Case Study: Re-consider Using Flow Cytometry for *In Vivo* Micronucleus Assay

Mengqi Zhang, Xiang Shan, Liwen Gao, Millie Chen
WuXi AppTec, Suzhou, China

药明康德 WuXi AppTec

Abstract: 3109

Purpose

In OECD guideline 474, Mammalian Erythrocyte Micronucleus Test, the traditional method of bone marrow (BM) micronucleus assay by microscopic analysis, as well as the flow cytometry (FCM) analysis method using peripheral blood are both considered acceptable, and the later one, which has the advantages of analyzing more cells, data robust and time-saving, are becoming more and more popular and used widely for both regulatory work and in other fields. However, in a special case, a difference of over ~8 folds in changes in cytotoxicity (RET% reduction and PCE% reduction) was observed using the above two different experimental methods in the same experimental design. Then we respectively analyze the data from our own laboratory, and observed that the FCM method for analyzing the peripheral blood samples should be re-considered under the circumstance of testing compounds for the treatment some diseases, such as diabetes, or obesity.

Methods and Materials

Flow cytometry (FCM) analysis method：After administration, 0.4 mL/rat of peripheral blood was collected from the jugular vein of rats, blood samples were diluted by anticoagulant, fixed and labeled, after which 20,000 reticulocytes are collected by flow cytometry for micronucleus analysis.

Microscopic analysis: After administration, at least one femur was collected from animals, bone marrow cells were processed until spreading on smears. After fixation and staining, 4000 polychromatic erythrocytes were scored under fluorescence microscope for micro-nucleus frequency analysis.

Total 318 *in vivo* micronucleus assay studies conducted at WuXi AppTec (Suzhou) from 2017 to 2022 were collected for the retrospective analysis, 227 were analyzed by flow cytometry analysis and 91 were analyzed by bone marrow analysis.

Results and Discussion

In the retrospective analysis, twelve cases of test compounds with the indication of diabetes, or obesity were identified, where most of which were administered via SC injection (7/12), several others were by oral gavage (4/12) or IV injection (1/12).

Under the circumstances of such cases, where SC or IV injection were used and when peripheral blood were used for FCM analysis, the number of collected RET cells was greatly decreased dramatically from a standard 20,000 (the default setting at WuXi AppTec) to a low level of cell numbers, generally several thousands to even hundreds, due to the high cytotoxicity observed, as a consequence, the depression in RET% is generally greater than 80% even at a very low dosage, which might leads to fake increase in background MN-RET cells or even the invalidity of assays. This issue usually triggers re-testing or additional repeating of the assay, such as 4/12 (~33.3%) of these tested compounds in our respective analysis were retested.

However, when taking bone marrow cells for microscope analysis under the same study design, no obvious cytotoxicity was found in the changes of PCE% in any test article treatment group compared with the control group. One case from our laboratory was that, where animals were treated under the same regime and had both bone marrow and peripheral blood collected for microscopic and FCM analysis separately. There was an obvious reduction (by ~92%) in RET%, but only a slight decrease in PCE% (~11%) under the same dosage when compared with the negative control group, showing a ~8 fold difference in cytotoxicity.

Treatment (mg/kg/day)	RET% (Peripheral Blood)	PCE% (Bone Marrow)
Control Article	3.63 ± 0.42	66.5 ± 4.4
Test Article	0.26 ± 0.19	59.2 ± 4.1
RET% Reduction Ratio	92.84%	NA
PCE% Reduction Ratio	NA	10.98%

Differences in depression of PCE% (bone marrow) or RET% (peripheral blood) over the concurrent control group under the same study design and dose.

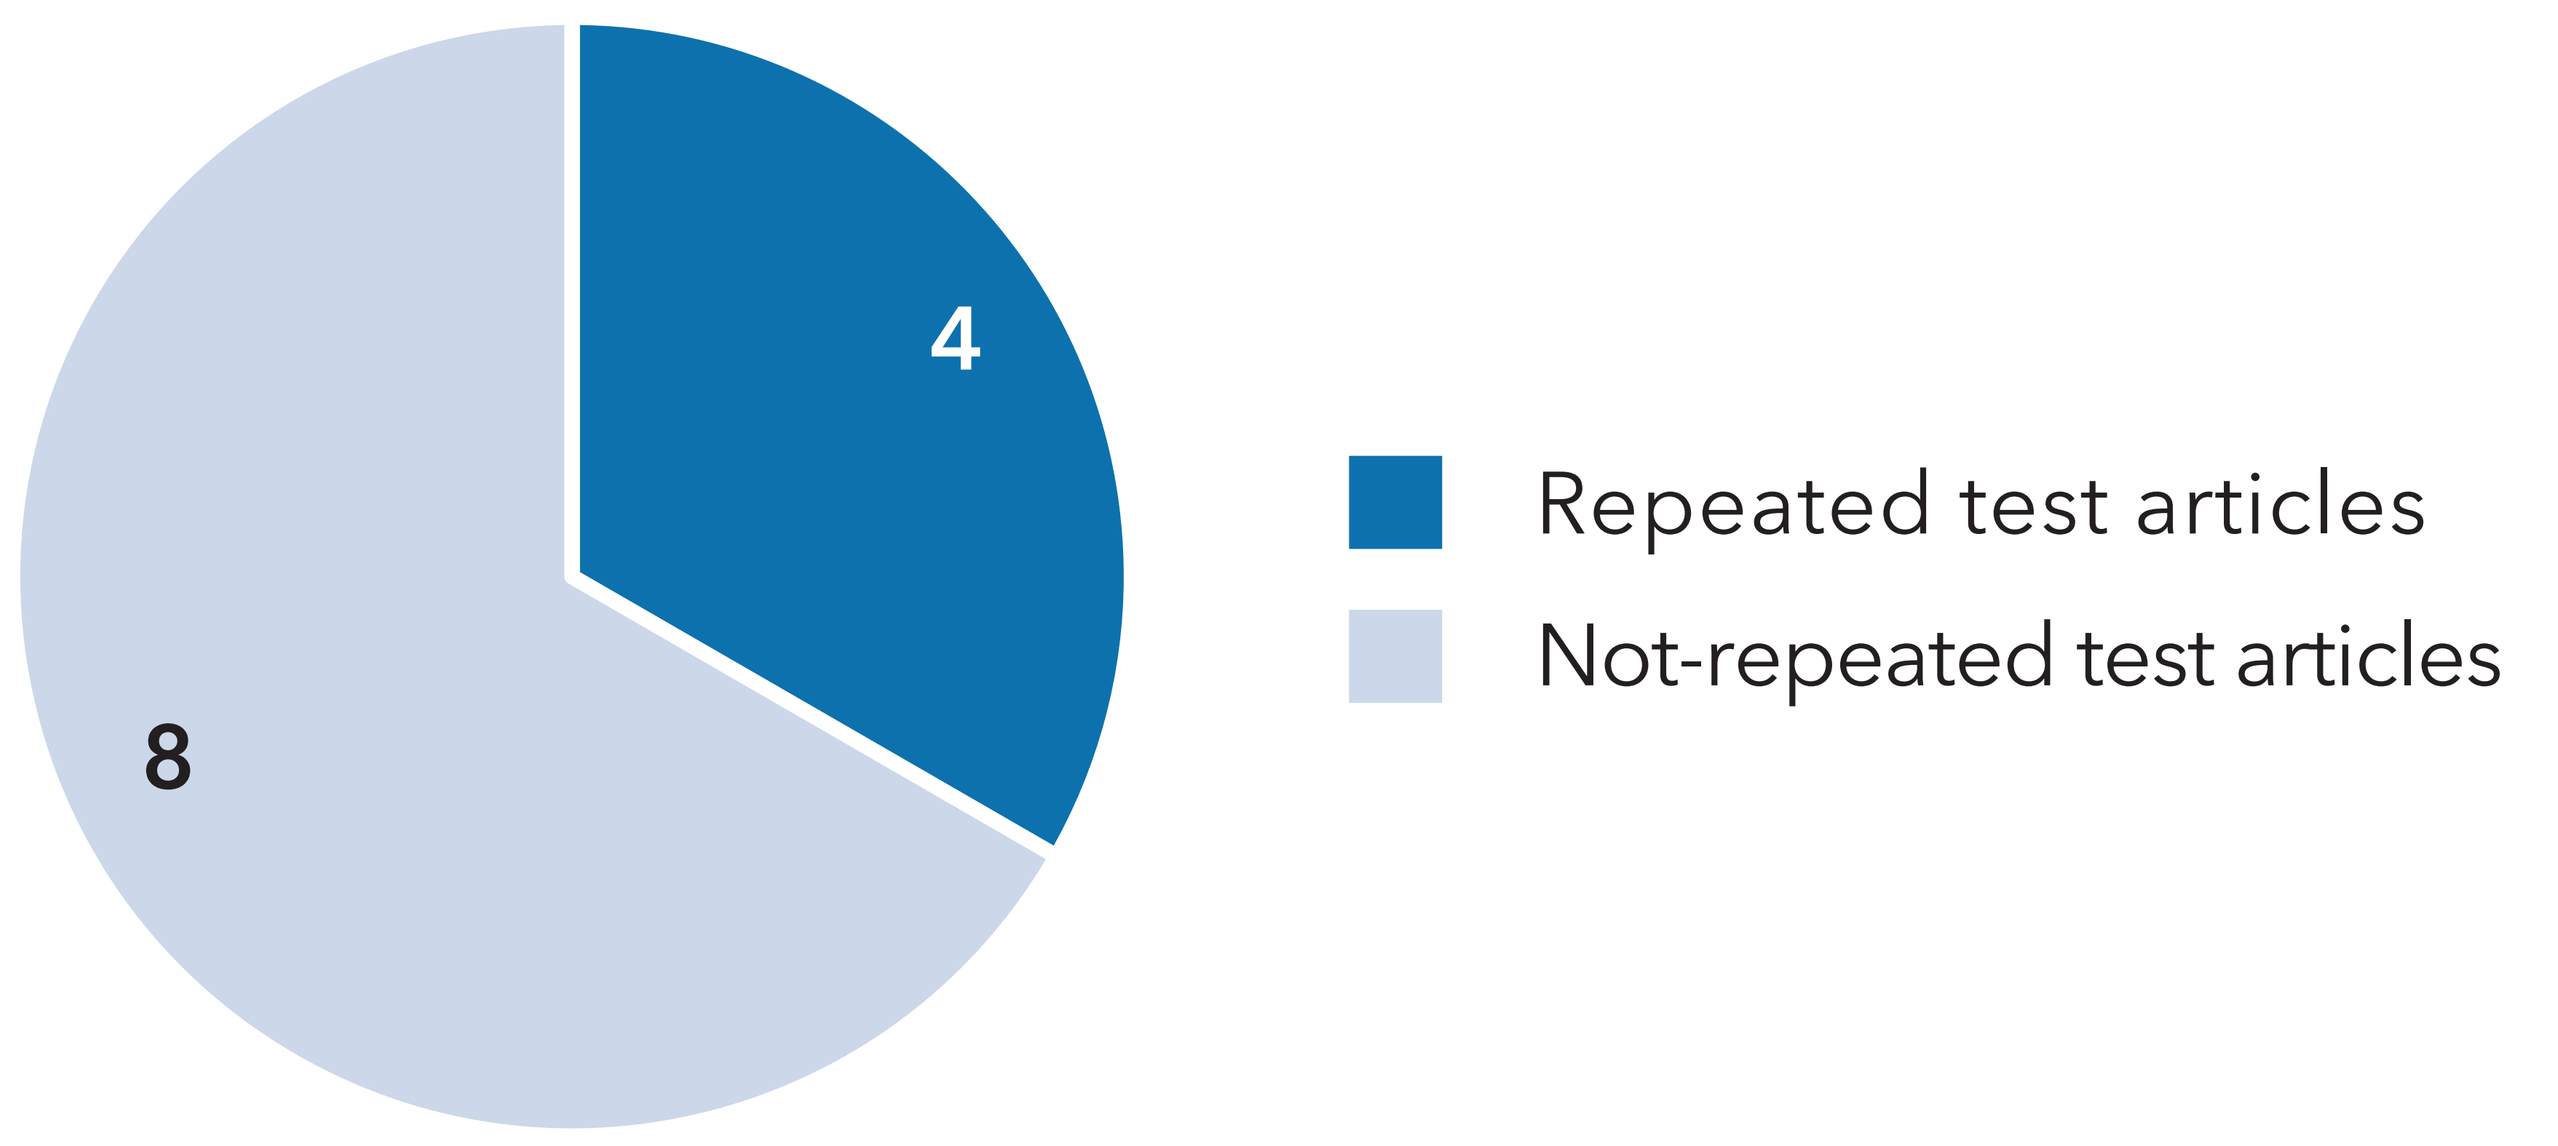
4
8
Repeated test articles
Not-repeated test articles

Differences in depression of PCE% (bone marrow) or RET% (peripheral blood) over the concurrent control group under the same study design and dose.

Conclusion

Given these findings in the *in vivo* peripheral blood micronucleus assay, we propose that BM micronucleus assay should be used when the test compounds are intended for the treatment of indication such as diabetes, or obesity.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou).