Establishing the Preclinical Immunotoxicity Reference Based on Immunophenotyping Data in Bama Minipigs

Qinlei Gu[1], Mingdong Fu[1], Xiaochen Guo[1], Lan Li[2], Jinpeng Li[1], Xiaoying Jin[1]
[1]WuXi AppTec, 1318 WuZhong Avenue, Suzhou, Jiangsu, China. [2]WuXi AppTec, 288 Fute Zhong Road, Waigaoqiao Free Trade Zone, Shanghai, China.

Abstract: 5128

Background and Purpose

Therapeutics with immune modulating effect may occasionally have unintended effects and cause collateral damage to normal tissues. During clinical trials of immunomodulatory therapeutics, instances with severe, and even fatal adverse effects have been reported. These occurrences underscore the importance of immunotoxicity assessment in preclinical studies. One of the recommended parameters for preclinical immunotoxicity evaluation is immunophenotyping, which helps to evaluate specific cell populations and supports identification of potential biomarkers for clinical studies.

Minipigs have emerged as a valuable alternative in regulated toxicology studies, particularly for their anatomical and physiological similarities to humans. As outlined in ICH S11 guideline, they are a versatile model feasible for all administration routes, except inhalation, and are particularly popular in dermal studies. Although minipigs have a long history of domestication and utilization, their historical control data in immunotoxicology studies is relatively less well-established compared with other large animals, such as dogs and non-human primates. In this study, we surveyed one widely used Bama minipig breed for immune cell populations at baseline, in order to establish a reliable reference range for lymphocyte immunophenotyping.

Methods and Test System

Animals

Bama Minipig used in this report were originated from China. All animals were naïve and maintained in compliance with the Guide for the Care and Use of Laboratory Animals. All animal research was approved by the Institutional Animal Care and Use Committee in WuXi AppTec.

Immunophenotyping

Blood samples were collected with anti-coagulant potassium EDTA and washed with 2 mL of DPBS by centrifugation of 5 min at 400 g. Red blood cell was then lysed with BD Pharm Lyse lysing solution. Live/dead stain was stained with Fixable Viability Stain 510 for 10-15 minutes. Antibody cocktail was added which includes: FITC anti-pig CD45, PE-Cy™7 anti-pig CD3ε, Alexa Fluor® 647 anti-pig CD16, and PE anti-pig CD79a. Cells were incubated with antibody cocktail at room temperature for 30 minutes protected from white light. Cells were then washed and re-suspended with DPBS and analyzed using BD LSRFortessa™ flow cytometer with FACS Diva software.

Statistics

Two-sample t test was to calculated p-value for normally distributed data, and Mann-Whitney test was used for datasets that are not normal.

Results

Lymphocyte immunophenotyping was conducted in peripheral blood collected from 46 untreated male and 45 untreated female Bama minipigs. The baseline percentage of lymphocyte subpopulations was reported as total T cell (CD45+CD3ε+), B cell (CD45+CD3-CD16-CD79a+), and natural killer (NK) cell (CD45+CD3-CD16+CD79a-). To assess the distribution of the data, a variety of parameters were calculated and presented in Table 1, including the mean, standard deviation, minimum, 5th percentile, 25th percentile, median, 75th percentile, 95th percentile, and maximum. In summary, the 90% reference range (from 5th to 95th percentile) for lymphocyte subset proportion was as follows: total T cells range from 79.4% to 94.1% in male and from 79.2% to 97.2% in female; B cells range from 2.4% to 11.5% in male and from 1.6% to 12.8% in female; NK cells range from 2.7% to 12.4% in male and from 1.0% to 9.4% in female. In addition, a difference between male and female minipig was observed in NK cell baseline percentage (p-value=0.0047), with a higher NK proportion in male minipigs. Histograms and density lines in Figure 1 showed that T cell percentage from Bama mini pigs demonstrated a right-heavy tail distribution, and the B cell percentage showed left-heavy tail distribution. Both T cell and B cell parameters appears not following normal (Gaussian) distribution, whereases NK cell percentage distributed normally. Histograms for both male and female Bama mini pigs are included, and their distributions for each parameter were similar.

Table 1 Reference range for the percentage of lymphocyte subpopulations in Bama mini pigs

Percentage of Lymphocyte Subpopulation						
	% T cells		% B cells		% NK cells	
Parameter	Female (n = 45)	Male (n = 46)	Female (n = 45)	Male (n = 46)	Female (n = 45)	Male (n = 46)
Mean	87.8	87.7	5.9	4.8	5.1	6.6
Standard deviation	6.9	4.1	4.2	2.6	2.2	2.6
Minimum	50.9	74.5	1.3	1.1	0.8	1.9
5th Percentile	79.2	79.4	1.6	2.4	1.0	2.7
25th Percentile	85.3	85.2	3.2	3.2	3.9	4.5
50th Percentile	89.2	88.1	4.9	4.2	4.9	6.3
75th Percentile	91.3	90.7	7.7	5.2	6.2	8.0
95th Percentile	97.2	94.1	12.8	11.5	9.4	12.5
Maximum	97.6	95.0	25.5	15.8	10.9	13.9
Reference range (90% Confidence interval)	79.2-97.2	79.4-94.1	1.6-12.8	2.4-11.5	1.0-9.4	2.7-12.5

Figure 1 Percentage of lymphocyte subpopulations in Bama mini pigs. Histogram and density plot of A T cell, B B cell and C NK cell percentages and violin plots for D females and E males are presented. Dotted lines represents 25th, 50th, and 75th percentile.

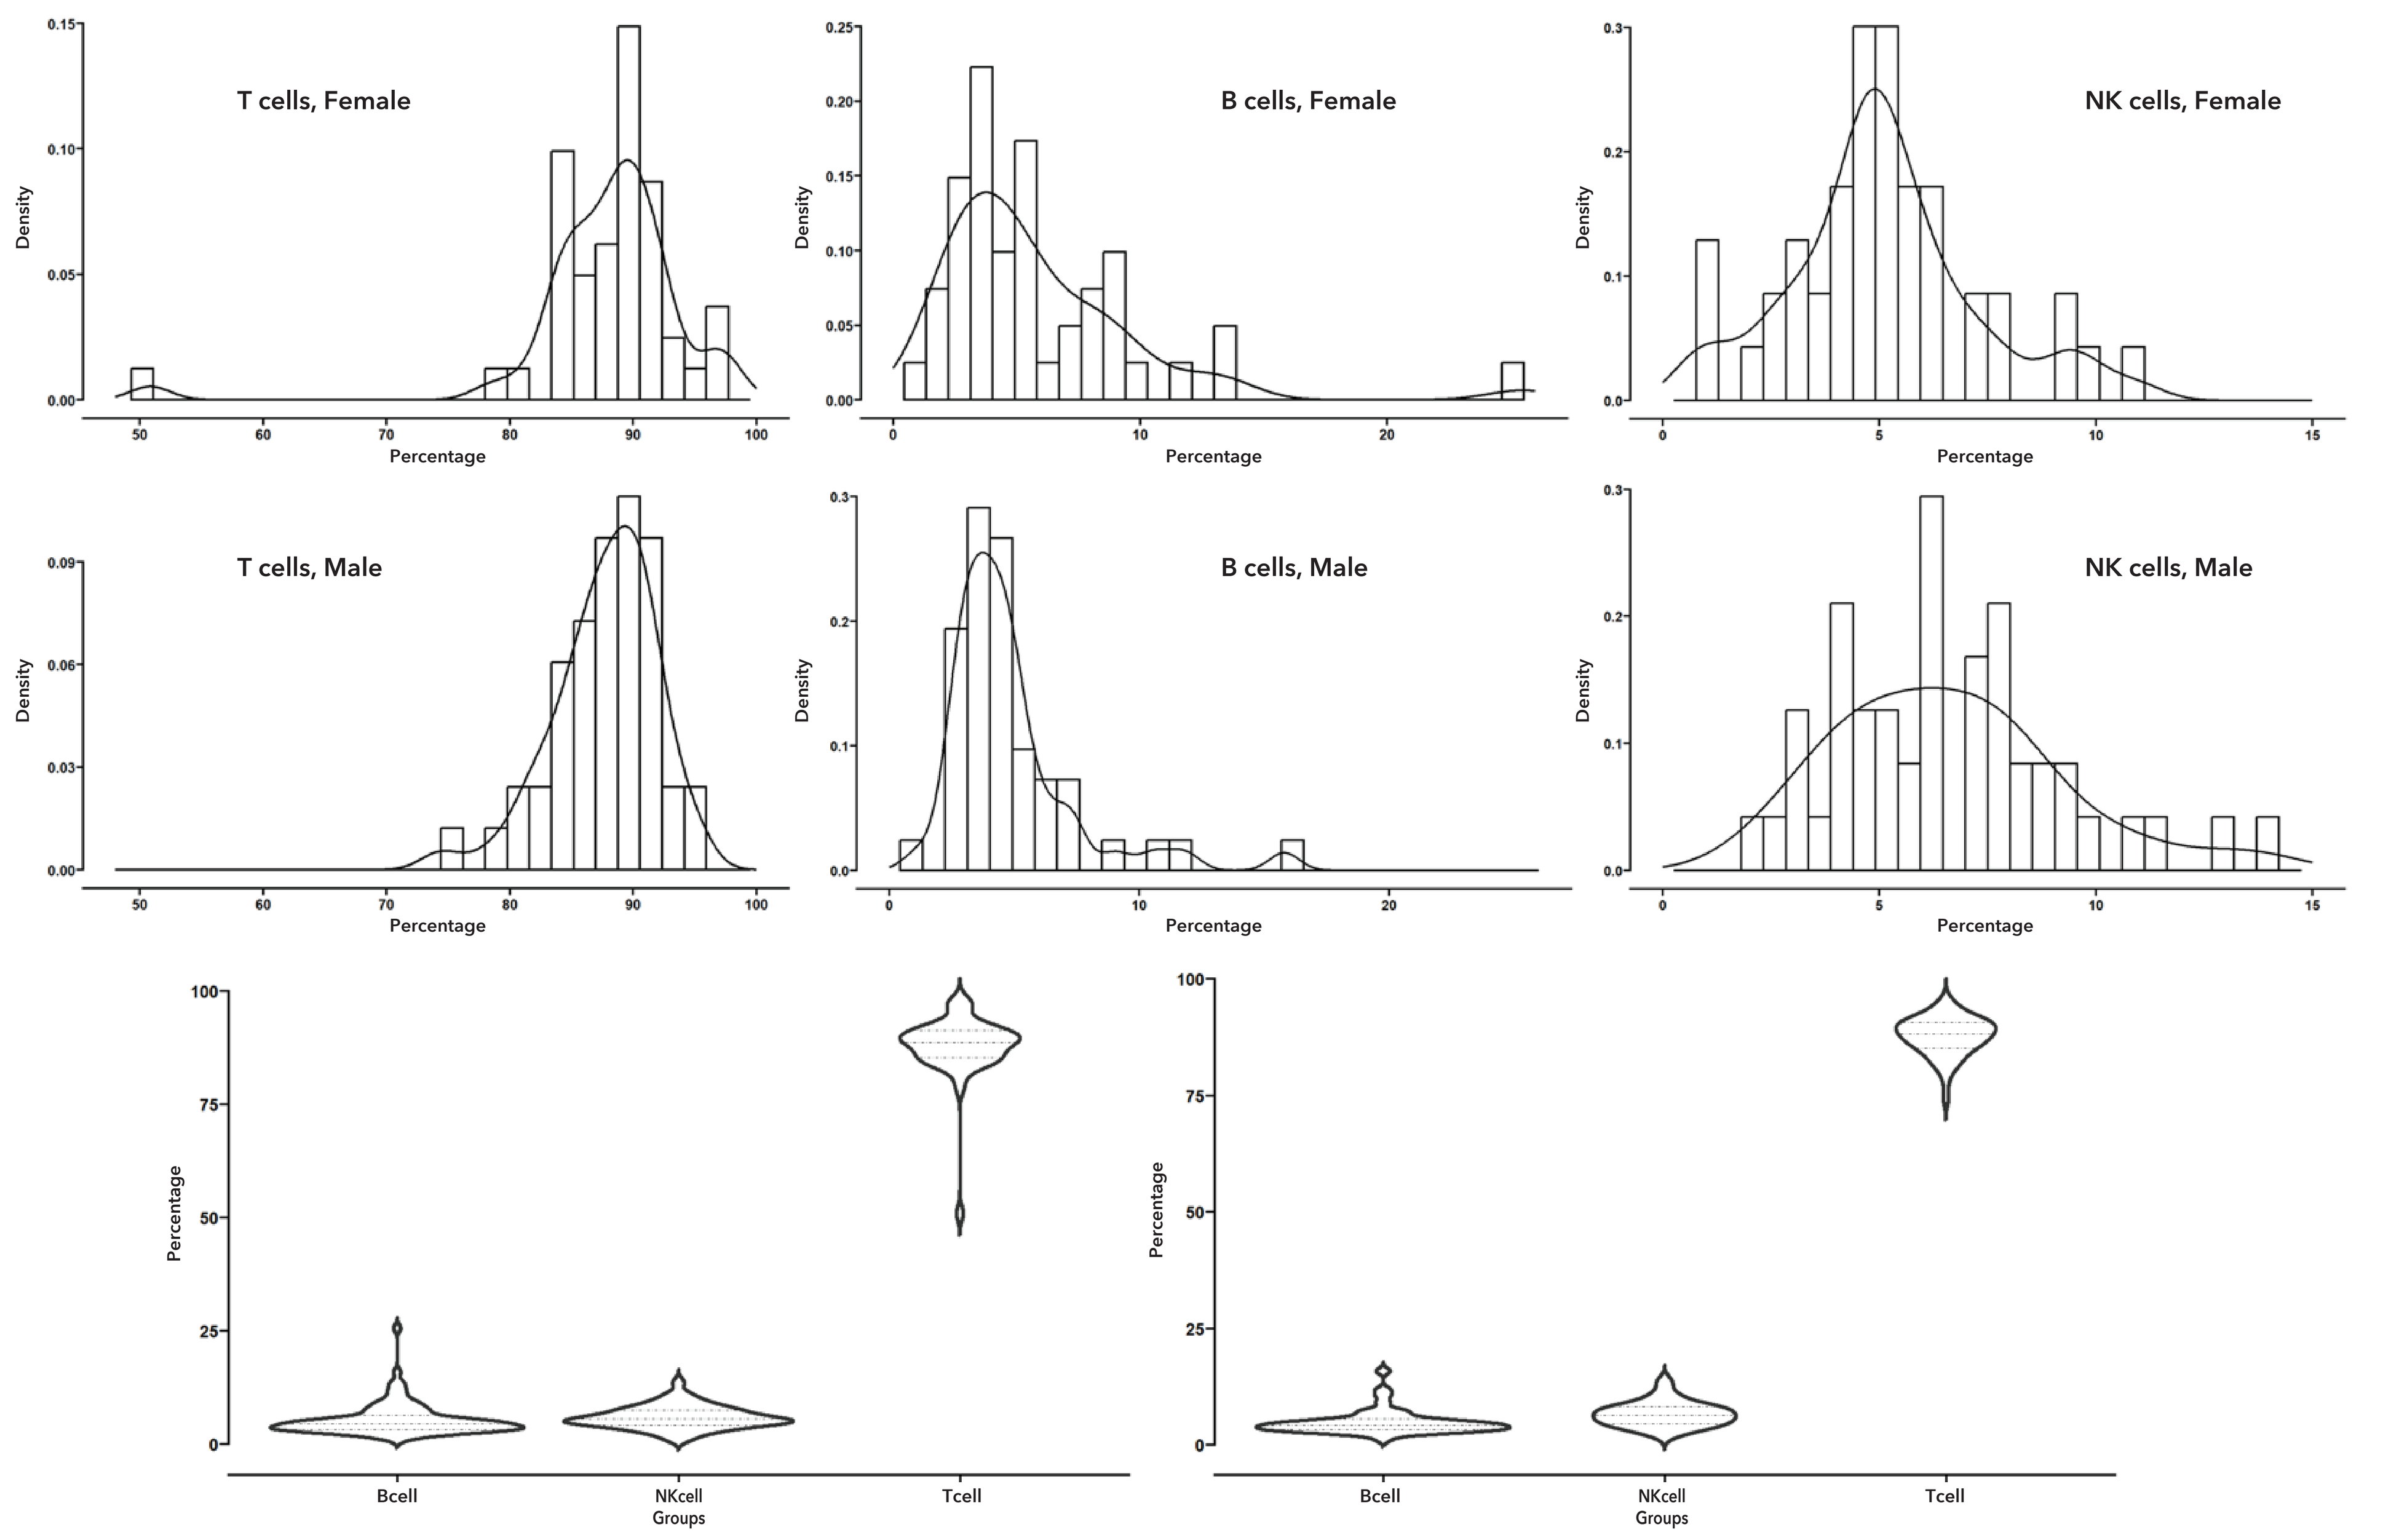
T cells, Female
B cells, Female
NK cells, Female
T cells, Male
B cells, Male
NK cells, Male
Density
Percentage
Bcell
NKcell
Tcell
Groups

Conclusion

Based on lymphocyte immunophenotyping data from 93 individual animals, a reference range for total T cells, B cells and NK cells in minipigs at homeostatic immune state has been established. This database could serve as a reliable reference, aiding the interpretation of preclinical immunotoxicity findings.

References

1. Tina Rubic-Schneider, Brigitte Christen, Dominique Brees, Michael Kammüller. Minipigs in Translational Immunosafety Sciences: A Perspective. 8-color whole blood immunophenotyping panel for the character. 2016. https://journals.sagepub.com/doi/full/10.1177/0192623315621628
2. Lunney JK, Van Goor A, Walker KE, Hailstock T, Franklin J, Dai C. Importance of the pig as a human biomedical model. Science Translational Medicine. 2021 Nov 24;13(621):eabd5758.
3. OMIP-107: rization and quantification of lymphocyte subsets and monocytes in swine - Arrigucci - 2024 - Cytometry Part A. https://onlinelibrary.wiley.com/doi/full/10.1002/cyto.a.