Spontaneous Ocular Lesions Observed in Laboratory Animals

Hongjing Cheng, Jonathan Gao, William M. Harrison, Yongdong Zhou
Toxicology, WuXi AppTec (Suzhou)

Abstract: 4162

Objective

To collect and analyze the data of spontaneous ocular lesions observed in laboratory animals at WuXi AppTec (Suzhou).

Methods

Ophthalmologic Examination (OE)

- Binocular indirect ophthalmoscope for fundus examination;
- Slit lamp microscopy for anterior segment of the eye.

Data Collection

- OE records for laboratory animals before experiments from 2008 to 2023 were collected through the Provantis system at WuXi AppTec (Suzhou) Co., Ltd. The animals included mice, rats, rabbits, dogs, mini-pigs, and non-human primates (NHPs).

Data Analysis

- Raw data from different documents were filtered by using the built-in query function in Google Sheets.
- A Google Apps Script was written to handle the multiple-source documents.
- Incidences of spontaneous ocular lesions in laboratory animals were calculated.

Results

Data were collected and analyzed from 50,484 Institute of Cancer Research (ICR) mice, 12,731 C57BL/6J mice, 692,159 Sprague-Dawley (SD) rats, 31,241 Wistar rats, 6,128 Newland rabbits, 6,663 Dutch Belted rabbits, 102,193 Beagle dogs, 5,295 Bama pigs, 75,763 Cynomolgus and 848 Rhesus .

Incidence of Spontaneous Ocular Lesions in Mice

- The incidence of lens opacity (primarily medium opacity), cornea opacity (primarily pinpoint opacity), eccentric pupil, and persistent hyaloid artery (PHA) in ICR mice was 23.13%, 8.93%, 6.09%, and 0.92%, respectively.
- The incidence of lens opacity (primarily pinpoint opacity), retinal color/pigmentation variation (RCPV), vitreous opacity/flare, and cornea opacity (primarily small opacity) in C57BL/6J mice was 75.66%, 5.89%, 1.46%, and 1.34%, respectively.

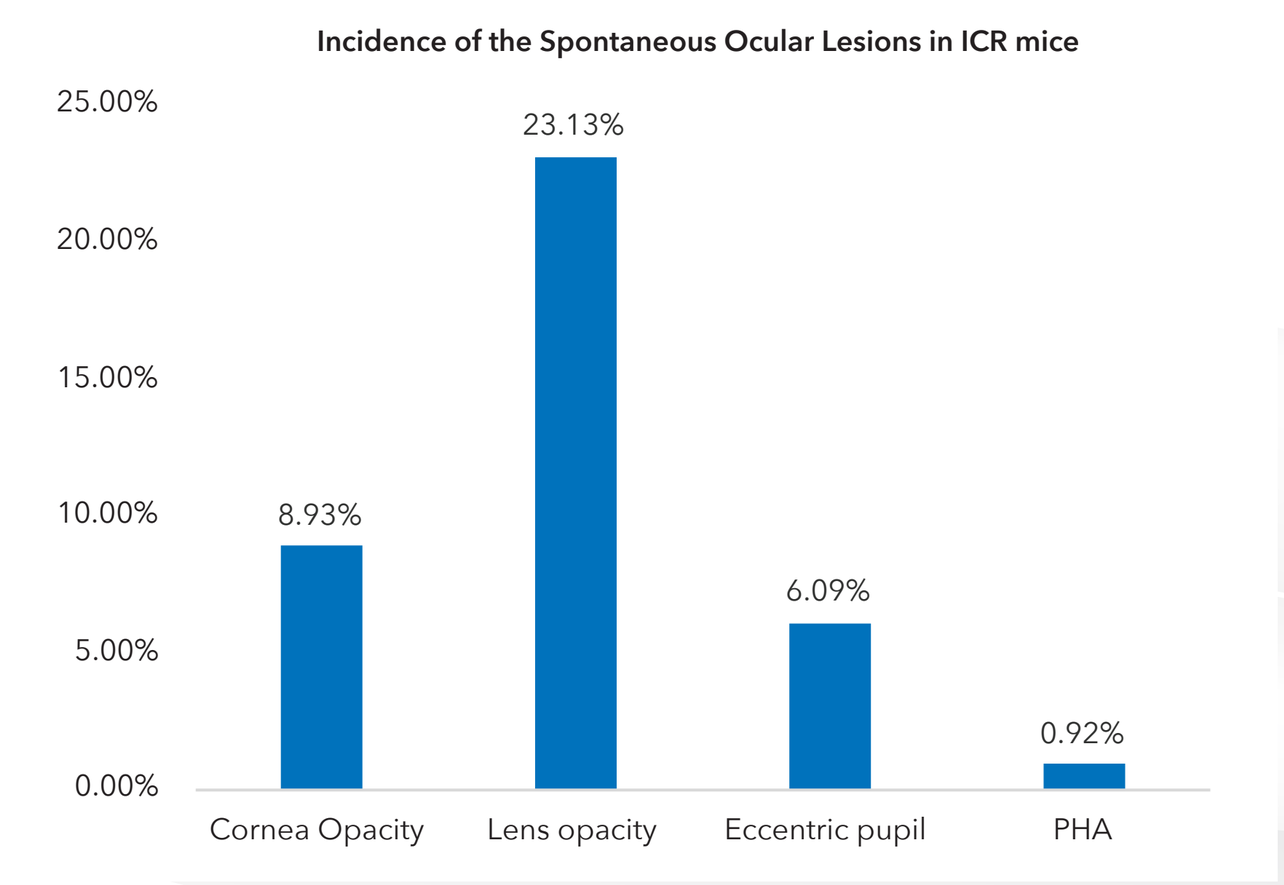
Incidence of the Spontaneous Ocular Lesions in ICR mice
8.93%
23.13%
6.09%
0.92%
Cornea Opacity
Lens opacity
Eccentric pupil
PHA

