Comparison of Ocular Features in Bama and Göttingen Minipigs

Xian Jing, Chao Guo, Yongbin Zhang, Yongdong Zhou
WuXi AppTec (Suzhou) Co., Ltd., Suzhou, China

WuXi AppTec

Abstract / Poster Number: 4337 / M836

Objective

Minipigs have become increasingly popular in ocular research due to their anatomical similarities to human eyes. In the USA and Europe, the Göttingen minipig is commonly used for ocular research, while the Bama minipig is typically the strain of choice in China. This study aims to provide comparative data on the ocular features between the two strains.

Methods

Bama minipigs aged 4–9 months (females: n=14 eyes; males: n=12 eyes) were used for ocular parameters collection. The data obtained from Bama minipigs was analyzed across three age groups (4–5, 6–7, and 8–9 months) and compared with the published data on Göttingen minipigs of comparable ages. The following parameters were evaluated in this study:

- The anatomical parameters—axial length, anterior chamber depth, lens thickness, and vitreous chamber depth—were measured with an ultrasound scanner; central corneal thickness was measured with a corneal pachymeter; retinal thickness was measured by Optical Coherence Tomography (OCT); vitreous volume was measured using ex vivo eyeballs.
- The physiological parameter, intraocular pressure (IOP), was measured with a tonometer.
- Fundus photographs (FP) were also collected.
- Toxicity bridging studies were also conducted to compare the test-article-related responses between the two strains.

Results

With increasing age, the measured anatomical parameters in Bama minipigs were as below: axial length was 16.39±0.44, 17.89±0.60, and 19.72±0.04 mm; anterior chamber depth was 1.55±0.23, 2.82±0.29, and 3.27±0.19 mm; lens thickness was 5.79±0.24, 6.40±0.22, and 7.20±0.10 mm; vitreous chamber depth was 8.02±0.32, 8.59±0.38, and 9.25±0.05 mm; central corneal thickness was 632±42, 674±58, and 776±13 μm; retinal thickness ranged from 264±21 to 273±12 μm; intraocular pressure ranged from 15±1 mmHg to 17±3 mmHg. The measured vitreous volume was 2.1±0.2 mL.

Compared with the published data, the ocular anatomical parameters, IOP, and FP in Bama minipigs were comparable to those in Göttingen minipigs[1-4] (As shown in Figures 1-6).

In toxicity bridging studies, the endpoints, including ophthalmologic examination, IOP, electroretinography (ERG), OCT, and histopathology examinations, were consistent in both strains.

Figure 1 Lens thickness (mm)

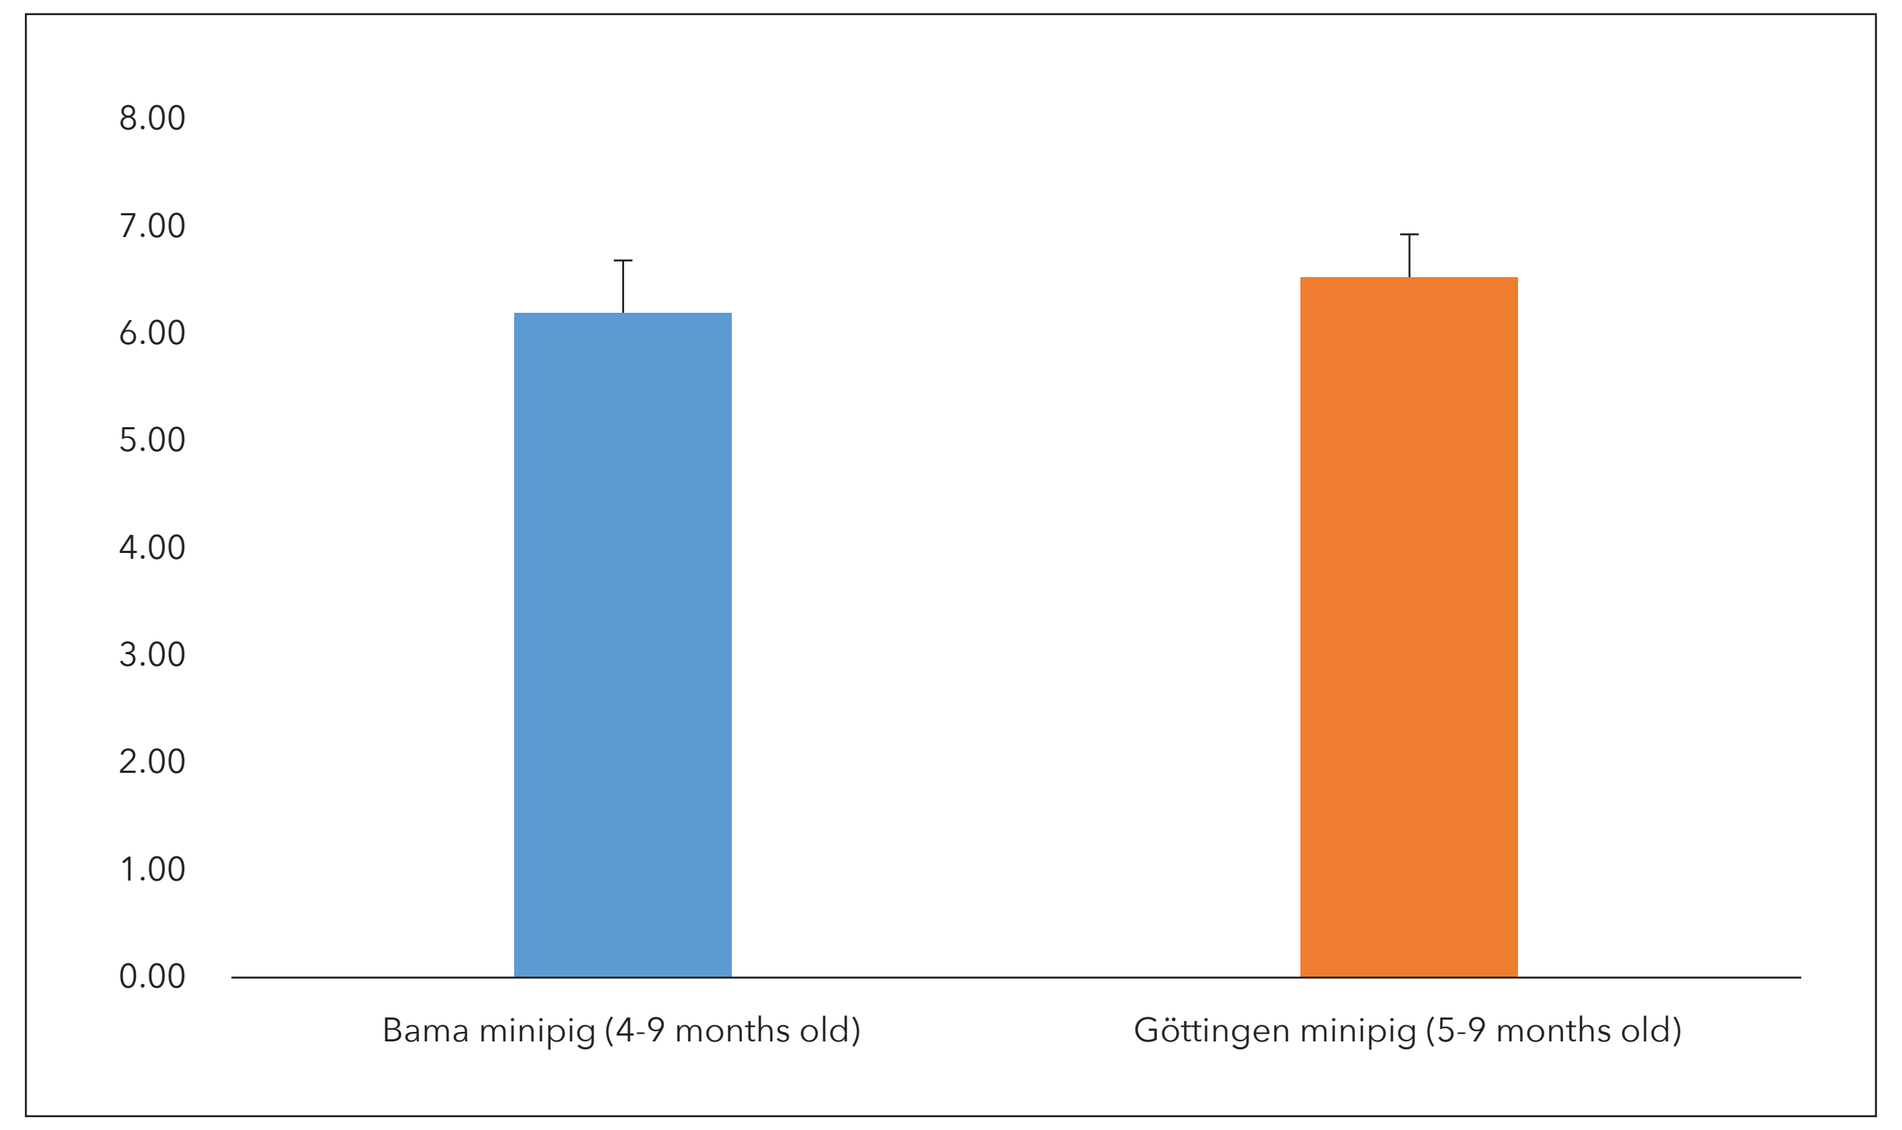
8.00
7.00
6.00
5.00
4.00
3.00
2.00
1.00
0.00
Bama minipig (4-9 months old)
Göttingen minipig (5-9 months old)

Figure 2 Axial length (mm)

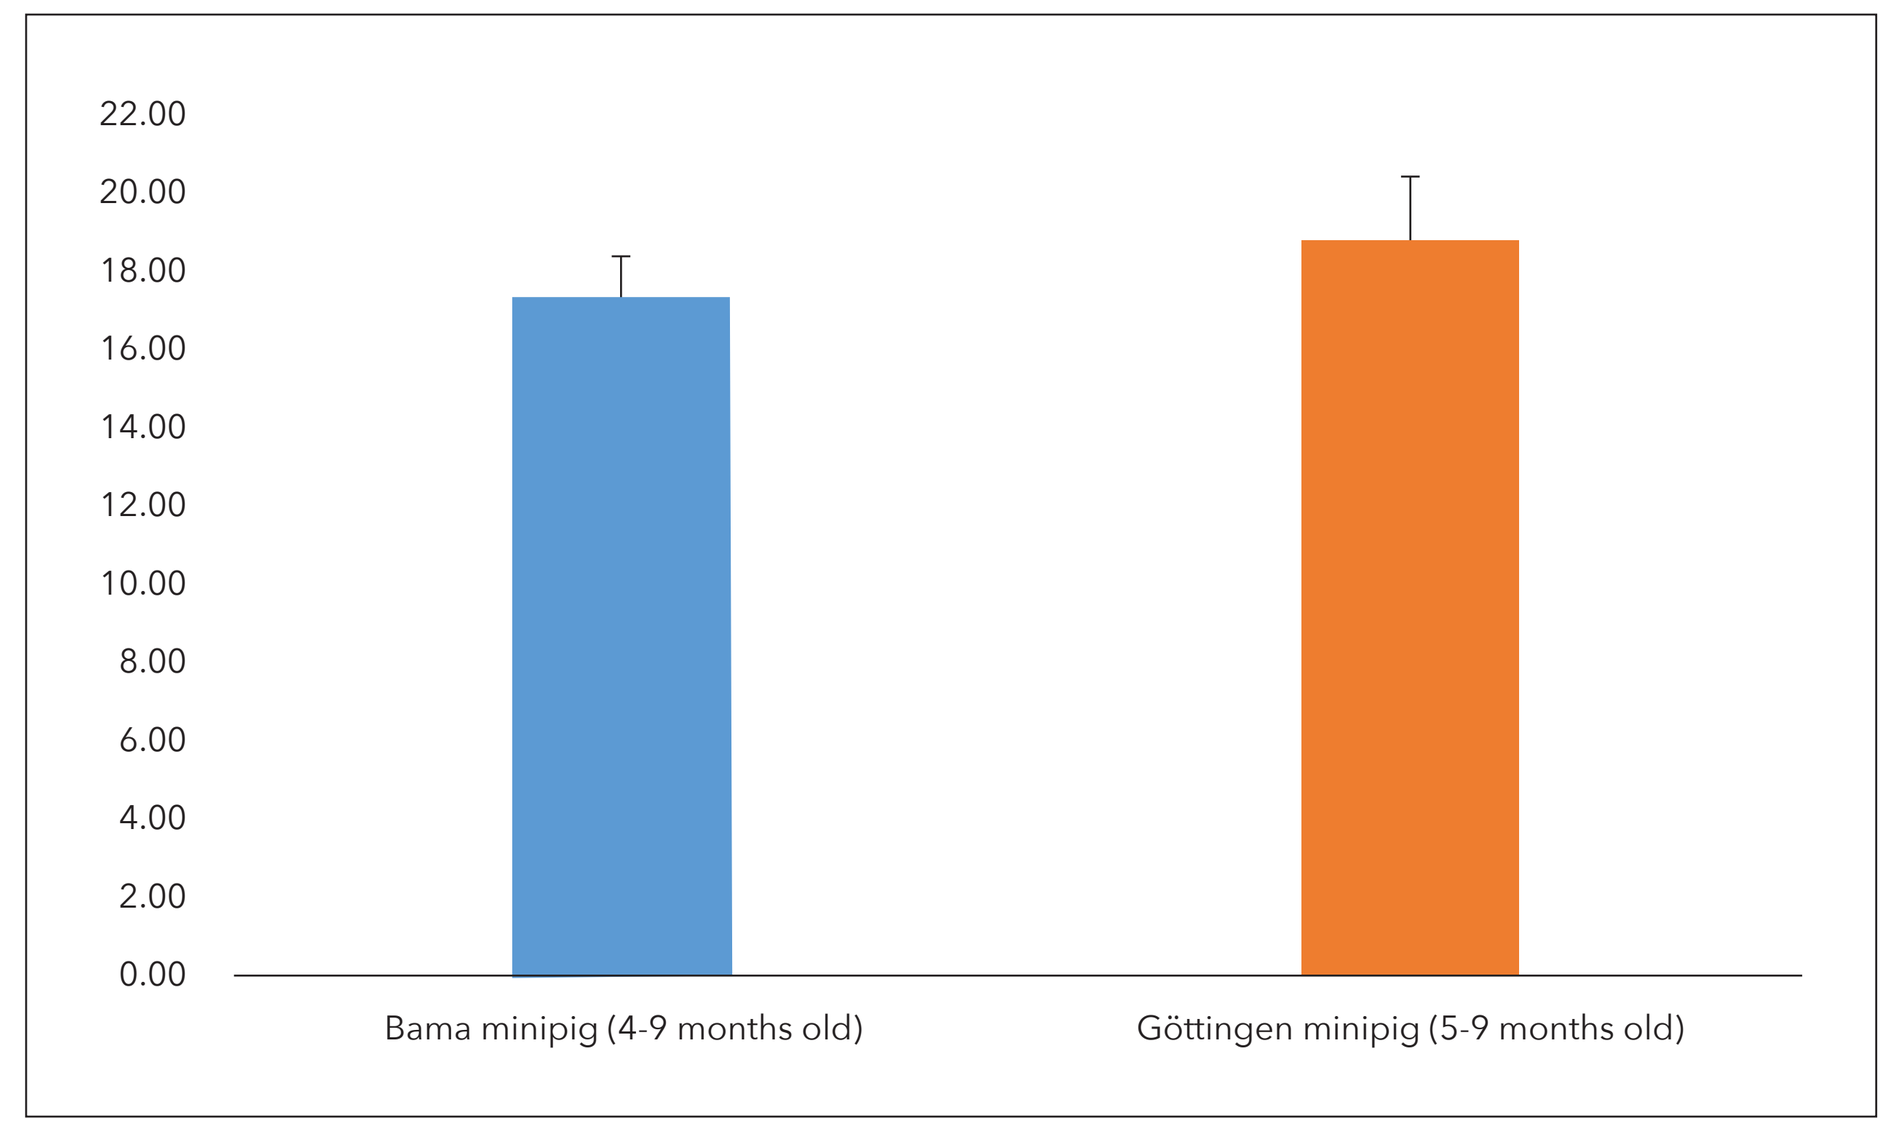
22.00
20.00
18.00
16.00
14.00
12.00
10.00
8.00
6.00
4.00
2.00
0.00
Bama minipig (4-9 months old)
Göttingen minipig (5-9 months old)

Figure 3 Retinal thickness (6mm from optic disc; μm)

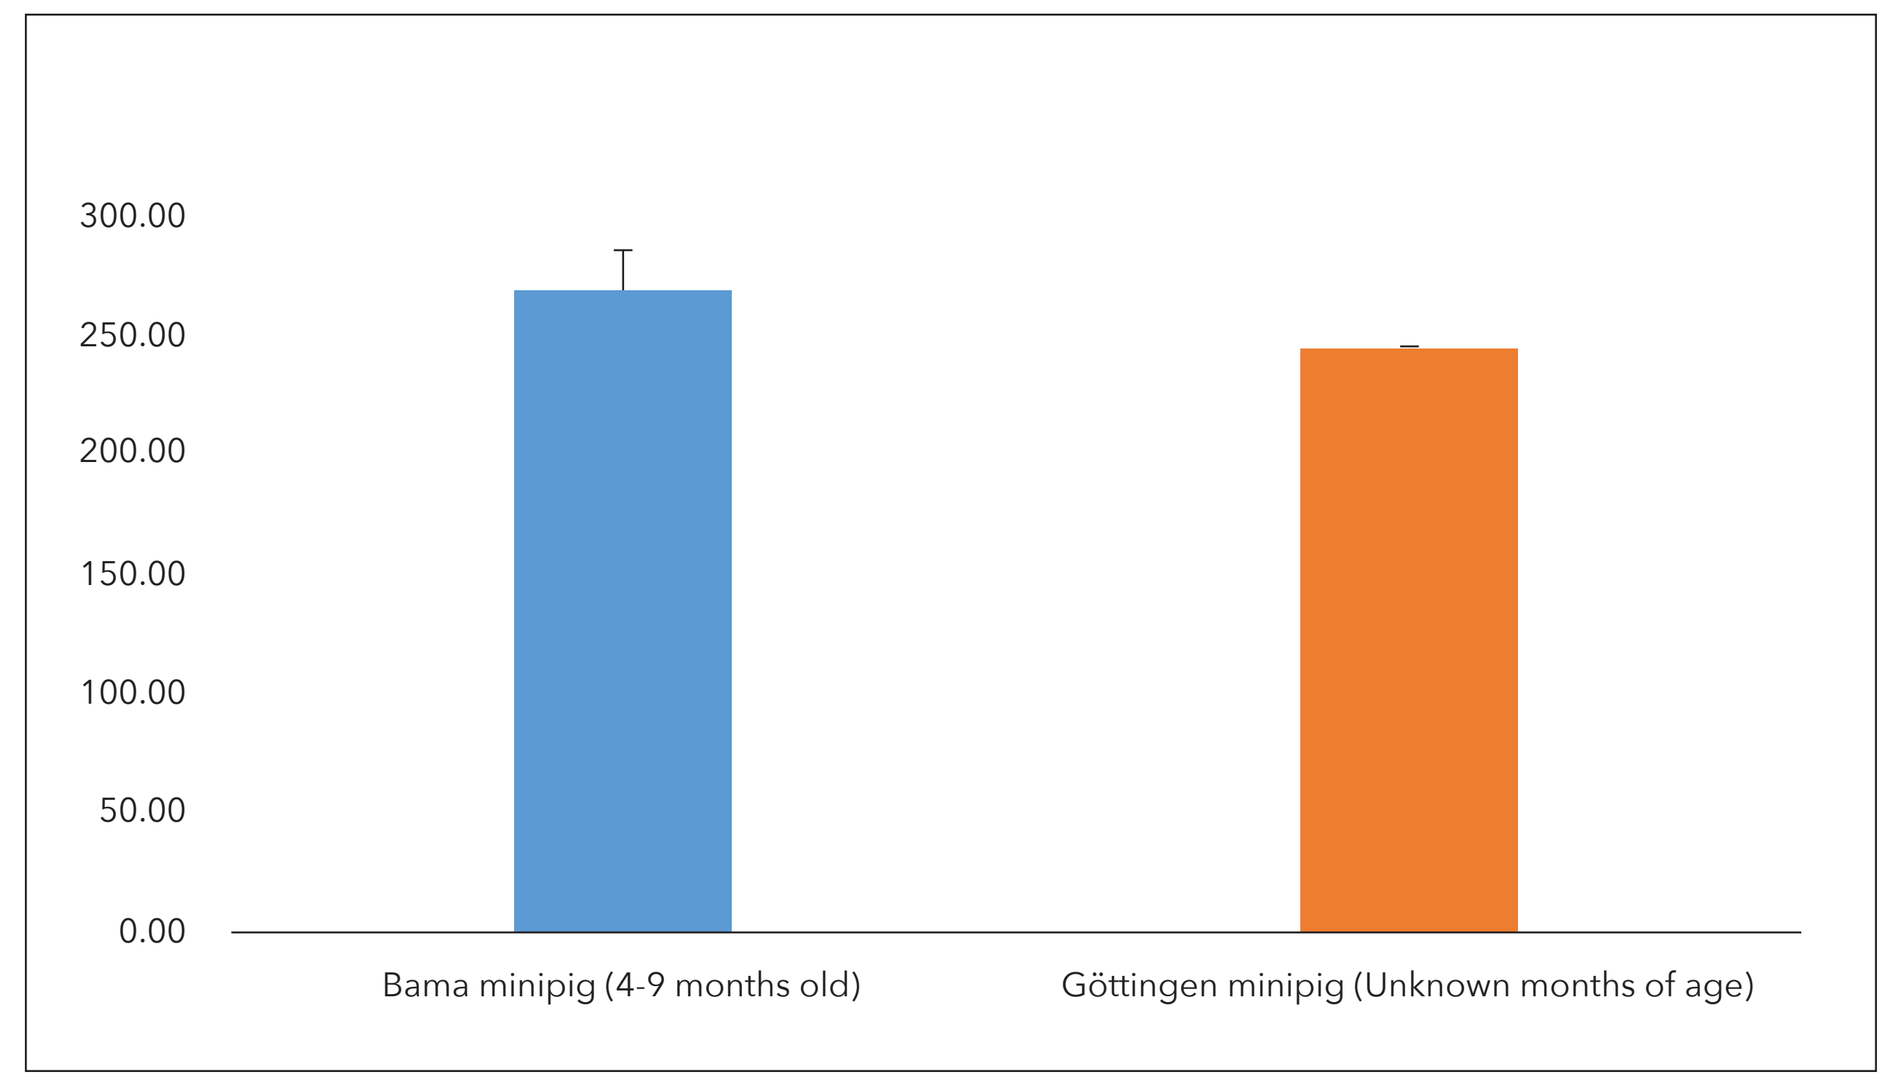
300.00
250.00
200.00
150.00
100.00
50.00
0.00
Bama minipig (4-9 months old)
Göttingen minipig (Unknown months of age)

Figure 4 Central cornea thickness (μm)

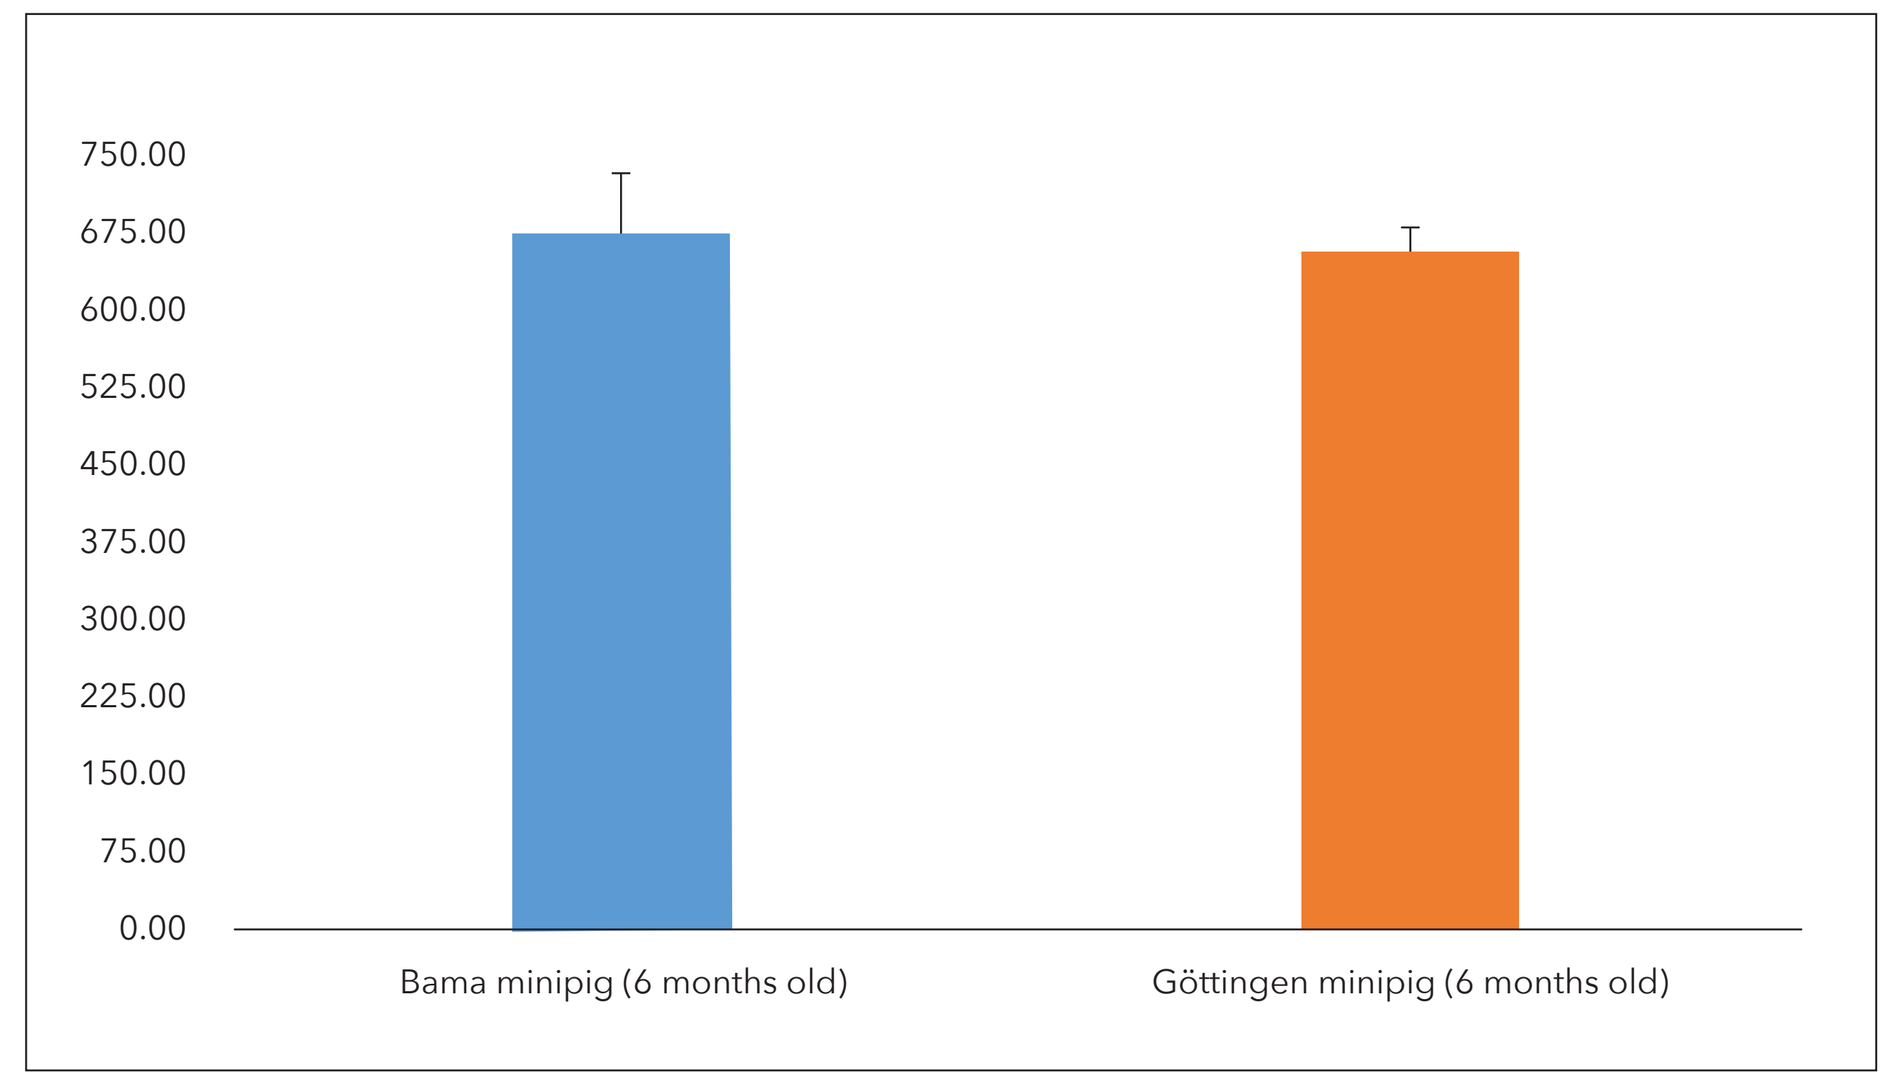
750.00
675.00
600.00
525.00
450.00
375.00
300.00
225.00
150.00
75.00
0.00
Bama minipig (6 months old)
Göttingen minipig (6 months old)

Figure 5 Vitreous volume (mL)

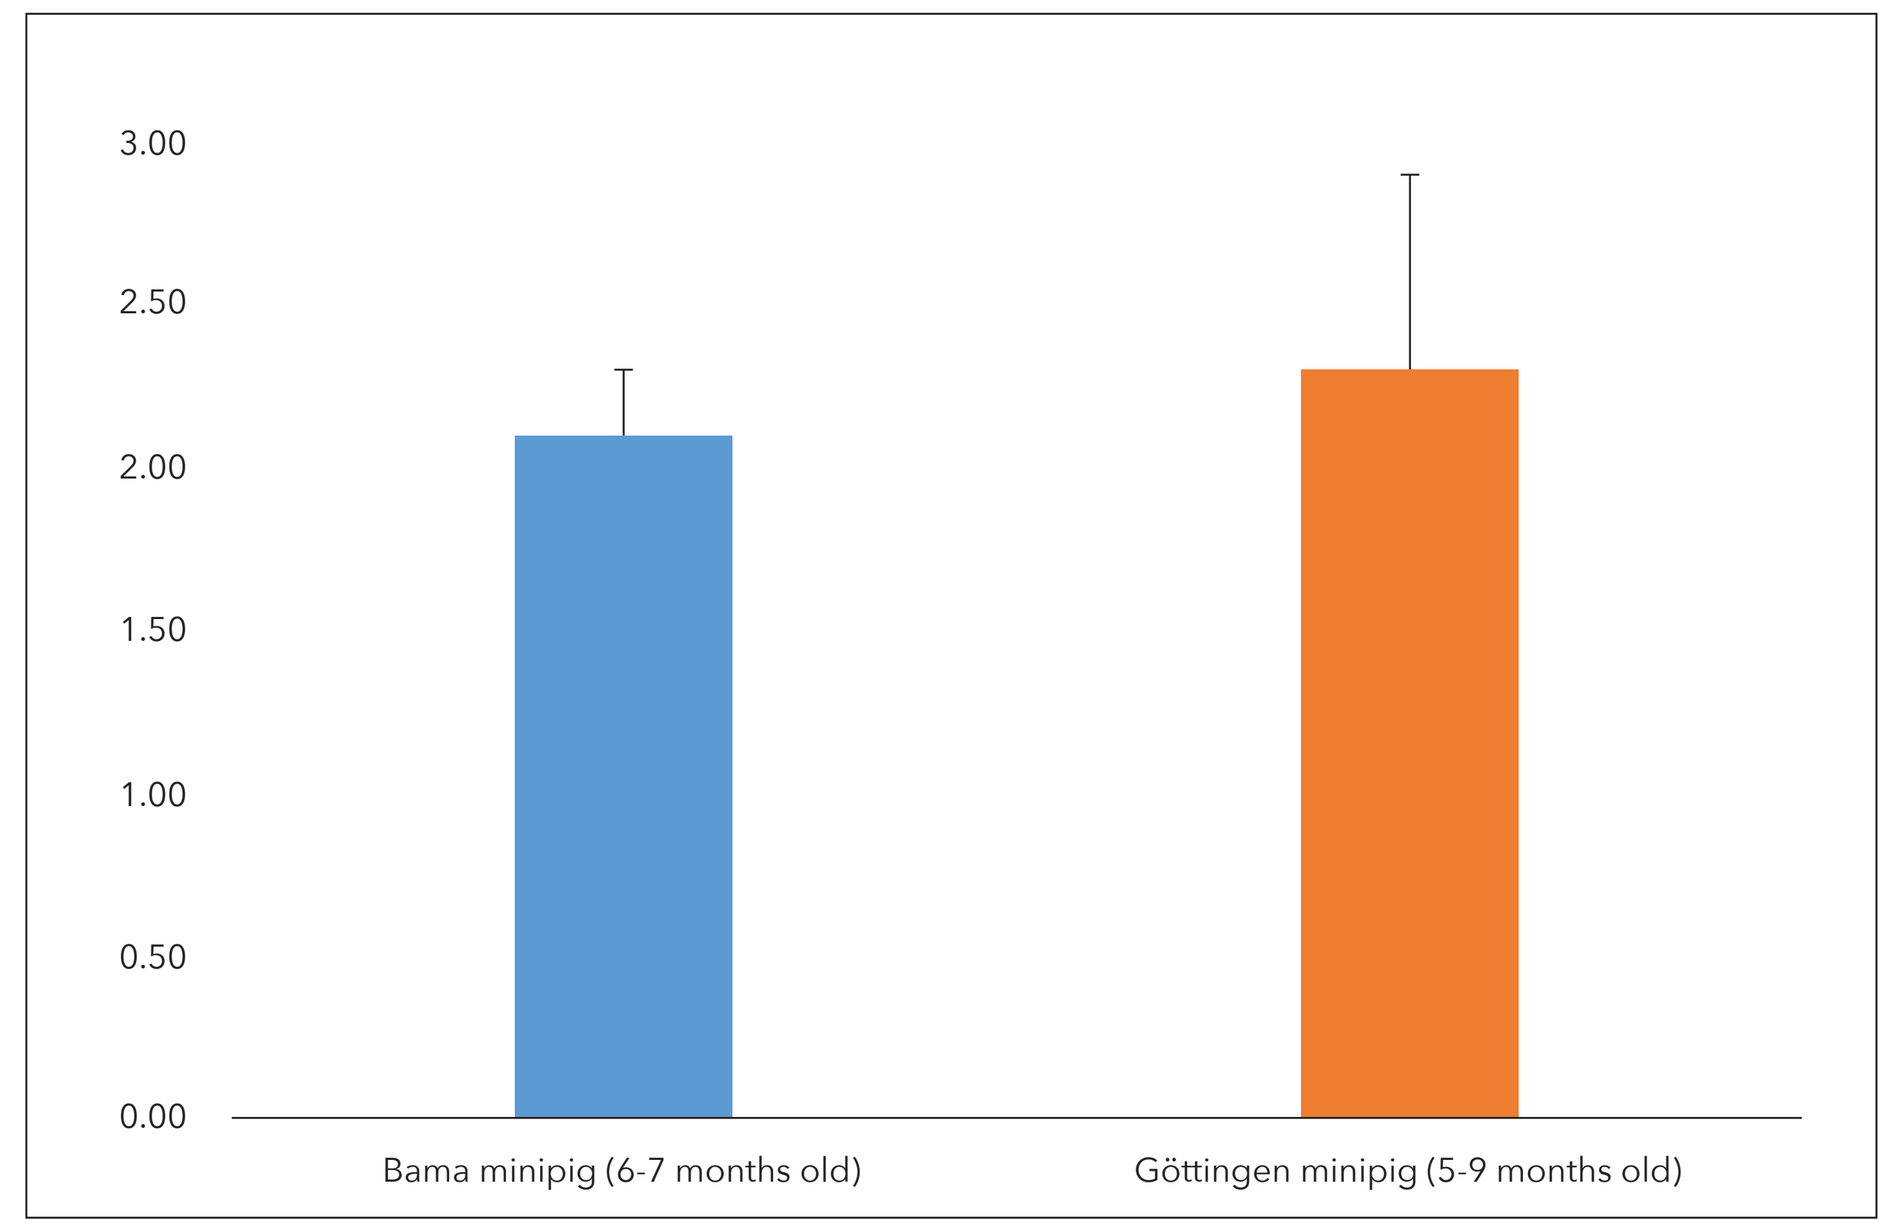
3.00
2.50
2.00
1.50
1.00
0.50
0.00
Bama minipig (6-7 months old)
Göttingen minipig (5-9 months old)

Figure 6 Intraocular pressure (mmHg)

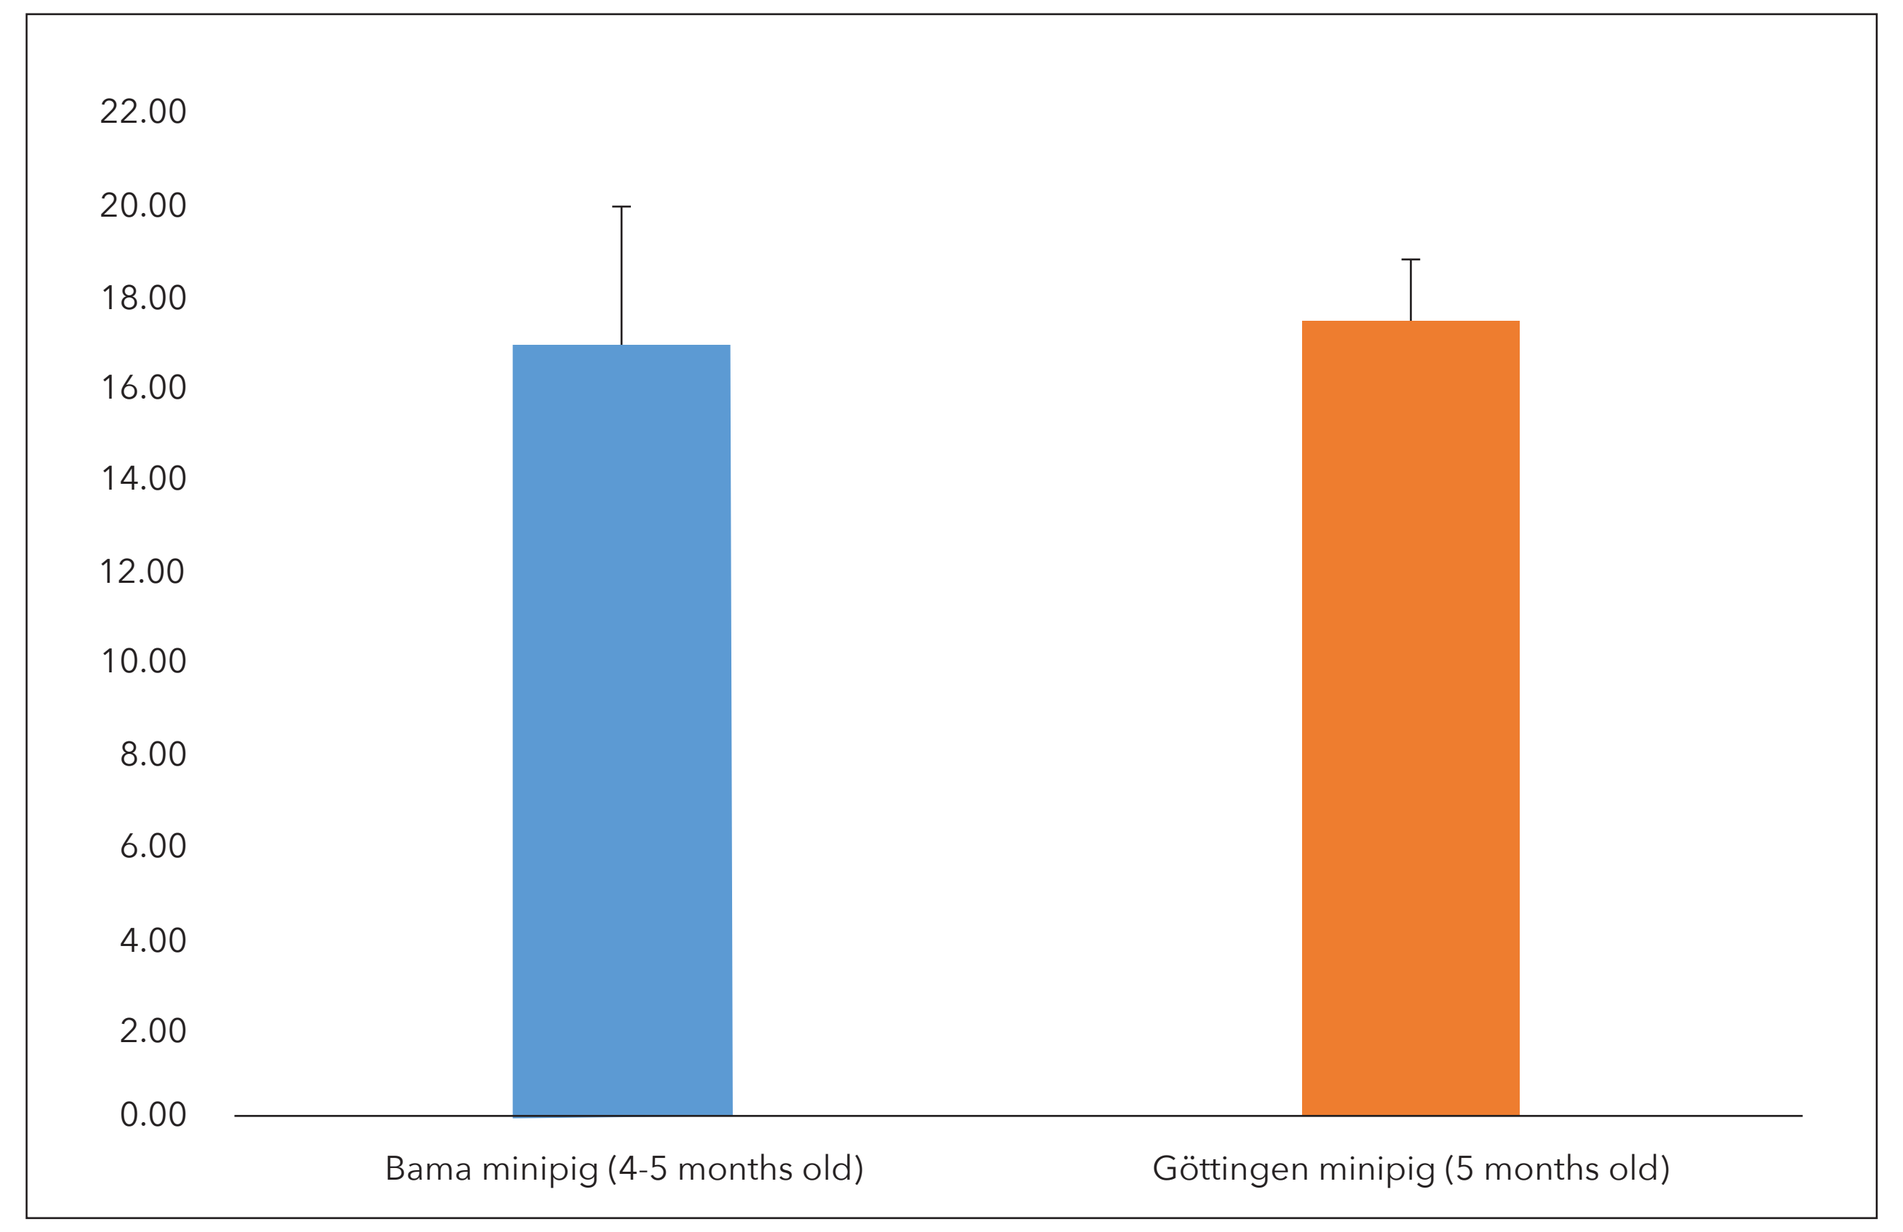
22.00
20.00
18.00
16.00
14.00
12.00
10.00
8.00
6.00
4.00
2.00
0.00
Bama minipig (4-5 months old)
Göttingen minipig (5 months old)

Conclusion

Based on the comparable ocular anatomical parameters, IOP, and FP results, and consistent test article-related responses observed between age-matched Bama and Göttingen minipigs, Bama minipigs could also serve as an appropriate species selection for ocular pharmacokinetics, pharmacodynamics, and toxicology research.

References

[1] Sakaida, T., Shimato, N., Kubota, T., Suzuki, Y., Atsumi, I., Kito, G., Kurata, M., & Kato, H. (2016). Ocular anatomical features in miniature pigs: Parameter measurement of the eyeball and parts of the eye. Animal Eye Research, 35, 1–7.

[2] Carpenter, C. L., Kim, A. Y., & Kashani, A. H. (2018). Normative Retinal Thicknesses in Common Animal Models of Eye Disease Using Spectral Domain Optical Coherence Tomography. Advances in experimental medicine and biology, 1074, 157–166.

[3] Rafat, M., Jabbarvand, M., Sharma, N., Xeroudaki, M., Tabe, S., Omrani, R., Thangavelu, M., Mukwaya, A., Fagerholm, P., Lennikov, A., Askarizadeh, F., & Lagali, N. (2023). Bioengineered corneal tissue for minimally invasive vision restoration in advanced keratoconus in two clinical cohorts. Nature biotechnology, 41(1), 70–81.

[4] Galdos, M., Bayón, A., Rodriguez, F. D., Micó, C., Sharma, S. C., & Vecino, E. (2012). Morphology of retinal vessels in the optic disk in a Göttingen minipig experimental glaucoma model. Veterinary ophthalmology, 15 Suppl 1, 36–46.

SCAN FOR A COPY