Comparison of Routine Parameters of Tg.rasH2 Mice and Non-Tg.rasH2 Mice

Lei Guo; Yan Zhang; Zhimei Wang; Sue McPherson
WuXi AppTec

药明康德 WuXi AppTec

Poster #: 118

Abstract

Tg.rasH2 mice are popular and sensitive models for rapid carcinogenicity testing of a test article, in which carcinogenic potential is determined within 26 weeks[1]. The Tg.rasH2 mice are hemizygous transgenic mice carrying the human prototype c-Ha-ras gene with its own promoter/enhancer, confers an unusually high susceptibility to tumor formation in Tg.rasH2 mice. Generally, 28-day dose range finding (DRF) studies are conducted in non-Tg.rasH2 mice, the wild type littermates of Tg.rasH2 mice, to establish the dosages for the 26-week Tg.rasH2 carcinogenicity study. Although closely related, the non Tg.rasH2 mouse has the same genetic background as the Tg.rasH2 mouse except for the omission of the transgene part, it is still important to understand two species' differences. Therefore, a comparison of routine parameters, including body weights, food consumption, hematology and serum chemistry parameters between Tg.rasH2 mice and non Tg.rasH2 mice was conducted in this study. The results from the two data sets were analyzed using non-Tg.rasH2 mice as reference control point. For body weight and food consumption, data were collected from the 28-day DRF studies in non-Tg.rasH2 mice and the first 4-week of the 26-week carcinogenicity studies in Tg.rasH2 mice. The body weight of Tg.rasH2 mice were lower than the non-Tg.rasH2 mice for about 15% in males and 10% in females, which correlated with lower food consumption (generally 20% to 30%) for Tg.rasH2 mice relative to non-Tg.rasH2 mice. The difference in body weight was similar to the reported data[2]. The hematology and serum chemistry data collected at the end of the 28-day DRF studies in non-Tg.rasH2 mice and at the end of the 26-week carcinogenicity studies in Tg.rasH2 mice were analyzed. Although these data were collected at different timeslot, they also had some significance for studying the differences between two species. The hematology data showed remarkable lower white blood cell count (WBC) and lymphocytes (LYMP) in both sexes of Tg.rasH2 mice when compared to non-Tg.rasH2 mice. Differences in clinical chemistry were restricted to relatively higher Total Bilirubin (TBIL) in male Tg.rasH2 mice, and lower alkaline phosphatase (ALP) and glucose in both sexes of Tg.rasH2 mice relative to non-Tg.rasH2 mice. It is strongly recommend to consider the above differences in body weight, food consumption and a couple of clinical pathology parameters between Tg.rasH2 mice and non-Tg.rasH2 mice when estimating the dosages for the 26-week carcinogenicity studies in Tg.rasH2 mice using the 28-day DRF studies in non-Tg.rasH2 mice.

Methods and Materials

- Tg.rasH2 mouse: CByB6F1-Tg(HRAS)2Jic (tg/wt)
 Non Tg.rasH2 mouse: CByB6F1-Tg(HRAS)2Jic (wt/wt)
 - CLEA Japan, Inc.
 - Approximately 7 weeks old at study initiation
- Data of control animals from completed toxicology studies were used
- Housing
 - Social housing- Females were pair- or trio-housed in solid bottom cages, with corncob bedding and water bottle
 - Individual housing- Males were housed individually in solid bottom cages, with corncob bedding and water bottle
- Feeding regimen- Rodent feed supplied by Beijing Keao Xieli Co., Ltd, ad libitu (except when investigations were being performed)
- All studies were maintained under the same conditions: Temperature 20°C to 26°C, Humidity 40% to 70% and Light Cycle 12 hours light and 12 hours dark.
- In-life data
 - Body weight: data were collected once weekly
 - Food consumption: data were collected once weekly
- Laboratory investigations
 - Blood samples for hematology and clinical chemistry evaluations were obtained from abdominal aorta
 - Hematology: K2EDTA
 - Serum Chemistry: Plain with separating gel
- Instrumentation
 - Hematology- Advia 2120 Hematology Analyzer
 - Serum Chemistry- Hitachi 7180 Biochemistry Analyzer
- Reporting and Analysis
 - Provantis V10.5.0.0

Results and Discussion

Hematology and serum chemistry: The hematology and serum chemistry data collected at the end of the 28-day DRF studies in non-Tg.rasH2 mice and at the end of the 26-week carcinogenicity studies in Tg.rasH2 mice were analyzed. Although these data were collected at different timeslot, they also had some significance for studying the differences between two species. The hematology data showed remarkable lower white blood cell count (WBC) and lymphocytes (LYMP) in both sexes of Tg.rasH2 mice when compared to non-Tg.rasH2 mice. Differences in clinical chemistry were restricted to relatively higher Total Bilirubin (TBIL) in male Tg.rasH2 mice, and lower alkaline phosphatase (ALP) and glucose in both sexes of Tg.rasH2 mice relative to non-Tg.rasH2 mice.

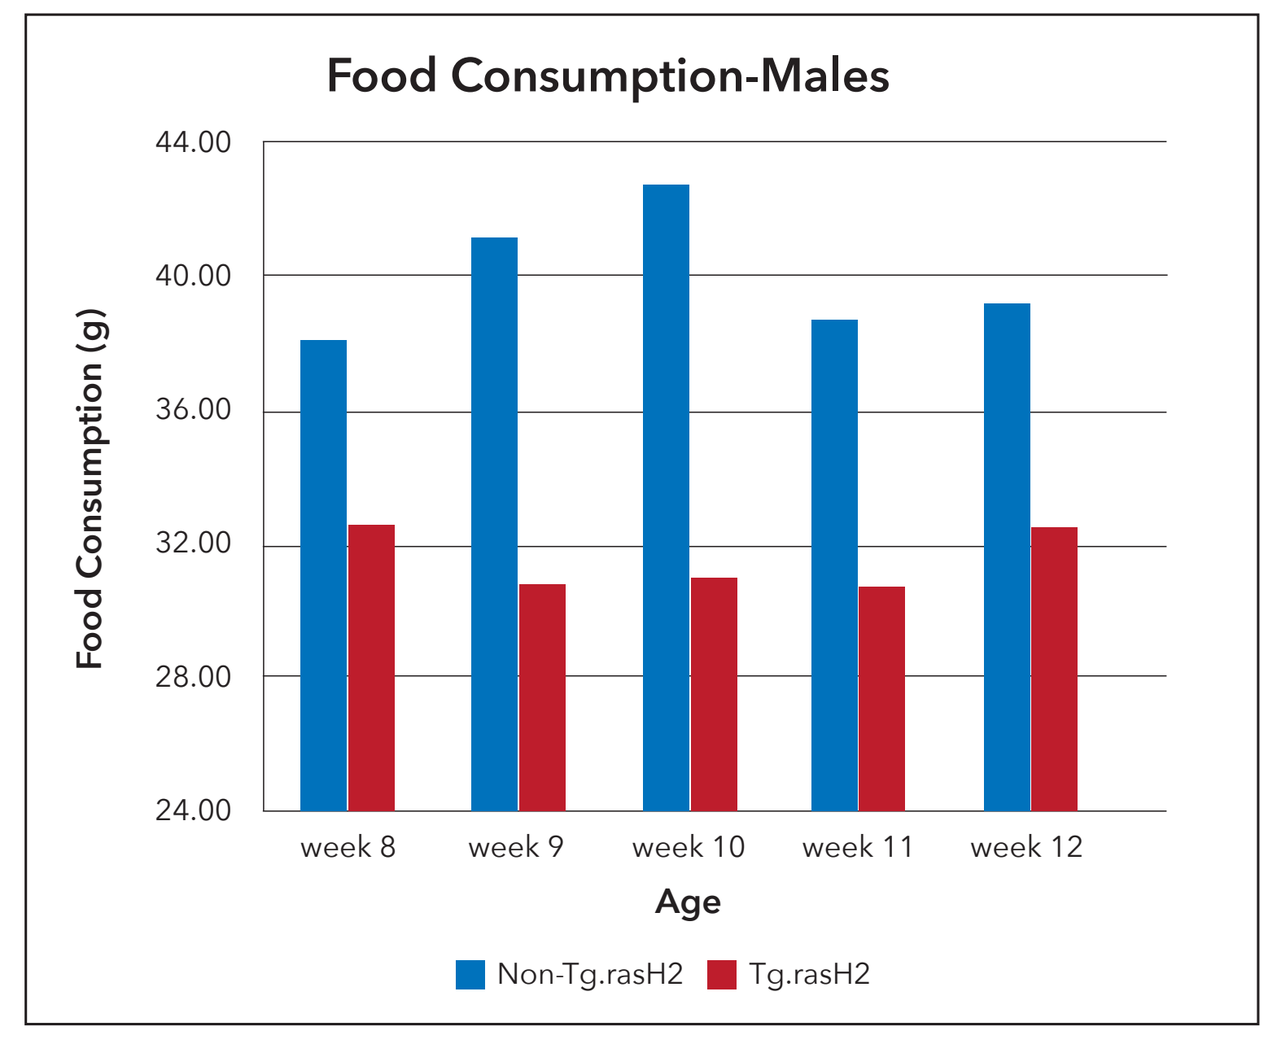
Food Consumption-Males
Food Consumption (g)
44.00
40.00
36.00
32.00
28.00
24.00
week 8
week 9
week 10
week 11
week 12
Age
Non-Tg.rasH2
Tg.rasH2

