Spontaneous Ocular Abnormalities in Sprague-Dawley Rats

HJ Cheng, WM Harrison, YD Zhou
Toxicology, WuXi AppTec, Suzhou, China

Abstract: 3458

Objective

To collect in-house data of spontaneous ocular abnormalities in Sprague-Dawley (SD) rats and compare them with reported data of SD rats from the Taisho Pharmaceutical Research Center (Saitama, Japan).

Methods

Data Collection

- In the WuXi AppTec (Suzhou, China), 111016 male and female SD rats (6 to 9 weeks of age) purchased from Vital River Laboratory Animal Technology Co., Ltd (Beijing or Zhejiang) from 2020-01-02 to 2023-07-31.
- The incidences of the spontaneous abnormalities in the eyes of all the animals were recorded in the pre-treatment ophthalmologic examinations in toxicity studies at our facility from 2020-01-02 to 2023-07-31.

Ophthalmologic Examination

- Binocular indirect ophthalmoscope
- Slit lamp

Data Comparison

- In the Taisho Pharmaceutical Research Center (Saitama, Japan), 2033 male and 1322 female SD rats (4 to 7 weeks of age) were purchased from Charles River Laboratories Japan (Tsukuba, Atsugi or Hino Breeding Center, Ibaraki, Kanagawa or Shiga, Japan) from 2004 through 2015.
- Compared and analyzed the incidences of the spontaneous ocular abnormalities of SD rats between the WuXi AppTec (Suzhou, China) and Taisho Pharmaceutical Research Center (Saitama, Japan).

Results

Incidences of the Spontaneous Abnormalities in the WuXi AppTec (Suzhou, China)

- The incidence of corneal opacity was 83%.
- The incidence of lens opacity was 10%.
- The incidence of persistent hyaloid artery was 6%.
- All other ocular abnormalities of the globe, conjunctiva, cornea, anterior chamber, lens, iris, vitreous, and choroid or retina occurred at an incidence of less than 1%.

