Retrospective Review of Tolerable Levels for Polyethylene Glycol in Non-Clinical Studies as a Vehicle Component

Jian Gu, Yan Zhang, Sue McPherson
WuXi AppTec, Suzhou, China

Abstract: 3014

Introduction

Polyethylene glycol (PEG) is a widely used polymer and characterized by its high water solubility and biocompatibility, making it a popular choice as a vehicle in various pharmaceutical and toxicological applications. This retrospective review aimed to determine the tolerable level for PEG of varying molecular weights used as vehicles in toxicology studies.

Methods

A retrospective review was performed regarding solvents used and their toxicity from approximately 1400 general toxicology studies conducted before 2021. Of these, approximately 80 studies utilized PEG as a vehicle component. The species involved primarily included rats, mice, monkeys, and dogs, with administration routes mainly including oral and intravenous dosing, and the studies were of varying duration from single dose to 39 weeks.

Results and Conclusions

For PEG-400 at concentrations of 40% to 100% with oral administration route, it could lead to abnormal animal feces (soft/watery/mucoid stool). Data showed that lower or equal to 30% is relatively safe with commonly used dosing volume of 10 mL/kg for rodents and 5 mL/kg for large animals. When intravenous (IV) injection for rats and dogs with dose volume of 6 or 5 mL/kg, greater or equal to 40% could lead to local irritation at the injection site with pathological changes (diffuse dark/ purple discoloration of the distal tail correlated microscopically with diffuse, marked necrosis of the vascular wall; diffuse, mild to marked necrosis of the epidermis and dermis, subcutaneous edema; and hemorrhage).

For PEG-300 and PEG-200 with oral administration route, the maximum proportion used was 40% and 25%, respectively, and well tolerated. And IV injection for 20% (v/v) PEG-200 was well tolerated. However, a potential effect of PEG-200 on the vacuolation in the adrenal zona fasciculata was noted in a 28-day rat study, in which 5% (v/v) Ethanol, 5% (v/v) Solutol, and 20% (v/v) PEG200 was used. Although it was a rare finding in our database and not excluded inducing by Solutol, the effect of PEG 200 was also noteworthy. The data were shown in the Table 1.

Therefore, PEG can serve as a favorable vehicle component for toxicology studies, with PEG-400 being the more commonly used. The primary adverse reactions observed are abnormal feces and local irritation. The review presents the maximum tolerable proportions of Polyethylene Glycol as a vehicle component and could be a useful tool in selecting suitable vehicles for toxicology studies.

Acknowledgments

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec.

Table 1 Tolerable Levels for Polyethylene Glycol

Species	Route	Duration	Volume (mL/kg)	Adverse Reactions/Toxicity	Vehicle Components
Rats	Oral	11 days	20	Well tolerated	40%(v/v) PEG300+5%(v/v) Ethanol+55%(v/v) [18.18% (w/v) E TPGS in purified water]
		8 weeks	5	Well tolerated	95% (v/v) PEG400 +5% (v/v) Tween 80
		28 days	10	Soft stool	40% (v/v) PEG400 in purified water
		28 days	10	Vacuolation in the adrenal zona fasciculata	5% (v/v) Ethanol+5% (v/v) Solutol+20% (v/v) PEG200 in purified water
	IV Injection	32 days	2	Well tolerated	20% (v/v) PEG 300+10% (v/v) Kolliphor EL+10% (v/v) ethanol+30% (v/v) 100 mM PBS (pH 7.4) in sterile water for injection
		7 days	6	Skin discolored (tail/distal, purple) and swelling (tail/distal, slight); diffuse dark/purple discoloration in the distal tail	5%(v/v) DMSO+45%(v/v) PEG400 +50%(v/v)[10%(w/v)HP-β-CD in water]
Mice	Oral	13 Weeks	10	Well tolerated	30%(v/v) PEG400+60%(v/v) PHOSAL 50PG+10%(v/v) Ethanol
	IP	28 Days	10	Well tolerated	20%(v/v) PEG400 in PBS, pH: 8.0±0.2
	IV Injection	14 Days	10	Well tolerated	10% (v/v) PEG400+90%(v/v)[10%(w/v) HP-β-CD in Sterile Water for Injection]
Dogs	Oral	14 days	10	Well tolerated	40%(v/v) PEG400+10%(v/v) Cremophor EL in purified water 0.5% (w/v) MC in purified water
		28 days	5	Well tolerated	5% (v/v) DMSO+30% (v/v) PEG400+65% (v/v) [20%(w/v) SBE-β-CD in purified water]
		13 weeks	5	Well tolerated	40% (v/v) PEG300+10% (v/v) Cremophor RH40+50% (v/v) purified water
		7 days	10	Soft/watery stool	40% (v/v) PEG400 in purified water
		14 days	5	Soft stool	40% (v/v) PEG400 in purified water
		28 days	5	Small vomitus, watery/soft/mucoid stool	40%(v/v) PEG400+60%(v/v) [50% (w/v) HP-β-CD in saline]
		28 days	9 5	Soft/watery stool	40% (v/v) PEG300+10% (v/v) Cremophor RH40+50% (v/v) purified water
		13 weeks	1.5	Soft/watery stool	Polyethylene glycol 400(PEG400)
	IV Injection	7 days	5	Soft stool and swelling of the tail	40%(v/v) PEG400 in 0.9% Sodium Chloride Saline Solution
Monkeys	Oral	39 weeks	4(BID)	Well tolerated	25%(v/v) PEG 200+75%[0.5%(w/v) MC (400cP) in purified water]
		7 days	10	Soft/watery stool	50%(v/v) PEG 400+10%(v/v) solutol in Purified Water
	IV Infusion	2 weeks	5	Well tolerated	1:10 ratio of Diluent A[50% Propylene Glycol (PG) +50% Ethanol (v/v)] in Diluent B [PEG 400 /Solutol HS 15/Purified water (5/7/88, v/v/v)]