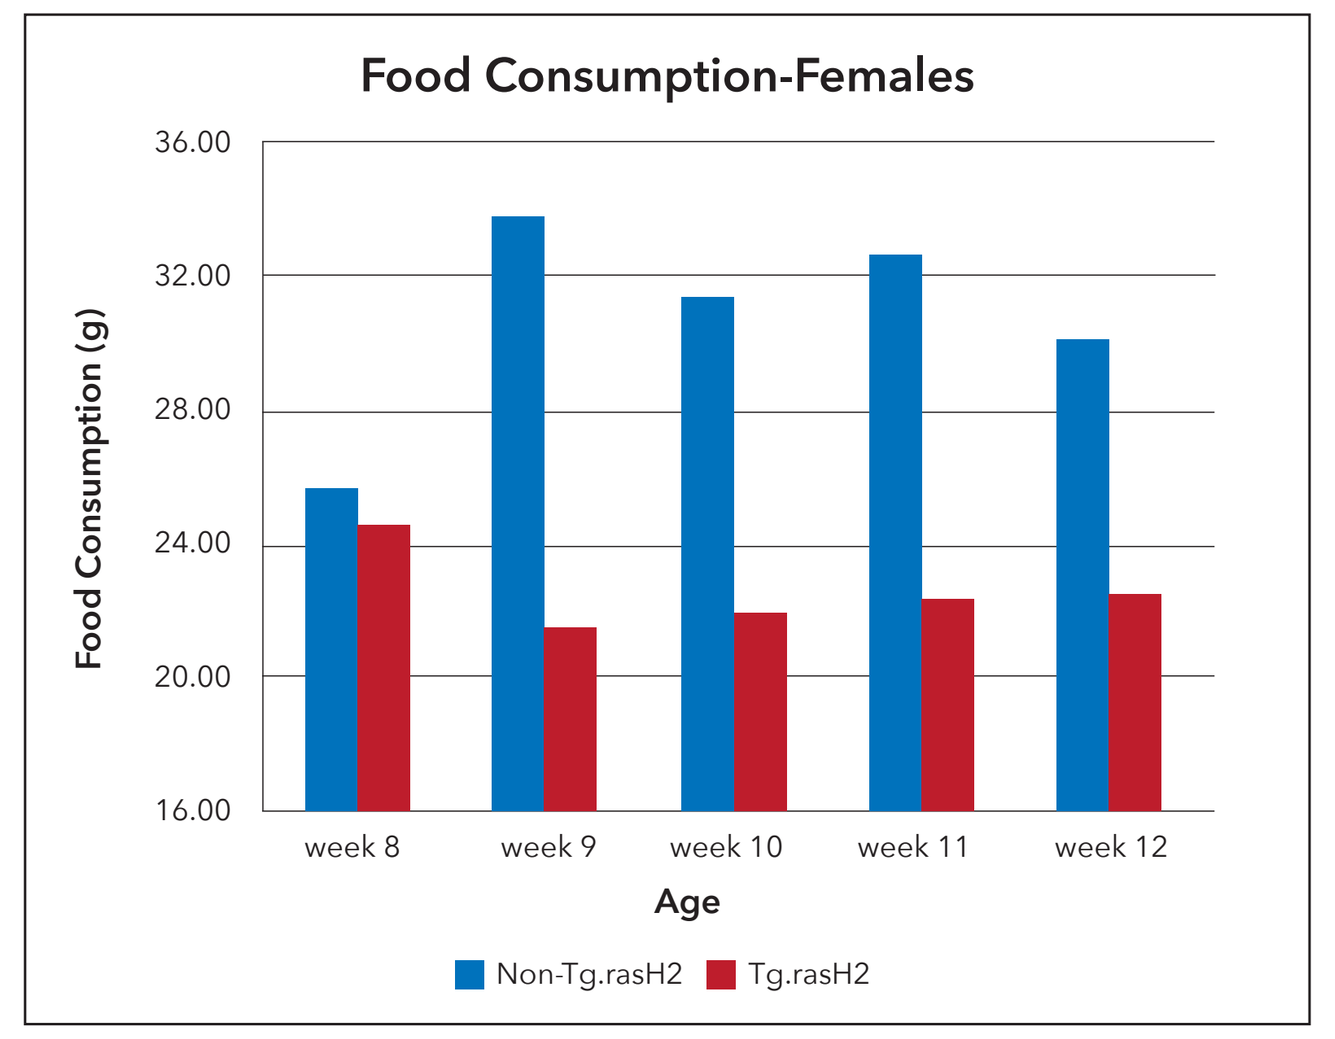
Food Consumption-Females
Food Consumption (g)
36.00
32.00
28.00
24.00
20.00
16.00
week 8
week 9
week 10
week 11
week 12
Age
Non-Tg.rasH2
Tg.rasH2

Body weight and food consumption: Data were collected from the 28-day DRF studies in non-Tg.rasH2 mice and the first 4-week of the 26-week carcinogenicity studies in Tg.rasH2 mice. The body weight of Tg.rasH2 mice were lower than the non-Tg.rasH2 mice for about 15% in males and 10% in females, which correlated with lower food consumption (generally 20% to 30%) for Tg.rasH2 mice relative to non-Tg.rasH2 mice. The difference in body weight was similar to the reported data2.

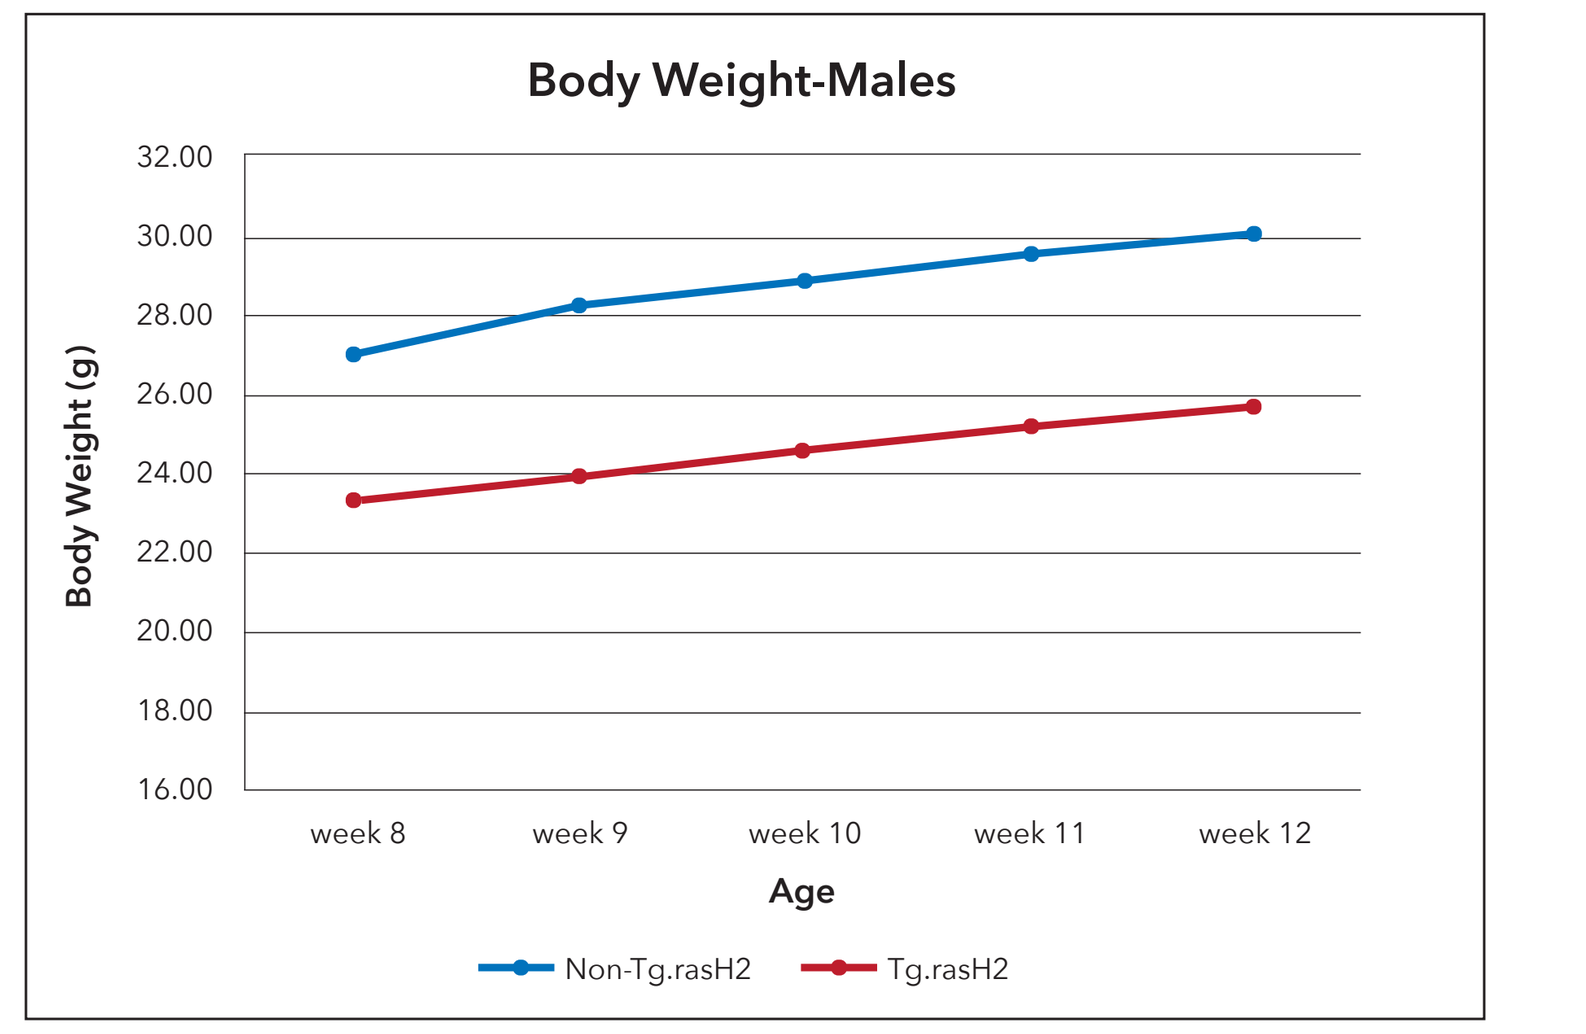
Body Weight-Males
Body Weight (g)
32.00
30.00
28.00
26.00
24.00
22.00
20.00
18.00
16.00
week 8
week 9
week 10
week 11
week 12
Age
Non-Tg.rasH2
Tg.rasH2

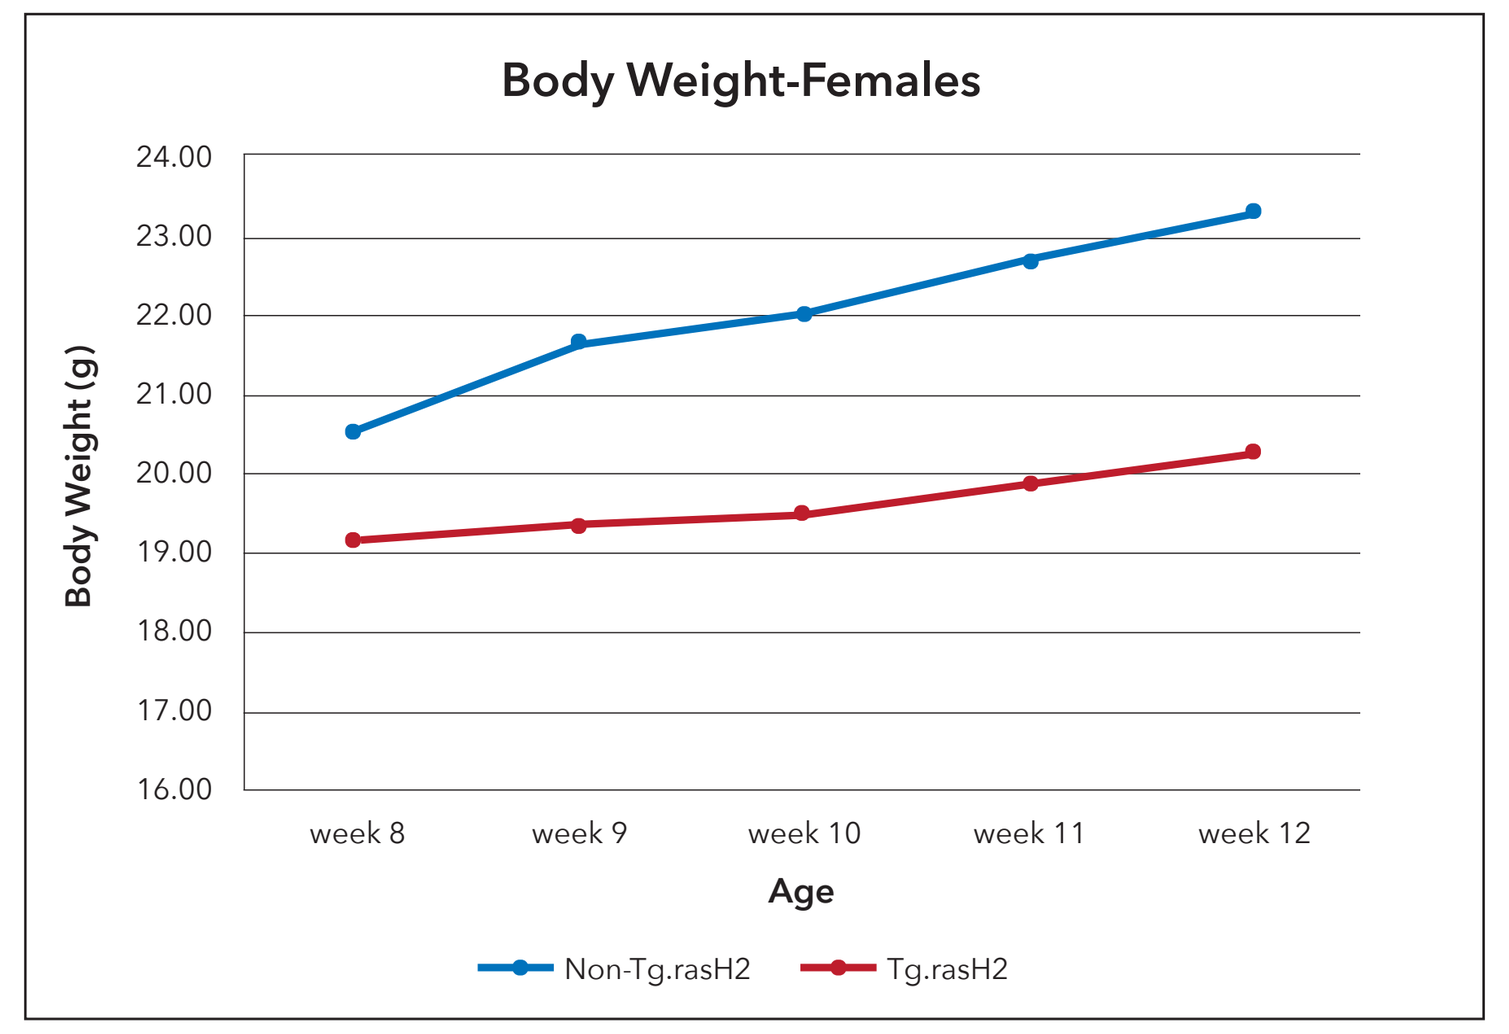
Body Weight-Females
Body Weight (g)
24.00
23.00
22.00
21.00
20.00
19.00
18.00
17.00
16.00
week 8
week 9
week 10
week 11
week 12
Age
Non-Tg.rasH2
Tg.rasH2

