Cytokine Profiling for Immunotoxicity Assessment: Comparison of MSD®, Luminex®, and CBA Multiplex Platforms in Preclinical Matrices

Qinlei Gu*, Jinpeng Li*, Tingting Yu, Wenwen Sheng, Bo Wei, Yufeng Zong, Xiaochen Guo, Chen Zhu, Mingming Wang, Xiaoying Jin
WuXi AppTec, 1318 WuZhong Avenue, Suzhou, Jiangsu, China
*Authors contributed equally.

Abstract / Poster Number: 4749 / G540

Background and Purpose

Cytokines are the key signaling molecules facilitating the inter-cellular communications in both innate and the adaptive immune system. These small protein/glycoprotein or peptide molecules mediate critical immunological activities, reflect immune activation, and indicate disease status. Cytokine profiling is central to immunotoxicity assessment during drug development. Ligand binding assays are routinely employed to detect cytokine release at the protein level. Multiplex cytokine analysis methods are widely adopted for capturing complex cytokine profiles, offering advantages over singleplex methods by reducing sample handling and minimizing total sample volume requirements.

Among these platforms, cytometric bead array (CBA), Meso Scale Discovery (MSD®) and Luminex® provide high-throughput analysis of cytokines for preclinical risk assessment purpose. Despite their popularity, choice of the right platform and interpreting results across different platforms remains challenging. In this study, we directly compare cytokine profiles generated by three multiplex platforms (MSD®, CBA, and Luminex®) using identical sample sets in a single-laboratory setting.

Methods and Test System

Sample

Stock naïve serum and positive control serum samples from non-human primates (NHPs) and in supernatants from human peripheral blood mononuclear cells (huPBMCs) stimulated with in-house positive control mixes were tested for cytokine measurement. Naïve and positive NHP serum were collected in the site facility and stocked in -70˚C freezer for < 3months. huPBMCs were purchased from qualified supplier and then incubated at 37˚C with in-house cytokine-stimulation mix or plain tissue culture medium. Supernatant were collected for cytokine measurement after 24 hours.

Cytokine Measurement

The cytokines IL-2, IL-6, TNF-α, and IFN-γ were quantified in all NHP samples using three multiplex platforms : ECL platform (MSD® U-plex), Luminex® platform (Thermofisher ProcartaPlex) and CBA platform (Biolegend LEGENDplex) . The cytokines IL-2, IL-6, IL-10, and IFN-γ in all human samples with MSD® and CBA platform (BD Flex set for human supernatant). Additional analytes were measured on specific platforms in the same serum: IP-10 was quantified in NHP serum in CBA and Luminex® panel in the same assay as other analytes. All assays were performed according to the manufacturers' protocols. Selectivity/ parallelism was performed to determine MRD. Accuracy and precision run was also evaluated and acceptance criteria were CV <25%. Results from the same sample group were compared across different platforms. Data values below the lower limit of quantification (LLOQ) were substituted with zero for comparative analysis.

Statistics

Statistical comparisons were performed using ANOVA for comparing the mean for more than 2 groups or by pairwise t-tests for 2 groups.

Results

In unstimulated human PBMC and naïve NHP samples, baseline levels were comparable across all platforms (Fig 1, Fig 2D, Fig 3A-D). However, platform-specific differences were observed for IL-2, IL-10, and IP-10 in positive NHP serum (Fig 2B-D) and for IL-10 detection in stimulated human PBMC supernatant (Fig 3D).

Luminex® and CBA platforms detected zero IL-2 release in positive NHP serum (Fig 2B), while the MSD® platform showed moderate IL-2 release in the sample serum samples, indicating potential differences in antibody binding between the two IL-2 assays. Additionally, IP-10 in stimulated NHP serum was detectable only by MSD®, but not by Luminex® (Fig 2D). Despite that the IL-10 release demonstrated a consistent trend after stimulation in both platforms (Fig 3D), IL-10 levels in the stimulated human PBMC supernatant were significantly higher when measured with CBA than with MSD® (p-value = 0.0391).

While IL-6 and IFN-γ levels in both human and NHP samples showed no significant statistical differences across platforms, absolute concentration values varied by over threefold (Fig 3A & 3C). Consequently, these platforms are suitable for relative quantification to identify the trends, but direct comparison of data values generated from different platforms is not recommended.

Figure 1 Maximal and minimal ranges of positive and naïve NHP serum for IFN-gamma, IL-2, IL-6 and TNF-alpha measured with the MSD®, Luminex®, and CBA multiplex immunoassay kits. n=5 for both stocked positive and naïve serum. Data below the LLoQ of each cytokine was substituted with 0. ANOVA test was performed with p-value = (A) 0.5352 , (B) 0.0193, (C) 0.1002, (D) 0.2441.

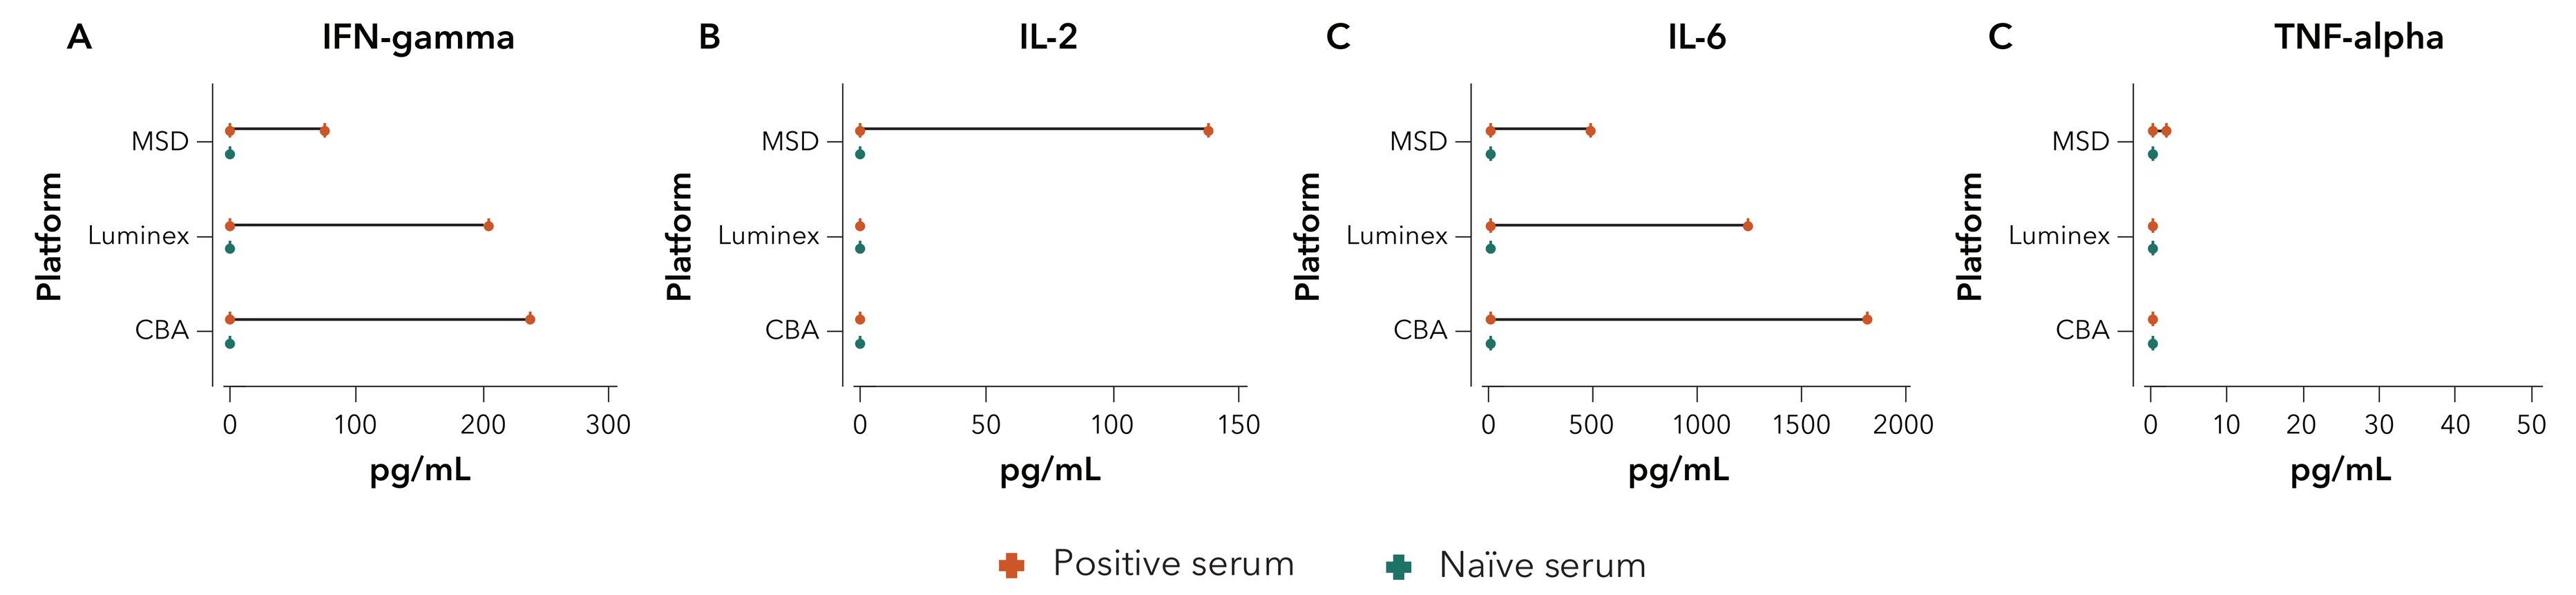
A IFN-gamma
B IL-2
C IL-6
C TNF-alpha
Platform
MSD
Luminex
CBA
pg/mL
Positive serum
Naïve serum

Figure 2 Dotplot depiction of relative analyte concentrations measured by the MSD®, Luminex®, and CBA multiplex cytokine immunoassay kits. (A) IFN-gamma, (B) IL-2 and (C) IL-6 levels were detected in positive NHP serum (n=5) with MSD®, Luminex®, and CBA platforms. IP-10 level was detected with Luminex and CBA assay. IP-10 level from both naïve and positive NHP serum was represented in (D). Connected lines represent the same sample. Concentrations (x-axis) in (A), (B) and (C) are on linear scale and in (D) are on log scale. Data below the LLoQ of each cytokine was substituted with 0.

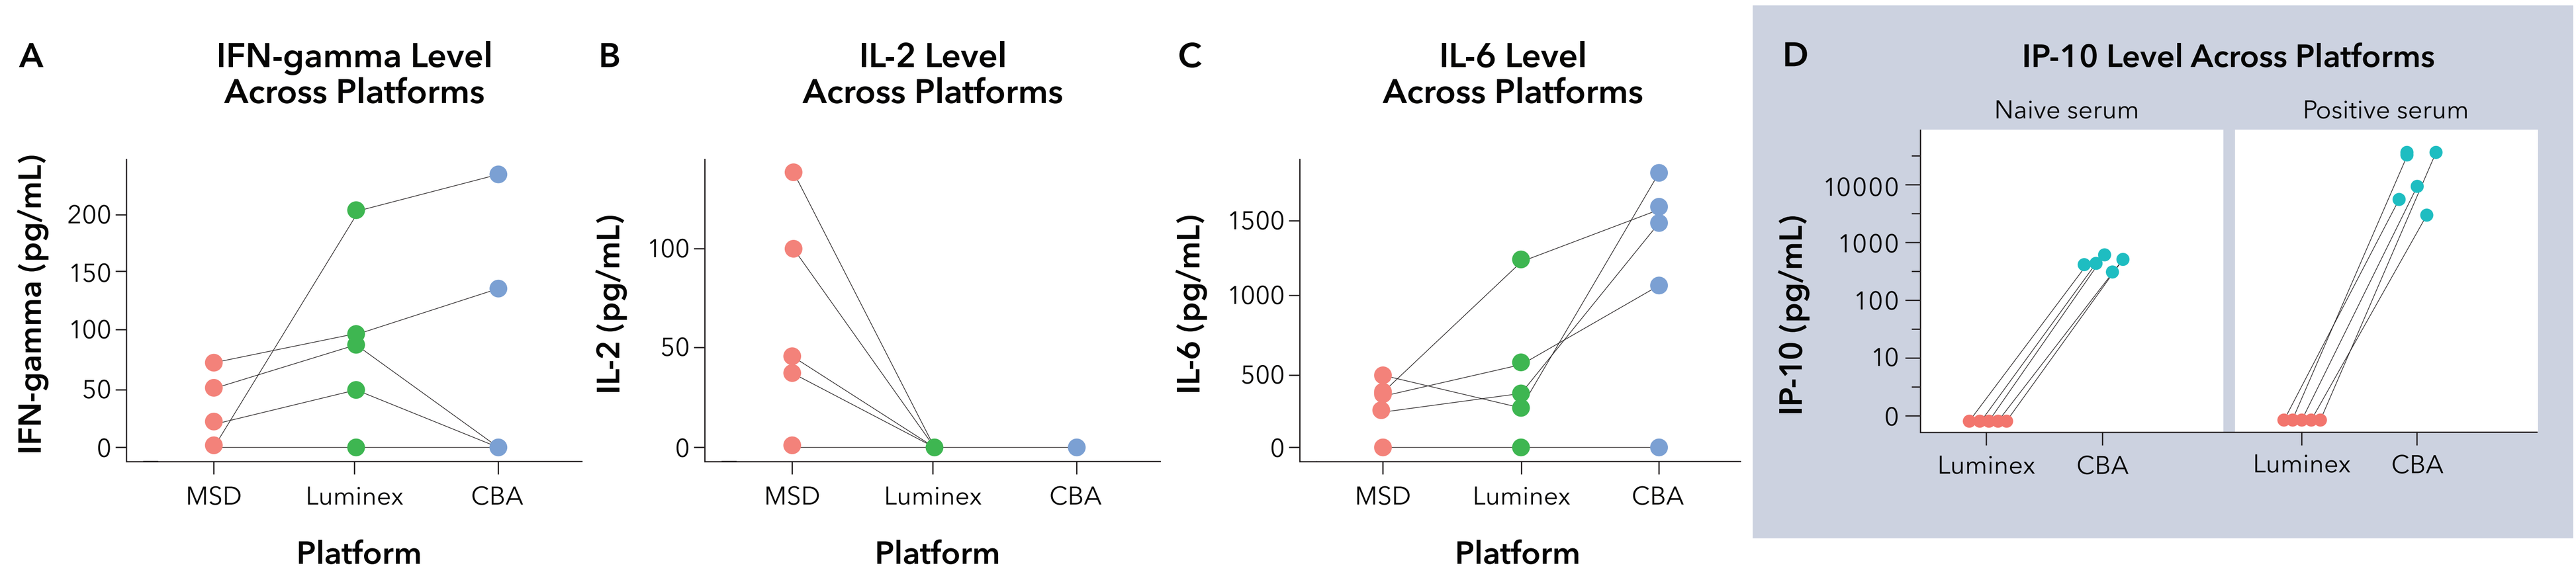
A IFN-gamma Level Across Platforms
B IL-2 Level Across Platforms
C IL-6 Level Across Platforms
D IP-10 Level Across Platforms
Naive serum
Positive serum
IFN-gamma (pg/mL)
IL-2 (pg/mL)
IL-6 (pg/mL)
IP-10 (pg/mL)
MSD
Luminex
CBA
Platform

Figure 3 Dotplot depiction of relative analyte concentrations measured by the MSD® and CBA multiplex cytokine immunoassay kits. IFN-gamma release from unstimulated human PBMC were shown in (A). IFN-gamma release in stimulated sample were above the detection range for both platforms, and was not plotted in the graph. Release of cytokine IL-2, IL-6 and IL-10 from both stimulated and unstimulated PBMC were shown in (B), (C), and (D). Concentrations (x-axis) in (A) are on linear scale and in (B), (C) and (D) are on log scale. Dot of each color represents one animal. Stimulated and unstimulated PBMC are colored as in figure legend.

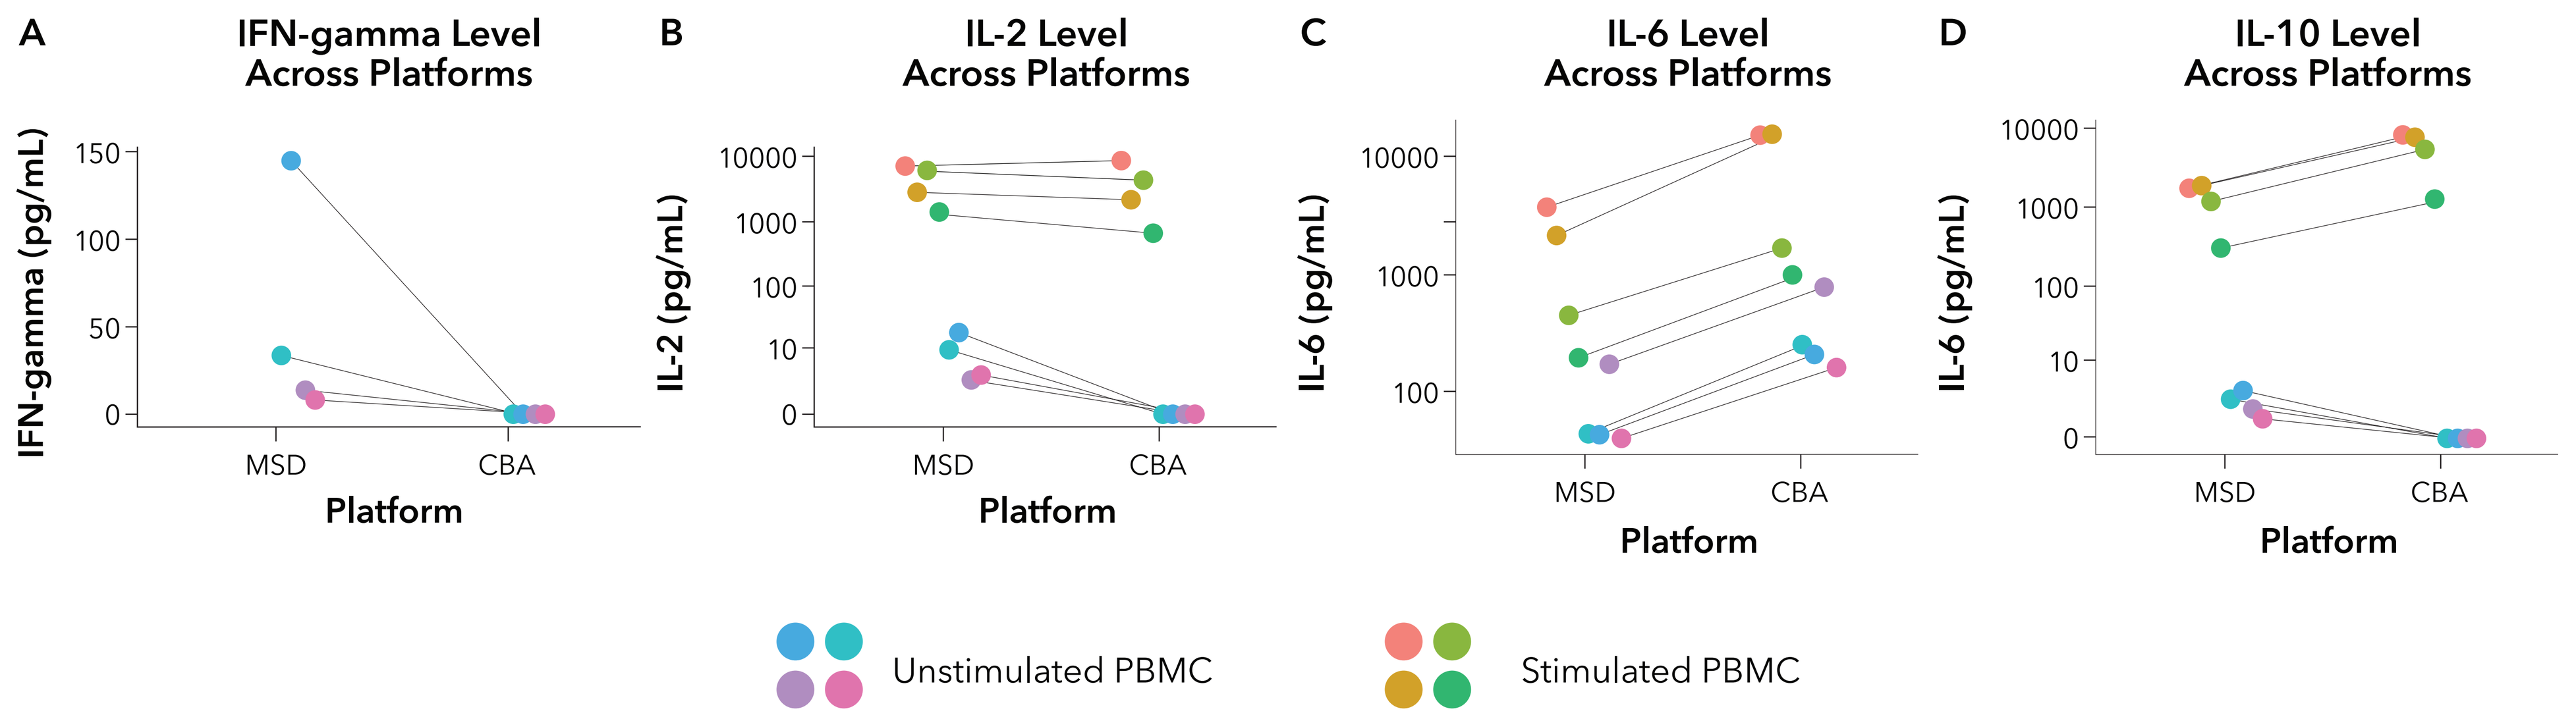
A IFN-gamma Level Across Platforms
B IL-2 Level Across Platforms
C IL-6 Level Across Platforms
D IL-10 Level Across Platforms
IFN-gamma (pg/mL)
IL-2 (pg/mL)
IL-6 (pg/mL)
MSD
CBA
Platform
Unstimulated PBMC
Stimulated PBMC

Conclusion

Our data demonstrated that selecting a multiplex cytokine platform (MSD®, Luminex®, or CBA) for preclinical immunotoxicity assessment requires a fit-for-purpose approach. When comparison between data set is expected, trend rather than data point values may be comparable for data generated across platforms. However, when direct comparison of absolute cytokine levels is crucial, data set from same platform may be required.

References

1. Günther A, Becker M, Göpfert J, Joos T, Schneiderhan-Marra N. Comparison of Bead-Based Fluorescence Versus Planar Electrochemiluminescence Multiplex Immunoassays for Measuring Cytokines in Human Plasma. Front Immunol. 2020;11:572634. doi:10.3389/fimmu.2020.572634

2. Platchek M, Lu Q, Tran H, Xie W. Comparative Analysis of Multiple Immunoassays for Cytokine Profiling in Drug Discovery. SLAS Discovery. 2020;25(10):1197-1213. doi:10.1177/2472555220954389

MSD® is a registered trademark of Meso Scale Diagnostics, LLC and Luminex® is a registered trademark of Luminex Software, Inc.

SCAN FOR A COPY