Validation of the Flow-cytometric Peripheral Blood Micronucleus Assay in Mouse

Mingzhu Kang, Ning Xu, Lingling Cao, Xiang Shan, Liwen Gao, Millie Chen
WuXi AppTec (Suzhou)

药明康德 WuXi AppTec

Poster #: P508

Purpose

Traditionally, the mammalian erythrocyte micronucleus assay employs bone marrow collection and manual scoring for micronucleus frequency detection, which is labor-costing and time-consuming. The flow cytometry (FCM) method, which owns the advantages of time-efficient and data robustness and consistency, was also adopted and recommended by ICH-S2(R1) guideline and OECD Testing Guideline 474. And although extensive work using this method has been performed in rats in pre-clinical studies, only a few tests had been done in the mouse in regular testing. This presentation shows the validations of mouse peripheral blood micronucleus assay using flow cytometry as the analysis method.

Methods and Materials

A routine three daily administration was determined, including negative control (0 mg/kg/day), hydroquinone (20, 40 and 80 mg/kg/day), positive control article cyclophosphamide monohydrate was administrated once at 75 mg/kg, samples were collected at 20-24 hours post last dosing and processed following Litron's commercial analysis kit manual. Percentage of reticulocyte among total erythrocytes (%RET) and micronucleus frequency (%MN-RET) were the parameters and evaluated over the evaluation criteria for intra-day and inter-day precision, carryover effect and stability.

Results and Discussion

a. The flow-cytometric based peripheral blood micronucleus assay in mouse was valid. The micronucleus frequency in the negative control group was comparable with literature.

b. The test article, hydroquinone, a well-know genotoxic compound, had induced statistically significant and dose-related increase in micronucleus frequency for both males and females. The positive control (CP) also induced statistically significant increases in micronucleus formation.

c. For evaluation, blood samples were stabile for at least for 30 days when stored in Fixative (absolute methanol) or LTSS (long term storage solution). No obvious carryover between samples were noted. Both intra-day and inter-day assay variation met acceptable criteria.

Validation Parameters

Characteristic	Procedure	Acceptance Criteria
Negative and positive	The kit-supplied positive and negative control samples were analyzed at least 2 times on different analyzing days.	The data obtained will be calculated for %RET and %MN-RET and should be within/comparable with the reference range provided by the supplier.
Carryover	Detect both the negative and positive blood samples in a sequence of positive 1 (P1) – positive 2 (P2) – positive 3 (P3) – negative 1 (N1) – negative 2 (N2) – negative 3 (N3) with automatic washing selection between each sample.	The data obtained will be calculated for %MN-RET, and the carryover will be calculated as [(N1-N3)/P3] ×100%. The carryover effect will be acceptable when it is less than 5%.
Precision	For the intra-day precision, three fixed blood samples from two animals were labeled separately for detection. For the inter-day precision, three fixed blood samples from two animals were labeled for detection daily for two consecutive days separately.	The variance estimates will be expressed as a coefficient of variation (%CV) relative to the overall mean of %RET or %MN-RET. Intra- and inter-day assay variation will be acceptable if the %CV≤20%.
Stability	Male samples of CP treatment dose group in the fixative and in the LTSS were tested separately after at least 1 month storage duration in the ultra-low temperature freezer.	The stability will be acceptable if the %RET and %MN-RET percent difference is ≤20% compared to the corresponding initial time point samples.

Table 1 Negative and Positive Control Data by Flow Cytometry Analysis

Sample	No. of RET	No. of MN-RET	No. of NCE	No. of MN-NCE	%RET	%MN-RET
Reference range supplied by the litron Lab						
Negative Control					1.41-2.20	0.12-0.27
Positive Control					0.77-1.20	1.42-3.19
Acquisition on 2024-04-01						
Kit-P1	19641	359	2183526	3972	0.91	1.80
Kit-P2	19672	328	2291793	3875	0.86	1.64
Kit-N1	19962	38	1199898	1607	1.64	0.19
Kit-N2	19958	42	1274691	1605	1.54	0.21
Acquisition on 2024-04-02						
Kit-P3	19572	428	2108637	4052	0.94	2.14
Kit-P4	19636	364	2163948	3687	0.91	1.82
Kit-N3	19962	38	1138080	1478	1.72	0.19
Kit-N4	19980	20	1179788	1345	1.67	0.10

Note: NCE=Normochromatic Erythrocyte; RET=Reticulocyte; MN-NCE=Micronucleated NCE; MN-RET=Micronucleated RET; %RET=(No. of RET+ No. of MN-RET)/(No. of RET+ No. of MN-RET+No. of NCE+No. of MN-NCE)×100%; %MN-RET=No. of MN-RET/(No. of RET+ No. of MN-RET) ×100%