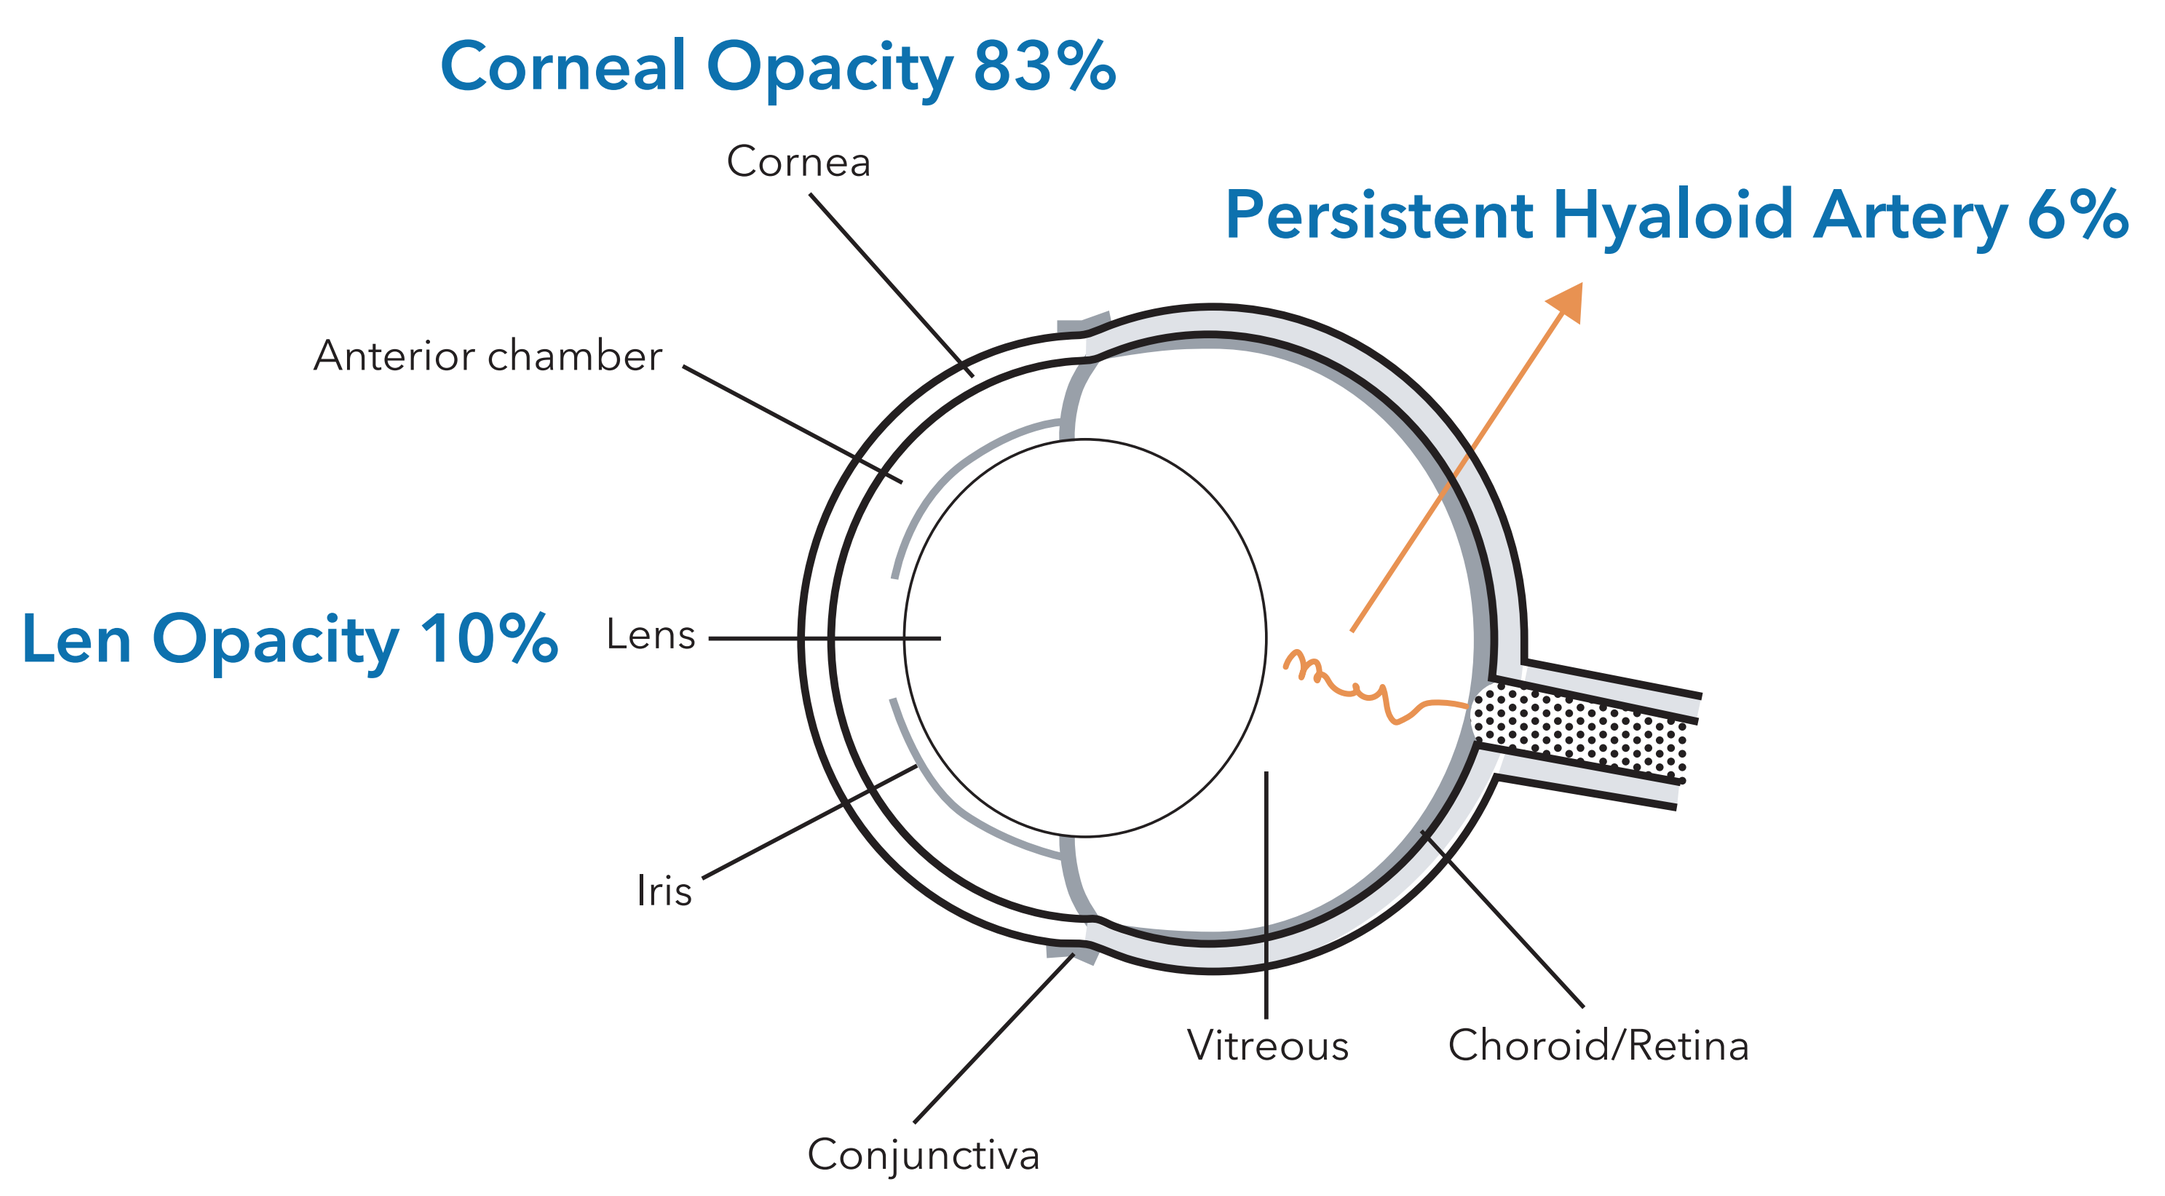
Corneal Opacity 83%
Cornea
Persistent Hyaloid Artery 6%
Anterior chamber
Len Opacity 10%
Lens
Iris
Vitreous
Choroid/Retina
Conjunctiva

