Combining Immunocapture and Microflow Liquid Chromatography-Tandem Mass Spectrometry to Advance Bioanalysis of Protein Biomarkers in Toxicological Studies

Zhuoheng Zhou, Pei Zhang, Xuejin Yang, Linkai Li, Mingzhu Yang, Jing Huang, Jinpeng Li*
Department of Toxicology, WuXi AppTec Ltd., Suzhou, China

WuXi AppTec

Abstract / Poster Number: 4632 / E410

Background and Purpose

Biomarkers play a critical role throughout the entire drug-development lifecycle, contributing to both safety and efficacy assessments. As industrial report [1] suggested, from 2011 to 2020, drugs developed with biomarker guidance led to 2 times higher success rate in Phase II trial and 3 times higher overall approval than those developed without. Proteins, as the functional end products of the central dogma, their expression profiles closely reflect the physiological activity, making them particularly suitable as biomarkers. The Early Detection Research Network (EDRN) currently catalogs 627 protein biomarkers across ten organs, corresponding to 45% of the total entries. So far nearly all clinical biomarkers approved by FDA are protein-based. Protein biomarkers are commonly measured by ligand-binding assays such as ELISA and electrochemiluminescence assay. These platforms however, have relatively limited assay ranges and rely upon specific antibody accessibility. By contrast, liquid chromatography–tandem mass spectrometry (LC–MS/MS) has emerged as an important alternative due to high specificity and methodological robustness. LC–MS/MS methods are also inherently high-throughput and can quantify multiple biomarkers in a single assay, thereby shortening laboratory turnaround time, and lowering cost.

Figure 1 The success rate of the clinical trials with or without biomarker monitoring in different phases from 2011 to 2020 [2]

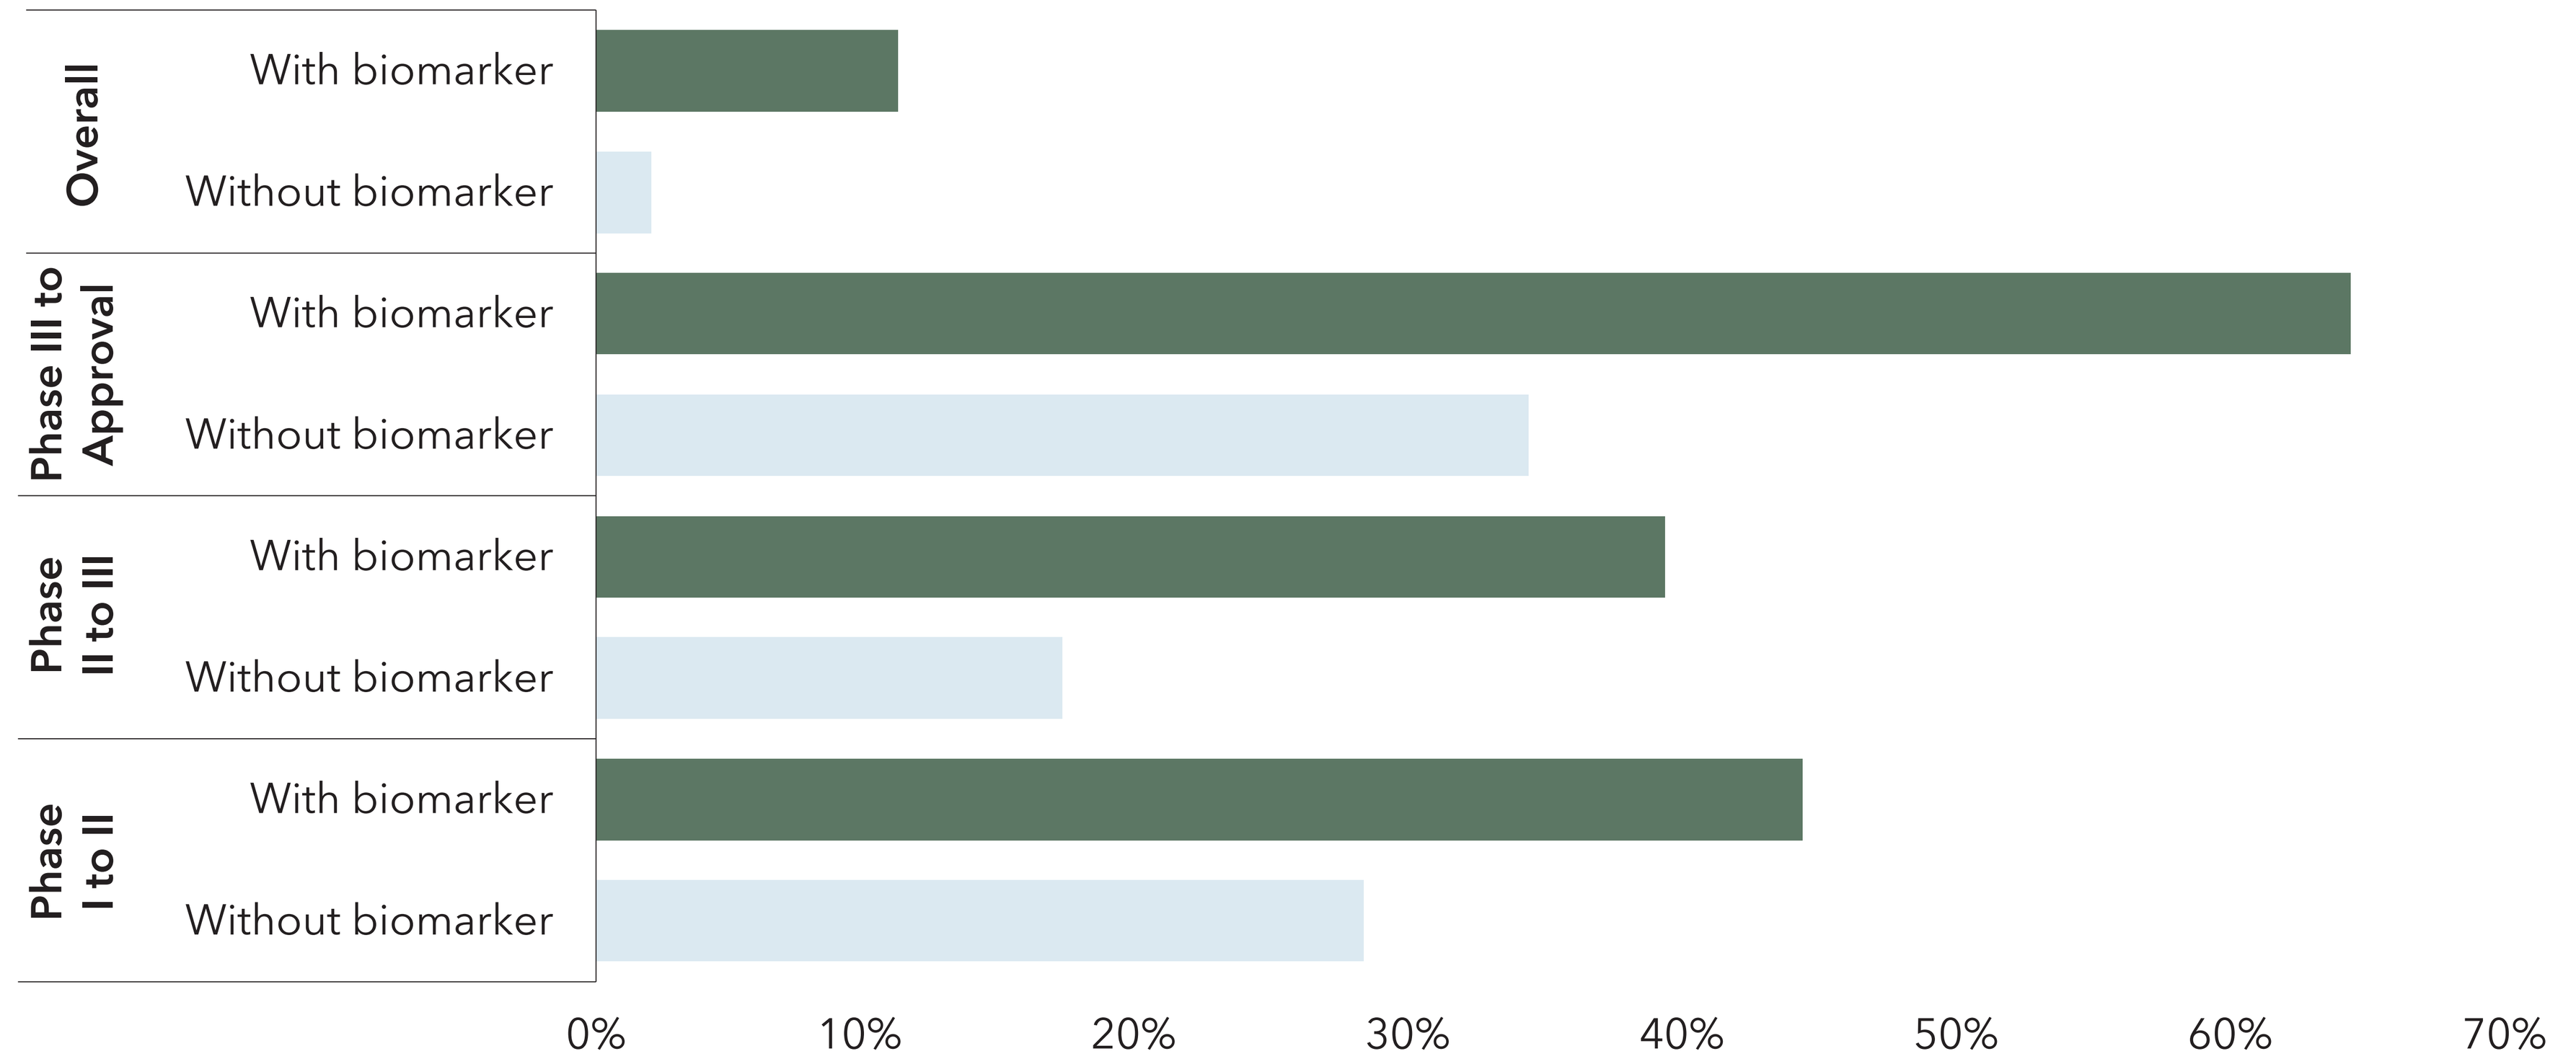
Overall
Phase III to Approval
Phase II to III
Phase I to II
With biomarker
Without biomarker
0%
10%
20%
30%
40%
50%
60%
70%