Table 2 Carryover Assay by Flow Cytometry Analysis

Treatment	Sample	No. of RET	No. of MN-RET	No. of NCE	No. of MN-NCE	%RET	%MN-RET
CP Dose (75 mg/kg)	P1	19684	316	3882052	6414	0.51	1.58
	P2	19700	300	3828142	6246	0.52	1.50
	N3	19680	321	4230071	5719	0.47	1.60
Negative Control Article Dose (0 mg/kg/day)	N1	19956	58	1300982	2603	1.51	0.29
	N2	19937	63	1284688	2538	1.53	0.32
	N3	19950	50	1287502	2715	1.53	0.25
Carryover=[(N1-N3)/P3]x100%=2.48%							

Note: NCE=Normochromatic Erythrocyte; RET=Reticulocyte; MN-NCE=Micronucleated NCE; MN-RET=Micronucleated RET; %RET=(No. of RET+ No. of MN-RET)/(No. of RET+ No. of MN-RET+No. of NCE+No. of MN-NCE)×100%; %MN-RET=No. of MN-RET/(No. of RET+ No. of MN-RET) ×100% CP=Cyclophosphamide monohydrate

Table 3 Intra-day Precision Assay by Flow Cytometry Analysis

Sample	No. of RET	No. of MN-RET	No. of NCE	No. of MN-NCE	%RET	%MN-RET
2004 Sample						
	19875	125	2874707	3351	0.69	0.63
	19872	128	2876621	3157	0.69	0.64
	19883	117	2915488	3144	0.68	0.59
2005 Sample						
	19813	187	2073032	2627	0.95	0.94
	19812	188	2097434	2657	0.94	0.94
	19819	181	1991381	2625	0.99	0.91
CV%					**1.75**	**3.33**

Note: NCE=Normochromatic Erythrocyte; RET=Reticulocyte; MN-NCE=Micronucleated NCE; MN-RET=Micronucleated RET; %RET=(No. of RET+ No. of MN-RET)/(No. of RET+ No. of MN-RET+No. of NCE+No. of MN-NCE)×100%; %MN-RET=No. of MN-RET/(No. of RET+ No. of MN-RET) ×100%

Table 4 Inter-day Precision Assay by Flow Cytometry Analysis

Acquisition Date	2004 Sample		2005 Sample	
	% RET	% MN-RET	% RET	% MN-RET
2024-04-01	0.69	0.63	0.95	0.94
	0.69	0.64	0.94	0.94
	0.68	0.59	0.99	0.91
2024-04-02	0.66	0.86	1.01	0.90
	0.65	0.69	0.99	0.96
	0.65	0.63	1.01	1.06
CV%	**3.66**	**11.26**	**2.82**	**3.23**

Note: NCE=Normochromatic Erythrocyte; RET=Reticulocyte; MN-NCE=Micronucleated NCE; MN-RET=Micronucleated RET; %RET=(No. of RET+ No. of MN-RET)/(No. of RET+ No. of MN-RET+No. of NCE+No. of MN-NCE)×100%; %MN-RET=No. of MN-RET/(No. of RET+ No. of MN-RET) ×100%

Table 5 Stability Assay by Flow Cytometry Analysis

Sample	No. of RET	No. of MN-RET	No. of NCE	No. of MN-NCE	%RET	Difference(%)	%MN-RET	Difference(%)
Acquisition on 2024-04-01 for samples of initial timepoint								
2001	19701	299	3841829	6046	0.52	NA	1.50	NA
2002	19762	238	1557218	2164	1.27	NA	1.19	NA
2003	19863	137	1751270	1850	1.13	NA	0.69	NA
2004	19862	138	2893234	3350	0.69	NA	0.69	NA
2005	19795	205	1944340	2733	1.02	NA	1.03	NA
Mean					**0.92**	**NA**	**1.02**	**NA**
Acquisition on 2024-05-01 for samples stored in fixative								
2001	19682	318	4535333	7224	0.44	-15.23	1.59	6.35
2002	19757	243	1854198	2237	1.07	-15.83	1.22	2.10
2003	19842	158	2101680	2227	0.94	-16.52	0.79	15.33
2004	19872	128	3792765	4152	0.52	-23.59	0.64	-7.25
2005	19802	198	2450337	3290	0.81	-20.48	0.99	-3.41
Mean					**0.76**	**-18.11**	**1.05**	**2.75**
Acquisition on 2024-05-01 for samples stored in LTSS								
2001	19694	306	4347088	6312	0.46	-11.56	1.53	2.34
2002	19802	198	1623647	2017	1.22	-4.03	0.99	-16.81
2003	19858	142	1766976	1745	1.12	-0.88	0.71	3.65
2004	19887	113	3266513	3313	0.61	-11.35	0.57	-18.12
2005	19822	178	2155123	2813	0.92	-9.68	0.89	-13.17
Mean					**0.86**	**-6.43**	**0.94**	**-7.87**

Note:NCE=Normochromatic Erythrocyte; RET=Reticulocyte; MN-NCE=Micronucleated NCE; MN-RET=Micronucleated RET; %RET=(No. of RET+ No. of MN-RET)/(No. of RET+ No. of MN-RET+No. of NCE+No. of MN-NCE)×100%; %MN-RET=No. of MN-RET/(No. of RET+ No. of MN-RET) ×100%；Difference(%)=(samples in stored in LTSS/Fixative)-samples in the initial time point)/samples in the initial time point×100%

Figure 1 Negative(left) and Positive (right) Control Data Plots

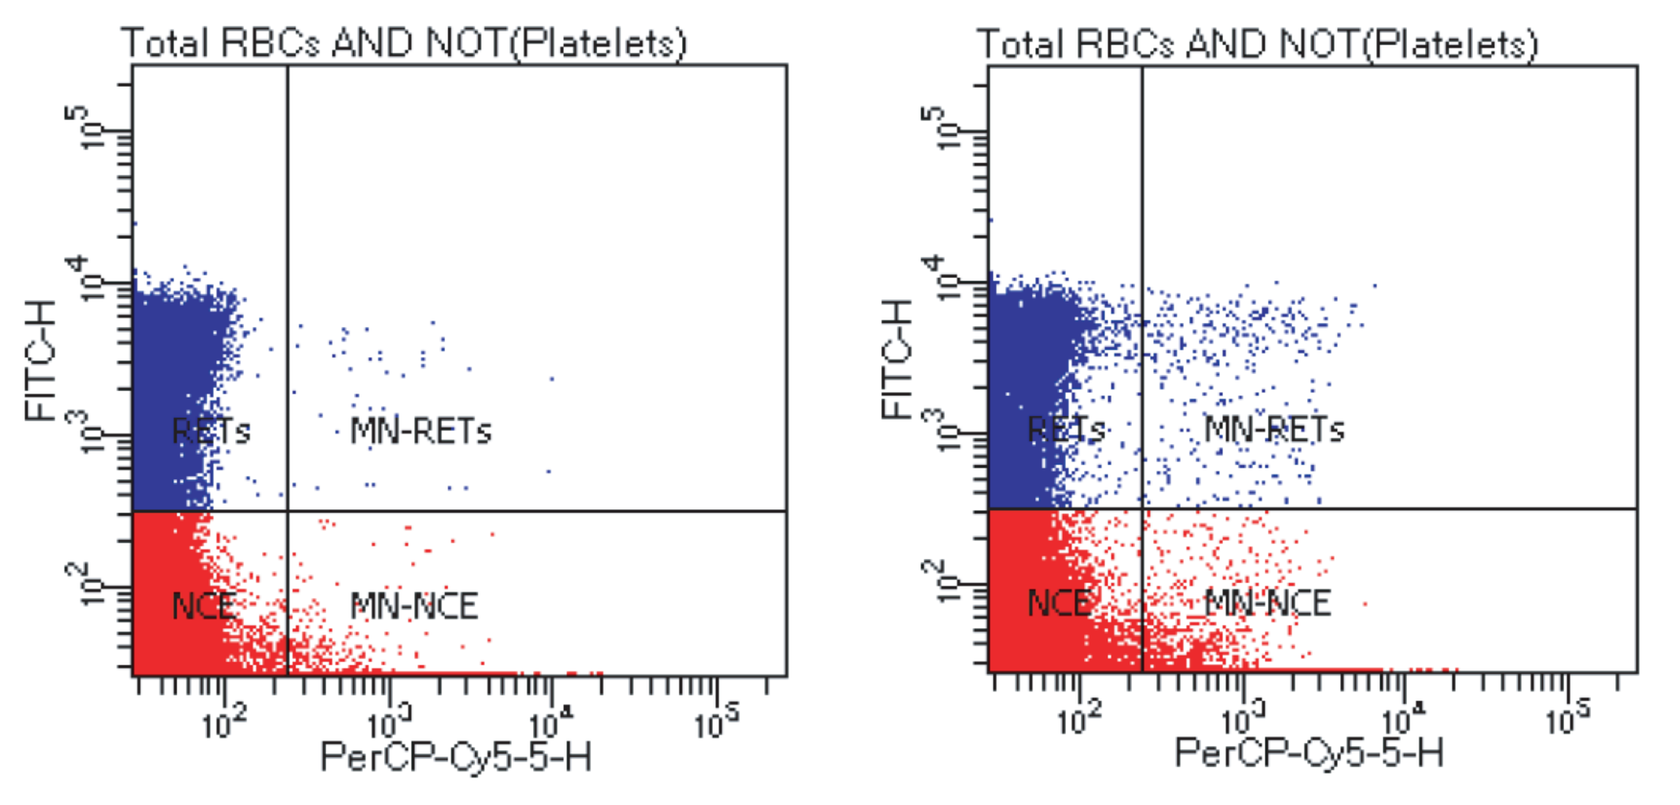
Total RBCs AND NOT(Platelets)
FITC-H
PerCP-Cy5-5-H
RETs
MN-RETs
NCE
MN-NCE
Total RBCs AND NOT(Platelets)
FITC-H
PerCP-Cy5-5-H
RETs
MN-RETs
NCE
MN-NCE

Conclusion

All validation specifications met the acceptance criteria. WuXi AppTec has validated and is able to perform flow cytometry based peripheral blood micronucleus assay in mouse under GLP condition.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and In Vitro Toxicology group at WuXi AppTec (Suzhou).