Regulatory-Compliant Genetic Toxicology Assessment for Tides

Mengqi Zhang; Liwen Gao; Millie Chen; Xiaoxia Li; Robert Zhang
WuXi AppTec (Suzhou) Co., Ltd., Suzhou, China

Abstract / Poster Number: 4317 / M815

Background and Purpose

Peptide and oligonucleotide therapeutics, commonly grouped as TIDES, are two of the fastest-growing and most promising modalities within the pharmaceutical industry. Oligonucleotides (ONs), comprising single or double strands of RNA or DNA, act via mechanisms such as antisense oligonucleotides (ASOs), small interfering RNAs (siRNAs), microRNAs (miRNAs), small activating RNAs (saRNAs), and aptamers. Peptides mean any polymer that are composed of 40 or fewer amino acids. With the unique properties, Tides have emerged as a prominent force in innovative drug development, to better understand the genotoxicity of Tides, we conducted a retrospective analysis of genotox data for both FDA-approved Tides and the Tides previously evaluated in our laboratory.

Methods

No single assay is able to detect all types of DNA damage, a test battery has been adopted to evaluate the genotoxic liability of novel pharmaceuticals. ICH S2(R1) guidance outlines appropriate *in vitro* and *in vivo* tests. The guidance provides two options for the standard genetic toxicology battery that includes an assessment of mutagenicity in a bacterial reverse mutation assay and genotoxicity evaluation in mammalian cells using *in vitro* and/or *in vivo* tests. Genotoxicity data of Tides that were either approved for marketing by the FDA between 2016 and 2025, or evaluated at WuXi AppTec (Suzhou) Co., Ltd. between 2018 and 2025, were collected for this retrospective analysis.

Results

Percentage of different classes of novel therapeutics approved by the FDA in the span of 2016–2025 of which Tides attribute for 11.2%. A total of 20 oligonucleotide drug products that includes 1 aptamer, 11 antisense oligonucleotides (ASOs), 7 small interfering ribonucleic acids (siRNAs), and 1 mixture of single-stranded and double-stranded polydeoxyribonucleotides have been identified. All oligonucleotide drug products were evaluated for genotoxicity and the results are all negative. A total of 33 peptides that includes 9 Peptide-Drug Conjugates (PDCs); 19 oligopeptides (2-9 amino acids); 1 polypeptide (PP), 32 of them are synthetic. 24/33 (73%) were evaluated for Ames: 1 positive, 2 false positive (due to feeding effect), others are negative. 23/33 (70%) were evaluated *in vitro* mammalian cell test, 16 run for chromosomal aberration assay (CAA), others are for MLA/micronucleus assay (MNT), and they are all negative. 22/33 (67%) were evaluated for *in vivo* micronucleus (12 in rats and 10 in mice), and they are all negative. From 2018 to 2025, there are 19 peptides and 35 oligonucleotides tested in our laboratory. For the 19 peptides, 12/19 (63%) were evaluated for Ames; 15/19 (79%) were evaluated in CAA, and 11/19 (58%) were evaluated for *in vivo* micronucleus, they are all negative. For the 35 oligonucleotides, 35/35 (100%) were evaluated for Ames and CAA, 31/35 (89%) were evaluated for *in vivo* micronucleus, 1 equivocal (WoE was used for evaluating the marginal increase), 2 false positives (one positive is contributed by the excipient, and the other is impacted by the binding between drug product and fluorescent dye), the others are negative.

Figure 1 Battery Approach for Genotoxicity

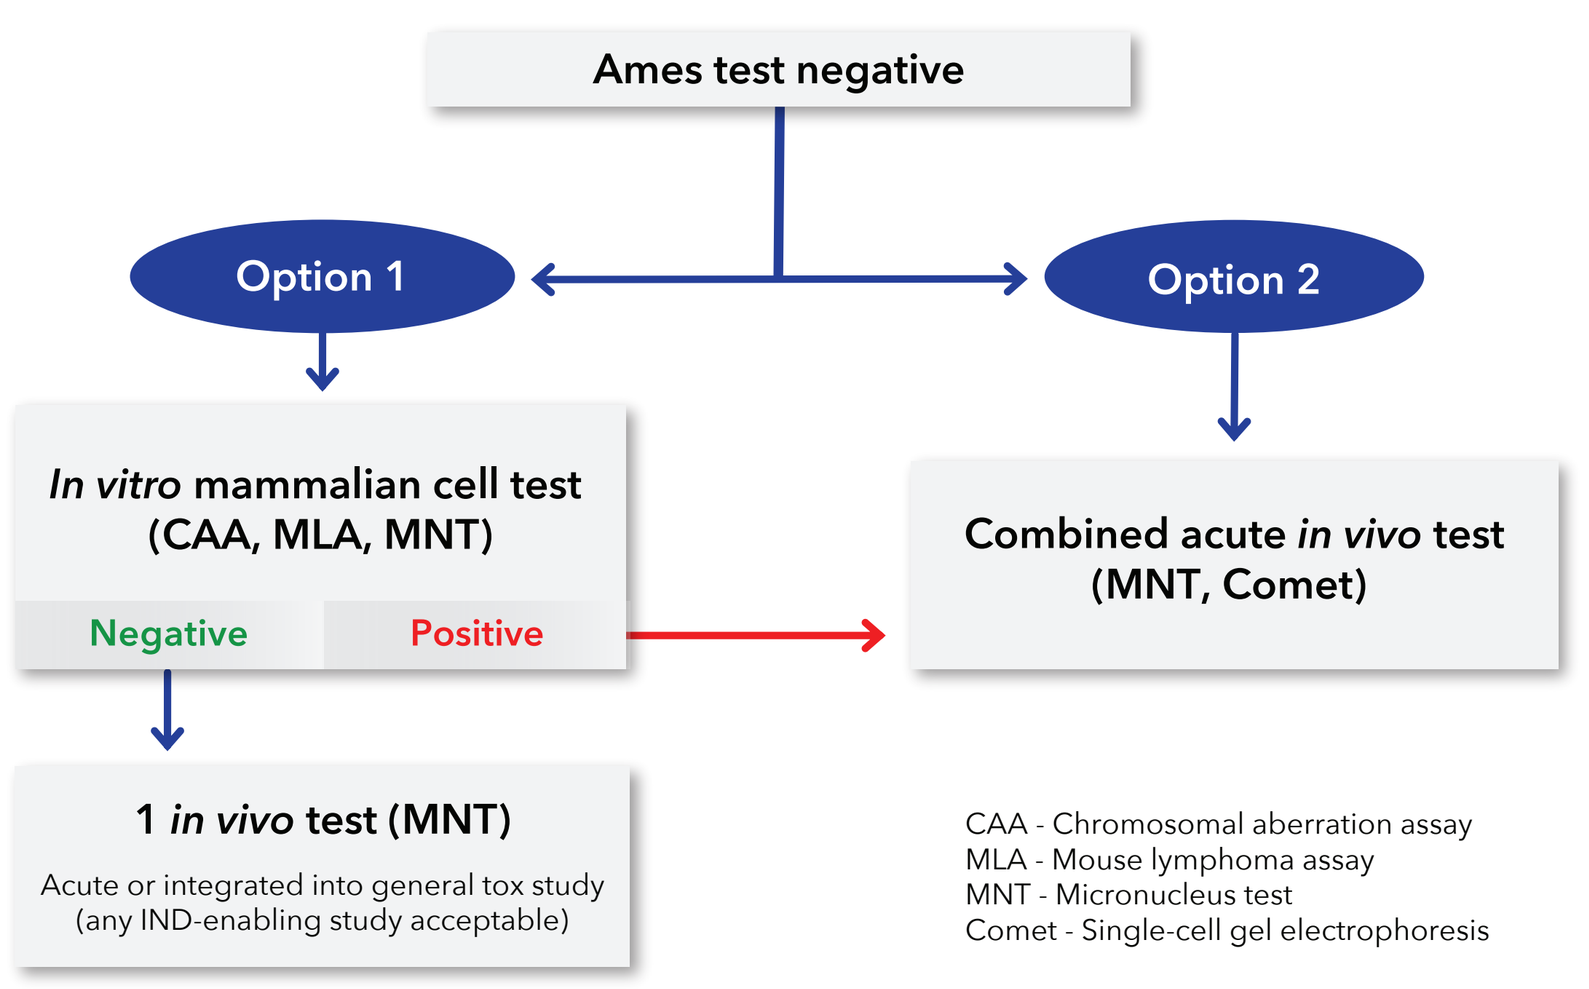
Ames test negative
Option 1
Option 2
In vitro mammalian cell test (CAA, MLA, MNT)
Negative
Positive
Combined acute in vivo test (MNT, Comet)
1 in vivo test (MNT)
Acute or integrated into general tox study (any IND-enabling study acceptable)
CAA - Chromosomal aberration assay
MLA - Mouse lymphoma assay
MNT - Micronucleus test
Comet - Single-cell gel electrophoresis

Figure 2 FDA-Approved Tides (2016–2025)

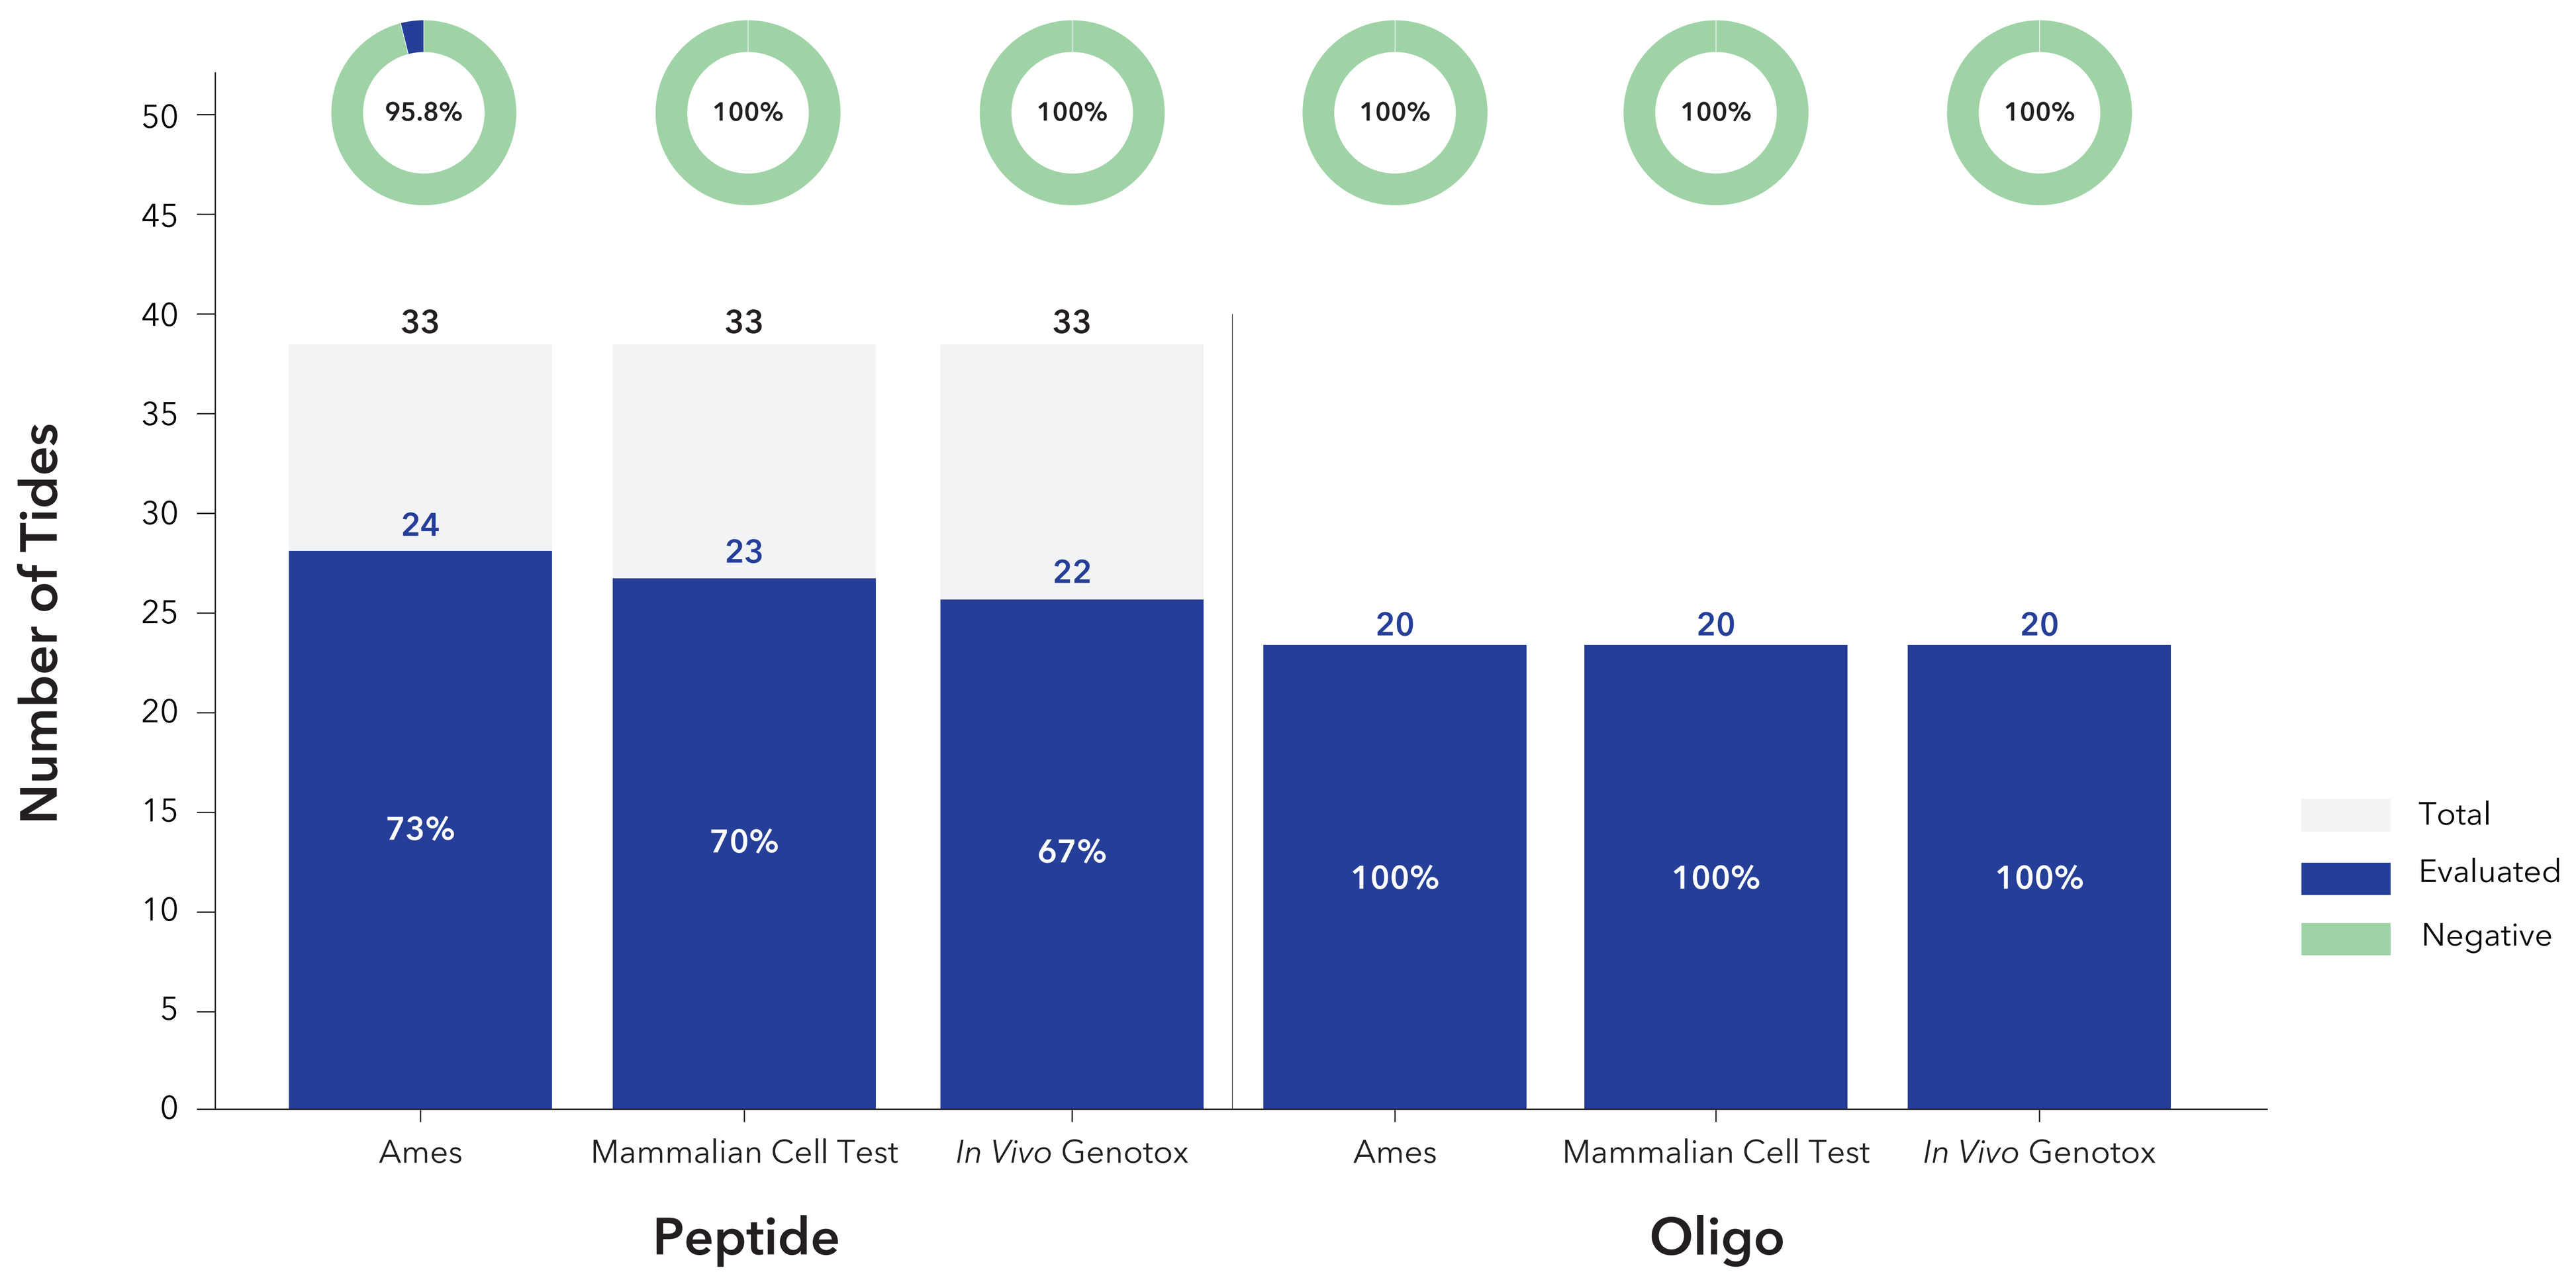
Number of Tides
0
5
10
15
20
25
30
35
40
45
50
95.8%
100%
100%
100%
100%
100%
33
33
33
24
23
22
20
20
20
73%
70%
67%
100%
100%
100%
Ames
Mammalian Cell Test
In Vivo Genotox
Ames
Mammalian Cell Test
In Vivo Genotox
Peptide
Oligo
Total
Evaluated
Negative

Figure 3 Tides Tested in Genetic and *In Vitro* Toxicology Laboratory (2018-2025)

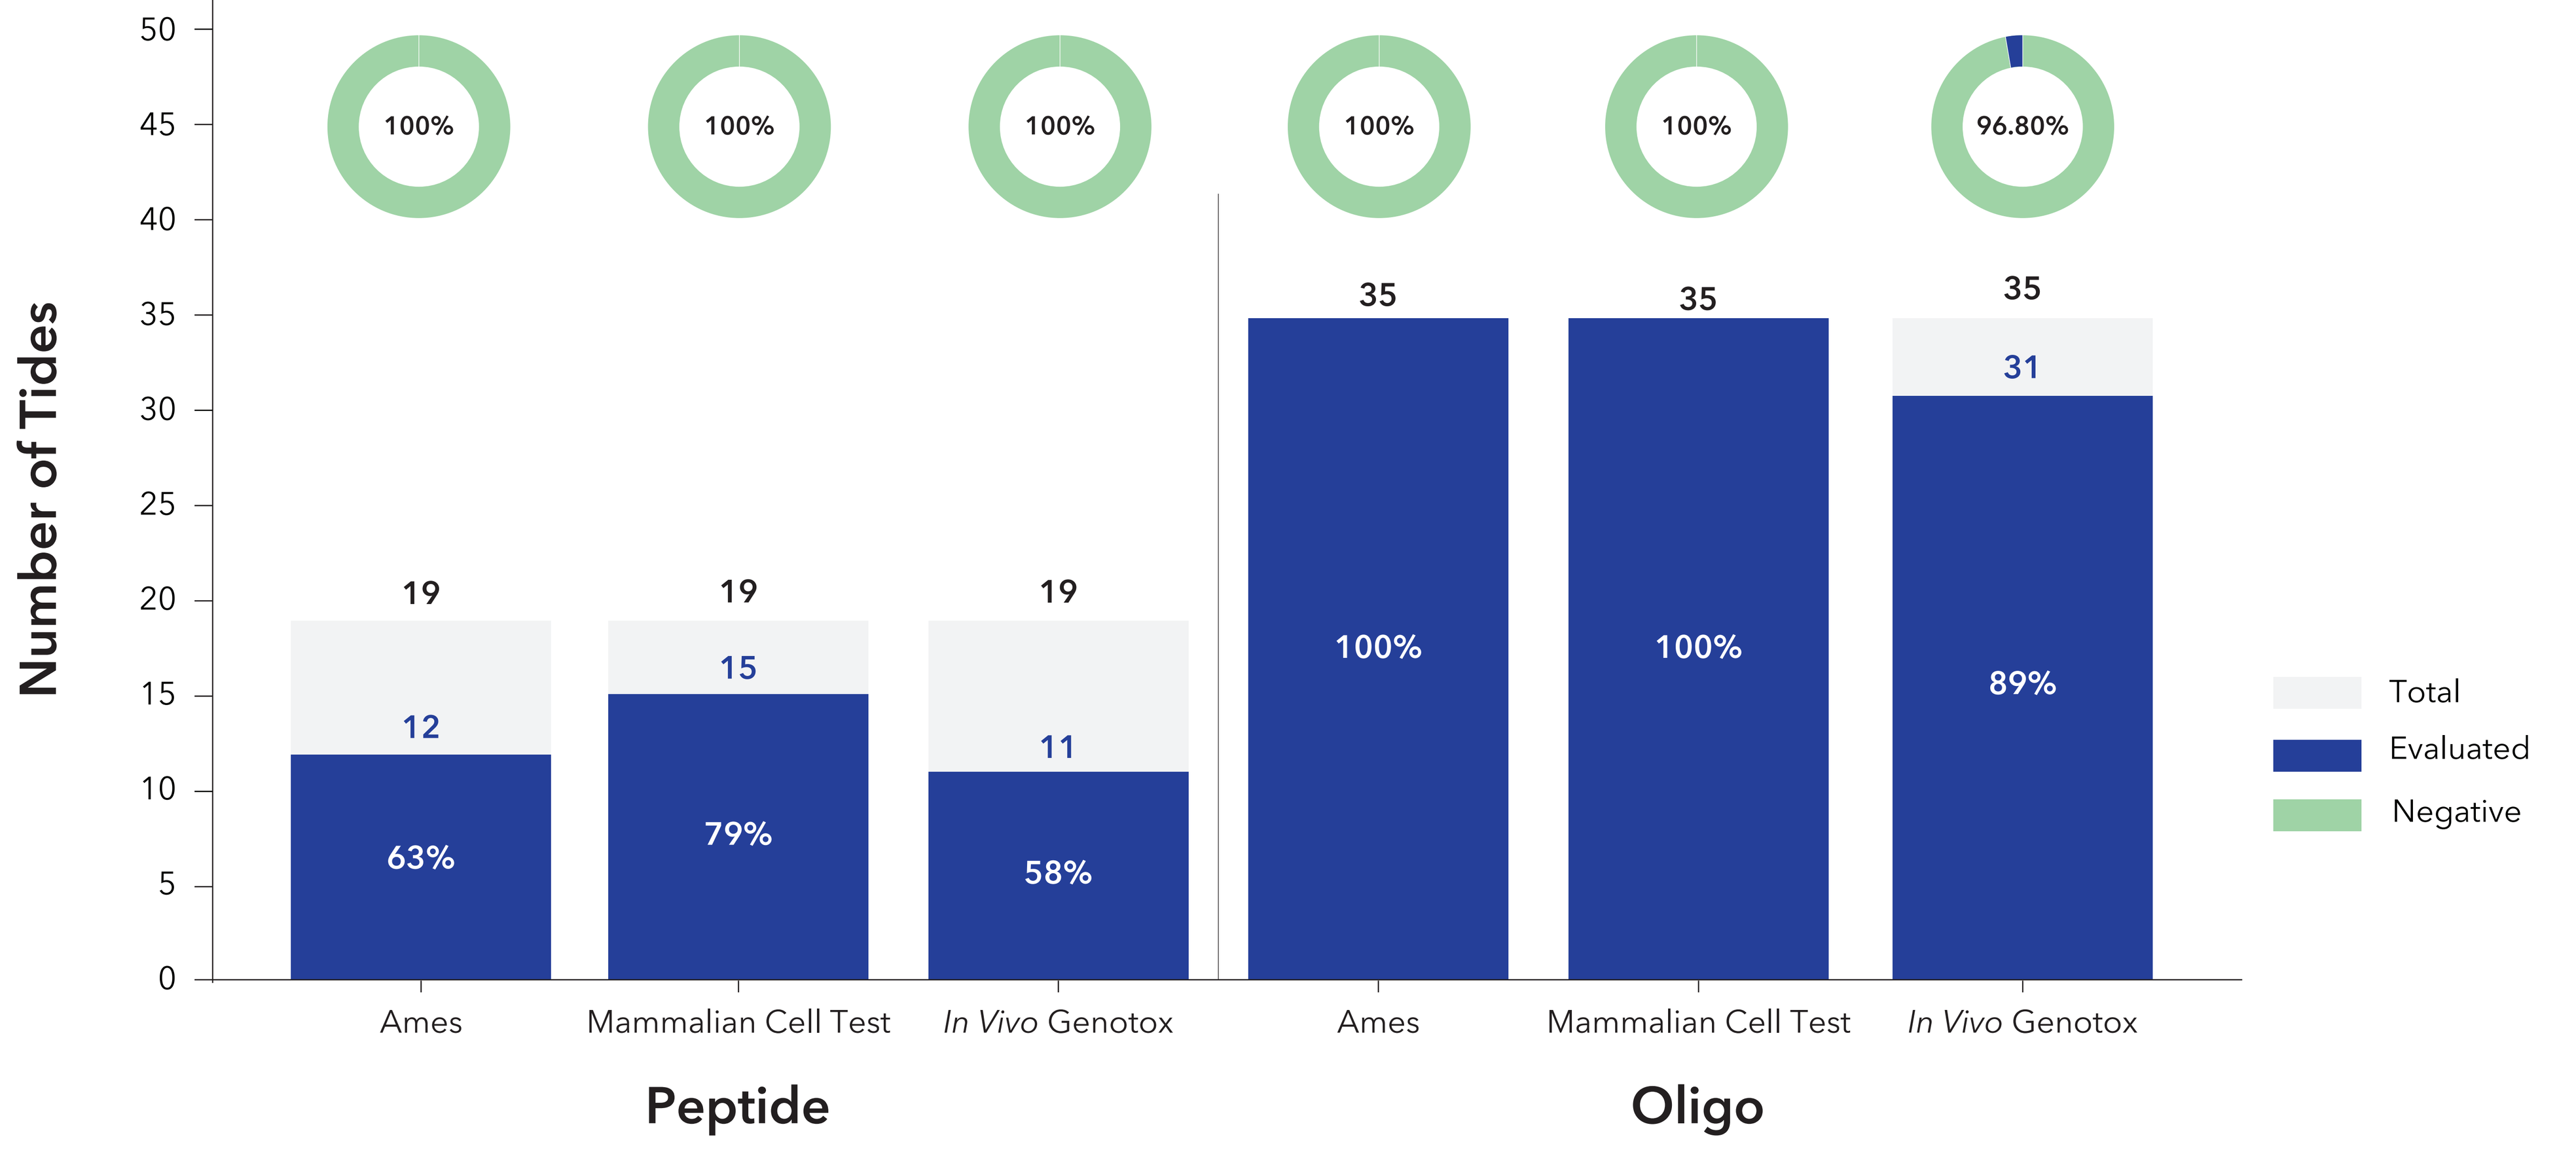
Number of Tides
0
5
10
15
20
25
30
35
40
45
50
100%
100%
100%
100%
100%
96.80%
19
19
19
35
35
35
12
15
11
31
63%
79%
58%
100%
100%
89%
Ames
Mammalian Cell Test
In Vivo Genotox
Ames
Mammalian Cell Test
In Vivo Genotox
Peptide
Oligo
Total
Evaluated
Negative

Conclusion

Consistent with reports in the literature, the Oligonucleotide Safety Working Group's genotoxicity testing subcommittee believes that therapeutic oligonucleotides are unlikely to be genotoxic and peptides are large with poor membrane permeability and are not expected to reach the cytoplasm or nucleus, thus they would not interact directly with DNA or the mitotic apparatus, and their degradation releases endogenous amino acids which should pose no genotoxic risk. In summary, most Tides have undergone genetic toxicology testing, and the results have generally been negative.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou) Co., Ltd.

SCAN FOR A COPY