Development of a Multiplex RT-qPCR Method to Quantify mRNA Transcript

Zhihua Fang, Nan Jia, Dan Wang, Jianbo Diao, Jing Shi*
WuXi AppTec (Shanghai) Ltd.co., 288 Fute Zhong Road, WaiGaoQiao Free Trade Zone, Shanghai, China 200131

WuXi AppTec

Poster: Day2-P17

Introduction

- The number of novel therapeutic modalities in development, for example CAR-T therapy and oligonucleotide drugs, has grown significantly in recent years.
- A recent FDA draft guidance has recommended to include an assessment of pharmacodynamics biomarkers, such as target mRNA, for the development of oligonucleotide therapeutics[1].
- To support clinical pharmacology assessment of gene therapies and related products that are designed to alter gene expressions we have attempted to establish a workflow to qualitatively and quantitatively analyze mRNA transcripts from human whole blood.
- Although regulatory agencies have provided few details on experimental designs and acceptance criteria for the bioanalysis of novel INDs, recommendations described in MIQE guidelines have been incorporated into the present workflow[2].

Materials and Methods

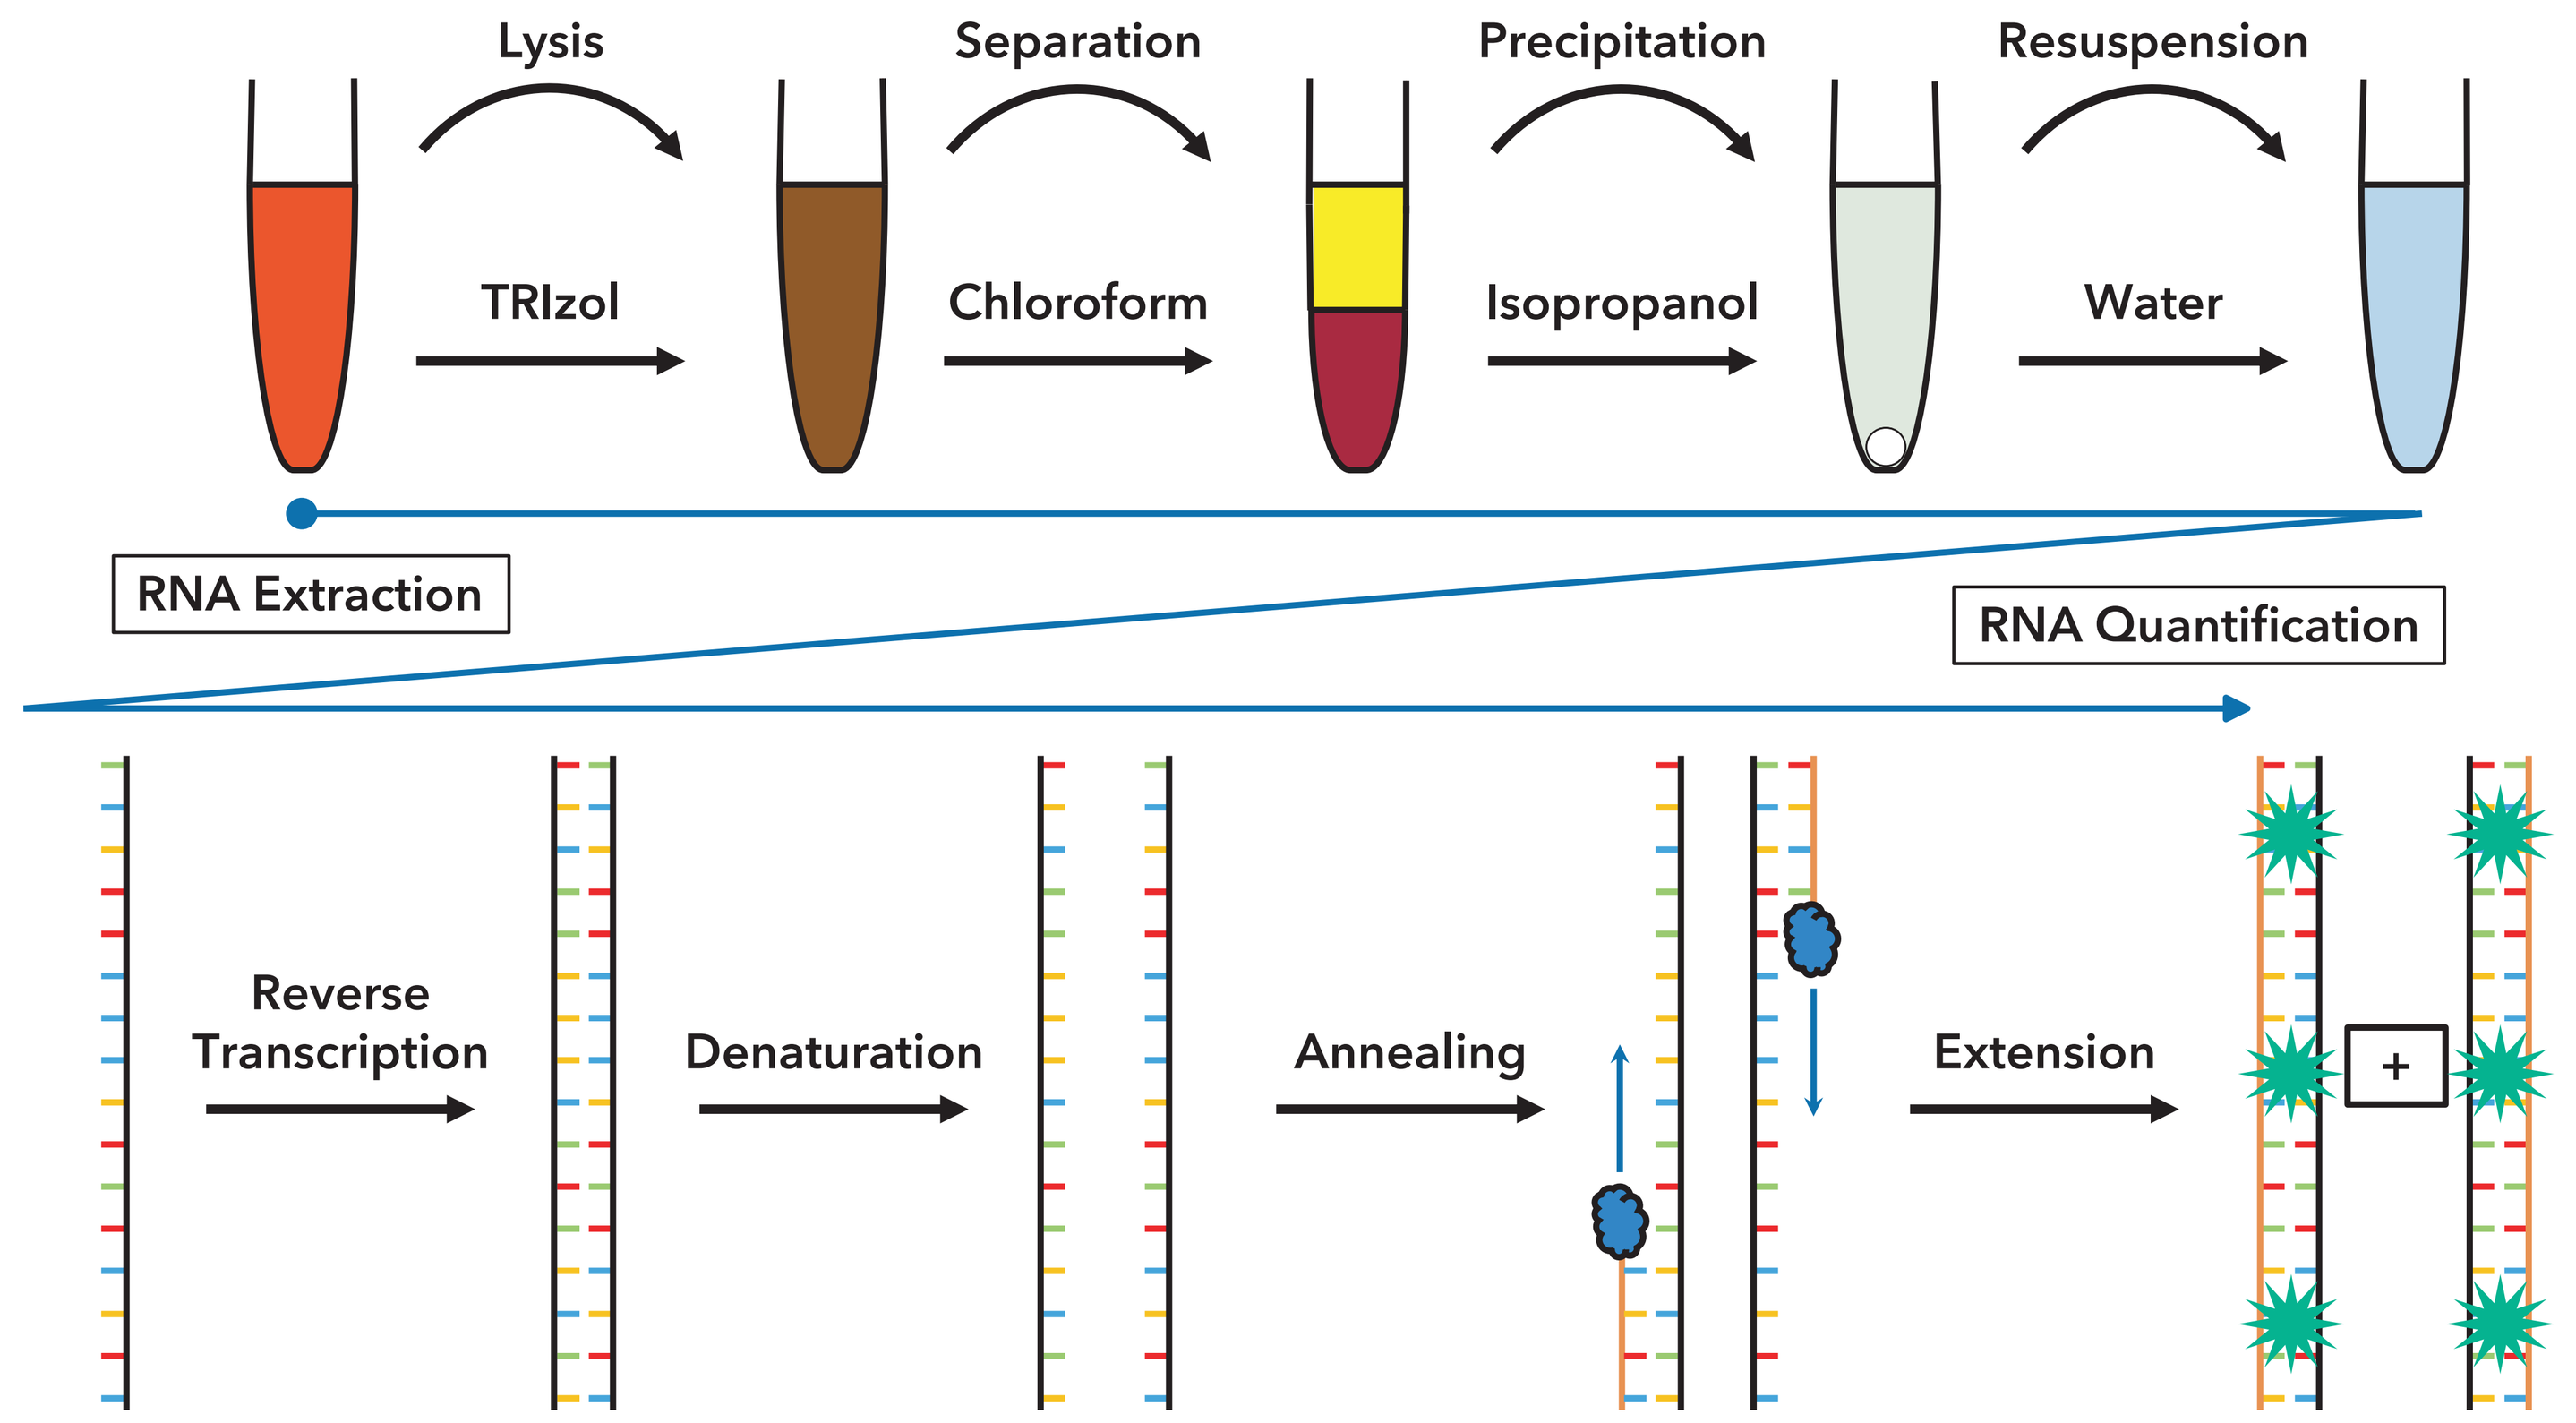
Lysis
Separation
Precipitation
Resuspension
TRIzol
Chloroform
Isopropanol
Water
RNA Extraction
RNA Quantification
Reverse Transcription
Denaturation
Annealing
Extension
+

Figure 1 An illustration showing the overall work flow of RNA extraction and quantification.

Experimental Design

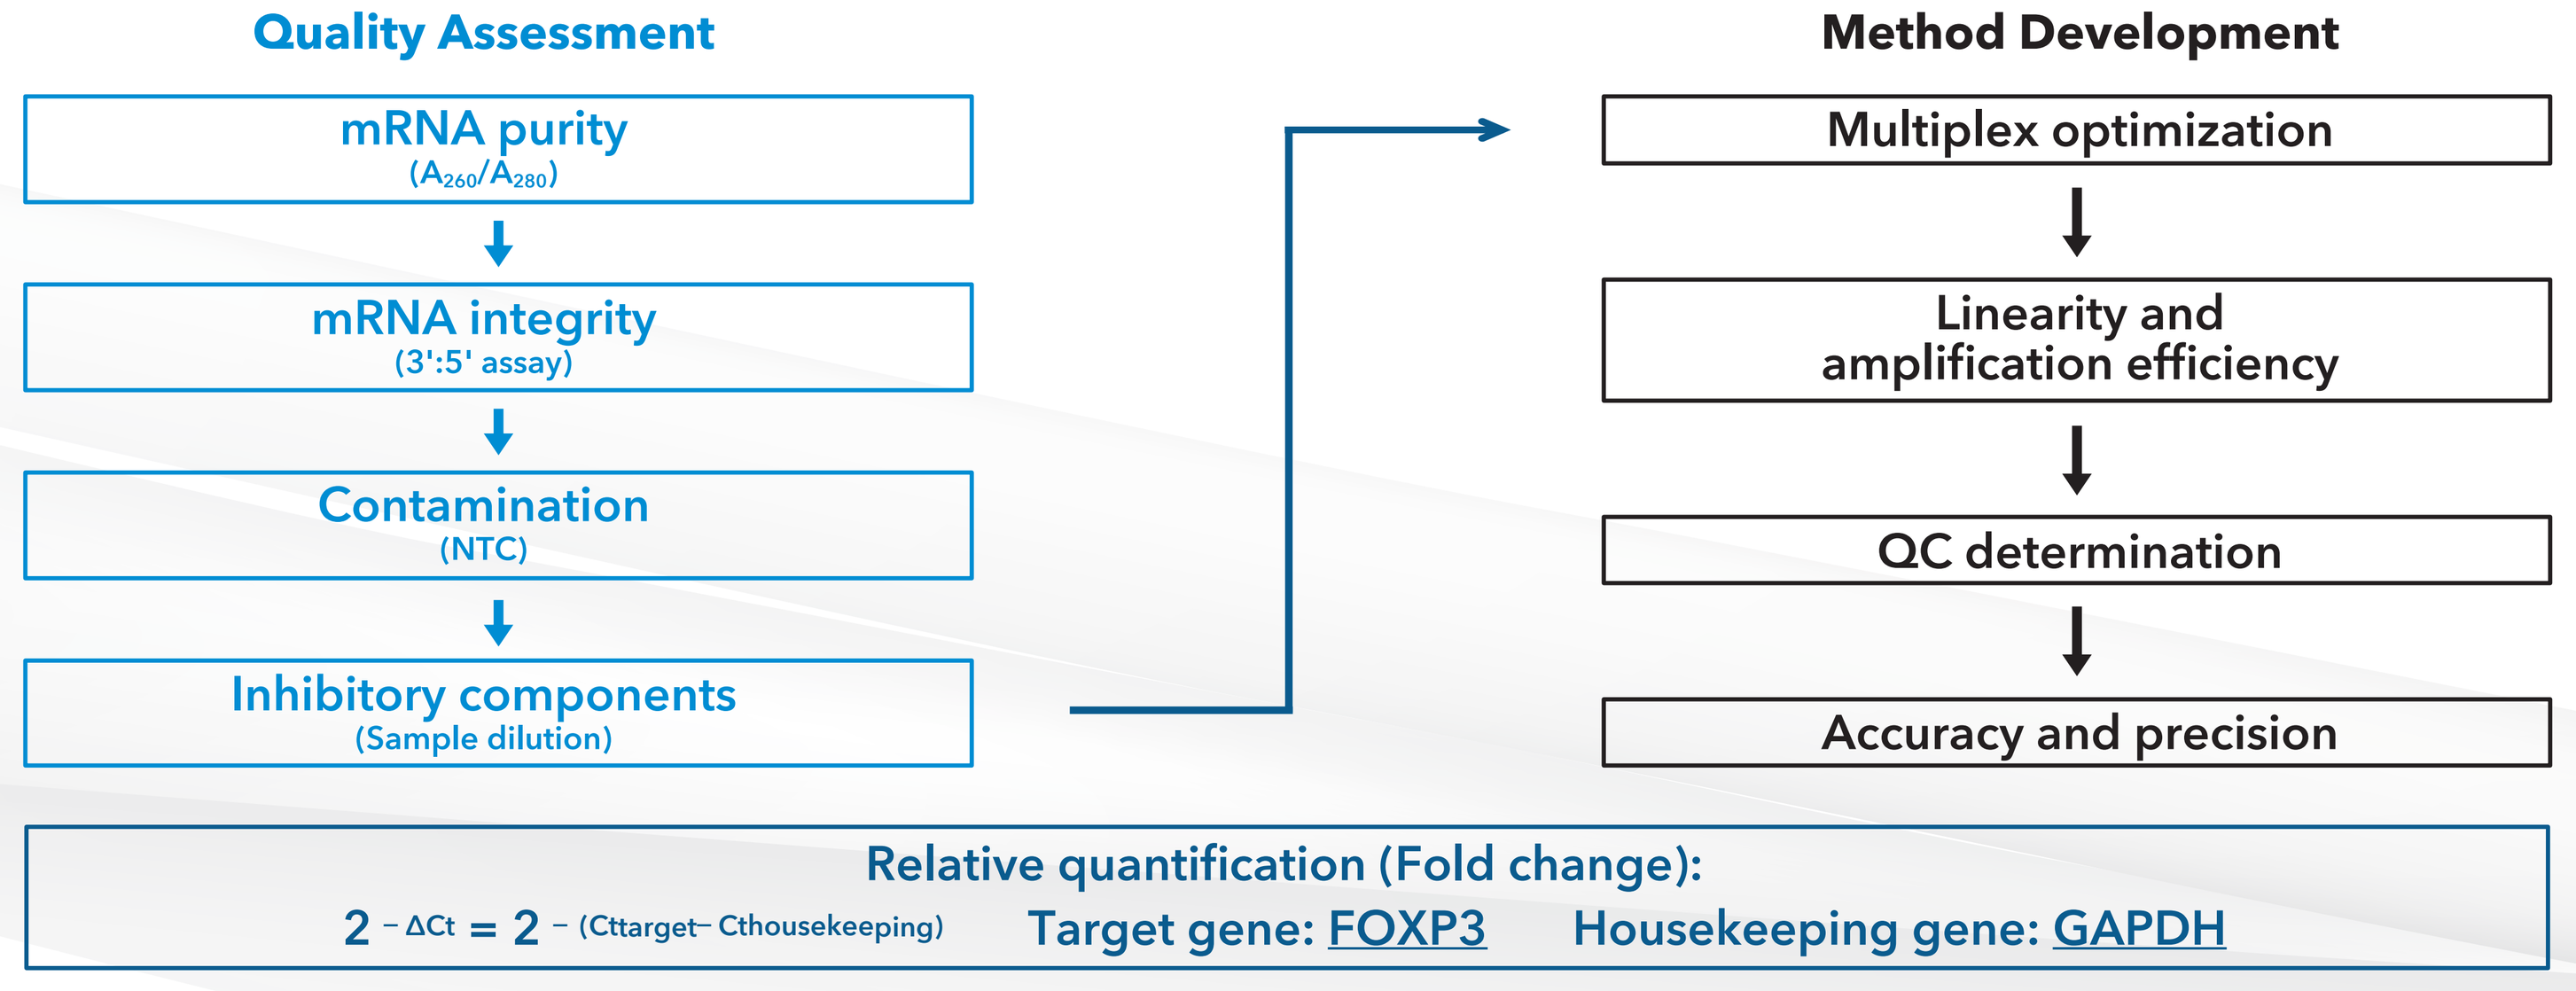
Quality Assessment
mRNA purity
(A_{260}/A_{280})
mRNA integrity
(3':5' assay)
Contamination
(NTC)
Inhibitory components
(Sample dilution)
Method Development
Multiplex optimization
Linearity and amplification efficiency
QC determination
Accuracy and precision
Relative quantification (Fold change):
$2^{-\Delta Ct} = 2^{-(Ct_{target} - Ct_{housekeeping})}$ Target gene: FOXP3 Housekeeping gene: GAPDH

Results

Quality Assessment

Parameter	Experiments	Criteria	Example
mRNA Putiry	UV-spectrophotometer (e.g., NanoDrop)	$1.85 \le A_{260}/A_{280} \le 2.10$	Conc. = 78.2 ng/µl A_{260}/A_{280} = 1.92
mRNA Integrity	3':5' assay(Ref. 2,3,4) (GAPDH gene)	0.2 ≤ 3':5' Ratio ≤ 5.0	3':5' Ratio = 1.6
Contamination	Include a triplicate of NTC in each run	$39.000 \le Ct_{(NTC)}$	Undetermined
Inhibitory Components	At least 2 serial dilution of input RNA	Bias% ($2^{-\Delta Ct}$) of a lower input RNA concentration is within ±30% of the undiluted input RNA	$2^{-\Delta Ct(200\ ng)}$ = 0.0128 $2^{-\Delta Ct(100\ ng)}$ = 0.0119 $2^{-\Delta Ct(50\ ng)}$ = 0.0139 Bias% $2^{-\Delta Ct(100\ ng)}$ = -6.9% Bias% $2^{-\Delta Ct(50\ ng)}$ = -8.0%

Table 1 Acceptance criteria and examples of experiments that are used to assess quality parameters. All samples are run in triplicates.

Multiplex Optimization

Lot #	Ct Value		
	$\Delta Ct_{(uniplex)}$	$\Delta Ct_{(multiplex)}$	$\Delta Ct_{(multiplex)} - \Delta Ct_{(uniplex)}$
Before Optimization			
Lot A	6.462	8.649	+2.187
Lot B	7.229	5.551	-1.678
Lot C	4.159	6.106	+1.947
Lot D	6.401	9.025	+2.624
After Optimization			
Lot 1	6.286	6.109	-0.177
Lot 2	5.621	5.978	+0.357
Lot 3	7.401	6.860	-0.541
Lot 4	7.051	6.874	-0.177
Lot 5	6.675	6.406	-0.269
Lot 6	7.353	6.980	-0.373

Table 2 A summary of data for multiplex optimization

- A multiplex system needs to be validated against a uniplex reaction.
- Optimization can be achieved by adjusting primer/probe concentration, annealing temperature, enzyme concentration and buffer concentration.
- Acceptance criteria: $|\Delta Ct(multiplex) - \Delta Ct(uniplex)| \le 1.000$.
- The dramatic difference in ΔCt between a uniplex and a multiplex system is optimized by doubling the concentrations of the reverse transcriptase and DNA polymerase in our experimental setup.

Linearity and Amplification Efficiency

Dilution Factor	Input RNA (ng/well)	FOXP3	GAPDH	Fold Change		
		Mean Ct	Mean Ct	ΔCt	$2^{-\Delta Ct}$	Bias%
1:1	400.0	30.524	22.755	7.769	0.0046	0.0
1:2	200.0	31.743	23.896	7.847	0.0043	-5.3
1:4	100.0	32.760	24.871	7.889	0.0042	-8.0
1:8	50.0	33.979	25.988	7.991	0.0039	-14.3
1:16	25.0	34.654	26.894	7.760	0.0046	0.6
1:32	12.5	35.390	27.947	7.443	0.0057	25.4

Table 3 A summary of data for the evaluation of linearity in multiplex format.

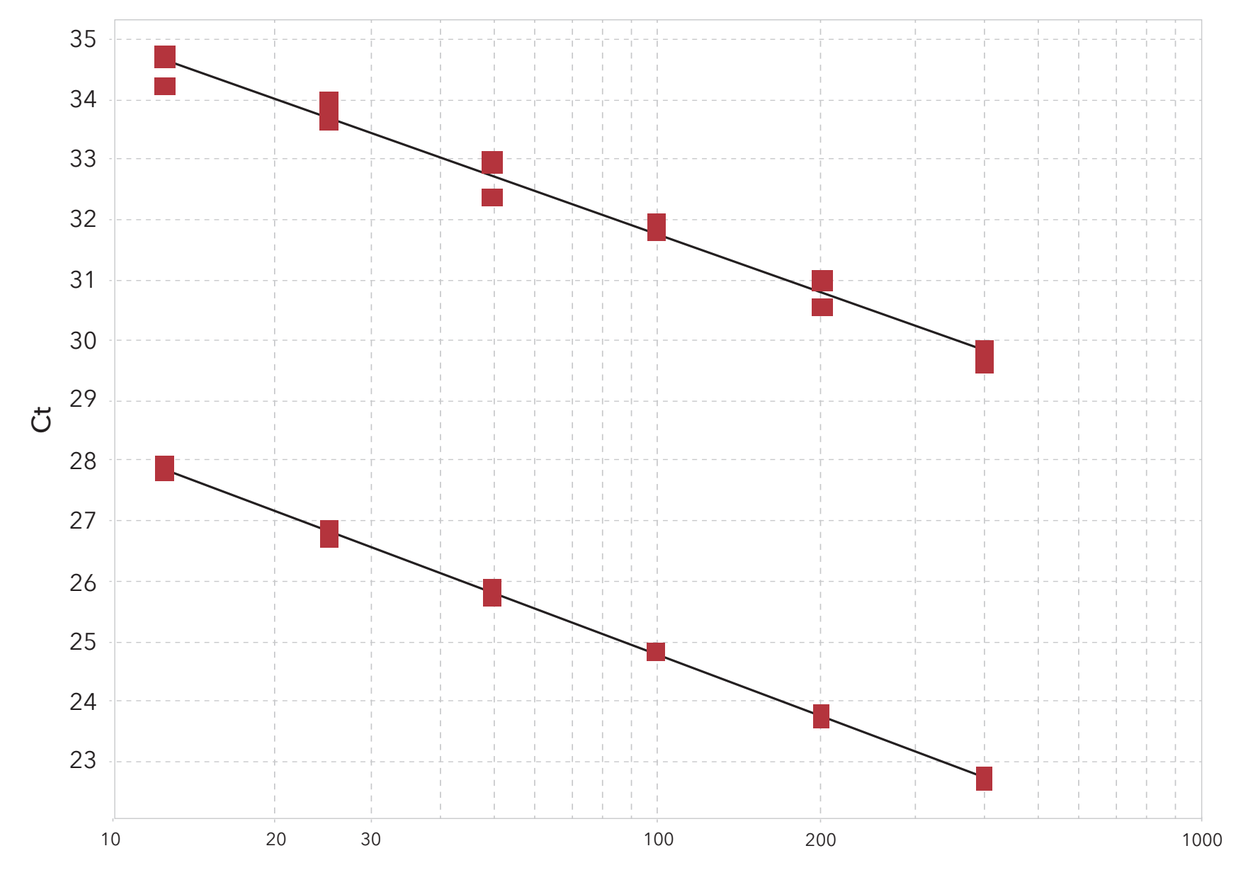
Ct
35
34
33
32
31
30
29
28
27
26
25
24
23
10
20
30
100
200
1000

Figure 2 Standard curves for amplification of FOXP3 and GAPDH genes. Variables for each curve are listed below:

Gene: FOXP3 Slope: -3.208 Y-Inter: 38.184 R^2: 0.984 Eff%: 104.994
Gene: GAPDH Slope: -3.411 Y-Inter: 31.591 R^2: 0.999 Eff%: 96.424

- Input RNA is prepared in a 2-fold serial dilution to assess linearity.
- Bias% = $(2^{-\Delta Ct(1:x)} - 2^{-\Delta Ct(1:1)})/2^{-\Delta Ct(1:1)}$ x 100%
- Bias% of all tested input RNA concentrations passed acceptance criteria.
- When multiplexing, reaction efficiencies of FOXP3 and GAPDH are highly comparable.

QC determination

Run #	HQC		MQC		LQC	
	Fold	Intra-CV%	Fold	Intra-CV%	Fold	Intra-CV%
Run 01	0.0143	4.5	0.0087	9.2	0.0058	7.6
	0.0132		0.0075		0.0051	
	0.0133		0.0074		0.0051	
Run 02	0.0150	9.1	0.0098	12.1	0.0073	16.7
	0.0147		0.0079		0.0057	
	0.0173		0.0081		0.0054	
Run 03	0.0209	4.1	0.0130	24.9	0.0086	6.5
	0.0218		0.0088		0.0077	
	0.0227		0.0085		0.0077	
Average	0.0170		0.0089		0.0065	
Inter-%CV	22.3		19.4		20.5	

Table 4 A summary of data for QC determination.

- Prior matrix screening selects three blood samples for QC determination.
- Three sets of QC samples are included in each run.
- The fold change of each QC level is determined by averaging 9 values.

Accuracy and Precision

Run #	HQC (0.0170)		MQC (0.0089)		LQC (0.0065)	
	Intra-CV%	Intra-Bias%	Intra-CV%	Intra-Bias%	Intra-CV%	Intra-Bias%
Run 01	2.3	-22.4	10.6	-9.0	8.6	4.6
Run 02	8.3	-24.1	13.0	13.5	6.4	-3.1
Run 03	2.5	-27.1	10.4	-13.5	12.2	-4.6
Run 04	10.9	-22.9	11.6	-30.3	18.5	-21.5
Inter-	6.5	-23.5	20.8	-10.1	14.7	-7.7

Table 5 A summary of intra- and inter-assay CV% and Bias%.

- Three sets of quality controls were prepared for each A&P run.
- Fold change at each QC level is used to calculate CV% and Bias%.
- Both of the inter-assay CV% and Bias% passed the acceptance criteria.

	Sample	Fold Change			
		ΔCt	$2^{-\Delta Ct}$	Mean	Bias%
Baseline	STL-1	8.260	0.0033	0.0035	NA
	STL-2	8.088	0.0037		NA
	STL-3	8.078	0.0037		NA
	STH-1	7.719	0.0047	0.0045	NA
	STH-2	7.875	0.0043		NA
	STH-3	7.841	0.0044		NA
Freeze-Thaw Stability (3 cycles at - 70°C)	STL-1	6.986	0.0079	0.0077	125.4
	STL-2	7.051	0.0075		115.5
	STL-3	7.017	0.0077		120.6
	STH-1	7.033	0.0076	0.0076	69.7
	STH-2	6.968	0.0080		77.5
	STH-3	7.118	0.0072		60.0

Table 6 A summary of freeze-thaw stability data.

- Freeze-thaw stability is assessed by comparing the $2^{-\Delta Ct}$ of RNA samples after 3 freeze-thaw cycles with that of the baseline values.
- Total RNA isolated from two separate individuals with a high and low expression of the target gene are regarded as stability high and low samples (STH and STL), respectively.
- Two sets of QC samples with pre-determined fold change levels (HQC, MQC and LQC) are included in each run.

Conclusion

- In the present work, we established an RT-qPCR workflow for qualitative and quantitative bioanalysis of mRNA transcripts isolated from human whole blood.
- Quality parameter need to be assessed while multiplex reactions need to be optimized prior to method development /validation runs.
- In our initial method development attempts, we have successfully conducted linearity evaluation, QC determination, A&P validation runs as well as tests for stability evaluation.

References

1. Clinical Pharmacology Considerations for the Development of Oligonucleotide Therapeutics-Guidance for Industry (Draft). U.S. Food and Drug Administration. 2. Bustin et al, The MIQE Guidelines: Minimum Information for Publication of Quantitative Real-Time PCR experiments. Clinical Chemistry 55(4): 611-622. 3. Nolan, Hands and Bustin, Quantification of mRNA using real-time RT-PCR. Nature Protocols Vol 1(3) 1559-1582. 4. Padhi et al, A PCR-based quantitative assay for the evaluation of mRNA integrity in rat samples. Biomolecular Detection and Quantification 15:18-23.