A Retrospective Study of Nonclinical Safety Data for siRNAs

Zhang Yan, Guo Lei, Yongbin Zhang
WuXi AppTec (Suzhou) Co., Ltd., Suzhou, China

Abstract / Poster Number: 4711 / F502

Background and Purpose

Small interfering RNA (siRNA) therapeutics represent a promising class of drugs. This poster aims to summarize the toxicological findings from a retrospective analysis to identify common patterns and assess overall safety.

Methods

A retrospective analysis was conducted on a database of 46 GLP-compliant, repeated-dose toxicity studies. The database encompassed 23 different siRNA molecules in preclinical development. Details on therapeutic areas, experimental design (species, duration, route), and extensive data collections were analyzed. The data included toxicokinetic parameters, clinical and anatomic pathology, immunological profiles (cytokines, complement, immunophenotyping), and immunogenicity assessments.

Results

The studies covered therapeutic areas including metabolic (35%), liver-targeted (30%), cardiovascular (22%), autoimmune (9%), and renal diseases (4%). All the studies included one control group and 3 treated groups with an 8-week (91%) or 13-week (9%) recovery period. Test species were primarily nonhuman primates (NHPs, 48%) and rats (39%), with fewer studies in dogs (2%) and mice (11%).

Figure 1 Number of Studies by Targeted Diseases

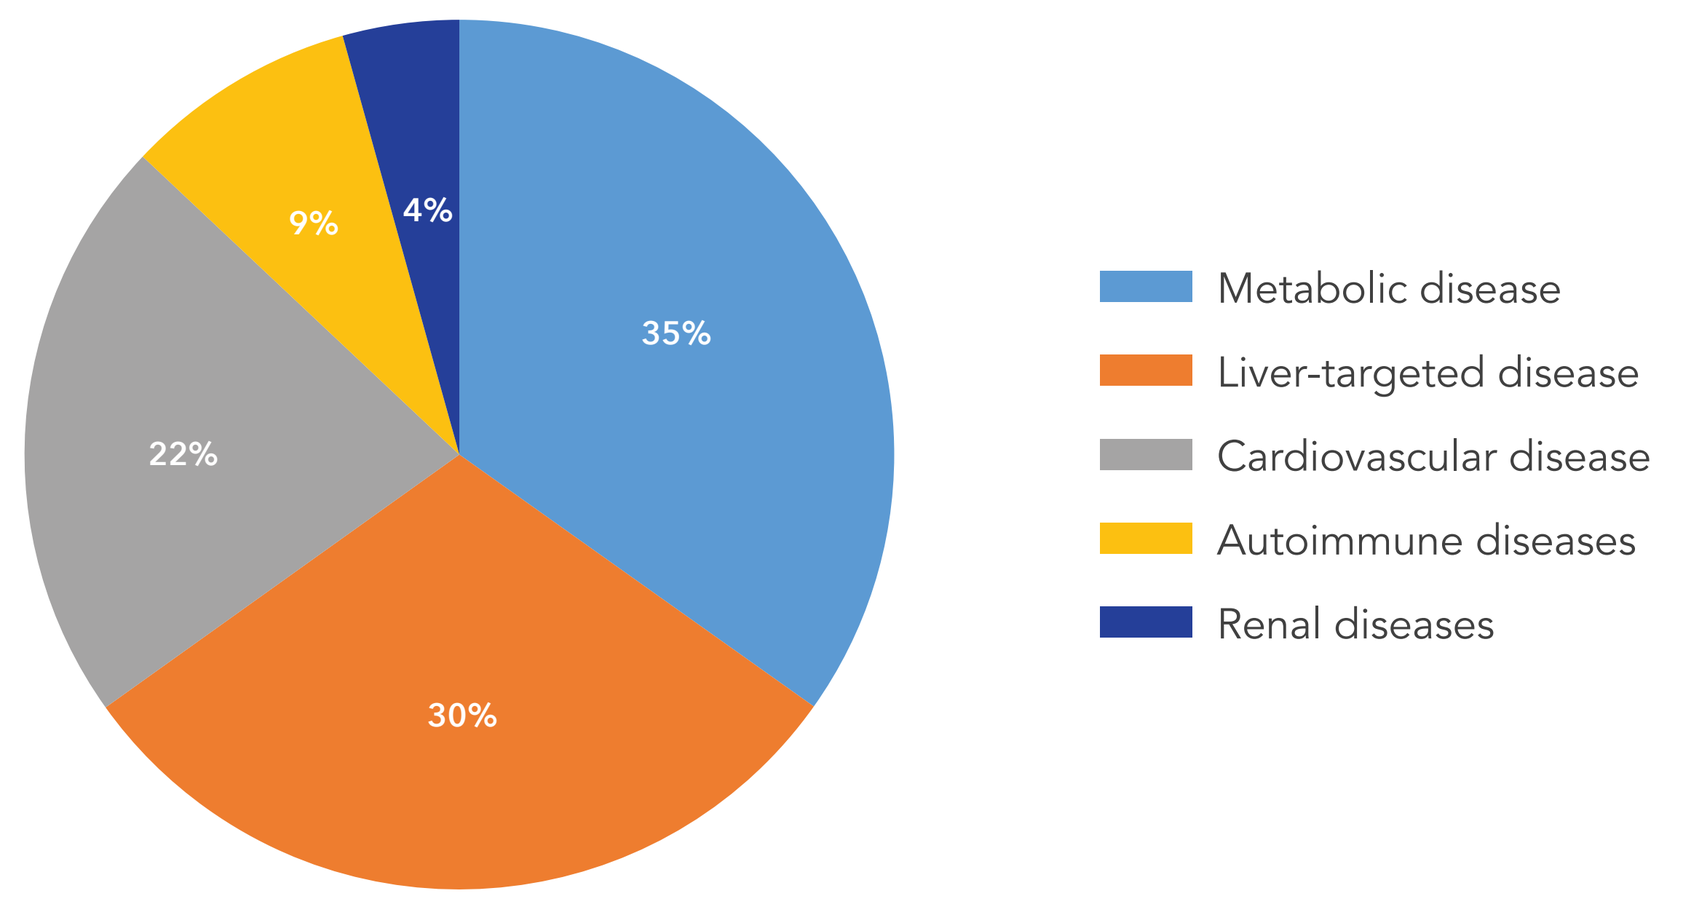
35%
30%
22%
9%
4%
Metabolic disease
Liver-targeted disease
Cardiovascular disease
Autoimmune diseases
Renal diseases

Most siRNAs utilized the GalNAc delivery system and were administered subcutaneously at doses of 10-600 mg/kg, generally with once weekly (26%) or once every two weeks (52%) dosing regimen. The time to maximum concentration (Tmax) was 0.5-2.0 hours in rodents and 1.0-8.0 hours in NHPs, with no observed accumulation. Higher drug exposure was noted in the liver and followed by the kidneys. Anti-drug antibodies (ADAs) were rarely detected and had no impact on systemic exposure.

Figure 2 Incidence of Dose Level

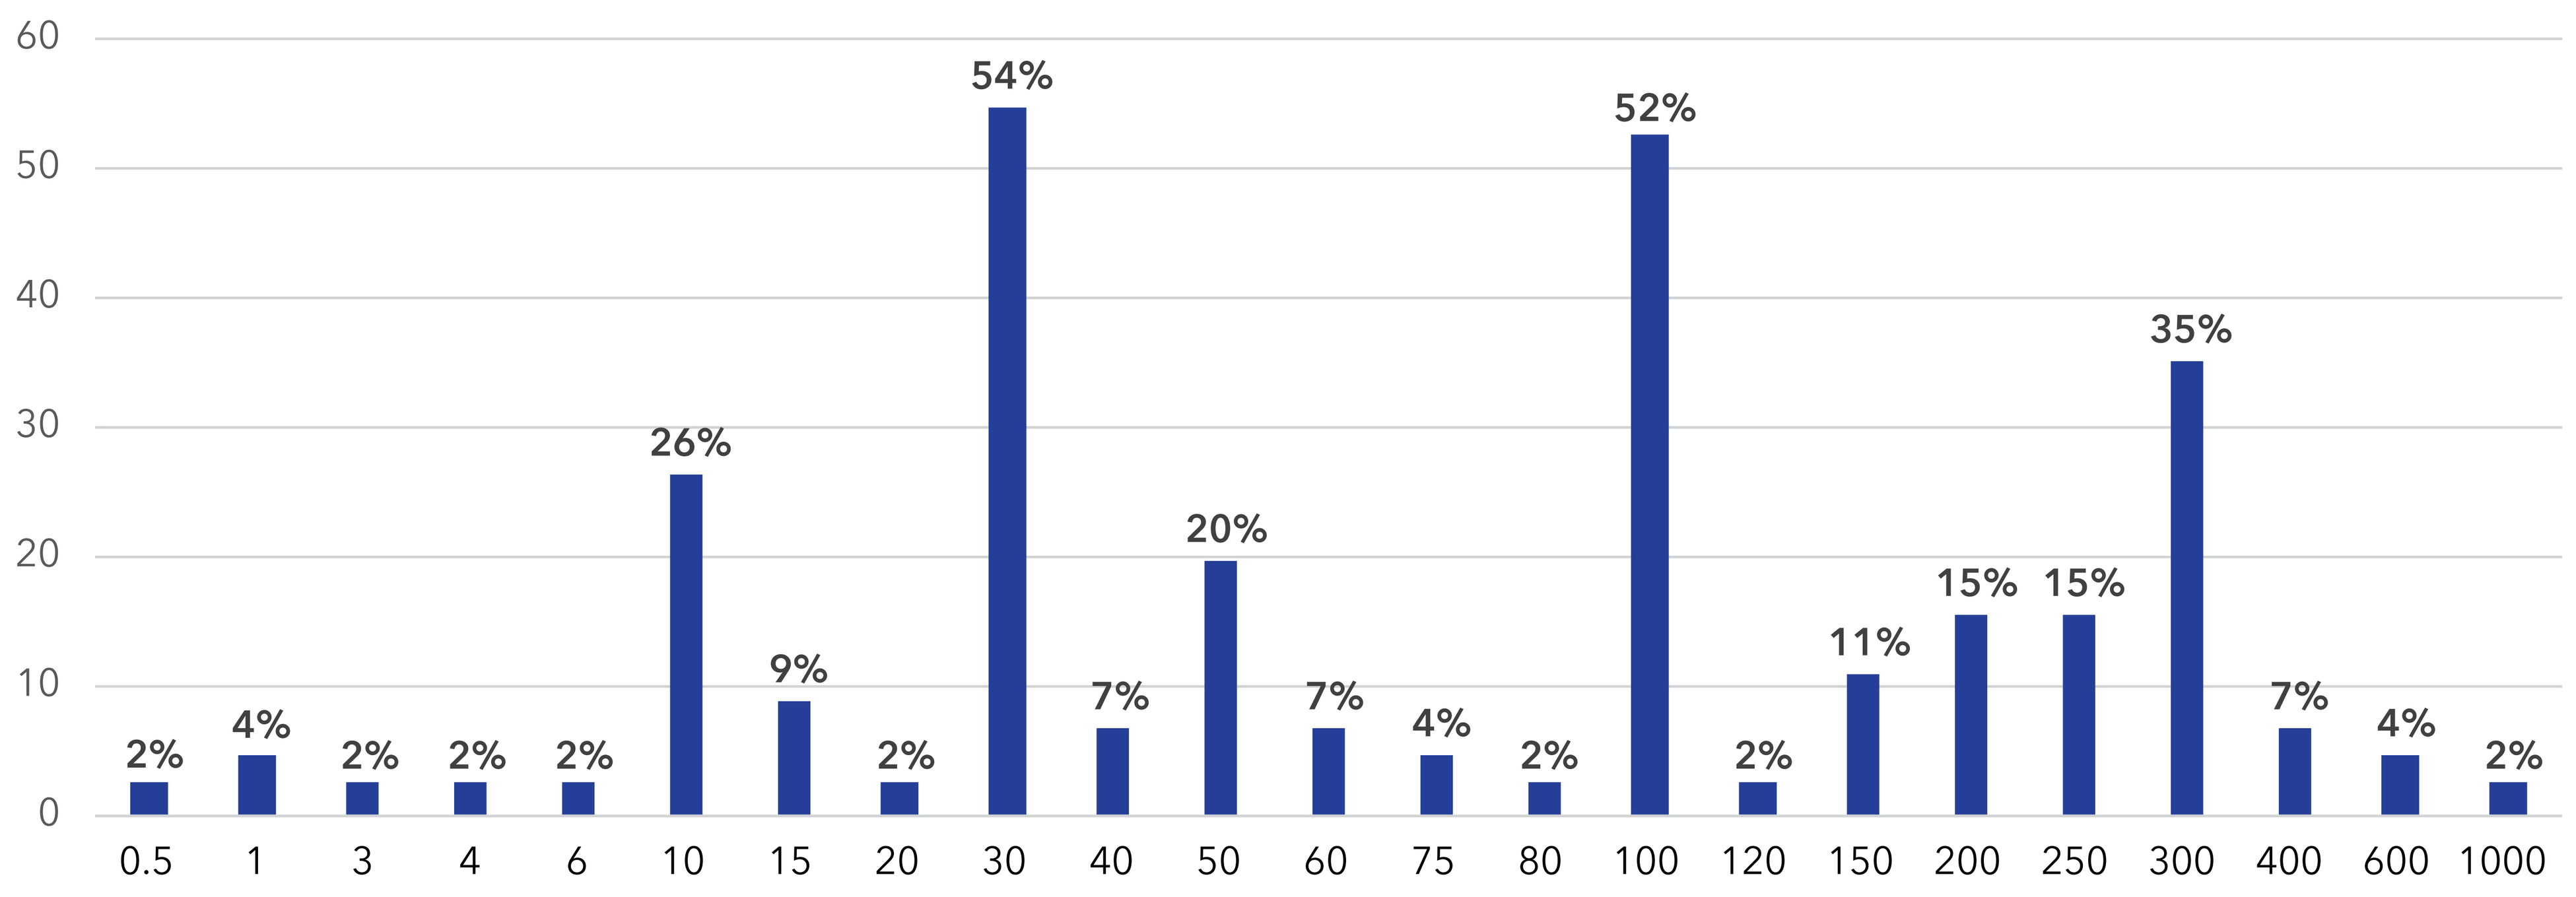
60
50
40
30
20
10
0
2% 4% 2% 2% 2% 26% 9% 2% 54% 7% 20% 7% 4% 2% 52% 2% 11% 15% 15% 35% 7% 4% 2%
0.5 1 3 4 6 10 15 20 30 40 50 60 75 80 100 120 150 200 250 300 400 600 1000

Changes were noted in hematology (22%), coagulation (28%), serum chemistry (59%, mainly were hepatic parameters), and pharmacological markers, while immunological effects were rare (13%). The primary target organs were the liver, kidneys, lymph nodes, and injection sites, with occasional involvement of the spleen, thymus, thyroid, and other organs. Overall, the observed effects were most frequently minimal to moderate in severity, and in approximately half of the studies, all these changes were either reversible or showed partial recovery.

Figure 3 Incidence of Target Organ

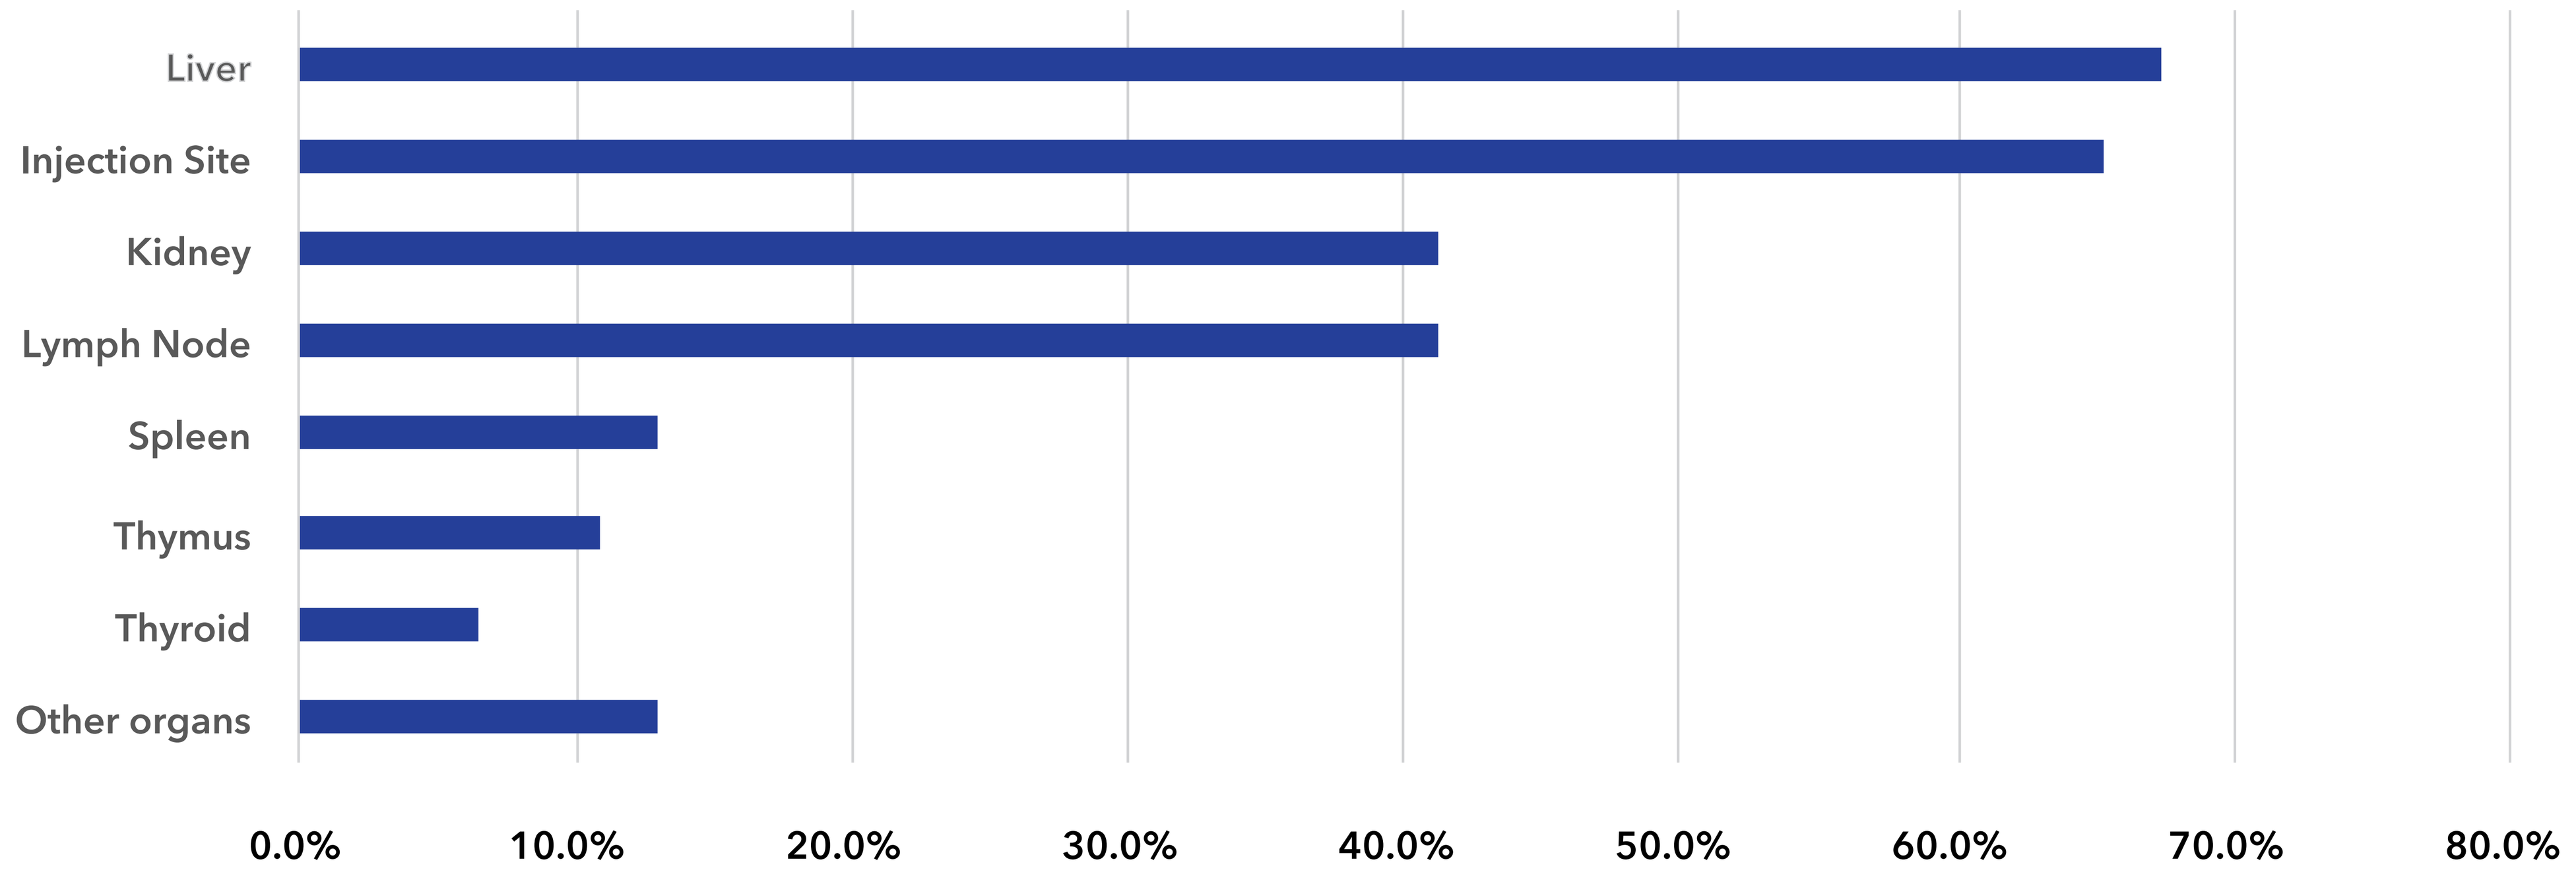
Liver
Injection Site
Kidney
Lymph Node
Spleen
Thymus
Thyroid
Other organs
0.0% 10.0% 20.0% 30.0% 40.0% 50.0% 60.0% 70.0% 80.0%

Conclusion

This retrospective analysis demonstrates a generally favorable and predictable safety profile for siRNA therapeutics in preclinical development. Toxicological findings were primarily confined to expected target organs, were typically mild to moderate, and often reversible. The establishment of a No Observed Adverse Effect Level (NOAEL) in 98% of the studies, coupled with rare and non-impactful immunogenicity, supports the continued development of siRNA drugs.

SCAN FOR A COPY