Not All Literature-Based Structural Alerts Lead to Mutagenicity

Liwen Gao; Dongping Li; Ruoya Zhou; Jiahui Wang; Xiaofen Ren; Suqin Jiang; Millie Chen; Xiaoxia Li; Robert Zhang
WuXi AppTec (Suzhou) Co., Ltd., Suzhou, China

Abstract / Poster Number: 4319 / M817

Background and Purpose

DNA-reactive substances are capable of directly damaging DNA even at low concentrations, which can result in mutations and potentially lead to cancer. According to reports, there is a strong correlation between structural alerts indicative of DNA reactivity and mutagenic potential, and structure-based assessments are useful for predicting bacterial mutagenicity outcomes. There are a variety of approaches to conduct this evaluation including a review of the literature-based structural alerts (LBSAs), and/or computational toxicology assessment. The mutagenic potential can be typically identified using bacterial reverse mutation assay. In order to gain a better understanding of the correlation between structural alerts and mutagenic potential, and to facilitate expert review using LBSAs, we conducted a retrospective analysis of bacterial reverse mutation data with structures generated in our laboratory.

Methods

The bacterial reverse mutation (Ames) assay was performed in compliance with ICH S2(R1) and OECD TG471, some studies are conducted using miniaturized versions of standard assays, such as the mini-Ames assay. The Ames studies conducted at WuXi AppTec (Suzhou) Co., Ltd. from 2018 to 2025 were collected for the retrospective analysis. Only data with available structural information are included, and these mainly pertain to impurities. The structures are classified based on LBSAs, such as those published by Ashby, Tennant and Müller.

Results

Of the 392 compounds tested in the Ames, 279 (71.2%) were concluded to be negative and 113 (28.8%) were mutagenic. Among these 392 compounds, there are 242 compounds containing LBSAs, and the remaining 150 compounds lack LBSAs. Out of 150 compounds without LBSAs, 14 (9.3%) were mutagenic. For 242 compounds containing LBSAs (totally 15 categories), only 99 (40.9%) were actually mutagenic in the Ames test, indicating that relying solely on LBSAs without taking mitigating factors into account can result in excessive sensitivity. The highest frequencies of LBSAs found were in three categories that together comprise over 70% of all alerts: aromatic amines and derivatives, alkyl halides and aromatic nitro compounds. The percentage (58.3%) was higher for aromatic nitro compounds, another two classes (aromatic amines and alkyl halides) that were between 40% and 45% Ames positive. Michael acceptor represents a LBSA with a positive predictivity of 20.0%, with only 3 of the 15 compounds in this class showing a positive Ames test result. The LBSA of epoxides represents a LBSA which serves as a suitable tool for the prediction of mutagenicity, with a positive predictivity of 66.7%. The mutagenicity of LBSAs is impacted by physicochemical properties (such as molecular weight), chemical reactivity and metabolic activation and these expertise can be useful in expert reviews of in silico predictions.

Each alerting structure has its own mutagenic mechanism and can also lead to positive results in specific strains. Two troubleshooting cases are presented here by utilizing these knowledge. Case 1: compound A is a small molecule for metabolic diseases. Compound A with lower purity is positive in TA98/TA100 with S9, but negative in the absence of S9 mix and other strains. Since no LBSAs are identified in compound A, considering the specific positive strains, it is suspected that the positive result is caused by impurities (such as aromatic amines). Aromatic amines are deemed to be specifically acting through the nitrenium ion mechanism of action and generally be positive in TA98/TA100 in the presence of S9 mix. The follow-up studies confirmed the suspicion. Case 2: Invalid Ames due to extreme high revertants even in negative control. Based on the specific positive strains and the alerting structure knowledge, ethylene oxide (EO), a common reagent for sterilization, was suspected (sensitive strains are TA100 and TA1535). The vendor confirmed that EO was used to sterilize the petri dishes. After we switched to petri dishes sterilized by gamma irradiation, everything returned to normal.

Figure 1 Metabolic conversion of aromatic nitro and aromatic amine into an electrophilic nitrenium ion and subsequent reaction with DNA (Nu = nucleophilic DNA center).

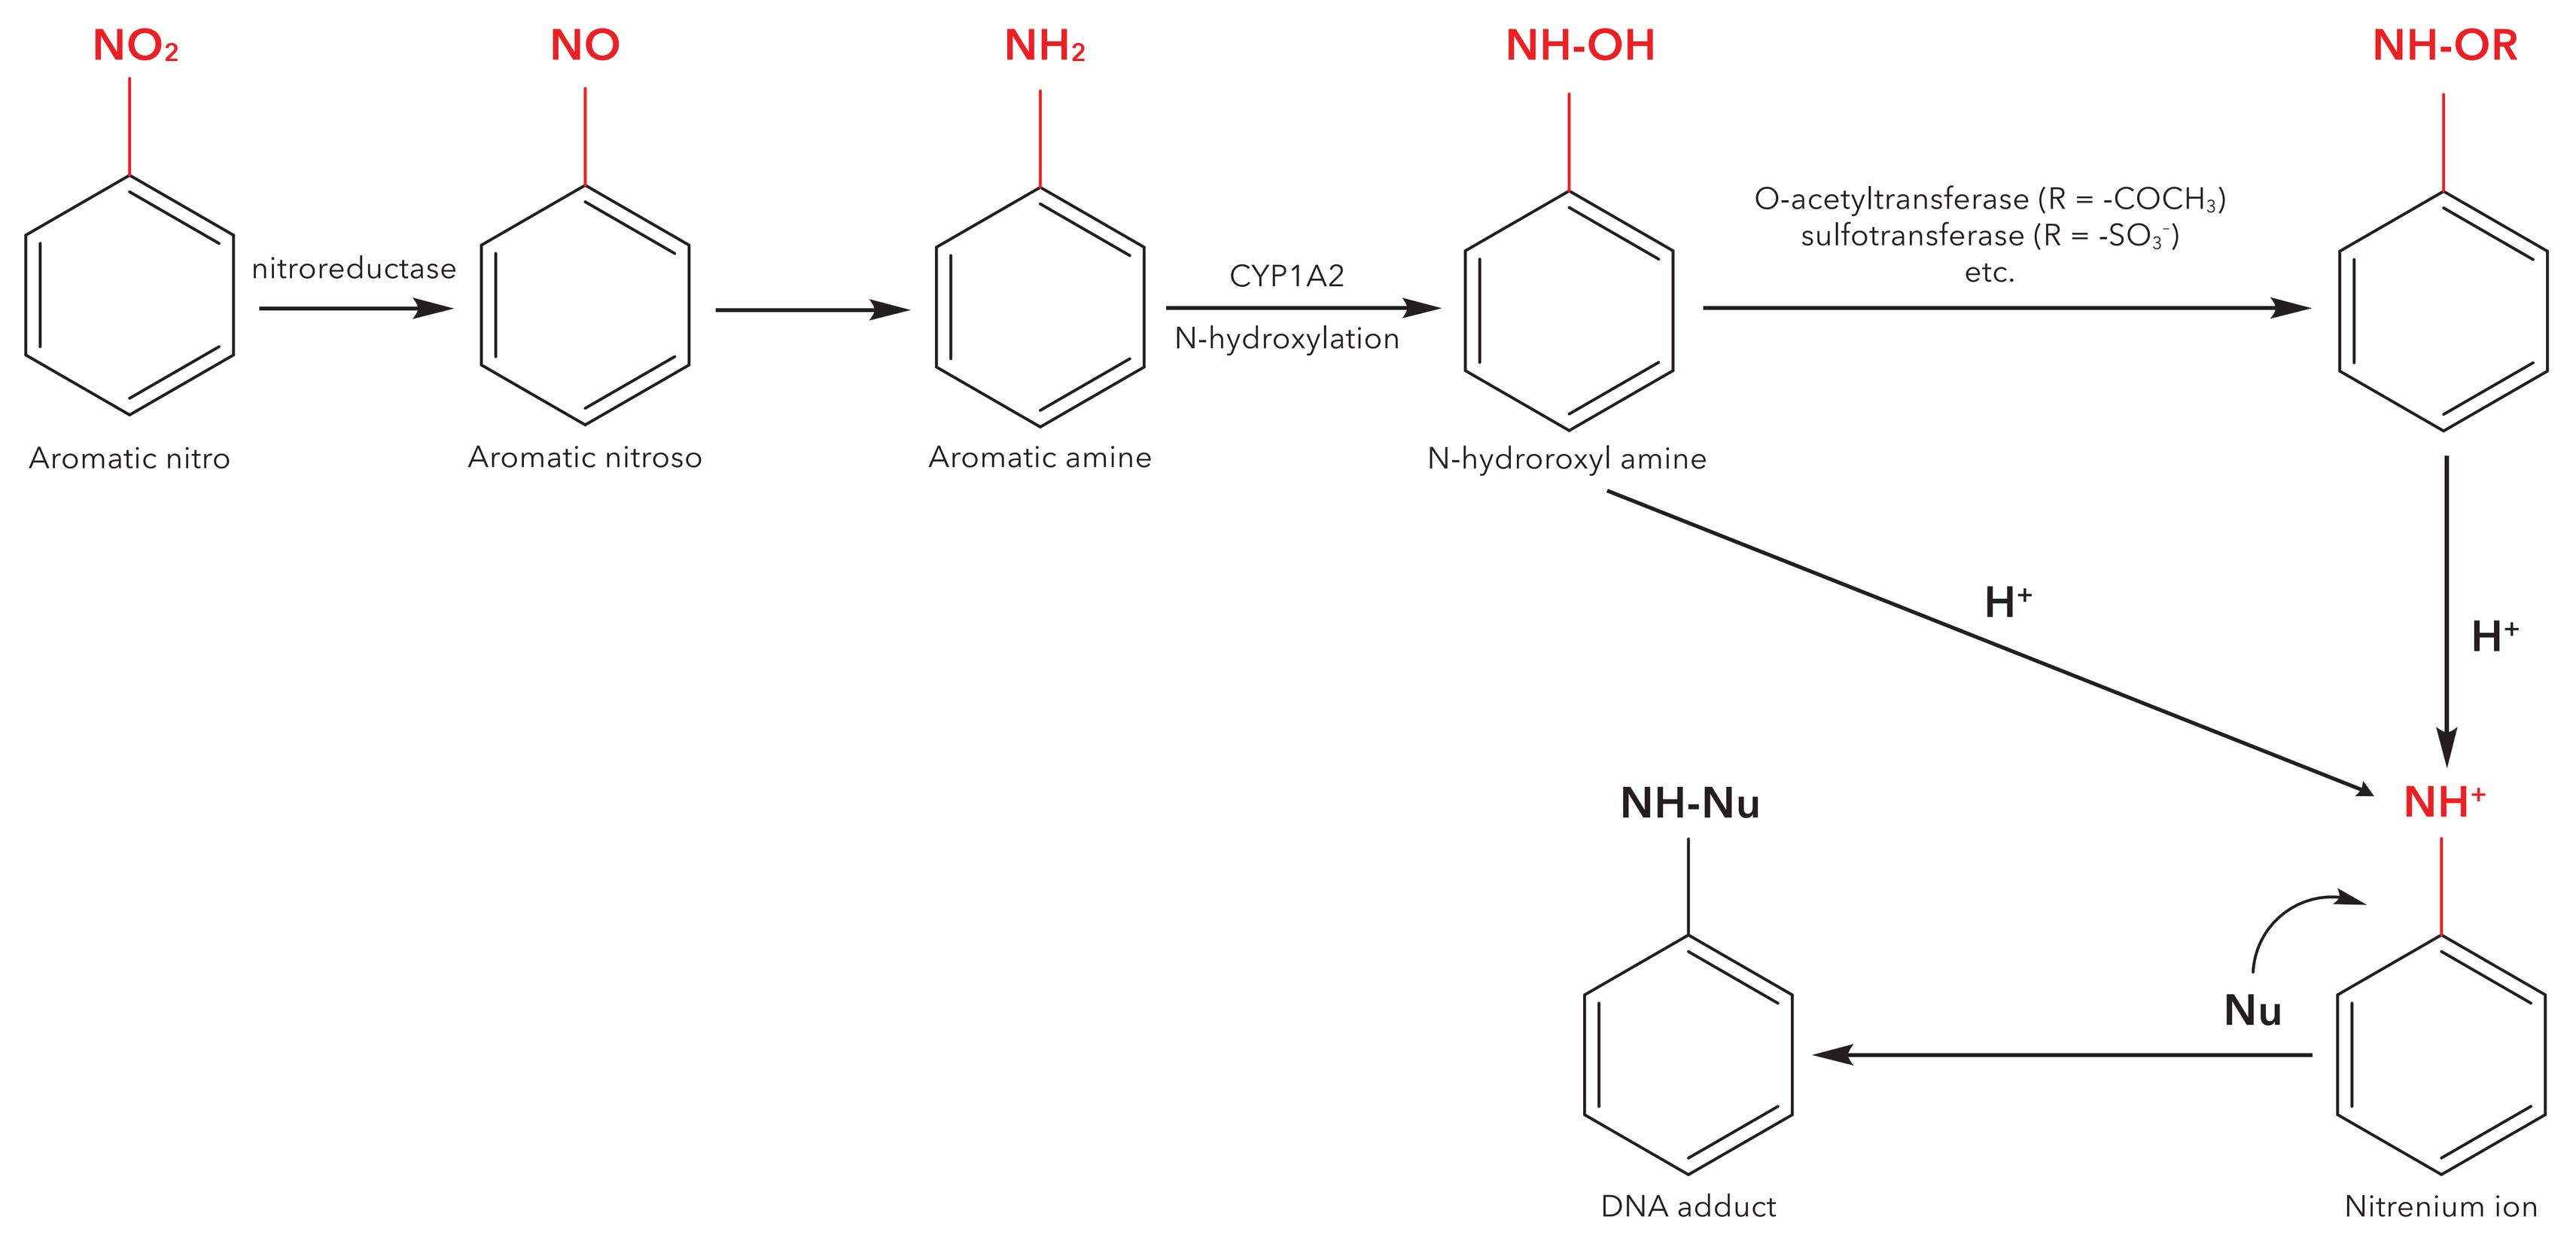
NO2
NO
NH2
NH-OH
NH-OR
nitroreductase
CYP1A2
N-hydroxylation
O-acetyltransferase (R = -COCH3)
sulfotransferase (R = -SO3-)
etc.
Aromatic nitro
Aromatic nitroso
Aromatic amine
N-hydroroxyl amine
H+
H+
NH+
NH-Nu
Nu
DNA adduct
Nitrenium ion

Figure 2 Alkylation of DNA by alkyl halide-containing compounds (Nu = nucleophilic DNA center).

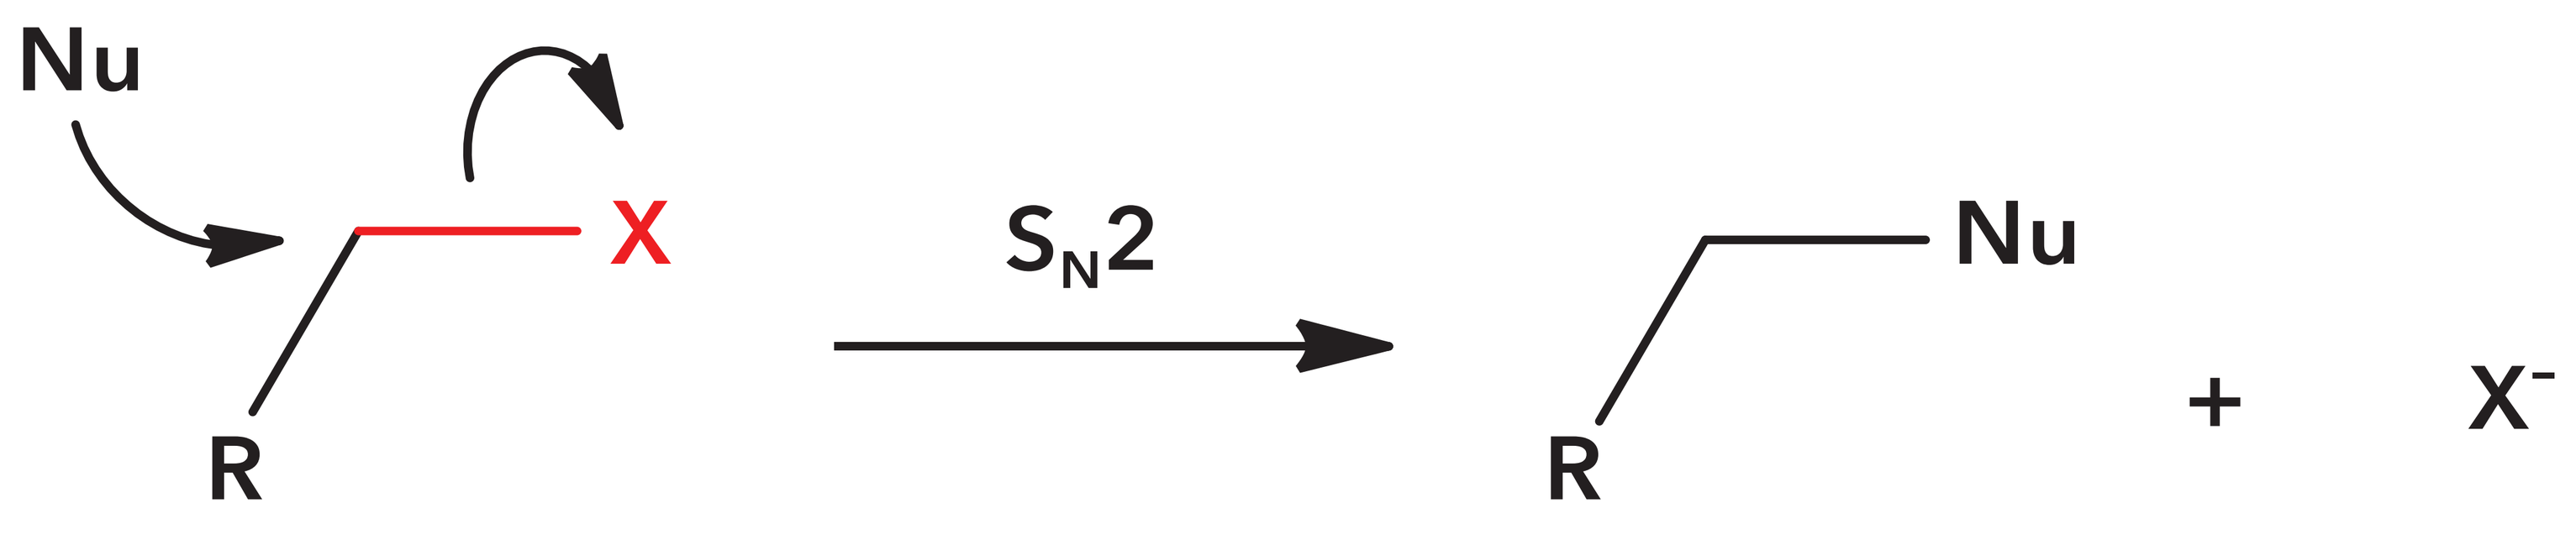
Nu
X
R
SN2
Nu
R
+
X-

Table 1 The Ames Assay Outcomes for the Structurally Alerting Compounds

Structural Alert Category	Ames Positive	Tested in Ames	%Positive
No structural alert	14	150	9.3
Aromatic amines	40	97	41.2
Alkyl halides	22	50	44.0
Aromatic nitro	14	24	58.3
Boracic acids	8	18	44.4
Michael acceptors	3	15	20.0
Epoxides	4	6	66.7
Aldehydes	0	6	0.0
Others (8 categories)	8	26	30.8
Total LBSA	99	242	40.9

Conclusion

In summary, 392 compounds were tested over an 8-year period in our laboratory, of which 113 (28.8%) were found to be mutagenic in the Ames test. The probability of a positive result for compounds containing LBSAs was 40.9%, compared to only 9.3% for those without LBSAs. LBSAs can be a useful tool in expert reviews of in silico predictions and in trouble-shooting. However, relying solely on LBSAs without taking mitigating factors into account may lead to a significantly higher rate of false positives.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and In Vitro Toxicology group at WuXi AppTec (Suzhou) Co., Ltd.

SCAN FOR A COPY