Evaluation of the Impact of the Use of Slide Imaging in the Histopathological Evaluation of Toxicology Studies

Jiajia Zhang; Kathryn Gropp; Long Jing; Zhimei Wang; Sue McPherson; Xixing Zhao; Tiansheng Zhou
WuXi AppTec

Abstract: 4239

Background and Purpose

In today's environment time is critical and key to getting drugs to the market/patients. The turnaround time of data at each stage of the drug development program will trigger the next phase of the development as well as the setting of dose levels for future studies. The last set of data to be evaluated is often the histopathological evaluation of the slides. Typically, the glass slides will be sent to the pathologist for evaluation, this may be a short distance of across the hallway or across the world. This study was done to see if there was any difference in the time or results, it took for the evaluation and reporting when evaluating slides from glass and using a scanner (Aperio GT 450, Picture 1).

Picture 1 Aperio GT 450 Digital Slide Scanner

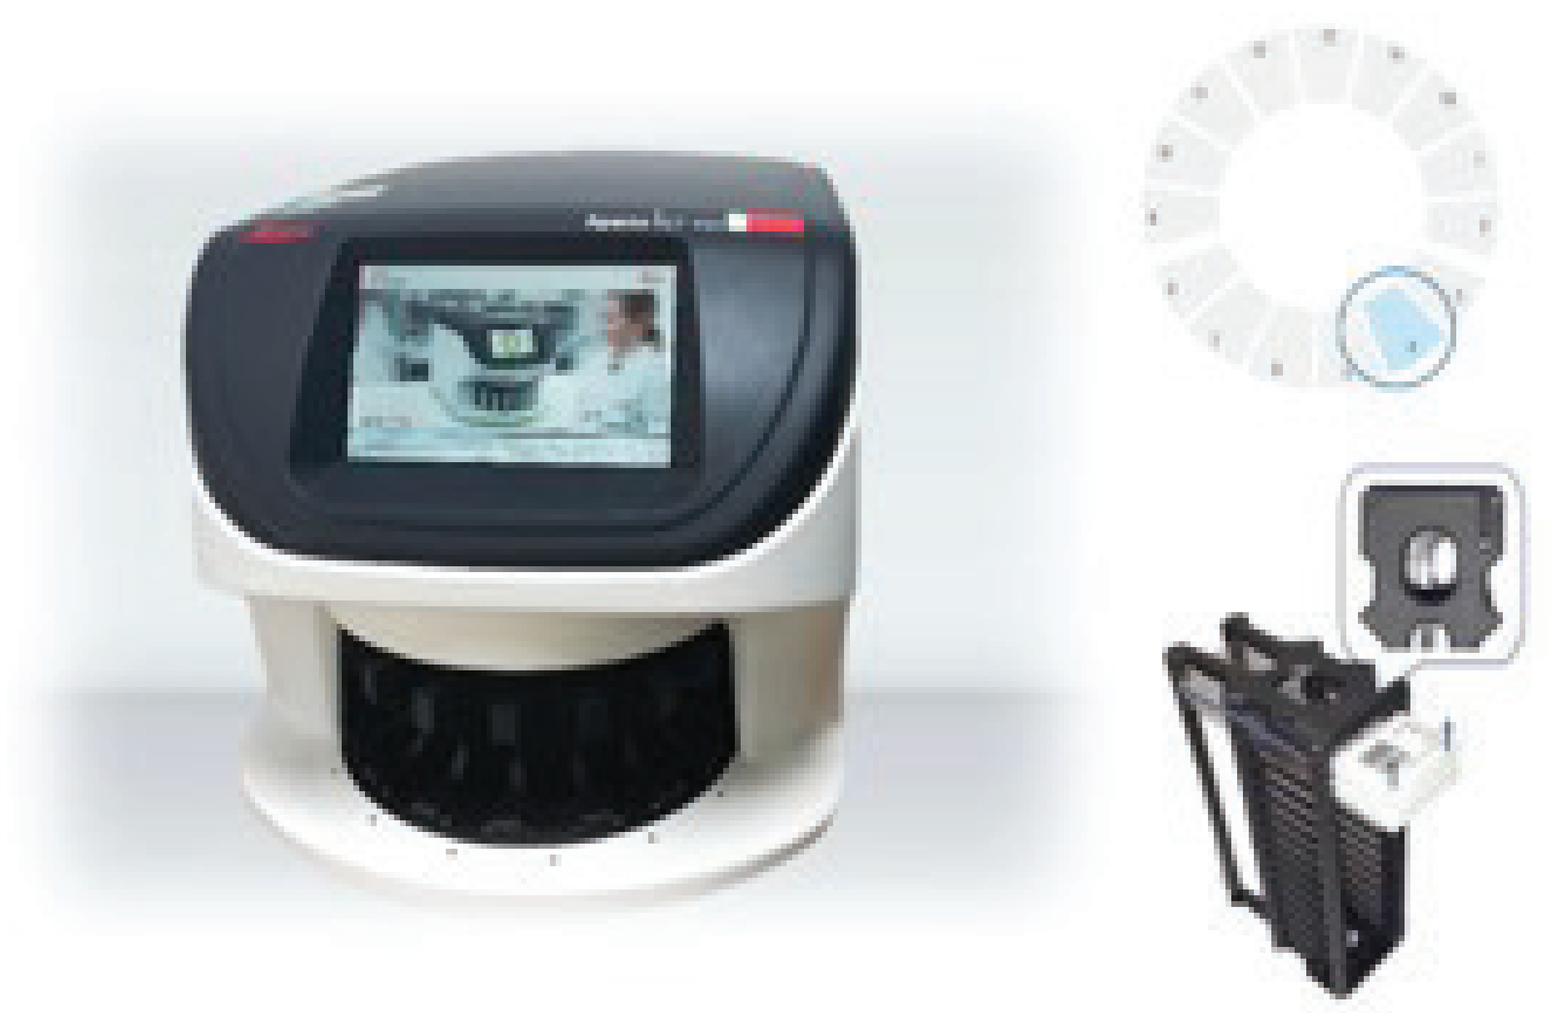

Methods

Four hundred and fifty slides from three validation studies (Table 1), including mice, rats, and dogs were used. Glass slides were prepared and evaluated as well as scanned to the pathologist. The time from necropsy to reading was measured as well as for reading/evaluation for both methods. In addition, the accuracy of the evaluations was also compared. One US pathologist (DACVP) completed the diagnosis on e-slide (electronic slides) through both eSlideManager online and slides saved in a hard drive. Three pathologists in China (two DCCVP, and one other pathologist with over 10 years working experience) made their diagnoses using glass slides, eSlideManager online, and slides saved in hard drive, respectively (Table 2).

Table 1 Validation Studies Information

Validation Studies	Study Type	Slides Number	Selection of Slides
Study 1	Oral dose phototoxicity study in Long-Evans rats	260 Slides	All slides from this study
Study 2	A single-dose intraperitoneal injection toxicity study in the mouse followed by a 14-day observation period	140 Slides	All slides from this study
Study 3	A T-cell dependent antibody response validation study in beagle dogs.	50 Slides	Part slides from this study

Table 2 Glass/Digital Slides Assigned for Pathologist

Sites	Glass Slides	Digital Slides Saved in the Hard Drive Reading by Software Download from Leica Official Website	Digital Slides Online Reading
SZ (Suzhou, China)	SZ local Pathologist	SZ local Pathologist	SZ local Pathologist
US (United States of America)	Not ship to US for review	US Pathologist (DACVP)	US Pathologist (DACVP)

Results

The results showed that was no significant difference amongst all the diagnoses from four pathologists who had their own separate diagnoses on glass slides and e-slides. E-Slides can clearly display in Aperio ImageScope as Image 1, 2, 3, the maximum objective lens magnification can be 40X that same as the microscope, the lesion can be clearly observed in Aperio ImageScope.

The overall time for glass slide and e-slides evaluation was different; reading/evaluation online was quicker. Comparing transfer the e-slide to pathologists abroad with glass slide delivery, there were time savings on preparation for documentation, packing materials, transportation, customs clearance and avoidance of possible damage or loss of glass slides during the shipping process.

Conclusion

Scanning is a valuable tool and will become more used in the future following wider acceptance to this medium.

Image 1 Overview of the liver e-slide in Aperio ImageScope

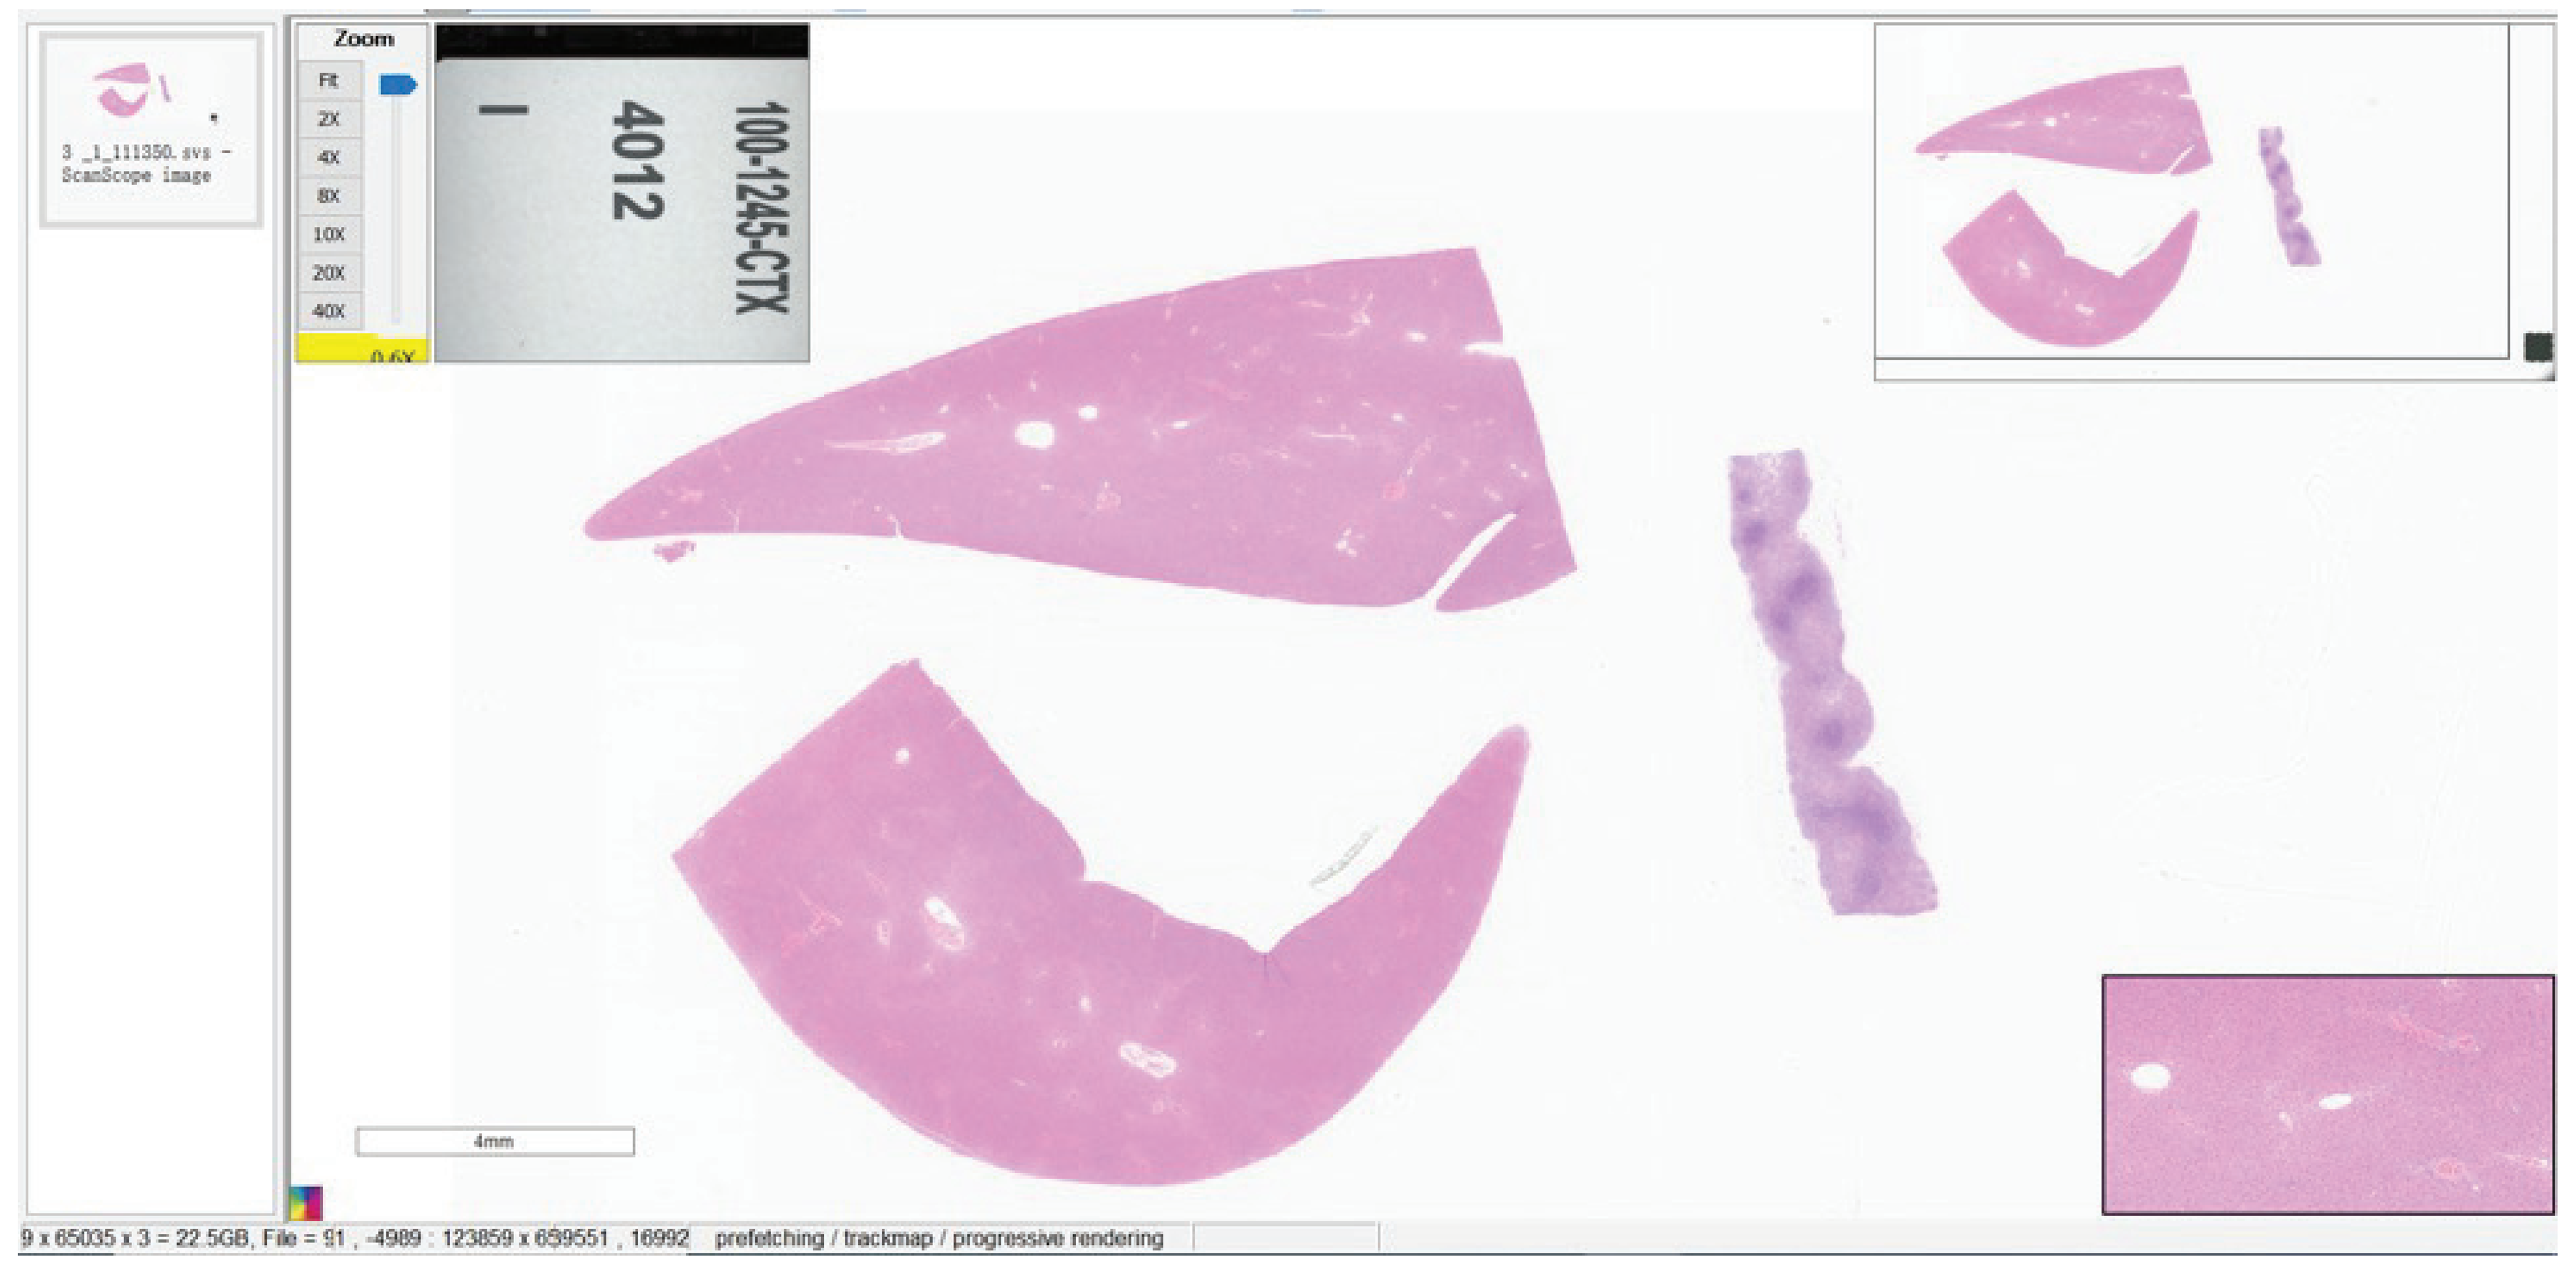
3 _1_111350.svs - ScanScope image
Zoom
Fit
2X
4X
8X
10X
20X
40X
4012
100-1245-CTX
4mm

Image 2 Control Group, Male, Liver, Normal. (Objective lens magnification=20 X; HE)

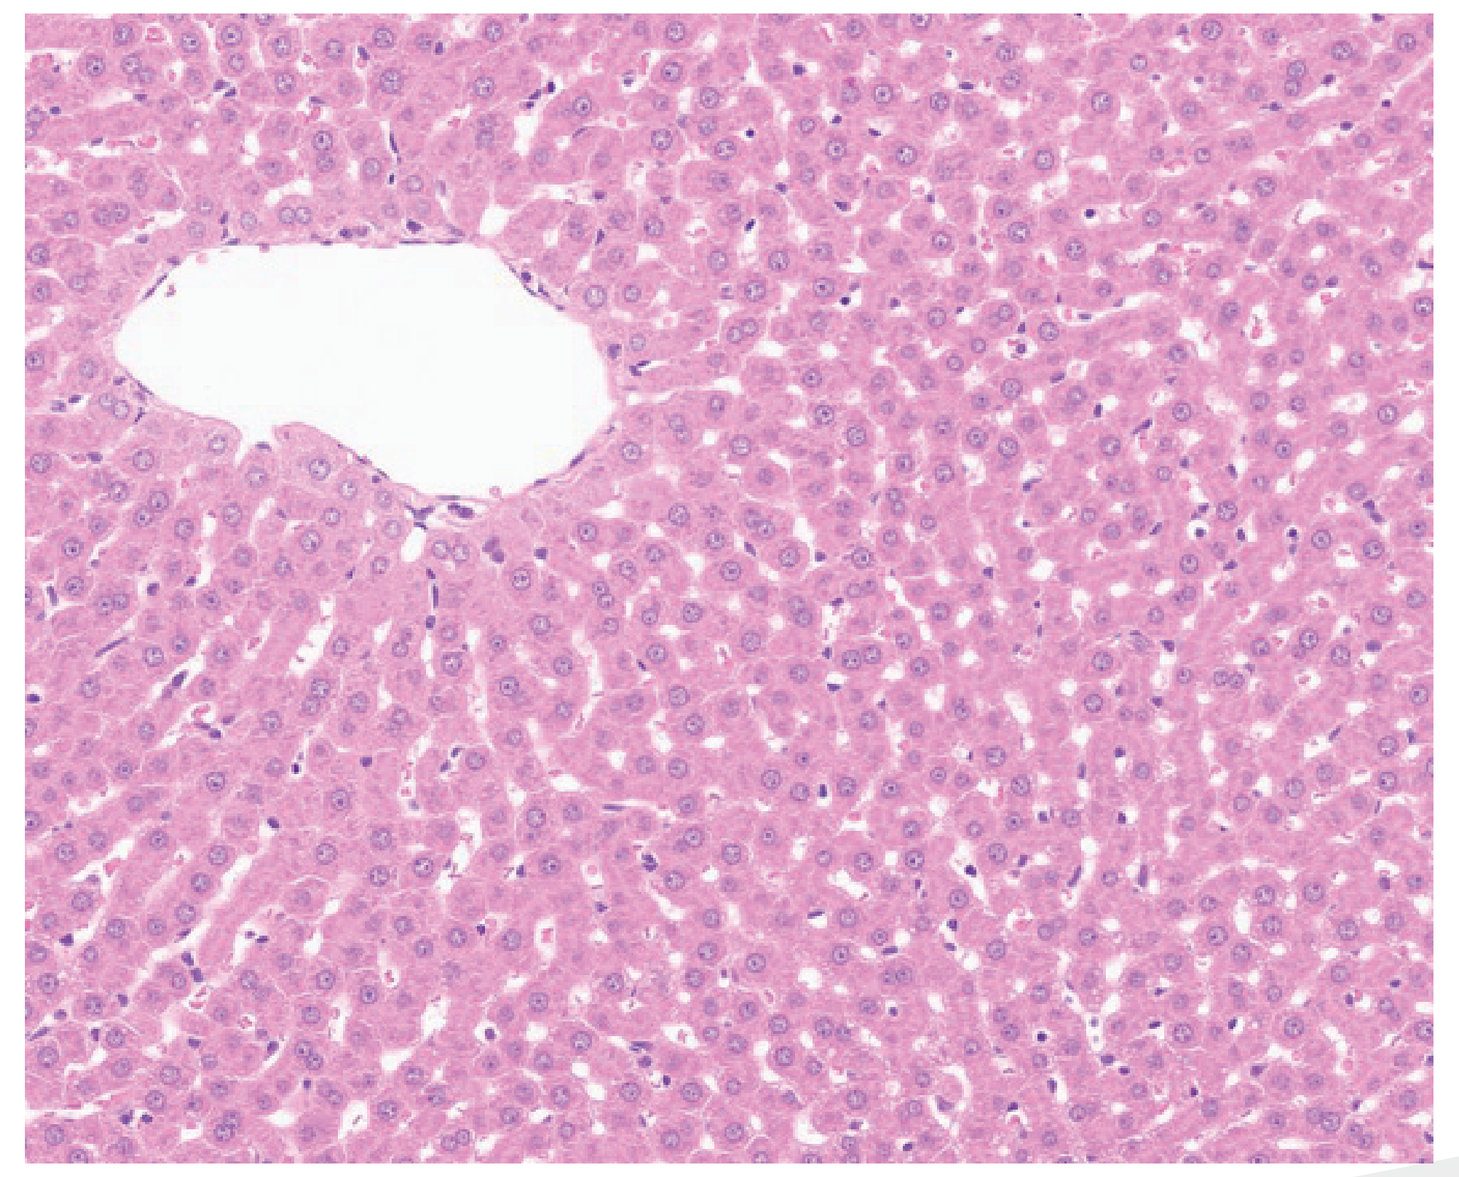

Image 3 High Dose Group, Male, Liver; Necrosis, Single Cell; Centrilobular, Hepatocyte (blue arrow) (Objective lens magnification=20 X; HE)

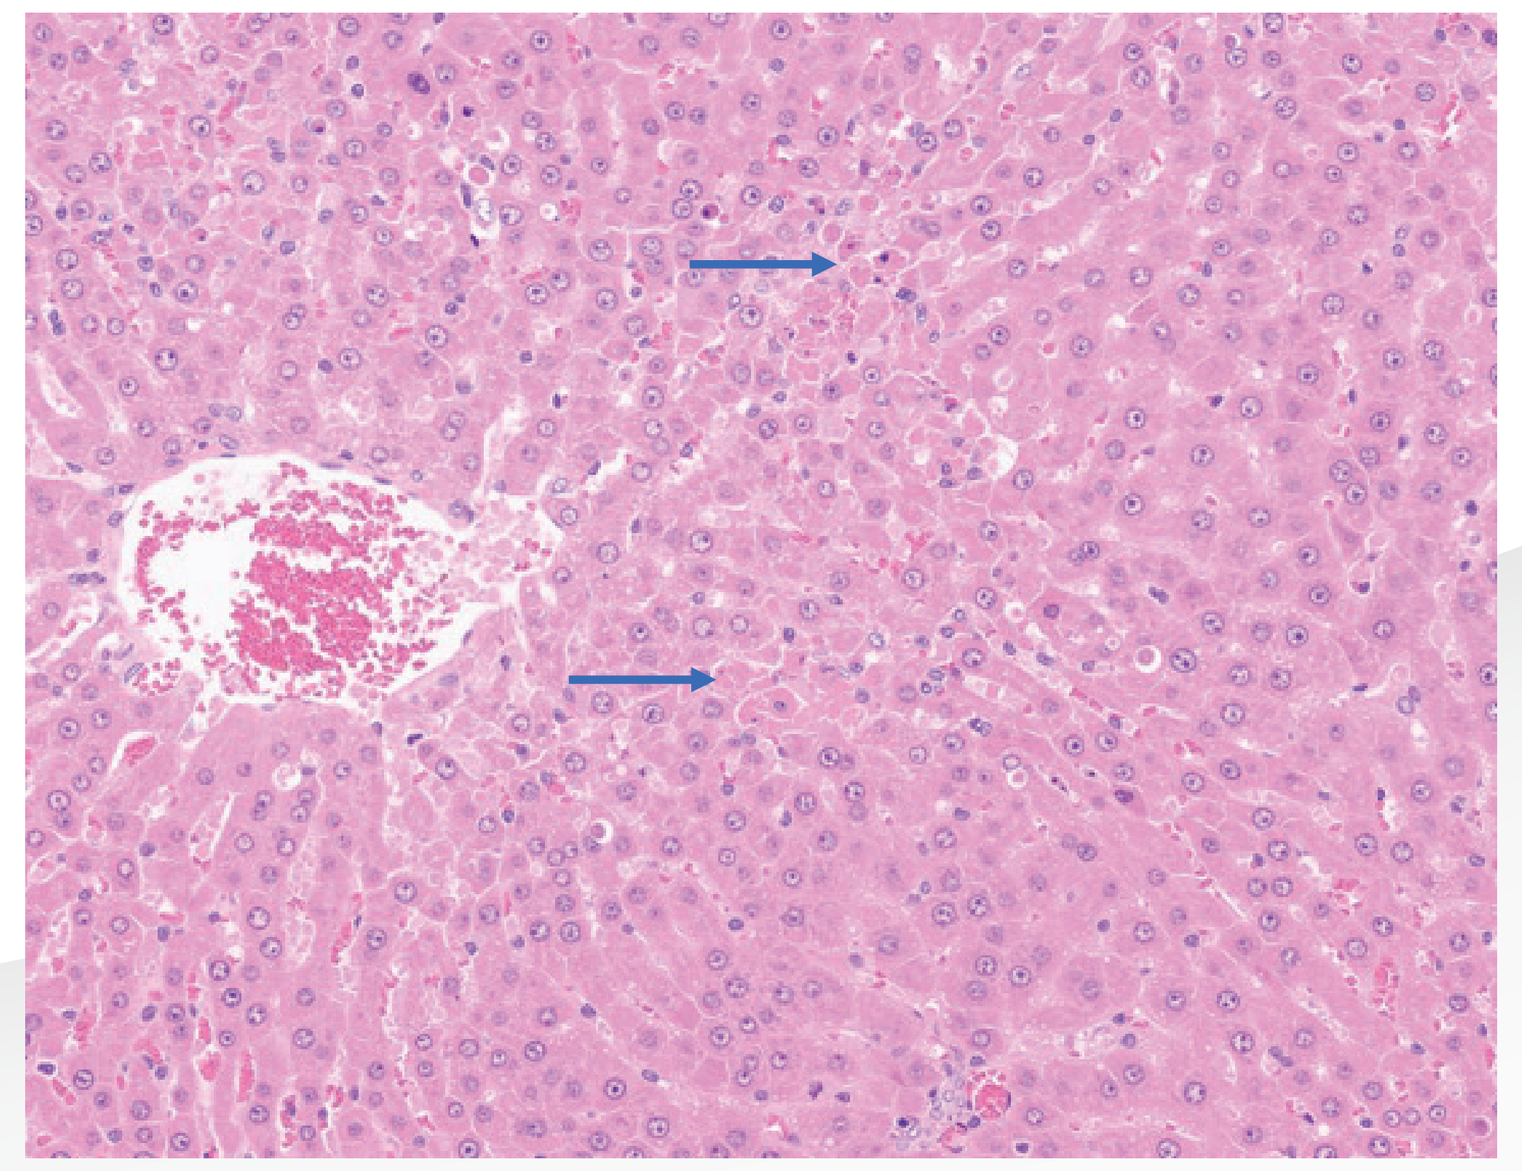

Acknowledgements

The authors would like to thank the technicians and pathology team at WuXi AppTec who provided their valuable support