Hematology

Males

Parameters		WBC (x10^9/L)	RBC (x10^6/μL)	HGB (g/dL)	HCT (%)	MCV (fL)	MCH (pg)	MCHC (g/dL)	RDW (%)	RET (x10^9/L)	#NEUT (x10^9/L)	%NEUT (%)	#LYMP (x10^9/L)	%LYMP (%)	#MONO (x10^9/L)	%MONO (%)	#EOS (x10^9/L)	%EOS (%)	#BASO (x10^9/L)	%BASO (%)	PLT (x10^9/L)	MPV (fL)
non-Tg	Mean	2.63	10.34	15.72	49.14	48.32	15.44	31.98	11.88	319.98	0.38	14.82	2.16	82.08	0.03	0.82	0.04	1.38	0.01	0.42	1227.00	8.14
	SD	0.67	0.05	0.52	2.17	1.00	0.10	0.61	0.22	24.49	0.11	4.35	0.59	3.95	0.01	0.26	0.03	0.85	0.00	0.16	34.55	0.59
	N	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25
Tg	Mean	1.33	10.03	15.20	47.05	46.85	15.20	32.35	12.10	273.30	0.32	24.60	1.02	70.95	0.02	0.90	0.05	2.85	0.01	0.45	1270.00	7.95
	SD	0.09	0.11	0.20	1.55	1.05	0.10	0.55	0.10	11.20	0.03	4.90	0.13	3.65	0.01	0.20	0.02	1.15	0.00	0.05	21.00	0.45
	N	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44
	Diff%	49.43	3.02	3.31	4.25	3.04	1.55	-1.16	-1.85	14.59	15.79	-65.99	53.05	13.56	42.31	-9.76	-25.00	-106.52	0.00	-7.14	-3.50	2.33

Females

Parameters		WBC (x10^9/L)	RBC (x10^6/μL)	HGB (g/dL)	HCT (%)	MCV (fL)	MCH (pg)	MCHC (g/dL)	RDW (%)	RET (x10^9/L)	#NEUT (x10^9/L)	%NEUT (%)	#LYMP (x10^9/L)	%LYMP (%)	#MONO (x10^9/L)	%MONO (%)	#EOS (x10^9/L)	%EOS (%)	#BASO (x10^9/L)	%BASO (%)	PLT (x10^9/L)	MPV (fL)
non-Tg	Mean	3.12	10.01	15.86	48.76	48.72	15.84	32.54	11.90	289.44	0.45	14.12	2.57	82.72	0.02	0.72	0.05	1.60	0.01	0.40	1137.20	7.88
	SD	0.67	0.23	0.48	1.81	1.03	0.26	0.53	0.23	27.30	0.23	7.61	0.73	7.38	0.01	0.37	0.02	0.40	0.00	0.17	24.98	0.55
	N	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25
Tg	Mean	1.67	9.74	14.80	46.10	47.45	15.20	32.10	12.60	337.05	0.39	23.35	1.20	71.95	0.02	1.20	0.05	2.85	0.01	0.30	1118.50	7.65
	SD	0.09	0.05	0.10	1.30	1.05	0.00	0.70	0.30	20.25	0.06	4.45	0.14	4.05	0.00	0.10	0.01	0.35	0.01	0.00	79.50	0.45
	N	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44	44
	Diff%	46.41	2.70	6.68	5.46	2.61	4.04	1.35	-5.88	-16.45	13.72	-65.37	53.38	13.02	16.67	-66.67	2.17	-78.13	16.67	25.00	1.64	2.92

Chemistry

Males

Parameters		ALT U/L	AST U/L	TP g/dL	ALB g/dL	TBIL mg/dL	ALP U/L	Glucose mg/dL	UREA mmol/L	CRE μmol/L	Ca mg/dL	P mg/dL	TCHO mg/dL	TG mmol/L	K mmol/L	Na mmol/L	Cl mmol/L	GLB g/dL	A/G	CK U/L
non-Tg	Mean	29.20	55.00	52.44	30.64	1.64	95.20	12.28	9.06	9.00	2.19	2.21	2.67	0.57	4.72	149.60	112.60	21.80	1.41	118.80
	SD	4.27	14.18	1.42	0.83	0.46	9.83	1.67	0.63	1.41	0.06	0.37	0.09	0.06	0.24	1.14	1.82	0.77	0.04	60.01
	N	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25.00
Tg	Mean	26.00	55.50	54.10	30.65	1.75	66.50	9.85	8.12	8.00	2.16	2.22	2.86	0.59	4.50	152.00	113.00	23.45	1.31	114.50
	SD	1.41	2.12	1.41	1.63	0.18	6.36	0.85	1.12	0.00	0.03	0.01	0.09	0.02	0.14	1.41	1.41	0.21	0.08	28.99
	N	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47.00
	Diff%	10.96	-0.91	-3.17	-0.03	-6.40	30.15	19.79	10.47	11.11	1.46	-0.05	-7.09	-3.36	4.66	-1.60	-0.36	-7.57	7.32	3.62

Females

Parameters		ALT U/L	AST U/L	TP g/dL	ALB g/dL	TBIL mg/dL	ALP U/L	Glucose mg/dL	UREA mmol/L	CRE μmol/L	Ca mg/dL	P mg/dL	TCHO mg/dL	TG mmol/L	K mmol/L	Na mmol/L	Cl mmol/L	GLB g/dL	A/G	CK U/L
non-Tg	Mean	22.80	54.80	50.06	31.20	1.68	127.00	11.72	8.49	8.80	2.21	2.25	1.99	0.43	4.28	150.40	114.40	18.82	1.69	76.20
	SD	4.15	14.69	1.72	0.64	0.17	10.93	1.33	1.08	1.48	0.06	0.23	0.09	0.03	0.18	1.34	1.14	1.13	0.06	35.87
	N	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25	25.00
Tg	Mean	28.00	69.50	52.50	31.80	2.57	101.50	9.16	7.89	8.50	2.16	1.97	1.94	0.58	4.45	150.00	114.50	20.70	1.55	199.50
	SD	2.83	0.71	0.71	0.14	0.52	12.02	1.77	0.08	0.71	0.10	0.01	0.11	0.18	0.49	0.00	0.71	0.85	0.06	135.06
	N	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47	47.00
	Diff%	-22.81	-26.82	-4.87	-1.92	-52.32	20.08	21.86	7.17	3.41	2.35	12.52	2.71	-33.64	-3.97	0.27	-0.09	-9.99	8.31	-161.81

It is strongly recommend to consider the above differences in body weight, food consumption and a couple of clinical pathology parameters between Tg.rasH2 mice and non-Tg.rasH2 mice when estimating the dosages for the 26-week carcinogenicity studies in Tg.rasH2 mice using the 28-day DRF studies in non-Tg.rasH2 mice.

Acknowledgements

The authors would like to acknowledge the contributions made by the technical groups at WuXi AppTec.

References

1. Yamamoto S., Hayashi Y., Mitsumori K., Nomura T. (1997) Rapid Carcinogenicity Testing System with Transgenic Mice Harboring Human Prototype c-HRAS Gene. Laboratory Animal Science, Vol. 47, pp. 121-126 2. Paranjpe MG, Denton MD, Vidmar TJ, Elbekai RH. Relationship of body weight parameters with the incidence of common spontaneous tumors in Tg.rasH2 mice. Toxicol Pathol. 2014;42(7):1143-1152