 28 days duration, ranged from 0.16 to 2 hours infusion) evaluated at the facility between 2019 and 2023. The survival rate was 97% (male) and 98% (female). For early terminated animals, the cause of death was attributed to the primary procedure-related macroscopic/microscopic findings of the single nodule (correlating microscopically with suppurative inflammation) in the injection site or major secondary embolic pneumonia in the lung/bridging hepatocellular necrosis in the liver/pyelonephritis or nephropathy in the kidney/suppurative inflammation in the urinary bladder/endocarditis and thrombosis in the heart. For schedule terminated animals, the most notable microscopic changes included intravascular thrombosis, intimal fibrosis, mixed cell infiltration of inflammatory cells and/or mineralization. The secondary procedure-related microscopic findings resulting from directly expanding or indirectly spreading through the blood/lymph circulation from the primary thrombosis/ suppurative inflammation consist direct expanding caused suppurative inflammation; indirectly spreading caused secondary findings in multiple organs; compensatory responses; stress-related non-specific responses. Such a data base will help both the toxicologist and pathologist to differentiate the study results so that the test article effects can be distinguished from procedural related findings.

Methods and Materials

Tissue samples from a total of 281 females and 306 male rats were obtained from control groups of thirty-two preclinical toxicological studies.

- The animals came from accredited suppliers Vital River, China.
- All control animals obtained had been dosed with an appropriate vehicle under same cannulation procedures (5 to 28 days duration, ranged from 0.16 to 2 hours infusion).
- At necropsy, standard tissue list as well as all injection sites, including the cannulated vein with cannula and surrounding tissue, were collected and preserved in 10% neutral buffered formalin.
- To be specific for injection site, three locations were collected (Fig. 1): location A: at approximately 0.5cm distal (posterior) to the end of the cannula in contact with the vein; location B: at approximately 0.2 to 0.5 cm proximal (anterior) to the end of the cannula in contact with the vein; location C: at approximately 0.5cm distances proceeding proximal (anterior) of location B.
- Two approximately 2- to 4-mm-thick pieces of the fixed tissue samples were processed for the routine paraffin embedding technique.
- Findings were graded as either minimal or mild based on the 5-scale grading system (minimal, mild, moderate, marked and severe).
- Slides were evaluated by qualified veterinary pathologists.

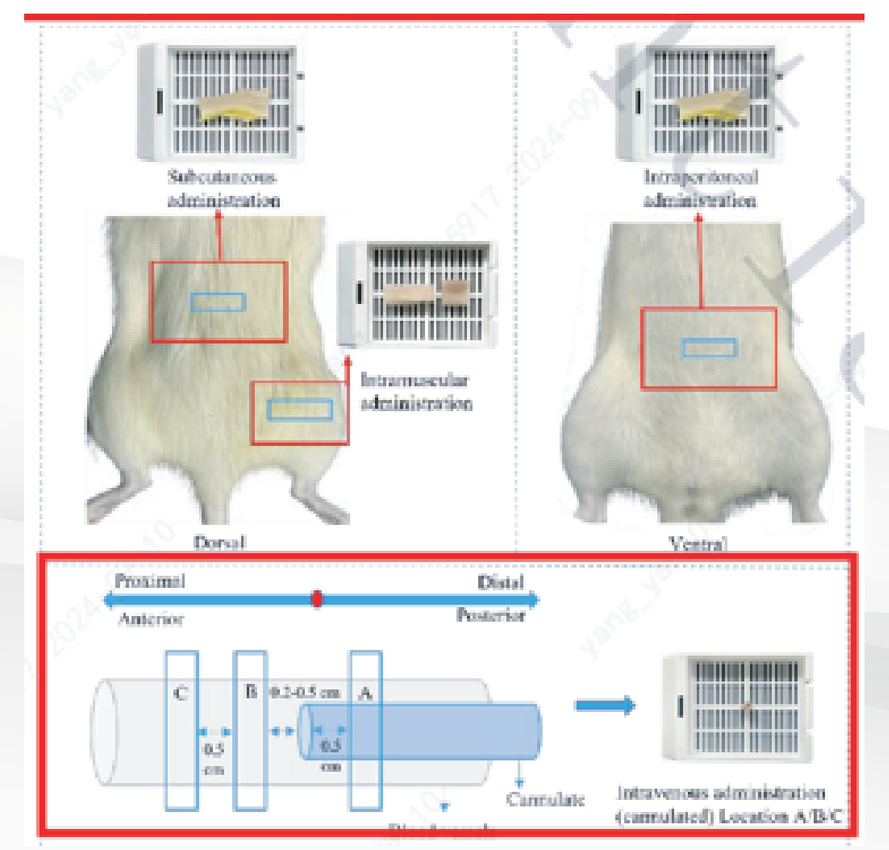
Subcutaneous administration
Intraperitoneal administration
Intramuscular administration
Dorsal
Ventral
Proximal
Anterior
Distal
Posterior
Cannulate

