Comparison of the Dermal Histopathological Structure Between the Bama and Göttingen Minipigs

Xixing Zhao, Tiansheng Zhou, Sue McPherson
WuXi AppTec, China

Abstract: 3114

Abstract

The use of the minipig as a non-rodent species in non-clinical research has continued to increase and although its use is still considerably less than that of the dog or the non-human primate (NHP), for certain areas of research, the physiological similarities between swine and humans make it the species of choice. A recent European RETHINK project has taken place to assess minipigs (in general not as a specific species) as models for the toxicity testing of new medicines and chemicals and concluded that there is great utility in their use in various areas of research. The minipig is often the species used for dermal studies. Swine skin shows many similarities to human skin. This has resulted in the extensive use of swine in burn and radiation research as the skin of other research animals such as rodents and dogs differs significantly from that of man. The epidermis of these animals is much thinner with a flat epidermal-dermal interface which does not have rete ridges and papillary projections, with relatively loose dermal structures and an underdeveloped vascular system. In comparison, swine skin is composed of three layers: epidermis, dermis, and subcutis. The epidermis contains the 5 strata seen in human skin while the dermis contains superficial venules and arteriolar plexuses, collagen, elastic fibers, hair follicles, sebaceous glands, and apocrine sweat glands and the subcutis contains loose connective tissue and fat tissue. A comparison of skin samples (H & E stained) taken from the back of Göttingen and Bama minipigs showed that the structure of the two strains was very similar. The only difference of note was a slight difference in thickness of the dermis (slightly thicker in the Göttingen animals), but this may be related to the age of the animals examined. In conclusion, both Bama and Göttingen minipigs have a similar skin histopathological structure and their structures are comparable to humans.

Materials and Methods

- Bama Miniature Swine / Bama Pig, conventional level
 Pearl Laboratory Animal Sci. & Tech. Co., Ltd. 3 Huanhu Road, Songshan Lake Sci. & Tech. Industrial Park, Dongguan, Guangdong province, China
 - The animals were maintained as controls in general toxicity studies
- Housing - Animals were socially housed (2 to 3 animals of the same sex and same dosing group together) per facility SOP in stainless steel cages equipped with a metal bar type floor and an automatic watering valve
- Feeding regimen - minipig feed from contract vendor, twice daily)
- All studies were maintained under the same conditions: Temperature 18 - 26°C, Humidity 40 - 70% and Light Cycle 12 hours light and 12 hours dark (except when TK or other investigations were being performed).

Eight normal control pigs (4/sex) were used for the comparison, this number was deemed sufficient to determine conclusive findings.

Tissue Collection and Processing

Back skin samples were collected according to SOP/Study Protocol. Tissues were trimmed and fixed in 10% neutral buffered formalin (NBF). Tissues were removed from fixative and embedded in paraffin, sectioned and stained with hematoxylin and eosin and examined microscopically.

Back skin HE slides from 8 normal control Gottingen minipigs (4/sex) were kindly provided by Charles River Laboratories at Mattawan, MI.

Results

A comparison of skin samples taken from the back of 8 Göttingen and Bama minipigs showed that the structure of the two strains was very similar. Other than the difference of note of slight difference in thickness of the dermis (slightly thicker in the Göttingen animals), this may be related to the age of the animals examined. These two breeds have similar microscopic morphology, architecture, and elements in the skin. These observations support the validity of using Bama pigs in skin toxicity as an alternative for Gottingen pigs.

Male Bama Pig 10x

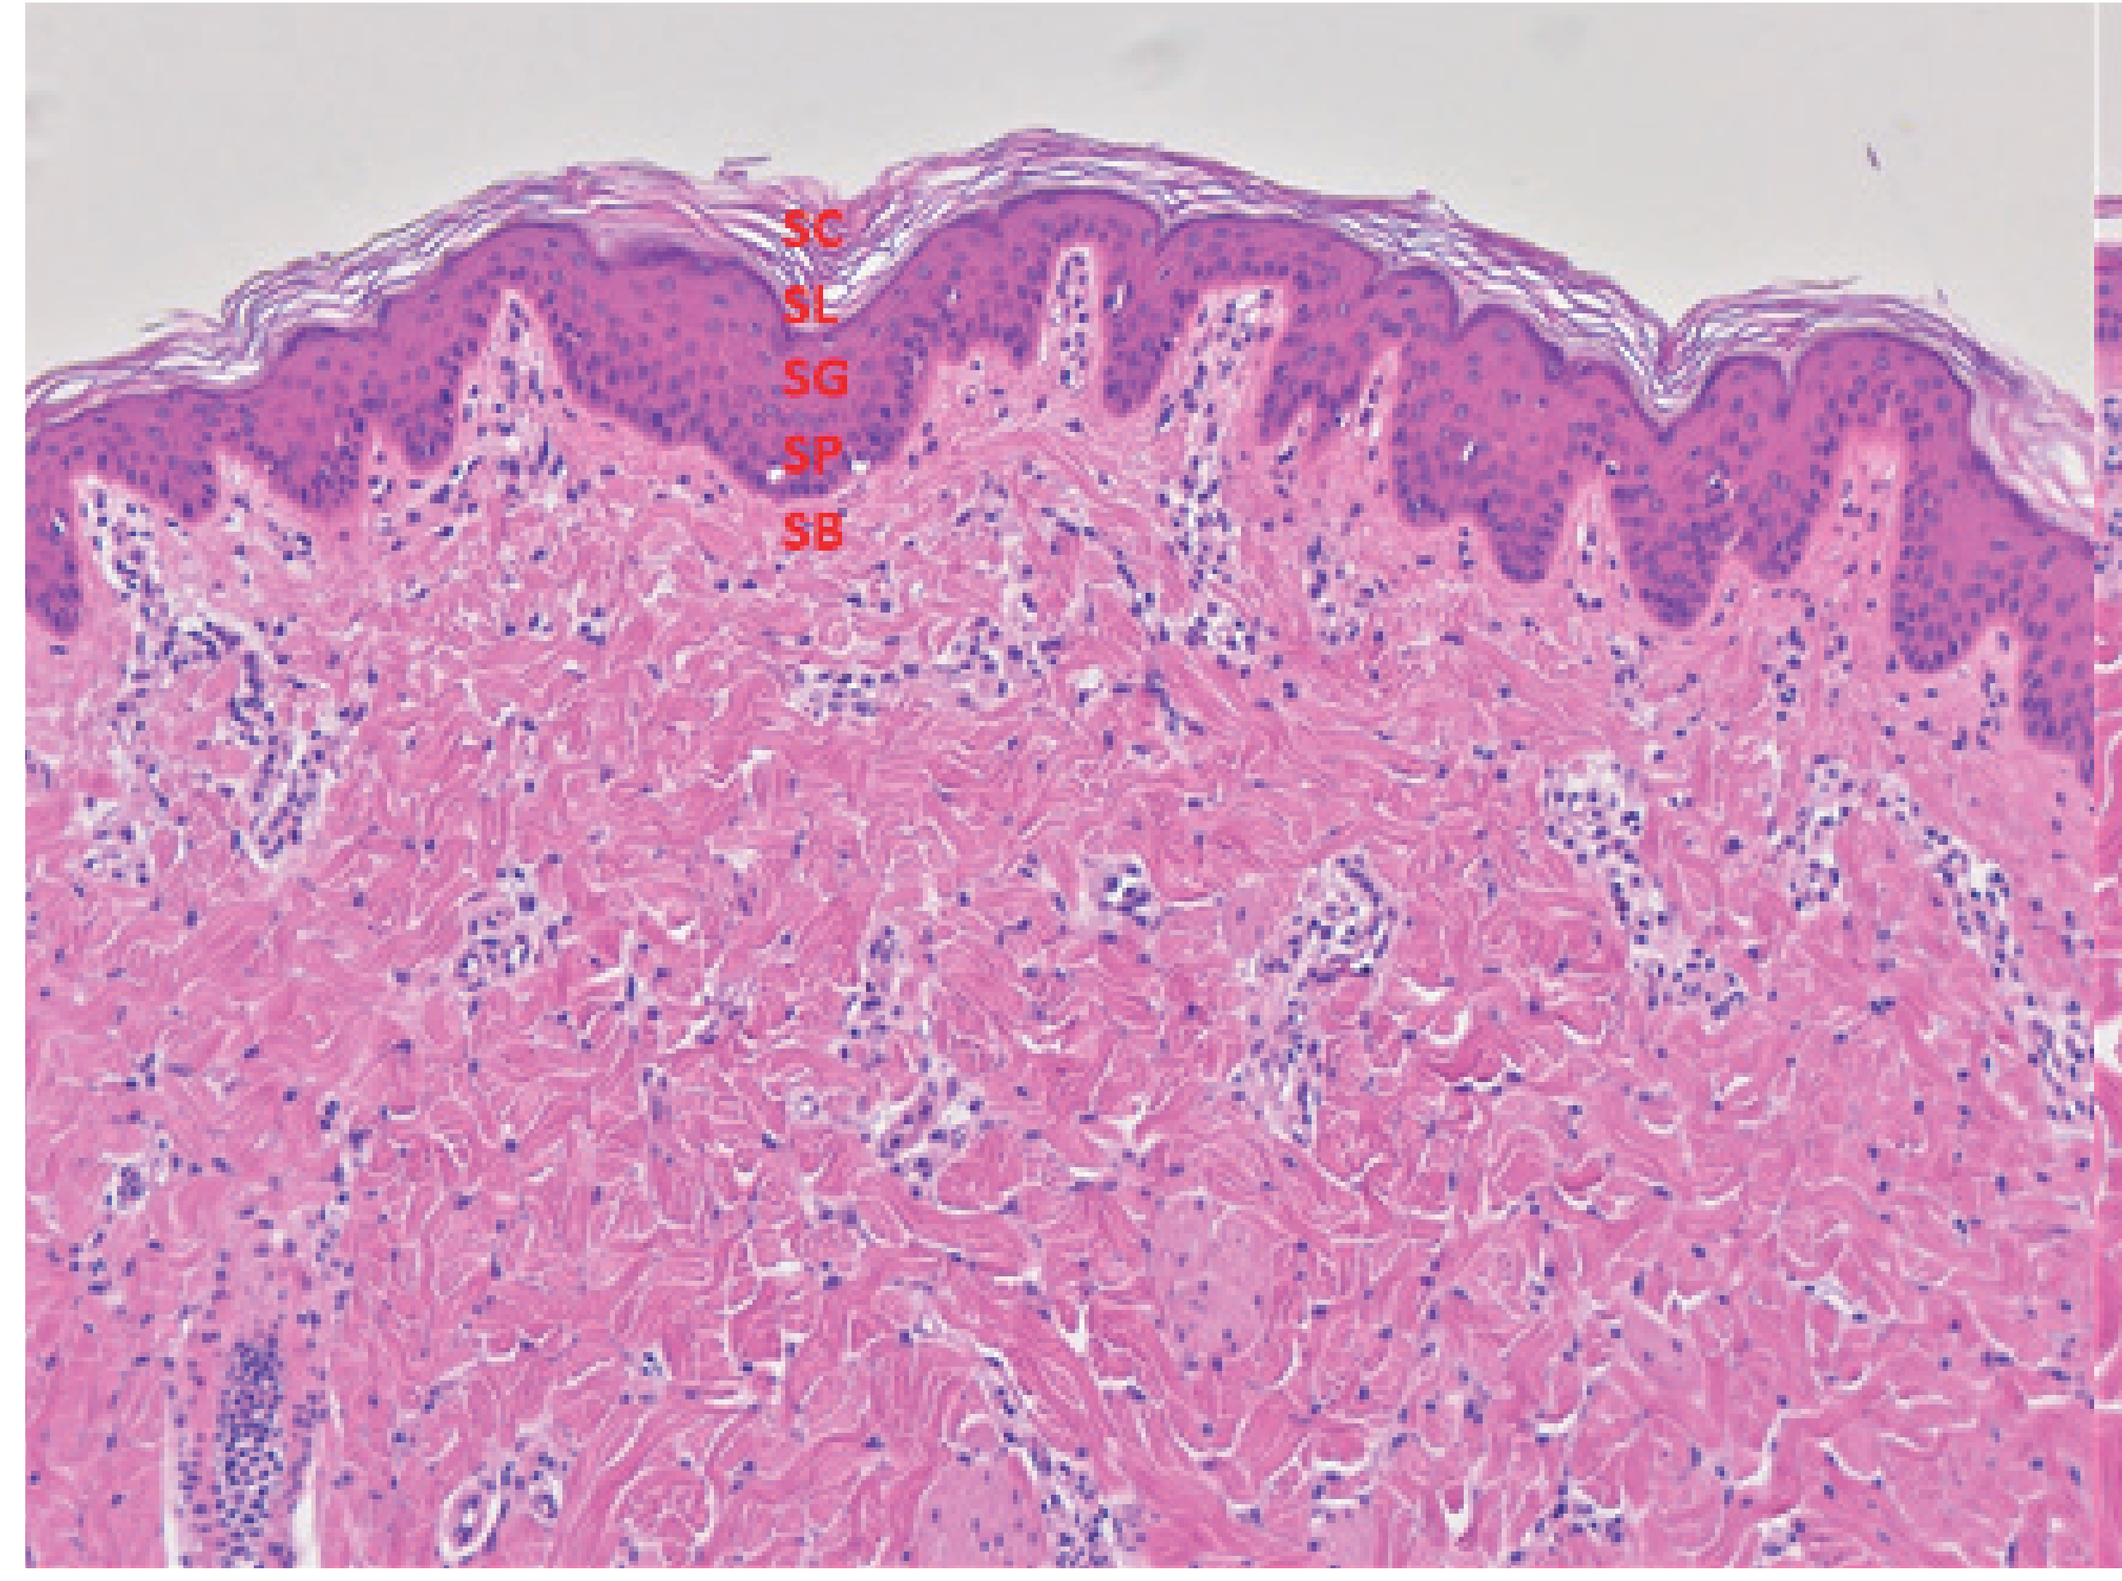
SC
SL
SG
SP
SB

Male Gottingen Pig 10x

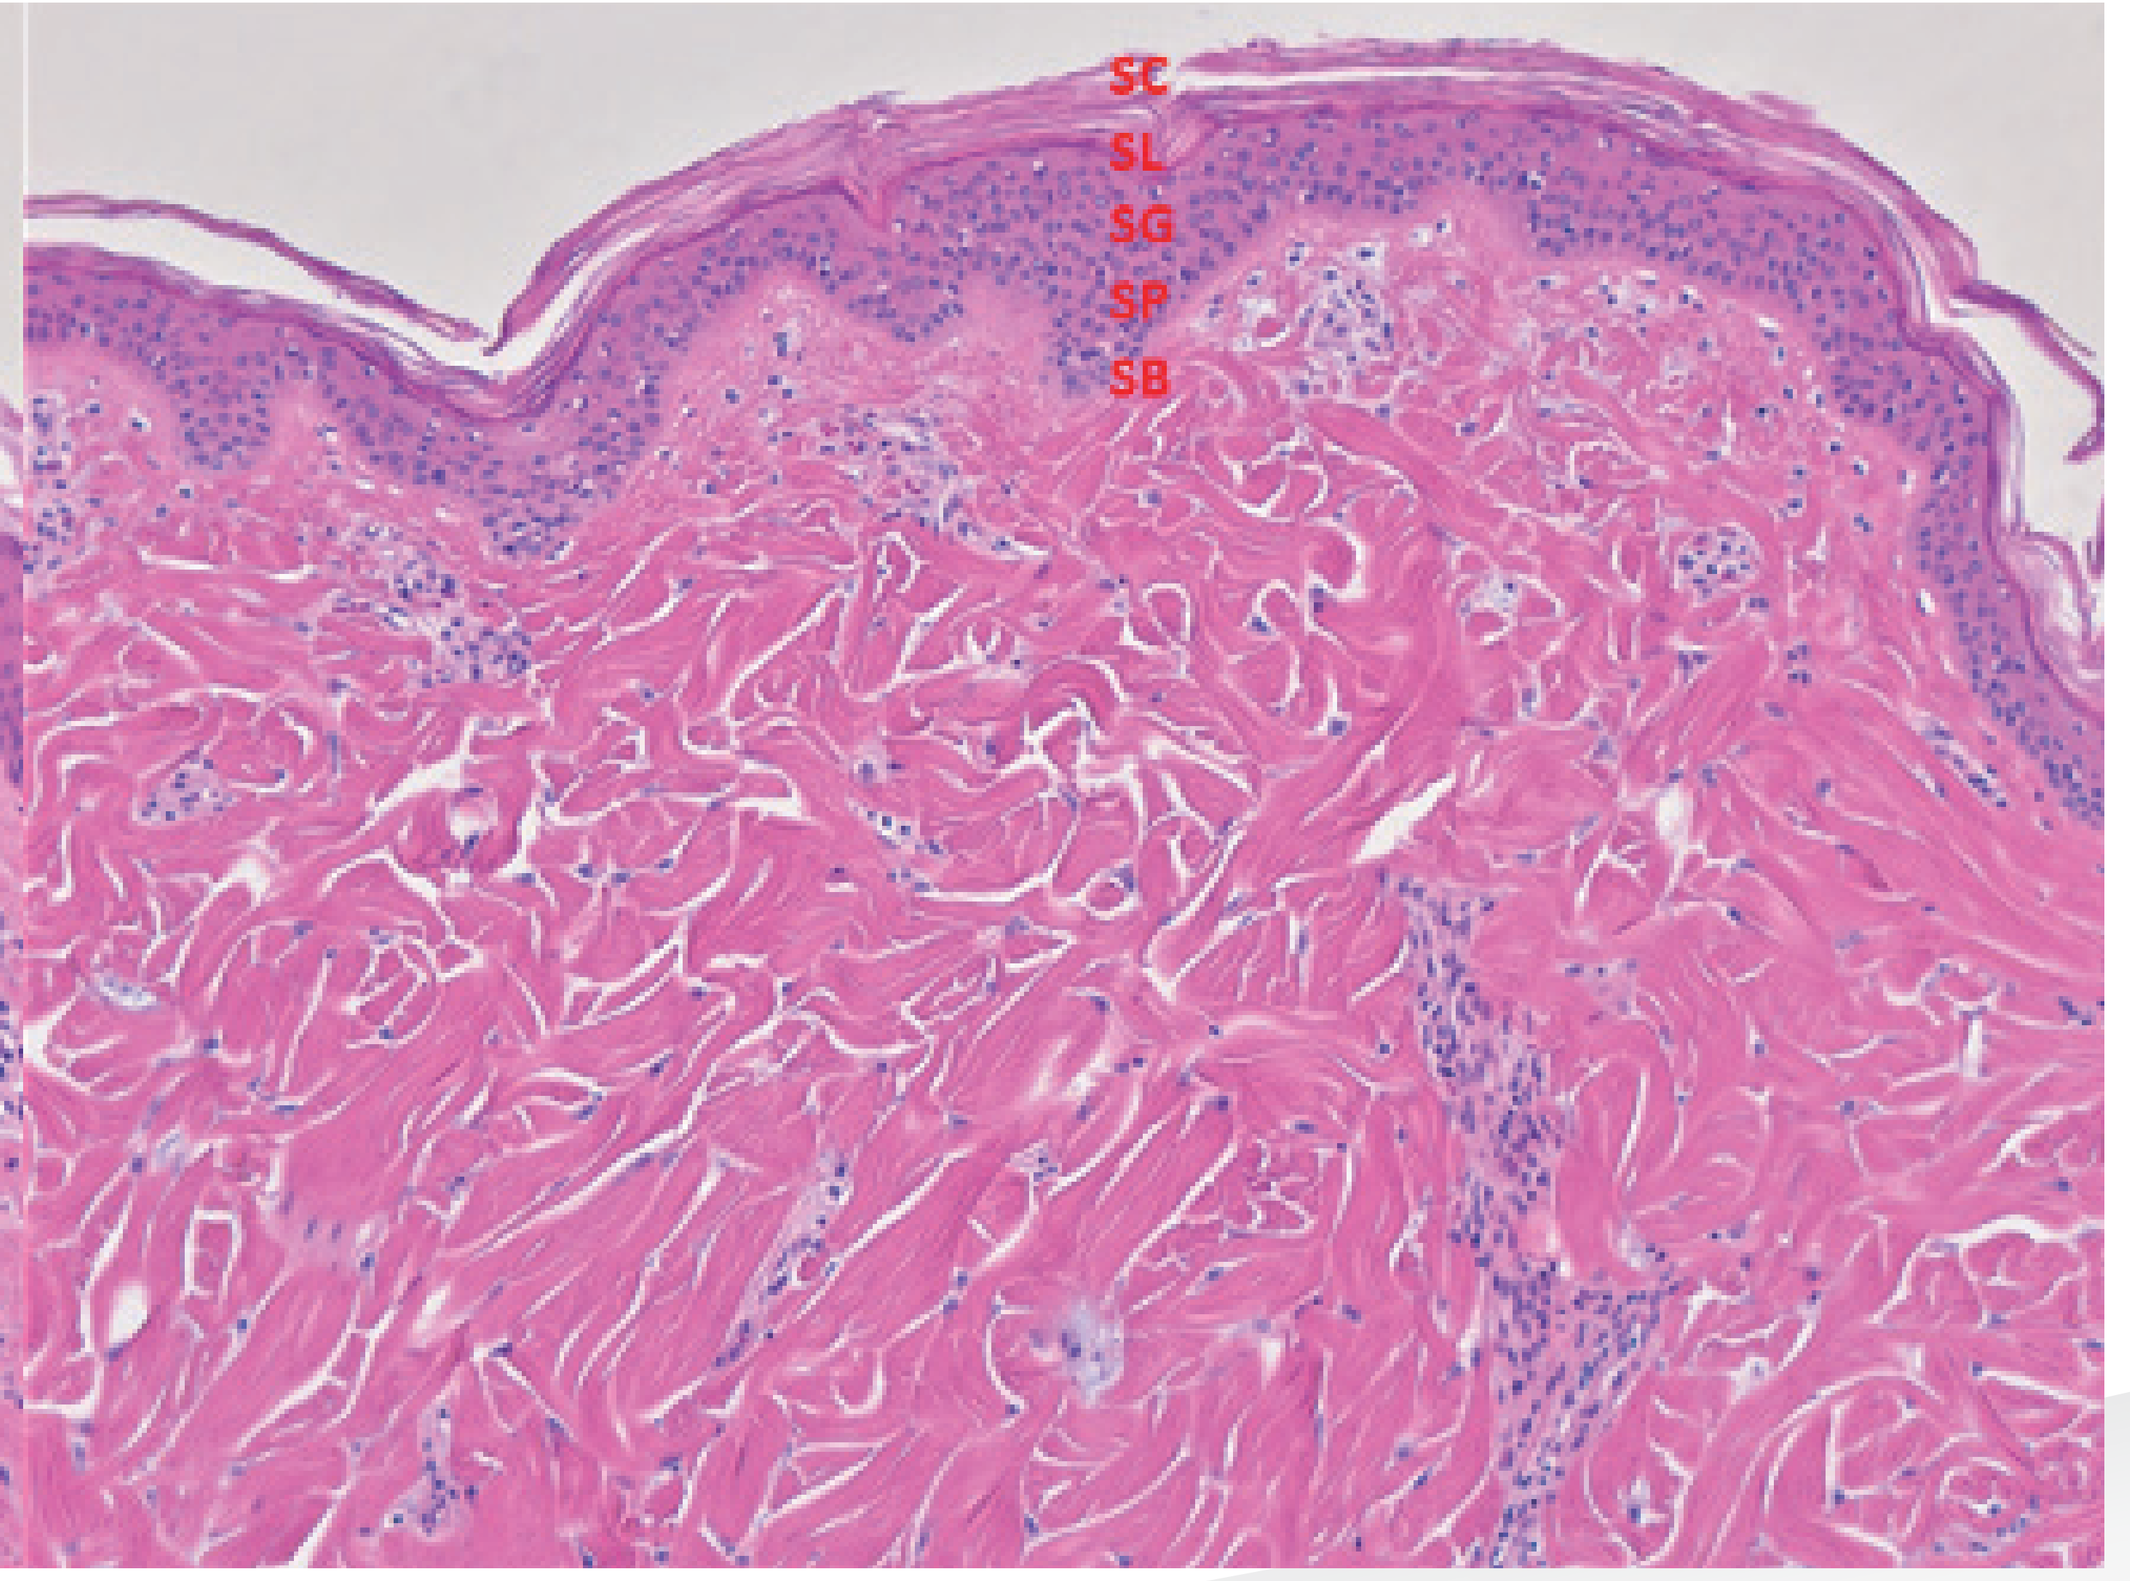
SC
SL
SG
SP
SB

SC=Stratum corneum; SL=Stratum lucidum, SG=Stratum granulosum; SP=Stratum spinosum; SB=Stratum basale

Acknowledgments

The authors would like to acknowledge the contributions made by the technical group at WuXi AppTec.