Method Validation for the Reactive Oxygen Species (ROS) Assay for Photoreactivity

Mengqi Zhang; Xiaohui Zhu; Yaohua Zhang; Liwen Gao; Xiaoxia Li; Millie Chen
WuXi AppTec, Suzhou, China

药明康德 WuXi AppTec

Abstract: 4107

Background and Purpose

As an in chemico screening tool for evaluating the photoreactivity of pharmaceuticals, the Reactive Oxygen Species (ROS) assay is recommended by the ICH guideline S10 for its advantages in high sensitivity in predicting direct in vivo phototoxicants. Since the introduction of the ICH guidance, the ROS assay has been used quite extensively within the pharmaceutical industry. This abstract presents the internal validation result of the ROS assay using 17 chemicals following recommendations study design of the OECD Test Guideline (TG) 495.

Methods and Materials

Three 96-well plates were used to test 17 test articles using a solar simulator (SOL500, Dr. Hönle AG) and a UV meter (p/n 086001, Dr. Hönle AG), the mixture was irradiated at 20°C to 29°C for 60 minutes (±5 minutes). The light from the solar simulator was filtered through an H2 filter, and the intensity of the light passing through the quartz plate cover was 4.6±0.3 mW/cm². The photoreactivity of the compound was assessed by measuring the bleaching of p-nitrosodimethylaniline (RNO) to detect singlet oxygen (SO) generation (decrease in absorbance of RNO at 440 nm) and the reduction of nitro blue tetrazolium (NBT) to detect superoxide anion (SA) generation (increase in absorbance of NBT at 560 nm) using spectrophotometry.

Results

The negative control (sulibenzone) and the positive control (quinine hydrochloride dihydrate) results in this test were comparable to those ecommended in guidelines and reported in the literature, demonstrating the validity of the test. For Octyl Salicylate, p-Aminobenzoic Acid, Benzocaine, and Erythromycin, no precipitation or color interference was observed at 20 µM (for Octyl Salicylate only) or 200 µM in DMSO. The SO values for these 4 compounds were 0.50, -0.90, 7.87, and 0.43, respectively, and the SA values were -14.20, -11.77, 7.97, and 2.50, respectively, all indicating non-photoreactive. For Fenofibrate, Doxycycline hydrochloride, Norfloxacin, 8-Methoxy Psoralen, Acridine, Chlorpromazine hydrochloride, Diclofenac, Furosemide, Ketoprofen, Nalidixic Acid, Omeprazole, and Promethazine Hydrochloride, no precipitation or color interference was observed at 20 µM (for Fenofibrate only) or 200 µM in DMSO. The SO values for these 12 compounds were 204.30, 140.60, 35.37, 184.37, -39.50, 231.13, 231.63, 118.67, 133.83, -102.03, 51.93 and 78.33, respectively, and the SA values were 380.27, 94.77, 81.57, 155.60, 74.90, 300.07, 47.40, 79.77, 210.03, 105.47, 36.53 and -9.30, respectively, all indicating photoreactive. For L-Histidine, no precipitation or color interference was observed at 200 µM in NaPB, the SO value was 0.53, and the SA value was 30.13, indicating weakly photoreactive. The obtained results were comparable with the values listed in OECD TG495.

Table 1 The ROS detection values for 17 proficiency substances and negative/positive controls using solar simulators and the OECD acceptable range

Chemical Name	Concentration	WuXi Result		OECD Acceptance Range		Conclusion	Compound Category
		SO	SA	SO	SA		
Benzocaine	200 µM	-0.90	-11.77	-7 to 9	-7 to 17	non-photoreactive	non-phototoxic compound
Erythromycin	200 µM	7.87	7.97	-15 to 11	-9 to 21	non-photoreactive	non-phototoxic compound
8-Methoxy Psoralen	200 µM	35.37	81.57	31 to 137	0 to 126	photoreactive	phototoxic compound
Furosemide	200 µM	231.63	47.40	31 to 225	-7 to 109	photoreactive	phototoxic compound
Ketoprofen	200 µM	118.67	79.77	120 to 346	77 to 151	photoreactive	phototoxic compound
Nalidixic Acid	200 µM	133.83	210.03	54 to 246	88 to 470	photoreactive	phototoxic compound
Fenofibrate	20 µM	78.33	-9.30	77 to 203	-31 to 11	photoreactive	phototoxic compound
Omeprazole	200 µM	-102.03	105.47	-221 to 103	30 to 216	photoreactive	phototoxic compound
Promethazine Hydrochloride	200 µM	51.93	36.53	20 to 168	-3 to 77	photoreactive	phototoxic compound
Acridine	200 µM	184.37	155.60	182 to 328	121 to 243	photoreactive	phototoxic compound
p-Aminobenzoic Acid	200 µM	0.50	-14.20	-8 to 12	-11 to 7	non-photoreactive	non-phototoxic compound
L-Histidine	200 µM	0.53	30.13	-8 to 12	8 to 120	weakly photoreactive	non-phototoxic compound
Octyl Salicylate	20 µM	0.43	2.50	-5 to 11	-8 to 20	non-photoreactive	non-phototoxic compound
Doxycycline Hydrochloride	200 µM	204.30	380.27	115 to 429	230 to 468	photoreactive	phototoxic compound
Norfloxacin	200 µM	140.60	94.77	131 to 271	57 to 161	photoreactive	phototoxic compound
Diclofenac	200 µM	231.13	300.07	34 to 416	47 to 437	photoreactive	phototoxic compound
Chlorpromazine Hydrochloride	200 µM	-39.50	74.90	-56 to 70	66 to 106	photoreactive	phototoxic compound
Sulisobenzone	200 µM	2.00	-20.57	-9 to 11	-20 to 2	non-photoreactive	non-phototoxic compound
Quinine Hydrochloride	200 µM	379.14	245.03	319 to 583	193 to 385	photoreactive	phototoxic compound

Conclusion

This test system has the capability to identify photoreactive substances from non-photoreactive substances, demonstrating the ROS assay for safety evaluation is well-validated in Genetic and *In Vitro* Toxicology laboratory of WuXi AppTec.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou).