Novel Approach to Mitigate Pharmacological Actions of a Novel Hedgehog (Hh) and Smoothened (Smo) Inhibitor to Successfully Complete a 26 Week Rat Study

Luis Dellamary and Miguel de los Rios, Endeavor BioMedicines, San Diego, CA
Sue McPherson, WuXi AppTec, Suzhou, China

Abstract: 3015

Introduction

The conduct of toxicology studies can be difficult enough with toxic responses from the test materials. However, these can be further compounded by the pharmacological responses of the test material which can result in studies terminating through these issues and not as a direct toxic response of the test material. This poster will illustrate the novel approaches taken for the successful evaluation of a potent small molecule Hedgehog (Hh) and Smoothened (Smo) inhibitor, that effectively downregulating the downstream transcription factor Gli-1. The Hh pathway is important in bone and teeth development, causing craniofacial abnormalities and misshapen tooth buds when disrupted (Hosoya et al, 2020). This exaggerated pharmacological effect on the bone (skeletal) and teeth development is more pronounced in rodents which have continuous teeth and bone development (Hosoya et al, 2020; Zhao et al 2014; Kimura et al, 2008; Lui et al, 2008).

Previous studies had been terminated early due to adverse clinical signs and effects on body weight and food consumption, secondary to effects on the oral cavity (tooth and bone). The previous 26-week study was terminated early on Day 42, following 40/39 days of oral administration of ENV-101 in male and female Sprague Dawley rats, respectively. In the terminated 26-week study, there were test-article related toxicities predominantly due to effects upon growing teeth which were considered adverse at the mid- and high-dose levels (2 and 6 mg/kg/day). In the terminated study, 5 high dose males and females died prematurely which preceded the subsequent termination of all animals in the study. Body weight losses of 16.6% to 19.9% in the mid- and high-dose females, respectively and 21.5% in high-dose males correlated with reduced food consumption of 25.20% in mid- and high-dose females, and 23.15% in high-dose males.

In the terminated study, the resulting adverse effects on body weight and food consumption were consistent with exaggerated pharmacology effects of ENV-101. Due to reduced food consumption, body weight loss and associated clinical observations. Effects on the bone and teeth may have been exacerbated by the use of relatively younger rats (not fully developed) in the terminated 26-week study.

The repeat study utilized 20-week old rats at dosing initiation which were 8 weeks or 13 weeks older than rats utilized in prior toxicology studies with ENV-101 and the same dose levels (0.6, 2 and 6 mg/kg/day), with frequent monitoring of body weights and supplementation with nutritious soft diets at the first observation of jaw and teeth effects affecting feeding behavior to help mitigate body weight loss and associated secondary toxicities. By such measures the 26-week dosing period was successfully completed, and the potential toxicity evaluated.

Materials and Methods

Animal strain	Rat (Rattus norvegicus)/Sprague Dawley (SD), SPF
Animal supplier	Beijing Vital River Laboratory Animal Technology Co., Ltd.
Age at initiation	21 weeks
Body weight at initiation	400 to 800 g for males and 200 to 600 g for females
Body weight and food consumption	Twice weekly
Diet	Rodent diet supplied ad libitum
Food supplementation	Nutra-Gel, or 60% Ready Jelly mixed with 40% Peanut Butter)
Opthal	Pretest and week 26
Clinical path	Week 13, and prior to scheduled necropsy
TK	Days 1, 91 and 182
Necropsy	Weeks 27 and Recovery 31

Body Weights

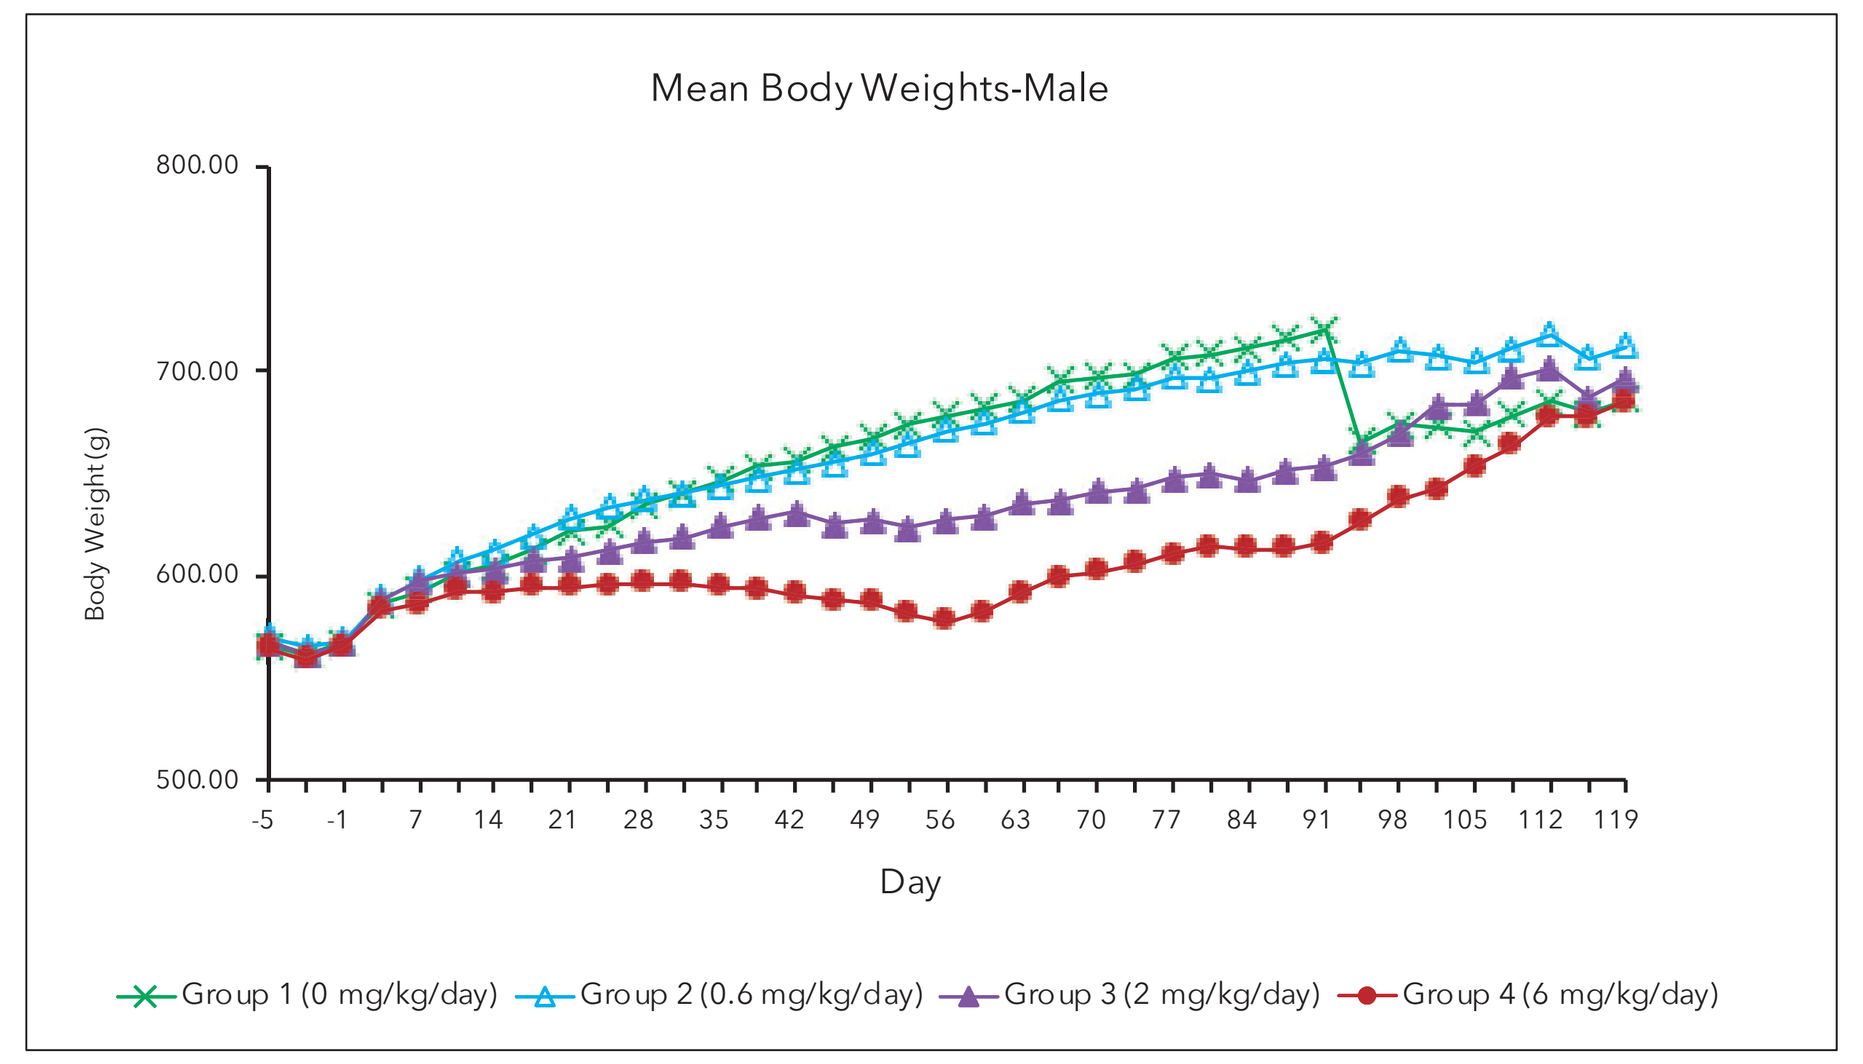
Mean Body Weights-Male
Body Weight(g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

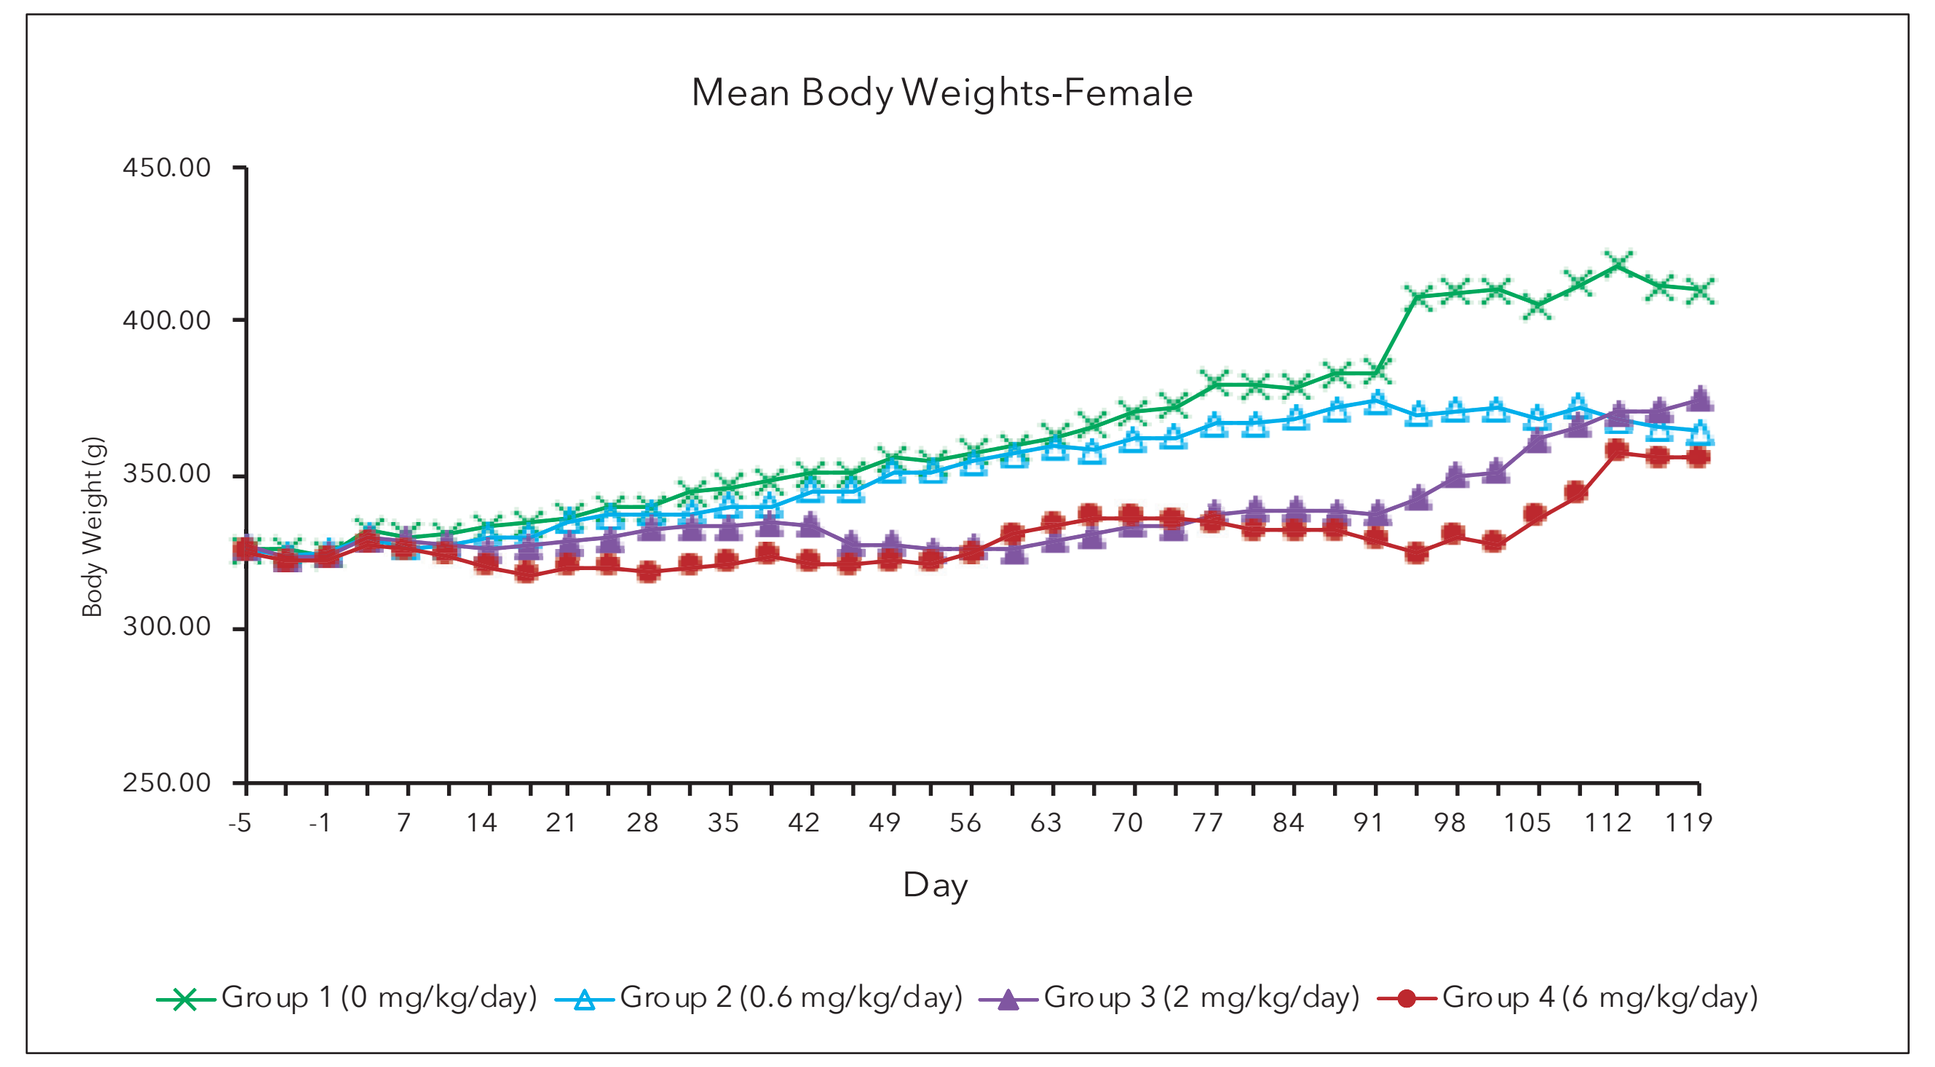
Mean Body Weights-Female
Body Weight(g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

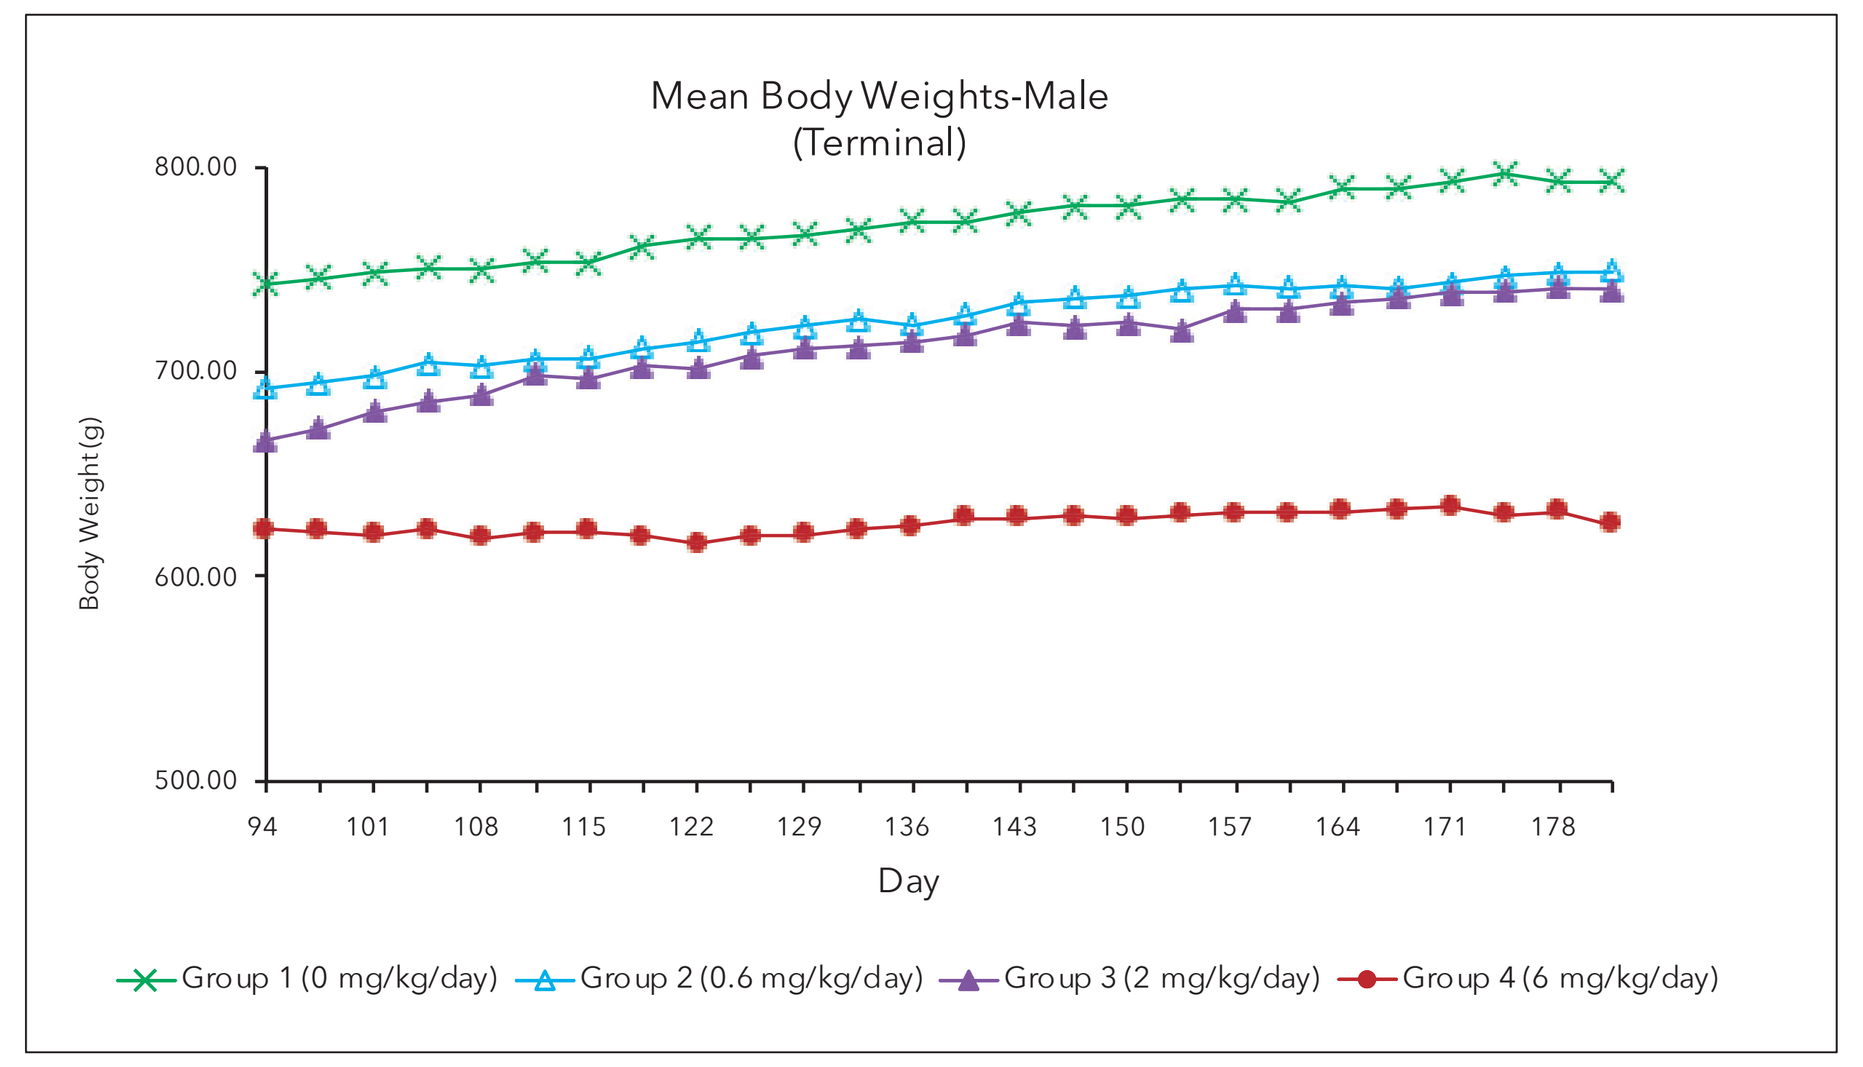
Mean Body Weights-Male (Terminal)
Body Weight(g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

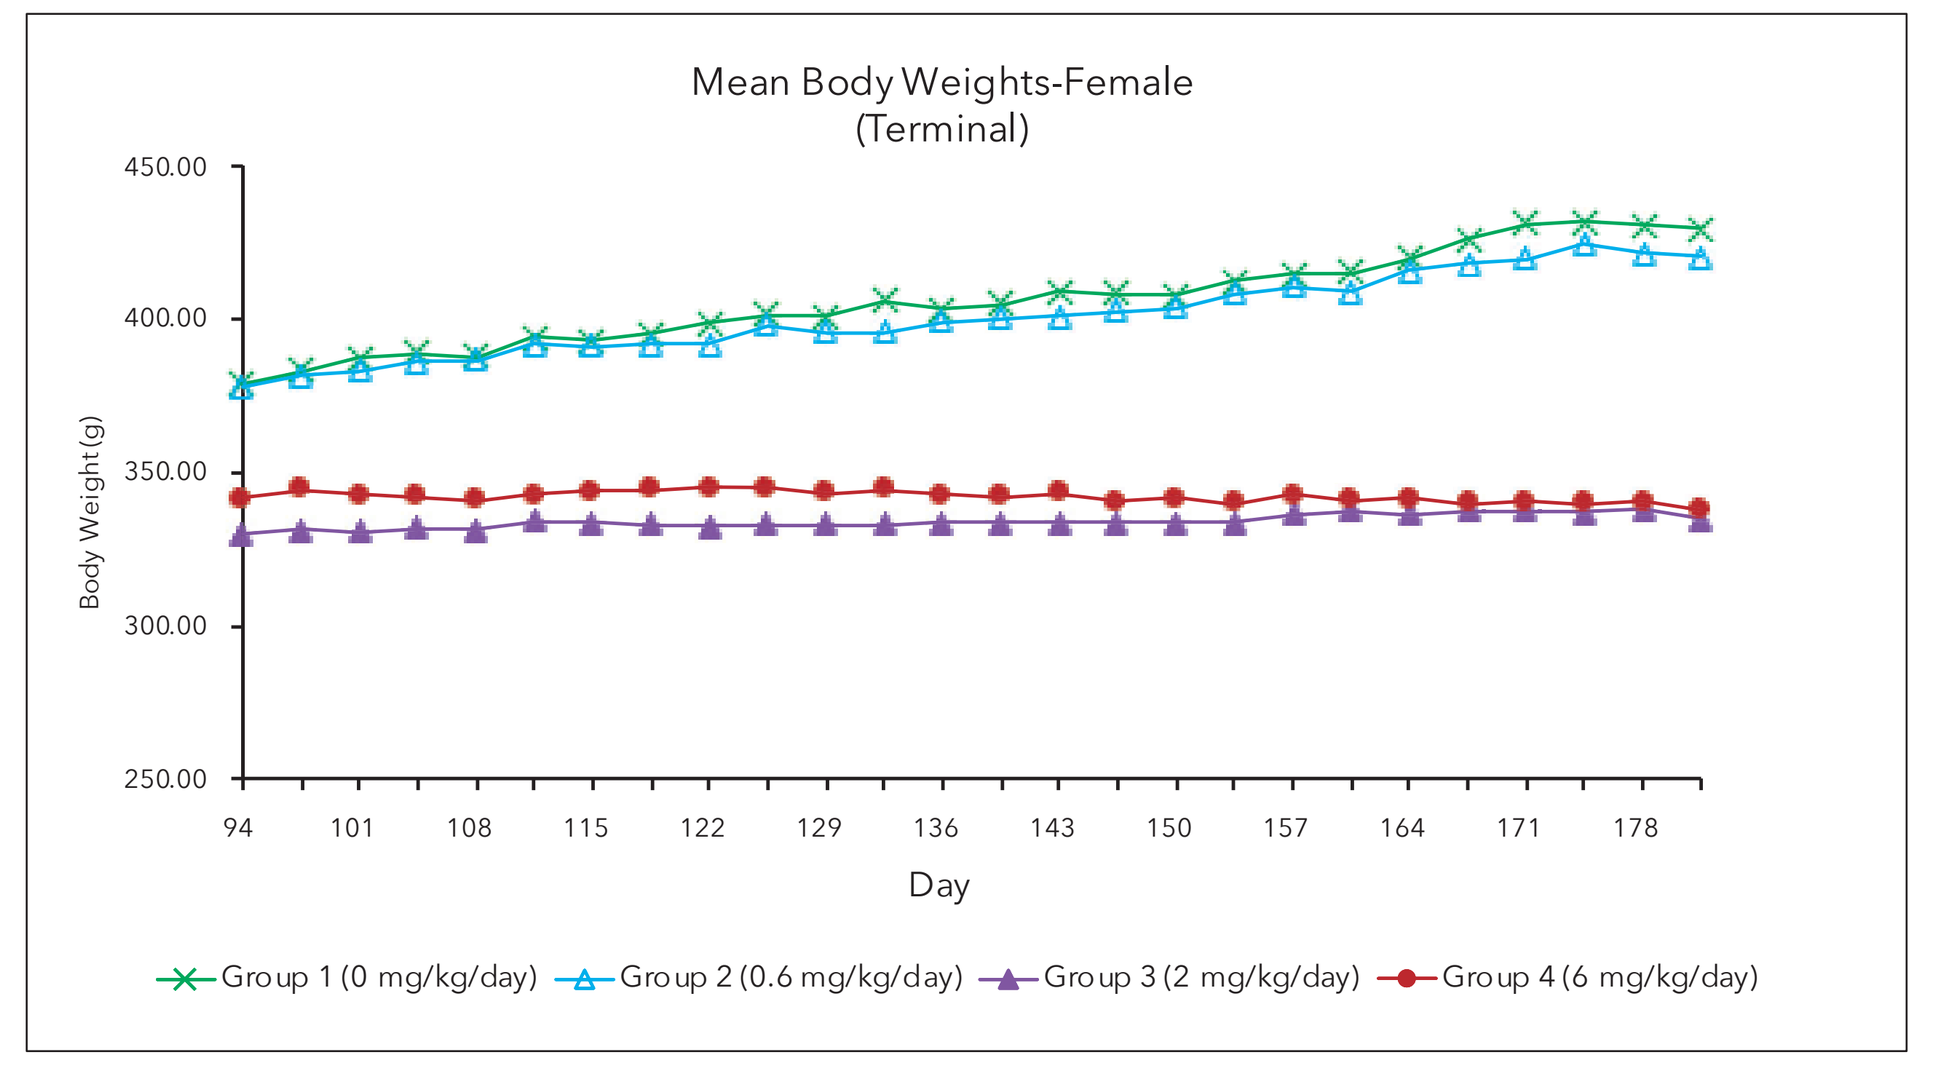
Mean Body Weights-Female (Terminal)
Body Weight(g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

Food Consumptions

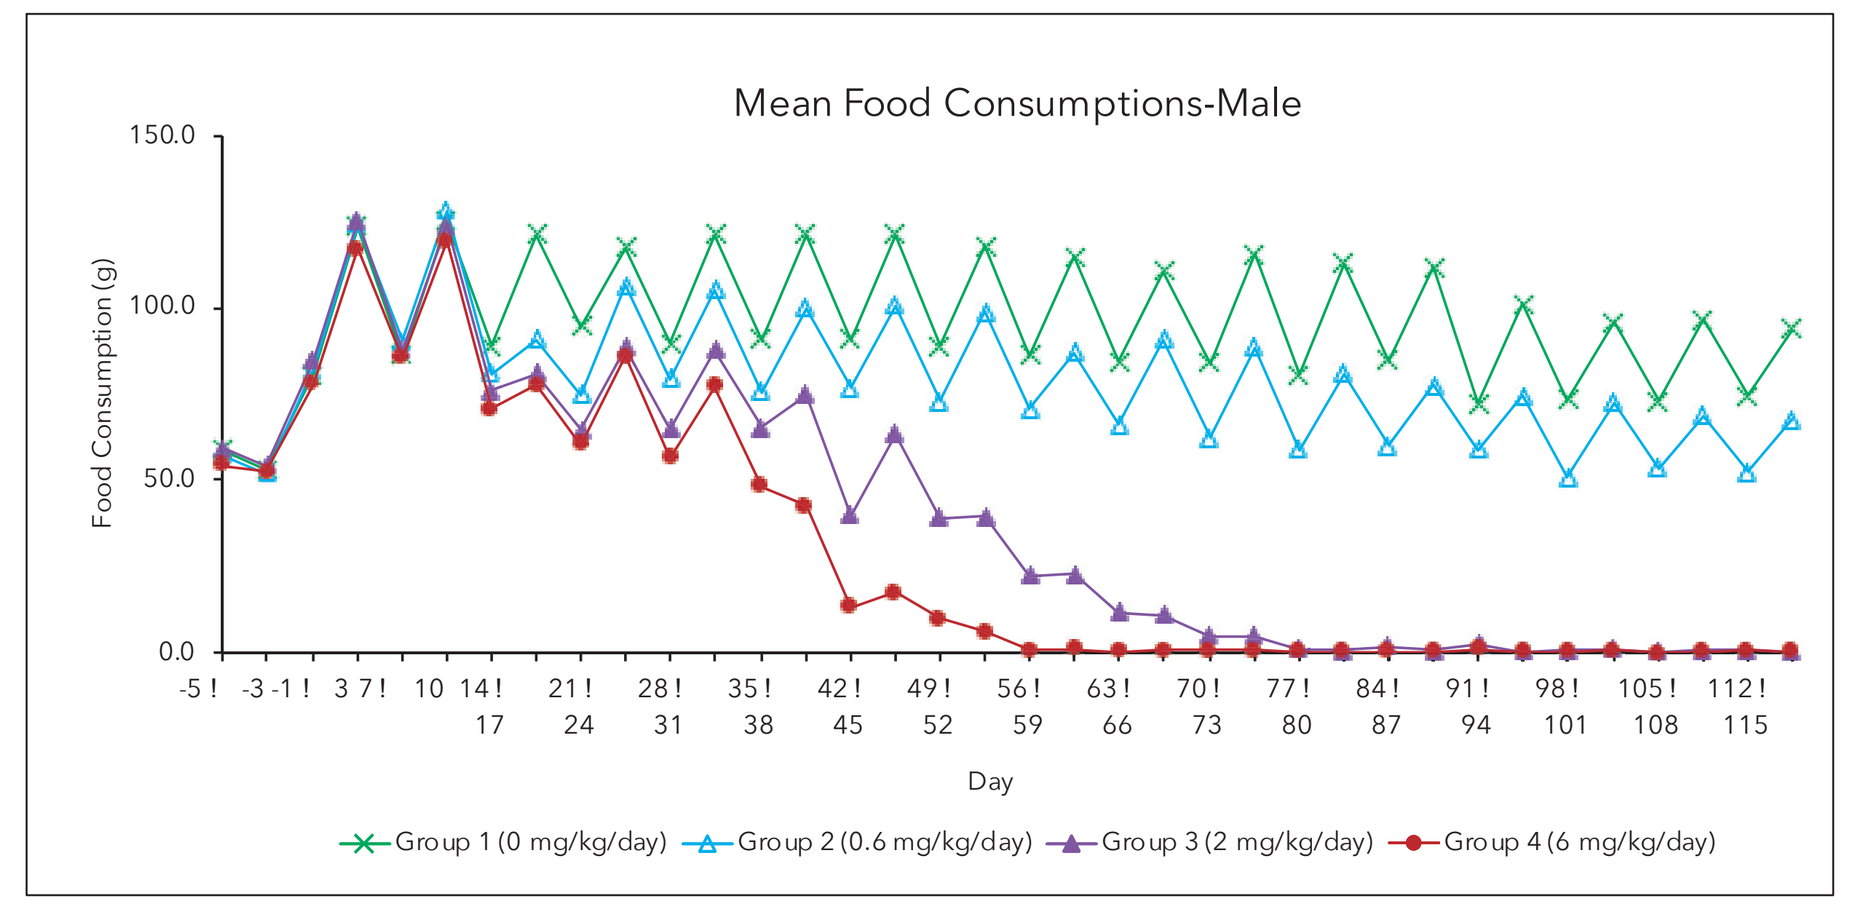
Mean Food Consumptions-Male
Food Consumption (g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

