A Retrospective Analysis of the Potential Associated Factors of Procedure-Related Findings in Intrathecal Injection Studies

Ruixuan Wang, Leo Pan, Sue McPherson
WuXi AppTec, Suzhou, China

Abstract: 3265

Background and Purpose

Despite ongoing breakthroughs in the field of drug therapy with the advancement of technology, the development of central nervous system (CNS) drugs still faces significant challenges, mainly because of the poor drug access to the CNS. It has been estimated that for greater than 98% of small molecules and nearly 100% of large molecules, systemic delivery to the CNS is not effective. The CNS system is protected by the blood-brain barrier (BBB) and the blood-cerebrospinal fluid barrier. Therefore, local or non-system drug delivery route, such as intrathecal (IT) injection – injection into the between the thin layers of tissue that cover the brain and spinal cord provides a direct means of administration the cerebrospinal fluid (CSF) to bypass the BBB and gain direct access to the CNS. With no meninges in the way of diffusion, the dose required is much smaller, and the onset of maximal effect is more rapid. The smaller dose also means that systemic toxicity is lower. This route of administration is not restricted to drugs for the CNS but also pain management where the drug can reach the supraspinal pathways. However, in most IT injection studies, alongside changes related to the test article, there are also some histopathology changes associated with the dosing administration procedures, which are always difficult for the study pathologists to distinguish from the test article related findings. This retrospective review aimed to analyze factors that may be related to these findings, with the goal of providing guidance to minimize such lesions in future study designs, and also provide a guide to the scientist to put the study findings into perspective and to determine what were truly related to the test material and what were procedural related.

Methods

A retrospective review was performed regarding all studies involving intrathecal administration as the dosing route conducted in the facility from 2021 to the present. Histopathological examinations were performed in 15 of these studies. The animal species involved consisted of rats and monkeys, with study durations ranging from single dose to 13 weeks. The procedure-related findings were analyzed with the following factors that might have been associated with these lesions such as the age and weight of the animals, the dosing volume, dosing frequency, the number of dosing administrations, the number and frequency of survival CSF collections.

Results

In these studies, whilst the study designs were not entirely the same, a lower rate of procedure-related findings was always observed in studies using older animals and with lower dosing frequencies. The occurrence of findings related to the procedure does not show any clear correlation with the weight of the animals. These details were summarized in Table 1 and Table 2.

Moreover, due to the differences between monkeys and rats, the design of monkey studies typically includes the collection of CSF during the in-life phase to aid in subsequent drug distribution and metabolism analysis. Analysis of the monkey studies with the same dosing frequency but with different survival CSF collection frequency, the results showed a more frequent survival CSF collections at survival were correlated with more procedure-related findings.

Table 1 Potential Associated Factors of Procedure-Related Findings in SD Rats

Species	Dosing Frequency	Dosing Numbers/ Animal	Age at Dosing Initiation (Years)	Dosing Volume (μL)	Body Weight at Dosing Initiation (g)		Rate[a]
					Males	Females	
SD Rats	Single	1	6-7	10	237.11-273.02	168.2-198.8	100.0%
			9-12	30	373.94-424.55	239.7-273.6	2.1%
	Q4W	4	7-8	20	248.46-277.24	176.3-218.0	20.0%
	Q2W	2	6-8	30	272.24-299.09	178.1-204.9	67.7%
			7-8	30	294.39-356.87	183.0-227.1	60.0%
			8-11	10	318.42-375.35	205.7-236.0	-
	QW	5	5-6	30	168.28-217.56	136.2-171.4	90.0%
		2	6-7	10	237.11-273.02	168.2-198.8	50.0%
			7-8	30	290.96-326.87	202.8-228.7	83.0%

Note: Q4W: once every four weeks Q2W: once every two weeks QW: once weekly -: No procedure-related findings were noted.
[a] : Rate = Animals numbers with procedure-related findings/Total animal numbers × 100%

Table 2 Potential Associated Factors of Procedure-Related Findings in Cynomolgus Monkeys

Species	Dosing Frequency	Total Dosing Numbers/ Animal	Age at Dosing Initiation (Years)	Dosing Volume (mL)	Body Weight at Dosing Initiation (kg)		Survival CSF Collection Numbers/Week		Rate[a]
					Males	Females	Maximum Numbers/ Week	Maximum Numbers	
Cynomolgus Monkey	Q2W	2	3-4	1	3.4-4.2	2.8-3.1	1	3	0%
				1	2.8-5.6	-	-	-	0%
				1	2.3-4.4	2.2-3.2	5	11	100.0%
				1	2.8-5.6	-	5	11	100.0%
		4	2.3-2.5	1	2.3-2.6	2.0-2.8	2	7	25.0%
			2.5-3.0	1	2.3-3.3	2.4-3.2	2	8	58.3%
	QW	5	1.5-1.7	1	1.9-2.7	1.8-2.3	1	2	100.0%
		4	3	1.67	2.6-3.5	2.4-2.9	1	6	0%

Note: Q2W: once every two weeks QW: once weekly -: No procedure-related findings were noted.
[a] : Rate = Animals numbers with procedure-related findings/Total animal numbers × 100%

Conclusion

In future studies, whenever possible, to avoid potential interference from these procedure-related injuries on the results, older animals should be selected, and the number and/or frequency of CSF collections at survival should be minimized during the study.

Acknowledgements

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec (Suzhou).

References

1. Nance E, Pun SH, Saigal R, Sellers DL. Drug delivery to the central nervous system. Nat Rev Mater. 2022 Apr;7(4):314-331.
2. Gribkoff VK, Kaczmarek LK. The need for new approaches in CNS drug discovery: Why drugs have failed, and what can be done to improve outcomes. Neuropharmacology. 2017 Jul 1; 120:11-19.