Comparison of Changes of Hematology & Coagulation in Fasted & Non-fasted Non-Human Primates

Haiyan Pei; Millie Chen; Xiaoxia Li
WuXi AppTec, Suzhou, China

WuXi AppTec

Abstract: 3282

Purpose

The non-human primates, such as cynomolgus monkey (macaca facicularis), are routinely used in preclinical toxicity testing, and clinical pathology data is a key index to evaluate drug toxicity. Generally, non-human primates are fasted overnight prior to blood collection for clinical pathology analysis. In some occasions, non-fasted blood samples need also to be taken, such as when blood clotting or insufficient blood volume, monkeys that have been fed…etc. Comparison of the changes of hematology and coagulation parameters in fasted and non-fasted is considered necessary in order to provide concrete evidence for data interpretation.

Method

Comparison test on hematology and coagulation analysis in non-human primates that were fasted overnight prior to blood collection (compared procedure) and feed at least one hour and two hours (test procedure) prior to blood collection were conducted. Evaluation Criteria: Based on CLSI EP15-A2, the bias in reportable percent ($\overline{b\%}$) between the two procedures was calculated, which should less than or equal to the quality requirement of laboratory (1/2 total allowable error).

Material

Non-Human Primates, 2-4 years old

- Guangdong Blooming-Spring Biological Technology Development Co., Ltd
- Data of animals from training were used
- Housing-Animals were singly housed in stainless steel cages equipped with a metal bar type -floor and an automatic watering valve
- All studies were maintained under the same conditions: Temperature 22 ± 4 °C, Humidity 50 ± 20% and Light Cycle 12 hours light and 12 hours dark

Laboratory investigations

- Blood samples for hematology and coagulation evaluations were obtained from a cephalic or saphenous vein
- Anticoagulant: Hematology- K2EDTA & Coagulation- Sodium Citrate
- Instrument: Hematology- Advia 2120i Hematology Analyzer & Coagulation- BE Thrombolyzer XRM Coagulation Analyzer

Hematology Comparison Test Results

Parameter	WBC			RBC			HGB			PLT			MCV		
Species	Test I	Test II	Compare	Test I	Test II	Compare	Test I	Test II	Compare	Test I	Test II	Compare	Test I	Test II	Compare
Monkey	15.94	26.72	17.14	5.36	5.29	5.20	13.9	13.5	13.3	413	398	487	81.8	81.3	83.0
	10.40	16.92	5.85	5.28	5.33	5.69	12.4	12.5	13.5	446	486	466	78.3	78.5	78.9
	13.00	14.15	10.25	5.58	5.54	5.73	14.2	14.1	14.8	376	359	386	83.4	83.4	82.9
	12.89	12.59	10.34	5.53	5.28	5.58	13.3	12.6	13.5	436	411	461	78.6	76.1	76.3
	23.75	24.39	12.96	5.81	5.61	5.68	14.0	13.5	13.7	265	224	233	82.5	80.1	82.0

Coagulation Comparison Test Results

Parameter	PT			APTT			FIB			TT		
Species	Test I	Test II	Compare	Test I	Test II	Compare	Test I	Test II	Compare	Test I	Test II	Compare
Monkey	11.3	10.7	14	20.3	20.1	20.7	2.47	2.37	2.51	12.2	11.9	12.3
	12.6	14.3	11.4	20.8	19.8	19.4	1.99	2.1	2.29	11.2	11.3	10.8
	11.8	12.5	11.4	18.7	18.8	18.8	1.97	1.99	2.34	12.8	12.9	12.4
	11.1	11	10.7	16.7	16.9	16.3	2.7	2.36	2.77	11.3	11.9	11.7
	11.8	10.7	12.7	19.3	18.8	18.6	2.38	1.81	2.33	10.8	10.4	11.3

Comparison	Bias in Reportable Percent for Hematology Parameter					Bias in Reportable Percent for Coagulation Parameter			
Parameter	WBC	RBC	HGB	PLT	MCV	PT	APTT	TT	FIB
Test I	41.1	-1.1	-1.4	-2.8	0.4	-1.7	2.2	-6.2	-0.3
Test II	78.6	-2.9	-3.8	-7.1	-0.9	-0.3	0.8	-13.2	-.02

Note: Test I: Test Procedure I: Animals were feed at least one hour ; Test II: Test Procedure II: Animals were feed at least two hours; Compare: Compared procedure: Animals were fasted overnight. Total allowable error for WBC, RBC, HGB, PLT, MCV, PT, APTT, FIB, TT is ±15%, ±6%. ±7%, ±25%, ±7%, ±15%, ±15%, ±15% and ±20%.

Comparison Results

The bias in reportable percent between animals that were fasted and animals were feed at least one hour and two hours were 41.1 and 78.6 for hematology parameters leukocyte count (WBC) , -1.1 and -2.9 for erythrocyte count (RBC), -1.4 and -3.8 for hemoglobin concentration (HGB), -2.8 and -7.1 for platelet count (PLT), and 0.4 and -0.9 for mean corpuscular volume (MCV) and -1.7 and -0.3 for coagulation parameters prothrombin time (PT), 2.2 and 0.8 for activated partial thromboplastin time (APTT), -6.2 and -13.2 for fibrinogen (FIB) and -0.3 and-0.2 for thrombin time (TT).

Comparison Conclusion

There is no significant difference for coagulation analysis between animals that were fasted overnight and were feed at least one hour prior to blood collection. The blood samples for coagulation analysis may be collected approximately 1h after non-human primates were fed. For hematology test, it is suggested that non-human primates will be fasted overnight prior to blood collection.