Validation of the Flow Cytometry Based Pig-a Gene Mutation Assay in Rats

Cong Yue, Mingzhu Kang, Ning Xu, Yating Di, Xiang Shan, Liwen Gao, Millie Chen, Xiaoxia Li
WuXi AppTec, Suzhou, China

WuXi AppTec

Abstract: 4105

Background and Purpose

Though assessment about gene mutagenicity is not a new concept, it wasn't until June 2022 that OECD Guideline 470 was officially released for regulatory purposes. The assay is recommended when positive result is observed in bacterial reverse mutation assay, and can act as a possible substitute of the traditional TGR assay. And like the mammalian micronucleus assay (OECD 474), the Pig-a assay uses flow cytometry as sample analysis method, which owns the advantages of time-efficient and data robustness and consistency.

Methods and Materials

OECD recommended study design was used, including elimination of jackpot mutation events (7 days before administration), selected animals were then administrated with negative control (0 mg/kg/day) and ethyl methanesulfonate (25, 50 and 100 mg/kg/-day) for 28 consecutive days, positive control N-Nitroso-N-ethylurea (ENU) was administrated to animals once daily for 3 consecutive days at 20 mg/kg/day (corrected by correction factor). On Day 29, blood samples were collected and processed following Litron's analysis kit manual. Additional parameters of carry-over effect, intra-day and inter-day precision and frozen sample stability were also evaluated.

Results

a) For jackpot mutation checkpoint, 4 animals were excluded from randomization for the notable high incidence of MUT RET per million RET or MUT RBC per million RBC.

b) Immediate analysis revealed dose related increase response and statistically significant increases in both MUT RBCs and MUT RETs ($p \leq 0.05$) in all EMS dose groups when compared with negative control article group.

c) For carryover effect evaluation, the variance of negative control samples over positive control samples were within 5%.

d) The coefficient of variation (%CV) of RET%, MUT RET and MUT RBC were acceptable for both Intra- day and inter-day precision analysis.

e) Samples were stability when stored under -85~-75°C for up to two months.

Validation Parameters

Characteristic	Procedure	Acceptance Criteria
Negative and Positive	The per 6 samples of male animals from negative control and positive control treatment were analyzed for both pre-column and post column samples.	The data obtained was calculated for RET%, MUT RET and MUT RBC of the negative control and positive control group was comparable to literature reported negative control and positive control data range.
Carryover	Detect both the negative and positive blood samples in a sequence of PC (positive control) 1-PC2-PC3-NC (negative control) 1-NC2-NC3 for both pre-column and post column samples, manual washing was performed between pre- and post-column sample.	The data obtained was calculated for RET%, MUT RET and MUT RBC. The carryover was calculated as [(NC3-NC1)/PC3]*100%. The carryover effect would be acceptable if the carryover value is no more than 5% for each parameter.
Precision	For the intra-day precision, two blood samples from test article treatment group were labeled two times (interval not more than 8 hours) for detection. For the inter-day precision, two blood sample from test article treatment group were labeled for detection once daily for two consecutive days.	The variance estimates were expressed as a coefficient of variation (%CV) relative to the overall mean of RET%, MUT RET and MUT RBC. Intra- and inter-day assay variation wouldbe acceptable if the %CV ≤20%.
Stability	The samples of male animals from test article treatment group were frozen on the day of collection, and stored for 1 or 2 months, then thawed for FCM analysis.	RET%, MUT RET per million RET, and MUT RBC per million RBC generated from fresh blood samples were compared to frequencies observed in corresponding, frozen blood samples (1 or 2 months). The parameters were evaluated using Bland–Altman plots to assess agreement and bias, all data fell in the consistency limit (lower and upper limits of agreement).

Table 1 Carryover Effect

		Pre-Column				POST-Column				
Treatment Group	Sample ID	No. of MUT RET	No. of WT RET	No. of MUT RBC	No. of WT RBC	No. of MUT RET	No. of MUT RBC	%RET	Mutant RBCs per 10^6 Total RBCs	Mutant RETs per 10^6 Total RETs
ENU Dose(20 mg/kg/day)	1	2	7546	26	155560	1464	18478	4.6	148.8	236.0
	2	1	8005	25	165438	1603	18952	4.6	126.6	213.9
	3	2	7142	29	150803	1703	20132	4.5	147.4	254.3
Negative Control Article Dose (0 mg/kg/day)	1	0	5768	2	157083	10	115	3.5	0.8	1.9
	2	0	4911	2	137411	11	98	3.5	0.9	2.5
	3	1	6478	0	167467	7	124	3.7	0.9	1.4
Carryover=[(NC3-NC1)/PC3]*100%								4.44%	0.07%	-0.20%

Note: WT= Wild type; MUT=Mutant; RET=Reticulocyte; RBC=Red blood cells; %RET = (No. of MUT RET of PRE Column + No. of WT RET of PRE Column)/(No. of MUT RET of PRE Column + No. of WT RET of PRE Column + No. of MUT RBC of PRE Column + No. of WT RBC of PRE Column)×100; Mutant RBC per 106 Total RBC = (No. of MUT RET of POST Column + No. of MUT RBC of POST Column)/ Total RBC Equivalents × 106; Mutant RET per 106 Total RET = No. of MUT RET of POST Column/ Total RET Equivalents × 106

Table 2 Intra-day Precision

		Pre-Column				POST-Column				
Treatment Group	Sample ID	No. of MUT RET	No. of WT RET	No. of MUT RBC	No. of WT RBC	No. of MUT RET	No. of MUT RBC	%RET	Mutant RBCs per 10^6 Total RBCs	Mutant RETs per 10^6 Total RETs
EMS Dose(100 mg/kg/day)	1	0	3720	2	103308	75	1107	3.5	12.6	22.9
	2	0	7162	6	166725	178	1493	4.1	13.2	34.1
Average								3.8	12.9	28.5
EMS Dose(100 mg/kg/day)	1	1	8098	4	187685	252	2049	4.1	13.0	34.5
	2	0	7313	4	197114	181	1937	3.6	11.2	26.7
Average								3.9	12.1	30.6
Coefficient of Variation (%CV)								1.84	4.53	5.03

Table 3 Inter-day Precision

		Pre-Column				POST-Column				
Treatment Group	Sample ID	No. of MUT RET	No. of WT RET	No. of MUT RBC	No. of WT RBC	No. of MUT RET	No. of MUT RBC	%RET	Mutant RBCs per 10^6 Total RBCs	Mutant RETs per 10^6 Total RETs
EMS Dose(100 mg/kg/day)	1	0	3720	2	103308	75	1107	3.5	12.6	22.9
	2	0	7162	6	166725	178	1493	4.1	13.2	34.1
Average								3.8	12.9	28.5
EMS Dose(100 mg/kg/day)	1	1	8166	3	185060	268	1895	4.2	12.8	37.4
	2	0	7811	3	207950	194	2051	3.6	12.5	29.9
Average								3.9	12.6	33.6
Coefficient of Variation (%CV)								1.84	1.66	11.61

Table 4 Stability Test - 1 Month

		Pre-Column				POST-Column				
Treatment Group	Sample ID	No. of MUT RET	No. of WT RET	No. of MUT RBC	No. of WT RBC	No. of MUT RET	No. of MUT RBC	%RET	Mutant RBCs per 10^6 Total RBCs	Mutant RETs per 10^6 Total RETs
Fresh Blood Sample Analysis Result:										
ENU Dose (20 mg/kg/day)	1	1	6376	17	136873	1164	12166	4.5	101.7	199.6
	2	0	6856	19	138477	728	12035	4.7	104.8	126.8
	3	0	5475	9	131323	629	10440	4.0	91.7	130.2
	4	3	7481	19	141959	875	11526	5.0	93.3	131.4
	5	0	5675	24	149730	827	13521	3.7	107.1	169.1
	6	0	6854	14	161810	482	11628	4.1	83.2	81.5
Mean								4.3	97.0	139.7
Lower LOA to Upper LOA								-0.3564~0.4564	-44.5349~-7.0318	-67.3790~16.7790
Frozen Sample Stored at <-80°C for 1 Month										
ENU Dose (20 mg/kg/day)	1	1	9895	24	219289	1242	12268	4.3	65.6	139.8
	2	2	9705	13	198640	943	13473	4.7	82.1	115.3
	3	1	7435	22	180860	804	11060	3.9	70.0	120.2
	4	0	9129	21	171162	982	11655	5.1	83.0	127.3
	5	2	8518	15	213463	1004	13882	3.8	73.0	128.3
	6	2	9032	13	193459	434	8855	4.5	53.4	55.9
Mean								4.4	71.2	114.5
Bias Value								0.0500	-25.7833	-25.3000

Note: WT= Wild type; MUT=Mutant; RET=Reticulocyte; RBC=Red blood cells.

Table 5 Stability Test - 2 Months

		Pre-Column				POST-Column				
Treatment Group	Sample ID	No. of MUT RET	No. of WT RET	No. of MUT RBC	No. of WT RBC	No. of MUT RET	No. of MUT RBC	%RET	Mutant RBCs per 10^6 Total RBCs	Mutant RETs per 10^6 Total RETs
Fresh Blood Sample Analysis Result:										
ENU Dose (20 mg/kg/day)	1	1	6376	17	136873	1164	12166	4.5	101.7	199.6
	2	0	6856	19	138477	728	12035	4.7	104.8	126.8
	3	0	5475	9	131323	629	10440	4.0	91.7	130.2
	4	3	7481	19	141959	875	11526	5.0	93.3	131.4
	5	0	5675	24	149730	827	13521	3.7	107.1	169.1
	6	0	6854	14	161810	482	11628	4.1	83.2	81.5
Mean								4.3	97.0	139.7
Lower LOA to Upper LOA								-0.3564~0.4564	-44.5349~-7.0318	-67.3790~16.7790
Frozen Sample Stored at <-80°C for 2 Montsh										
ENU Dose (20 mg/kg/day)	1	2	8601	18	183730	1135	12286	4.5	88.2	166.8
	2	1	9033	22	180005	840	12613	4.8	83.2	108.7
	3	4	7183	33	180796	710	11232	3.8	78.6	122.3
	4	0	8547	21	148603	970	10457	5.4	92.1	143.8
	5	1	7617	29	188710	996	14153	3.9	93.7	158.9
	6	0	8994	21	187919	562	11249	4.6	68.1	71.0
Mean								4.5	84.0	128.6
Bias Value								0.1667	-12.9833	-11.1833

Note: WT= Wild type; MUT=Mutant; RET=Reticulocyte; RBC=Red blood cells.

Figure 1 Plots of post column sample for Negative (Saline, left) and Positive (ENU, right) Control Article

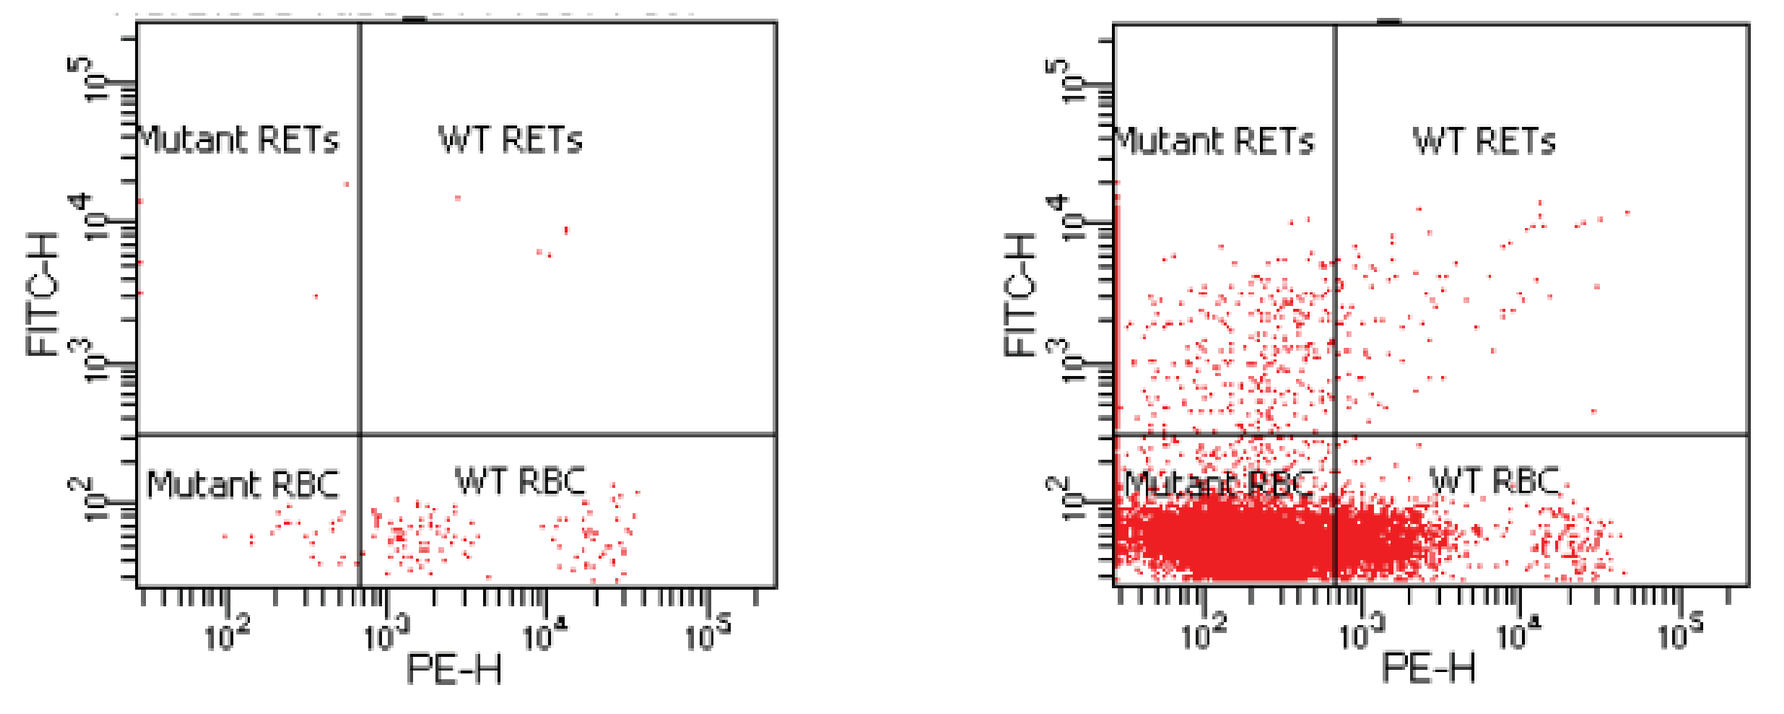
Mutant RETs
WT RETs
Mutant RBC
WT RBC
FITC-H
PE-H
Mutant RETs
WT RETs
Mutant RBC
WT RBC
FITC-H
PE-H

Conclusion

The test article EMS has induced statistically significant and dose-related increase in both MUT RET and MUT RBC, with no notable changes in RET% over the doses, thus, the testing system is considered can detect Pig-A gene mutation under the condition of this study. Additional validations of carryover effect, Intra- and inter-day precision and blood samples stability under -85~-75°C all demonstrated successful. The assay is fully validated and can be performed under GLP compliance in WuXi AppTec.

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and *In Vitro* Toxicology group at WuXi AppTec (Suzhou).