The conventional LC–MS/MS platforms however, generally achieve lower limits of quantification (LLOQ) for proteins/peptides in the order of 1–10 ng/mL, which is insufficient for many endogenous protein biomarkers (expressed at pg/mL even fg/mL level). Improving LC–MS/MS sensitivity is therefore the key to further advance protein biomarker bioanalysis. In this work, we streamlined an analytical workflow integrating immunocapture and microflow liquid chromatography (microLC), aiming to push further the protein quantitation limit, while maintaining a wide dynamic range. The established methods were validated and applied in real-world preclinical toxicology studies.

Methods and Test System

The serum samples were shaking-incubated at room temperature overnight, together with in-house prepared anti-biomarker beads based on biotin-streptavidin interaction. After repeatedly washing the beads with PBST buffer, the captured proteins were eluted with water containing 0.1% v/v TFA. Following centrifugation, dithiothreitol and iodoacetamide were added to the supernatant sequentially for protein reduction and alkylation. The proteins were digested by trypsin with an 1:10 enzyme-to-substrate ratio at 37 °C for 3 h before injection. The chromatographic separation was running at 12 μL/min on an ACQUITY M-Class System equipped with a BEH C18 column (130 Å, 1.7 μm, 300 μm * 100 mm). The chromatograms were acquired in MRM mode on a SCIEX 7500 system, with transitions of the signature peptide preselected by empirical validation accessing public databases. The quantitation of endogenous protein biomarker was carried out with external standard method using surrogate matrices.

Figure 2 Schematics of the immunocapture workflow followed by enzymatic digestion and microLC-MS/MS quantification of protein biomarkers in biological matrices

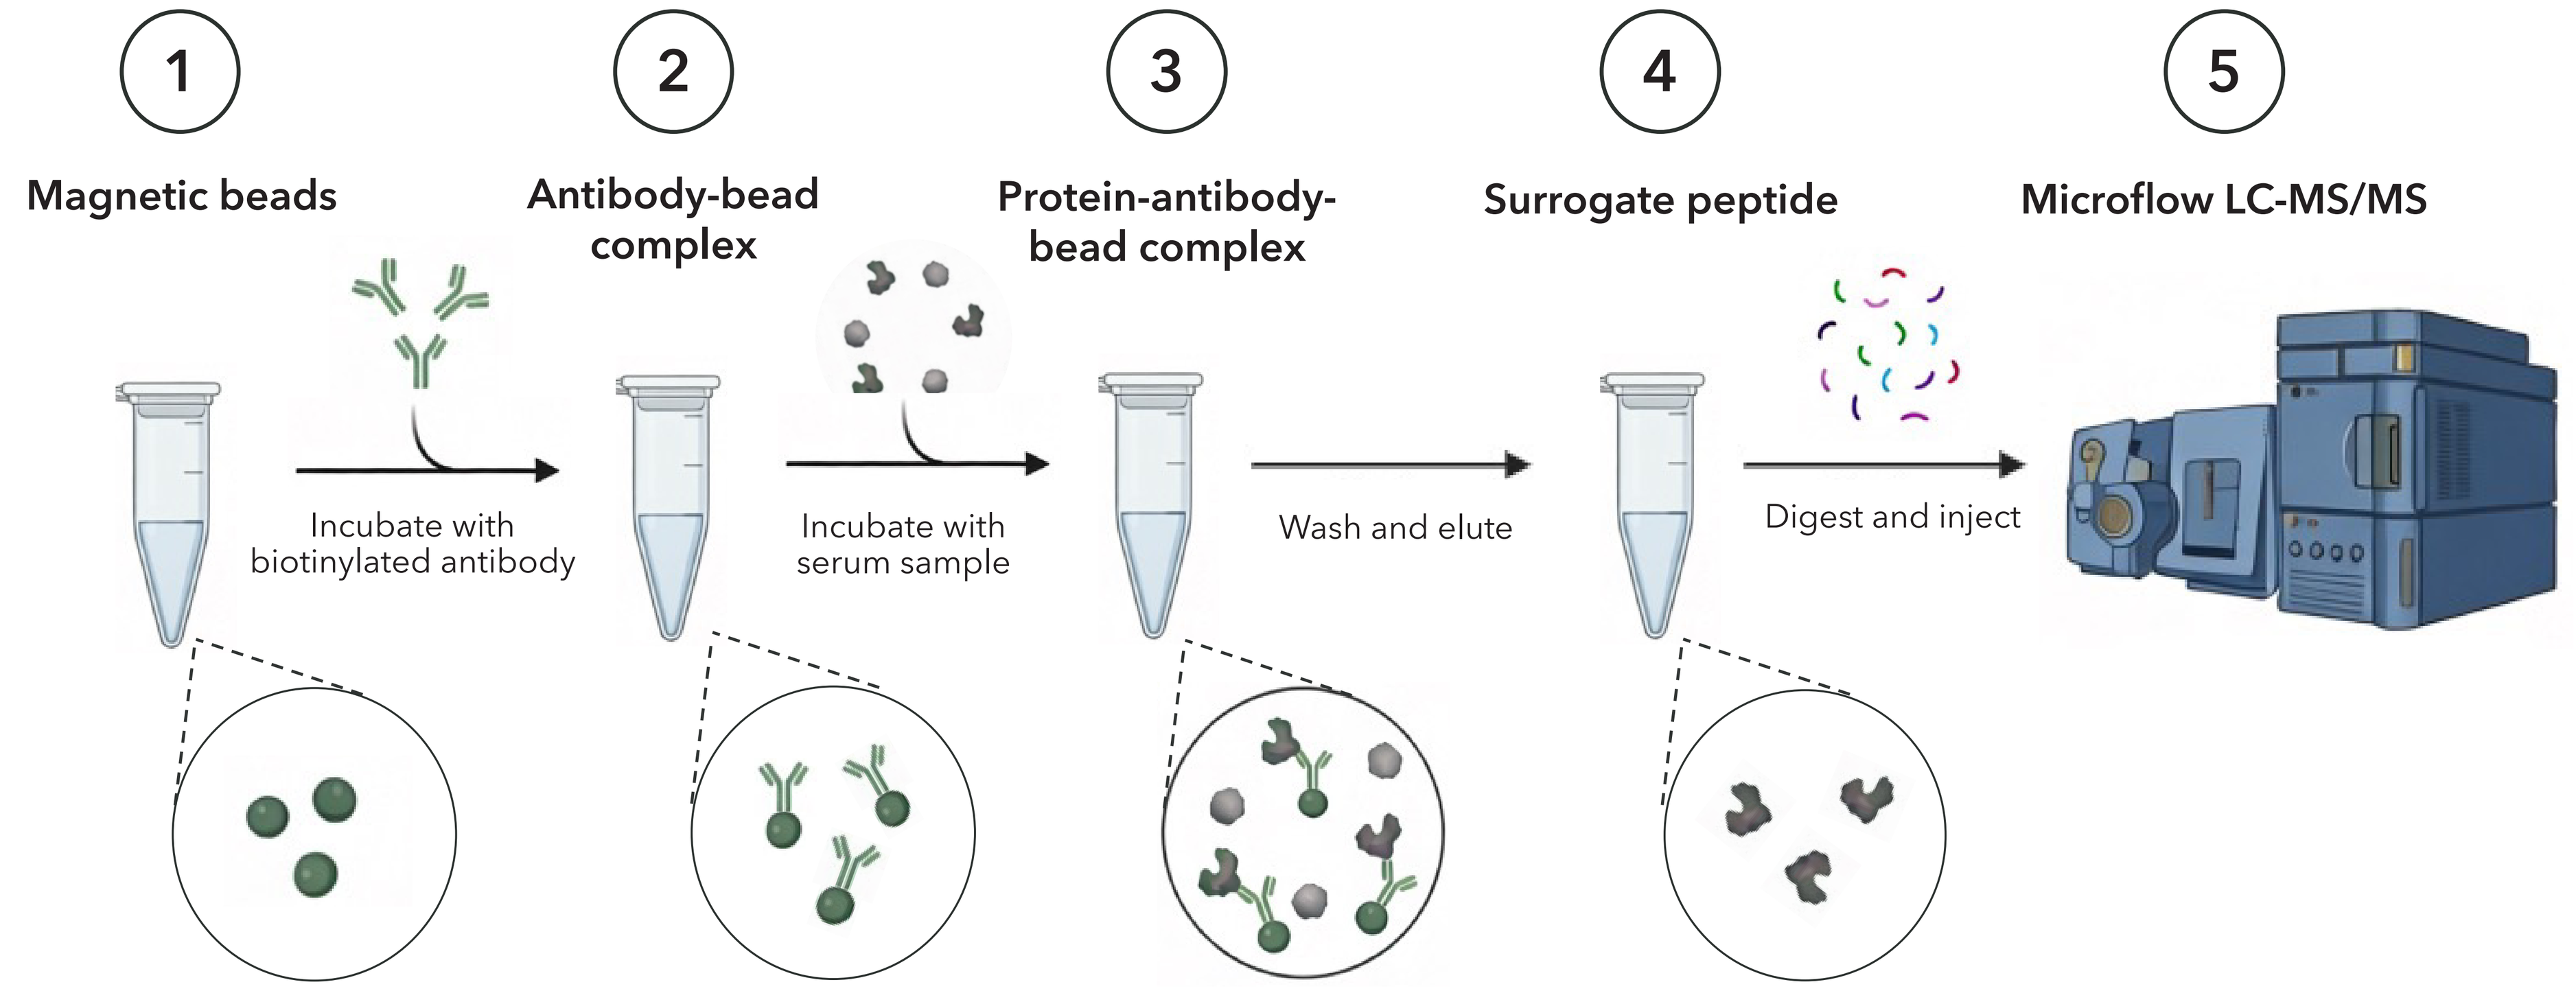
1
Magnetic beads
2
Antibody-bead complex
3
Protein-antibody-bead complex
4
Surrogate peptide
5
Microflow LC-MS/MS
Incubate with biotinylated antibody
Incubate with serum sample
Wash and elute
Digest and inject

Results

Systematic method optimization was implemented regarding immunocapture recovery, digestion efficiency, chromatographic resolution and MS detectability. Among all parameters, beads-to-protein ratio, enzyme-to-substrate ratio, signature peptide selection, and chromatographic flowrate were found most impactful for sensitivity improvement, while surrogate matrices selection was critical to assure quantification precision and accuracy. The applicability and figure of merits of the developed workflow were showcased with bioanalysis of secreted TNF-like ligand 1A (sTL1A) in NHP serum in general toxicology studies, to monitor the immunomodulation of DR3-TL1A pathway upon dosage. The LLOQ was achieved at 50 pg/mL with linearity across three orders of magnitude. The precision, accuracy as well as method robustness have been validated fully adhering GLP regulatory guidance.

Figure 3 Systematic optimization on the antibody-bead complex suspension volume (A), immunocapture incubation time (B), digestive enzyme amount (C) and digestion time (D) to enhance the recovery of the surrogate peptide for quantification of sTL1A in serum samples.

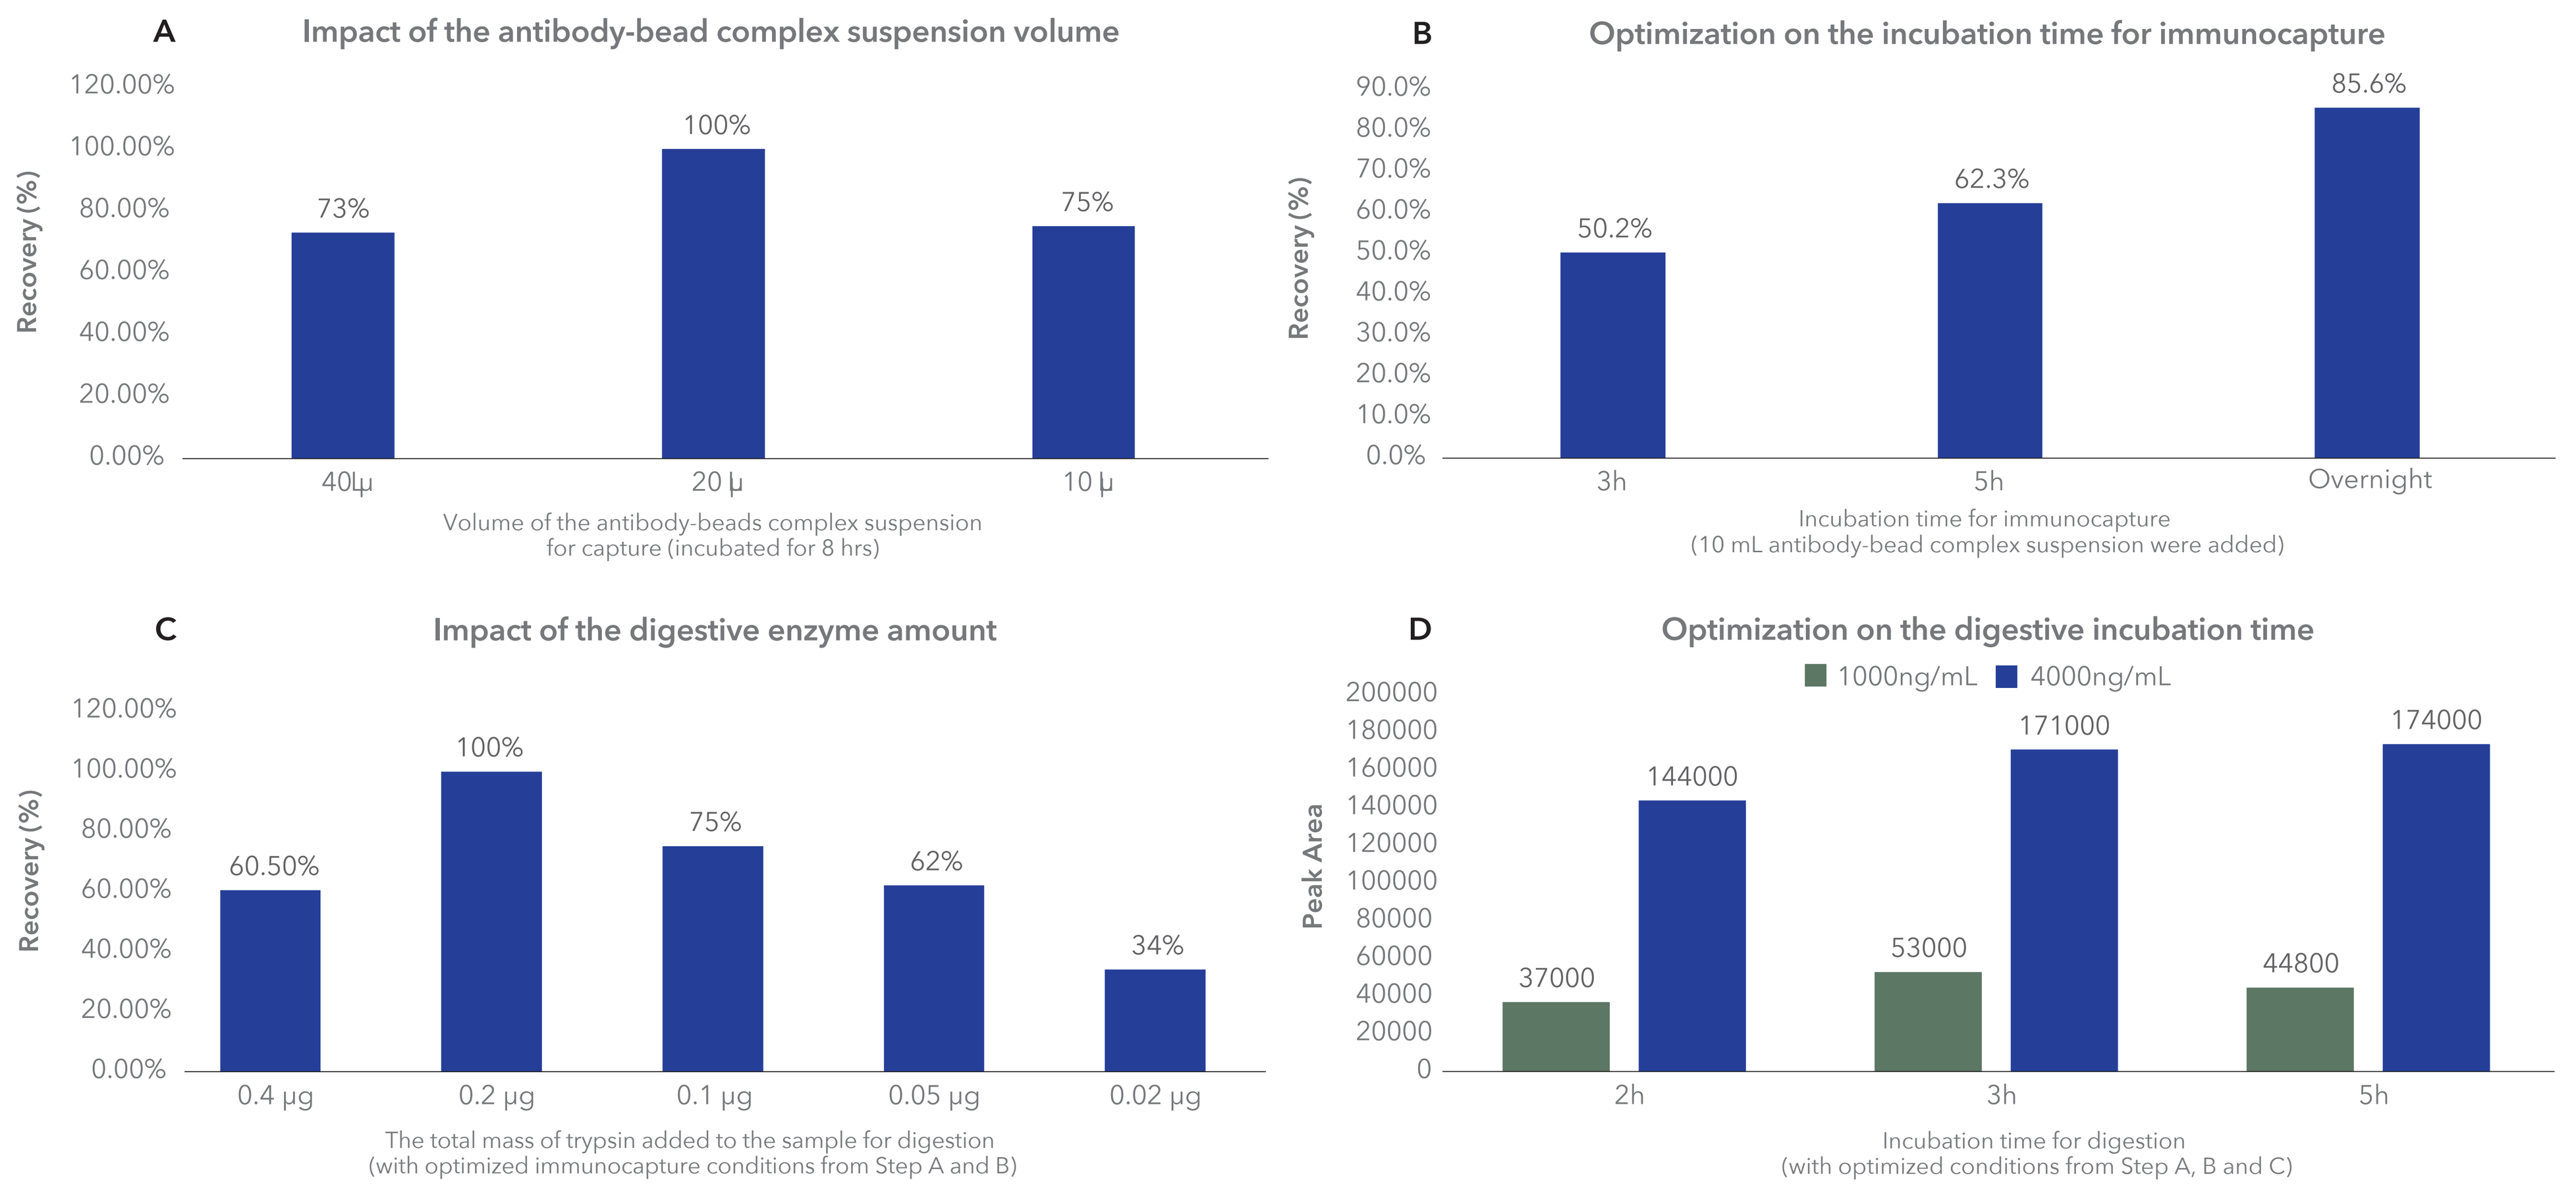
A
Impact of the antibody-bead complex suspension volume
Recovery (%)
120.00%
100.00%
80.00%
60.00%
40.00%
20.00%
0.00%
73%
100%
75%
40μ
20 μ
10 μ
Volume of the antibody-beads complex suspension for capture (incubated for 8 hrs)
B
Optimization on the incubation time for immunocapture
Recovery (%)
90.0%
80.0%
70.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
50.2%
62.3%
85.6%
3h
5h
Overnight
Incubation time for immunocapture (10 mL antibody-bead complex suspension were added)
C
Impact of the digestive enzyme amount
Recovery (%)
120.00%
100.00%
80.00%
60.00%
40.00%
20.00%
0.00%
60.50%
100%
75%
62%
34%
0.4 μg
0.2 μg
0.1 μg
0.05 μg
0.02 μg
The total mass of trypsin added to the sample for digestion (with optimized immunocapture conditions from Step A and B)
D
Optimization on the digestive incubation time
1000ng/mL
4000ng/mL
Peak Area
200000
180000
160000
140000
120000
100000
80000
60000
40000
20000
0
37000
144000
53000
171000
44800
174000
2h
3h
5h
Incubation time for digestion (with optimized conditions from Step A, B and C)

Figure 4 (A) illustrates the linearity of the developed immunocapture-microLC-MS/MS assay ranges from 0.05 to 4 ng/mL for the sTL1A concentration in monkey serum, while (B) and (C) depicts the chromatographic peaks of sTL1A in blank matrix and at the LLOQ level in spiked matrix, respectively.

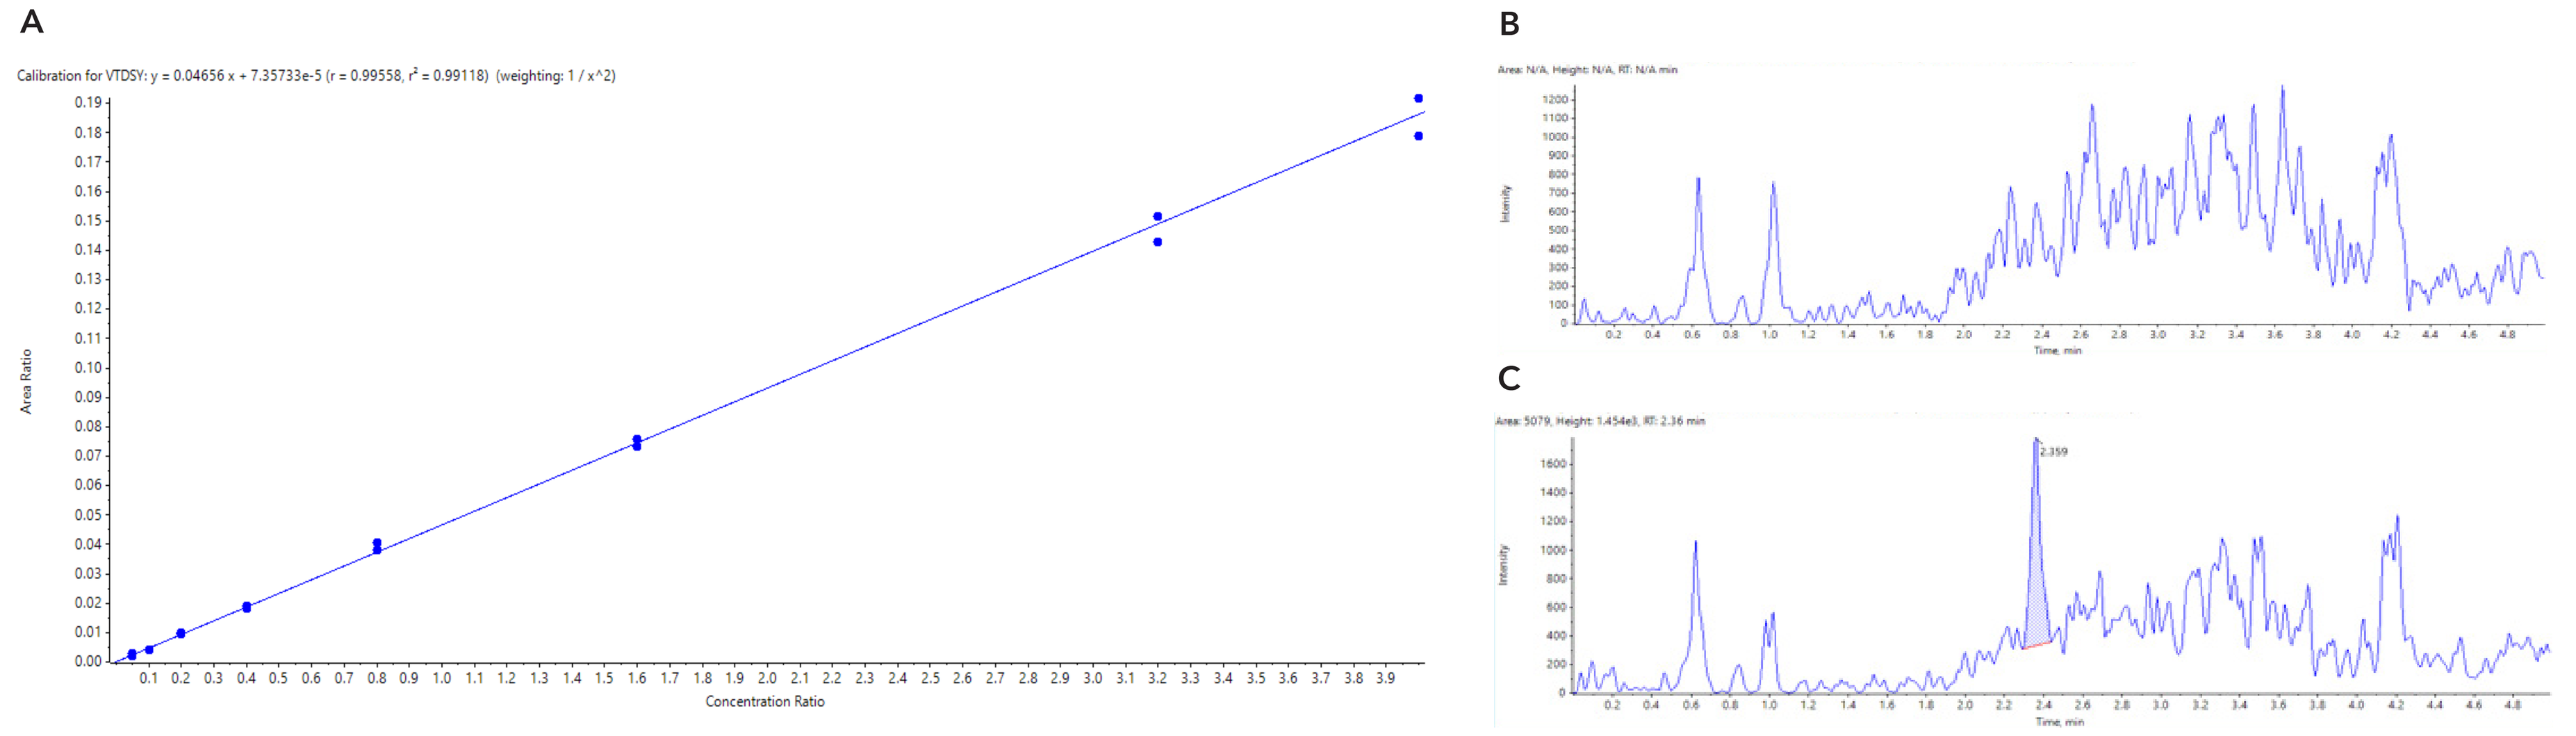
A
Calibration for VTDSY: y = 0.04656 x + 7.35733e-5 (r = 0.99558, r² = 0.99118) (weighting: 1 / x^2)
Area Ratio
Concentration Ratio
B
Area: N/A, Height: N/A, RT: N/A min
Intensity
Time, min
C
Area: 5079, Height: 1.454e3, RT: 2.36 min
2.359
Intensity
Time, min

Conclusion

A streamlined bioanalytical workflow has been established combining immunocapture and microLC-MS/MS towards highly sensitive protein biomarker quantitation in biological matrices, with significantly reduced analytical turnaround time and reagent costs as well as improved method robustness. The applicability and advantages have been demonstrated in real-world general toxicological studies with full GLP compliancy. It also suggests great potential in multiplexing. By monitoring multiple canonical surrogate peptides, this workflow can potentially quantify a larger biomarker panel simultaneously in "single-shot", providing in-depth physiological information towards preclinical development.

References

[1] https://www.bio.org/clinical-development-success-rates-and-contributing-factors-2011-2020 [2] https://www.technologynetworks.com/proteomics/blog/using-protein-biomarkers-increases-the-chances-of-success-in-clinical-trials-329018

SCAN FOR A COPY