Spontaneous Lesions Associated with Test Article-Unrelated Deaths in Repeated Dose Mouse Studies

Junli Zhao, Pan L, McPherson S, Tiansheng Zhou
WuXi AppTec, Suzhou, China

药明康德
WuXi AppTec

Abstract: 3286

Introduction

Distinguishing test article-related and –unrelated deaths is critical in interpreting nonclinical study data as deaths of unknown cause that may be test article-related warrant further understanding of the mechanism of the drug candidate and bring more challenges to risk assessment and ultimately regulatory submission and approval. Conventionally and scientifically, the cause of the deaths occurred in animal studies is judged by comprehensive considerations including comparison with the concurrent control, consistency of dose dependency (more applicable to small molecule chemical drugs), time and incidence of deaths, reference to background pathology lesions of the specific species, and comparison of findings with animals that survived to scheduled termination. The purpose of this investigation is to analyze the spontaneous lesions that resulted in test article-unrelated deaths in mouse studies conducted at the facility and provide references as background data. A retrospective analysis was performed on approximately 50 repeated dose mouse studies of 4-week duration or longer completed so far.

Methods

Repeated dose mouse studies of 4-week duration or longer were reviewed retrospectively. The animals used on studies were CD-1, C57BL/6J, and BalB/c mouse, sourced from BioLASCO Taiwan Co., Ltd. and Vital River Laboratory Animal Technology Co., Ltd. (Beijing and Zhejiang).

For each study, the number of animals, number of unscheduled deaths that were considered related to spontaneous lesions and unrelated to the test article, and the detailed in-life and pathology findings associated with the deaths were tabulated. These data were further analyzed and categorized based on the cause of deaths identified and compared with published literatures.

Results

- Spontaneous lesions that resulted in treatment unrelated deaths were noted in approximately 48.7% studies and represented approximately 4.20‰ of total animals used on the studies, as shown in Table 1.
- The mortality rate was higher in 26-week study than 13-week and 28-day studies being 75%, 58.7%, and 25% of the total studies at each period, respectively.
- Among these, about half of the incidences were undetermined (due to multiple organs appeared autolysis, or TK animals without histopathology examination, or no obvious findings in gross and microscopic evaluation).
- Spontaneous tumors including lymphosarcoma, histiocytic sarcoma, adenocarcinoma, and other solid tumors (masses) were noted as early as of age Week 14 and e reported elsewhere[1, 2] as spontaneous tumors that could occur in laboratory mice.
- The remaining incidences included urinary obstruction, sepsis, and bacterial infection. These and associated animal information were summarized in Table 2. Other findings listed in Table 2 were not commonly observed and may represent incidental cases.

Table 1

Species	Vendor	Number of Animals Used on Studies	Number of Test Article-Unrelated Deaths with Spontaneous Lesions
CD-1	Vital River	16932	89
	Biolasco	548	0
C57BL/6J	Vital River	2792	0
BalB/c	Vital River	1174	1

Biolasco: BioLASCO Taiwan Co., Ltd.; Vital River: Vital River Laboratory Animal Technology Co., Ltd. (Beijing and Zhejiang)

Table 2

Cause of Death (Number)	Species	Vendor	Sex	Age at Death (Week)	Mode of Death	In-Life Findings	Pathology Findings	Remarks	Dosing Route
Undetermined (59)	CD-1	Vital River	M and F	8 to 31	EU/FD	Slight thinness, decreased activity, and labored breathing, Hunched posture, discolored skin (pale) of bilateral ears, unkempt, red/tan stool, cold to touch, and slight thin	No noteworthy findings associated with mortality	1. Multiple organs appeared autolysis 2. TK animal. Microscopic evaluation not performed	IV bolus, SC, and PO
Fight or Injury (8)	Balb/c (1)	Vital River	M	9 to 11	EU/FD	Scab on the dorsal thorax, loss of left eye	None	None	PO
Reflux (4)	CD-1	Vital River	M/F	9 to 12	EU/FD	Abnormal respiratory sounds, cold to touch, slightly thin, decreased activity, and unkempt	Erosion/ulceration in the olfactory epithelium; multifocal, moderate neutrophilic inflammation in the lumen	None	PO
Hyaline Glomerulopathy (1)	CD-1	Vital River	F	2²	EU	Hunched posture and slight thinness	Marked hyaline glomerulopathy; Moderate tubule Degeneration;	None	SC
Mass (2)	CD-1	Vital River	F	19	EU	Abnormal gait, bilateral hindlimb atonia and were worsened; Hard mass of right hindlimb, discolored skin (pale) of bilateral ears, cold to touch, decreased activity, increased respiratory rate, and labored breathing	Lymphosarcoma in various tissues	None	PO
0Neutrophilic inflammation in the meninges of brain (1)	CD-1	Vital River	M	10	EU	Decreased activity, unkempt, prostration, and atonia of bilateral hindlimbs	Presence of neutrophilis in the meninges,vascular thrombi and fibrinoid material and neutrophils in the walls of the meningeal vessels and in the vessels adjacent neuropil	None	PO
Bacterial Infection (3)	CD-1	Vital River	F	CD-1	EU	None	Multifocal to coalescing scabs on the dorsal abdomen that correlated to ulceration of the epidermis, subcutaneous inflammation, and bacterial colonies	None	PO
Sepsis (6)	CD-1	Vital River	F	13	EU/FD	Decreased activity, thin, cold to touch, piloerection, and pale skin	Inflammation or inflammation/ necrosis in multiple organs with bacterial colonies	None	PO
Obstructive Uropathy (4)	CD-1	Vital River	M	18-21	EU/FD	Cold to touch, activity decreased, skin discolored (pale, both ears, both forelimbs, and both hind limbs, and tail), partial loss of prepuce	Bilateral dilation of the renal pelvis, ectasia and mild to moderate degeneration/regeneration and necrosis of the renal tubules, and degeneration/regeneration of the smooth muscle in the urinary bladder, prostate, and seminal vesicles	None	PO
Long-established ulceration of the tail and perineal area jejunum, with peritonitis (1)	CD-1	Vital River	M	8	EU	Cold to touch, decreased activity, atonia in bilateral hindlimbs, unkempt, discolored skin in tail (purple)	Inflammation of the rectum and unilateral nephropathy with bladder hemorrhage	None	PO
Histiocytic sarcoma in the heart (1)	CD-1	Vital River	CD-1	10	FD	None	Histiocytic Sarcoma in heart	None	PO

EU: Euthanized; FD: Found dead; M: Male; F: Female; TK: Toxicokinetics. PO: Oral gavage; SC: Subcutaneous injection. Vital River: Vital River Laboratory Animal Technology Co., Ltd.

Acknowledgements

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec.

References

1. Ancuta A, Michela L, Lan T, Radu A, Sydney M, and Vasanthi Mowat. Causes of Mortality and Profile of Spontaneous Tumors in Young CD-1 Mice. Toxicologic Pathology. 1–11, 2022.
2. Son WC, Gopinath C. Early occurrence of spontaneous tumors in CD-1 mice and Sprague–Dawley rats. Toxicol Pathol. 2004;32(4):371-374.