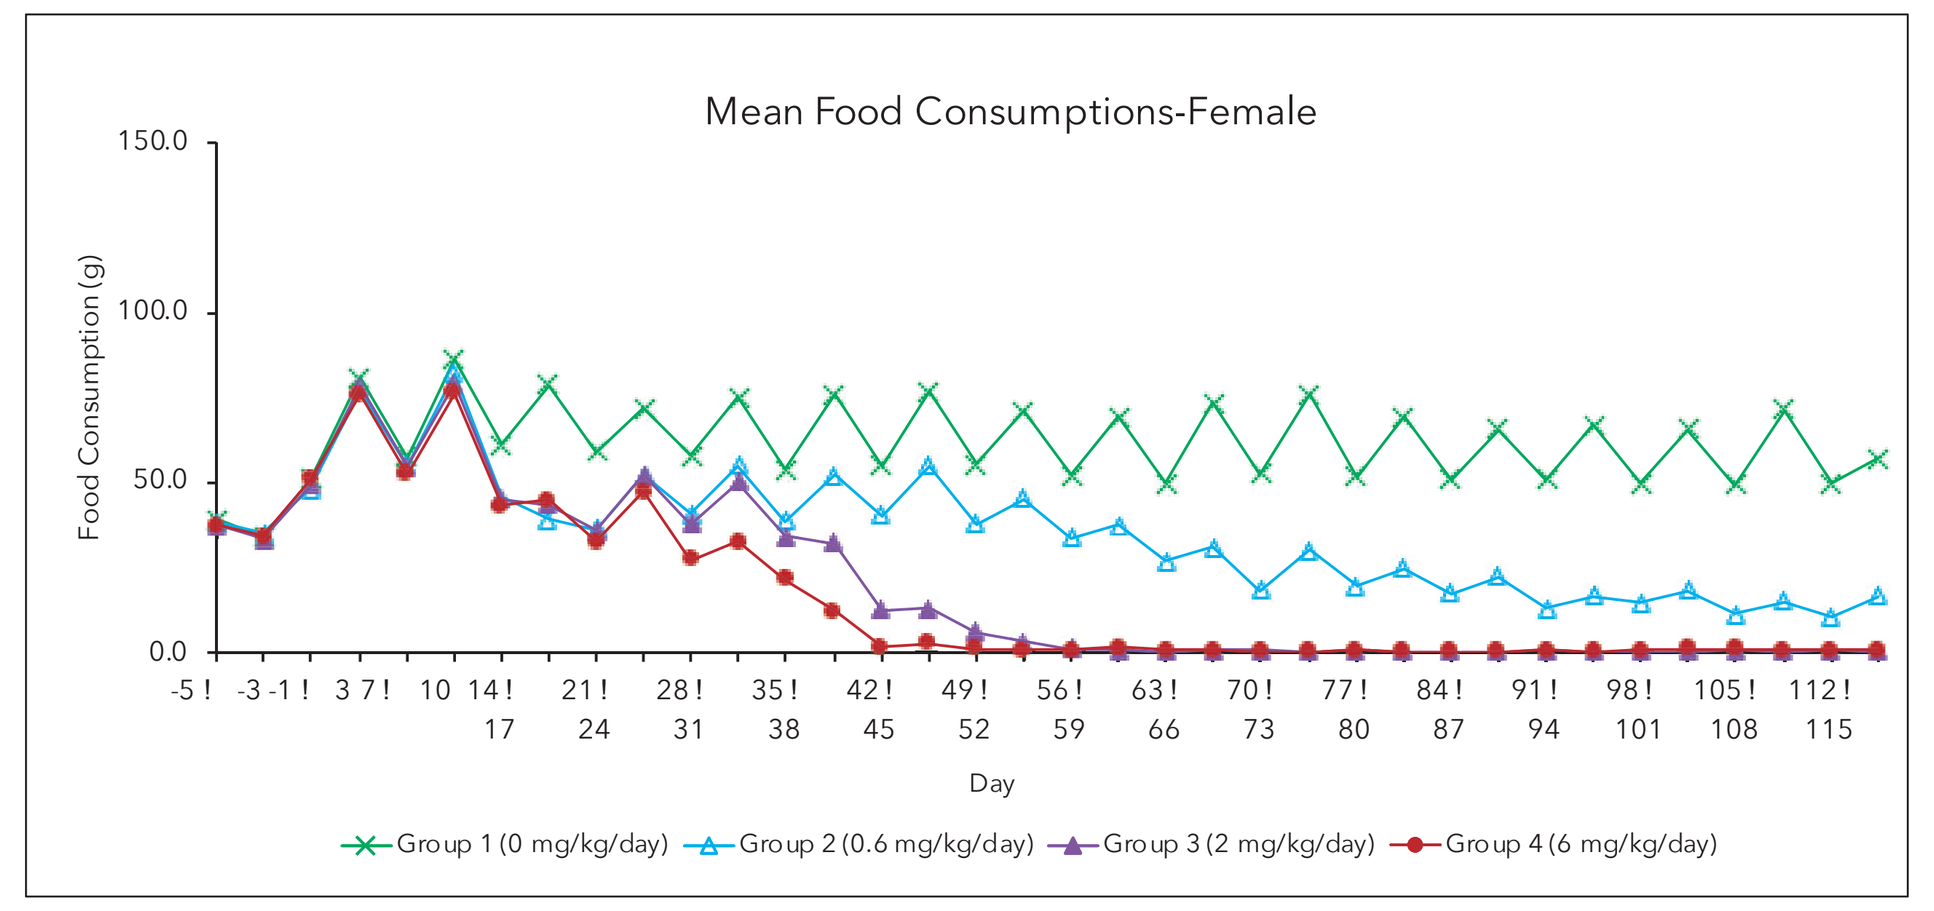
Mean Food Consumptions-Female
Food Consumption (g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

