Non-Drug Induced Spontaneous Convulsions in Different Nonclinical Laboratory Animals Used for Non-Clinical Drug Safety Evaluation

Zhimei Wang
WuXi AppTec (Suzhou) Co., Ltd., Suzhou, China

WuXi AppTec

Abstract / Poster Number: 4702/ F493

Background and Purpose

Convulsions are occasionally observed during long-term toxicity and carcinogenicity studies in laboratory animals, but distinguishing whether these events are related to the test article or are spontaneous/procedural is crucial for accurate data interpretation. This review aimed to summarize the incidence and characteristics of convulsions in various commonly used laboratory animal species at the Suzhou testing facility, and to provide a reference for toxicologists and pathologists in differentiating test article-related effects from procedural or spontaneous findings.

Method

A retrospective review was conducted of studies performed at the Suzhou testing facility between 2018 and 2025. The analyzed studies included 7-day to 2-year carcinogenicity and toxicity studies on Sprague-Dawley (SD) rats, CD-1 mice, Tg.rasH2 mice, Beagle dogs, non-human primates (NHPs), Bama minipigs, Hartley guinea pigs, and New Zealand white rabbits. The occurrence, timing, and circumstances of convulsions were recorded and analyzed, with attention to study duration, sex, procedures (e.g., blood collection, dosing), and histopathological findings.

Results

The results indicated that convulsions were most frequently observed in rats, particularly in the 2-year studies, with a highest incidence of spontaneous convulsions at 5.9% (males) and 4.41% (females). The average percentages of convulsions in the 26-week were up to 1.88% (males) and 4.27% (females), respectively. Additionally, in the 26-week study, convulsions with the highest incidence reaching 13% or 14% were observed from 3 studies. In these studies, the dosing frequency was greater than or equal to twice per day or using nasal dosing. The average percentages of convulsions in the 13-week were up 1.02% (males) and 5.10% (females), respectively, with maximum percentages up to 5.26% (males) and 8.70% (females), respectively. In the 4-week studies, symptoms were noted in only one male rat. In CD-1 mice and Tg.rasH2 mice during the 26-week studies, convulsions occurred mainly during blood collection or formulation administration procedures, suggesting procedural-related. In NHPs, convulsions were noted once each in a 2-month and a 6-month Intravenous Infusion study, while no occurrences were observed in Beagle dogs, guinea pigs, rabbits, or minipigs. Convulsions first appeared at 20 weeks in males and 14 weeks in females, with an average onset age of approximately 51 weeks across all animals during 2-Year Carcinogenicity study. Some animals had convulsions only once, while others experienced multiple episodes. No deaths were related to convulsions, and no related histopathological abnormalities were found in the brain upon examination.

Conclusion

SD rats exhibited a higher frequency of convulsions compared to other species, especially in longer-duration studies. The convulsions observed in mice were mostly procedural, and other species showed minimal or no occurrences. No convulsion-related deaths or brain pathology were detected. The increased frequency of convulsions in SD rats may be influenced by environmental factors, age, and handling frequency. The compiled database will support toxicologists and pathologists in distinguishing between test article-related effects and spontaneous or procedural findings in future studies.

Acknowledgements

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec (Suzhou) Co., Ltd.

SCAN FOR A COPY