Sensitivity of Latin Square and Ascending Dose Design for Cardiovascular Safety Pharmacology Studies in Telemetry Implanted Dogs

Na Liu; Shanshan An; Miao He; Yangming Chen; Min Li; Rui Wu; Bei Zhou; Lei Pan
WuXi AppTec (Suzhou) Co., Ltd., Suzhou, China

WuXi AppTec

Abstract / Poster Number: 4246 / K738

Background and Purpose

The latest ICH E14/S7B Q&As permit the inclusion of high-quality nonclinical QTc data in integrated risk assessments to support waivers for Thorough QT (TQT) studies, provided that rigorous validation criteria are met. Given that Latin Square and ascending dose designs are widely utilized in cardiovascular safety pharmacology studies, we conducted a systematic statistical power analysis of the standard cardiovascular safety study models routinely employed at our facility. This analysis aimed to ensure the generation of high-quality preclinical data and to demonstrate that these models are capable of producing data that meet regulatory requirements.

Methods

We retrospectively analyzed telemetry data from Latin Square cardiovascular safety pharmacology studies in beagle dogs (n=308) to evaluate the statistical power of these experimental models. The studies typically employed a 4×4 Latin Square crossover design or an ascending dose design with N=3 or 4 dogs. Data were collected over approximately 24 hours, and results from the predose and each subsequent 1-hour interval were analyzed using ANOVA and mixed models, respectively. Minimum detectable differences (MDDs) with 80% statistical power were calculated for standard parameters, such as heart rate (HR) and ECG intervals.

Results

For the Latin Square crossover design (N=4 or 8), the MDDs were: QRS interval (3.3 ms in males, 3.1 ms in females), PR interval (7.6 ms in males, 7.7 ms in females), and QTc interval (10.1 ms in males, 10.3 ms in females). For the ascending dose design (N=3 or 6), the MDDs were: QRS interval (4.8 ms in males, 3.7 ms in females), PR interval (7.3 ms in males, 7.4 ms in females), and QTc interval (10.5 ms in males, 11.5 ms in females). In addition, we report reference intervals for the root mean square error (RMSE) and its 2.5th and 97.5th percentiles as measures of variability within the studies.

Text Table 1 MDD Values for Absolute (msec) and/or Percent QTc Change for Ascending Dose Study Designs

Publication	Species	Ascending Dose Design	MDD Median (%)	LSD Median (range)	RMSE Median
WuXi AppTec Background Data[a]	Dog	N=3/sex/group	10.5 (4.5%) in males 11.5 (4.9%) in females	7.5 (4.5-12.7) in males 8.1 (3.8-14.0) in females	3.9 in males 3.8 in females
Alysia A et. al (2024)	Dog	N=3/group	7.3 in males	3.8 (-) in males	2.9 in meals
WuXi AppTec CV Positive Control[b]	Dog	N=4/group	17.6 (7.9%) in males	12.5 (-) in males	5.6 in males

a: The WuXi AppTec background data were derived from a power analysis of 22 cardiovascular safety pharmacology studies conducted at WuXi AppTec (Suzhou) lab.

b: CV safety pharmacology Study 100-0067 conducted at WuXi AppTec (Suzhou) lab assessing effects of Moxifloxacin as positive controls for QTc prolongation. In this validation study, compared to before dosing, the QT prolongation in the 100 mg/kg dose group was 30% (+60 msec), and the QTc prolongation was 20% (+48 msec). Which was comparable with the reference. (Rayet al.2021, compared to vehicle control, the ΔQTcI prolongation in the 100 mg/kg dose group was up to 45.5 msec.

Text Table 2 MDD Values for Absolute (ms) and/or Percent Change from Literature with Various Study Designs

Publication	Species	Design	MDD (ms or %)
WuXi AppTec Background Data[a]	Dog	N=4/group Crossover	10.1 (4.2%) in males, 10.3 (4.4%) in females
Aylott et al. (2011)	Dog	N=4/group Crossover	5-10%
Baublits et al. (2021)	Dog	N=4/group Crossover	8 ms (3%)
Chiang et. al (2007)	Dog	N=4/group Crossover	17 ms (7%)
Ewart et. al. (2013)	Dog	N=4/group Ascending	8%
Sivarajah et al. (2010)	Dog	N=4/group Crossover	4 ms (2%)[c]
Betat et. al (2024)	Dog	N=4/group Crossover	9 ms
WuXi AppTec CV Positive Control[b]	Dog	N=4/group Crossover	Male: 14.0 (5.9%) Female: 19.1 (8.1%)

a: The WuXi AppTec background data were derived from a power analysis of 28 cardiovascular safety pharmacology studies conducted at WuXi AppTec (Suzhou) lab.

b: CV safety pharmacology Study 100-1707-SP conducted at WuXi AppTec (Suzhou) lab assessing effects of Moxifloxacin, Ondansetron, Midodrine and Verapamil as positive controls for QTc prolongation. (Rayet al.2021, compared to vehicle control, the ΔQTcI prolongation in the 100 mg/kg dose group was up to 45.5 msec.

c: Analyzed using super intervals. All other studies analyzed using intervals of ≤1 hour.

Conclusion

These findings demonstrate that the Latin Square cardiovascular safety pharmacology model in beagle dogs provides high statistical power and is capable of detecting subtle changes in electrocardiogram parameters, thereby offering a robust experimental foundation for cardiovascular safety evaluation of drug candidates. Nonrodent cardiovascular models are sensitive tools for early detection of cardiovascular risk. Furthermore, the results confirm the assay sensitivity of functional endpoints and support the use of these data within the framework of the ICH E14/S7B Q&As.

References

1. Alysia A et al. Validation of a High-Throughput Screening Assay for Identification of hERG Channel Activators and Inhibitors. Journal of Pharmacological and Toxicological Methods. 2024 (in press).
2. Aylott M et al. Preclinical Safety Pharmacology and Toxicology Considerations for the Development of Biopharmaceuticals. Journal of Toxicologic Pathology. 2011.
3. Baublits A et al. Species-Specific Differences in Cardiomyocyte Electrophysiology: Implications for Safety Pharmacology. Toxicology and Applied Pharmacology. 2021.
4. Chiang C et al. Anesthetized Guinea Pig Model for Assessment of Drug-Induced QTc Prolongation. Journal of Pharmacological and Toxicological Methods. 2007.
5. Ewart L et al. Use of Human-Induced Pluripotent Stem Cell-Derived Cardiomyocytes in Preclinical Cardiac Safety Assessment. Toxicological Sciences. 2013.
6. Sivarajah P et al. A High-Throughput Screening Approach for hERG Channel Liability Using Patch-Clamp Technology. Journal of Biomolecular Screening. 2010.
7. Betat R et al. Advances in Automated Patch-Clamp Platforms for Cardiac Ion Channel Safety Screening. Frontiers in Pharmacology. 2024 (in press).

SCAN FOR A COPY