Method Development Optimization for Intravesical Instillation in the Rat

Na Liu, Leo Pan, Fuqiang Wang, Sue McPherson
WuXi AppTec, Suzhou, China

Abstract: 3071

Introduction

Bladder cancer is one of the 10th most common cancers globally and ranks 13th in terms of deaths with approximately 550,000 new cases annually. Intravesical instillation therapy is aneffective alternative treatment in the treatment of such cancers. Preclinical evaluation of these therapies requires intravesical installation. This is not a straightforward route of administration, and the following describes the optimization of the instillation to enable the successful evaluation of these drugs.

Methods

By using anesthesia isoflurane, it was possible to extend the formulation dwelling time to 1.5 hours. To refine the method, the following three measures were taken: (1) the 18 Gauge endovenous catheter was replaced with 22 Gauge sterile plastic feeding tube (22 gavage × 25 mm) to allow a more smooth insertion into the urethra; (2) a heparin cap was connected with the needle to avoid the leakage of infused formulation when the syringe was removed from the infusion needle; (3) a custom-made rat stretcher (28 cm×10 cm) was used to avoid the indwelling needle falling from the bladder when the anesthetized rat was transferred to the isoflurane anesthesia chamber. During the 1.5 hours anesthesia period, the animals were closely monitored until removal 1.5 hours post dose. At recovery the needle fell out from bladder along with urine with spontaneous micturition.

Results and Conclusions

Procedure-related findings included transient red urine discoloration noted on the first 3 weekly dosing days, increased incidence in urine occult blood, protein and red blood cells (Table 1) and microscopic findings of minimal or mild multifocal mixed infiltration in the lamina propria and urothelium of pelvis of the unilateral or bilateral kidney, minimal or mild multifocal neutrophilic infiltration in the mucosa of the urinary bladder, minimal multifocal mixed/ neutrophilic infiltration and focal hemorrhage in the mucosa, and minimal increased mitotic figures in the urothelium of the urethra in the dosing phase.

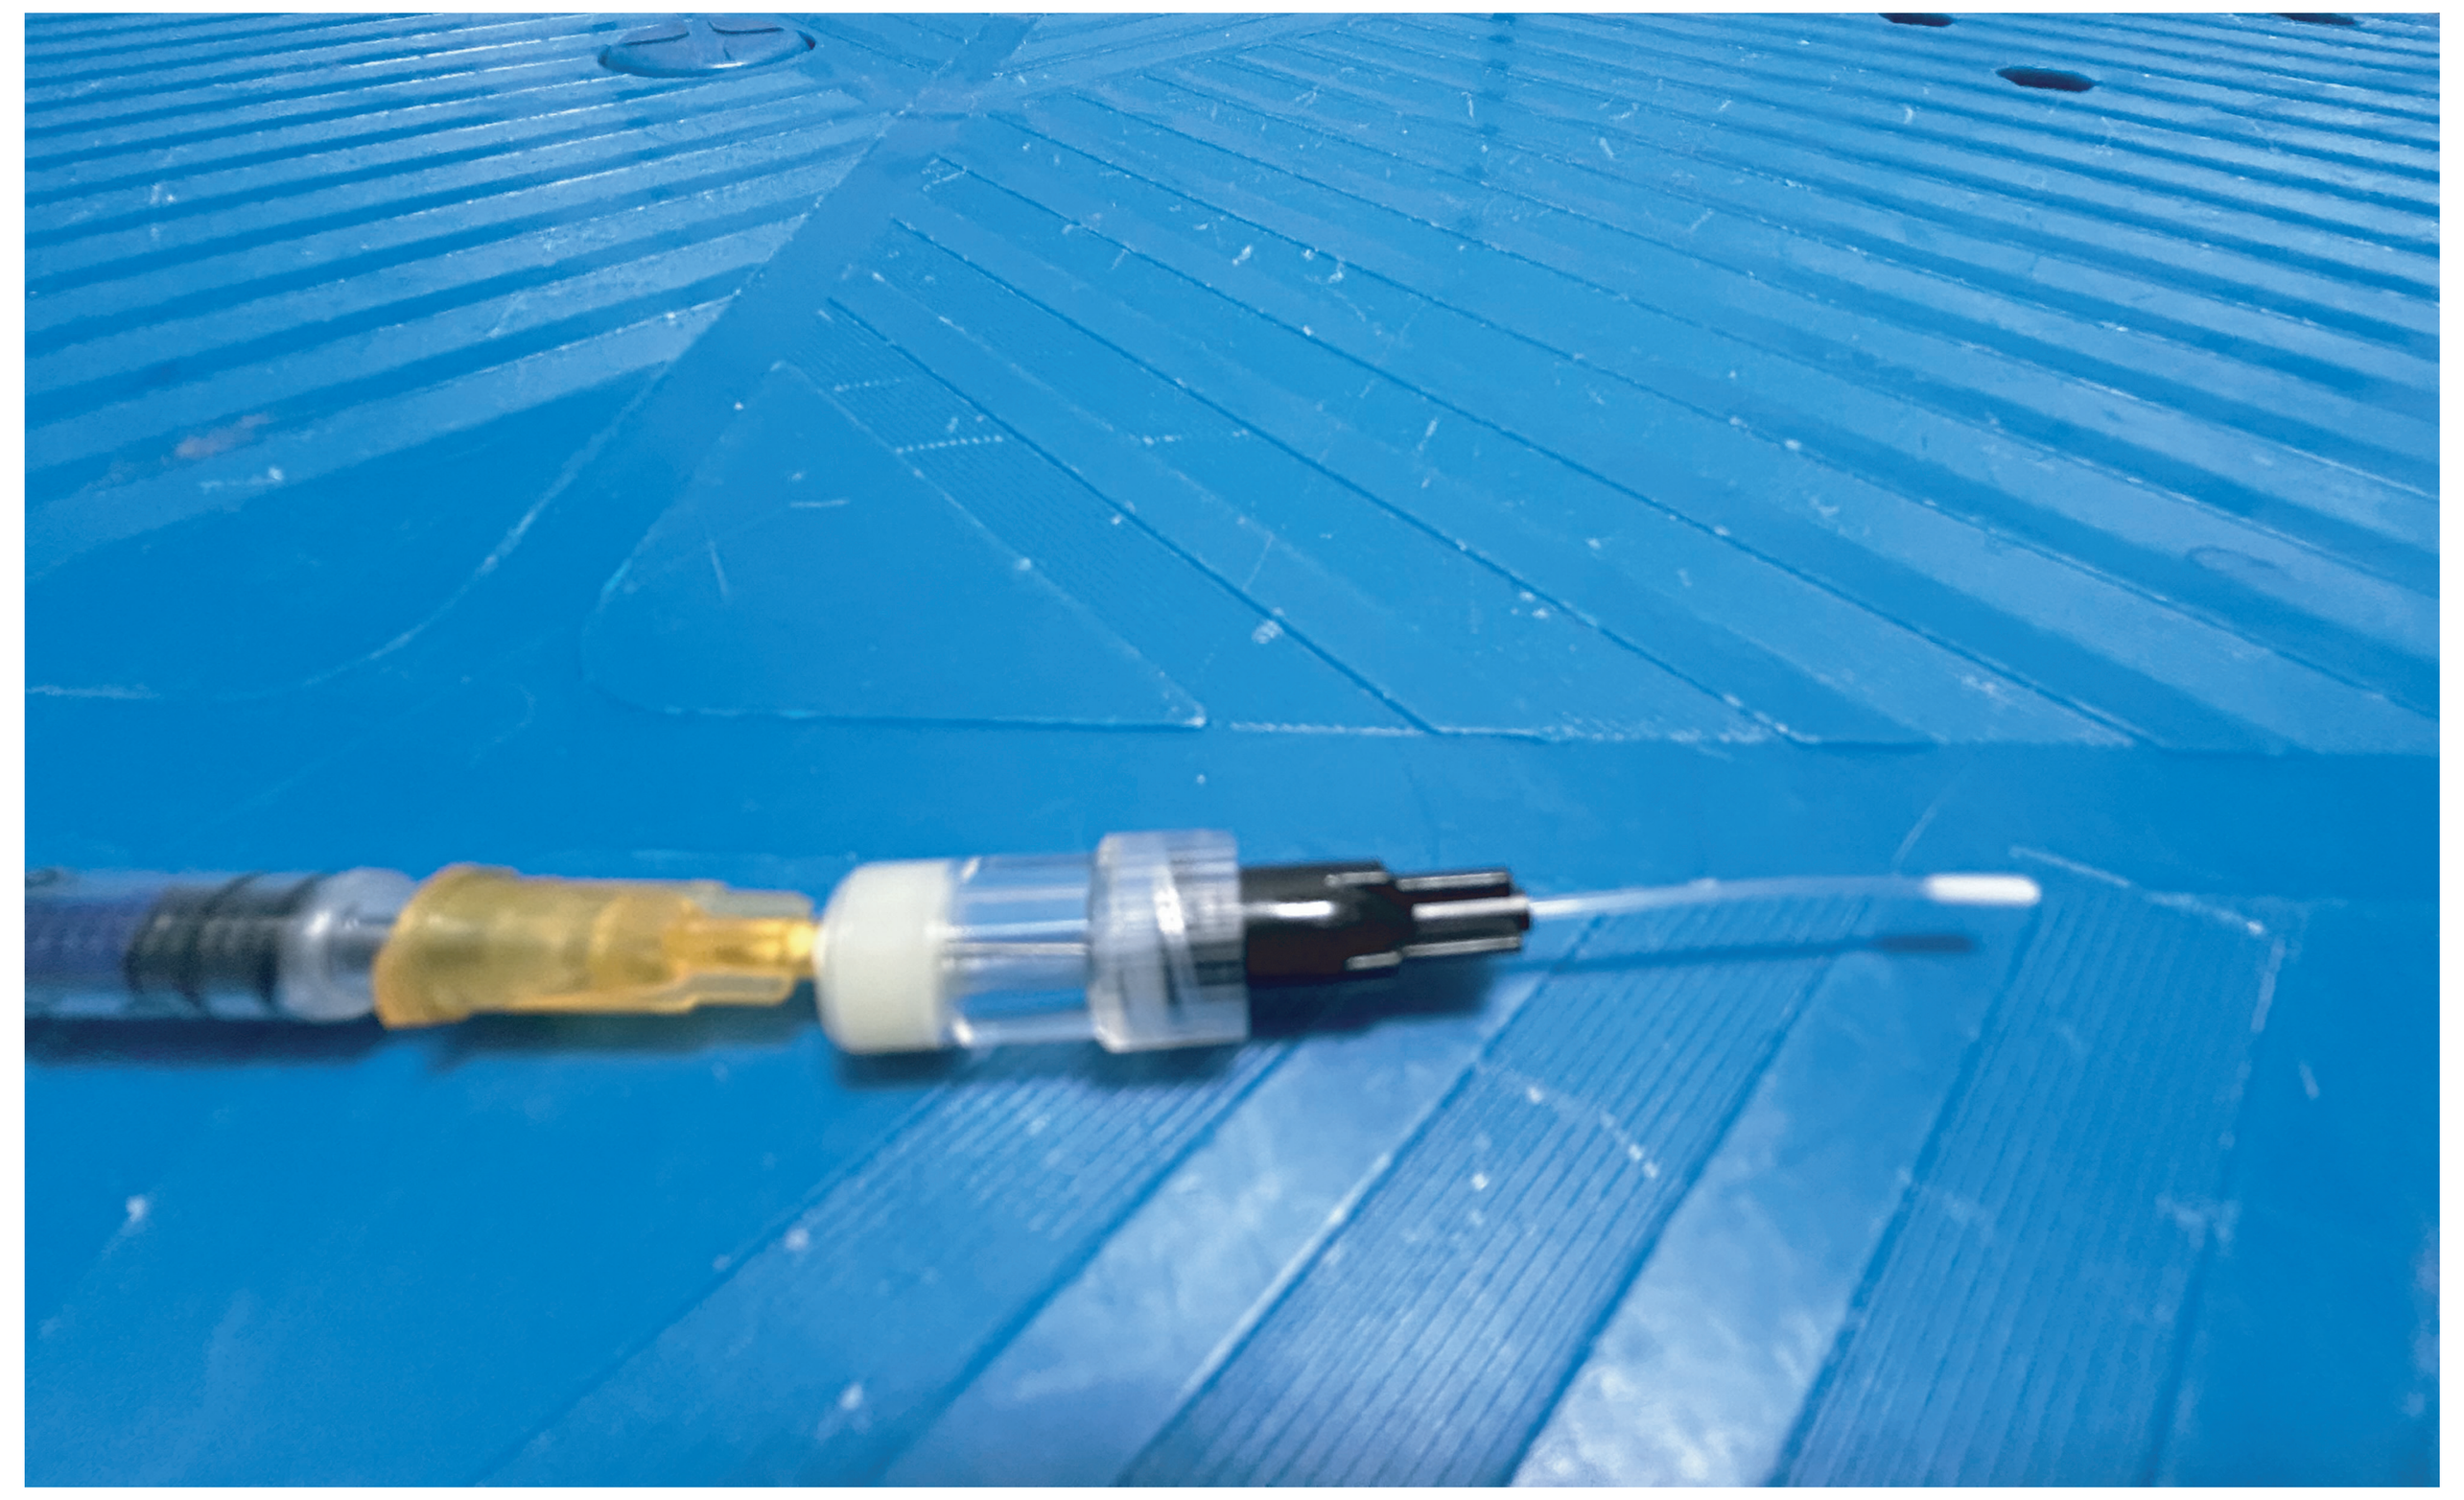

A heparin cap connected with the needle of 22 Gauge sterile plastic feeding tube

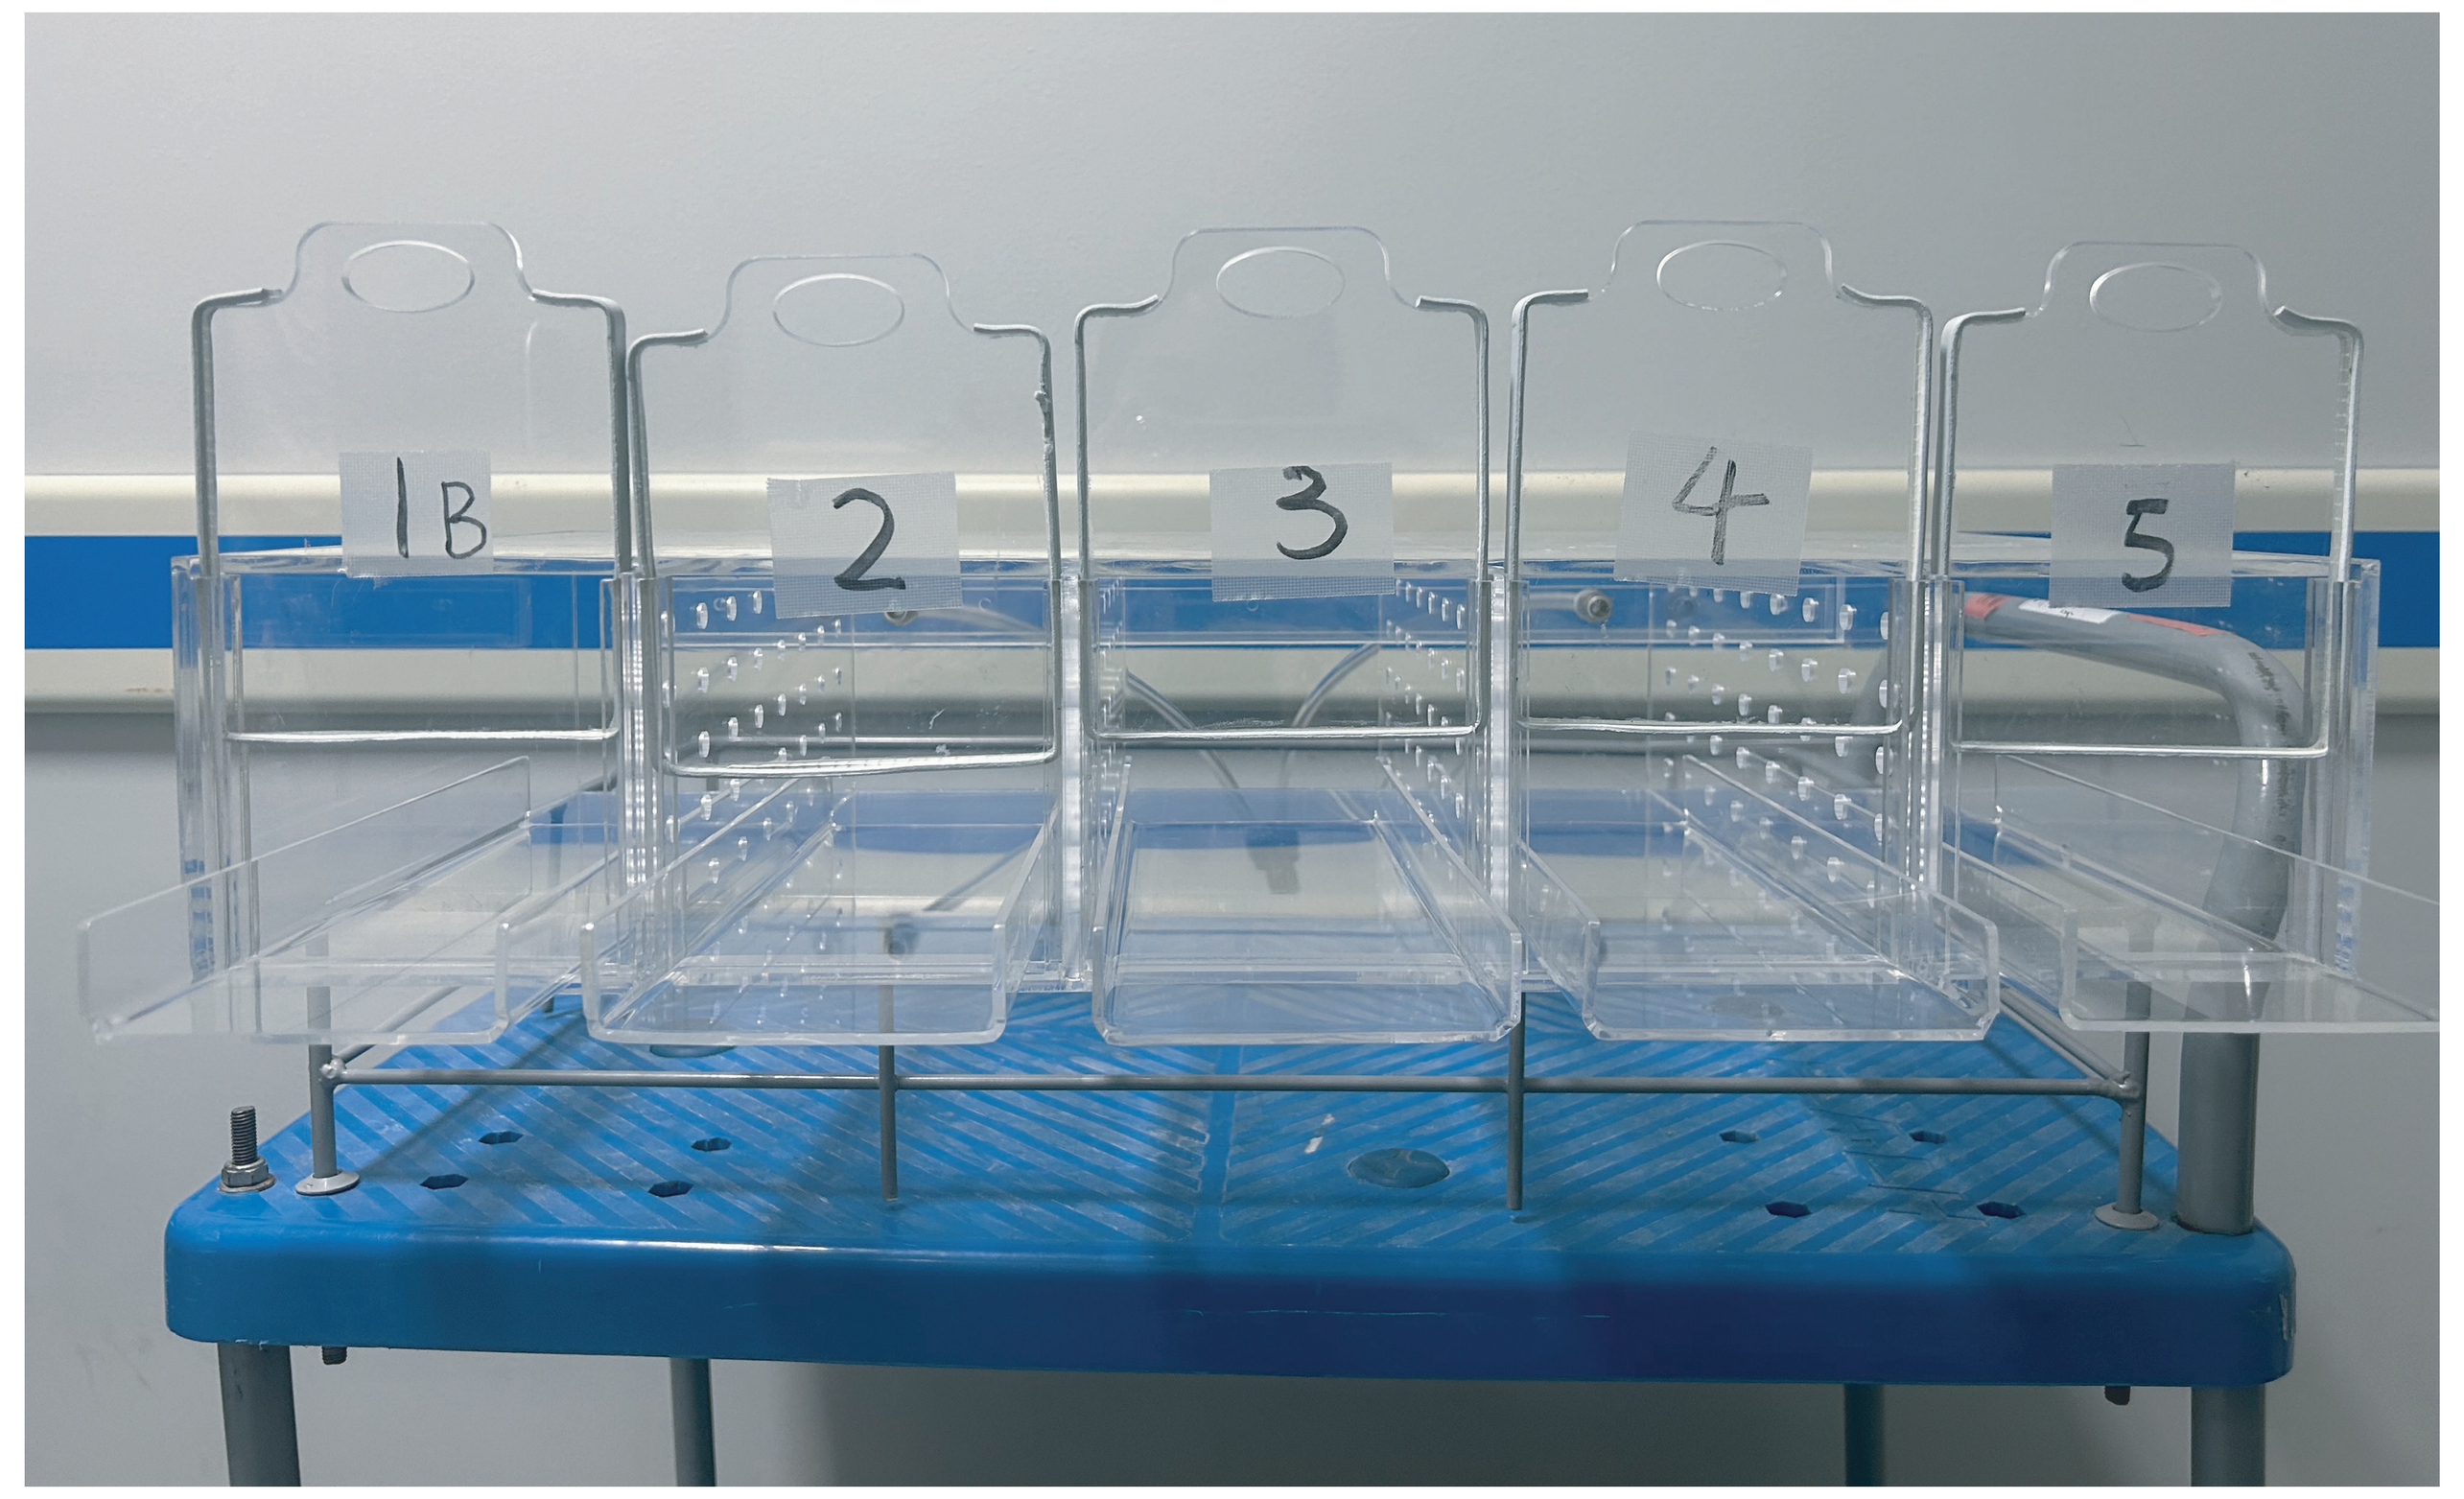
1B
2
3
4
5

The isoflurane anesthesia chamber with a custom-made rat stretcher (28 cm×10 cm)

There were no in life changes, or organ weight and macroscopic changes observed throughout the dosing and recovery phases. Indicating that with these method refinements intravesical instillation is considered suitable for the evaluation of drugs using this route of administration.

Table 1 Procedure-Related Urinalysis and Urine Sediment Changes in Dosing Phase

BLD	PRO	RBCu
2/15: +- 1/15: ++ 1/15: +++	5/15: +-; 1/15: +	2/15: ++ 1/15: +++

Note: Occult Blood (BLD): - = 0.00 mg/dL; +- = 0.03 mg/dL; + = 0.06 mg/dL; ++ = 0.15 mg/dL; +++ = 0.75 mg/dL.
Protein (PRO): - = 0 mg/dL; +- = 15 mg/dL; + = 30 mg/dL; ++ = 100 mg/dL; +++ = 300 mg/dL; ++++ = 1000 mg/dL.
Red Blood Cell/Erythrocytes (RBC/RBCu): -: negative; +: 1-10 (few); ++: 11-50 (moderate); +++: >50 (many).

Acknowledgments

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec.