Normal, But Not Normal, Review of Some Vehicle-Related Histopathology Findings in the Concurrent Control Animals in the General Toxicity Studies

Fei Zhou[1], Yan Zhang[2], Xixing Zhao[2], Tiansheng Zhou[2], Long Jing[1], Lin Zhu[2], Meixi Zhang[2], Angelique Leone[2]
[1] Laboratory Testing Division, WuXi AppTec (Nantong) Co., Ltd, Safety Evaluation Center, 2588, Qiantang River Road, Qidong Economic Development Zone, Nantong 226200, Jiangsu, China
[2] Laboratory Testing Division, WuXi AppTec (Suzhou) Co., Ltd, 1318 Wuzhong Avenue, Wuzhong District, Suzhou 215104, Jiangsu, China

WuXi AppTec

Abstract / Poster Number: 3759 / B192

Background and Purpose

Concurrent control groups are essential in general toxicity studies and are typically composed of animals administered the vehicle. The vehicle is usually the same solvent used to dissolve the test article, such as saline, which is the most commonly used vehicle. However, some solvents, including surfactants or excipients, may be employed to meet the solubility and absorption requirements of pharmaceutical formulations. This report describes toxicity findings from three vehicles used in general toxicity studies, highlighting the need for toxicologic pathologists and toxicologists to recognize that the concurrent control group may not always represent a completely normal baseline.

Methods

Histopathology data for vehicles, dimethylacetamide (DMAC), β-cyclodextrin, and caprylocaproyl macrogol-8 glyceride (Labrasol), used in the control groups from preclinical general toxicity studies, were reviewed at our facility.

Results

DMAC
Intravenous administration resulted in observed toxicities in the liver of SD rats and Beagle dogs, as well as in the testes and eyes of SD rats. Specifically, centrilobular mixed cell infiltration and occasional single-cell necrosis of hepatocytes were noted in the liver. In the testes, seminiferous tubular degeneration and atrophy were observed. Additionally, in the eyes, single-cell necrosis of the corneal epithelium accompanied by fibroplasia in the corneal stroma was observed.

β-cyclodextrin
For oral gavage studies, hepatocellular necrosis in ICR mice and vacuolation of the tubular epithelial cells in the kidneys of the SD rats were observed.

Labrasol
Orally, toxicities were observed in the lungs from a rat study, and the histopathology changes consisted of bronchiolo-alveolar mixed cell infiltration and obliterative bronchiolitis with foreign material. The foreign material was lightly basophilic, fibrillar, and predominantly found in the terminal bronchioles, although it was occasionally observed in the alveoli and bronchi. Mortalities were also noted in this case.

DMAC

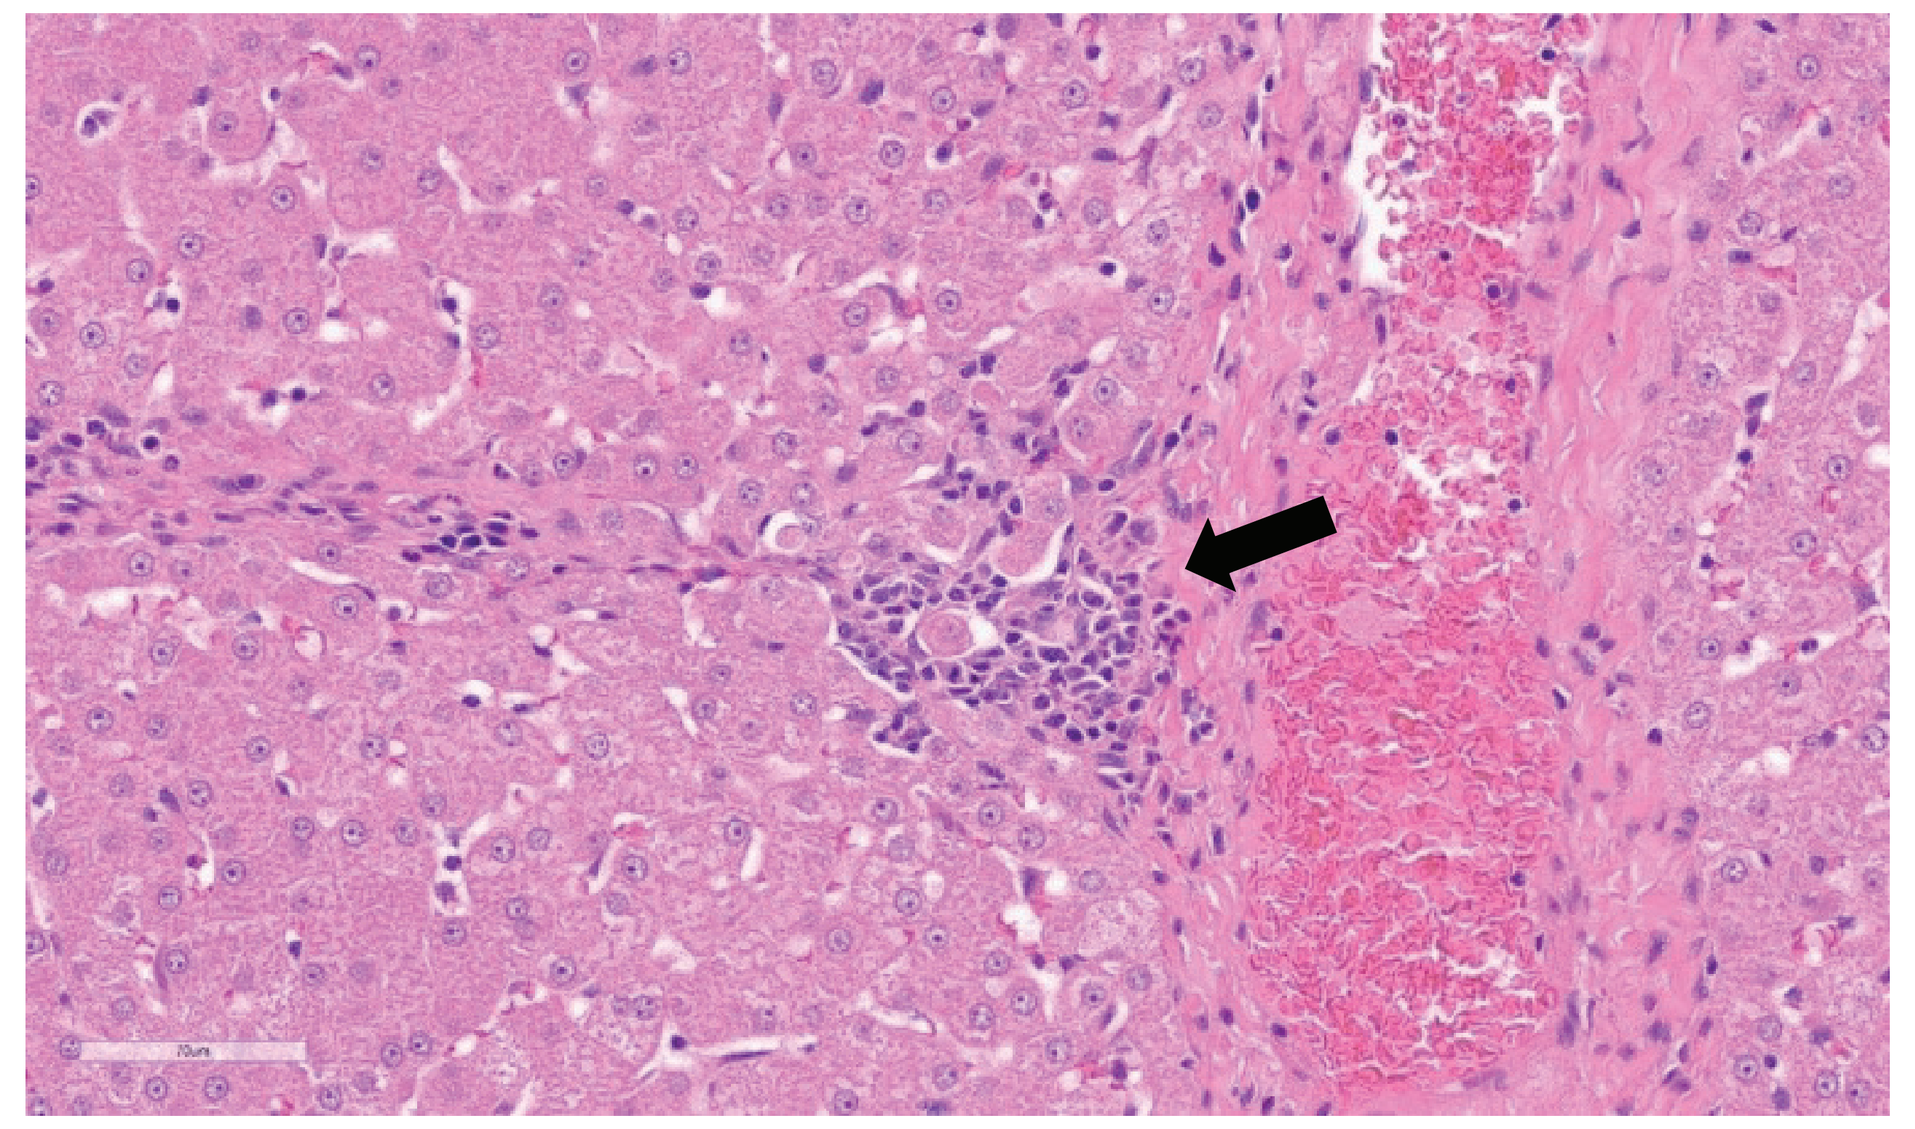

Liver, Centrilobular mixed cell infiltration (arrow)

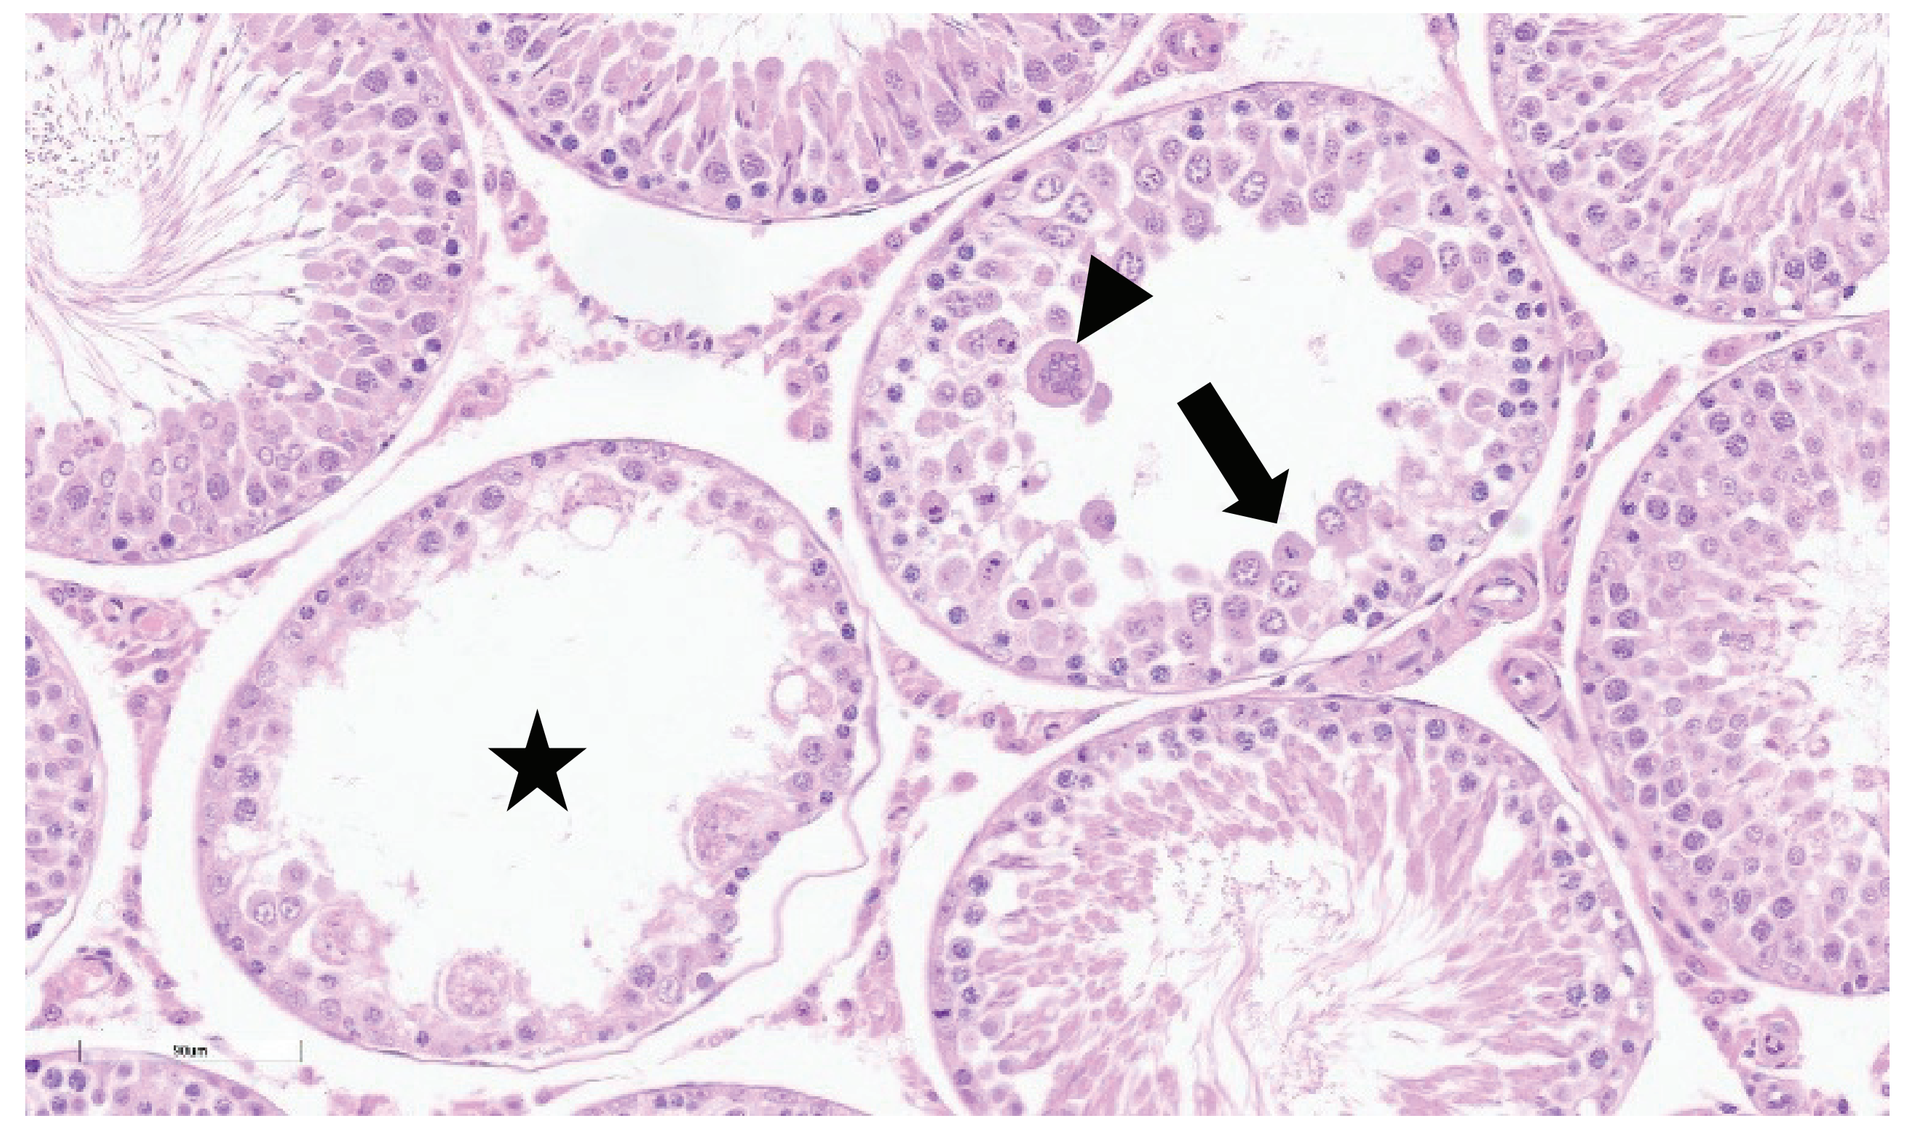

Testes: Atrophic tubule (star), germ cell degeneration (arrow), multinucleated germ cells (arrowhead)

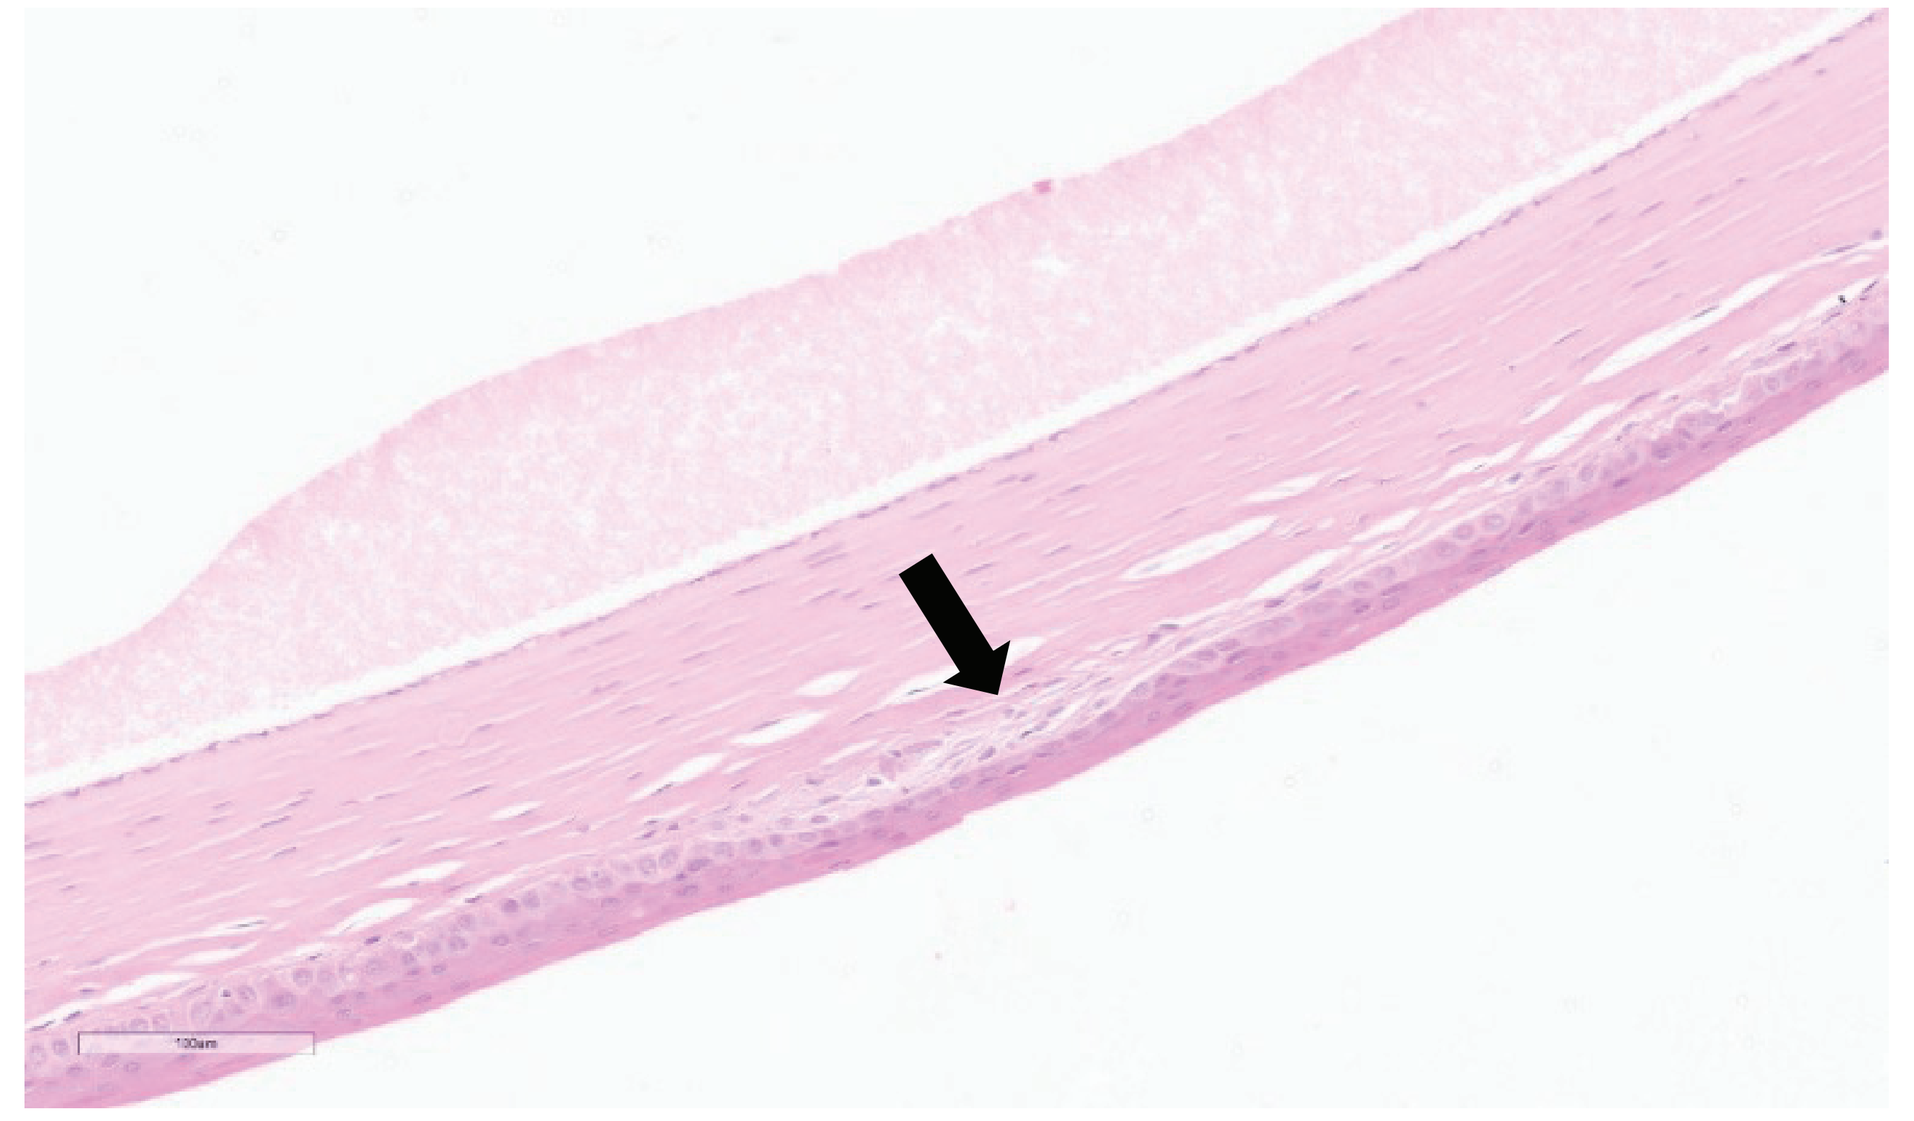

Cornea, Fibroplasia in corneal stroma (arrow)

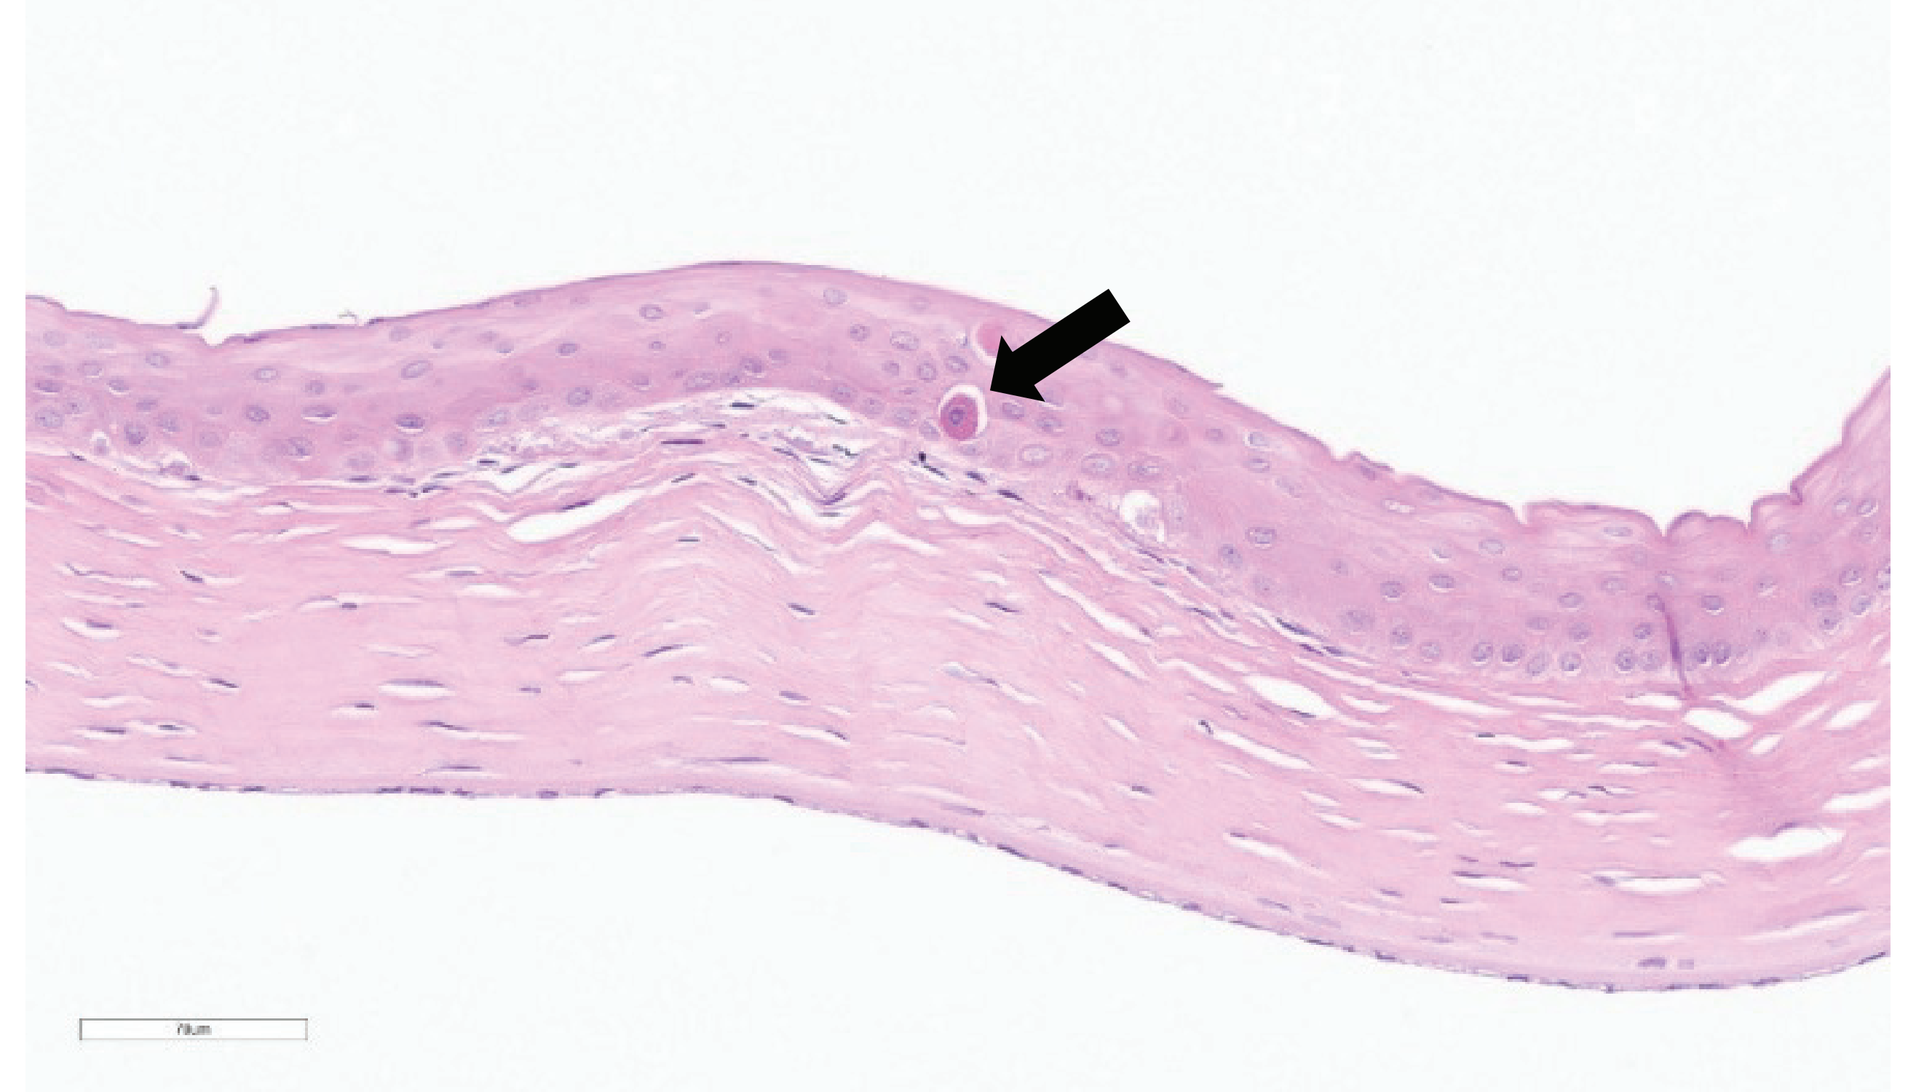

Cornea: Single-cell necrosis of corneal epithelium (arrow)

β-cyclodextrin

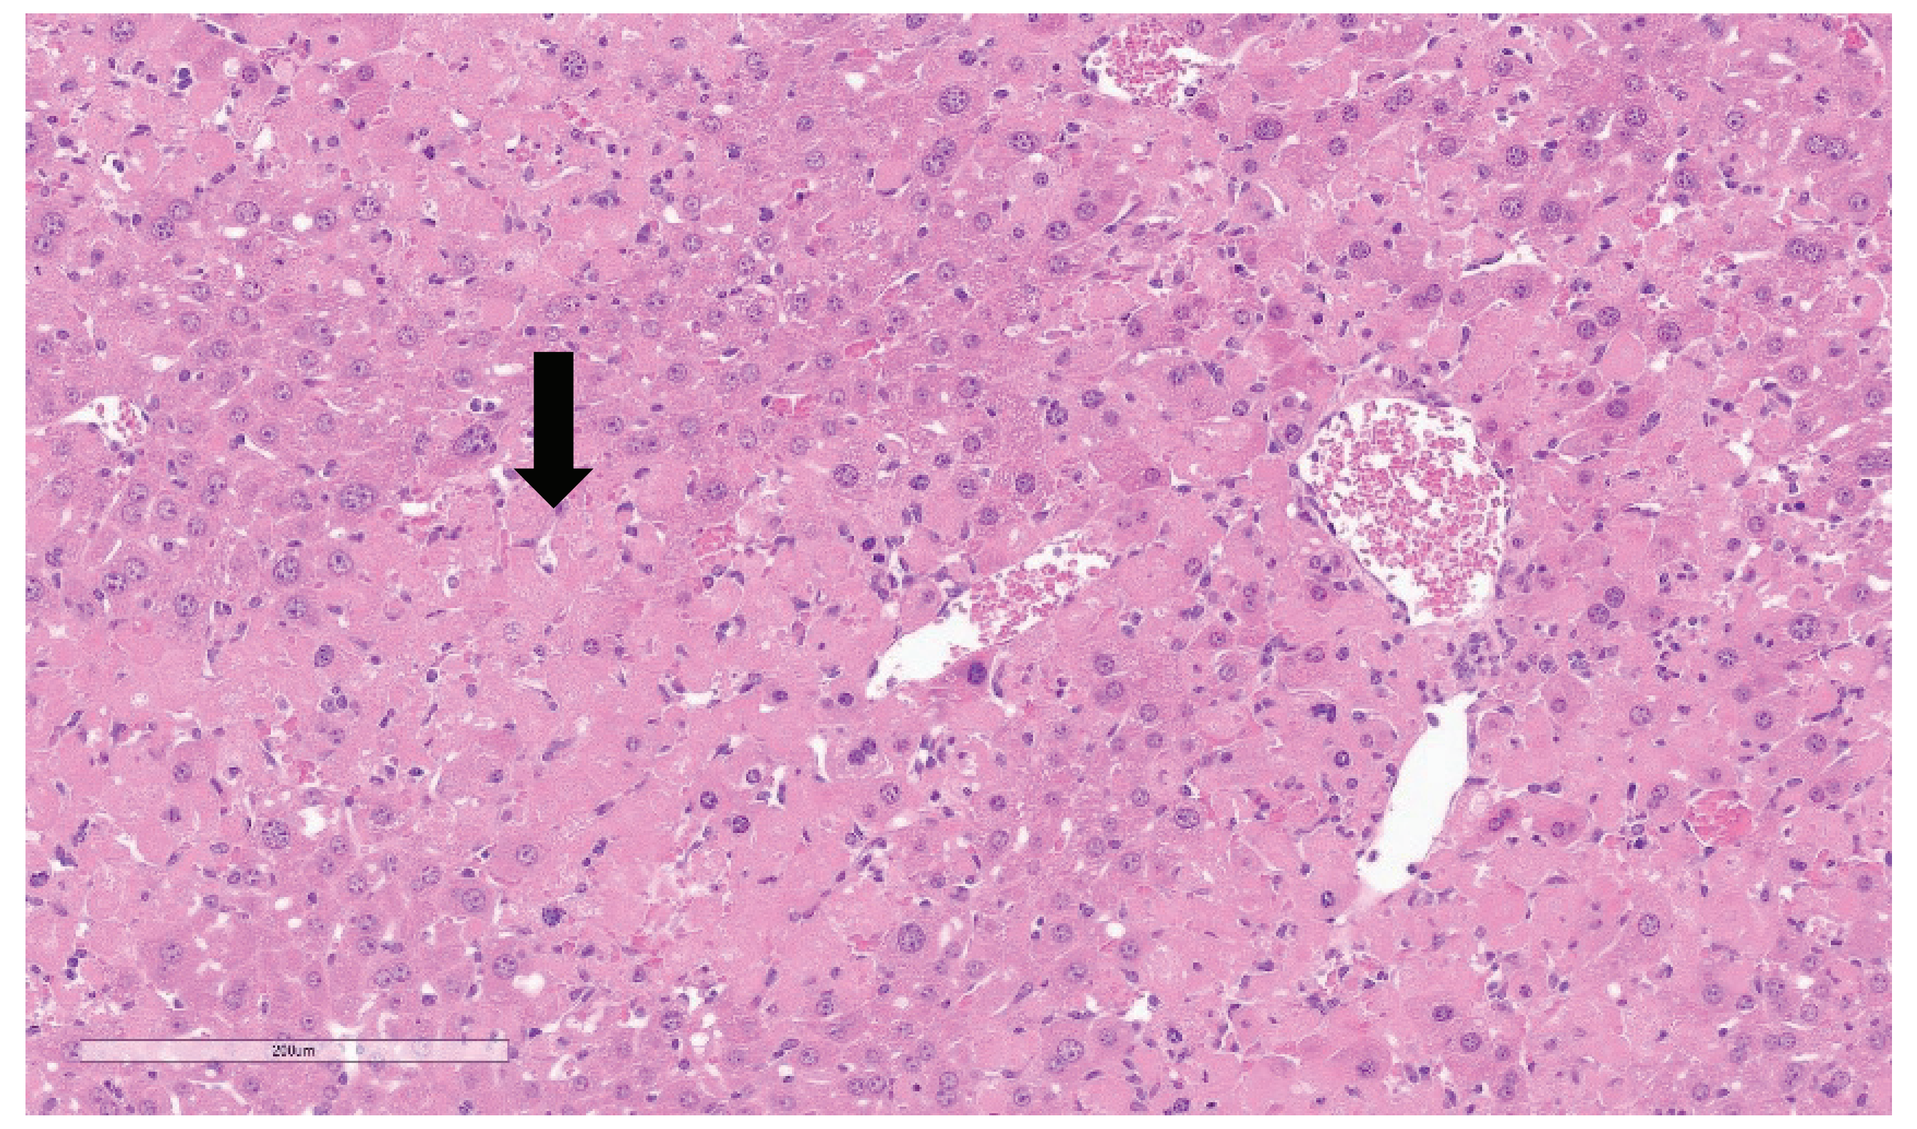

Liver, Coagulative necrosis of hepatocytes (arrow)

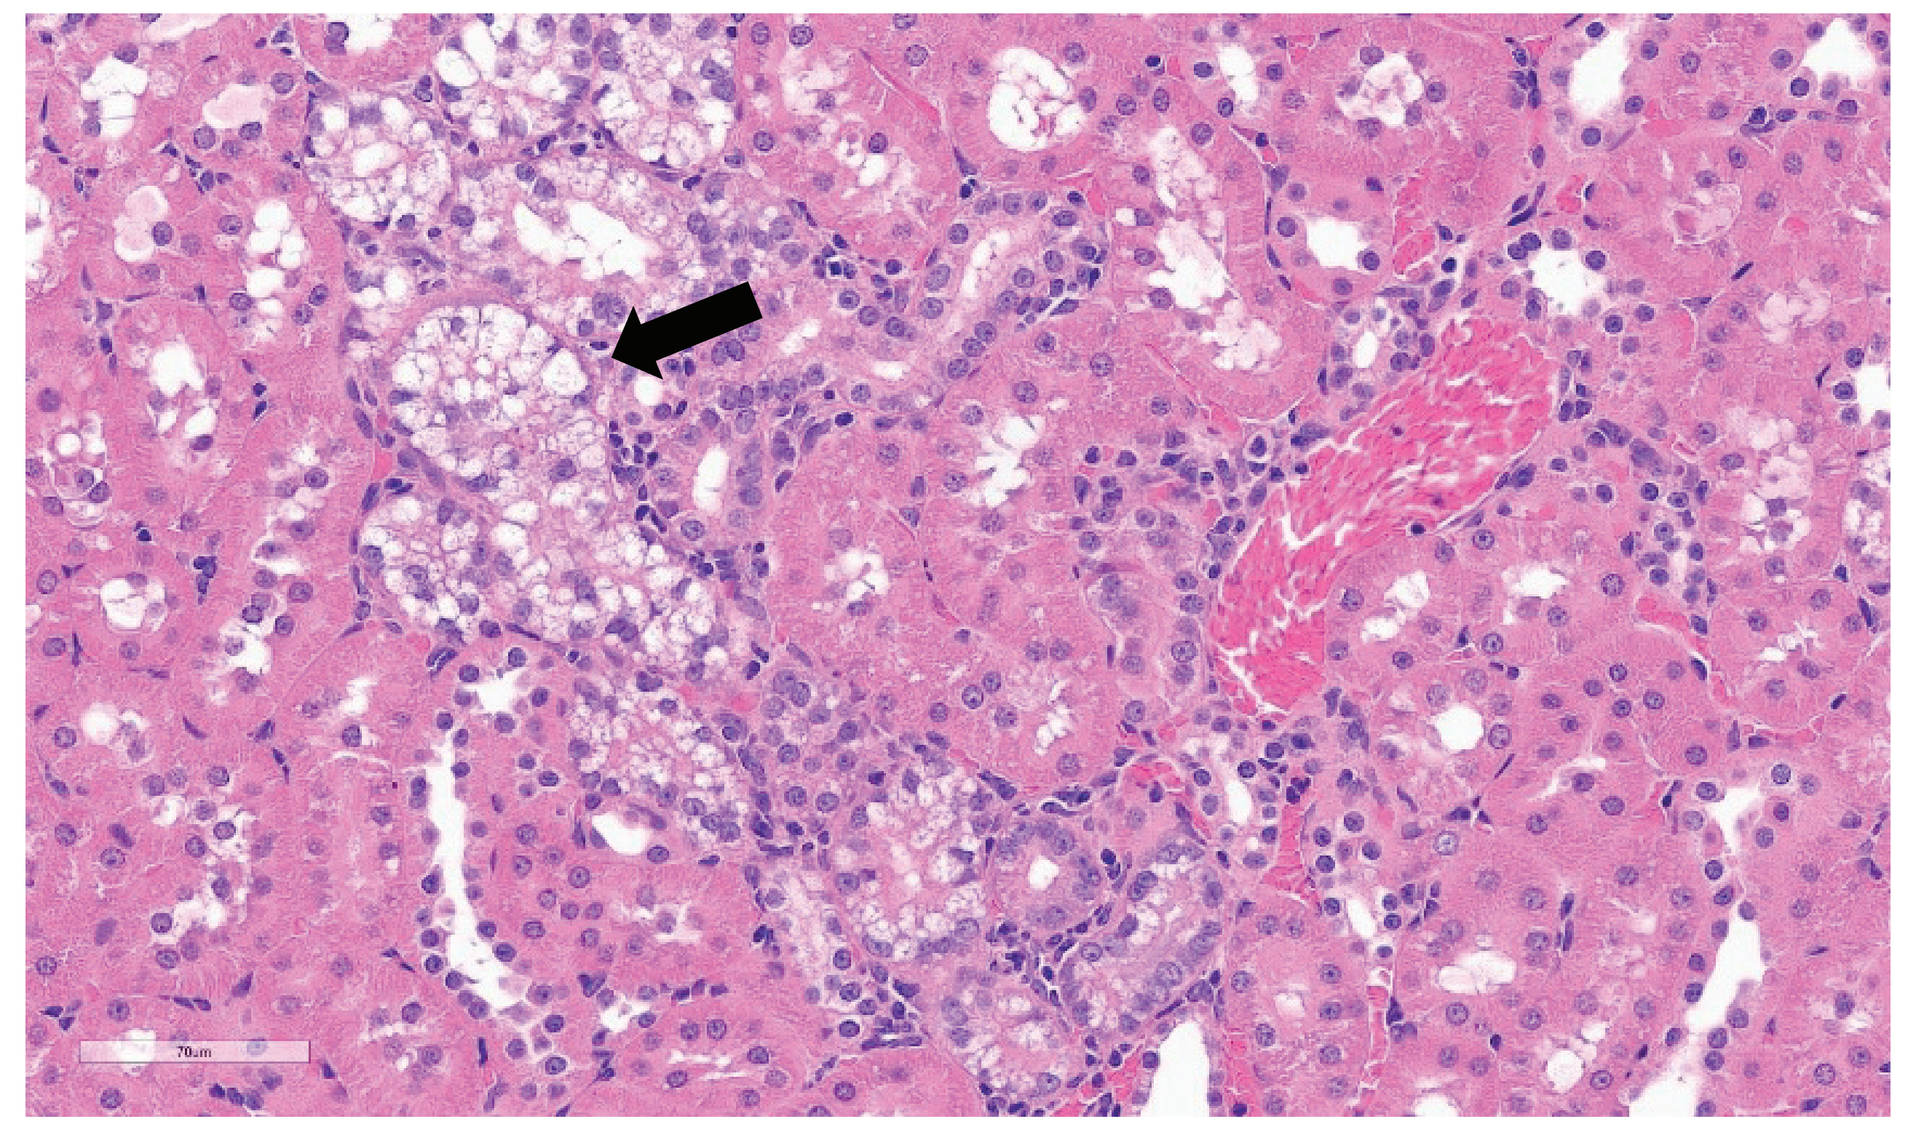

Kidneys, Tubular epithelium vacuolation (arrow)

Discussion

DMAC
Dimethylacetamide is widely used for its superior solvent properties. It has been reported that DMAC poses low to moderate hepatotoxicity risks to the occupational health of employees. Hepatic toxicity has been explored using human normal hepatocytes (LO-2), where apoptosis of the hepatocytes was attributed to increased ROS generation and activation of the Bcl-2 pathway. Regarding testicular toxicity, it has been reported that DMAC acts in a post-meiotic manner to inhibit spermatogenesis, resulting in a decreased number of elongated spermatids. Additionally, DMAC distorts the mitochondrial and microtubular structure of sperm, which may lead to infertility in rats. Finally, the OECD SIDS has reported that this vehicle causes mild and reversible irritation to the eyes (cornea).

Cyclodextrins
Cyclodextrins (CDs) serve as effective formulation vehicles, enhancing the solubility of drugs in aqueous environments and thereby improving the delivery of various medicinal agents to biological systems. Regarding the renal alterations observed in this study, it has been documented that HP-β-CD forms stable complexes with cholesterol in the bloodstream. The absence of free cholesterol in the urine leads to the dissociation of these complexes in the kidney, resulting in the accumulation of cholesterol and cholesterol esters in the urinary bladder and renal pelvis. This accumulation is responsible for the observed renal alterations. Additionally, another study has indicated that the vacuoles observed represent giant lysosomes formed by cyclodextrin microcrystals embedded within the lysosomal matrix. In contrast, there are no published references concerning the liver changes observed in ICR mice, indicating that further investigation is warranted.

Labrasol
Labrasol is frequently used in the course of the early toxicological characterization of new candidate drugs to enhance the intestinal absorption of poorly soluble compounds. The toxicities reported in the publication included pneumonia with foamy contents in the airways, emphysematous lungs, gas and/or distension of the gastrointestinal tract, and hyperemia of the gastric mucosa. Furthermore, death could be caused by this vehicle. The findings in our case are largely consistent with those early reports, including observed mortalities. In our case, death was attributed to changes in the lungs. The presence of foreign material in the lungs suggests that these changes were most likely due to reflux followed by aspiration pneumonia, resulting from foam formation by the vehicle. Additionally, Labrasol is inherently cytotoxic, which may have further contributed to the lung changes when aspirated.

Conclusion

Certain vehicles, such as DMAC, β-cyclodextrin, and Labrasol, may exhibit toxicities. It is crucial for study pathologists and toxicologists to closely examine the histopathological findings in the concurrent control groups when these vehicles are used in studies. They must differentiate these findings from those related to the test articles.

References

1. Xin Liu, Wei Gong, Yan Qiong Xu, Bao Li Zhu. 2016. Dimethylacetamide-Induced Hepatic Injury in Vitro: Mechanism and Potential Preventive Strategy. Biomed Environ Sci. 29(2):153-157.
2. Nupur Khera, Chafik Ghayor, et al. 2020. N, N-Dimethylacetamide, an FDA Approved Excipient, Acts Post-Meiotically to Impair Spermatogenesis and Cause Infertility in Rats. Chemosphere. 256:1-9.
3. Y. Saavedra, J. Benito, F. Cabello, et al. 2010. Toxic Effects Associated to The Use of the Delivery Enhancer Caprylocaproyl Macrogol-8 Glyceride (Labrasol) In Early Toxicological Studies In Rats. An Activity of the Melius Project. Abstracts/Toxicology Letters. S37–S351.
4. Sarah Gould, Robert C Scott. 2005. 2-Hydroxypropyl-Beta-Cyclodextrin (HP-Beta-CD): A Toxicology Review. Food Chem Toxicol. 43(10):1451-1459.

SCAN FOR A COPY