A Retrospective Analysis of Internal In Vitro Chromosomal Aberration Data

Liwen Gao; Mengqi Zhang; Millie Chen
WuXi AppTec, Suzhou, China

Abstract: 3103

Purpose

The *in vitro* chromosomal aberration assay (CAA) is to identify substances that cause structural chromosomal aberrations and is a part of the recommended regulatory test battery for genotoxicity required by most regulatory agencies. Structural chromosomal aberrations are a consequence of failure or mistakes in the DNA repair process such that chromosomal breaks either do not rejoin or rejoin in abnormal configurations. There are published data indicating that the CAA has an unacceptably high incidence of positive results, to better understand this assay, we retrospectively evaluated the *in vitro* chromosomal aberration data generated in our laboratory.

Materials and Methods

The CAA was conducted in compliance with ICH S2(R1) and OECD TG473. The CAA studies conducted at WuXi AppTec (Suzhou) Co., Ltd. from 2012 to 2021 were collected for the retrospective analysis. The survey only focus on pharmaceuticals, other test articles, such as impurities, food additives, medical devices, were excluded.

The Chinese hamster ovary cell line, CHO-WBL, was employed in this assay. Each study contained two phase: a dose range-finding assay and the definitive chromosomal aberration assay. A dose range-finding assay was performed to establish the dose range for the definitive chromosomal aberration assay. The definitive CAA was used to evaluate the clastogenic potential of the test article. The CAA was conducted using standard procedures by exposing cultures of CHO-WBL cells to series concentrations of the test article as well as to concurrent positive and negative controls. Three treatment series were included. In the S9-activated treatment series, treatment will be for 3 hours; in the non-activated treatment series, treatment will be for 3 hours and for 22 hours. At predetermined intervals after treatment to the test article, the CHO-WBL cells are treated with a metaphase-arresting substance, then harvested and stained, and metaphase cells are analyzed microscopically for the presence of chromosomal aberrations.

For a valid assay, responses observed in the assay are evaluated as follows: the conclusion is usually based on the statistical analysis of the proportion of cells with structural aberrations in treated cultures as compared with the concurrent negative controls, but this is not the only parameter used. The types of aberrations, the occurrence of cells with multiple aberrations, concentration-related response and the historical negative control percentages of cells with structural aberrations are also taken into account. The tests with and without S9 and the different treatment lengths will be considered separately. If any of the tests with S9 activation or without S9 is positive, the overall conclusion is positive. The final evaluation of the test article is based on scientific judgment.

Microscope images depicting several common abnormalities observed in metaphase spreads using CHO-WBL cells.

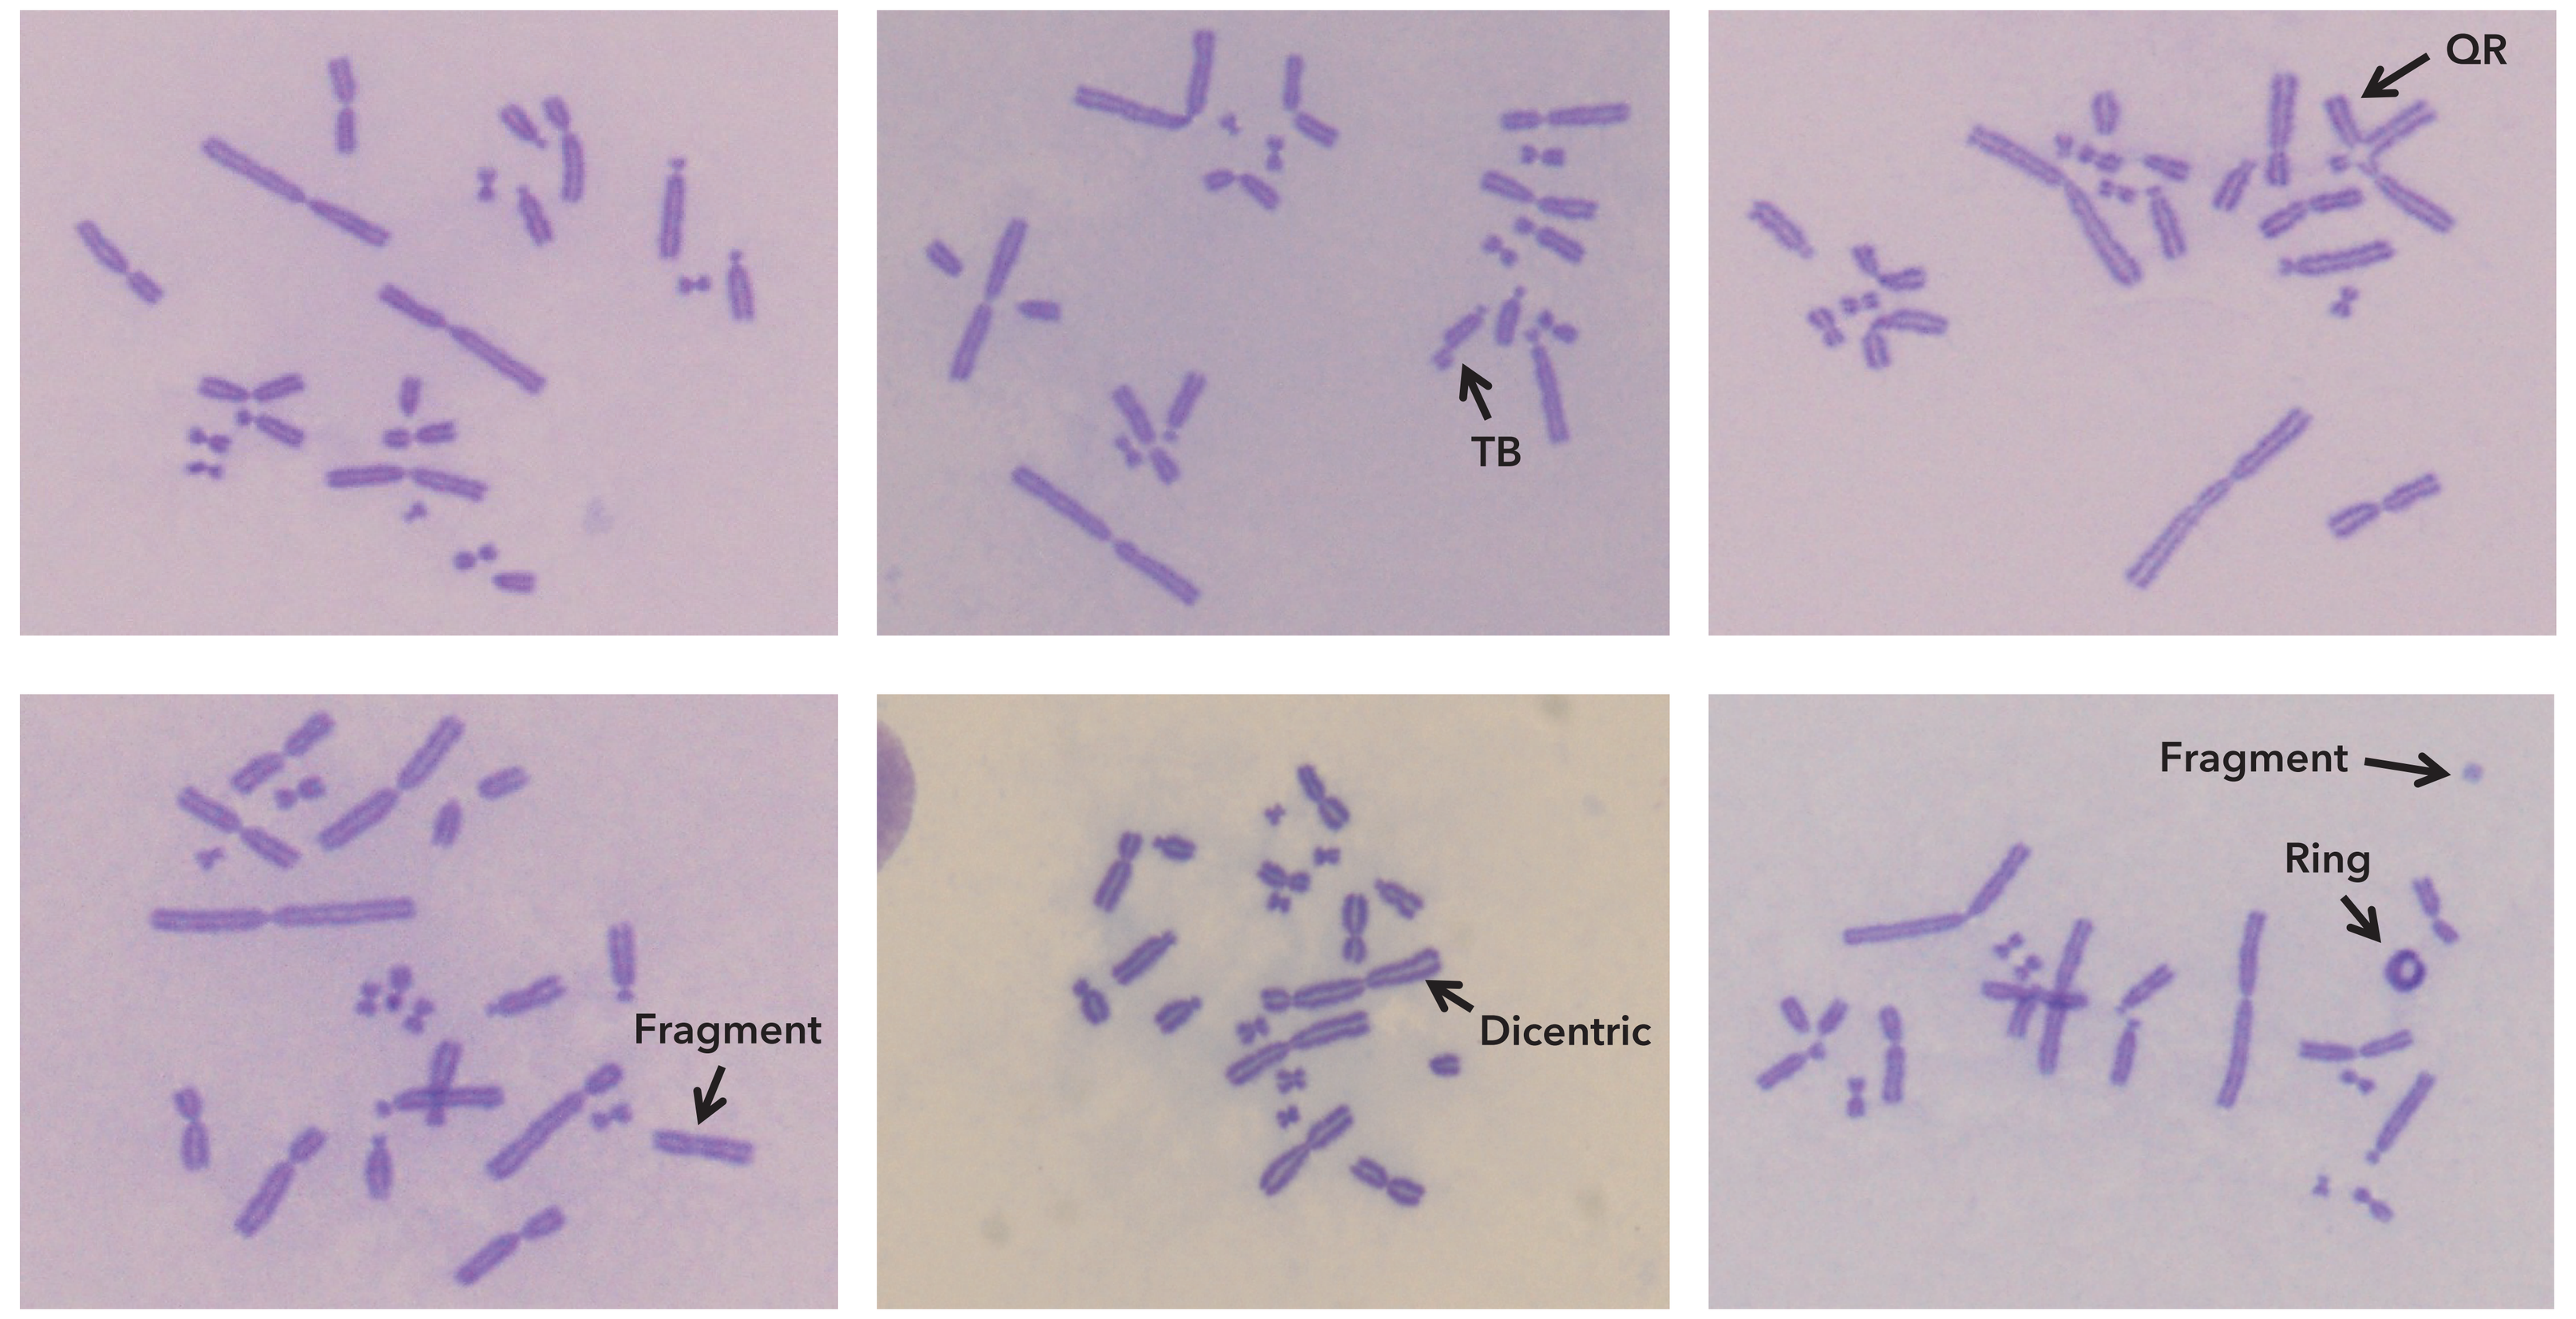
TB
QR
Fragment
Dicentric
Fragment
Ring

Results of 291 compounds tested in the *in vitro* chromosomal aberration assay at WuXi AppTec between 2012 and 2021.

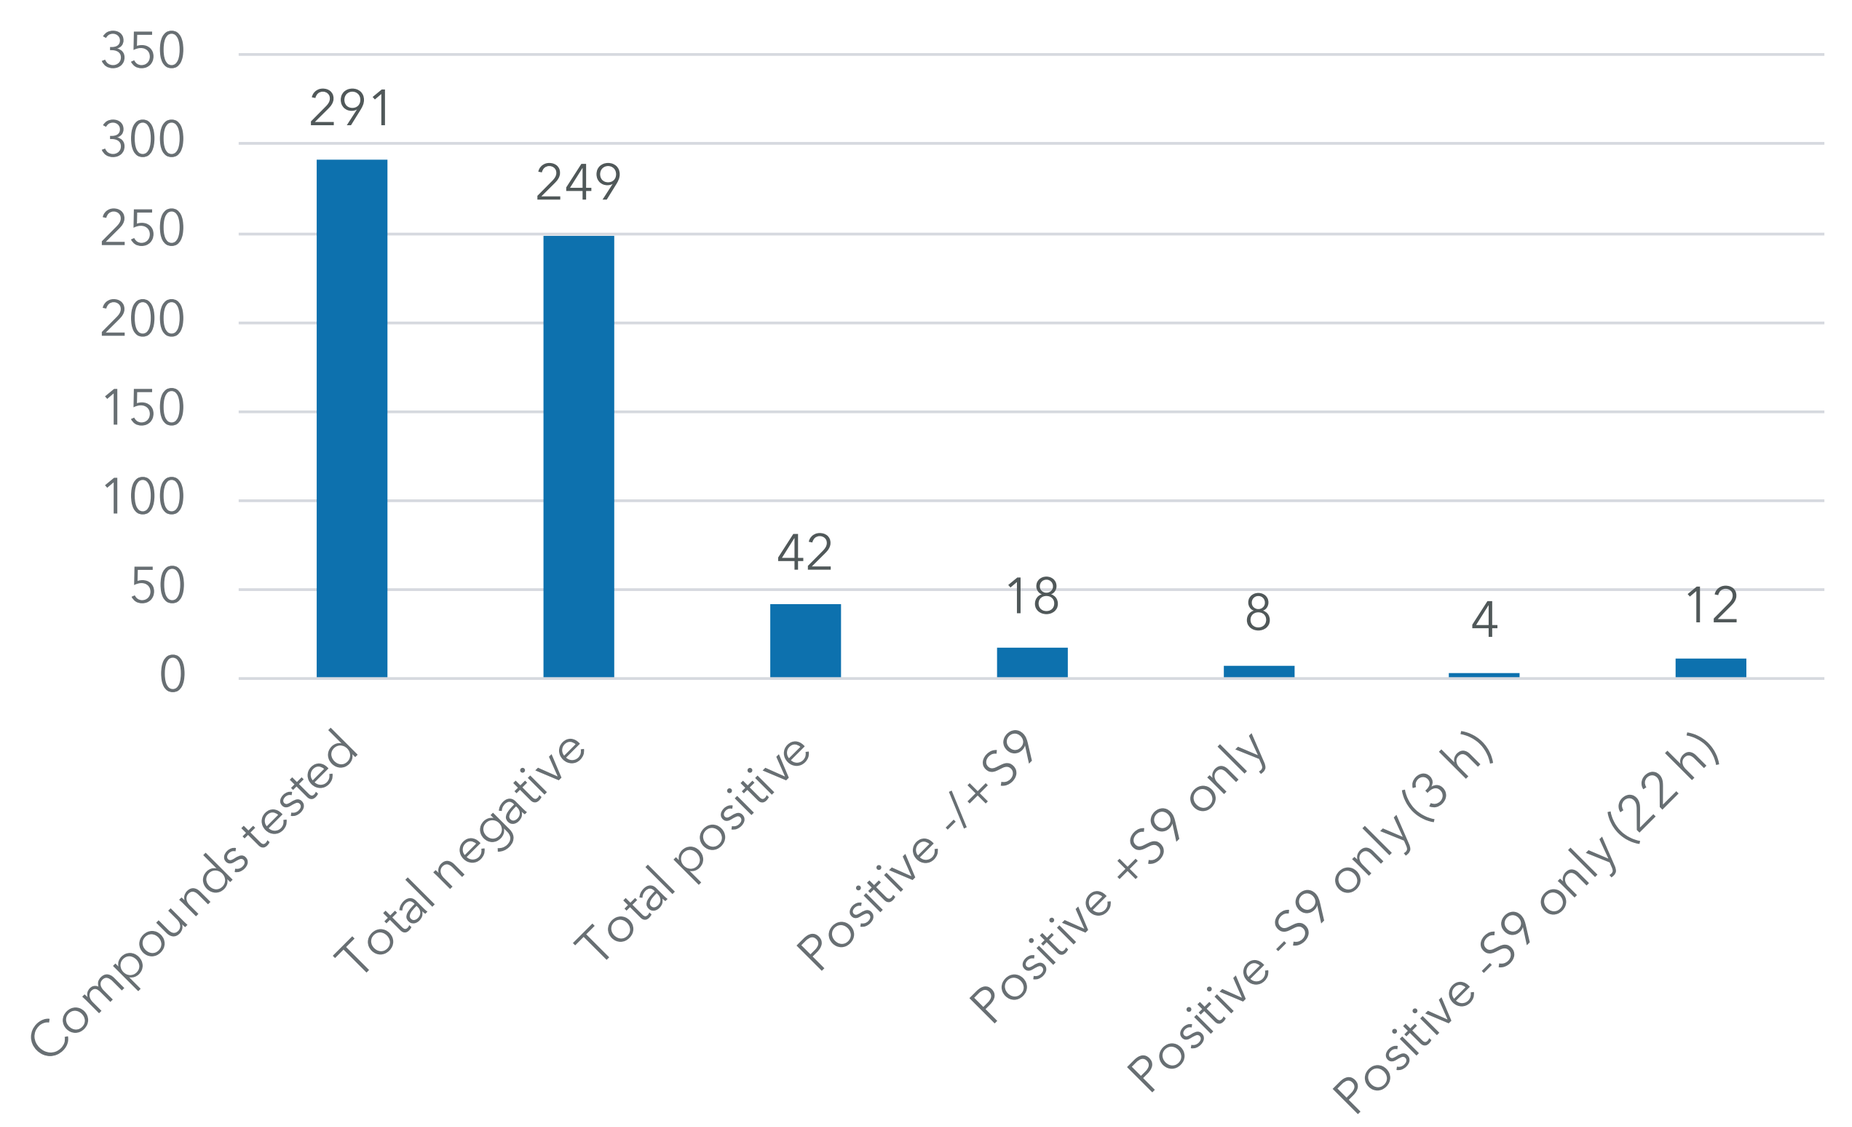
350
300
250
200
150
100
50
0
291
249
42
18
8
4
12
Compounds tested
Total negative
Total positive
Positive -/+S9
Positive +S9 only
Positive -S9 only (3 h)
Positive -S9 only (22 h)

Positive analysis for *in vitro* chromosomal aberration assay at WuXi AppTec between 2012 and 2021.

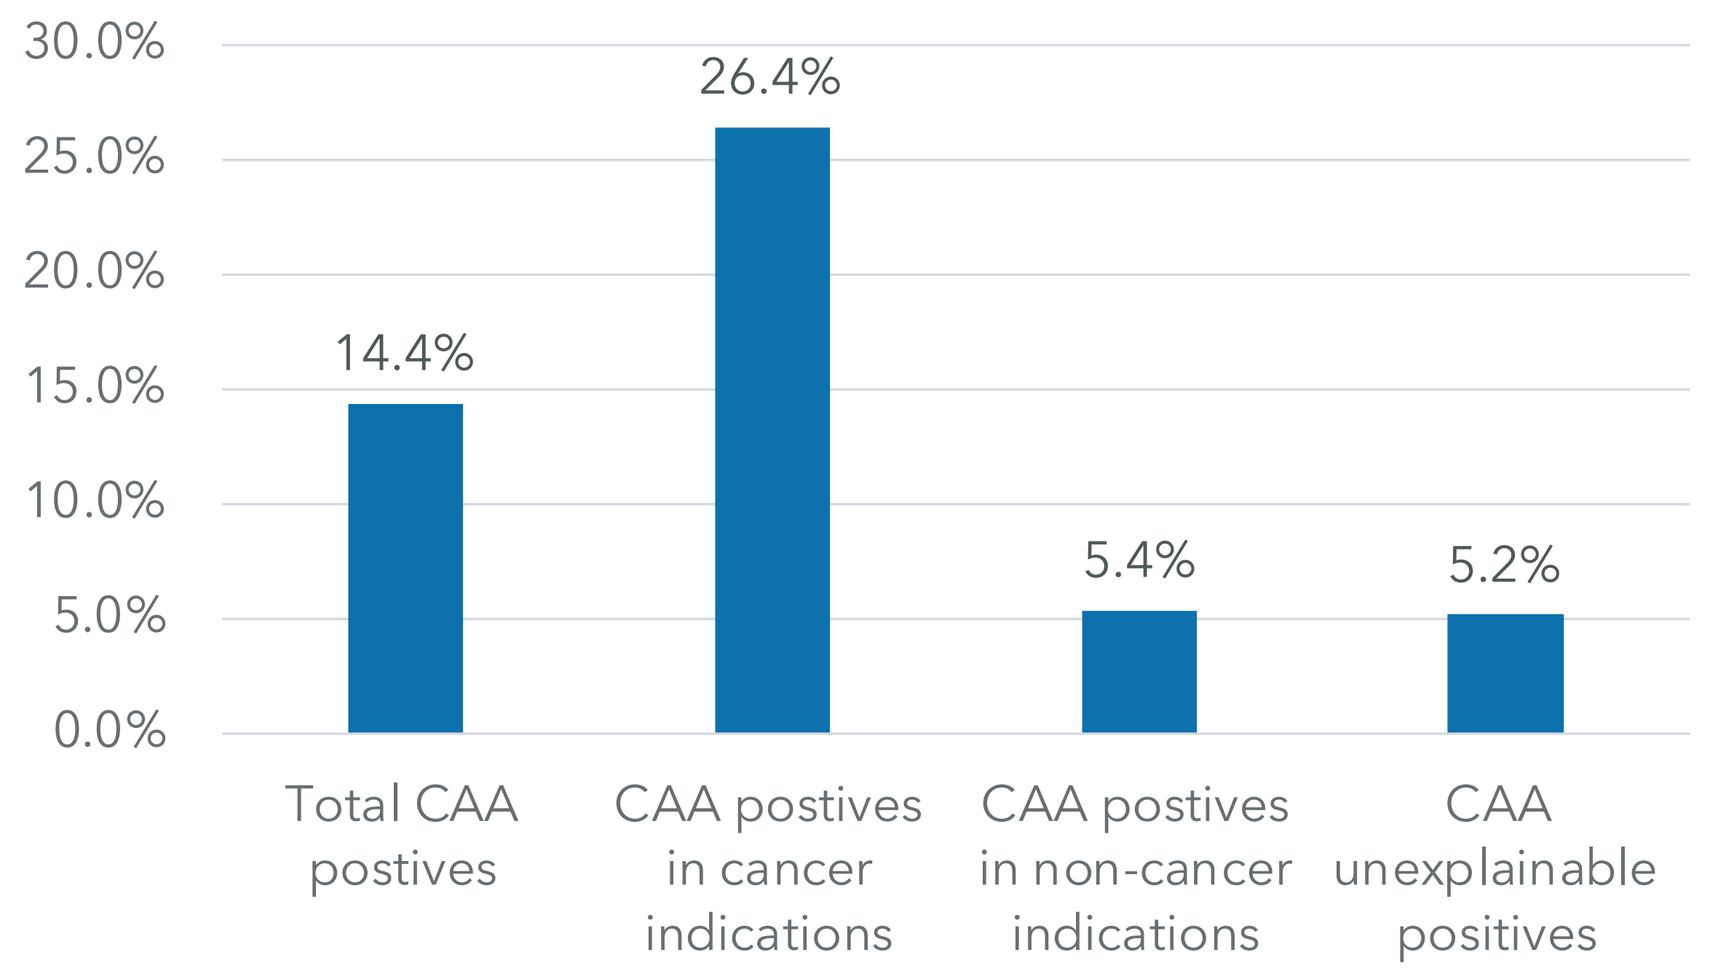
30.0%
25.0%
20.0%
15.0%
10.0%
5.0%
0.0%
14.4%
26.4%
5.4%
5.2%
Total CAA postives
CAA postives in cancer indications
CAA postives in non-cancer indications
CAA unexplainable positives

Compound classification for genotoxicity at WuXi AppTec.

Classification	Definition
1	The compound interacts with DNA directly by covalent binding. No pharmaceuticals but some impurities belong to this class in previous studies performed in our laboratory.
2A	The compound is involved in DNA metabolism, such as the inhibitors of enzymes that are essential for normal DNA synthesis, processing, and repair, and the positive calls are highly reproducible in previous studies, such as PARP, ATM, ATR inhibitors.
2B	The compound is involved in DNA metabolism, such as the inhibitors of enzymes that are essential for normal DNA synthesis, processing, and repair, but the consistent positive calls were not achieved in previous studies, such as PI3K, EGFR inhibitors.
3	Compounds that do not initially affect DNA or its metabolism may indirectly cause damage after disruption of other cellular physiological processes, frequently associated with cytotoxicity.

Results and Discussion

Of the 291 compounds tested in the presence and absence of S9, 249 (85.6%) were concluded to be negative and 42 (14.4%) were positive/equivocal. Of the 42 compounds that were positive/equivocal under any treatment series, 18 (42.9%) were active in the presence and absence of S9, 8 (19.0%) were active only in the presence of S9 and 16 (38.1%) were active only in the absence of S9, using 3-h or 22-h treatment. Based on the target and the reproducible call in previous studies, the active compounds were classified into 4 categories in our laboratory: 1, 2A, 2B and 3. For class 1, the compound interacts with DNA directly by covalent binding. No pharmaceuticals but some impurities belong to this class in previous studies performed in our laboratory. For class 2A, the compound is involved in DNA metabolism, such as the inhibitors of enzymes that are essential for normal DNA synthesis, processing, and repair, and the positive calls are highly reproducible in previous studies, such as PARP, ATM, ATR inhibitors. For class 2B, the compound is involved in DNA metabolism, such as the inhibitors of enzymes that are essential for normal DNA synthesis, processing, and repair, but the consistent positive calls were not achieved in previous studies, such as PI3K, EGFR inhibitors. For class 3, compounds that do not initially affect DNA or its metabolism may indirectly cause damage after disruption of other cellular physiological processes, frequently associated with cytotoxicity. In total, of the 42 positive/equivocal compounds, 27 compounds (17 in class 2A and 10 in class 2B) had pharmacological activity that has been associated with genotoxicity in mammalian cells. Hence, only 15 (5.2%) of 291 compounds tested gave positive/equivocal results that could not be explained by known mechanisms. For class 3 compounds, 11 were followed up with in vivo micronucleus and Comet assays, all are negative except one compound tested equivocal in Comet.

Conclusion

In summary, of 291 compounds tested over a 10-year period in our laboratory, 42 (14.4%) were positive/equivocal in the CAA and only 15 (5.2%) could be described as genuinely unexpected, which the results could be refuted with very high probability by the two appropriate in vivo genotox assays, such integrated micronucleus and Comet study.

Acknowledgments

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou) Co., Ltd.