Consideration of Tongue Toxicity in Ocular Toxicity Studies

ZQ Tan, YQ Ji, XD Qiao, WM. Harrison, YD Zhou
Toxicology, WuXi AppTec, Suzhou, China

药明康德 WuXi AppTec

Abstract: 3457

Background

Dysgeusia, or tongue related side effects, are not rarely reported in clinical cases of many eye drops, while changes in tongue are rarely noted in nonclinical studies. It indicates that the tongue may be ignored in the tissue list when performing histopathology in many nonclinical studies.

This increases the risk of missing tongue toxicity for ophthalmic products, especially eye drops.

Purpose

To present a case of ocular toxicity study with tongue abnormalities in histopathology, and emphasize that tongue should be listed for histopathology study in ocular toxicity studies.

Methods

A 28-day repeated dose ocular toxicity study of eye drops in rabbits was reviewed. The OE findings and histopathology results were reviewed and analyzed.

Study Design

Treatment

- The eye drops was a reformulated small molecule drug. Vehicle and Test article at three dose levels (0.08, 0.12, 0.16 mg/eye/day) was delivered to a unilateral eye of New Zealand white rabbit by topical ocular instillation, four times daily, for 28 consecutive days followed by a 14-day recovery period.

Ocular Examinations

- Ophthalmic Examinations - handhold slit lamp, and indirect ophthalmoscope, Pre, Week 1, Week 4 and end of recovery;
- Intraocular Pressure (IOP): Pre, Week 4 and end of recovery;
- Entire Retinal Function: fERG, Pre, Week 4 and end of recovery;

Termination and Histopathology

- Animals were necropsied on Day 29 and Day 43, respectively.
- Histopathology evaluation was conducted on a full panel of tissues in animals from the control and the high-dose groups, all gross lesions observed in animals from all groups, and ocular tissues and the potential target organ (tongue) in animals from the low and the mid-dose groups.

Results

Ophthalmic Examination Findings

- Slight conjunctiva hyperemia and congestion were observed during the experimental period. They were noted in both untreated and treated eyes and were in low frequency. It deemed to be not test article or vehicle article related.

Intraocular Pressure (IOP)

- There were no test article-related or vehicle-related changes in intraocular pressure in this study.

Electroretinography (ERG)

- There were no test article-related or vehicle-related changes in ERG.

Histopathology Findings

- No ocular abnormalities or systemic toxicity were noted except some histologic findings in the tongues of animals given ≥0.08 mg/eye/day.
- Epithelium degeneration on the dorsal surface of the tongue was minimal to moderate and characterized by swollen, pale-stained epithelial cells, and parakeratotic hyperkeratosis;