Figure 1 Schematic Diagram: Injection Site Collection from Femoral Vein Cannulated Rats

Results and Discussion

Key Macroscopic and Microscopic Findings: Injection Site

The mortality rate was 3% (male) and 2% (female). The primary procedure-related macroscopic finding of a single nodule in the injection, correlating microscopically with mild to marked suppurative inflammation. Primary procedure-related microscopic findings were identified in the injection sites (peri-catheter thrombosis or progressing to suppurative inflammation with/without perivascular hemorrhage). Data refers to Table 1.

Table 1 Major Gross and Histopathology Findings at Injection Site(s) - (Combined Sexes)

Injection Site(s)	Macroscopic Finding	Injection Site(s)	Histopathology Findings
	Discoloration; Red, Locally Extensive		Abscess; Focal
	Gel-Like Material		Accumulation; Fibrin, Subcutaneous
	Inflammation; Neutrophilic, Middle Portion; Severe		Degeneration/Necrosis; Vein
	Mass; Multifocal/single		Edema
	Mass; Single		Fibrosis/Intima/Perivascular; Focal,
	Nodule; Single		Foamy Macrophage; Intima
	Nodule; Firm, Single/Soft, Single, Subcutaneous		Hemorrhage
	Thick; Focal, Vascular Wall		Hyperplasia; Intima; Diffuse/Vascular Wall/Tunica Intima; Locally Extensive/Tunica Intima
	Thrombosis; Middle Portion		Infiltration, Mixed
			Inflammation; Vascular/Perivascular/Locally Extensive
			Mineralization; Vascular/Perivascular, Focal/Intima/Multifocal
			Necrosis; Minimal/Inflammation; Vein
			Pigmentation, Hemosiderin; (Vascular)/ Pigmented Macrophage; Intima
			Thrombosis/Acute, Focal/Chronic, Focal/Intravascular/Pericathether

Key Macroscopic and Microscopic Findings: Non-Injection Site

Table 2 below present the most common gross findings at non-injection site(s).

Macroscopic findings were noted in the adrenal glands (bilateral enlarged) (0.34%), lungs (discoloration), liver (enlarged) (0.17%), kidney (discoloration/pelvic dilation) (0.34%), urinary bladder (fluid-filled contents) (0.17%), spleen (enlarged) (0.34%), thymus (small) (0.17%), femur including stifling joint (swelling) (0.17%), adhesion among/between multiple organ/tissues (0.51%), etc. Also, 3 of the unscheduled deaths were attributed to sepsis which related to cannulation. Affected animals had a range of findings including meningitis, and/or pneumonia characterized by neutrophilic infiltration into affected tissues and often with associated bacteria in lungs, and brain.

Histopathology changes of adjacent tissues attributed by inflammatory infiltration or inflammatory response at the injection site as follows: sternum, bone marrow (hematopoietic); adrenal (hematopoietic); seminal vesicles (inflammation); kidney (inflammation); large intestine, rectum (inflammation); liver (hematopoietic); muscle, biceps femoris (inflammation); sciatic nerve (infiltrate); skin, (inflammation); spleen (hematopoietic); bladder (inflammation).

Table 2 Non-injection Site(s) Major Gross and Histopathology Findings at Injection Site(s) - (Combined Sexes)

Tissues/Organs	Gross Findings	Histopathology Findings
Adrenal Glands	Enlarged; Bilateral Swelling; Unilateral	Hypertrophy; Cortex; Diffuse, Bilateral
Bone, Femur, Including Stifle Joint	Dilation; Ventricle;	Etramedullary Hematopoiesis/Hematopoietic Hypercellularity
Brain	Cerebrum; Bilateral Discoloration; Dark Red, Focal, Right	Agenesis; Cerebrum; Cortex; Focal, Unilateral/ Dilatation; Ventricle
Kidneys	Acute; Focal, Unilateral, Mild Adhesion 1) Dilation; Pelvis; Bilateral/Right, Unilateral/Pale, Focal, Bilateral/Right/Multifocal To Coalescing, Bilateral/Multifocal, Bilateral 2) Discoloration; Yellow, Focal	1) Infiltration, Mixed; Pelvis/Mononuclear; 2) Inflammation, Cortex/Acute; Capsule/Mixed Cell, Cortex; Medulla/Neutrophilic, Cortex; Medulla And Papilla; Locally Extensive 3) Mineralization; Cortex; Multifocal/Tubular 4) Nephroblastematosis; Incidental/ Nephropathy; Cortex; Diffuse, Bilateral/Medulla; Diffuse, 5) Pyelonephritis; Bilateral/Cortex; Medulla; Bilateral/Medulla; 6) Tubulointerstitial Nephritis; 7) Vacuolation; Tubular, Cortex; Bilateral/Epithelium/Focal, Unilateral/Cortex
Liver	Adhesion Discoloration; White, Focal/Red, Focal	1) Fibrosis; Thrombosis 2) Hypertrophy/ Hyperplasia; Kupffer Cell 3) Increased Mitotic Figure; Hepatocyte 4) Infiltration, Lymphocytic/Mixed/Mononuclear; Perivascular; 5) Microgranuloma; Necrosis/Inflammation; Hepatocellular 6) Vacuolation; Kupffer Cell; Diffuse/Macrophage, Sinusoid/Microvesicular, Hepatocyte; 7) Vacuolation; Microvesicular, Hepatocyte;
Lungs With Mainstem Bronchi	Discoloration; Dark Red, Focal/Multifocal	1) Accumulation; Alveolar Macrophage/ Aggregation; Macrophage 2) Embolism 3) Hemorrhage; Alveolus; 4) Hyperplasia; Epithelial, Pneumocyte Type ii/Vascular, Tunica Media 5) Increased; Alveolar Macrophage/Macrophage, Alveolus; 6) Infiltration(Eosinophilic; Periarterial; Histiocytic; Alveolus; Inflammatory Cell; Perivascular) 7) Inflammation, Chronic Active; Focal/Multifocal; Interstitium 8) Inflammation, Granulomatous/Interstitium; Alveolus/Neutrophilic 9) Macrophage, Increased/ Metaplasia; Osseous 10) Microgranuloma; Multifocal/ Multinucleated Giant Cell 11) Necrosis/Inflammation; Vascular/Perivascular 12) Pneumonia; Embolic
Seminal Vesicles	Adhesion	Inflammation; Suppurative, Vesicle; Diffuse
Skin, Inguinal	Alopecia; Focal/Multifocal Discoloration; Yellow Focus; Depressed, Focal Scab. Multifocal/Focal/Multifocal to Coalescing	1) Crust/ Fibrosis; Dermis/Granuloma/ Hyperplasia 2) Infiltration; Foamy Macrophage, Subcutaneous; Dermis/ Inflammation; Suppurative, Focal 3) Ulceration; Diffuse/ Ulceration; Epidermis; Focal
Spleen	Enlarged; Diffuse	1) Extramedullary Hematopoiesis; Increased, Red Pulp; Diffuse 2) Hematopoiesis; Increased/Hyperplasia; Lymphoid, White Pulp 3) Increased Extramedullary Hematopoiesis/ Increased; Macrophage, Red Pulp; Scattered 4) Vacuolation; Macrophage, Red Pulp Depletion; Lymphoid, White Pulp/Hyperplasia; Diffuse 5) Extramedullary Hematopoiesis; Increased, Red Pulp 6) Erosion; Fundic Mucosa
Stomach	Discoloration; Dark Red, Multifocal, Mucosa; Glandular	Ulceration; Non-Glandular; Mucosa/ Ulceration; Pylorus
Urinary Bladder	Contents; Fluid-Filled	Dilation/ Hyperplasia; Mucosal, Diffuse/ Infiltration, Mixed; Diffuse
Uterus, Including Cervix	Adhesion	Fibroplasia; Adventitia; Focal/ Hyperplasia; Adventitia; Diffuse

Conclusion

The cause of death or adverse clinical signs were attributed to the primary procedure-related microscopic findings of suppurative inflammation in the injection site and its related major findings occurred in the heart, lungs, liver, kidney, and/or urinary bladder.

Canulation related primary procedure-related microscopic findings of suppurative inflammation in the injection site and its related major findings occurred in the heart, lungs, liver, kidney, and urinary bladder, etc organ.

Access to such a data base will help the scientist toxicologist discern the true test article related effects.

Acknowledgements

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec.

References

Nancy E. Everds et al., Interpreting Stress Responses during Routine Toxicity Studies: A Review of the Biology, Impact, and Assessment[J], Toxicologic Pathology, 2013. 41:560-614.

Elizabeth F. McInnes, Background Lesions in Laboratory Animals: A Color Atlas. Saunders Elsevier, 2012. p. 17-35, p. 103.