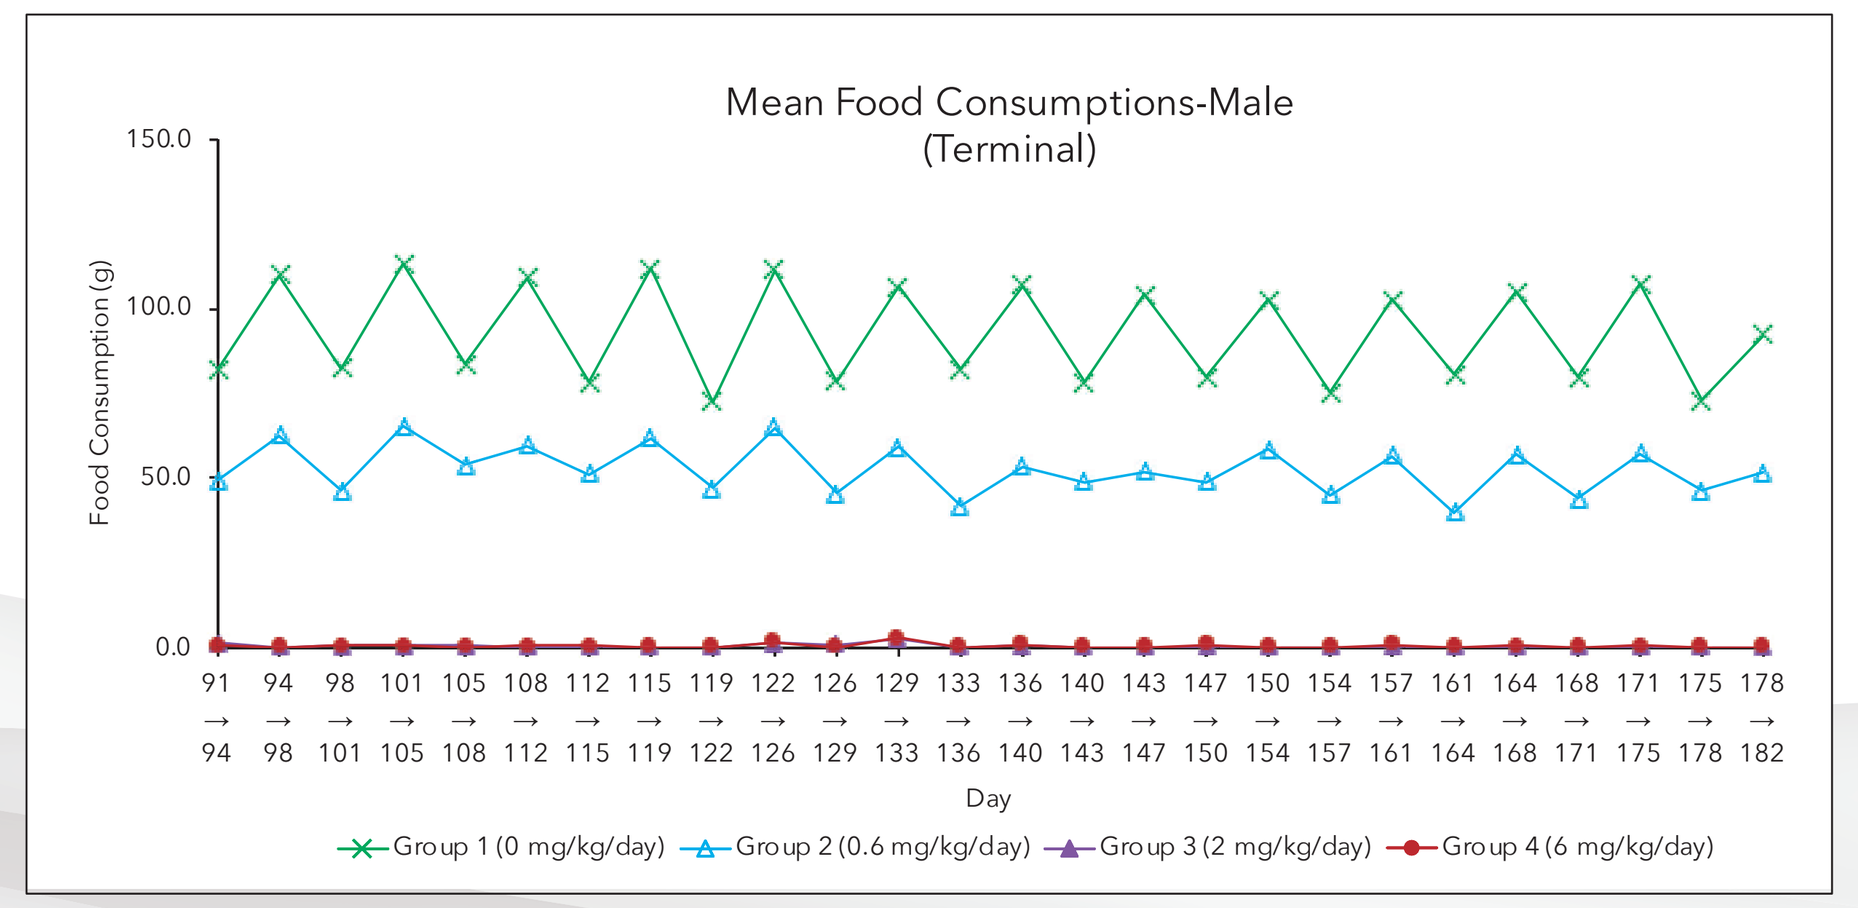
Mean Food Consumptions-Male (Terminal)
Food Consumption (g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

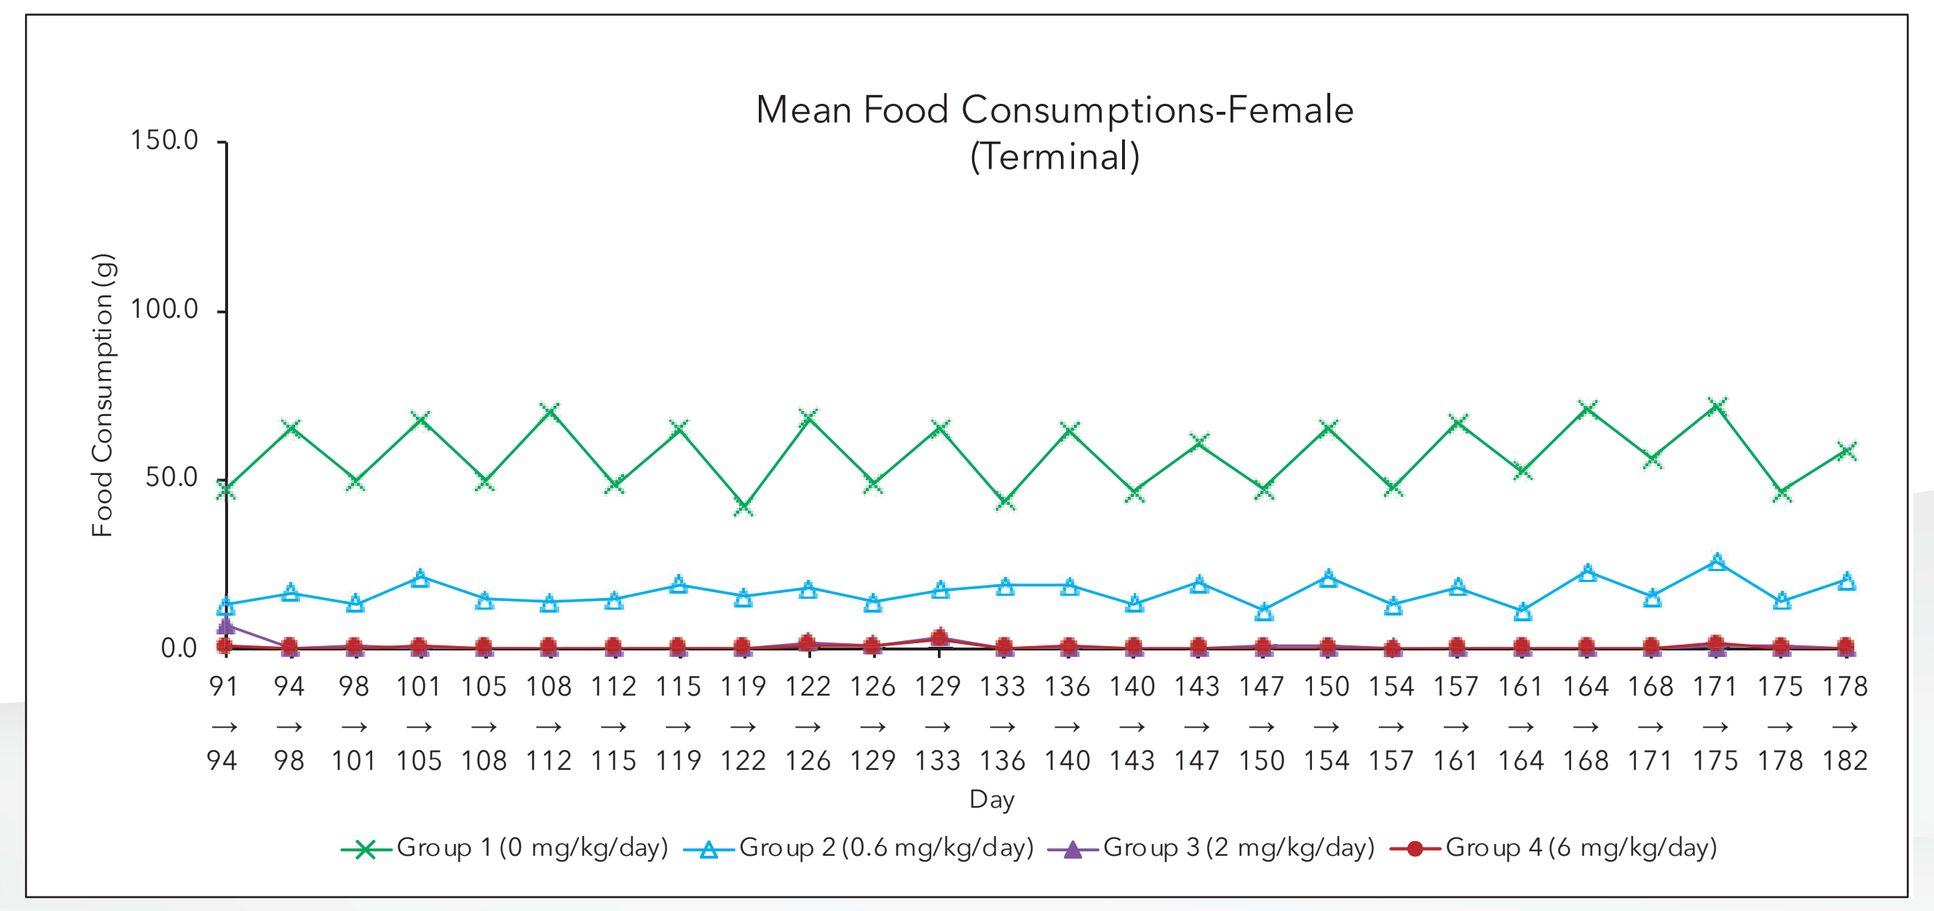
Mean Food Consumptions-Female (Terminal)
Food Consumption (g)
Day
Group 1 (0 mg/kg/day) Group 2 (0.6 mg/kg/day) Group 3 (2 mg/kg/day) Group 4 (6 mg/kg/day)

Results

Clinical signs
Dose related incidence of malocclusion, tooth/teeth missing, tooth/teeth broken, and abnormally shaped teeth.

Swelling in mouth, ocular discharge, alopecia in nose and periorbital area (also noted at necropsy), thin appearance, tremors, and abnormal gait were observed in males and/or females at ≥2 mg/kg/day.

Body weight
Significantly decreased mean body weight was observed in males and females at ≥2 mg/kg/day, starting from Week 8 and Week 4 for the mid (2 mg/kg/day) and high (6 mg/kg/day) dose groups, respectively. The decreases in body weight were considered a consequence of clinical changes in teeth that made it difficult for the animals to eat the pelleted feed.

Anatomic and clinical pathology
All direct effects of the test article were considered non-adverse, with the exception of those in the teeth (where dysplastic changes resulted in tooth breakage and secondary periodontal abscessation, peri-oral skin lesions, decreased food consumption and poor body weight gain), bones (where gait abnormalities were noted clinically) and stomach following 26 weeks.

All other anatomic and clinical pathology changes were interpreted as secondary to low food consumption and poor body weight gain at ≥2 mg/kg/day.

Conclusion
Findings - in-life and pathology findings were consistent with the hedgehog inhibiting properties of ENV-101. Exaggerated pharmacological-based effects involved decreased body weight and inappetence in males and females at ≥2 mg/kg/day, histopathology findings in teeth (in males at ≥2 mg/kg/day and in females at all doses), bones in males and females ≥2 mg/kg/day and stomach in females at 6 mg/kg/day.

It is also concluded that by understanding the pharmacology of the drug it is possible to overcome these findings by food suplementation and to successfully evaluate new drugs and bring to the market.

Acknowledgments

The authors acknowledge the technical contributions made by the technical team at Suzhou.