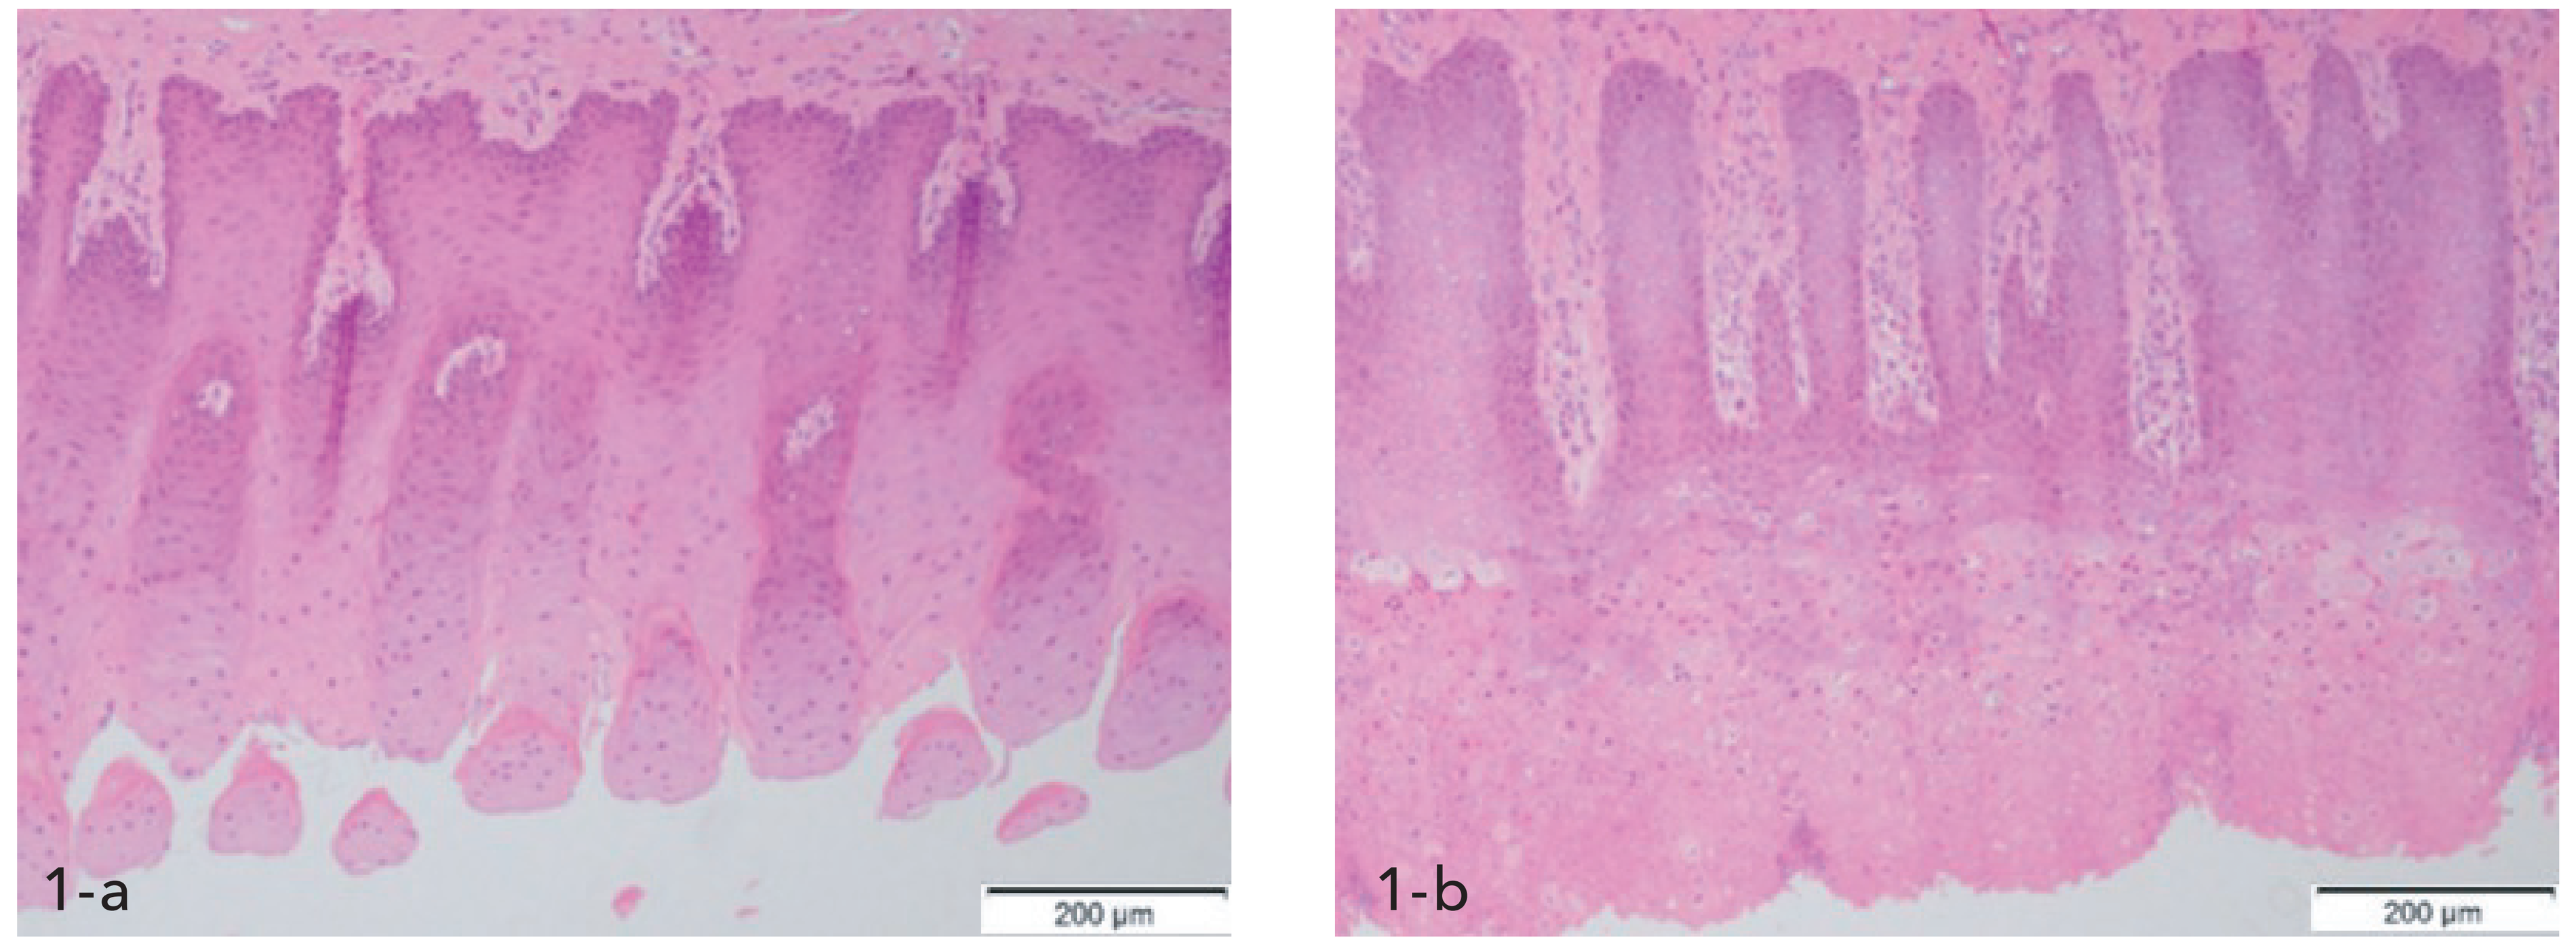
1-a
200 μm
1-b
200 μm

Figure 1 a. Vehicle control, epithelium of the tongue, normal; b. high dose group, the tongue was noted with epithelium degeneration, characterized by swollen, pale-stained epithelial cells, and parakeratotic hyperkeratosis.

- In addition, when moderate severity, the tongue degeneration was accompanied with necrosis and inflammation.
- But there was a complete reversal of the tongue changes observed at the end of the dosing phase followed by a 14-day recovery period.

Discussion & Conclusion

A dose-response relationship of microscopic findings in both the incidence and the severity was observed. Based on this case, definite test article related changes in the tongue were provided though these abnormalities recovered at the end of the study.

In the nonclinical report of lifitegrast[1], the tongue was identified as a potential target in both dogs and rabbits in the 39-week ocular toxicity studies. In dogs, minimal granulomatous inflammation of the tongue was noted in one high-dose male and one high-dose female at the end of the dosing phase and one high-dose female at the end of the recovery phase. In rabbits, a dose dependent increase in the incidence and severity of myofiber regeneration of the tongue was observed at all dose levels. It is unclear whether these findings are related to clinically observed dysgeusia.

As for learning points, attention should be paid to tongue toxicity in nonclinical safety evaluation of eye drops, and the tongue should be also added in the routine tissue list for histopathology study in ocular toxicity studies.

Reference

[1] FDA CDER, Pharmacology Reviews of Lifitegrast Ophthalmic Solution, 2016, 1-22.

Financial Disclosure

There are no potential conflicts of interest to disclosure.

Contact Information

zhou_yongdong@wuxiapptec.com