Case Study-Strategy of Follow-up Assays in Genotoxicity

Liwen Gao, Cong Yue, Xiang Shan, Millie Chen, Xiaoxia Li
WuXi AppTec, Suzhou, China

Abstract: 4116

Background and Purpose

According to ICH guideline S2 (R1), when positive results are observed in *in vitro* mammalian cell assay, mechanistic information can be used on a weight of evidence approach for evaluation of lack of genotoxicity, one of which is to follow up with two *in vivo* assays in two different tissues (usually bone marrow and liver) with proper demonstration in systemic exposure. Mechanistic experiments generally not performed in daily practice, so no mechanistic information about the testing compound are obtained.

This abstract provides new sight into combinational study cases for both genotoxicity potential evaluation and indirect mechanistic research, and is especially applicable for cases when rats are used, for it's lack of commercially available specific probes.

Methods

In the cases showing below, standard OECD recommendation study designs were employed, including *in vitro* and *in vivo* assays.

1. In *in vitro* mouse lymphoma (MLA) assay, L5178Y cell line were used and treated with test article in S9-activated 3-hour, non-activated 3-hours or non-activated 24-hours treatment series;
2. *In vitro* mammalian chromosomal aberration assay (CAA), CHO-WBL cells were treated with test article in S9-activated 3-hour, non-activated 3-hours and non-activated 24-hours treatment series;
3. *In vitro* micronucleus (MN) assay, CHO-WBL or TK 6 cells were treated in S9-activated 3-hour, non-activated 3-hours and non-activated 24-hours treatment series;
4. *In vivo* micronucleus (MN) assay, animals were administrated at a 24-hour interval for 3 consecutive days, bone marrow or peripheral blood samples were collected at 22 (20-24) hours post last dosing;
5. *In vivo* alkaline comet assay, animals were given daily treatments over a duration of 3 days (at approximately 24 hour intervals), and samples (liver or the duodenum) were collected once at 2-6 h (or Tmax) after the last treatment;
6. *In vivo* CAA assay, animals were administered once with test articles, followed by injection of metaphase-arresting agent at proper interval prior to sampling, then bone marrow samples were taken twice, 1.5 normal cell cycle post dosing and 24 hours following the first sampling.

Results

Different circumstances were encountered and the recommended strategies as follow-up assays were based on a case-by-case basis.

In the first case, a test article with the intended indication of myelofibrosis was performed in a battery of Ames assay, *in vitro* MLA assay and *in vivo* MN assay, among which Ames and MLA were both negative, while the *in vivo* MN assay was evaluated as positive but the notable increase in micronuclei formation was only observed in the highest dose group. Follow-up assays employed one integrated *in vivo* MN and comet (tissues, liver and duodenum) and one *in vitro* MN assay, follow-up assay results turned out be negative for micronuclei endpoints in the integrated assay, liver was evaluated as negative and duodenum was positive, positive finding was also found in the *in vitro* micronucleus assay (with proper cytotoxicity), these results were in accordance with the DNA repair related mechanistic information.

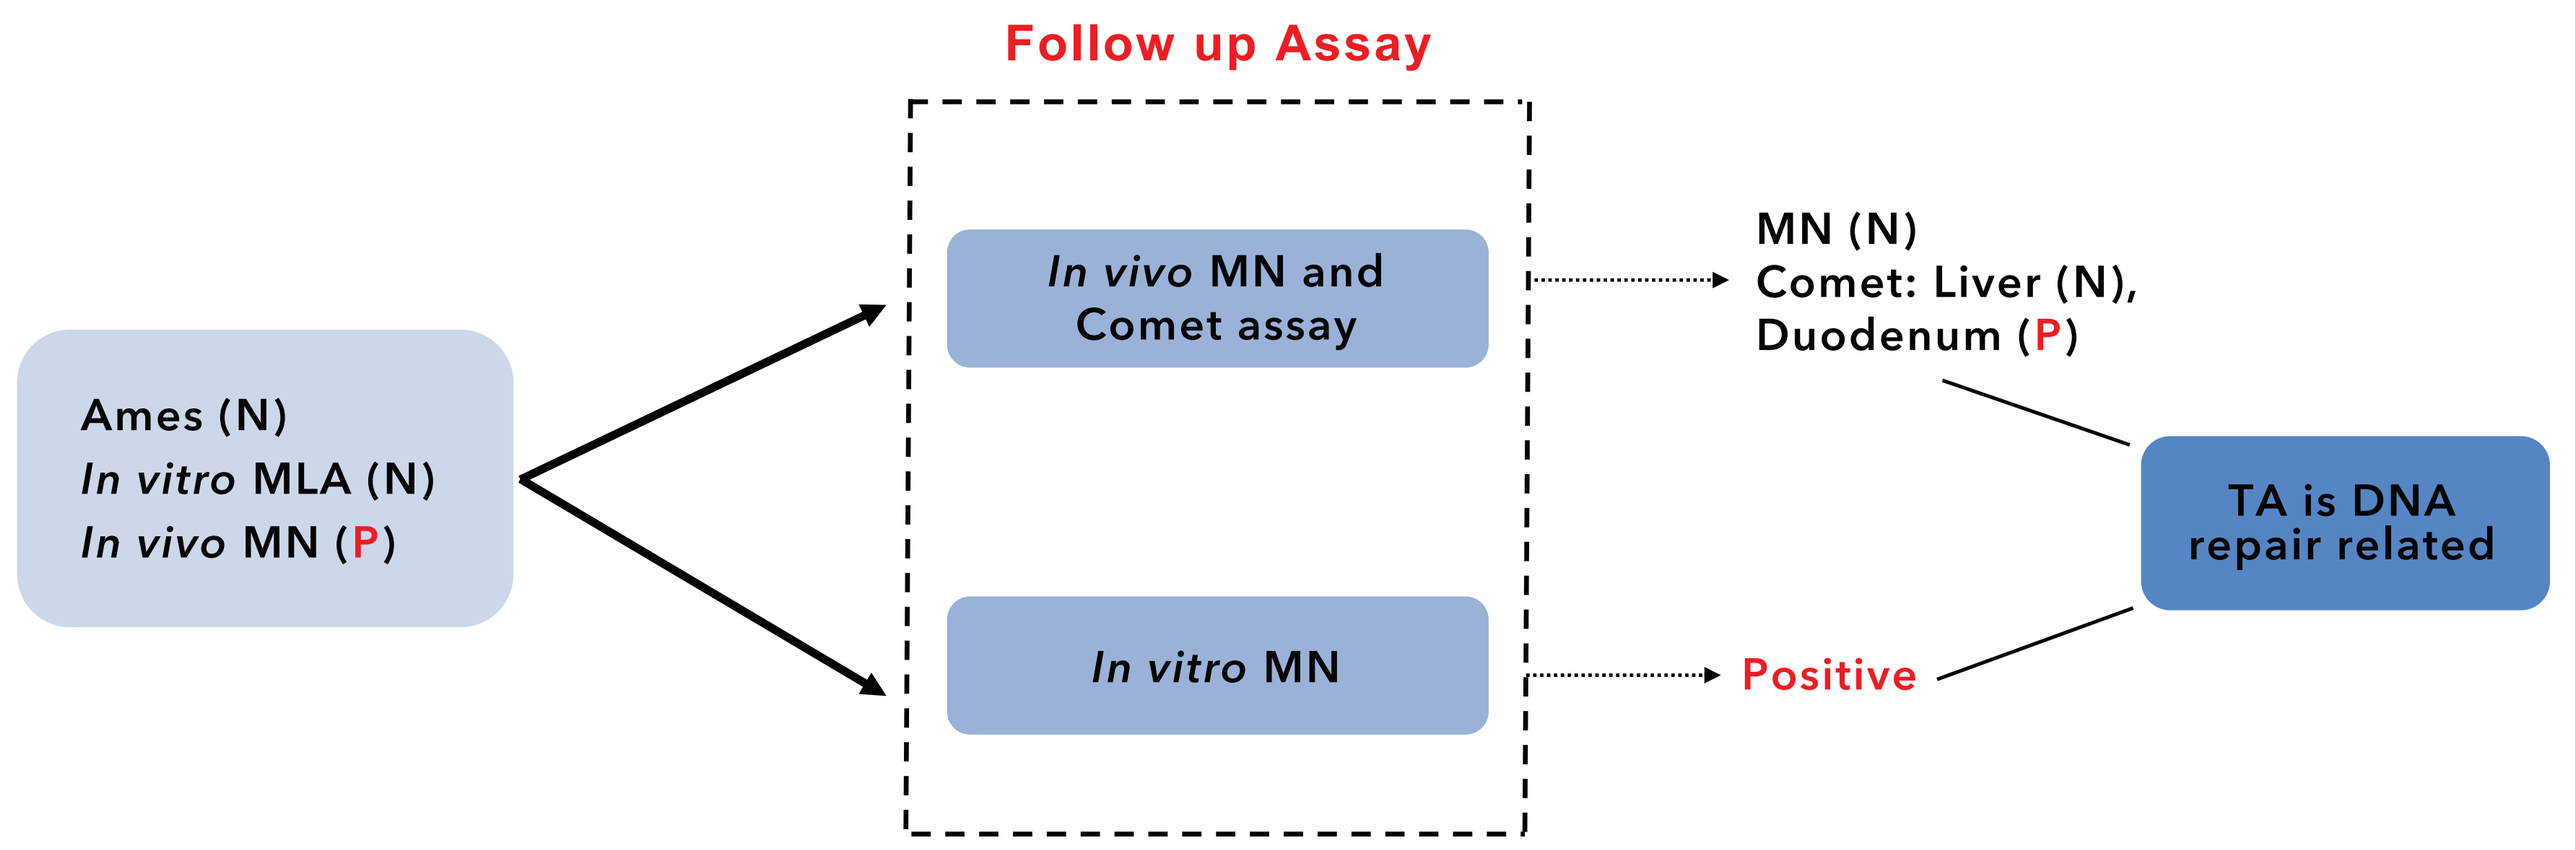
Follow up Assay
Ames (N)
In vitro MLA (N)
In vivo MN (P)
In vivo MN and Comet assay
In vitro MN
MN (N)
Comet: Liver (N),
Duodenum (P)
Positive
TA is DNA repair related

In the second case, Ames, *in vitro* CAA, one preliminary assay and one *in vivo* MN assay were performed, Ames and *in vitro* CAA were both negative, the preliminary assay was evaluated as positive, followed by an equivocal conclusion with optimized doses in the definitive assay. Because of the equivocal/positive signal *in vivo*, a follow-up *in vivo* comet assay (same dose as the definitive *in vivo* MN assay) and an *in vitro* MN were performed. The *in vitro* MN assay turned out to be positive, and the comet assay negative for both tissues. Thus, the consistent positive finding in *in vivo* and *in vitro* system strongly implied that the test article acts through an aneugenic mode of action.

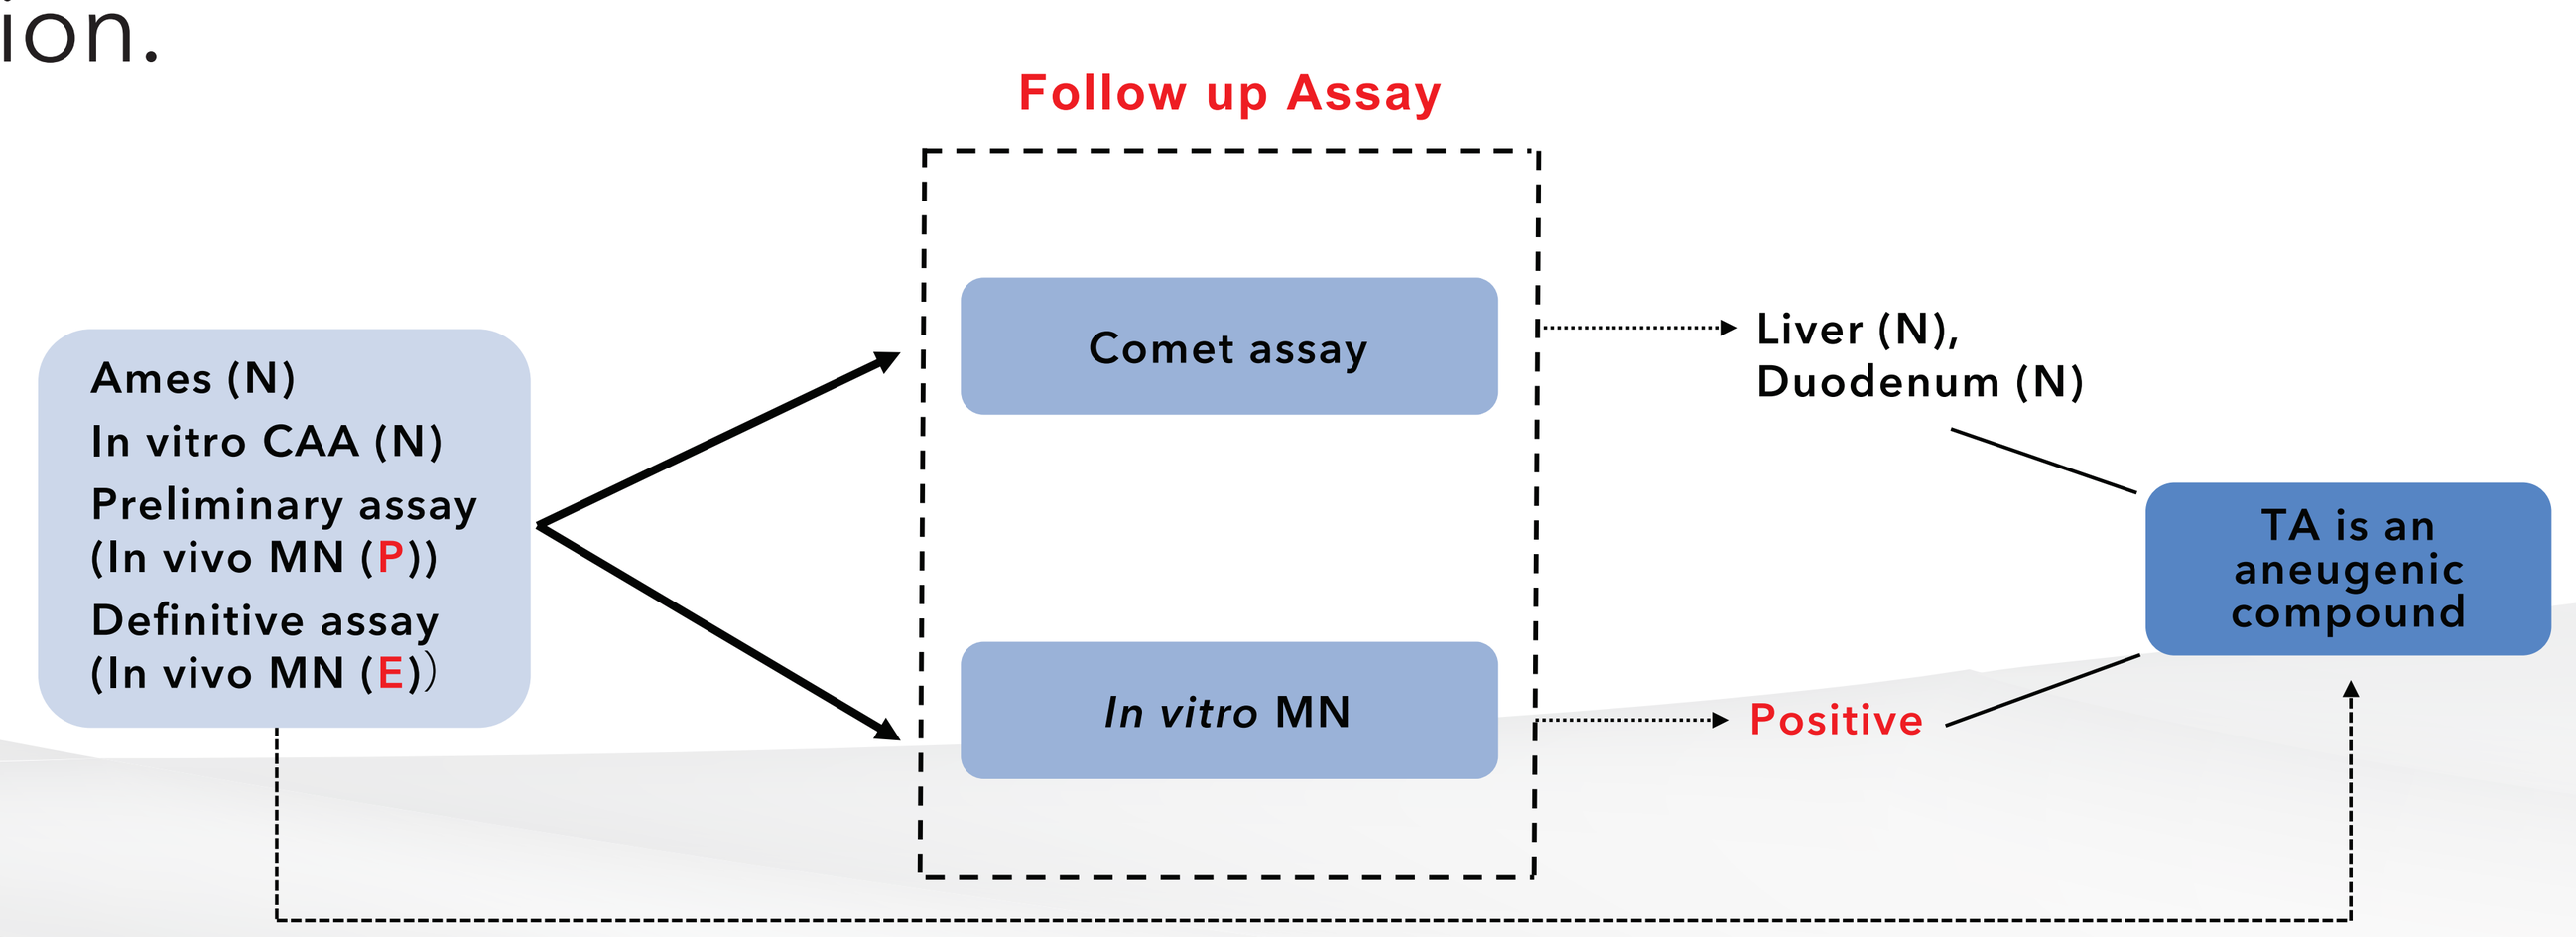
Follow up Assay
Ames (N)
In vitro CAA (N)
Preliminary assay
(In vivo MN (P))
Definitive assay
(In vivo MN (E))
Comet assay
In vitro MN
Liver (N),
Duodenum (N)
Positive
TA is an aneugenic compound

In the next case, Ames, *in vitro* CAA and one *in vivo* MN assay were performed, all were negative except the *in vivo* MN assay, which showed a strong increase in micro-nuclei formation, a follow-up *in vivo* chromosomal aberration assay was introduced, and the result was negative, which also point to the aneugenic potential of the test compound.

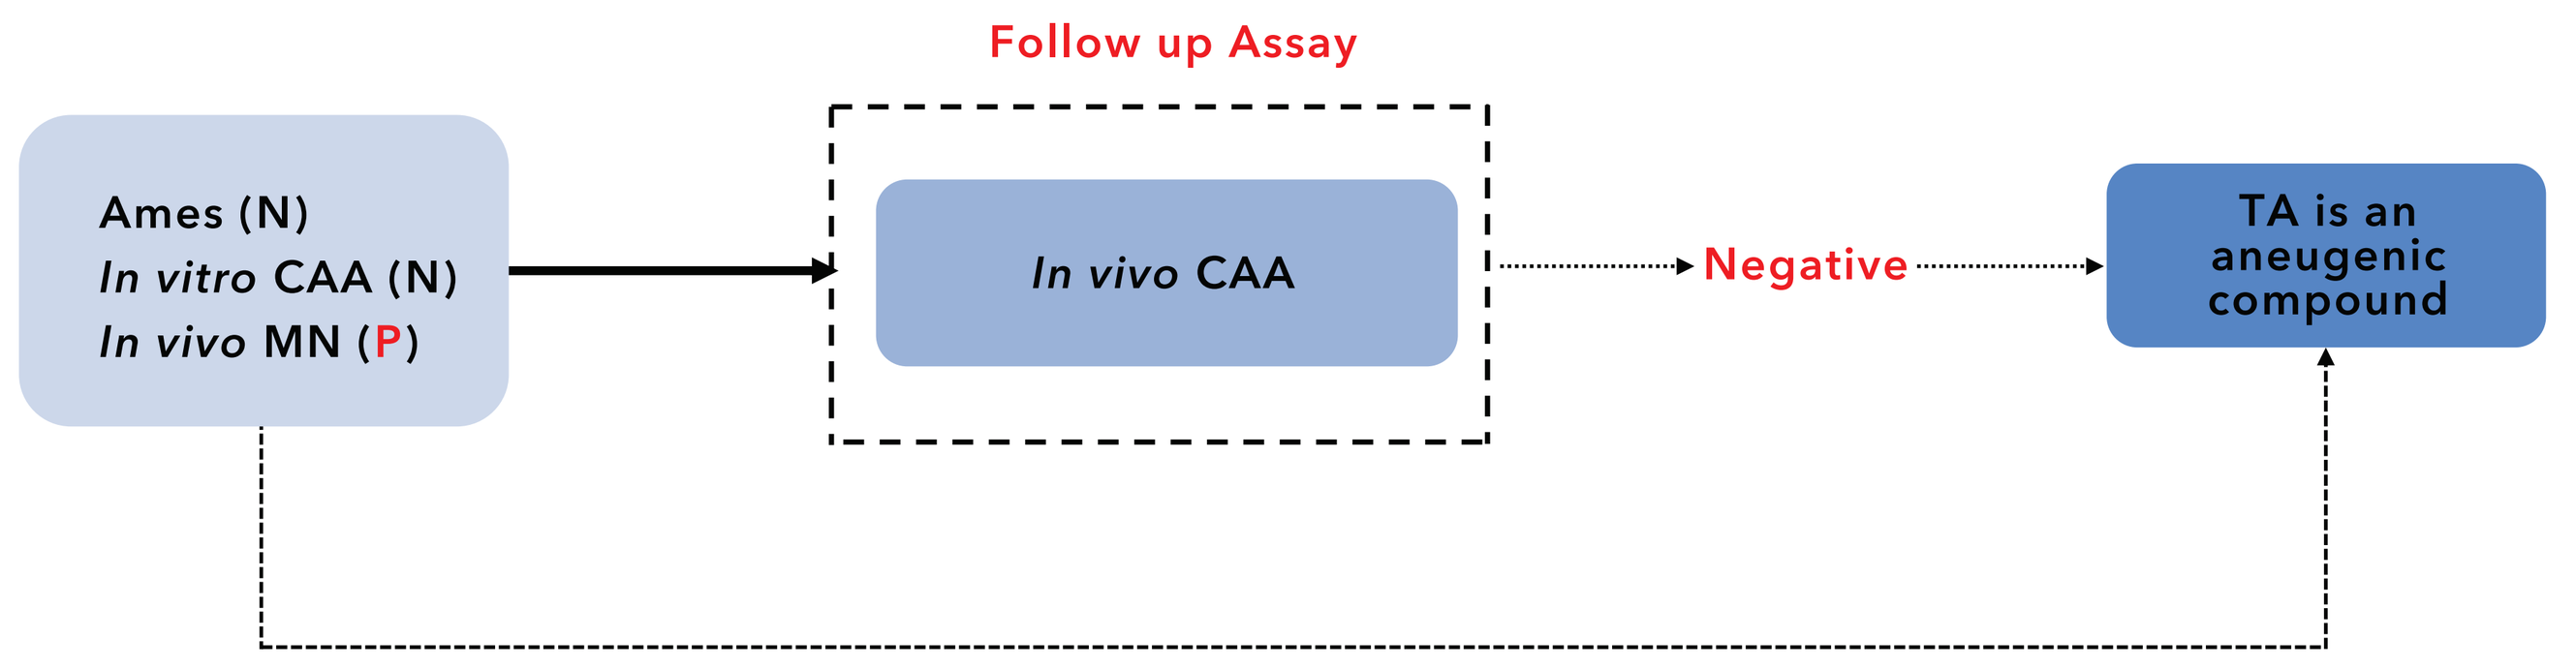
Follow up Assay
Ames (N)
In vitro CAA (N)
In vivo MN (P)
In vivo CAA
Negative
TA is an aneugenic compound

In the last case, Ames assay, *in vitro* CAA assay and *in vivo* MN assay were performed, except for Ames assay, both *in vitro* CAA and *in vivo* MN assay were evaluated as positive, a background research revealed that the target of test article is DNA damage repair related, the results were considered as on target effect, so no further follow-up assays were performed under this case.

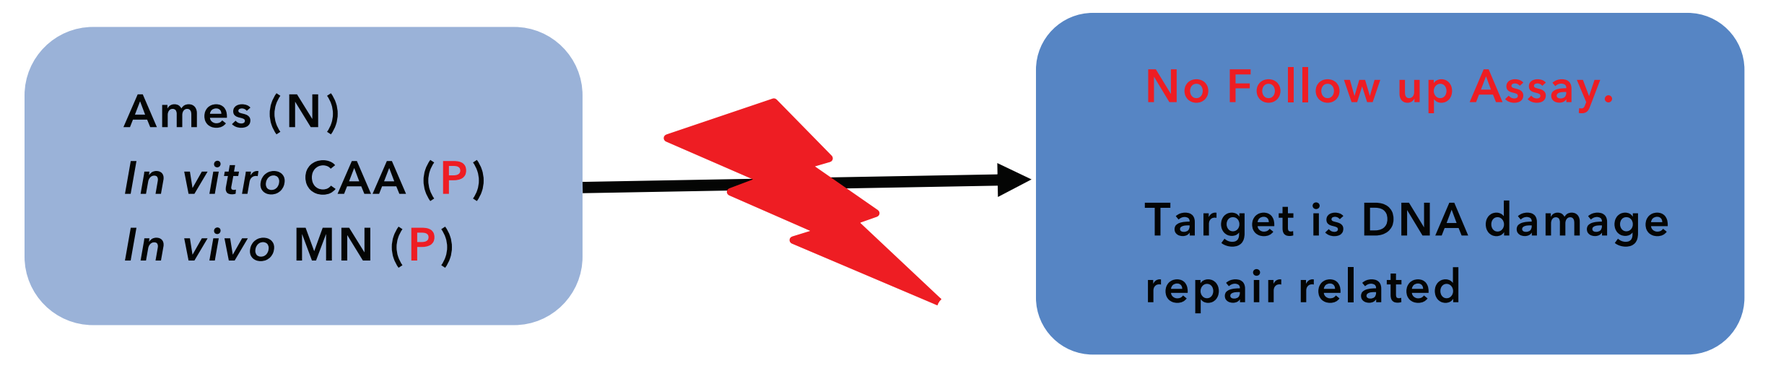
Ames (N)
In vitro CAA (P)
In vivo MN (P)
No Follow up Assay.
Target is DNA damage repair related

Conclusion

In summary, combinational *in vivo* and *in vitro* assays should be used flexibly for the purpose of mechanistic research (although maybe indirect evidence) and overall evaluation.

Some other methods, such as Fluorescence in Situ Hybridization can also be used for that purpose per the ICH guideline.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou).