A Review of the Incidence of Common Arrhythmias in Bama Minipigs Observed in Stand-Alone Safety Pharmacology and General Toxicology Studies

Yangming Chen; Rui Wu; Xinru Ji; Sue McPherson
WuXi AppTec, China

Abstract: 3115

Abstract

The use of the minipig as a non-rodent species in non-clinical research has continued to increase and although its use is still considerably less than that of the dog or the non-human primate (NHP), for certain areas of research the physiological similarities between swine and humans make it the species of choice. A recent European RETHINK project has taken place to assess minipigs (in general not as a specific species) as models for the toxicity testing of new medicines and chemicals and concluded there is great utility in their use in various areas of research. A retrospective review on the incidence of common arrhythmias in Bama Minipigs dosed with vehicle controls during both the conduct of stand-alone safety pharmacology and from general toxicology studies was performed. In stand-alone studies, the ECG data were continuously recorded for at least 20 hours for each animal by Physio Tel Digital technology of DSI Physiologic Data Acquisition and Analysis System with Lead II in conscious and unrestrained Bama Minipigs. In the general toxicology studies, the ECG data were continuously recorded for at least 11 seconds for each animal with a Schiller ECG machine with Leads I, II, III, aVR, aVL and aVF in conscious and restrained Bama Minipigs. The Bama minipigs were between 3 and 9 months of age. The incidence of arrhythmias (Ventricular Premature and Atrial Premature Complexes) was compared between the two collection methods. The results showed that the same incidence of 1% was noted between stand-alone and general toxicology studies, indicating that both methods are sensitive enough to detect background arrythmias in the Bama minipig.

Methods and Materials

Stand-Alone Studies	
Age	4 to 9 months
Bodyweight	17.9 to 29.7 kg
Animal Number	18 males and 20 females
Animal Source	Pearl Laboratory Animal Sci. & Tech. Co., Ltd. Wujiang Tian Yu Biological Tech. Co., Ltd.
ECG Recording	Telemetry with internal implants
	Conscious and unrestrained animals
	Lead II
	Continuous recording at least 20 hours
	DSI Physiologic Data Acquisition and Analysis System

General Toxicology Studies	
Age	3 to 5 months
Bodyweight	8.5 kg to 11.9 kg
Animal Number	143 males and 131 females
Animal Source	Pearl Laboratory Animal Sci. & Tech. Co., Ltd. Wujiang Tian Yu Biological Tech. Co., Ltd.
ECG Recording	External electrode
	Conscious and restrained animals
	Leads I, II, III, aVR, aVL and aVF
	Continuous recording at least 11 seconds
	Schiller ECG machine

Summary of ECG Data

- Summary of arrhythmias in stand-alone studies and general toxicology studies are presented in Table 1, Table 2, Figure 1, Figure 2, Figure 3 and Figure 4.
- The example arrhythmias in stand-alone studies and general toxicology studies are presented in Figure 5, Figure 6 and Figure 7.

Table 1 Summary of Arrhythmias in Stand-Alone Studies

	Number of Animals	Ventricular Premature Complexes (VPCs)	Time
Male	18	0	NA
Female	20	5	Two VPCs occurred at 14:00-15:00 One VPC occurred at 16:00-17:00 One VPC occurred at 03:00-04:00 One VPC occurred at 06:00-07:00
Total	38	5[a]	NA

[a]: 5 VPCs occurred in 3 females. One had 2 VPCs at 03:00-04:00 and 06:00-07:00, one had 2 VPCs at 14:00-15:00 and the other one had 1 VPC at 16:00-17:00.

Table 2 Summary of Arrhythmias in General Toxicology Studies

	Number of Animals	Atrial Premature Complexes (APCs)	Ventricular Premature Complexes (VPCs)
Male	143	4	4
Female	131	2	0
Total	274	6[b]	4[c]

[b]: 6 APCs occurred in 2 males and 1 female. One male had 3 APCs and the other male had 1 APC. The female had 2 APCs. [c]: 4 VPCs occurred in 3 males, in which 2 had 1 VPC each and 1 had 2 VPCs.

Figure 1 VPCs in Bama Minipigs of Stand-alone Studies

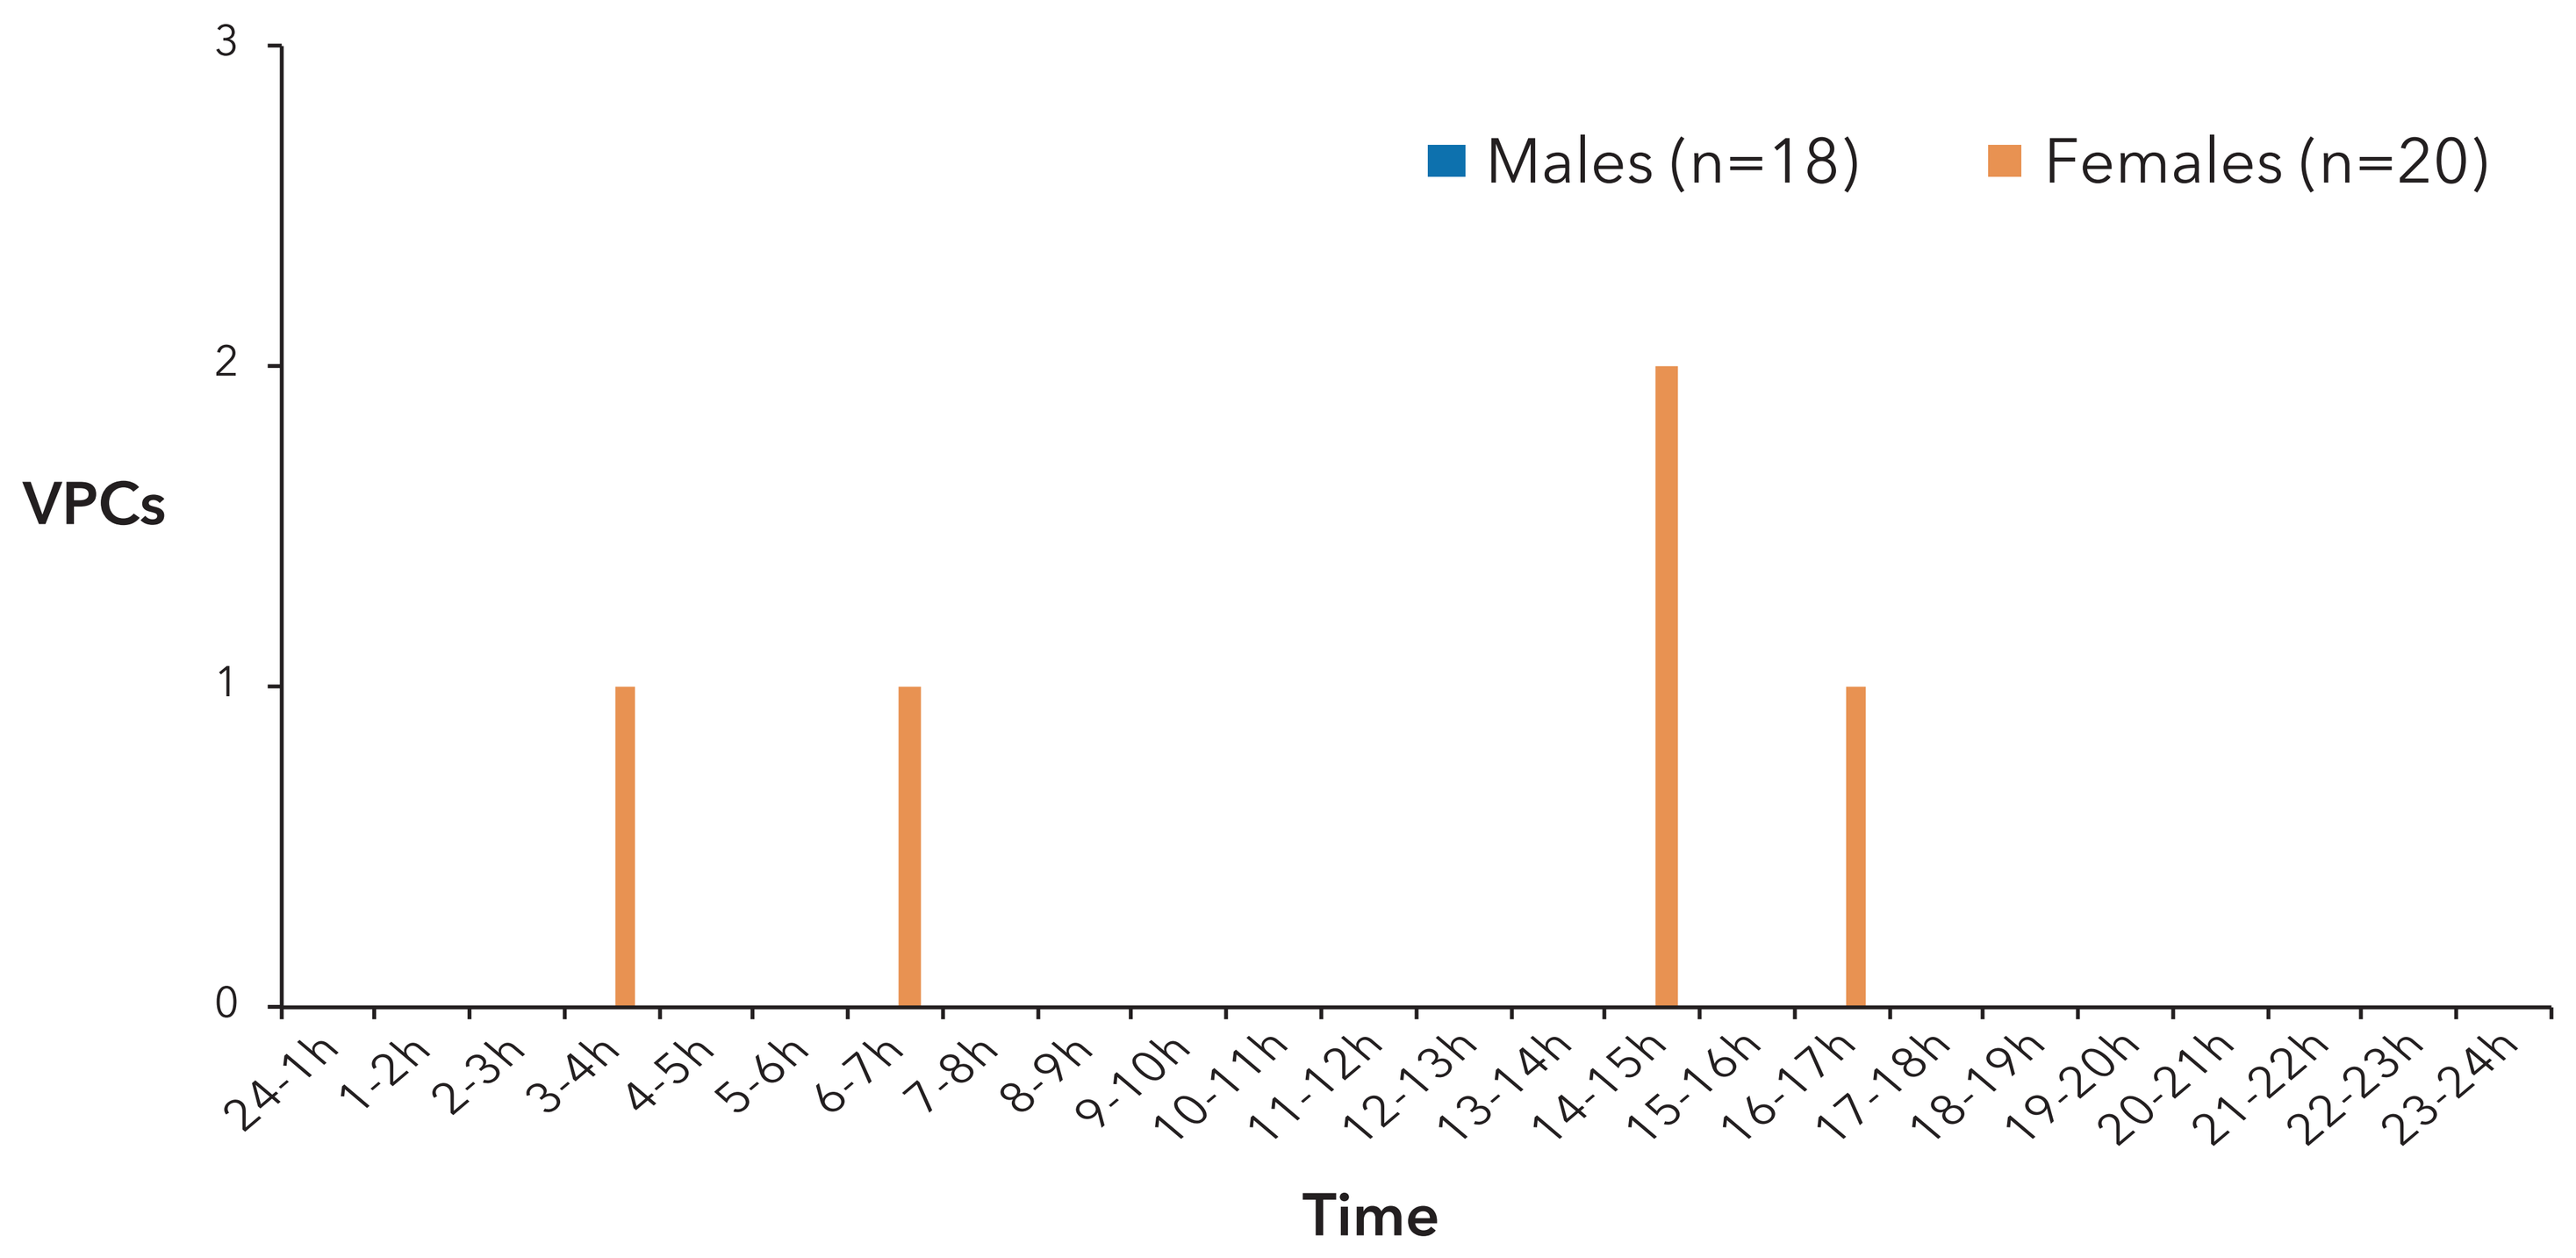
Males (n=18)
Females (n=20)
VPCs
0
1
2
3
24-1h
1-2h
2-3h
3-4h
4-5h
5-6h
6-7h
7-8h
8-9h
9-10h
10-11h
11-12h
12-13h
13-14h
14-15h
15-16h
16-17h
17-18h
18-19h
19-20h
20-21h
21-22h
22-23h
23-24h
Time

Figure 2
VPCs in Bama Minipigs of Stand-alone Studies (n=38)

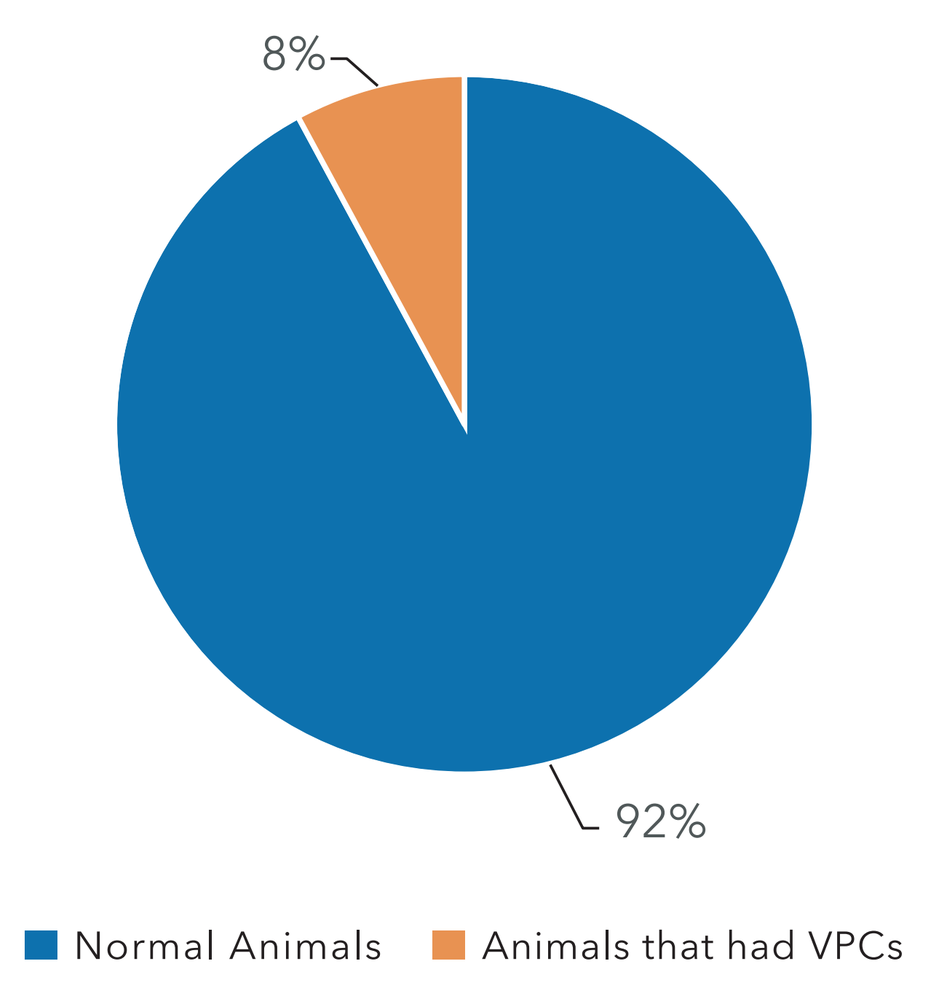
8%
92%
Normal Animals
Animals that had VPCs

Figure 3
APCs in Bama Minipigs of General Toxicology Studies (n=274)

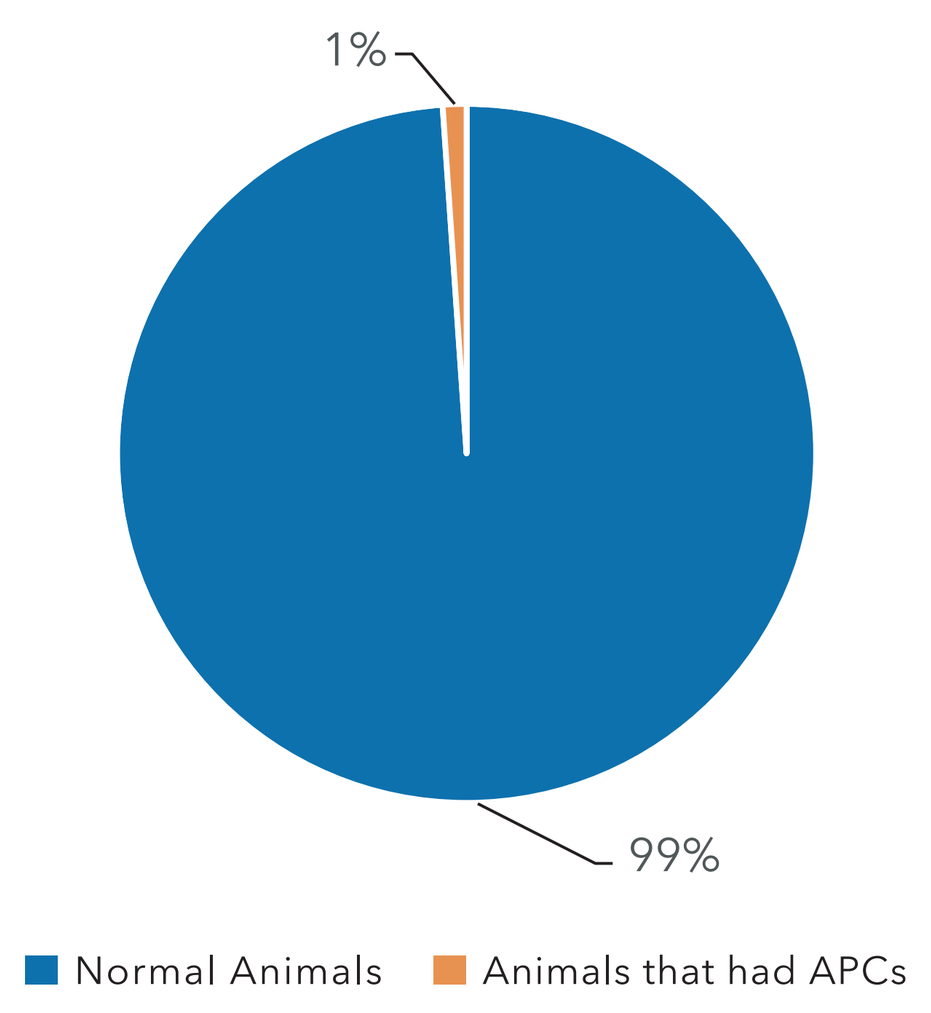
1%
99%
Normal Animals
Animals that had APCs

Figure 4
VPCs in Bama Minipigs of General Toxicology Studies (n=274)

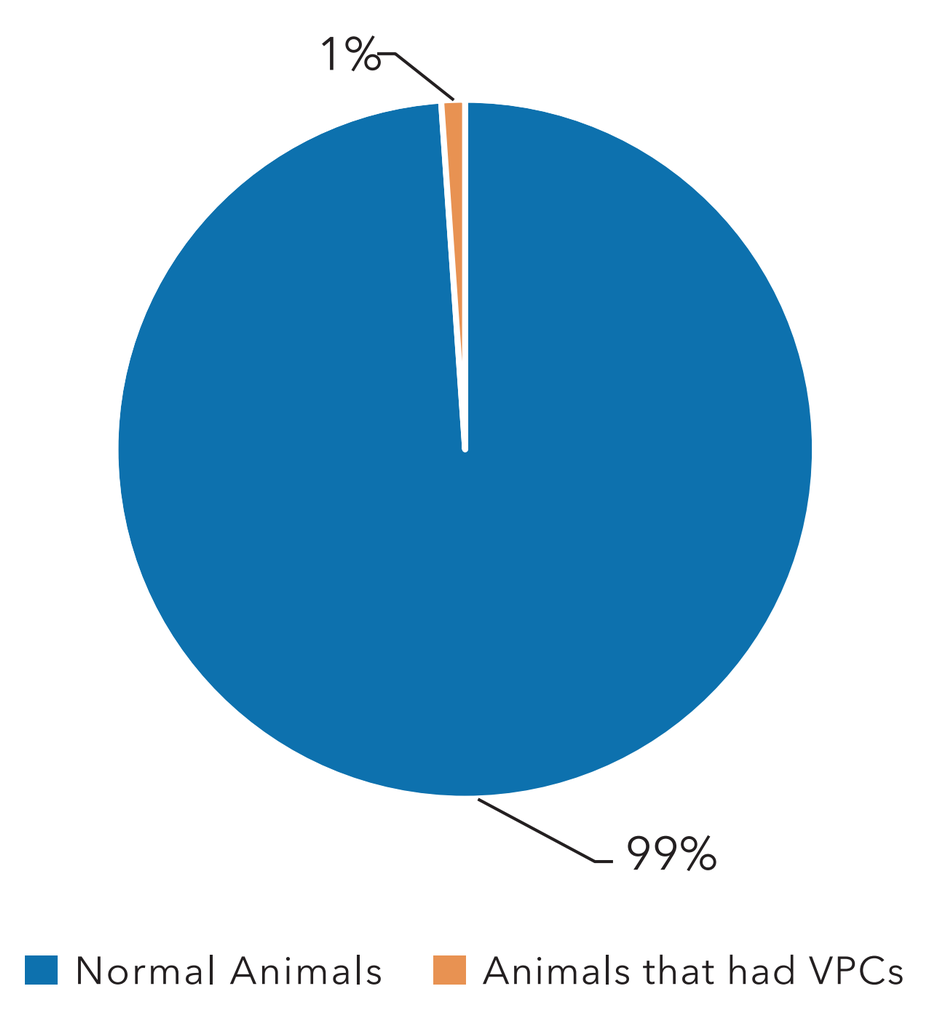
1%
99%
Normal Animals
Animals that had VPCs

Figure 5 Ventricular Premature Complex (VPC) in Bama Minipigs of Stand-alone Studies

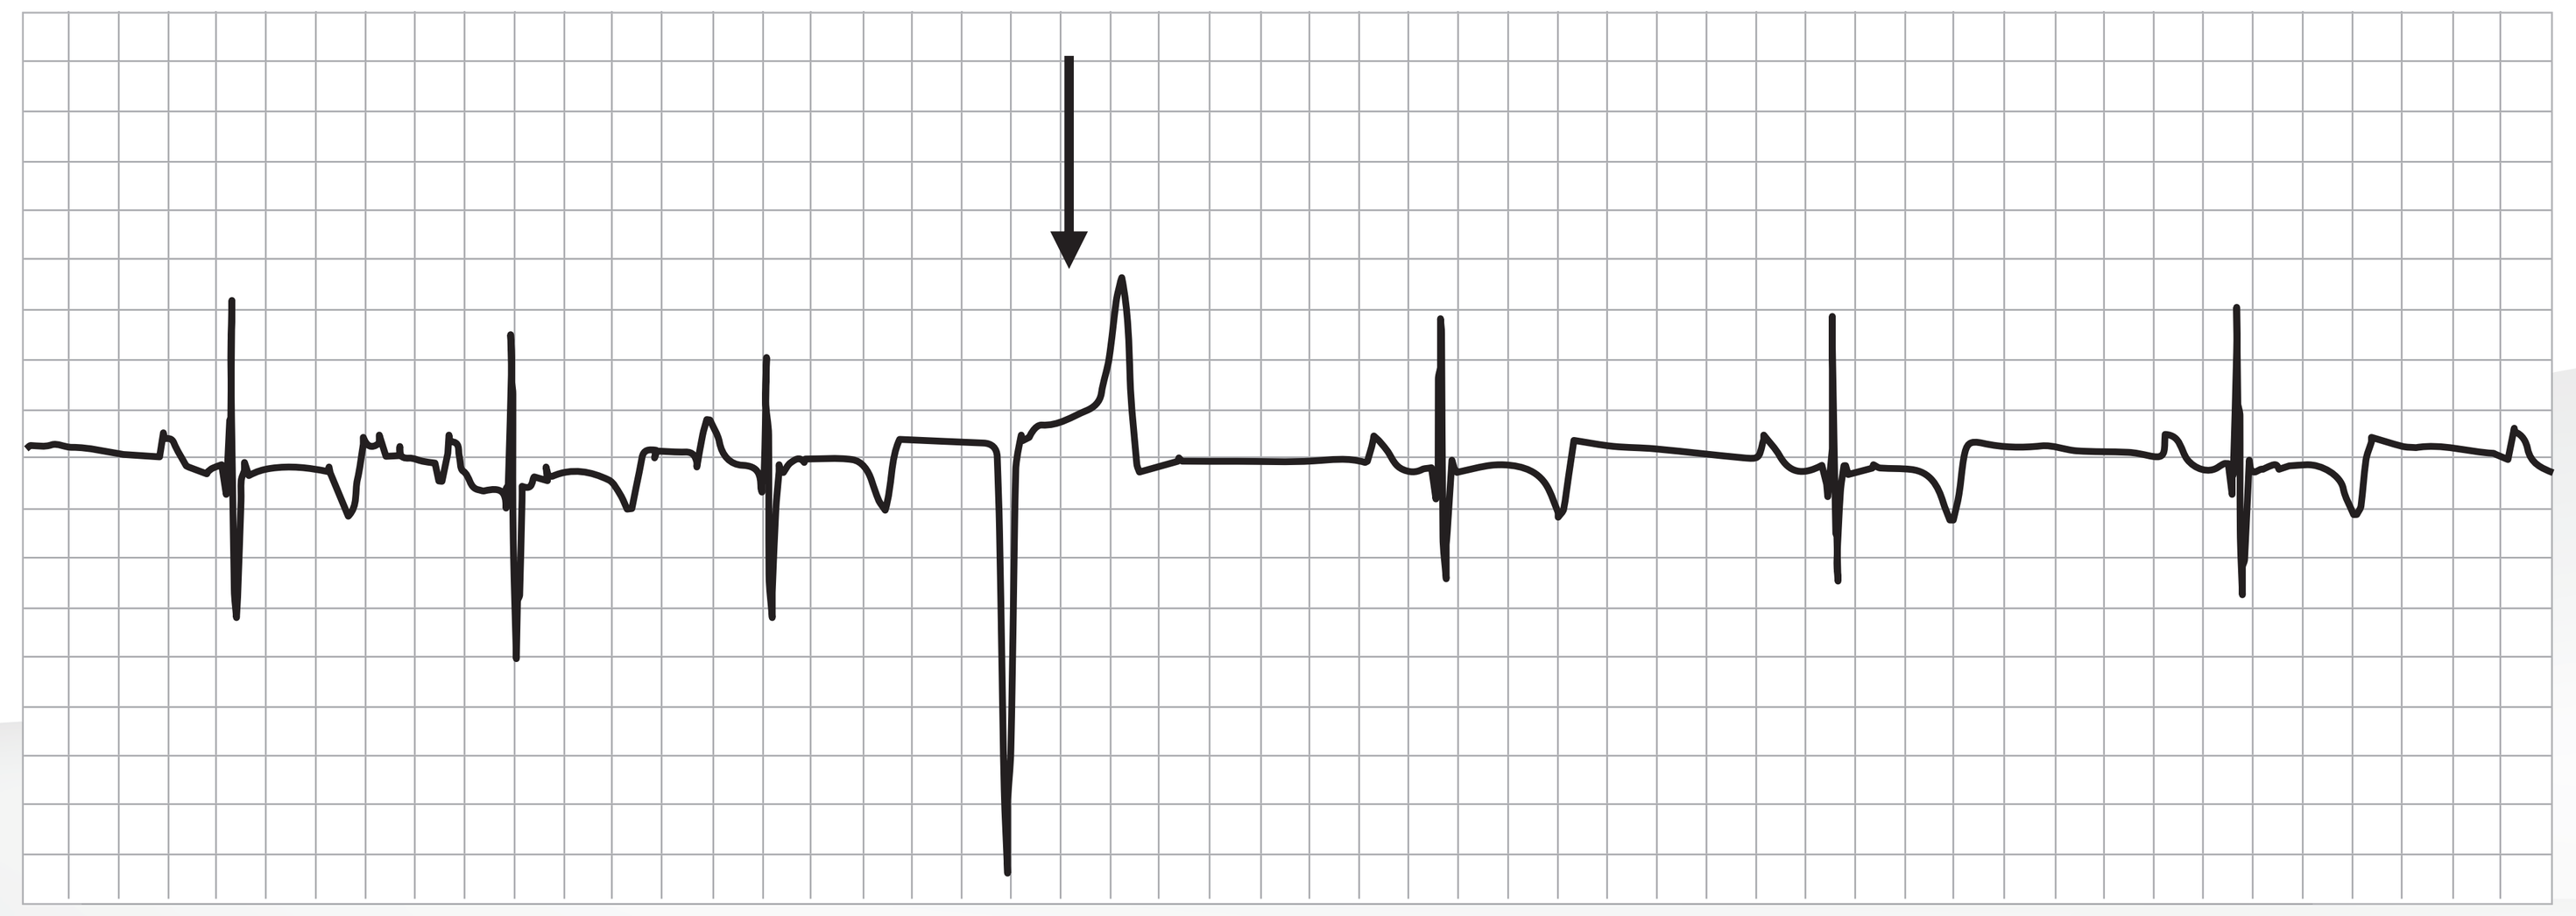

Figure 6 Atrial Premature Complex (APC) in Bama Minipigs of General Toxicology Studies

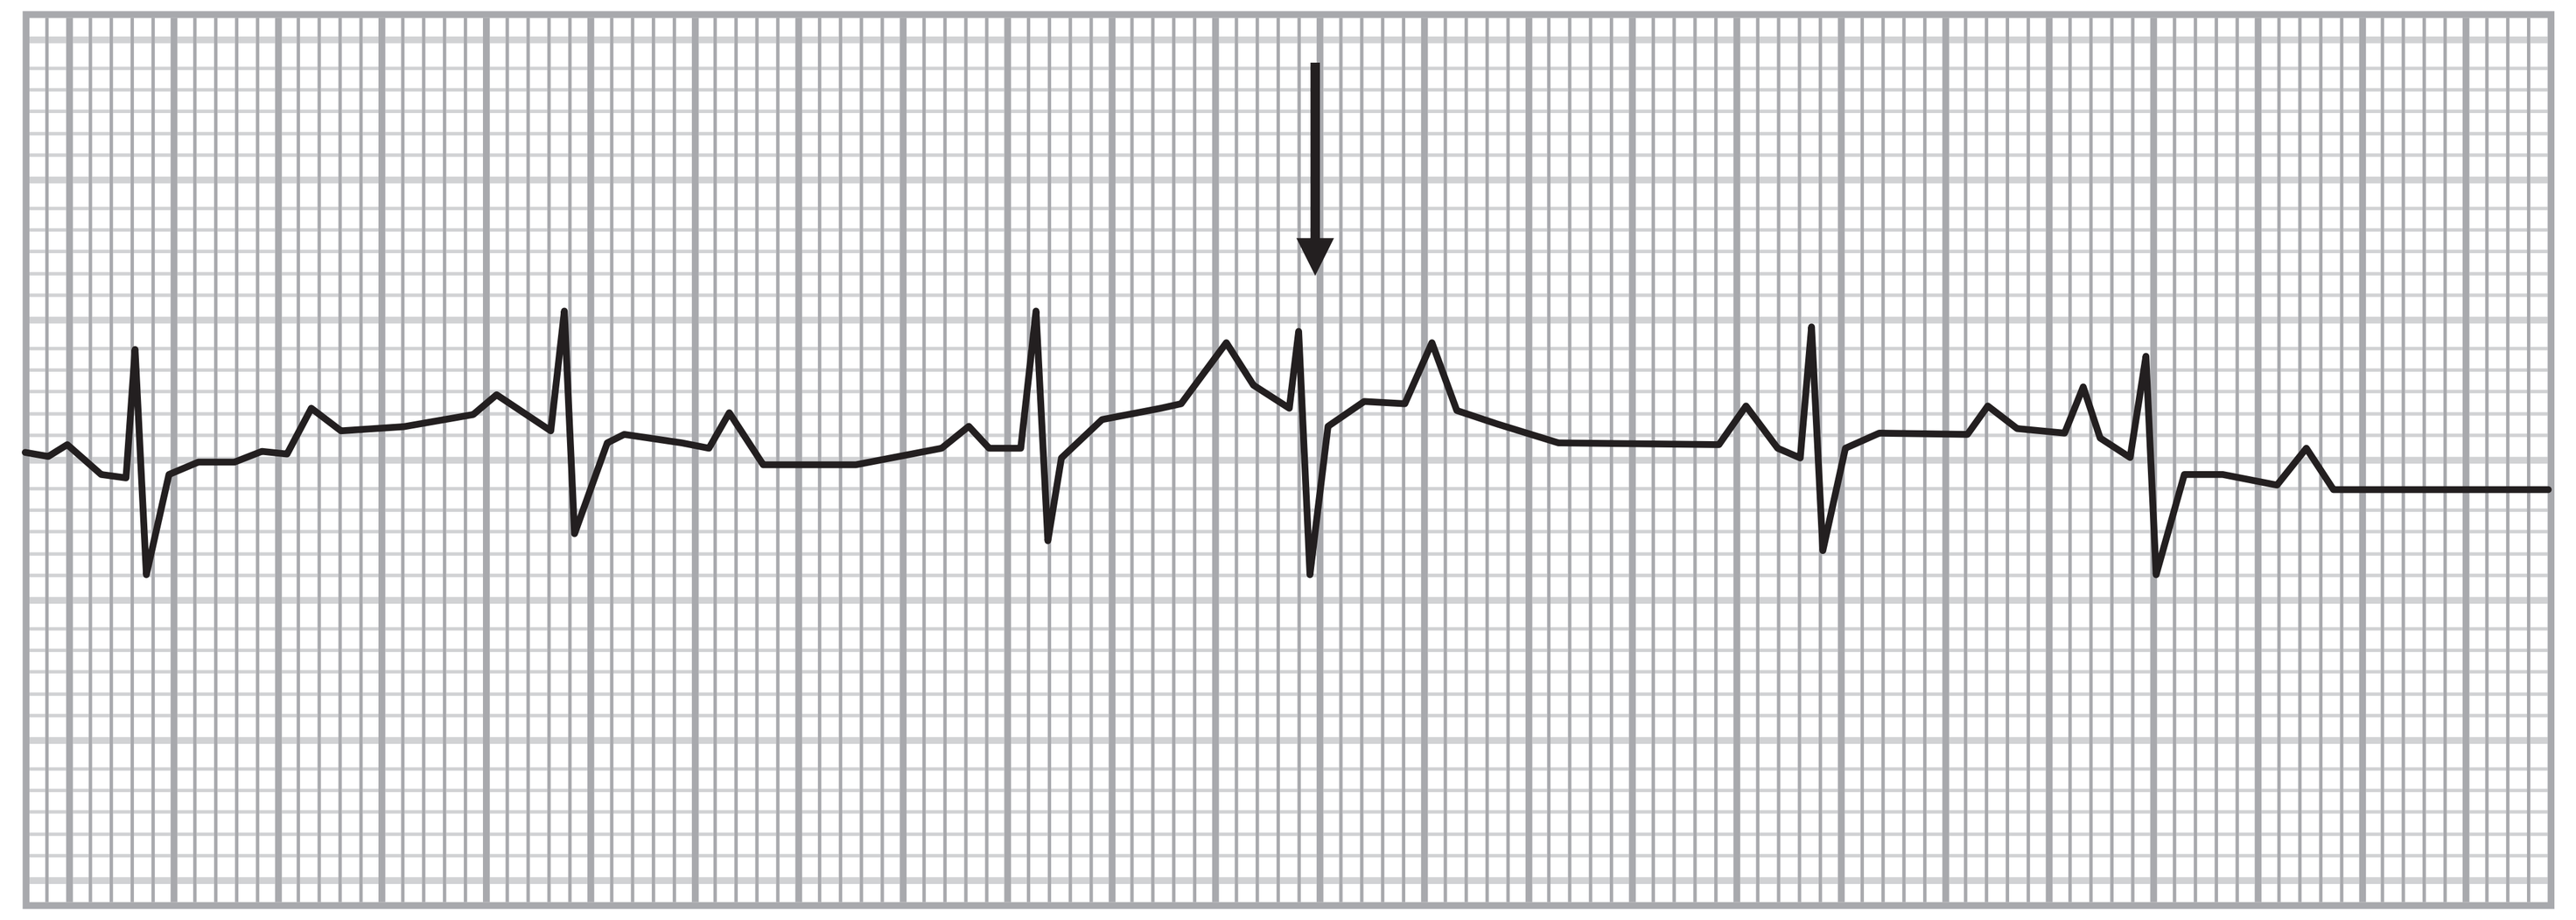

Figure 7 Ventricular Premature Complex (VPC) in Bama Minipigs of General Toxicology Studies

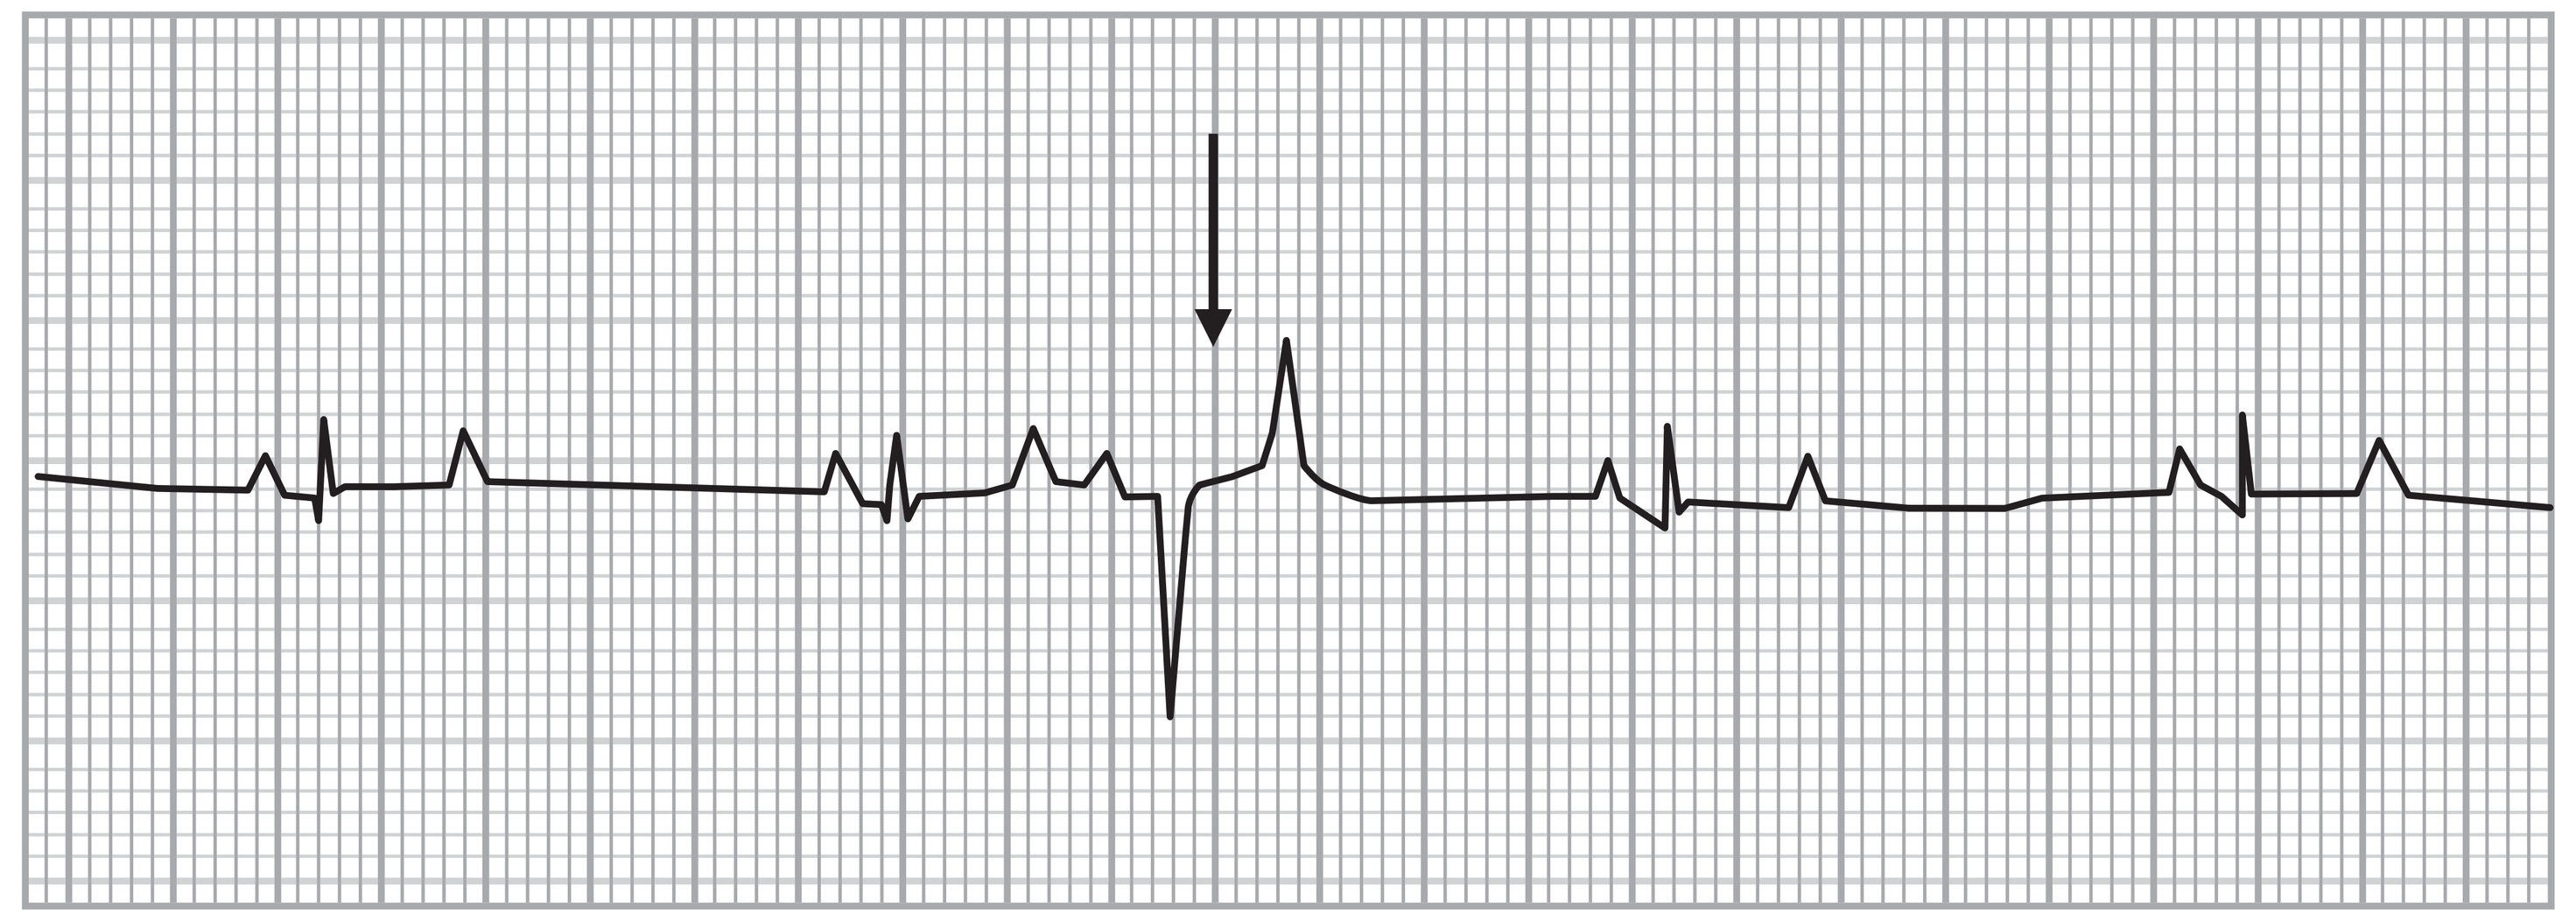

Conclusion

Under the condition of these data collections, the following conclusions were made.

In stand-alone safety pharmacology studies, the incidence of VPCs in Bama minipigs dosed with vehicle controls was 8%. APCs and VPCs had approximately the same incidence of 1% in general toxicology studies. Gender differences and the time of occurrence in the incidence of arrhythmias cannot be determined due to the limited amount of data. The infrequent VPCs and APCs are inherited from the Bama minipigs.