Validation of Fluorescence In Situ Hybridization Method in Mouse - With a Novel Staining Strategy

Yaoyao Chen, Yuanyuan Chen, Ning Xu, Xiang Shan, Liwen Gao, Millie Chen, Xiaoxia Li
WuXi AppTec, Suzhou, China

Abstract: 4106

Background and Purpose

Micronuclei (MN) originate from chromosomal fragments or whole chromosomes that fail to be incorporated into daughter nuclei, both *in vivo* and *in vitro*. Being one of the standard battery assays for regulatory purpose, the *in vivo* micronucleus (MN) assay serves a better understanding about the genotoxic potential of test compound and positive results can be induced by both clastogenic and aneugenic mode of action, will raise concerns about the carcinogenic and teratogenic risks of candidate compounds in drug development. As a common understanding of the genotoxicity community, the existence of threshold for aneugen is accepted. And according to ICH guideline S2 (R1), when positive findings are observed in *in vitro* cytogenetic assays, mechanistic research or two *in vivo* assays are recommended as a follow-up strategy. FISH, known for its high specificity, easiness, and time-efficiency, is available for both *in vitro* and *in vivo* conditions due to pan-centromeric probes. We performed almost the first standalone study to validate the FISH method in mouse using the cellulose column separation and a novel staining method.

Methods and Materials

Standard OECD study design of *in vivo* MN assay was employed, which includes three daily administration for the negative control article, 0.9% sodium chloride injection, positive control articles included cyclophosphamide monohydrate (CP) and vinblastine sulfate (VIN), and both were administrated once at 75 mg/kg or 2 mg/kg respectively, bone marrow was collected at 20-24 hours post last dosing. For micronucleus analysis, fresh bone marrow cells were fixed and then stained in Acridine Orange (AO) staining solution; for FISH analysis, the bone marrow cells were filtered through a self-made cellulose column, then slides were prepared and processed via an optimized FISH methods (Genetic Toxicology Testing-A Laboratory Manual), denaturing, annealing and hybridizing. For FISH slide staining, a new combination of PI and DAPI staining solution were applied to slide simultaneously. The micronucleus frequency (MN-PCE) is calculated by scoring ~4000 polychromatic erythrocytes (PCE) from each animal; and the ratio of polychromatic erythrocyte (PCE) to total erythrocytes was determined by counting 1000 to 2000 erythrocytes (polychromatic erythrocyte (PCE) and normochromatic erythrocyte (NCE)) for each animal; for FISH analysis, preferably 100 MN from animal were scored to with the discrimination of centromeric positive (C+) and negative (C-) ones.

Results and Discussion

Table 1 Summary result of Micronucleus Data and Fluorescence In Situ Hybridization Data

Treatment	Dose (mg/kg/day)	% PCE (Mean ± S.D., %)	MN-PCE Frequency (Mean ± S.D., ‰)	Percentage of C+ MN-PCE (Mean ± SD, %)	Percentage of C- MN-PCE (Mean ± SD, %)
Negative Control Article	0	71.6 ± 5.5	2.0 ± 1.1	40.97 ± 25.34	59.03 ± 25.34
Cyclophosphamide Monohydrate	75	64.1 ± 3.1	38.6 ± 9.4	21.86 ±6.59	78.14 ±6.59
Vinblastine Sulfate	2	51.1 ± 4.0	15.4 ± 2.6	74.22 ±7.67#	25.78 ±7.67

p≤0.05.

Note:

1. For CP and Vinblastine sulfate group, the animals were singly dosed via IP injection and collected for bone marrow at 20-24 hours afterwards.
2. Clear discrimination of PCE (red) over NCE cells (not stained) were noted.
3. Literature reference data (C+ MN-PCE%) for VIN and CP is 67.9% and 25.3±1.3% respectively.

Conclusion

1. First, the data had demonstrated the clastogenic mode of action of the CP, and the aneugenic mechanism of VIN for the induction of micronuclei formation.
2. Second, unlike to paper reported method, which uses cellulose column separation for all collected bone marrow cells, we only applied this step after micronucleus slide preparation and before FISH slide preparation because of poor discrimination between PCE cells and NCE cells after acridine orange (AO) staining.
3. Finally, the result also showed that the optimized staining method provided good discrimination of PCE cells over NCE cells, and the centromeric DNA signal was cleared distinguished.
4. Evidence supported that the optimized FISH method was successfully developed and validated in GIVT laboratory of WuXi AppTec (Suzhou).

Figure 1 Representative images of micronuclei (MN) in mice bone marrow analyzed using fluorescence in situ hybridization (FISH) method

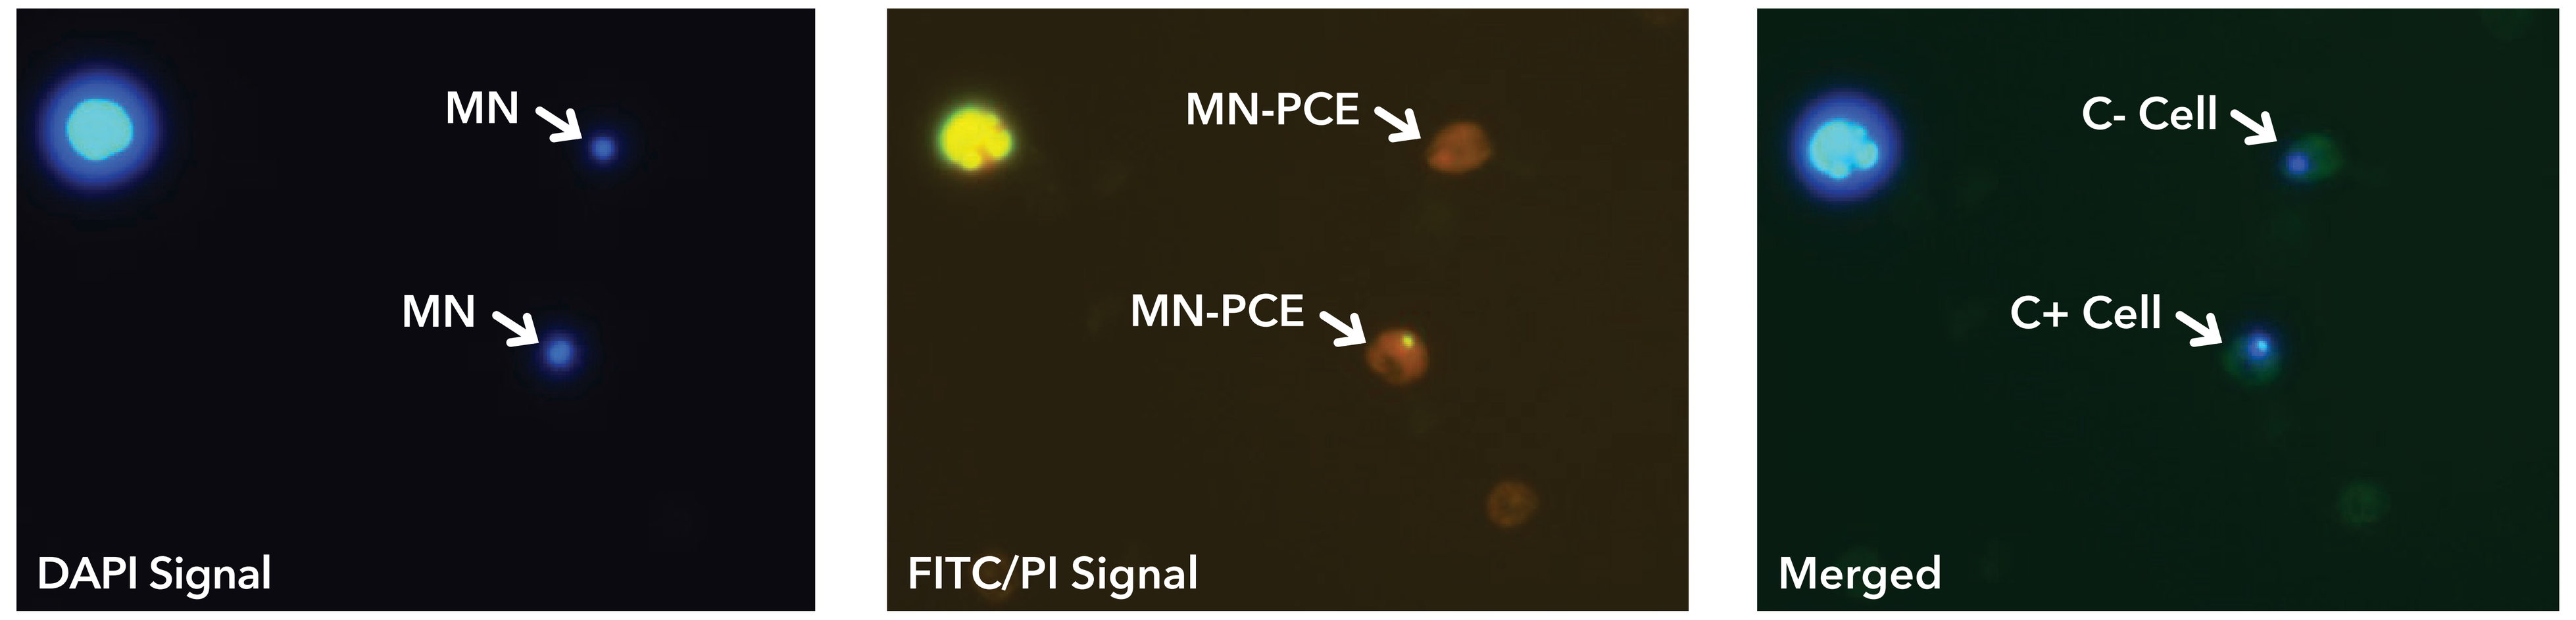
MN
MN
DAPI Signal
MN-PCE
MN-PCE
FITC/PI Signal
C- Cell
C+ Cell
Merged

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou).