Comparison of Systemic Exposure in Pregnant and Non-Pregnant Sprague-Dawley Rats

Shuijin Yang, Lei Pan, Sue McPherson
WuXi AppTec, Suzhou, China

药明康德 WuXi AppTec

Abstract: 5096

Introduction

It is well known that pregnancy could theoretically induce changes in drug Pharmacokinetic or Toxicokinetic (TK) profiles by affecting the absorption, distribution, metabolism or renal excretion. However, there is little or no information available regarding whether pregnant animals have altered PK/TK as compared to non-pregnant animals. The Sprague-Dawley Rat is widely used in preclinical general toxicology studies and embryo-fetal developmental (EFD) studies and there have lots of available TK data in pregnant and non-pregnant rats to be analyzed.

Materials and Methods

Twenty small molecular projects (the relevant species was Sprague-Dawley rat) performed at the Testing Facility since 2018 were retrospectively reviewed to evaluate the potential differences of systemic exposure between pregnant and non-pregnant rats by calculating the $AUC_{0\text{-}24h}$, C_0 and/or C_{max} ratios. Dosing route was oral gavage or intravenous administration. There were 14 projects with the same dose levels and 6 projects with different dose levels in repeated dose studies and EFD studies. For the projects with different dose levels, the dose normalized $AUC_{0\text{-}24h}$ and C_{max} or C_0 ($AUC_{0\text{-}24h}$/D and C_{max} or C_0/D) were calculated first and then used for the systemic exposure comparison. Differences of more than 0.5 fold and less than 2 fold in TK parameters of both $AUC_{0\text{-}24h}$ and C_{max} or C_0 were not considered to indicate significant difference.

Results

The comparison data showed there was only 1 project that the pregnant rats had higher systemic exposure than non-pregnant rats (the C_{max} ratios ranged from 2.7 to 4.1 and $AUC_{0\text{-}24h}$ ratios ranged from 3.7 to 5.8). No significant differences in systemic exposure (in terms of $AUC_{0\text{-}24h}$, and C_0/C_{max} ratios) for all remaining 19 projects.

Table 1 Systemic Exposure Comparison

Data List	C_0 and/or C_{max} ratios	$AUC_{0\text{-}24h}$ ratios
1*	0.3~1.2	0.6~1.0
2	0.5~1.1	1.0~1.2
3	0.5~0.8	0.6~1.3
4	0.4~0.9	0.8~1.5
5	0.6~1.7	0.8~1.4
6	0.3~0.8	0.6~1.5
7	0.4~1.5	0.9~1.5
8*	1.0~1.2	1.2~1.5
9	0.5~0.7	0.7~0.9
10*	0.5~1.5	0.6~1.2
11	0.4~1.5	0.9~1.9
12	0.6~1.1	0.8~1.4
13	0.3~1.1	0.6~1.2
14*	0.5~0.8	0.5~0.9
15	0.8	0.8~1.5
16*	**2.7~4.1**	**3.7~5.8**
17	0.9~1.2	0.7~1.1
18*	2.1~2.3	1.3~1.6
19	1.4~2.4	0.7
20	0.9~1.4	1.3~1.8

*Projects with different dose levels in repeated dose studies and EFD studies.

Conclusion

In conclusion, for most of drug candidates, there were no significant differences in systemic exposure between the two different physiological status (pregnancy vs non-pregnancy) in rats. However, it is still recommended to perform dose range finding study in pregnant animals prior to initiation of definitive EFD study to avoid unexpected maternal toxicity, embryotoxicity or fetotoxicity due to potential higher systemic exposure induced by pregnancy.

Acknowledgements

The authors would like to acknowledge the contributions made by the team at WuXi AppTec.