A Retrospective Analysis of Internal In Vitro Phototoxicity Data

Liwen Gao; Mengqi Zhang; Xiaohui Zhu; Ke Xu; Ning Xu; Millie Chen
WuXi AppTec

Poster #: 403

Purpose

The 3T3 Neutral Red Uptake Phototoxicity (NRU-PT) assay is an established in vitro assay used to evaluate the potential phototoxicity of a test article. Since recommended by EMA and ICH guidance, the 3T3 NRU-PT assay has been used quite extensively in the pharmaceutical industry. However, there are some limitations of this assay including high false positive rate and compound solubility issue. To better understand the assay, we retrospectively analyzed the data generated in our laboratory.

Methods and Materials

The in vitro 3T3 neutral red uptake (NRU) phototoxicity assay was conducted in compliance with ICH S10 and OECD TG432. The NRU phototoxicity studies conducted at WuXi AppTec 2017 to 2022 were collected for the retrospective analysis.

The NRU phototoxicity assay is based on a comparison of the cytotoxicity of a test article when tested in the absence and in the presence of exposure to a non-cytotoxic dose of simulated solar light.

The mouse fibroblast cell line, BALB/c 3T3 with a population doubling time of 20 to 25 hours will be employed in this assay. BALB/c 3T3 cells (abbreviated as 3T3) were originally obtained from the American Type Culture Collection (ATCC), Manassas, VA, USA. The cell stocks are stored in liquid nitrogen. Every batch of the cell stocks will be tested and determined to be free from mycoplasma contamination. Irradiation sensitivity of cells will be determined for each batch of the cell working stocks. Cells will not be used after the passage number 100.

Complete medium will be DMEM medium, supplemented with 10% new born calf serum, 4 mM L-glutamine, 100 units/mL penicillin, and 100 μg/mL streptomycin. BALB/c 3T3 cells will be cultured in a humidified incubator of 7.5% CO_2 in air at 37C.

Once the criteria for a valid assay are met, responses observed in the assay are evaluated as follows: If suitable concentration-response curves are obtained for the test article in the absence (-Irr) and presence (+Irr) of irradiation, the PIF is used to assess the phototoxic potential. If PIF value is not obtained, the MPE will be used to assess the phototoxic potential.

1. A test article is considered to be "Phototoxic" in this assay if PIF values of ≥5 (or MPE values ≥0.15, where appropriate) are obtained.
2. A test article is considered to be "Non-phototoxic" in this assay if PIF values of <2 (or MPE values of <0.1, where appropriate) are obtained.
3. A test article is considered to be equivocal phototoxic in this assay if PIF values of ≥2 and <5 (or MPE values of ≥0.1 and <0.15) are obtained. The MPE value is used to further support the conclusion if based on the PIF value:
 - MPE values ≥0.15 indicate a phototoxic response.
 - MPE values <0.1 indicate a non-phototoxic response.
 - MPE values ≥0.1 and <0.15 indicate an equivocal phototoxic response.

Figure 1 96-microwell plate for the 3T3 NRU phototoxicity assay

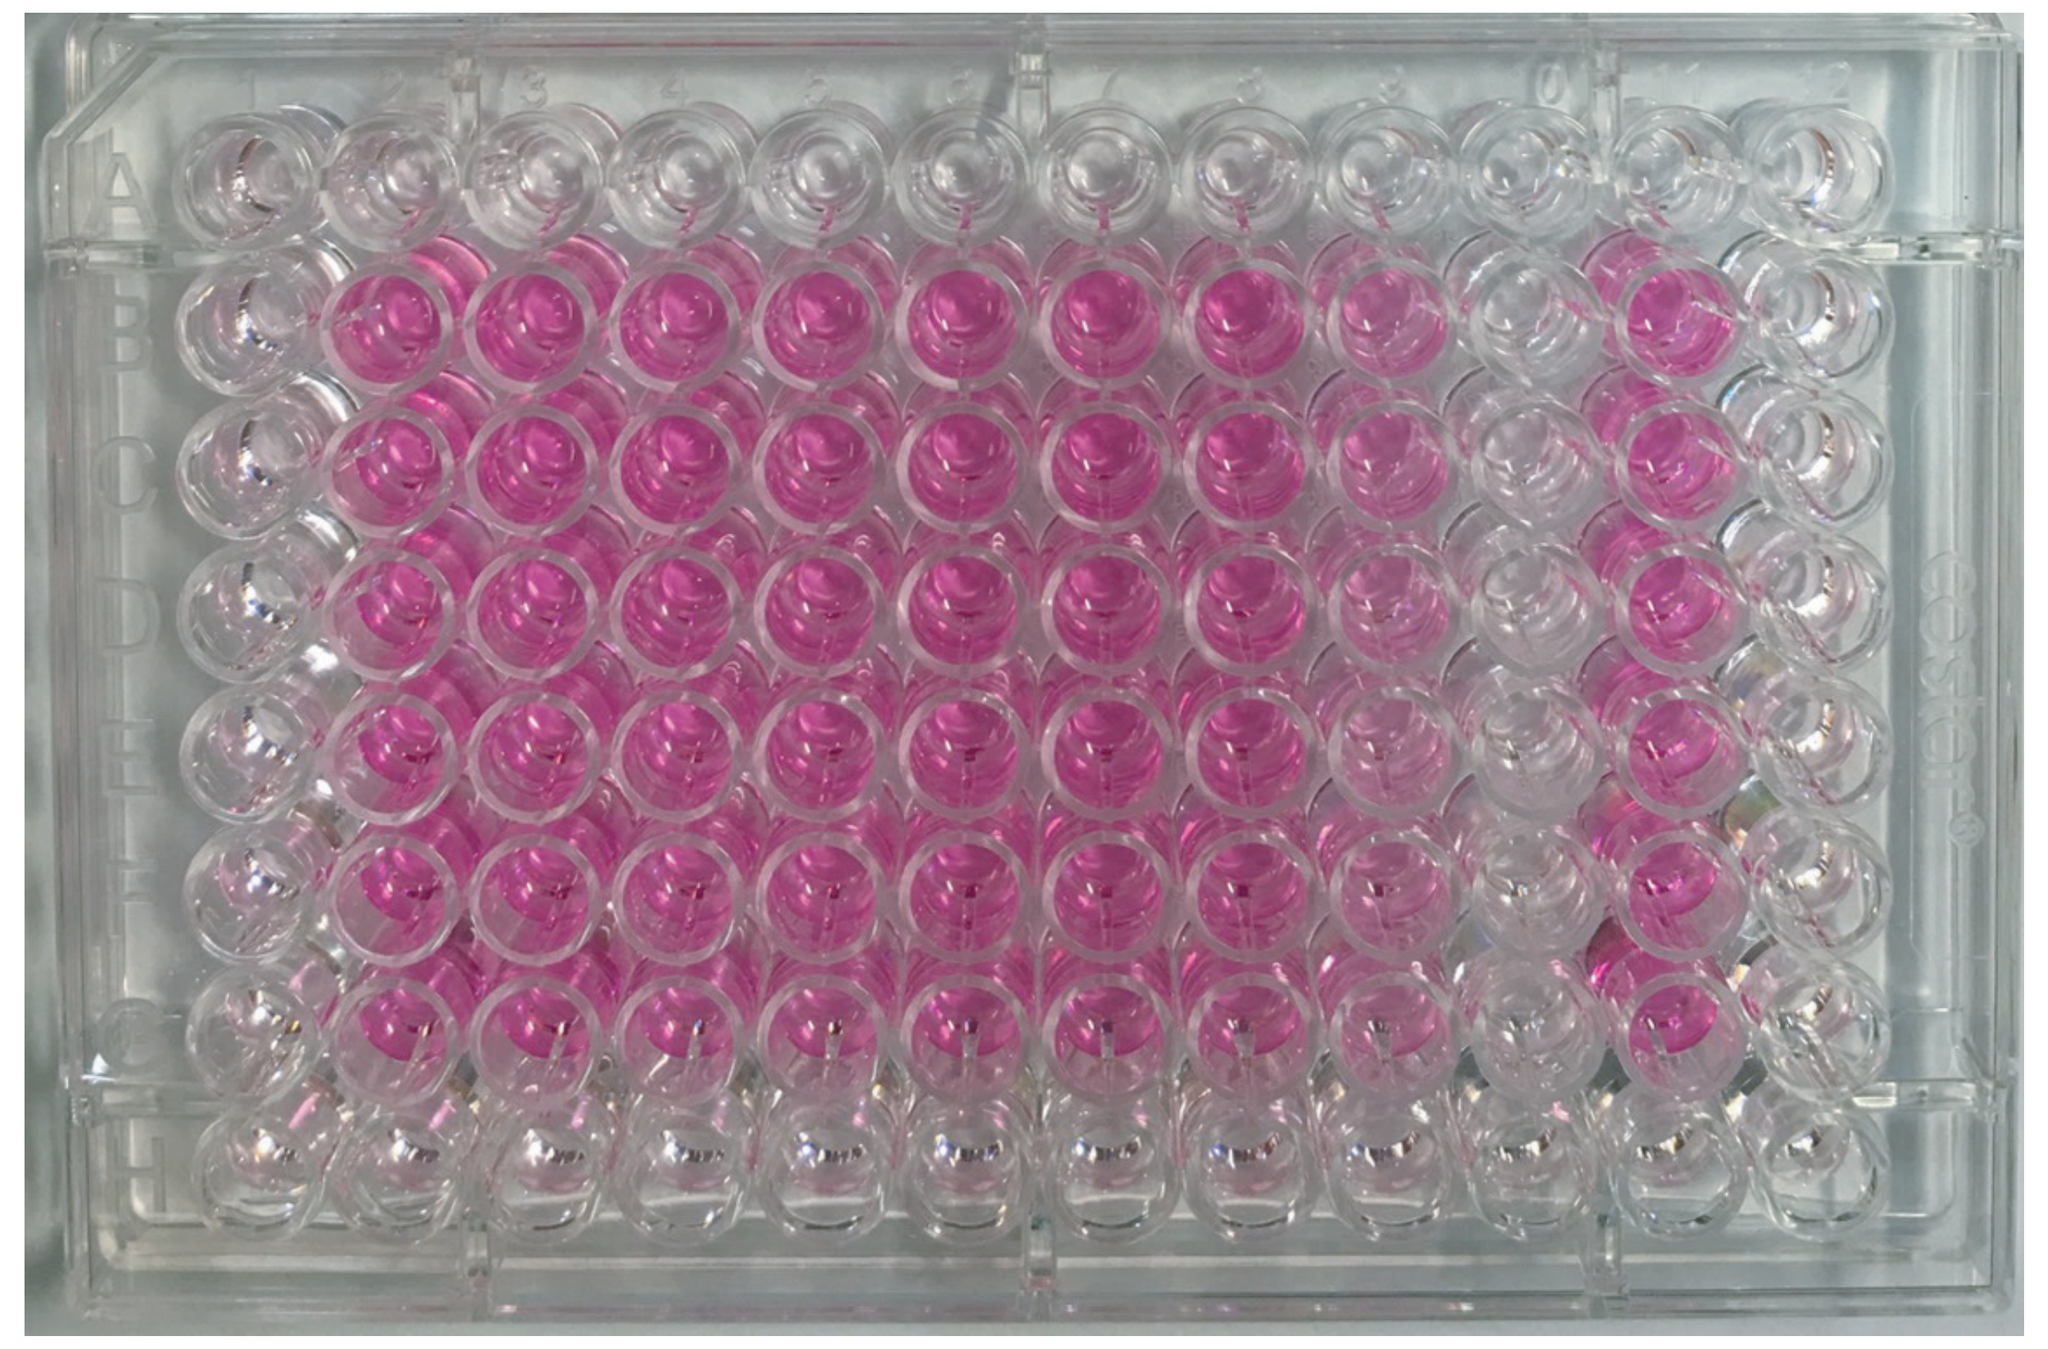

Results and Discussion

Data were collated for 100 compounds from 2017 to 2022. Of these, 52 compounds produced a photo irritation factor (PIF) value <2 (or mean photo effect, MPE value of <0.1) and were considered negative (52%), 8 compounds produced a PIF value of 2 to 5 (or MPE value of 0.1 to 0.15) and were considered equivocal (8%), whilst 40 compounds produced a PIF value ≥5 (or MPE value ≥0.15) and were considered positive (40%). Thus, the overall positive/equivocal ratio in the 3T3 NRU-PT assay is approaching to 50%, which is consistent with the value reported in the literatures. Among these studies, only 28% of final calls were made based on PIF values, most conclusions (72%) were made based on the MPE values, since the IC50 values were not determined due to solubility issue (43%) or low toxic effect (29%).

Based on these results, it appears that this assay is substantially oversensitive for predicting photosafety hazard (as opposed to high negative predictivity) and MPE is a good supplementary parameter if no IC50 values can be derived.

Experience from the Pharmaceutical Industry

In Vitro Phototoxicity (n = 361)

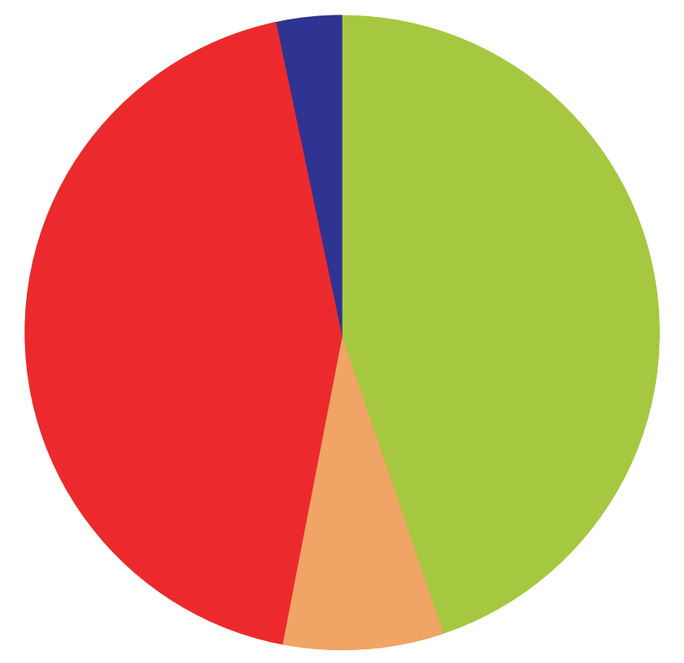

Figure 2 EFPIA photosafety survey based on 361 compounds from 10 labs: PIF<2, negative (45% green); PIF>5, positive (44% red); PIF 2-5, equivocal (8% orange); and not tested (3% blue). Cited from Lynch and Wilcox, Exp Toxicol Path, 2011.

In Vitro Phototoxicity (n = 100)

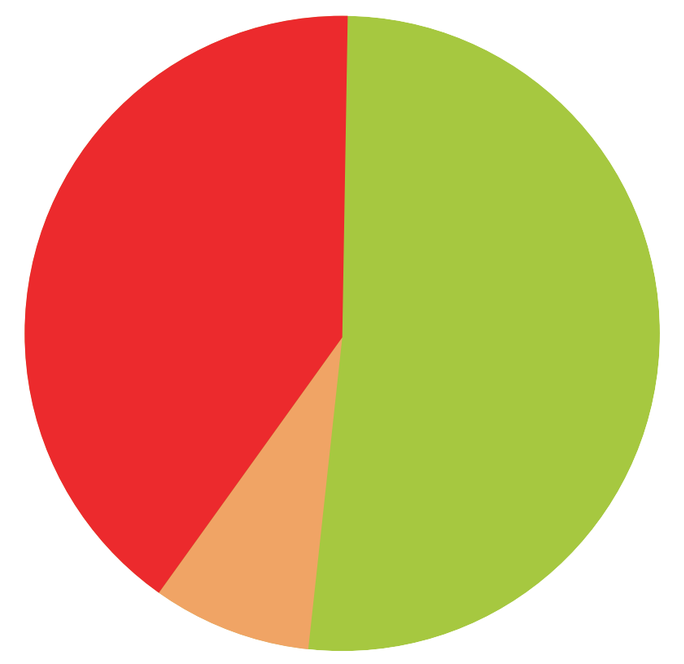

Figure 3 An internal retrospective analysis based on 100 compounds from WuXi AppTec: PIF<2, negative (52% green); PIF ≥ 5, positive (40% red); PIF 2-5, equivocal (8% orange).

MPE is a good supplementary parameter

Figure 4 Only 28% of final calls were made based on PIF values, most conclusions (72%) were made based on the MPE values, since the IC50 values were not determined due to solubility issue (43%) or low toxic effect (29%).

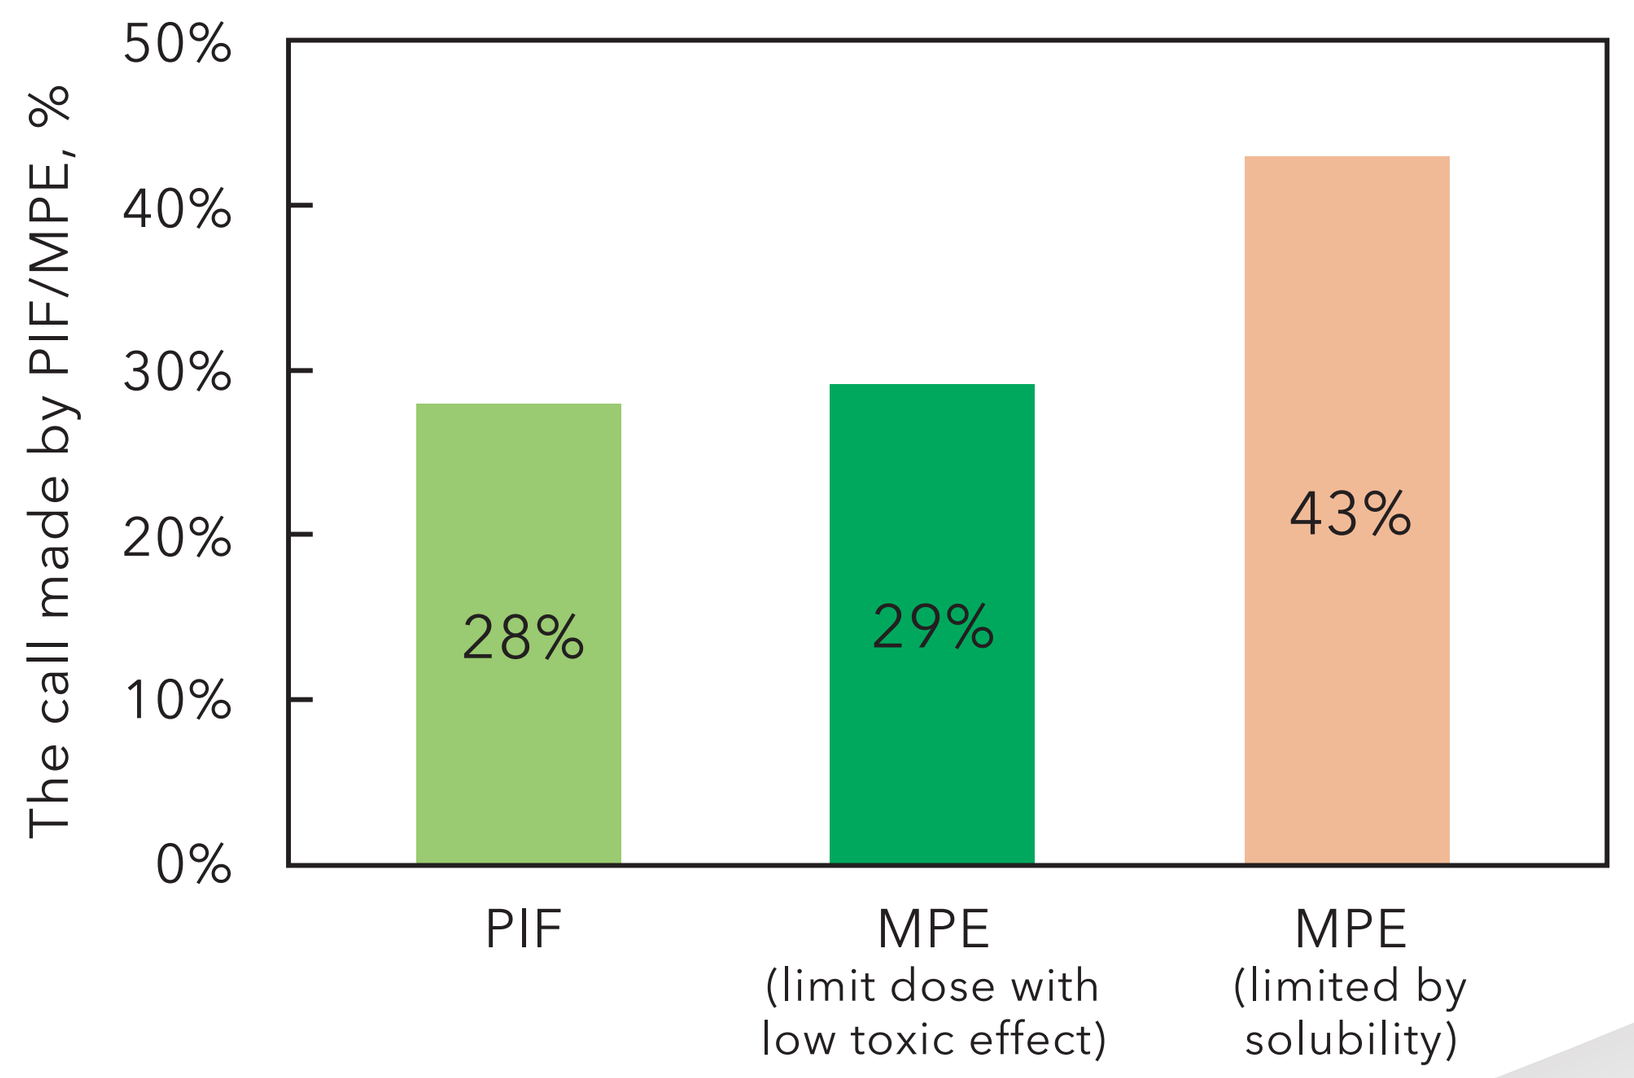
The call made by PIF/MPE, %
50%
40%
30%
20%
10%
0%
28%
29%
43%
PIF
MPE (limit dose with low toxic effect)
MPE (limited by solubility)

Acknowledgements

The authors would like to acknowledge the contributions made by the Genetic and In Vitro Toxicology group at WuXi AppTec.