Comparison of Systemic Exposure in Pregnant and Non-Pregnant New Zealand White Rabbits

Lei Guo, Anting Xu, Shuijin Yang, Sue McPherson
WuXi AppTec, Suzhou, China

Abstract: 3868

Introduction

It is well known that pregnancy could theoretically induce changes in drug Pharmacokinetic (PK) or Toxicokinetic (TK) profiles by affecting the absorption, distribution, metabolism or renal excretion. However, there is little or no information available regarding whether pregnant animals have altered PK/TK as compared to non-pregnant animals. The New Zealand White Rabbit is widely used in preclinical general toxicology studies and Embryo-Fetal Developmental (EFD) studies and there have lots of available TK data in pregnant and non- pregnant rabbits to be analyzed.

Materials and Methods

Fourteen small molecular projects (with non-pregnant single dose PK or repeat dose range finding studies in female rabbits) performed at the Testing Facility between 2021-2024 were retrospectively reviewed to evaluate the potential differences of systemic exposure between pregnant and non-pregnant rabbits by calculating the $AUC_{0\text{-}24h}$, C_0 and/or C_{max} ratios. Dosing route was oral gavage, subcutaneous injection or intravenous administration. There were 6 projects with the same dose levels and 8 projects with different dose levels in repeated dose range findings/single dose PK studies and EFD studies. For the projects with different dose levels, the dose normalized $AUC_{0\text{-}24h}$ and C_{max} or C_0 ($AUC_{0\text{-}24h}/D$ and C_{max} or C_0/D) were calculated first and then used for the systemic exposure comparison of first dosing. Differences of more than 0.5 fold and less than 2 fold in TK parameters of both $AUC_{0\text{-}24h}$ and C_{max} or C_0 were not considered to indicate significant difference.

Results

The comparison data showed there was only 1 project that the pregnant rabbits had higher systemic exposure than non-pregnant rabbits (the C_{max} ratio and $AUC_{0\text{-}24h}$ ratios was 3.5 and 4.8 respectively). No significant differences in systemic exposure (in terms of $AUC_{0\text{-}24h}$, and C_0/C_{max} ratios) for all remaining 13 projects.

Table 1 Systemic Exposure Comparison

Data List	C_0 and/or C_{max} Ratios	$AUC_{0\text{-}24h}$ Ratios
1*	0.3	0.6
2	0.4~1.0	0.4~0.9
3	0.7~1.1	0.7~1.0
4*	0.8~1.2	0.9~1.4
5	1.5	1.3
6*	0.8~0.9	0.8~0.9
7	3.5	4.8
8*	0.8	1.0
9*	0.7~0.9	0.7~0.9
10	1.6	1.5
11*	0.7~1.3	0.8~1.1
12	0.7	0.7
13*	1.2~1.5	1.9~2.1
14*	0.4~0.6	0.8~1.2

* Projects with different dose levels in PK/repeated dose range finding studies and EFD studies.

Conclusion

In conclusion, for most of drug candidates, there were no significant differences in systemic exposure between the two different physiological status (pregnancy vs non-pregnancy) in rabbits. However, it is still recommended to perform dose range finding study in pregnant animals prior to initiation of definitive EFD study to avoid unexpected maternal toxicity, embryotoxicity or fetotoxicity due to potential higher systemic exposure induced by pregnancy.

Acknowledgements

The authors would like to acknowledge the contributions made by the team at WuXi AppTec.