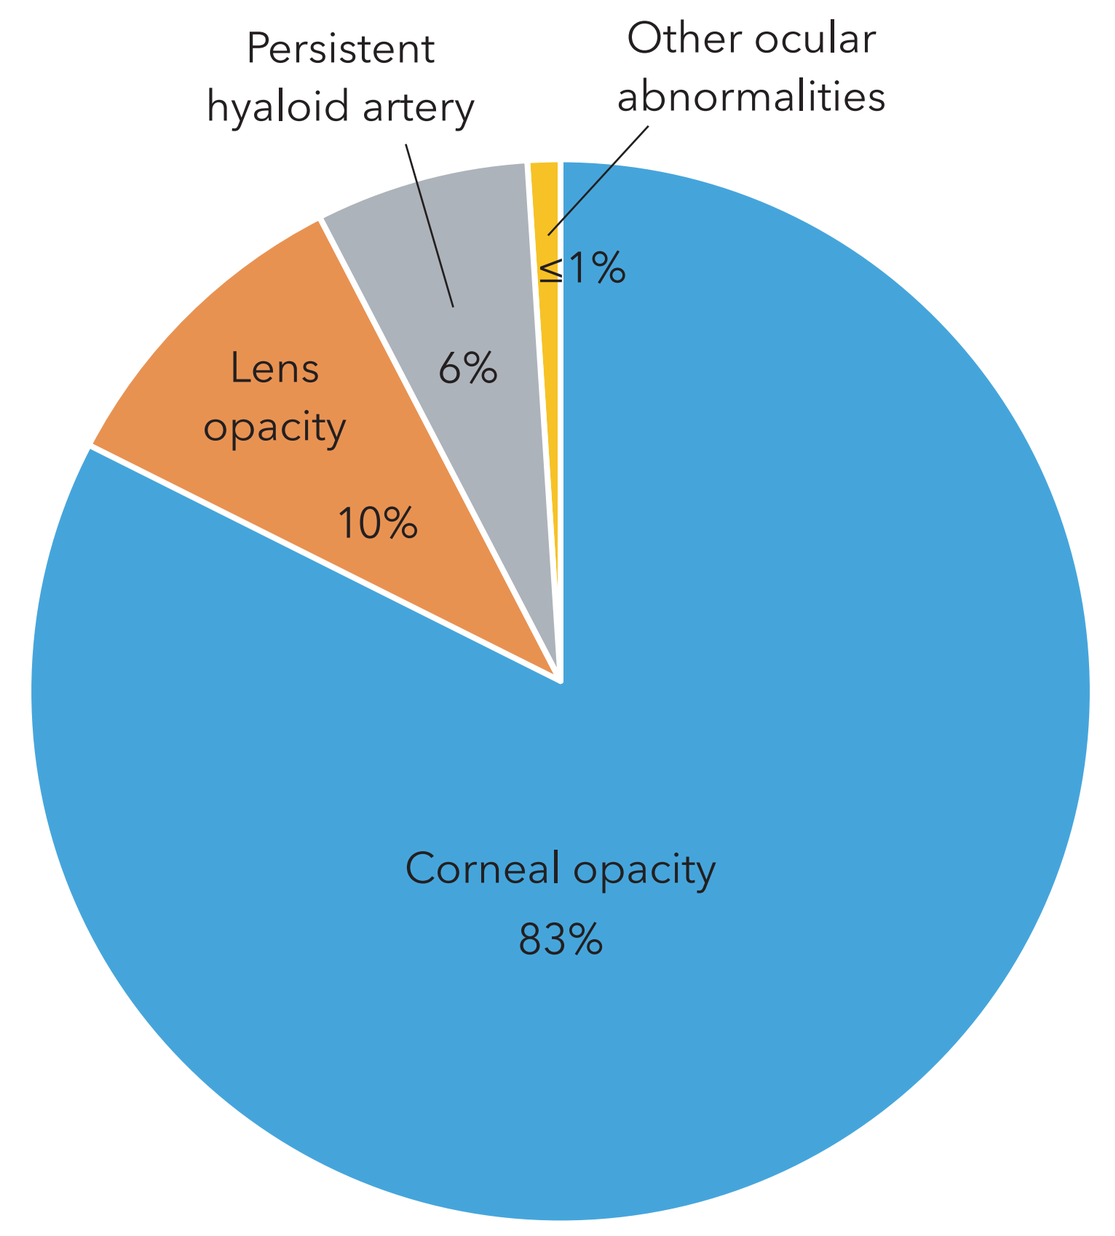
Persistent hyaloid artery
Other ocular abnormalities
≤1%
6%
Lens opacity
10%
Corneal opacity
83%

Incidence of Lens Opacity in Five Areas in the WuXi AppTec (Suzhou, China)

- The incidence of lens opacity in the nuclear areas was 83%.
- The incidence of lens opacity in the posterior capsule areas was 9%.
- The incidence of lens opacity in the anterior capsule areas was 7%.
- The incidence of lens opacity in the posterior cortical areas was 0.08%.
- The incidence of lens opacity in the anterior cortical areas was 0.05%.

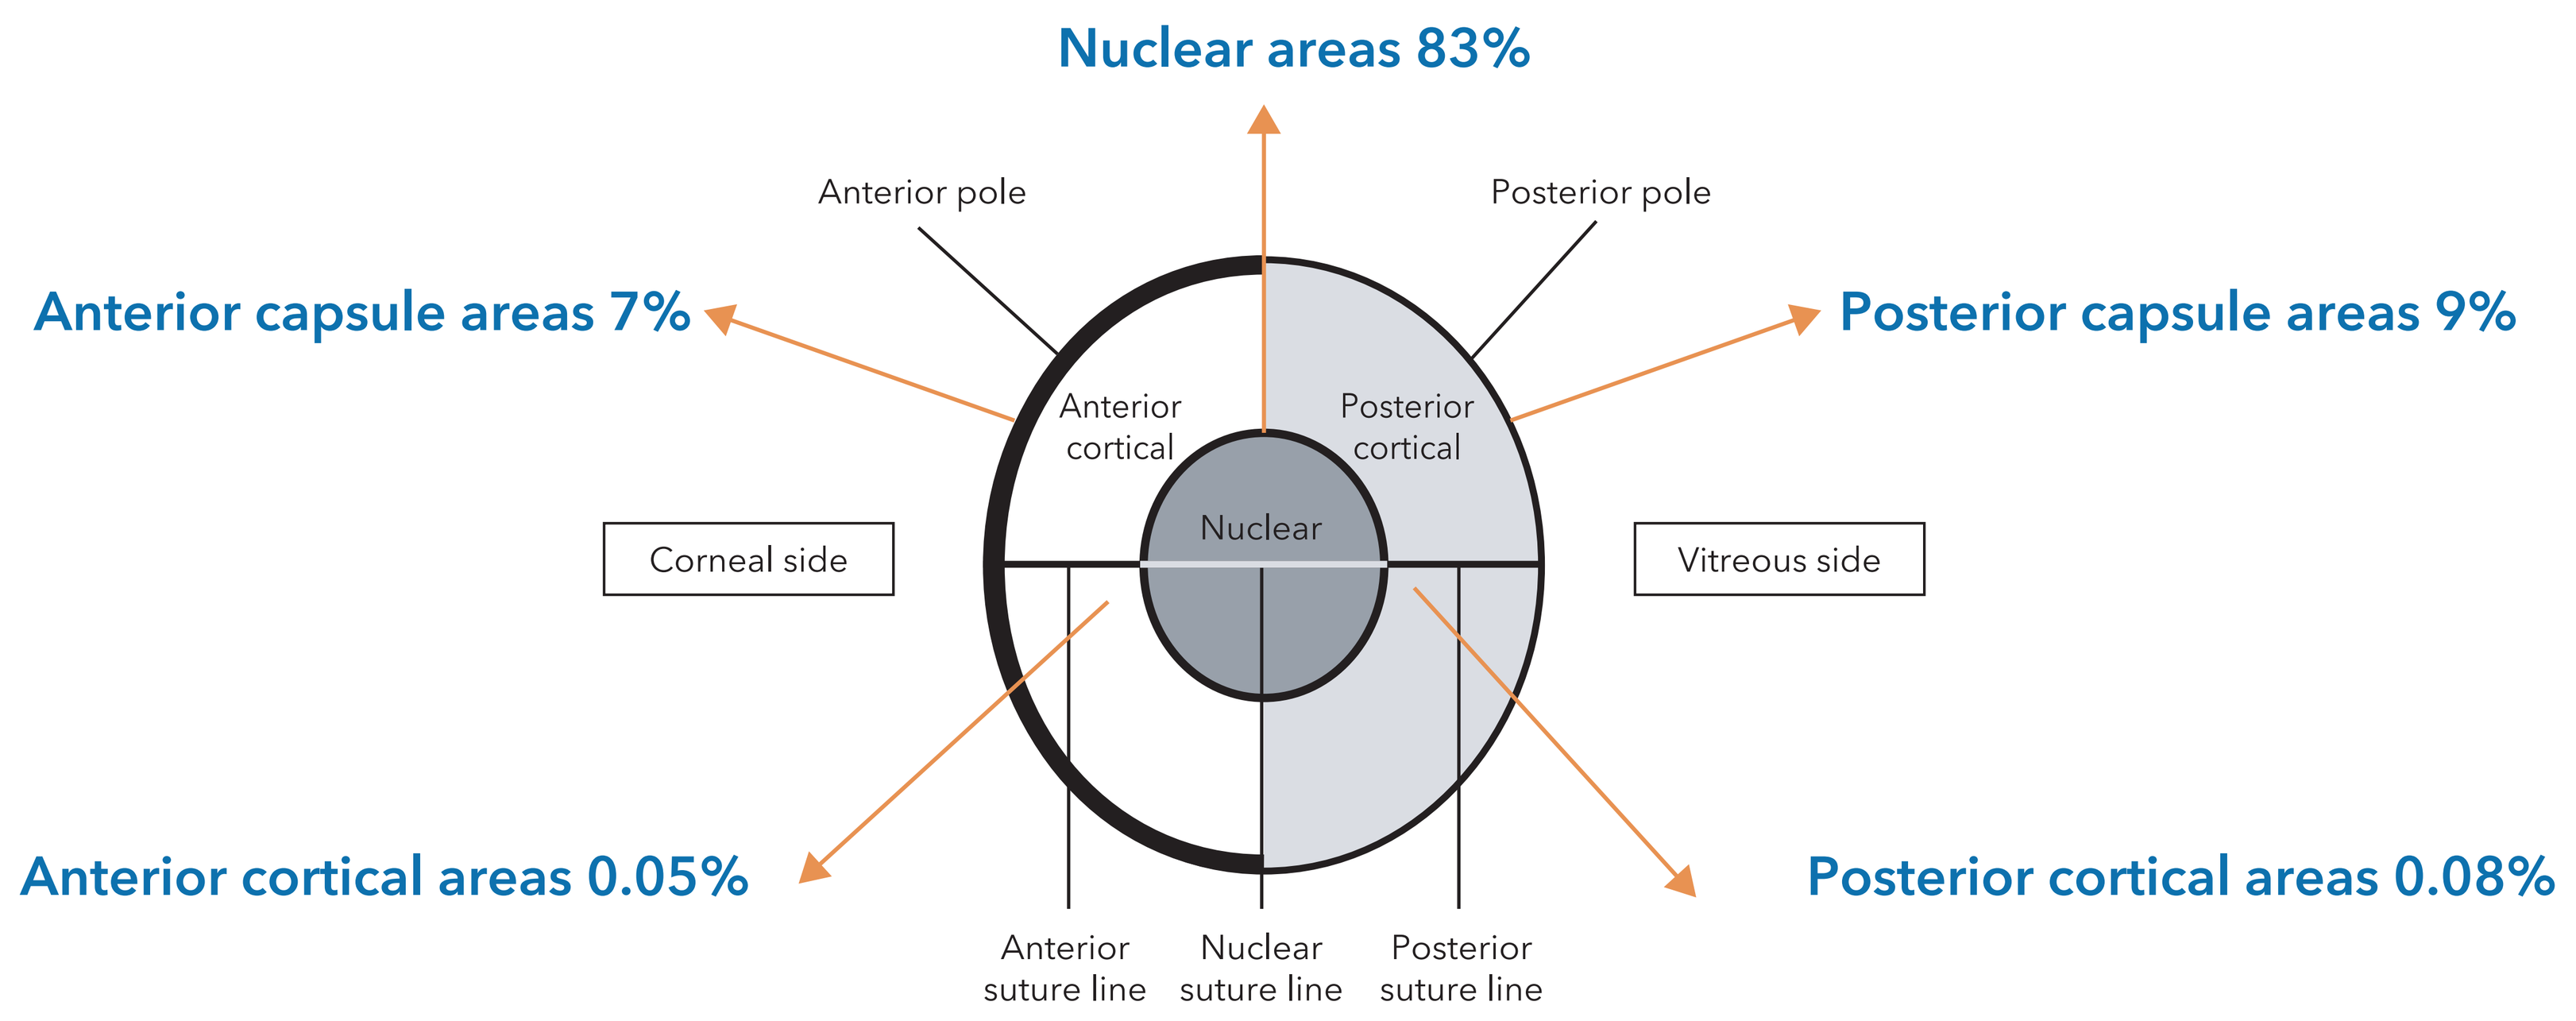
Nuclear areas 83%
Anterior pole
Posterior pole
Anterior capsule areas 7%
Posterior capsule areas 9%
Anterior cortical
Posterior cortical
Nuclear
Corneal side
Vitreous side
Anterior cortical areas 0.05%
Posterior cortical areas 0.08%
Anterior suture line
Nuclear suture line
Posterior suture line

Financial Disclosure

There are no potential conflicts of interest to disclosure.

Contact Information

zhou_yongdong@wuxiapptec.com

Compared Incidence of the Spontaneous Abnormalities between WuXi AppTec (Suzhou, China) and Taisho Pharmaceutical Research Center (Saitama, Japan)

- Higher incidences of lens opacity (43% and 47%) and persistent hyaloid artery (21% and 17%) in male and female, respectively, were noted in the Taisho Pharmaceutical Research Center (Saitama, Japan)[1].
- While a lower incidence of corneal opacity (61% and 60% in male and female, respectively) was noted in the Taisho Pharmaceutical Research Center (Saitama, Japan)[1].
- In WuXi AppTec (Suzhou), the incidences of corneal opacity（78% and 80%）, lens opacity (11% and 8%) and persistent hyaloid artery (3% and 2%) were similar in male and female and not shown in the below graph.

Incidences of the Spontaneous Abnormalities

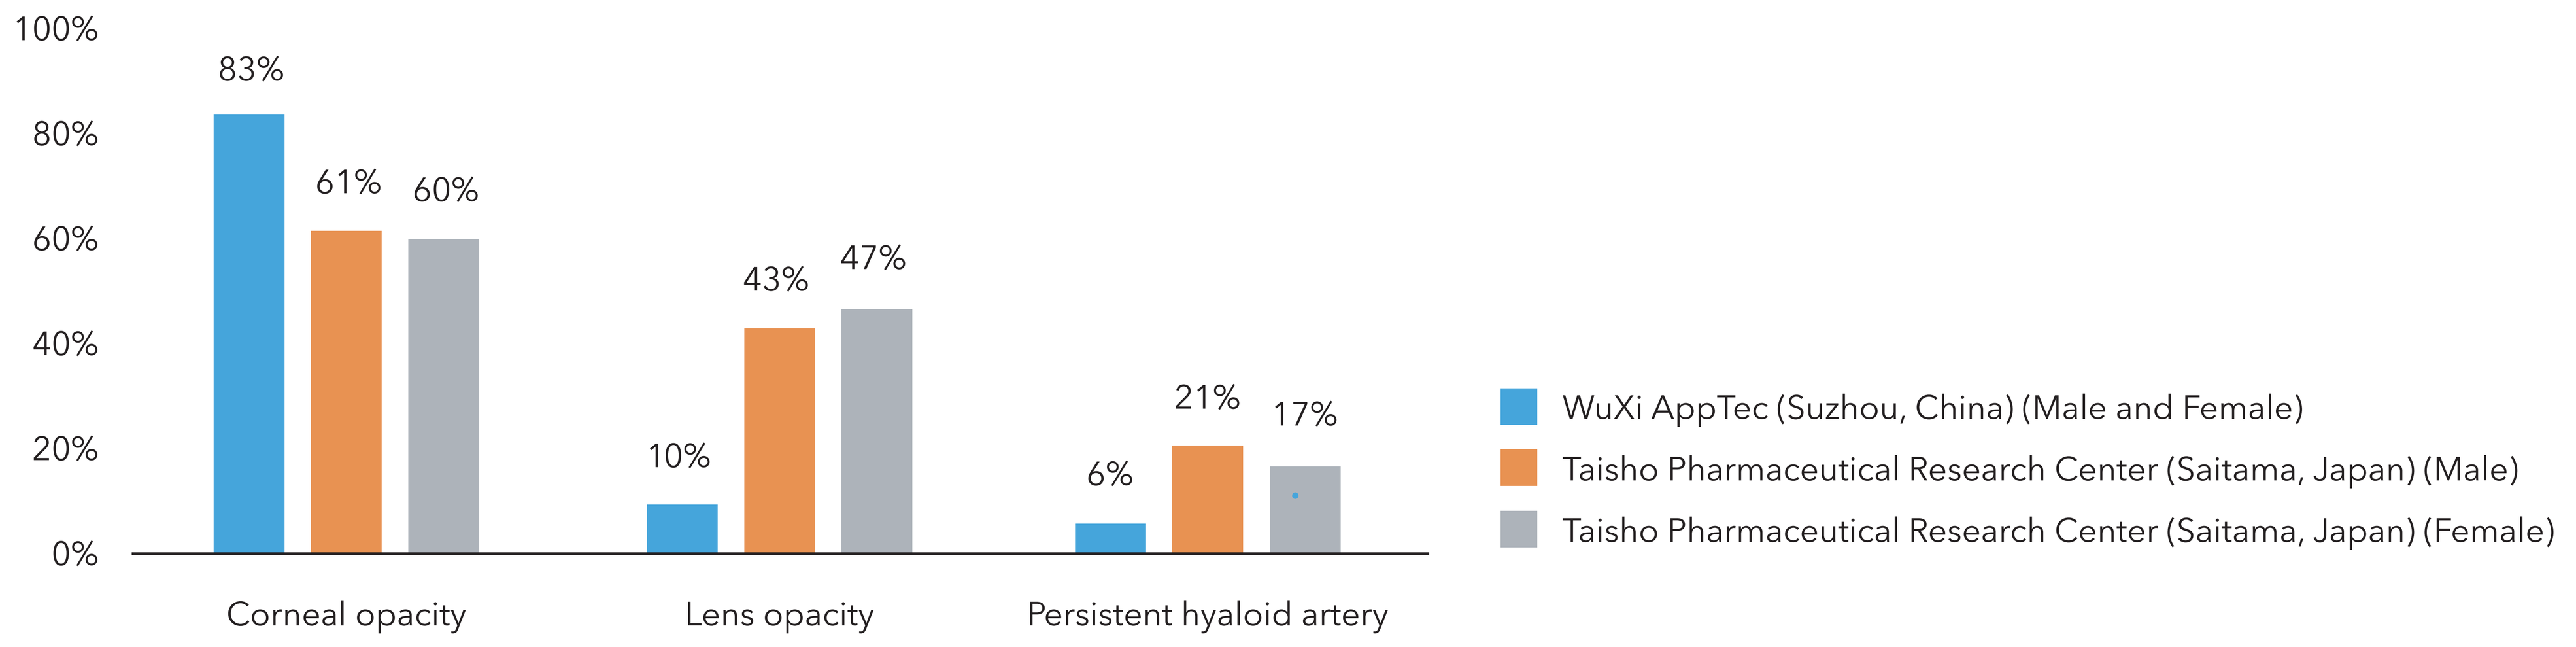
100%
80%
60%
40%
20%
0%
83%
61%
60%
10%
43%
47%
6%
21%
17%
Corneal opacity
Lens opacity
Persistent hyaloid artery
WuXi AppTec (Suzhou, China) (Male and Female)
Taisho Pharmaceutical Research Center (Saitama, Japan) (Male)
Taisho Pharmaceutical Research Center (Saitama, Japan) (Female)

Discussion & Conclusion

The incidences of the spontaneous abnormalities maybe vary in animals from different vendors, strains and different years.

Indicate the importance of establishing in house data and updating historical data regularly to identify the spontaneous abnormalities of animals.

Toxicologists and ophthalmologists could differentiate treatment-related changes from spontaneous lesions by referring to the in house data specific to the vendors and strains.

Reference

[1] Junya Morita, Haruhiro Yamashita, Kazunari Sugihara, Masaki Wakamatsu, and Minoru Sasaki. Spontaneous Ocular Abnormalities in Sprague-Dawley Rats. Comparative Medicine. 2020 April 70;2:140-144.