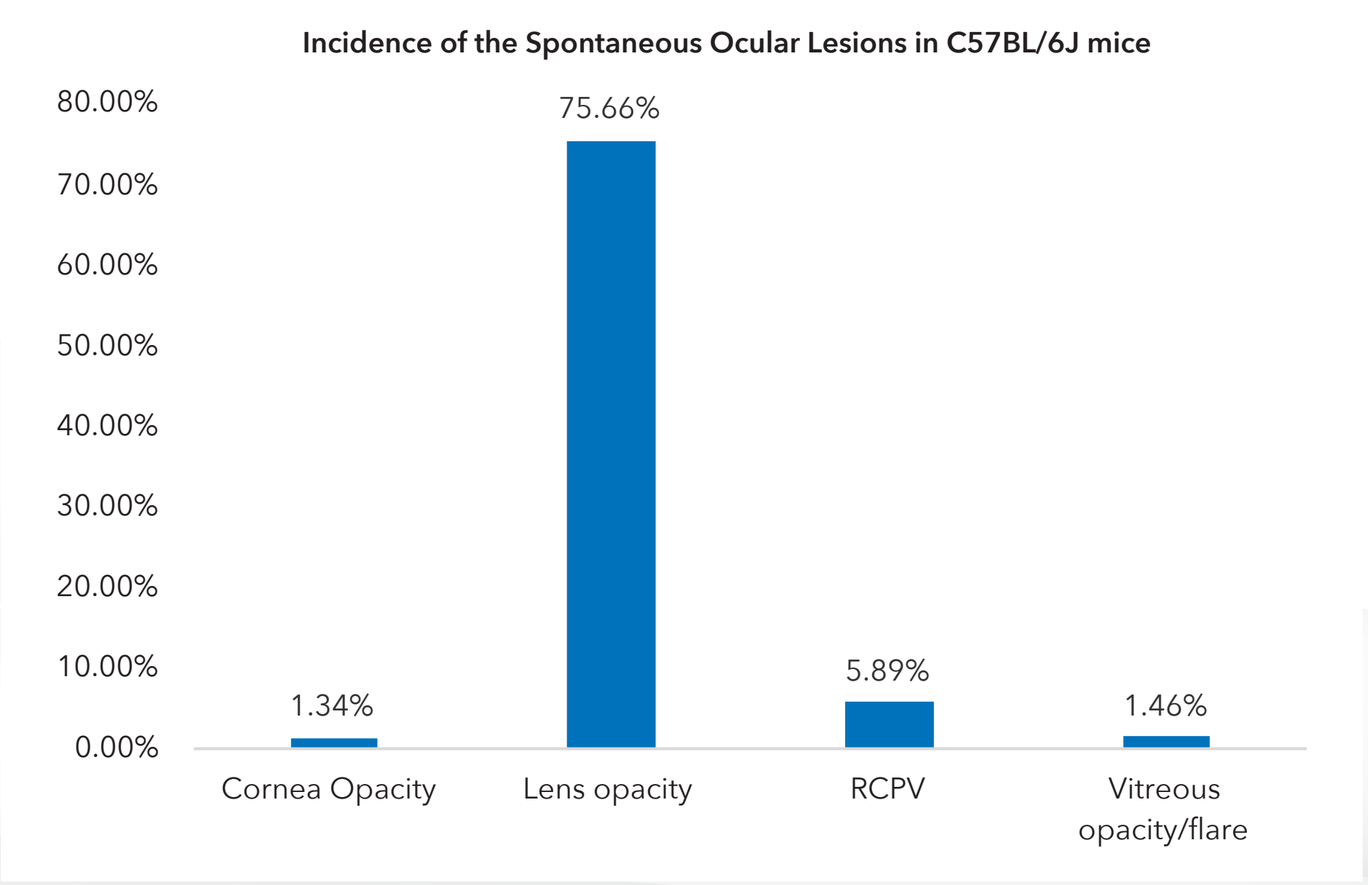
Incidence of the Spontaneous Ocular Lesions in C57BL/6J mice
1.34%
75.66%
5.89%
1.46%
Cornea Opacity
Lens opacity
RCPV
Vitreous opacity/flare

Contact Information: zhou_yongdong@wuxiapptec.com

Financial Disclosure There are no potential conflicts of interest to disclosure.

Incidence of Spontaneous Ocular Lesions in Rats

- The incidence of cornea opacity (primarily pinpoint opacity), lens opacity (primarily small opacity), persistent hyaloid artery (PHA), and persistent pupillary membrane (PPM) in SD rats was 70.71%, 12.02%, 2.09%, and 0.49%, respectively.
- The incidence of cornea opacity (primarily pinpoint opacity), lens opacity (primarily pinpoint opacity), persistent pupillary membrane (PPM), and persistent hyaloid artery (PHA) in Wistar rats was 79.57%, 9.81%, 2.11%, and 1.58%, respectively.

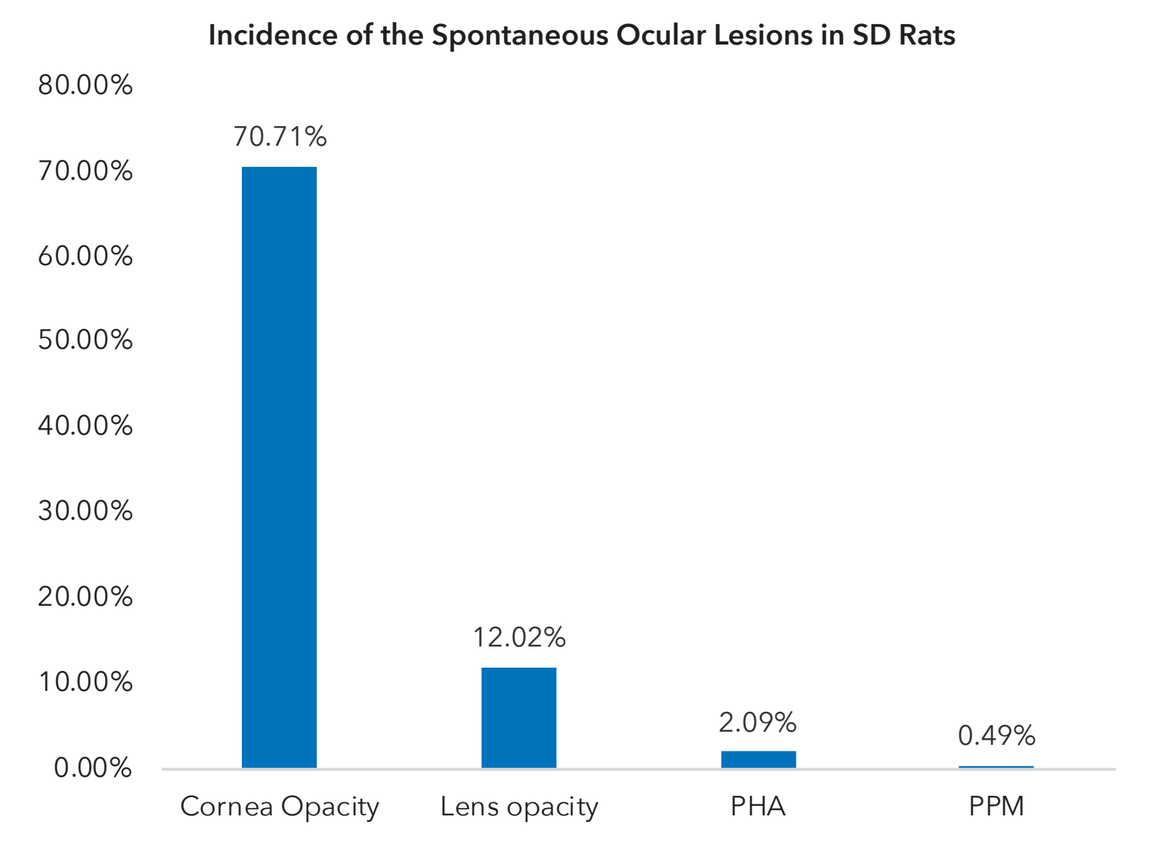
Incidence of the Spontaneous Ocular Lesions in SD Rats
70.71%
12.02%
2.09%
0.49%
Cornea Opacity
Lens opacity
PHA
PPM

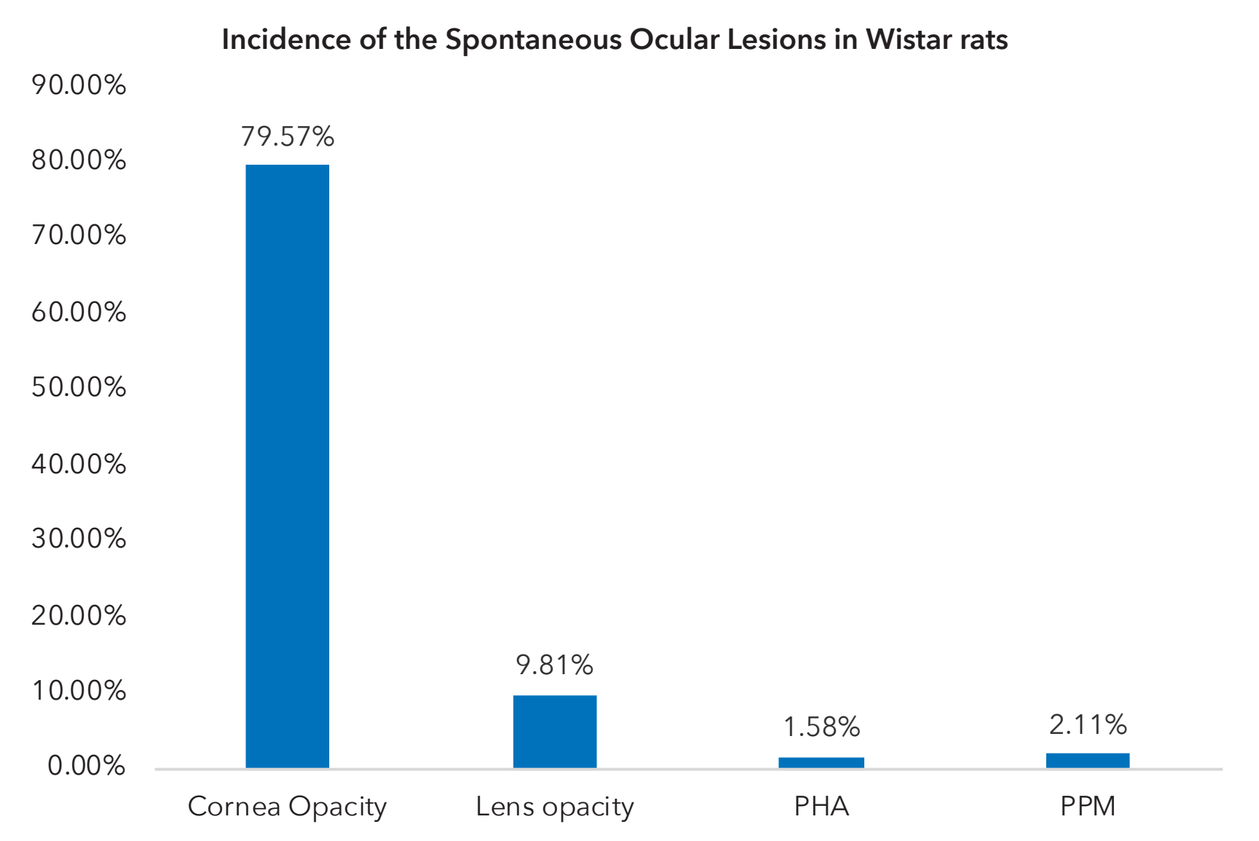
Incidence of the Spontaneous Ocular Lesions in Wistar rats
79.57%
9.81%
1.58%
2.11%
Cornea Opacity
Lens opacity
PHA
PPM

Incidence of Spontaneous Ocular Lesions in Rabbits

- The incidence of vitreous opacity/flare, lens opacity (primarily small opacity), conjunctiva hyperemia, cornea opacity (primarily small opacity) and ocular discharge in New Zealand rabbits was 13.12%, 11.03%, 3.28%, 2.27%, and 1.96%, respectively.
- The incidence of vitreous opacity, lens opacity (primarily small opacity), cornea opacity (primarily small opacity), conjunctiva hyperemia, ocular discharge, retinal edema, and conjunctiva swelling in Dutch Belted rabbits was 14.41%, 8.06%, 3.27%, 2.70%, 1.98%, 1.85%, and 1.56%, respectively.

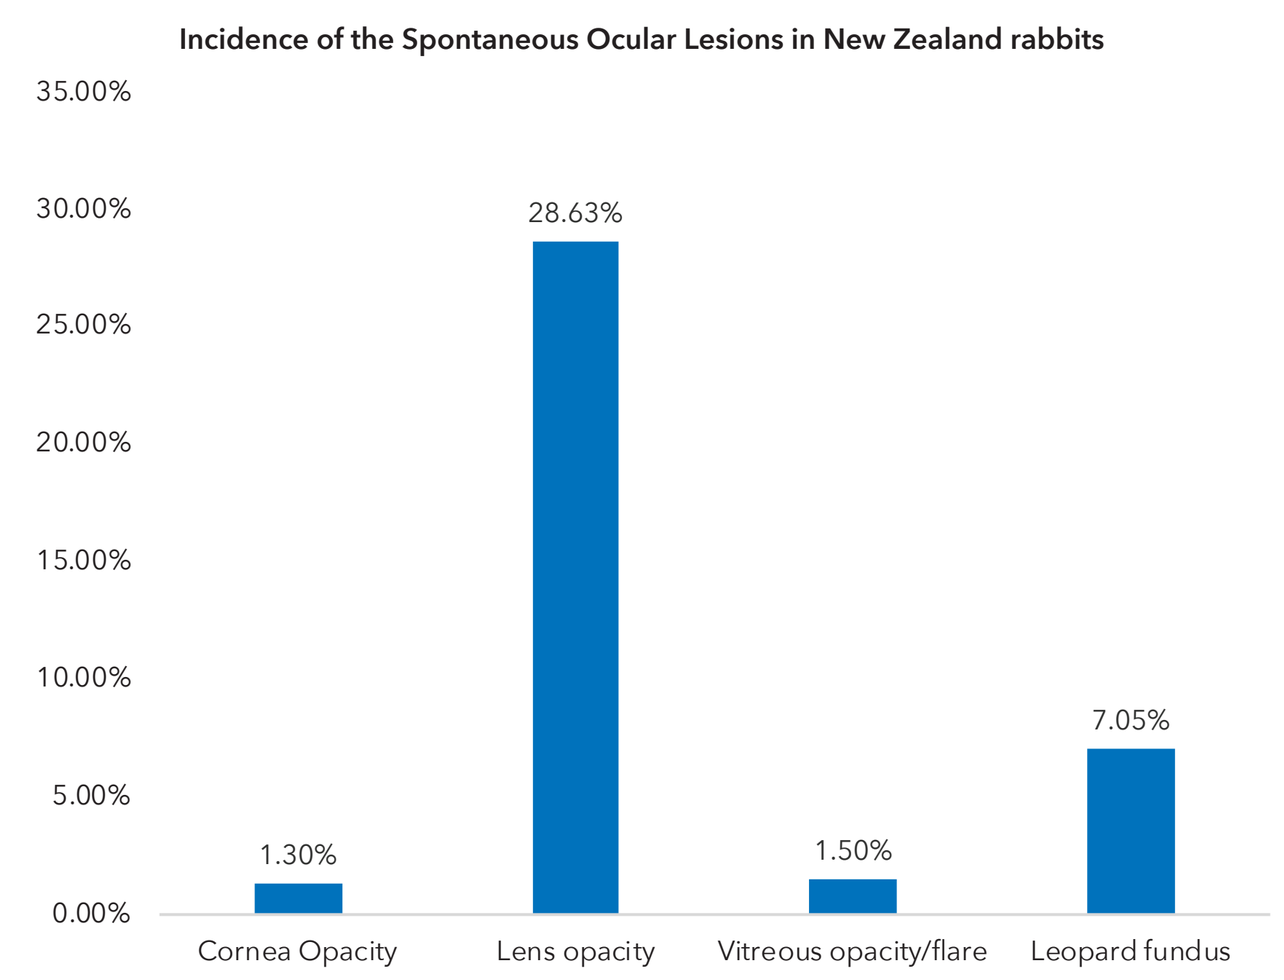
Incidence of the Spontaneous Ocular Lesions in New Zealand rabbits
1.30%
28.63%
1.50%
7.05%
Cornea Opacity
Lens opacity
Vitreous opacity/flare
Leopard fundus

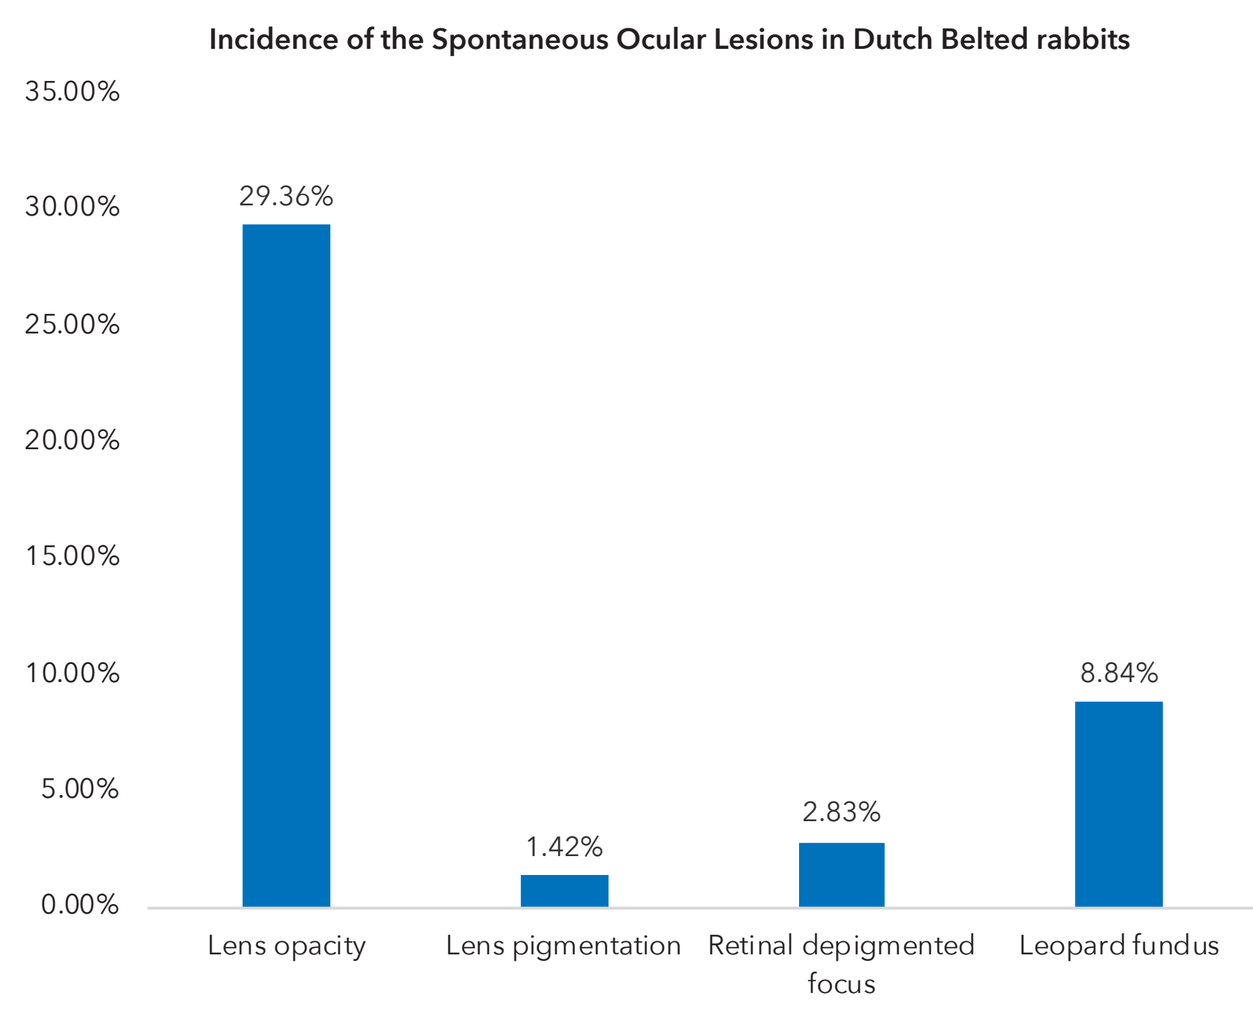
Incidence of the Spontaneous Ocular Lesions in Dutch Belted rabbits
29.36%
1.42%
2.83%
8.84%
Lens opacity
Lens pigmentation
Retinal depigmented focus
Leopard fundus

Incidence of Spontaneous Ocular Lesions in Beagle Dogs

- The incidence of lens opacity (primarily pinpoint opacity), cornea opacity (primarily small opacity), non-tapetal redness, small optic papilla, and ocular discharge in Beagle dogs was 21.62%, 3.75%, 2.47%, 2.44%, and 1.90%, respectively.

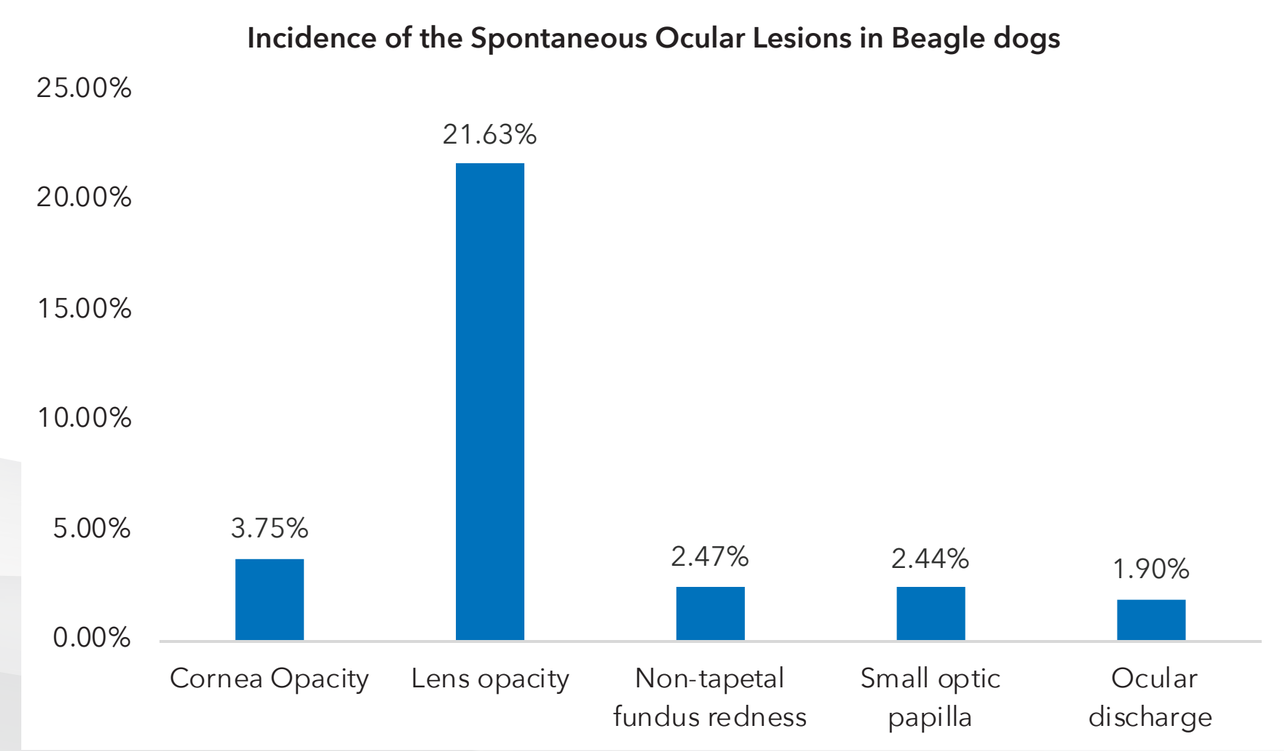
Incidence of the Spontaneous Ocular Lesions in Beagle dogs
3.75%
21.63%
2.47%
2.44%
1.90%
Cornea Opacity
Lens opacity
Non-tapetal fundus redness
Small optic papilla
Ocular discharge

Incidence of Spontaneous Ocular Lesions in Bama Pigs

- The incidence of cornea opacity (primarily pinpoint opacity), lens opacity (primarily small opacity), vitreous opacity/flare, ocular discharge, leopard fundus, lens pigmentation, and esotropia in Bama pigs was 9.18%, 9.06%, 3.78%, 2.66%, 2.25%, 1.57%, and 1.40%, respectively.

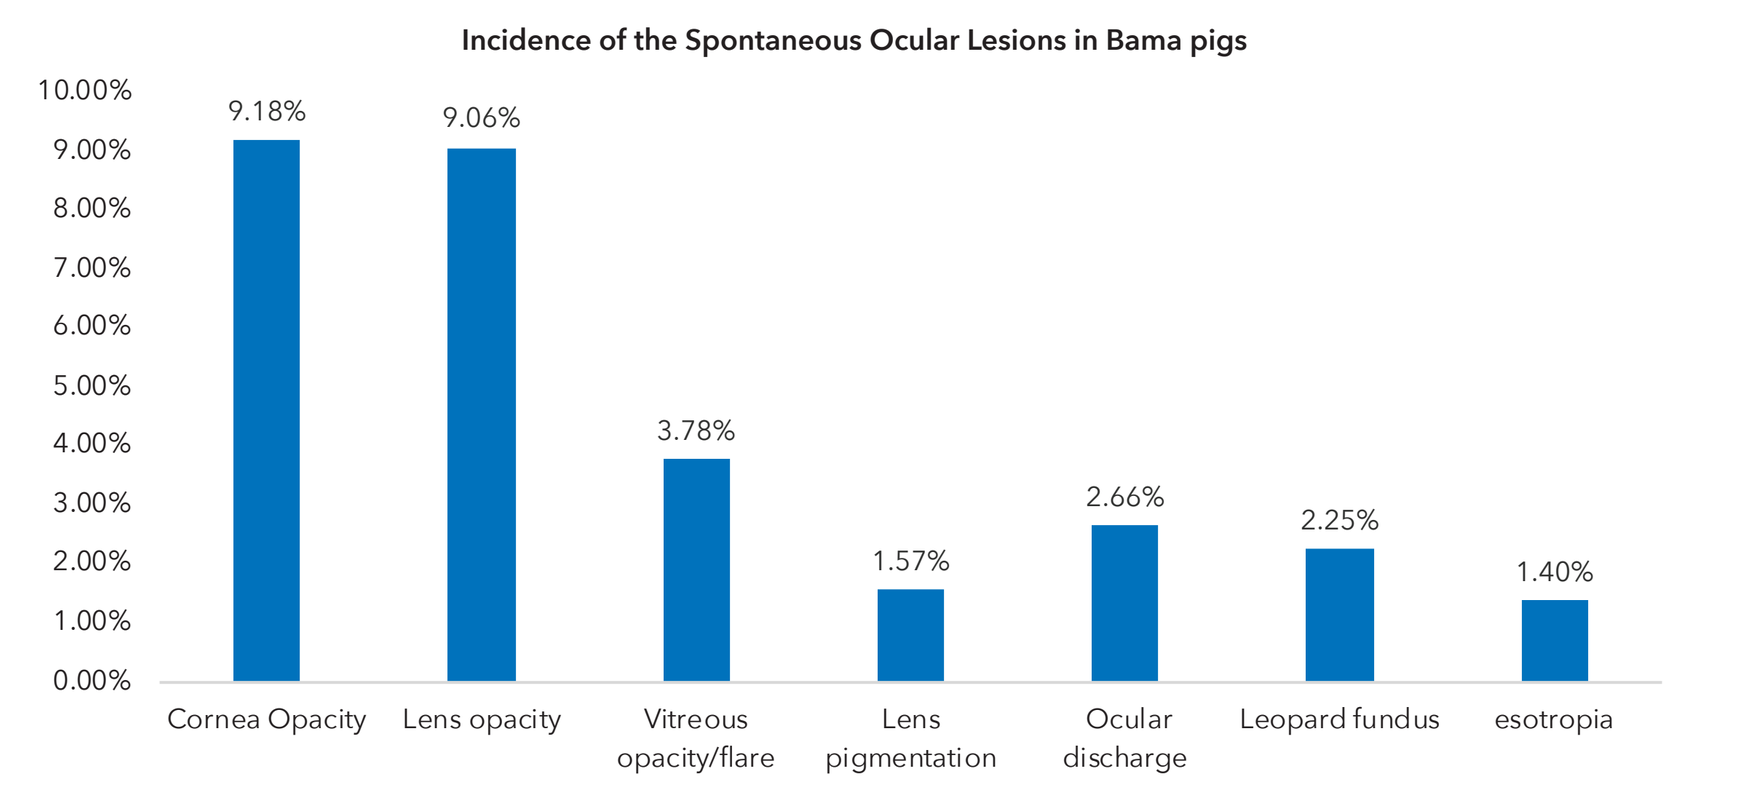
Incidence of the Spontaneous Ocular Lesions in Bama pigs
9.18%
9.06%
3.78%
1.57%
2.66%
2.25%
1.40%
Cornea Opacity
Lens opacity
Vitreous opacity/flare
Lens pigmentation
Ocular discharge
Leopard fundus
esotropia

Incidence of Spontaneous Ocular Lesions in Non-Human Primates

- The incidence of lens opacity (primarily pinpoint and small opacity), leopard fundus, vitreous opacity/flare, and cornea opacity (primarily small opacity) in Cynomolgus was 28.63%, 7.05%, 1.50%, and 1.30%, respectively.
- The incidence of lens opacity (primarily pinpoint opacity), leopard fundus, retinal depigmented focus, and lens pigmentation in Rhesus was 29.36%, 8.84%, 2.83%, and 1.42%, respectively.

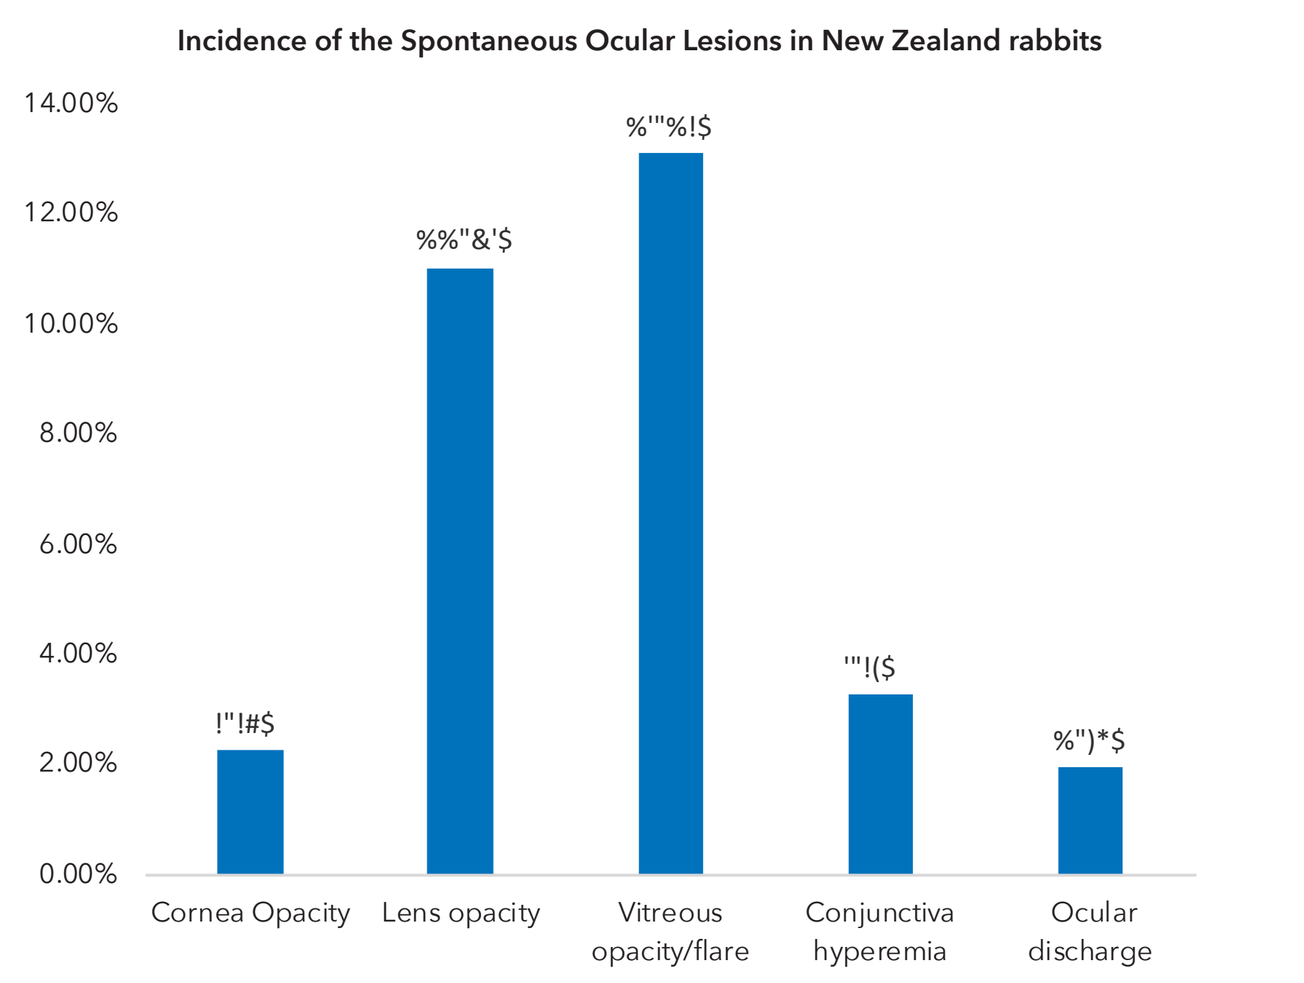
Incidence of the Spontaneous Ocular Lesions in New Zealand rabbits
Cornea Opacity
Lens opacity
Vitreous opacity/flare
Conjunctiva hyperemia
Ocular discharge

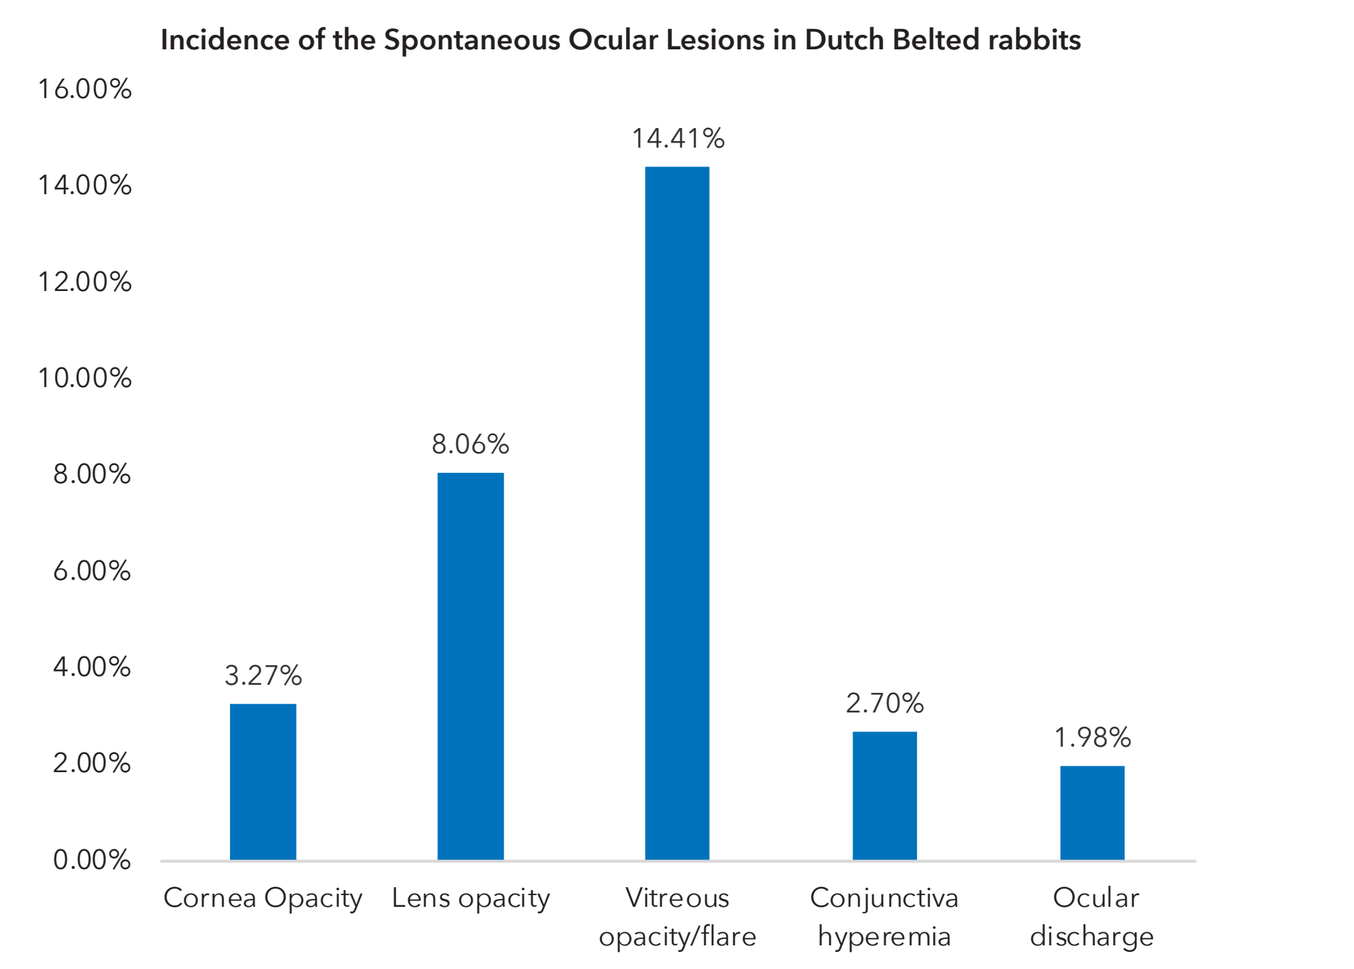
Incidence of the Spontaneous Ocular Lesions in Dutch Belted rabbits
3.27%
8.06%
14.41%
2.70%
1.98%
Cornea Opacity
Lens opacity
Vitreous opacity/flare
Conjunctiva hyperemia
Ocular discharge

Discussion

- Lens opacity was the primary spontaneous ocular lesion in mice, dogs, and non-human primates; in contrast, rats and Bama pigs primarily exhibited cornea opacity, while rabbits showed vitreous opacity/flare.
- The importance of selecting appropriate species in toxicologic studies.
- The data on these spontaneous ocular lesions was static; however, symptoms may develop as animals age.
- The differences in the incidences of a same ocular lesion were noted among animals of the same strain sourced from multiple vendors (data not shown), which could potentially be sample size.
- The data exhibited certain discrepancies compared to the literature, which may be attributed to factors such as age, sample size, and the source of animals.

Conclusion

- Toxicologists and ophthalmologists can differentiate treatment-related changes from spontaneous lesions by referring to the in-house data specific to the vendors and strains.
- Further analysis is necessary to explore the relationship between other factors such as sample size, vendor, and the incidences of spontaneous lesions.